Characterization of Zanubrutinib Safety/Tolerability Profile and Comparison With Ibrutinib Profile in Patients With B-Cell Malignancies: Post Hoc Analysis of a Large Clinical Trial Safety Database

Jennifer R. Brown,[1] Barbara Eichhorst,[2] Paolo Ghia,[3] Wojciech Jurczak,[4] Brad Kahl,[5] Nicole Lamanna,[6] Tadeusz Robak,[7] Mazyar Shadman,[8] Constantine Tam,[9] Lugui Qiu,[10] Aileen Cohen,[11] Meng Zhang,[11] Tommi Salmi,[12] Jason Paik,[11] Liping Wang,[11] Jun Zhang,[11] Han Ma,[11] Alessandra Tedeschi[13]

[1]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, USA; [2]Department of Internal Medicine, University of Cologne, Center for Integrated Oncology Aachen Bonn Köln Düsseldorf, Cologne, Germany; [3]Università Vita-Salute San Raffaele and IRCCS Ospedale San Raffaele, Milan, Italy; [4]Maria Sklodowska-Curie National Research Institute of Oncology, Krakow, Poland; [5]Siteman Cancer Center, Washington University School of Medicine, St. Louis, MO, USA; [6]Herbert Irving Comprehensive Cancer Center, Columbia University, New York, NY, USA; [7]Medical University of Lodz, Lodz, Poland; [8]Fred Hutchinson Cancer Research Center, University of Washington School of Medicine, Seattle, WA, USA; [9]Alfred Health, Melbourne, VIC, Australia; Monash University, Melbourne, VIC, Australia; [10]Department of Lymphoma and Myeloma, Blood Diseases Hospital and Institute of Hematology, Chinese Academy of Medical Sciences, Tianjin, China; [11]BeiGene USA, Inc, San Mateo, CA, USA; [12]BeiGene International GmbH, Basel, Switzerland; [13]ASST Grande Ospedale Metropolitano Niguarda, Milan, Italy

BACKGROUND

- Bruton tyrosine kinase inhibitors (BTKis) have revolutionized treatment of B-cell malignancies[1,2]
- Use of the first-generation BTKi ibrutinib may be limited by toxicities—including cardiovascular and gastrointestinal side effects and rash—attributed to off-target kinase inhibition[3-6]
- Zanubrutinib is a potent and selective next-generation BTKi that has been designed to improve tolerability by maximizing BTK occupancy and minimizing off-target effects[7]
- In a previous analysis of pooled data of 779 patients from 6 clinical trials, zanubrutinib was generally well tolerated and showed a consistent safety profile[8]
- Here we present an updated pooled analysis that characterizes the overall safety/tolerability profile of zanubrutinib in 1550 patients from 10 clinical studies, including 2 that compared zanubrutinib head-to-head with ibrutinib

METHODS

- Clinical trials (N=10) of zanubrutinib monotherapy included in these post hoc safety analyses are shown in **Table 1**
 - Patients with chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL), mantle cell lymphoma, marginal zone lymphoma, Waldenström macroglobulinemia, follicular lymphoma, and other B-cell malignancies were included
 - ASPEN (cohort 1) and ALPINE compared zanubrutinib head-to-head with ibrutinib

Table 1. Clinical Trials Included in the Pooled Analysis

Clinical trial	NCT number	Phase	Disease state	Zanubrutinib dose	Location	No. of patients treated with zanubrutinib (N=1550)
BGB-3111-1002	03189524	1	B-cell malignancies	160 mg BID 320 mg QD	China	44
BGB-3111-205	03206918	2	R/R CLL/SLL	160 mg BID	China	91
BGB-3111-206	03206970	2	R/R mantle cell lymphoma	160 mg BID	China	86
BGB-3111-210	03332173	2	Waldenström macroglobulinemia	160 mg BID	China	44
BGB-3111-AU-003	02343120	1/2	B-cell malignancies	160 mg BID 40 mg QD 80 mg QD 160 mg QD 320 mg QD	Global	373[a]
BGB-3111-214	03846427	2	Marginal zone lymphoma	160 mg BID	Global	68
BGB-3111-302 (ASPEN)[b]	03053440	3	Waldenström macroglobulinemia	160 mg BID	Global	129
BGB-3111-304 (SEQUOIA)	03336333	3	TN CLL/SLL	160 mg BID	Global	391
BGB-3111-305 (ALPINE)[b]	03734016	3	R/R CLL/SLL	160 mg BID	Global	324
BGB-3111-LTE1	04170283	3	B-cell malignancies	160 mg BID	Global	337[c]

[a] This value reflects the number of patients who received zanubrutinib 160 mg BID or 320 mg QD and were thus included in this analysis. [b] Compared zanubrutinib head-to-head with ibrutinib (420 mg QD). [c] The 337 patients in this long-term extension study previously participated in 1 of the other studies and were counted in the parent studies.

- Treatment-emergent adverse events (TEAEs) were summarized using MedDRA preferred terms
- Adverse events of special interest (AESIs) were defined using grouped terms
 - AESIs included anemia, atrial fibrillation/flutter, hemorrhage, hypertension, infections, neutropenia, second primary malignancies, and thrombocytopenia
- Incidence rates of TEAEs, exposure-adjusted incidence rates (EAIRs), and prevalence of AESIs over time were assessed
- EAIRs in units of persons per 100 person-months were calculated as follows:

 (Number of patients who experienced a TEAE of interest/total treatment exposure time in months for all patients) × 100
- An EAIR of 0.5 persons per 100 person-months indicates that if 1000 patients were each treated for a month, 5 would be estimated to experience the TEAE of interest

RESULTS

Patients and Exposure

- Data from 1550 patients treated with zanubrutinib and 422 patients treated with ibrutinib were included (**Table 2**)

Table 2. Demographics and Baseline Disease Characteristics

	All zanubrutinib (N=1550)	ASPEN/ALPINE[a] Zanubrutinib (n=425)	ASPEN/ALPINE[a] Ibrutinib (N=422)
Age, median (range), years	67.0 (20-95)	68.0 (35-90)	68.0 (35-90)
Sex, n (%)			
Male	1027 (66.3)	280 (65.9)	295 (69.9)
Female	523 (33.7)	145 (34.1)	127 (30.1)
Race, n (%)			
Asian	424 (27.4)	49 (11.5)	44 (10.4)
White	1032 (66.6)	348 (81.9)	357 (84.6)
Other	51 (3.3)	11 (2.6)	4 (0.9)
Not reported or missing	43 (2.8)	17 (4.0)	17 (4.0)
ECOG performance status, n (%)			
0	692 (44.6)	174 (40.9)	164 (38.9)
1	763 (49.2)	239 (56.2)	238 (56.4)
2	95 (6.1)	12 (2.8)	20 (4.7)
Diagnosis, n (%)			
CLL/SLL	938 (60.5)	324 (76.2)	324 (76.8)
Mantle cell lymphoma	140 (9.0)	0	0
Waldenström macroglobulinemia	249 (16.1)	101 (23.8)	98 (23.2)
Marginal zone lymphoma	93 (6.0)	0	0
Follicular lymphoma	59 (3.8)	0	0
Diffuse large B-cell lymphoma	45 (2.9)	0	0
Other[b]	26 (1.7)	0	0
Prior treatment status, n (%)			
Treatment naive	482 (31.1)	19 (4.5)[c]	18 (4.3)[c]
Relapsed/refractory	1068 (68.9)	406 (95.5)	404 (95.7)
No. of prior lines of therapy			
1	496 (32.0)	237 (55.8)	231 (54.7)
2	275 (17.7)	99 (23.3)	86 (20.4)
≥3	297 (19.2)	70 (16.5)	87 (20.6)

[a] Head-to-head randomized trials of zanubrutinib compared with ibrutinib in patients in ASPEN cohort 1 and ALPINE. Zanubrutinib includes patients with Waldenström macroglobulinemia from ASPEN cohort 1 (n=101) and patients with CLL from ALPINE (n=324); ibrutinib includes patients with Waldenström macroglobulinemia from ASPEN cohort 1 (n=98) and patients with CLL from ALPINE (n=324). [b] Includes patients with Richter's transformation (n=13), hairy cell leukemia (n=11), B-lineage lymphoma (n=1), and indolent lymphoma (n=1). [c] Patients with Waldenström macroglobulinemia from ASPEN cohort 1.

- Median exposure was 34.4 months among all patients who received zanubrutinib; 45.0% received zanubrutinib for ≥36 months (**Table 3**)
- The most frequent TEAEs leading to dose modifications were infection events
- In the pooled zanubrutinib population, deaths attributed to TEAEs occurred in 7.3% of patients; most (3.7%, n=57) were due to infections, including COVID-19–related TEAEs
- Cardiac-related TEAEs leading to death were lower with zanubrutinib than with ibrutinib (0.2% vs 1.7%) in the head-to-head trial populations

Table 3. Exposure, Dose Adjustments, and Deaths

	All zanubrutinib (N=1550)	ASPEN/ALPINE[a] Zanubrutinib (n=425)	ASPEN/ALPINE[a] Ibrutinib (N=422)
Duration of treatment, median (range), months	34.4 (0.1-90.0)	32.6 (0.4-68.7)	25.7 (0.1-59.3)
<12 months, n (%)	280 (18.1)	41 (9.6)	78 (18.5)
12 to <24 months, n (%)	235 (15.2)	96 (22.6)	106 (25.1)
24 to <36 months, n (%)	338 (21.8)	163 (38.4)	131 (31.0)
≥36 months, n (%)	697 (45.0)	125 (29.4)	107 (25.4)
Patients with ≥1 TEAE leading to, n (%)			
Dose reduction	156 (10.1)	59 (13.9)	81 (19.2)
Dose interruption	791 (51.0)	230 (54.1)	249 (59.0)
Treatment discontinuation	211 (13.6)	60 (14.1)	93 (22.0)
Treatment discontinuation due to cardiac TEAE	16 (1.0)	2 (0.5)	18 (4.3)
Deaths, n (%)			
Any TEAE	113 (7.3)	37 (8.7)	43 (10.2)
Cardiac TEAE	12 (0.8)	1 (0.2)	7 (1.7)

[a] Head-to-head randomized trials of zanubrutinib vs ibrutinib.

Safety and Tolerability of Zanubrutinib

- The most common nonhematologic TEAEs in patients who received zanubrutinib were upper respiratory tract infection and diarrhea (**Figure 1**)
 - No grade ≥3 TEAEs occurred in >10% of patients; the most common grade ≥3 TEAEs were pneumonia (8.4%) and hypertension (8.1%)
- Serious AEs occurred in 49.2% of patients who received zanubrutinib
 - The only serious AE in ≥5% of patients in the pooled zanubrutinib population was pneumonia (8.2%)

Figure 1. Most Common Nonhematologic TEAEs With Zanubrutinib[a]

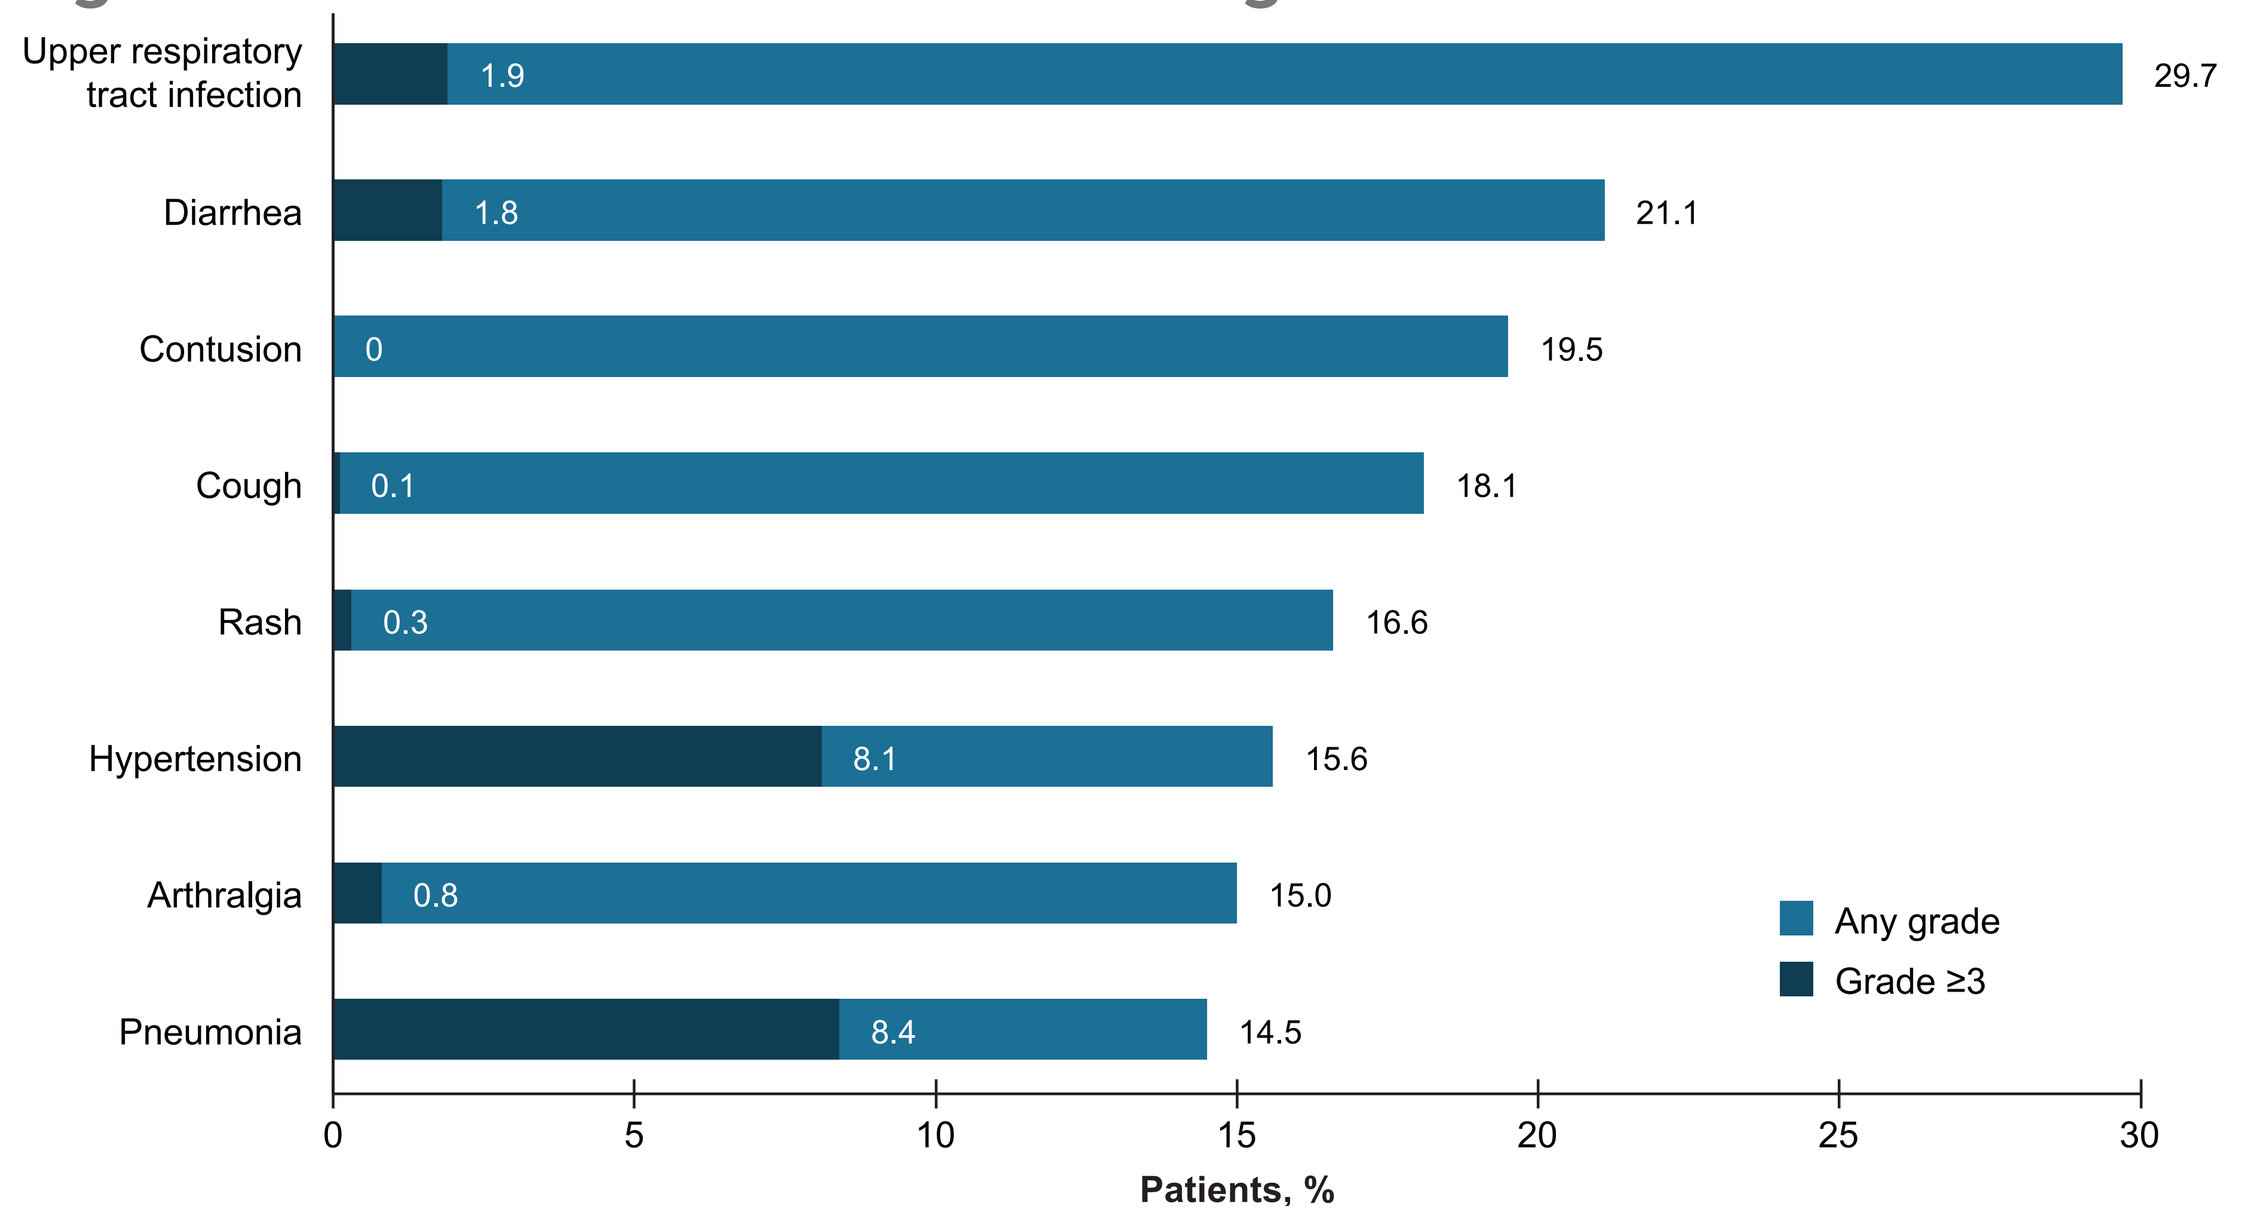
Upper respiratory tract infection 1.9 29.7
Diarrhea 1.8 21.1
Contusion 0 19.5
Cough 0.1 18.1
Rash 0.3 16.6
Hypertension 8.1 15.6
Arthralgia 0.8 15.0
Pneumonia 8.4 14.5
Any grade
Grade ≥3
0 5 10 15 20 25 30
Patients, %

[a] Any-grade TEAEs reported in ≥15% of patients; grade ≥3 TEAEs reported in ≥5% of patients in the pooled zanubrutinib population (N=1550).

AESIs

- EAIRs of AESIs, including infections, with zanubrutinib were numerically lower than with ibrutinib in head-to-head comparisons of the ASPEN/ALPINE study populations, except for neutropenia (**Figure 2**)
 - EAIRs of atrial fibrillation and infections were significantly lower with zanubrutinib vs ibrutinib (P<.0001 and P=.0098, respectively)
 - With the exception of ALPINE, EAIRs for hypertension with zanubrutinib were low (pooled EAIR <0.5 persons per 100 person-months) and consistent across studies
- The prevalence of AESIs tended to remain constant or decrease over time with zanubrutinib (**Figure 3**)
 - In head-to-head comparisons of the ASPEN/ALPINE study populations, hypertension tended to increase over time with ibrutinib, whereas it remained relatively stable with zanubrutinib
 - The prevalence of atrial fibrillation with zanubrutinib remained lower than with ibrutinib over time

Figure 2. EAIRs of AESIs

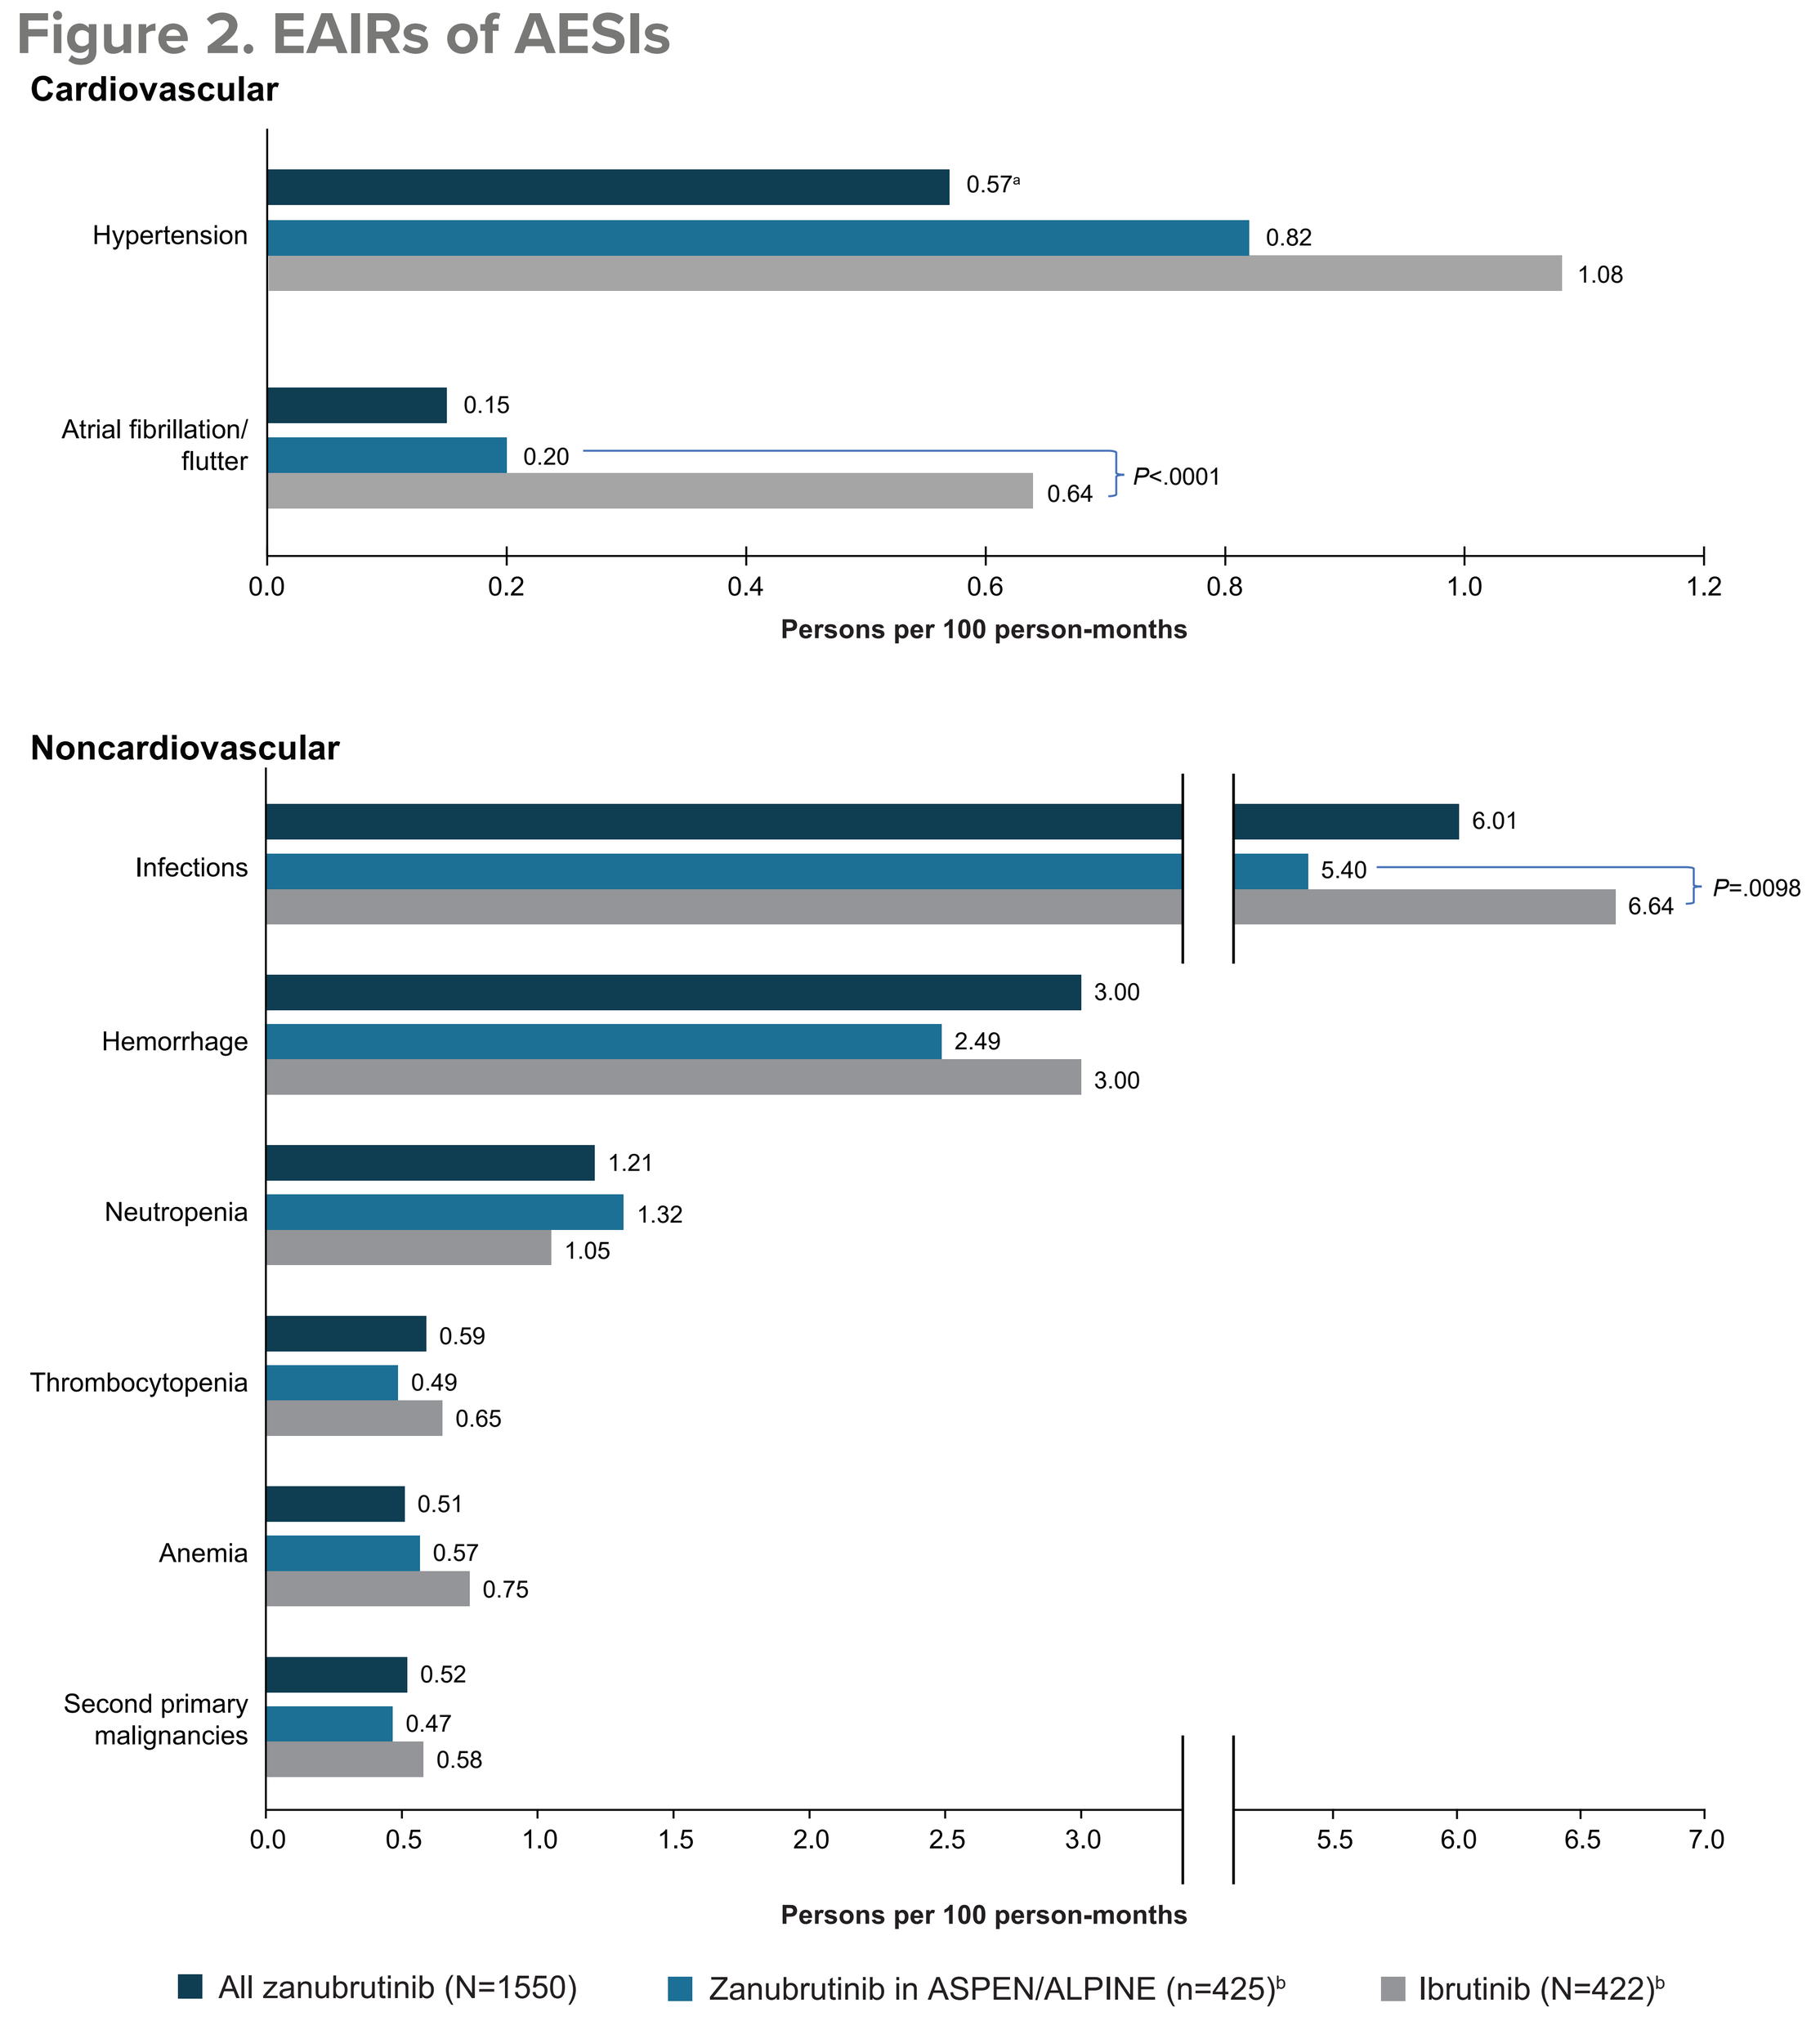
Cardiovascular
Hypertension 0.57[a] 0.82 1.08
Atrial fibrillation/flutter 0.15 0.20 0.64 P<.0001
0.0 0.2 0.4 0.6 0.8 1.0 1.2
Persons per 100 person-months
Noncardiovascular
Infections 6.01 5.40 6.64 P=.0098
Hemorrhage 3.00 2.49 3.00
Neutropenia 1.21 1.32 1.05
Thrombocytopenia 0.59 0.49 0.65
Anemia 0.51 0.57 0.75
Second primary malignancies 0.52 0.47 0.58
0.0 0.5 1.0 1.5 2.0 2.5 3.0 5.5 6.0 6.5 7.0
Persons per 100 person-months
All zanubrutinib (N=1550)
Zanubrutinib in ASPEN/ALPINE (n=425)[b]
Ibrutinib (N=422)[b]

[a] The EAIR for hypertension is 0.48 persons per 100 person-months in the subset of patients (n=1226) that excluded patients from ALPINE. [b] Zanubrutinib was compared head-to-head with ibrutinib in the phase 3 ASPEN (cohort 1) and ALPINE trials.

Figure 3. Prevalence of Selected AESIs Over Time

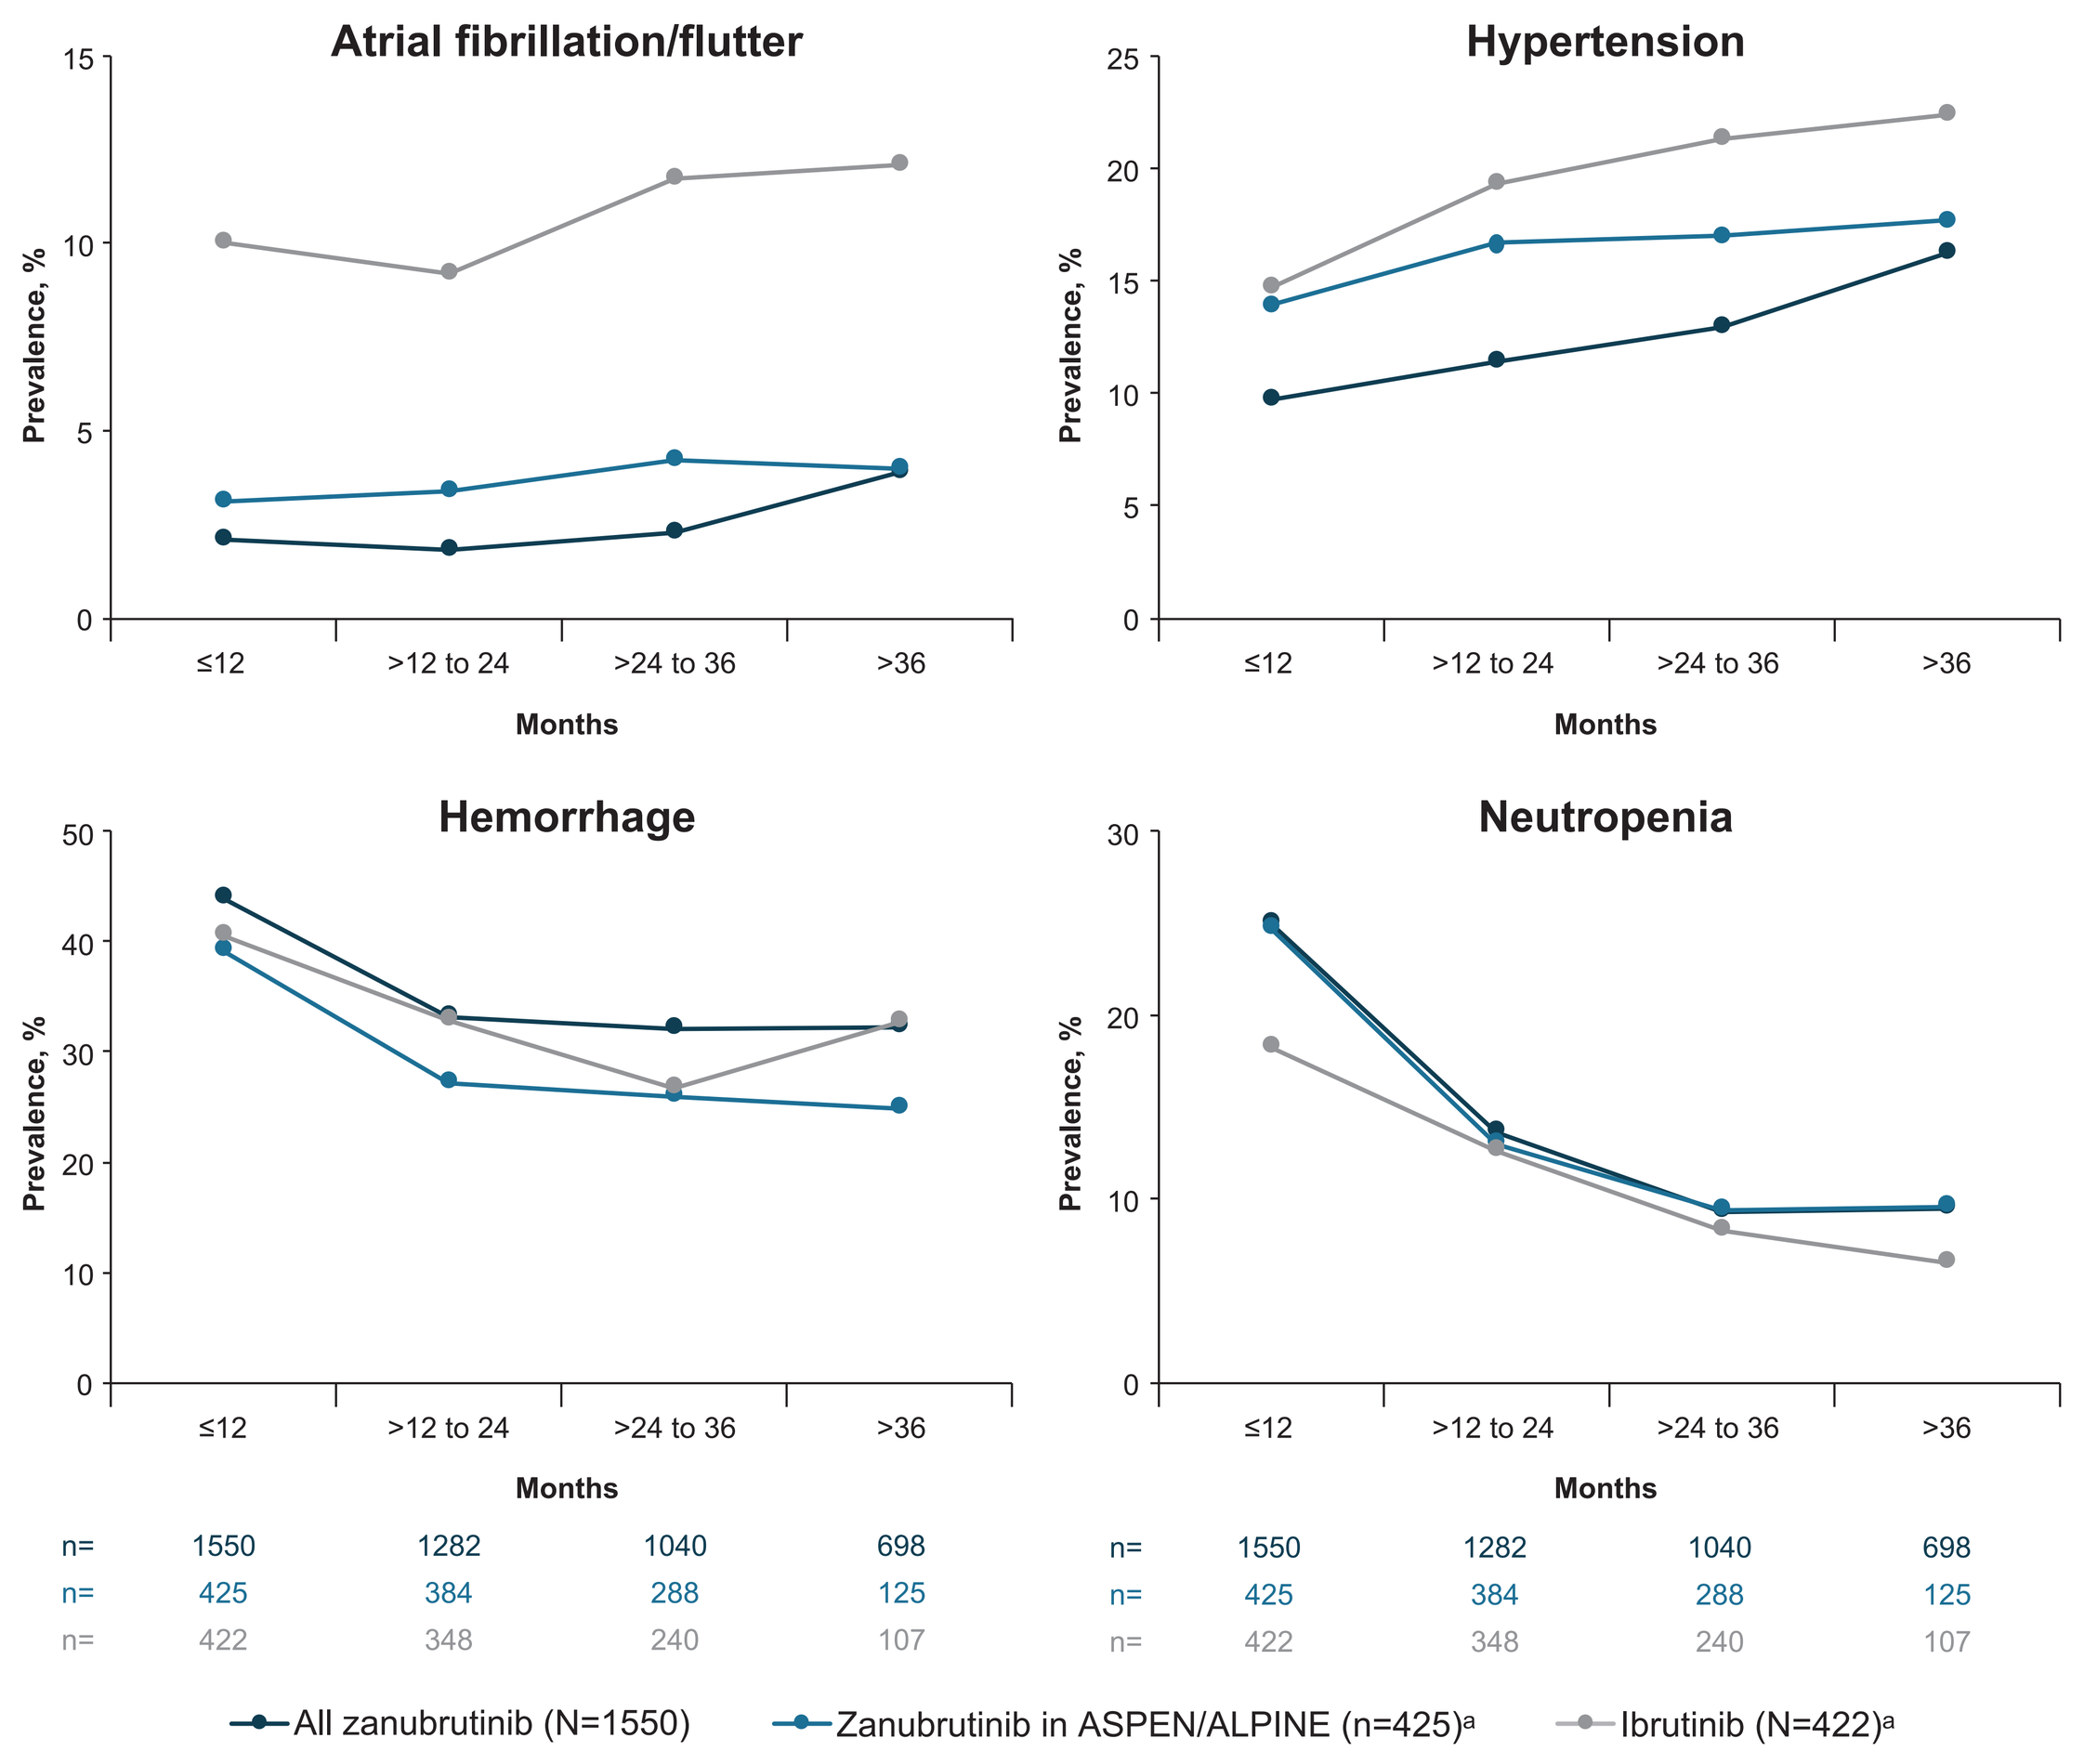
Atrial fibrillation/flutter
Hypertension
Hemorrhage
Neutropenia
Prevalence, %
Months
≤12 >12 to 24 >24 to 36 >36
n= 1550 1282 1040 698
n= 425 384 288 125
n= 422 348 240 107
All zanubrutinib (N=1550)
Zanubrutinib in ASPEN/ALPINE (n=425)[a]
Ibrutinib (N=422)[a]

[a] Zanubrutinib was compared head-to-head with ibrutinib in the phase 3 ASPEN (cohort 1) and ALPINE trials.

CONCLUSIONS

- These pooled safety analyses showed that zanubrutinib is well tolerated in patients with B-cell malignancies, with TEAEs (upper respiratory tract infection, diarrhea, contusion) that were generally mild to moderate in severity
- When comparing zanubrutinib vs ibrutinib in pooled head-to-head studies, rates of TEAEs or cardiac TEAEs leading to discontinuation were lower with zanubrutinib
 - In pooled head-to-head comparisons, rates of death due to cardiac events were lower with zanubrutinib than ibrutinib (0.2% vs 1.7%), which is comparable to findings in the phase 3 ALPINE study[9]
- EAIRs for AESIs, especially for hypertension and atrial fibrillation/flutter, were lower with zanubrutinib than ibrutinib in head-to-head comparisons
- In this pooled analysis, the EAIR for hypertension was largely influenced by data from ALPINE, which was an outlier from the rates observed in other studies of zanubrutinib[9,10]
 - Although hypertension rates were higher in ALPINE, cardiac events (eg, atrial fibrillation/flutter) were low and consistent with other zanubrutinib studies
- The prevalence of AESIs tended to decrease over time, without the emergence of new safety signals
- Due to the continuous dosing of BTKis in most B-cell malignancies, long-term tolerability and low treatment discontinuation rates with BTKis are important
- These analyses support zanubrutinib as an appropriate long-term treatment option for patients with B-cell malignancies

REFERENCES

1. Fowler N, Davis E. *Hematology Am Soc Hematol Educ Program.* 2013;2013(1):553-560.
2. Burger JA. *Cancer J.* 2019;25(6):386-393.
3. Coutre SE, et al. *Blood Adv.* 2019;3(12):1799-1807.
4. O'Brien S, et al. *Clin Lymphoma Myeloma Leuk.* 2018;18(10):648-657.
5. Jain P, et al. *J Clin Oncol.* 2021;40(2):202-212.
6. Buske C, et al. *J Clin Oncol.* 2022;40(1):52-62.
7. Guo Y, et al. *J Med Chem.* 2019;62(17):7923-7940.
8. Tam CS, et al. *Blood Adv.* 2022;6(4):1296-1308.
9. Brown JR, et al. *N Engl J Med.* 2023;338(4):319-332.
10. Ma H, et al. Presented at: British Society for Haematology Annual Meeting, April 23-25, 2023, Birmingham, UK. Abstract BSH23-OR19.

DISCLOSURES

JB: Consultancy: AbbVie, Acerta/AstraZeneca, BeiGene, Bristol Myers Squibb/Juno/Celgene, Catapult, Lilly, Genentech/Roche, Grifols Worldwide Operations, Hutchmed, iOnctura, Janssen, MEI Pharma, Pfizer, Pharmacyclics; Research funding: BeiGene, Gilead, iOnctura, Loxo/Lilly, MEI Pharma, SecuraBio, Sun, TG Therapeutics. **BE:** Research funding: Janssen, Gilead, Roche, AbbVie, BeiGene, AstraZeneca; Honoraria: Janssen, BeiGene, Roche, AbbVie, Novartis, Celgene, AstraZeneca; Speakers bureau: Roche, AbbVie, MSD, BeiGene, AstraZeneca; Travel, accommodations, expenses: BeiGene. **PG:** Honoraria: AbbVie, ArQule/MSD, AstraZeneca, BeiGene, Celgene/Juno/Bristol Myers Squibb, Janssen, Lilly/Loxo, MEI Pharma, Roche, Sanofi; Research funding: AbbVie, AstraZeneca, Janssen, Sunesis. **WJ:** Consultancy: Janssen, AstraZeneca, MEI Pharma, Lilly, Takeda, Roche, AbbVie, BeiGene; Research funding: AbbVie, Bayer, BeiGene, Celgene, Janssen, Roche, Takeda, TG Therapeutics, AstraZeneca, MEI Pharma, Lilly. **BK:** Research funding: BeiGene to Washington University School of Medicine (St Louis, MO, USA); Consulting fees: AbbVie, AstraZeneca, BeiGene, Janssen, Pharmacyclics. **NL:** Consultancy: AbbVie, AstraZeneca, BeiGene, Lilly/Loxo, Genentech, Janssen, Pharmacyclics; Research funding: AbbVie, AstraZeneca, BeiGene, Lilly/Loxo, Genentech, Octapharma, Oncternal, MingSight, TG Therapeutics. **TR:** Research funding: BeiGene, Octapharma, AstraZeneca, Janssen, Regeneron, GSK; Honoraria: AstraZeneca, BeiGene, Janssen, AbbVie, Octapharma, Regeneron, GSK; Travel, accommodations, expenses: AstraZeneca. **MS:** Consultancy: AbbVie, Genentech, AstraZeneca, Sound Biologics, Pharmacyclics, BeiGene, Bristol Myers Squibb, Morphosys/Incyte, TG Therapeutics, Innate Pharma, Kite Pharma, Adaptive Biotechnologies, Epizyme, Lilly, Adaptimmune, Mustang Bio, Regeneron, Merck, Fate Therapeutics, MEI Pharma, Atara Biotherapeutic; Research funding: Mustang Bio, Celgene, Bristol Myers Squibb, Pharmacyclics, Gilead, Genentech, AbbVie, TG Therapeutics, BeiGene, AstraZeneca, Sunesis, Atara Biotherapeutics, Genmab, Morphosys/Incyte, Vincerx. **CT:** Research funding: Janssen, AbbVie, BeiGene; Honoraria: Janssen, AbbVie, BeiGene, Loxo, AstraZeneca. **LQ:** Janssen, AstraZeneca, Takeda, Roche, AbbVie, BeiGene. **AC:** Employment: BeiGene; Equity holder: BeiGene; Travel, accommodations, expenses: BeiGene. **MZ:** Employment: BeiGene. **TS:** Employment: BeiGene Switzerland GmbH; Equity holder: BeiGene Ltd. **JP:** Employment: BeiGene. **LW:** Employment: BeiGene. **JZ:** Employment: BeiGene. **HM:** Employment: BeiGene. **AT:** Consultancy: BeiGene, AstraZeneca, AbbVie, Janssen; Honoraria: BeiGene, AstraZeneca, AbbVie, Janssen; Speakers bureau: BeiGene, AstraZeneca, AbbVie, Janssen; Travel, accommodations, expenses: BeiGene, AstraZeneca, AbbVie, Janssen.

CORRESPONDENCE

Jennifer R. Brown
Department of Medical Oncology, Dana-Farber Cancer Institute
Boston, MA, USA
jennifer_brown@dfci.harvard.edu

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeiGene Co, Ltd. Medical writing support was provided by Jackie Highland, PhD, and Lise Barnard, PhD, of Articulate Science, LLC, and supported by BeiGene.

Presented at the 2nd Bermuda Translational Summit on Hematologic Malignancies (DAVA) July 26-30, 2023, Bermuda
Previously presented at EHA 2023 Hybrid Congress; June 8-15, 2023; Frankfurt, Germany