UPDATED RESULTS OF THE ASPEN TRIAL FROM A COHORT OF PATIENTS WITH *MYD88* WILD-TYPE WALDENSTRÖM MACROGLOBULINEMIA

Abstract 2022

Meletios Dimopoulos, MD[1]; Ramon Garcia Sanz, MD, PhD[2]; Hui-Peng Lee, MBChB, FRACP, FRCPA[3]; Marek Trneny, MD, CSc[4]; Marzia Varettoni, MD[5]; Stephen Opat, MBBS, FRACP, FRCPA[6,7]; Shirley D'Sa, MD, MRCP, FRCPath[8]; Roger G. Owen, MD[9]; Gavin Cull, MB, BS, FRACP, FRCPA[10,11]; Stephen Mulligan, MBBS, PhD, FRACP, FRCPA[12]; Jaroslaw Czyz, MD, PhD[13,14]; Jorge Castillo, MD[15,16]; Marina Motta, MD[17]; Tanya Siddiqi, MD[18]; Mercedes Gironella Mesa, MD[19]; Miquel Granell Gorrochategui, MD[20]; Dipti Talaulikar, PhD, FRACP, FRCPA, MBBS[21]; Pier Luigi Zinzani, MD, PhD[22]; Elham Askari, MD[23]; Sebastian Grosicki, MD, PhD[24]; Albert Oriol, MD[25]; Simon Rule, MD[26]; Janusz Kloczko, MD[27]; Alessandra Tedeschi, MD[28]; Christian Buske, MD[29]; Veronique Leblond, MD[30]; Wai Y. Chan, PhD[31]; Jingjing Schneider, PhD[31]; Sunhee Ro, PhD[31]; Aileen Cohen, MD, PhD[31]; Jane Huang, MD[31]; and Constantine S. Tam, MBBS, MD, FRACP, FRCPA[32-35]

[1]National and Kapodistrian University of Athens, Athens, Greece; [2]Hospital Universitario de Salamanca, Salamanca, Spain; [3]Flinders Medical Centre, Adelaide, South Australia, Australia; [4]Vseobecna Fakultni Nemocnice v Praze, Prague, Czech Republic; [5]Fondazione IRCCS Policlinico San Matteo, Pavia, Italy; [6]Monash Health, Clayton, Victoria, Australia; [7]Monash University, Clayton, Victoria, Australia; [8]University College London Hospital Foundation Trust, London, United Kingdom; [9]St James University Hospital, Leeds, United Kingdom; [10]Sir Charles Gairdner Hospital, Perth, Western Australia, Australia; [11]University of Western Australia, Perth, Western Australia, Australia; [12]Royal North Shore Hospital, Sydney, New South Wales, Australia; [13]Szpital Uniwersytecki nr 2 im dr. Jana Biziela, Kujawsko-pomorskie, Bydgoszcz, Poland; [14]Department of Hematology, Collegium Medicum in Bydgoszcz, Nicolaus Copernicus University in Toruń, Bydgoszcz, Poland; [15]Dana-Farber Cancer Institute, Boston, MA, USA; [16]Harvard Medical School, Boston, MA, USA; [17]AO Spedali Civili di Brescia, Lombardia, Italy; [18]City of Hope National Medical Center, Duarte, CA, USA; [19]Hospital Universitario Vall d'Hebrón, Barcelona, Spain; [20]Hospital de La Santa Creu i Sant Pau, Barcelona, Spain; [21]Australian National University, Canberra, ACT, Australia; [22]Institute of Hematology "Seràgnoli" University of Bologna, Bologna, Italy; [23]Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [24]Department of Hematology and Cancer Prevention, Health Sciences Faculty, Medical University of Silesia, Katowice, Poland; [25]Institut Català d'Oncologia-Hospital Universitari Germans Trias i Pujol, Barcelona, Spain; [26]Plymouth Hospitals NHS Trust, Derriford Hospital, Devon, United Kingdom; [27]Uniwersytecki Szpital Kliniczny w Bialymstoku, Podlaskie, Poland; [28]ASST Grande Ospedale Metropolitano Niguarda, Milan, Italy; [29]CCC Ulm - Universitätsklinikum Ulm, Ulm, Baden-Württemberg, Germany; [30]Sorbonne University, Pitié Salpêtrière Hospital, Paris, France; [31]BeiGene USA, Inc., San Mateo, CA, USA; [32]Peter MacCallum Cancer Centre, Melbourne, Victoria, Australia; [33]St Vincent's Hospital, Fitzroy, Victoria, Australia; [34]University of Melbourne, Parkville, Victoria, Australia; and [35]Royal Melbourne Hospital, Parkville, Victoria, Australia

INTRODUCTION

- Bruton tyrosine kinase (BTK) plays a critical role in B-cell receptor signaling, which mediates B-cell proliferation, migration, and adhesion[1-3]
- First-generation BTK inhibitor ibrutinib has shown activity in Waldenström macroglobulinemia (WM) and has become a standard of care[4]
 - However, lower response rates,[5] no major responses,[5,6] and shorter survival[7] have been reported in patients (pts) who lack *MYD88^{L265P}* or other activating mutations (*MYD88^{WT}*)
- Zanubrutinib is a next-generation BTK inhibitor designed to maximize BTK occupancy and minimize off-target inhibition of TEC- and EGFR-family kinases (**Figure 1**)
 - **Potent, selective, and irreversible**[8]
 - **Equipotent against BTK compared with ibrutinib**; higher selectivity vs EGFR, ITK, JAK3, HER2 and TEC[9]
 - **Advantageous pharmacokinetic/pharmacodynamic properties**: complete and sustained BTK occupancy in peripheral blood mononuclear cells and lymph nodes[8]
 - **Favorable drug-drug interaction properties**: can be co-administered with strong/moderate CYP3A inhibitors at a reduced dose, proton pump inhibitors, acid-reducing agents, and anti-thrombotic agents[10,11]
 - **Approved for treatment of pts with relapsed/refractory (R/R) mantle cell lymphoma in the United States 11/2019**[12]

Zanubrutinib (BGB-3111)

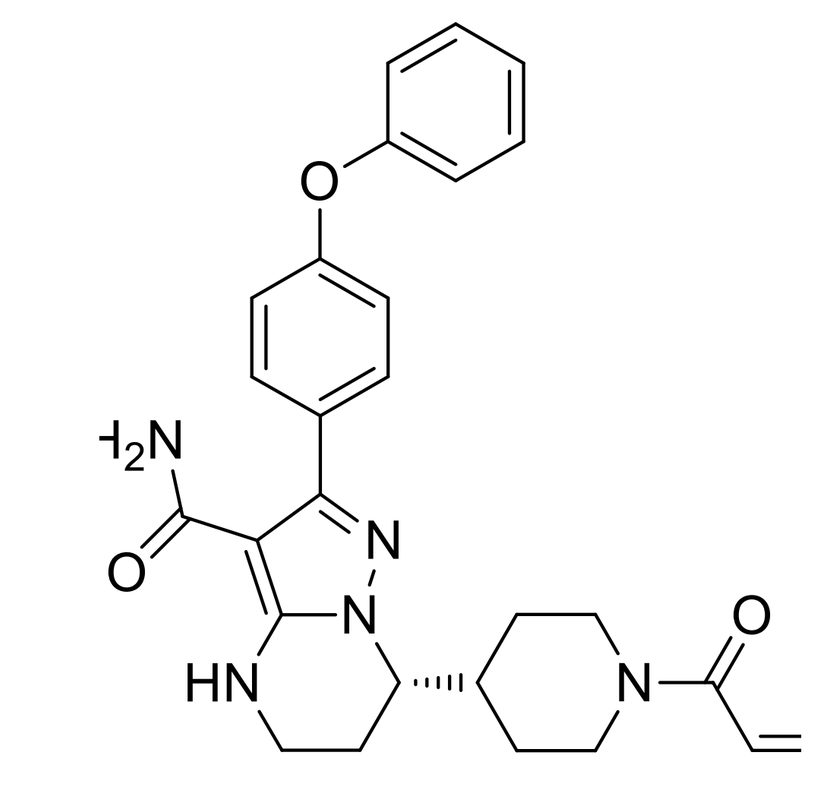

Figure 1a. Zanubrutinib: A Potent and Selective BTK Inhibitor[8,9]

	Targets	Assays	Zanubrutinib IC_{50} (nM)	Ibrutinib IC_{50} (nM)	Ratio (Zanubrutinib:Ibrutinib)
ON TARGET	BTK	BTK-pY223 Cellular Assay	1.8	3.5	0.5
		Rec-1 Proliferation	0.36	0.34	1.1
		BTK Occupation Cellular Assay	2.2	2.3	1
		BTK Biochemical Assay	0.22	0.2	1.1
OFF TARGET	EGFR	p-EGFR HTRF Cellular Assay	606	101	6
		A431 Proliferation	3210	323	9.9
	ITK	ITK Occupancy Cellular Assay	606	189	17
		p-PLCγ1 Cellular Assay	3433	77	45
		IL-2 Production Cellular Assay	2536	260	9.8
		ITK Biochemical Assay	30	0.9	33
	JAK3	JAK3 Biochemical Assay	200	3.9	51
	HER2	HER2 Biochemical Assay	661	9.4	70
	TEC	TEC Biochemical Assay	1.9	0.8	2.4

Figure 1b. Complete, Sustained BTK Occupancy With bid or qd Dosing[8,9]

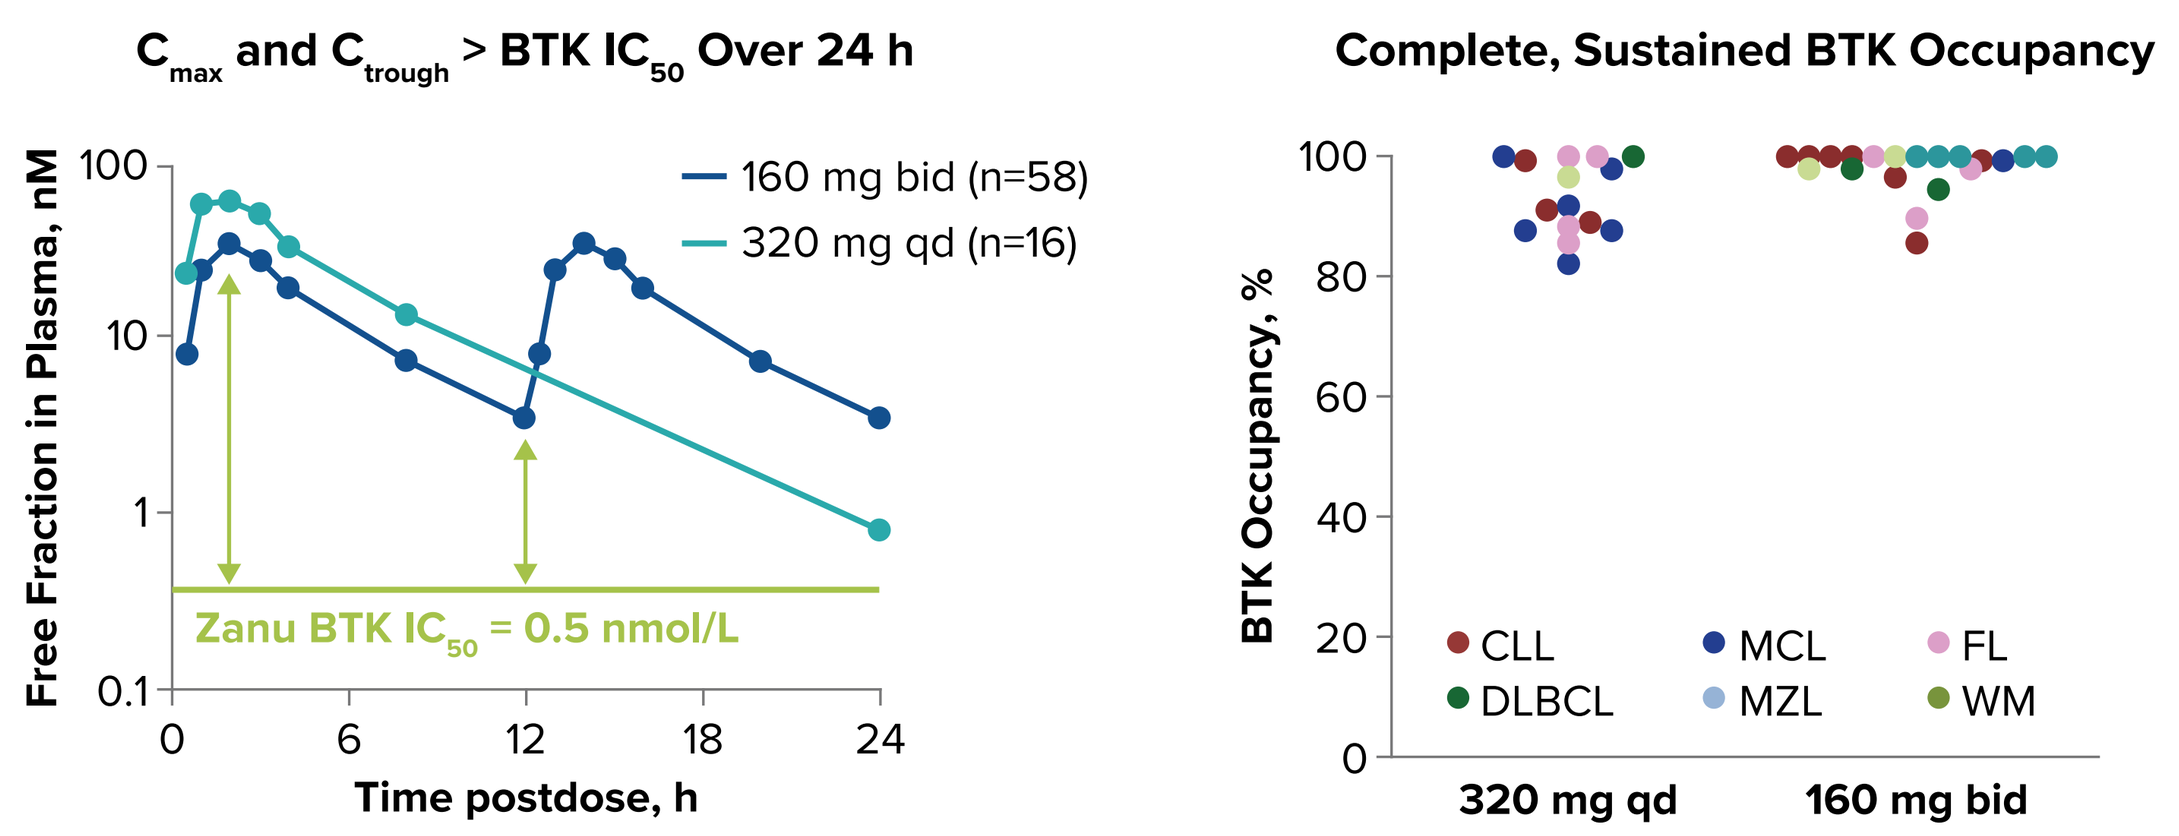

C_{max} and C_{trough} > BTK IC_{50} Over 24 h
Free Fraction in Plasma, nM
100
10
1
0.1
160 mg bid (n=58)
320 mg qd (n=16)
Zanu BTK IC_{50} = 0.5 nmol/L
0
6
12
18
24
Time postdose, h
Complete, Sustained BTK Occupancy
BTK Occupancy, %
CLL
DLBCL
MCL
MZL
FL
WM
320 mg qd
160 mg bid

bid, twice daily; BTK, Bruton tyrosine kinase; CLL, chronic lymphocytic leukemia; C_{max}, maximum concentration; C_{trough}, trough concentration; DLBCL, diffuse large B-cell lymphoma; EGFR, epidermal growth factor receptor; FL, follicular lymphoma; HER2, human epidermal growth factor receptor 2; HTRF, homogeneous time resolved fluorescence; IC_{50}, half maximal inhibitory concentration; ITK, IL-2–inducible T-cell kinase; JAK3, Janus-associated kinase 3; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; PLC, phospholipase C; qd: once daily; WM, Waldenström macroglobulinemia; Zanu, zanubrutinib.

OBJECTIVE

- To assess the safety and efficacy of zanubrutinib in WM pts with *MYD88^{WT}* from an exploratory cohort of the ongoing phase 3 study of zanubrutinib vs ibrutinib in pts with WM (ASPEN; NCT03053440)

METHODS

- ASPEN is an open-label, multicenter, randomized, phase 3 study of zanubrutinib vs ibrutinib in pts with WM (**Figure 2**)

Figure 2. Phase 3 ASPEN Trial Design

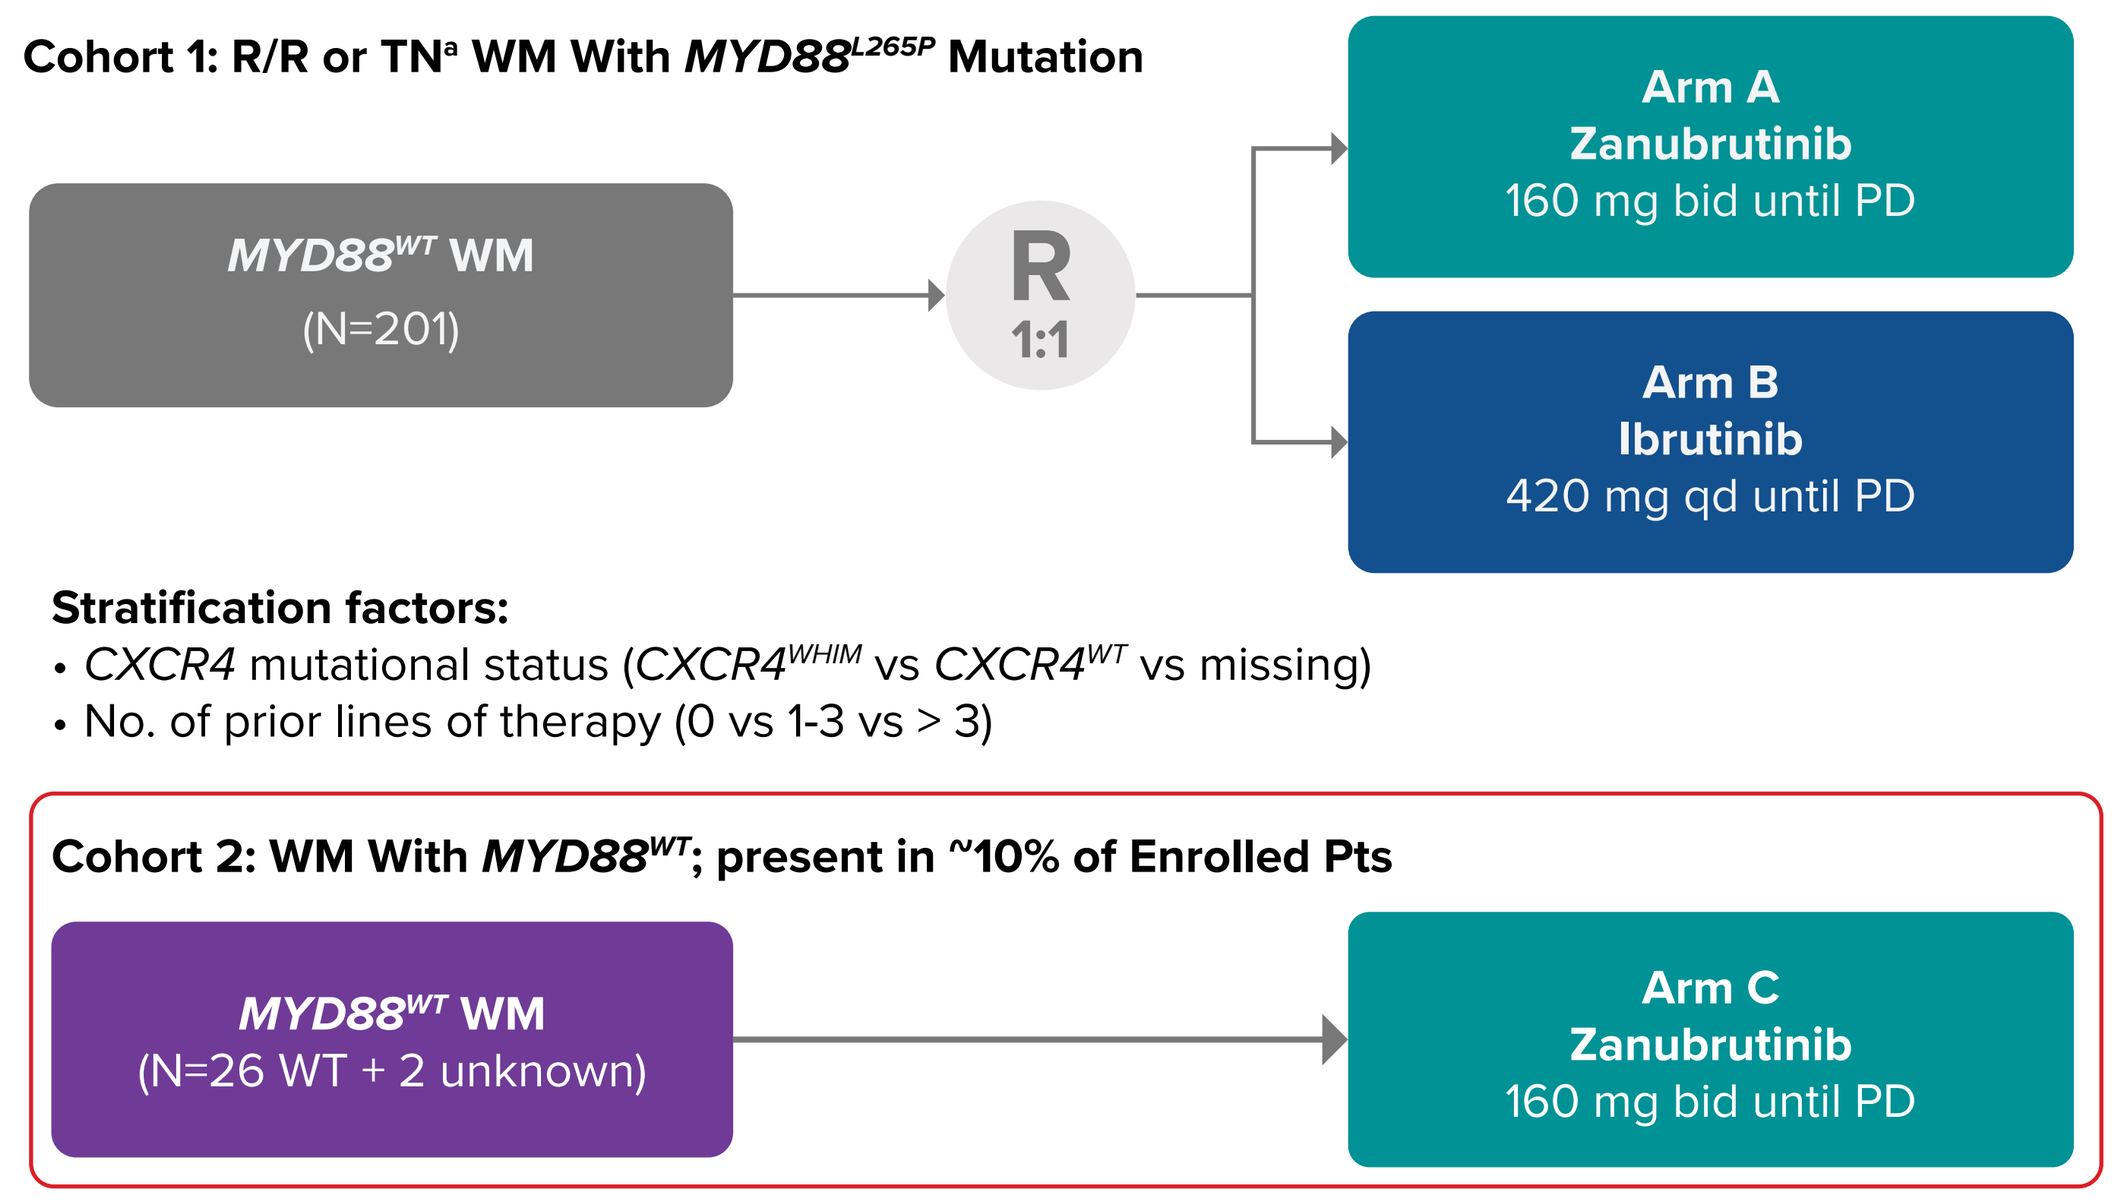

Cohort 1: R/R or TN[a] WM With *MYD88^{L265P}* Mutation
MYD88^{WT} WM
(N=201)
R
1:1
Arm A
Zanubrutinib
160 mg bid until PD
Arm B
Ibrutinib
420 mg qd until PD
Stratification factors:
• *CXCR4* mutational status (*CXCR4^{WHIM}* vs *CXCR4^{WT}* vs missing)
• No. of prior lines of therapy (0 vs 1-3 vs > 3)
Cohort 2: WM With *MYD88^{WT}*; present in ~10% of Enrolled Pts
MYD88^{WT} WM
(N=26 WT + 2 unknown)
Arm C
Zanubrutinib
160 mg bid until PD

EUDRACT 2016-002980-33; NCT03053440.
bid, twice daily; PD, progressive disease; pt, patient; qd, once daily; R/R, relapsed/refractory; TN, treatment-naïve.
[a]TN must be unsuitable for standard chemoimmunotherapy.

Eligibility

- Clinical and definitive histologic diagnosis of WM, with measurable disease (serum IgM >0.5 g/dL), and meeting ≥1 criterion for treatment according to consensus panel criteria from the Seventh International Workshop on WM[13]
- If treatment naïve, must be considered by treating physician unsuitable for standard chemoimmunotherapy regimens
- Eastern Cooperative Oncology Group performance status 0-2
- Absolute neutrophil count ≥750/µL, platelets ≥50000/µL (independent of growth factor/transfusions)
- Adequate renal, hepatic, and coagulation function
- No significant cardiac disease, active central nervous system involvement, or prior BTK inhibitors

Cohort Assignment

- Bone marrow *MYD88* and *CXCR4* mutations were assessed centrally at study entry (NeoGenomics Laboratory, Aliso Viejo, CA, USA)[14]
 - The *MYD88* mutation assay used detects all mutations in the region encompassing amino acid Ala^{260}-Pro^{278}, which includes the predominant mutation in WM, *MYD88^{L265P}* (**Figure 3**)
- Pts were assigned to cohort 1 (*MYD88* mutated; randomized) or exploratory cohort 2 (*MYD88^{WT}* or *MYD88* unknown, nonrandomized) based on the central laboratory *MYD88* mutation assay results

Figure 3. *MYD88*-Activating Mutations in Pts With WM

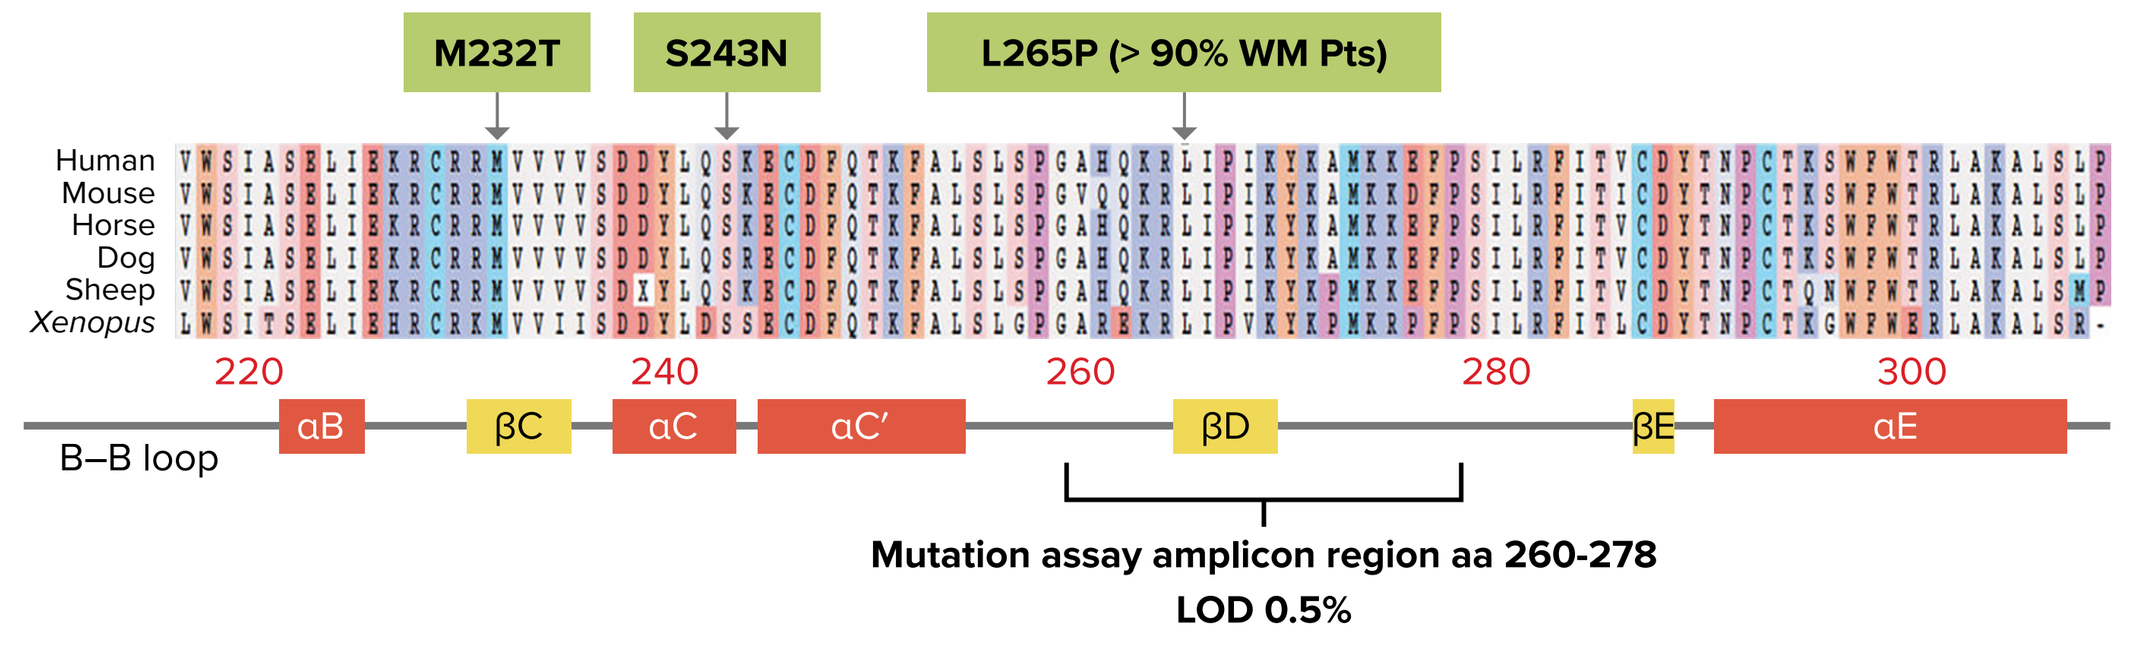

M232T
S243N
L265P (> 90% WM Pts)
Human
Mouse
Horse
Dog
Sheep
Xenopus
220
240
260
280
300
B–B loop
αB
βC
αC
αC′
βD
βE
αE
Mutation assay amplicon region aa 260-278
LOD 0.5%

- Detection in the *MYD88* amplicon (Ala^{260}-Pro^{278}) by the NeoGenomics LDT assay includes a wild-type-allele–blocking approach (limit of detection [LOD], 0.5%)[14] versus standard polymerase chain reaction/bidirectional Sanger sequencing assay used to detect *CXCR4* mutations (LOD, 10%-15%)
- For *MYD88^{WT}* pts with available samples (12 of 26), *MYD88* mutations were also evaluated by next-generation sequencing (200×; LOD, 5%); no other activating mutations were detected

Adapted from Treon et al[6] and Ngo et al[15]
LOD, limit of detection; pt, patient; WM, Waldenström macroglobulinemia.

Exploratory End Points for Cohort 2

- Responses were assessed monthly by immunoglobulin M (IgM) with extramedullary disease assessment every 3 months, according to response criteria in the National Comprehensive Cancer Network WM guidelines[16] and modified Owen criteria[17] as assessed by the independent review committee
- Efficacy: response rates (overall and major response rate), duration of response, progression-free survival, and overall survival; safety assessed according to National Cancer Institute Common Terminology Criteria for Adverse Events version 4.03

RESULTS

- In total, 28 pts (n=26 *MYD88^{WT}*; n=2 *MYD88* mutation status unknown) were enrolled into cohort 2
- The safety analysis set includes all 28 pts, and the efficacy analysis set includes 26 *MYD88^{WT}* pts, with a median follow-up of 17.9 months (range, 2.3-27.8; **Figure 4** and **Table 1**)

Figure 4. Disposition of Pts in Cohort 2

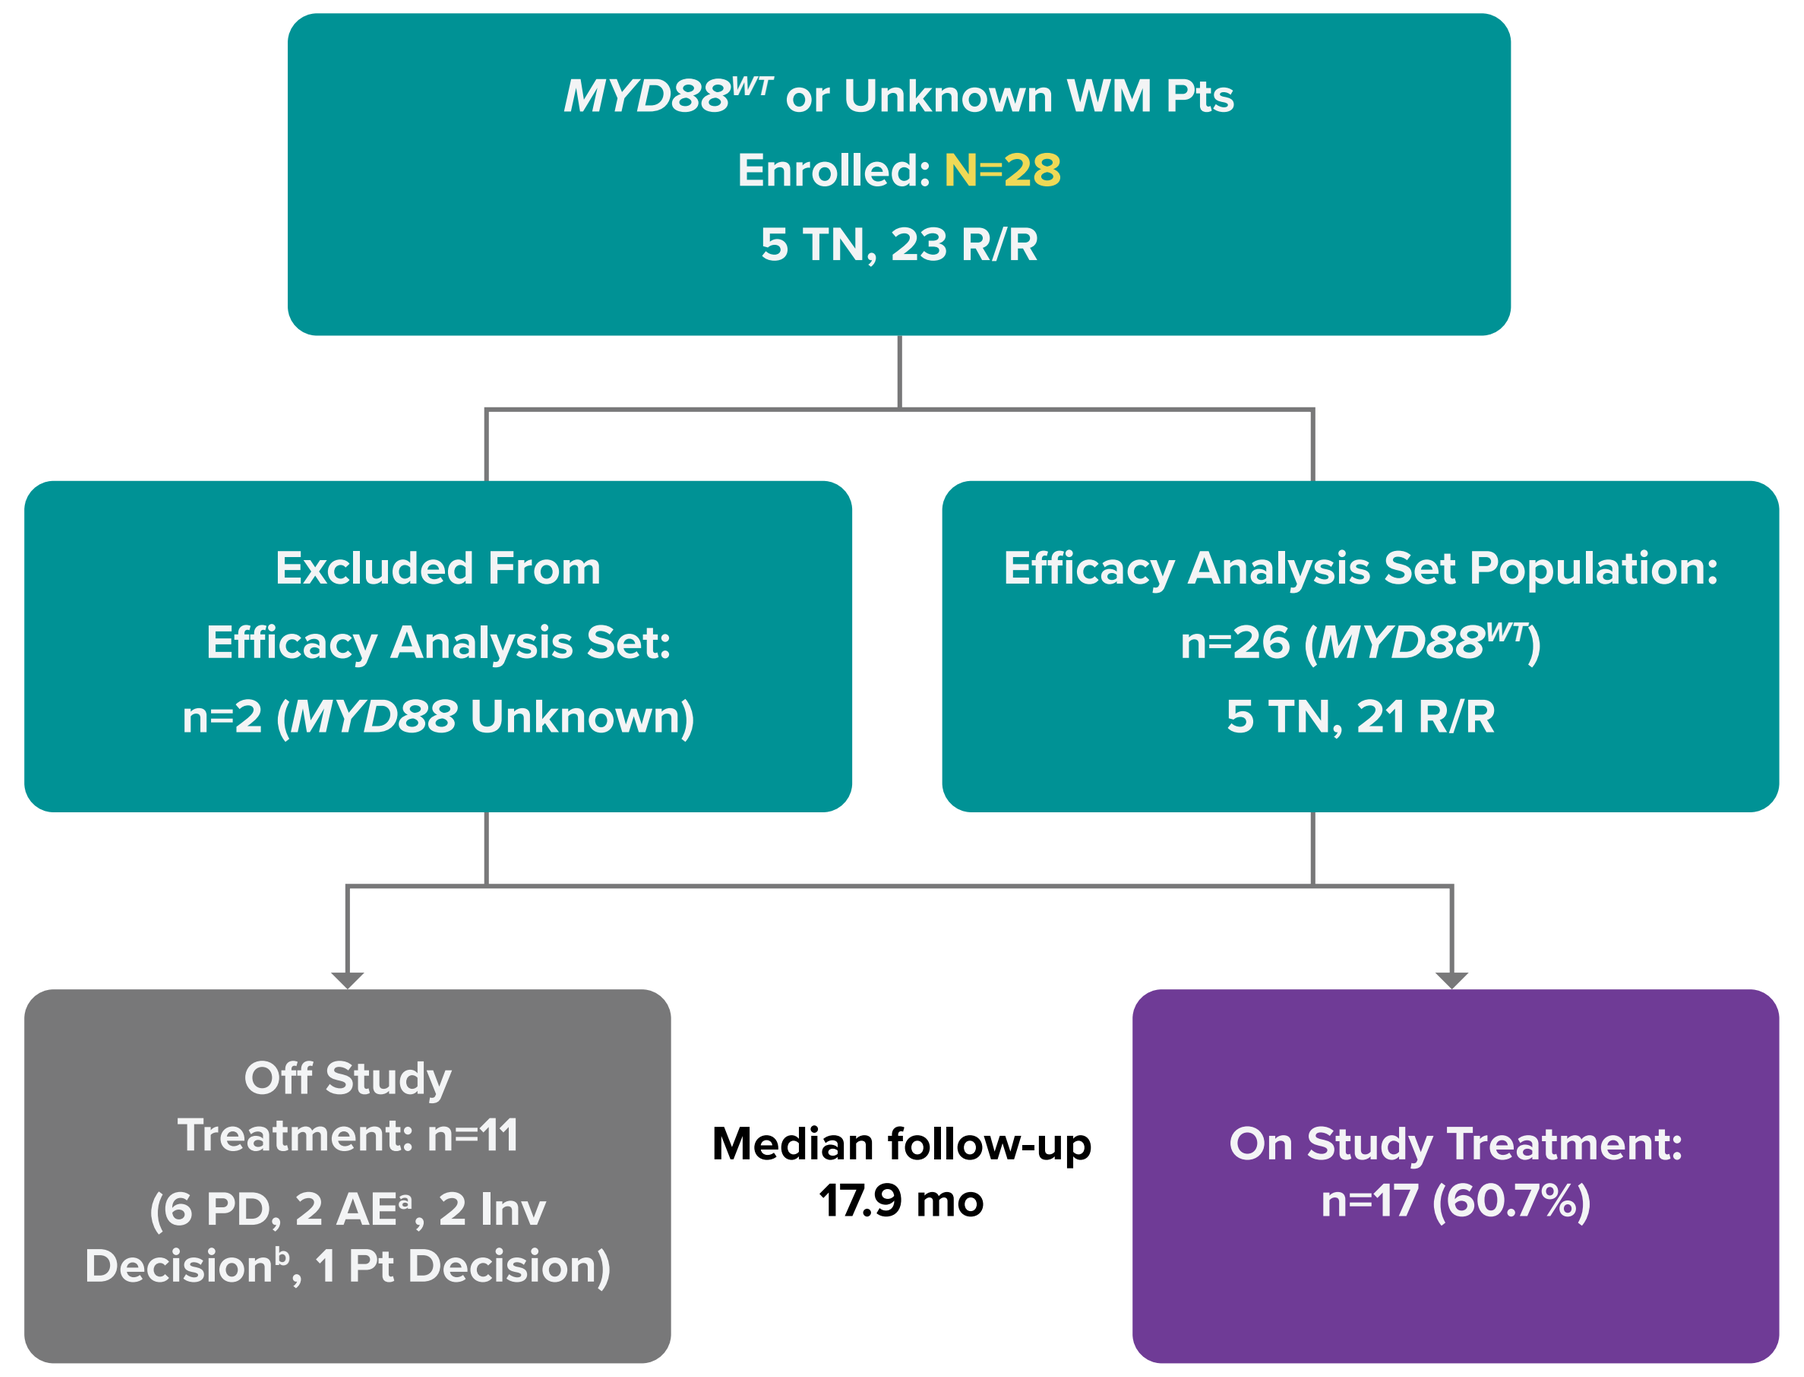

MYD88^{WT} or Unknown WM Pts
Enrolled: N=28
5 TN, 23 R/R
Excluded From
Efficacy Analysis Set:
n=2 (*MYD88* Unknown)
Efficacy Analysis Set Population:
n=26 (*MYD88^{WT}*)
5 TN, 21 R/R
Off Study
Treatment: n=11
(6 PD, 2 AE[a], 2 Inv Decision[b], 1 Pt Decision)
Median follow-up
17.9 mo
On Study Treatment:
n=17 (60.7%)

Data cutoff date: 31 Aug, 2019.
AE, adverse event; F/U, follow-up; Inv, investigator; PD, progressive disease; pt, patient.
[a]Grade 4 subdural hemorrhage; grade 3 diarrhea.
[b]Investigator decided no further treatment needed (n=1); pt discharged to hospice for palliative care (n=1).

Table 1. Pt and Disease Characteristics

Characteristic	Total (N=28)
Age, median (range), y	70.1 (39-87)
>65 years, n (%)	19 (67.9)
>75 years, n (%)	12 (42.9)
Sex, n (%)	
Men	14 (50)
Women	14 (50)
IPSSWM, n (%)	
Low	5 (17.9)
Intermediate	11 (39.3)
High	12 (42.9)
Prior treatment status	
Treatment-naïve, n (%)	5 (17.9)
R/R, n (%)	23 (82.1)
No. of prior therapies for R/R pts, median (range)	1 (1-5)
Extramedullary disease present at baseline by IRC, n (%)	21 (75.0)
Genotype, n (%)	
MYD88^{WT}/CXCR4^{WT}	23 (82.1)
MYD88^{WT}/CXCR4^{WHIM}	1 (3.6)
MYD88^{WT}/CXCR4 unknown	2 (7.1)
MYD88 unknown/*CXCR4* unknown	2 (7.1)
Hemoglobin ≤110 g/L, n(%)	15 (53.6)

IPSSWM, International Prognostic Scoring System Waldenström macroglobulinemia; pt, patient; R/R, relapsed/refractory; IRC, independent review committee.

Safety (N=28)

Table 2. AE Overview

Treatment Emergent AE	n (%)
Pts with ≥1 AE grade ≥3	18 (64.3)
Pts with ≥1 serious AE	11 (39.3)
AE leading to death	0
AE leading to treatment discontinuation	2[a] (7.1)
AE leading to dose reduction	2[b] (7.1)

AE, adverse event; pt, patient; SAE, serious adverse event.
[a]Grade 4 subdural hemorrhage (related) and grade 3 diarrhea (related).
[b]Grade 3 pneumonitis resolved and followed by grade 2 pneumonia (n=1); grade 1 diarrhea (n=1).

- No treatment-emergent AEs leading to death (**Table 2**)
- 2 Pts discontinued because of AEs
 - Grade 4 subdural hemorrhage
 - Grade 3 diarrhea
- Major hemorrhage occurred in 2 pts (**Table 3**)
 - Gastric ulcer haemorrhage
 - Periorbital hematoma, subdural hematoma, and subdural hemorrhage; treatment was permanently discontinued per protocol
- Atrial fibrillation/flutter occurred in 1 pt (grade 1)
- Most common adverse events (AEs; in >15% pts) were diarrhea, anemia, contusion, pyrexia, upper respiratory tract infection, respiratory tract infection, and cough (**Figure 5**)

Table 3. AE Categories of Interest (BTKi Class AEs)

AE Categories (Pooled Terms), n (%)	All Grade	Grade ≥3
Atrial fibrillation/flutter	1 (3.6)	0
Diarrhea (PT)	8 (28.6)	2 (7.1)
Hemorrhage	11 (39.3)	2 (7.1)
Major hemorrhage[a]	2 (7.1)	2 (7.1)
Hypertension	3 (10.7)	3 (10.7)
Neutropenia[b]	5 (17.9)	3 (10.7)
Infection	21 (75.0)	8 (28.6)
Second malignancy[c]	4 (14.3)	0

No tumor lysis syndrome or opportunistic infection was reported.
AE, adverse event; BTKi, Bruton tyrosine kinase inhibitor; PT, preferred term.
[a]Defined as any grade ≥3 hemorrhage or any grade central nervous system hemorrhage: gastric ulcer hemorrhage; and 1 patient had periorbital hematoma, subdural hematoma, and subdural hemorrhage.
[b]Including PT terms of neutropenia, neutrophil count decreased, febrile neutropenia, agranulocytosis, neutropenic infection, and neutropenic sepsis.
[c]Basal cell carcinoma (n=3) and Queyrat erythroplasia (n=1).

Figure 5. Common AEs (Any Grade >10% or Grade ≥3 in >1 Pt), Regardless of Causality

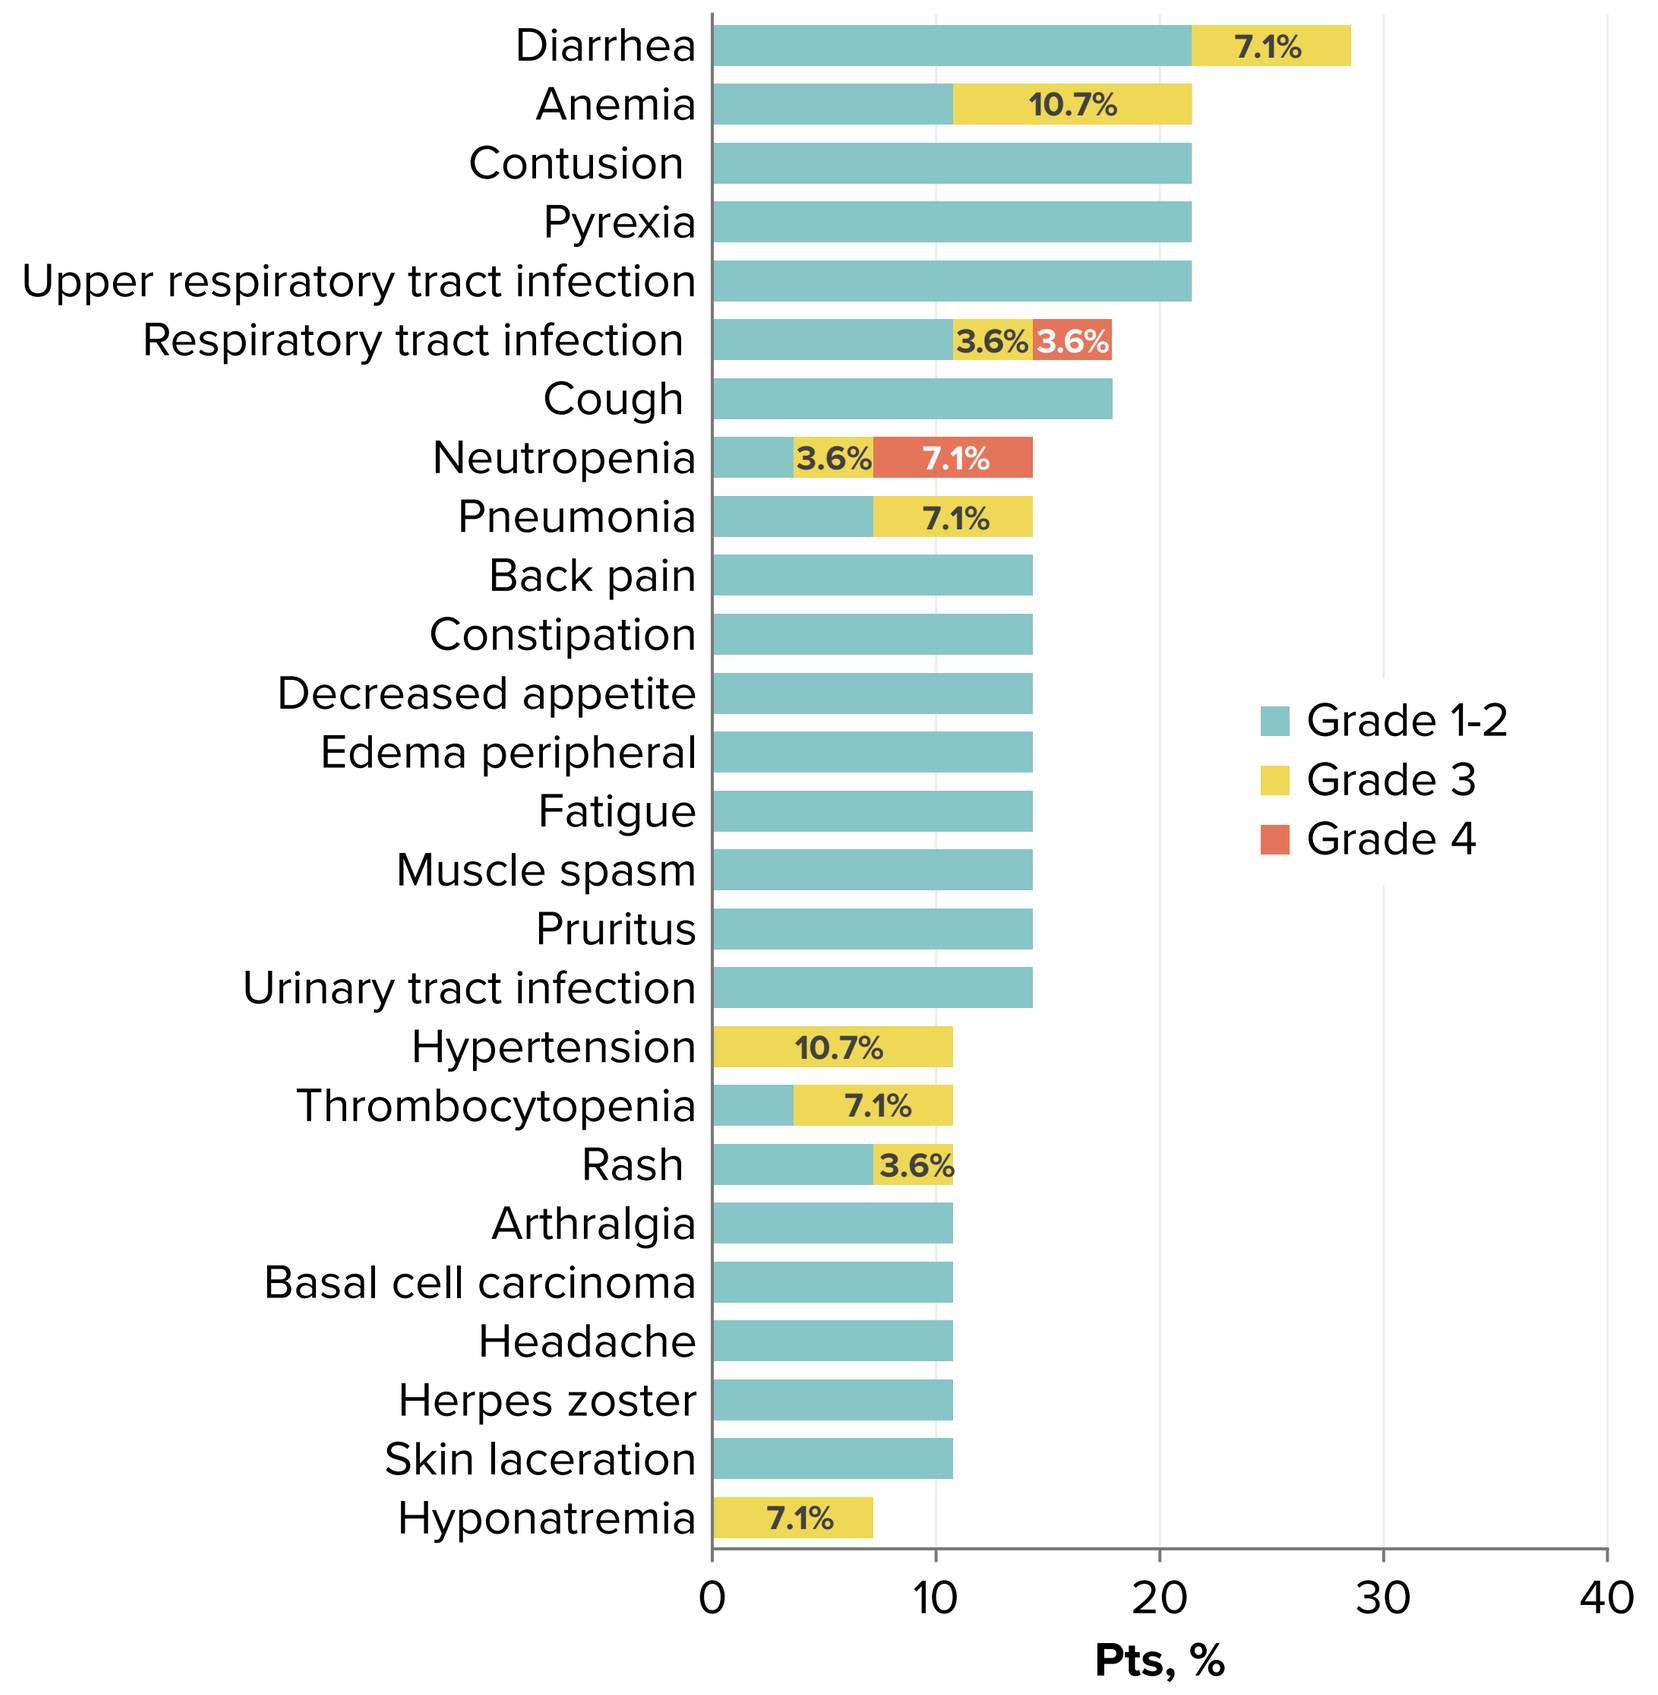

Diarrhea
Anemia
Contusion
Pyrexia
Upper respiratory tract infection
Respiratory tract infection
Cough
Neutropenia
Pneumonia
Back pain
Constipation
Decreased appetite
Edema peripheral
Fatigue
Muscle spasm
Pruritus
Urinary tract infection
Hypertension
Thrombocytopenia
Rash
Arthralgia
Basal cell carcinoma
Headache
Herpes zoster
Skin laceration
Hyponatremia
7.1%
10.7%
3.6%
3.6%
3.6%
7.1%
7.1%
10.7%
7.1%
3.6%
7.1%
Grade 1-2
Grade 3
Grade 4
0
10
20
30
40
Pts, %

AE, adverse event; pt, patient.

Efficacy (n=26)

- Major response rate of 50.0% including 26.9% with VGPR (**Figure 6**)
- Median time to first major response (partial response or better, requiring reduction in extramedullary disease if present at baseline) was 2.9 mo (range, 1.9-16.1; **Figure 7**)
- IgM complete response (requiring normal IgM and immunofixation negative) was achieved in 1 pt
- Median progression-free and overall survival were not yet reached (**Figure 8**)

Figure 6. Best Responses by IRC in Patients With *MYD88^{WT}* WM

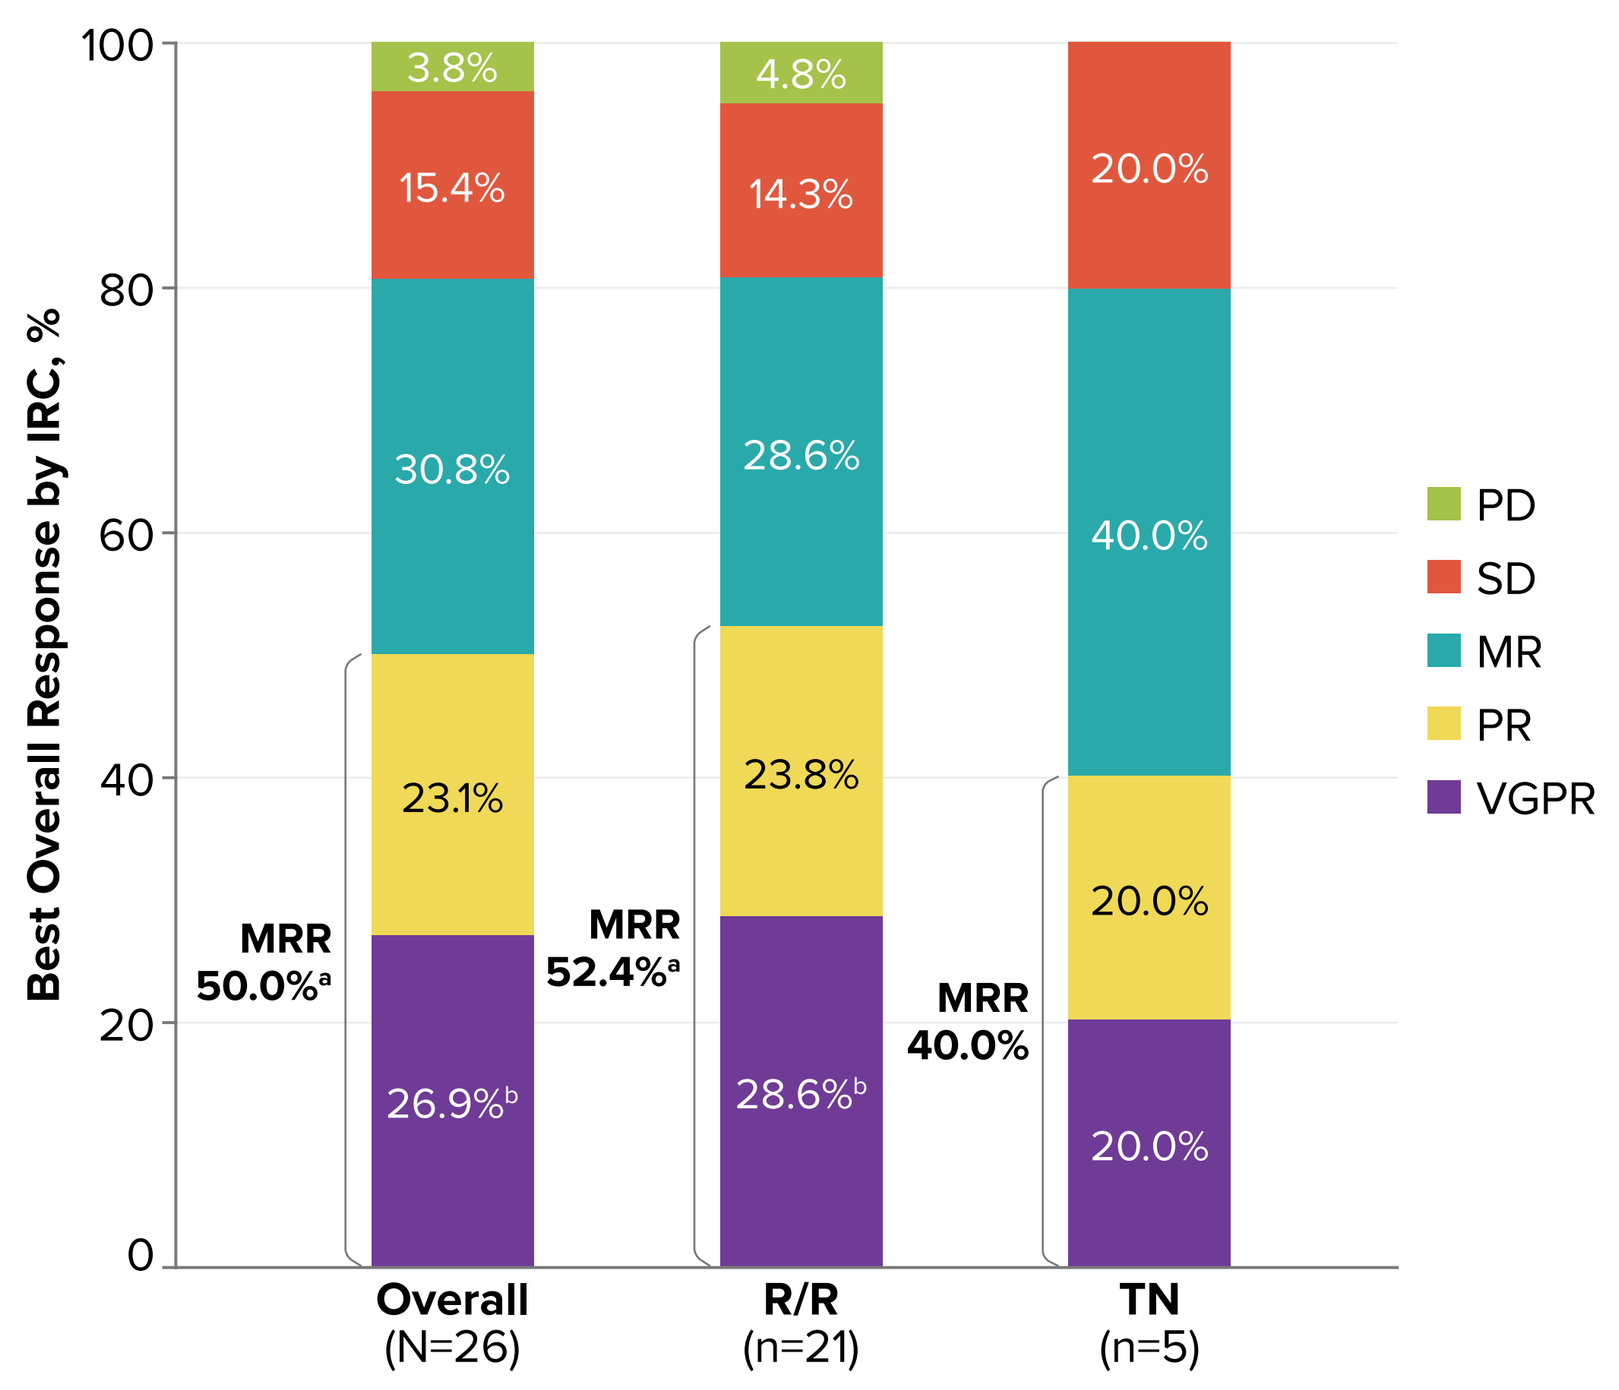

Best Overall Response by IRC, %
100
80
60
40
20
0
3.8%
15.4%
30.8%
23.1%
26.9%[b]
4.8%
14.3%
28.6%
23.8%
28.6%[b]
20.0%
40.0%
20.0%
20.0%
MRR 50.0%[a]
MRR 52.4%[a]
MRR 40.0%
Overall (N=26)
R/R (n=21)
TN (n=5)
PD
SD
MR
PR
VGPR

CR, complete response; IgM, immunoglobulin M; IRC, independent review committee; MR, minor response; MRR, major response rate (≥PR); PD, progressive disease; PR, partial response; pt, patient; R/R, relapsed/refractory; SD, stable disease; TN, treatment-naïve; VGPR, very good PR.
[a]Including pts confirmed by next-generation sequencing of no other activating *MYD88* mutations.
[b]One pt achieved IgM complete response (normalized IgM and negative immunofixation since cycle 11, with bulky extramedullary disease improving).

Figure 7. Responses Over Time on Treatment

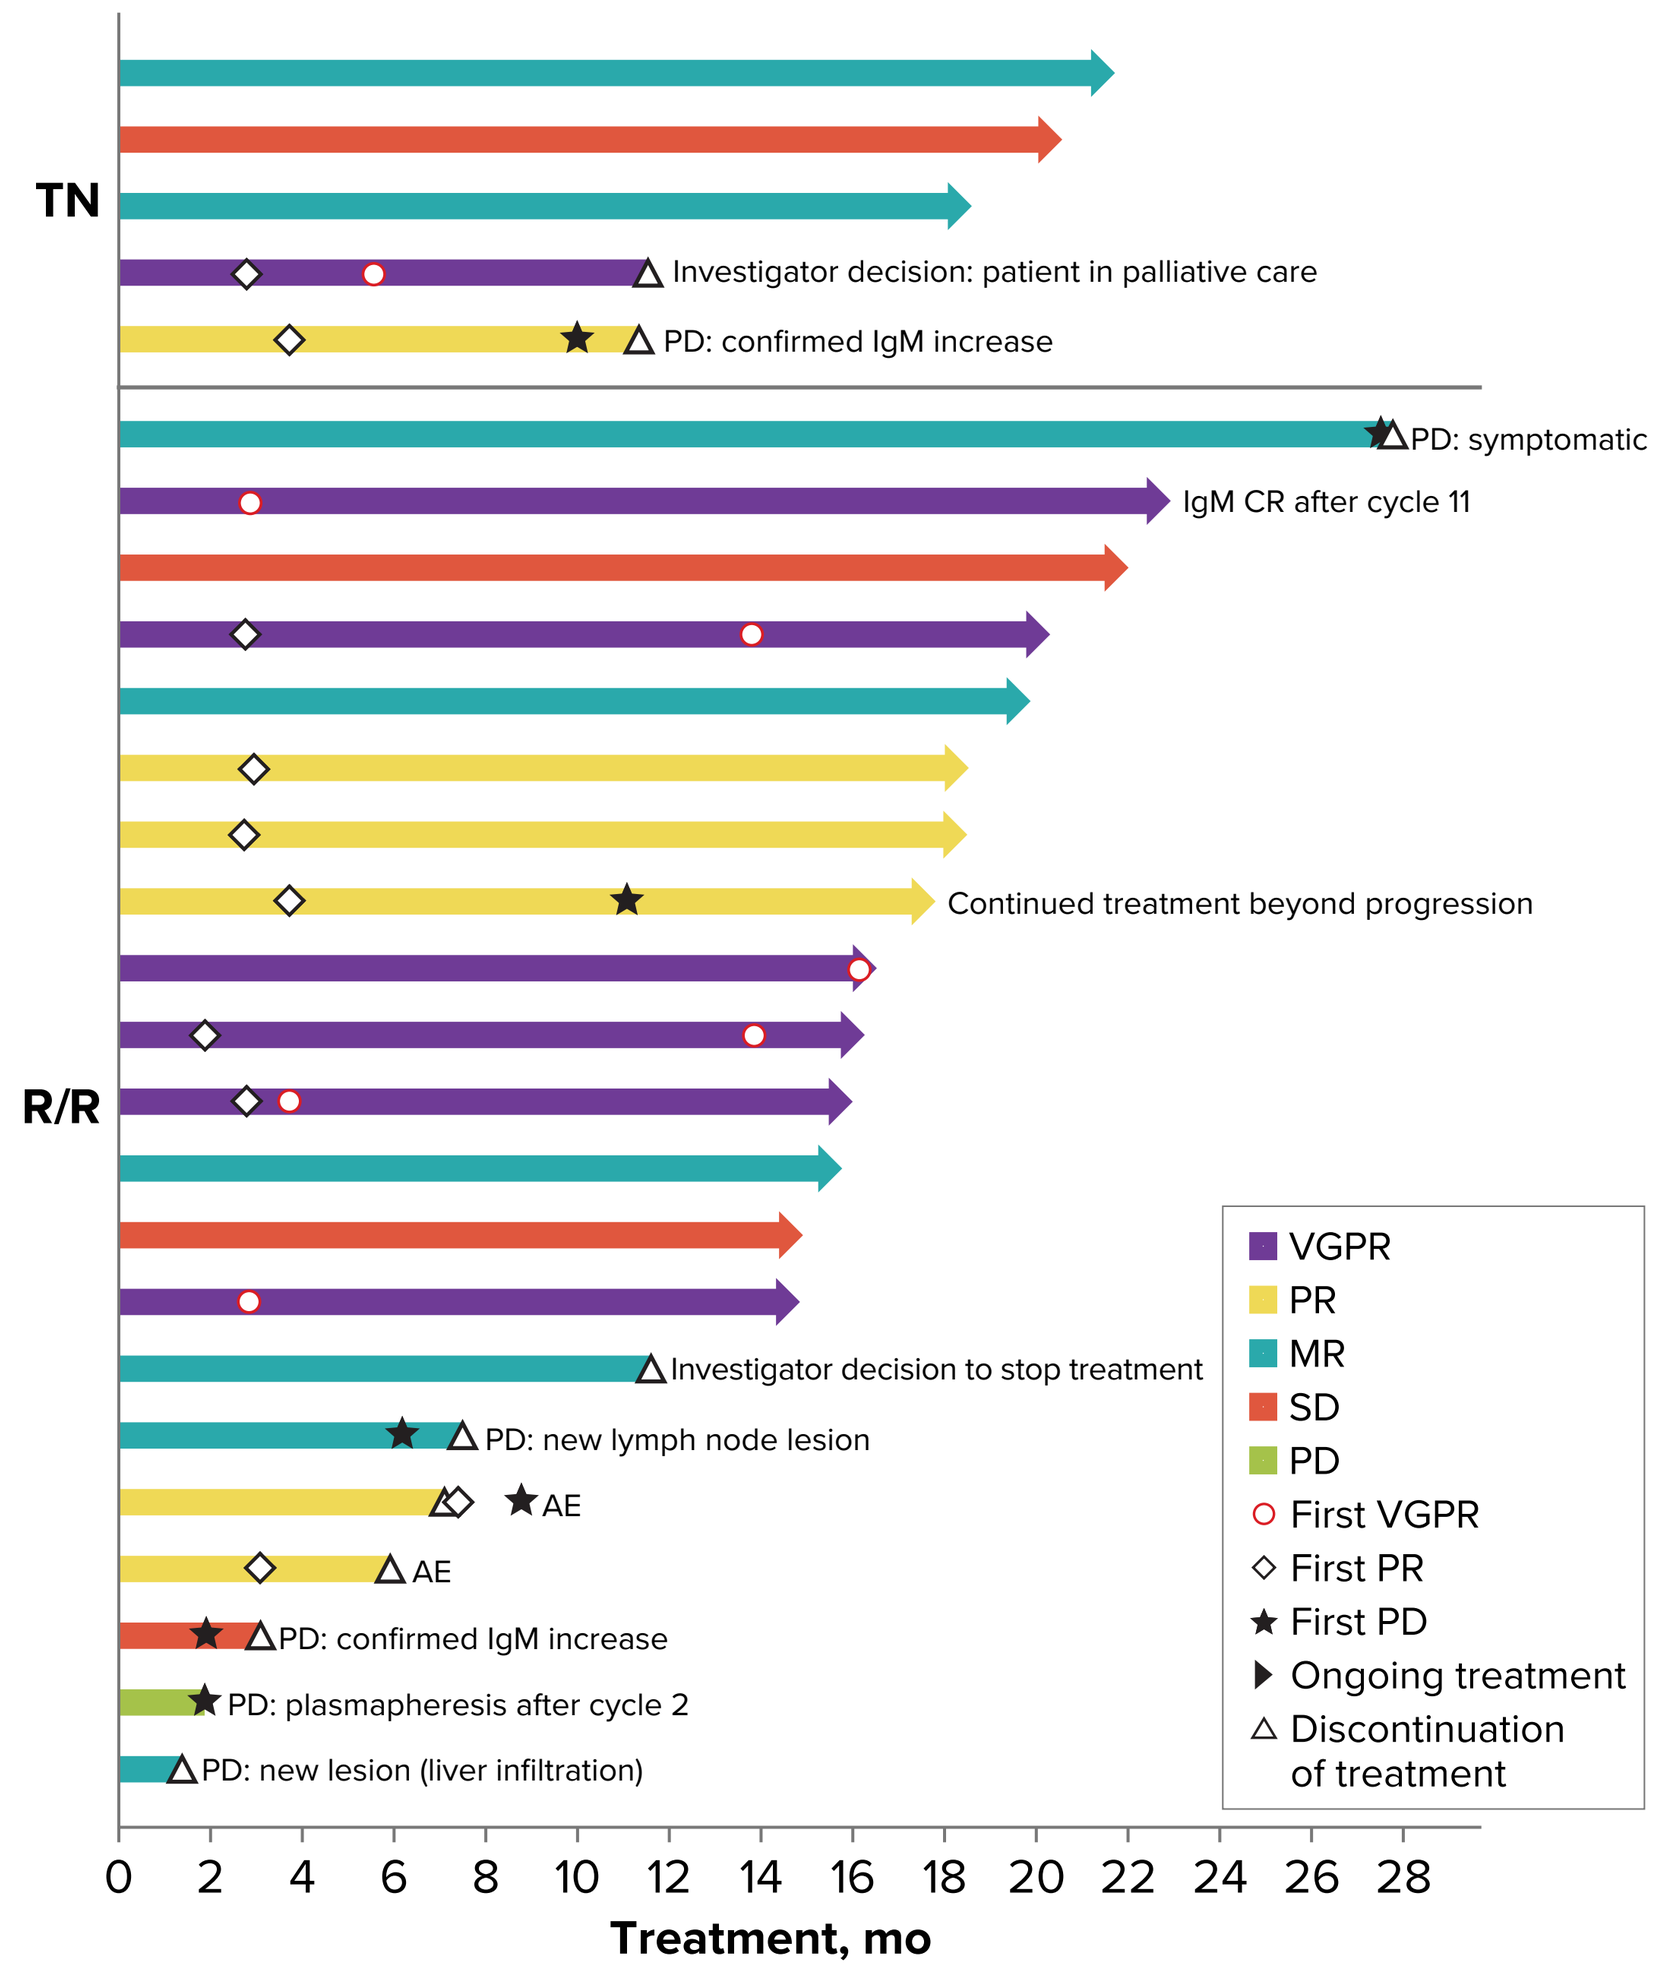

TN
R/R
Investigator decision: patient in palliative care
PD: confirmed IgM increase
PD: symptomatic
IgM CR after cycle 11
Continued treatment beyond progression
Investigator decision to stop treatment
PD: new lymph node lesion
AE
AE
PD: confirmed IgM increase
PD: plasmapheresis after cycle 2
PD: new lesion (liver infiltration)
VGPR
PR
MR
SD
PD
First VGPR
First PR
First PD
Ongoing treatment
Discontinuation of treatment
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Treatment, mo

Note: color of bars represents the best response for each patient.
AE, adverse event; MR, minor response; PD, progressive disease; PR, partial response; R/R, relapsed/refractory; SD, stable disease; TN, treatment-naïve; VGPR, very good PR.

Figure 8. Progression-Free and Overall Survival

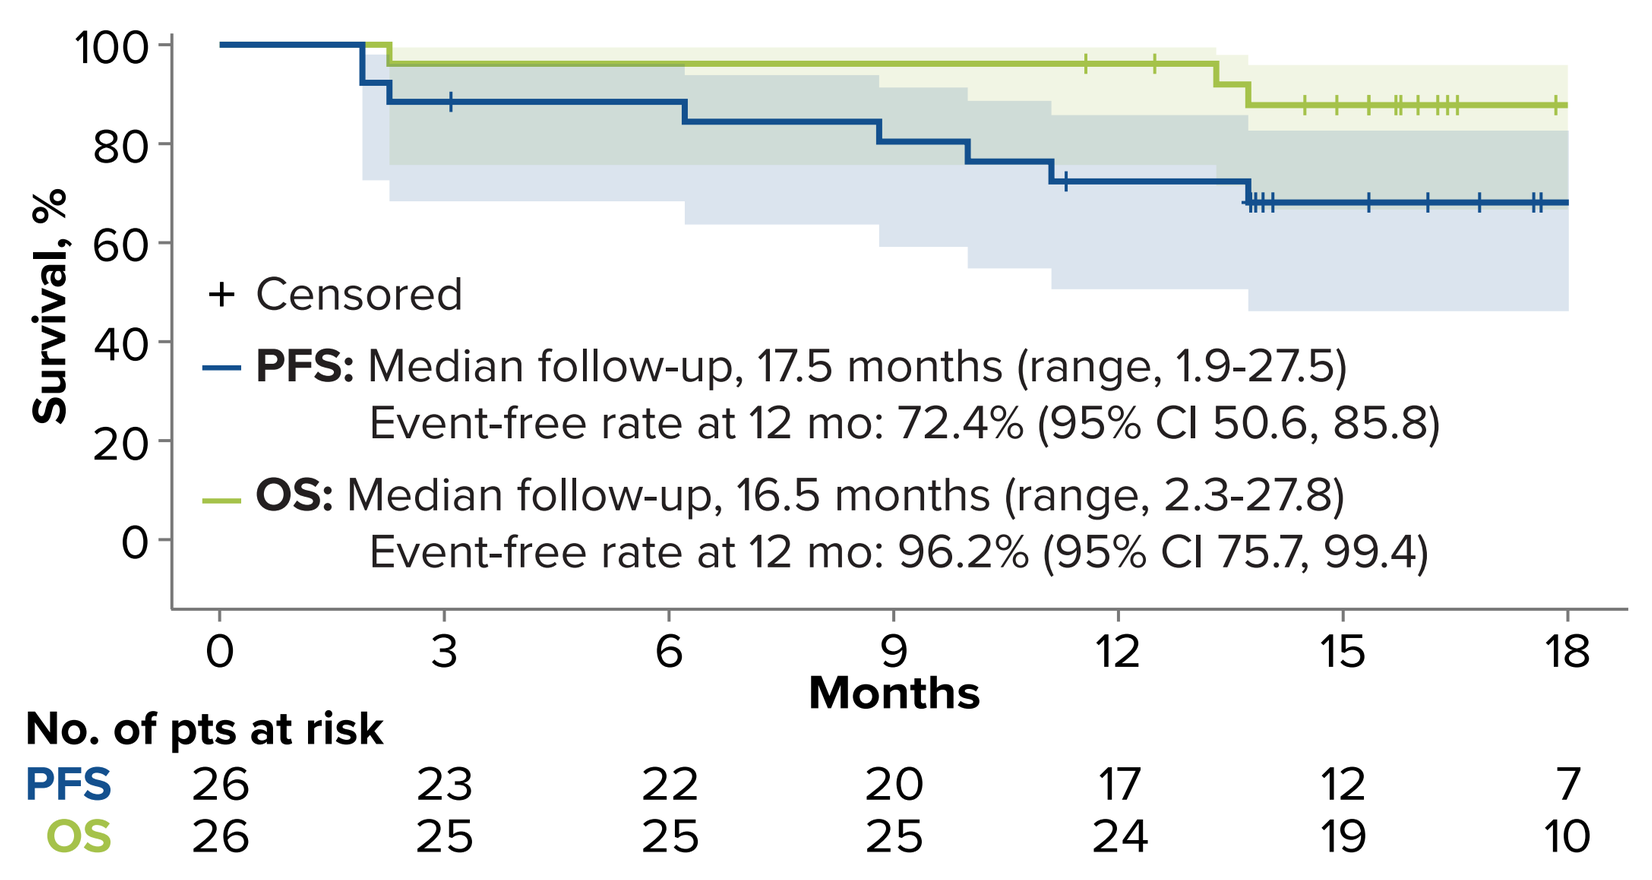

Survival, %
100
80
60
40
20
0
+ Censored
PFS: Median follow-up, 17.5 months (range, 1.9-27.5)
Event-free rate at 12 mo: 72.4% (95% CI 50.6, 85.8)
OS: Median follow-up, 16.5 months (range, 2.3-27.8)
Event-free rate at 12 mo: 96.2% (95% CI 75.7, 99.4)
0 3 6 9 12 15 18
Months
No. of pts at risk
PFS 26 23 22 20 17 12 7
OS 26 25 25 25 24 19 10

Shaded areas show the 95% CI.
OS, overall survival; PFS, progression-free survival.

CONCLUSIONS

- Largest cohort of pts with WM with confirmed *MYD88^{WT}* (n=26) studied in terms of safety and efficacy of BTKi treatment
- Single-agent zanubrutinib resulted in major responses (including very good partial response)
 - Major response rate of 50.0% including 26.9% with very good partial response
 - IgM complete response achieved in 1 pt
 - Median time to first major response was 2.9 months (range, 1.9-16.1)
- Zanubrutinib was well tolerated
 - Discontinuation because of AEs occurred in 7.1% of pts (2/28)
 - Primary reason for discontinuation was progressive disease (3 of 6 within first 3 cycles)
 - No fatal AEs reported
 - Low incidences of atrial fibrillation
 - AE profile is consistent with cohort 1 finding in the ASPEN study [abstract S225]

REFERENCES

1. Rickert RC. *Nat Rev Immunol*. 2013;13:578-591.
2. Choe H, Ruan J. *Oncology (Williston Park)*. 2016;30:847-858.
3. Yang G, et al. *Blood*. 2013;122:1222-1232.
4. Treon SP, et al. *J Clin Oncol*. 2020;38:1198-1208.
5. Treon SP, et al. *N Engl J Med*. 2015;372:1430-1440.
6. Treon SP, et al. *N Engl J Med*. 2015;373:584-586.
7. Palomba ML, et al. IWWM-9 Session 9, October 7, 2016 [abstr].
8. Tam CS, et al. *Blood*. 2019;134:851-859.
9. Guo Y, et al. *J Med Chem*. 2019;62:7923-7940.
10. Mu S, et al. *Cancer Chemother Pharmacol*. 2020;85:391-399.
11. BeiGene, data on file.
12. Brukinsa (zanubrutinib) [package insert]. San Mateo, CA: BeiGene USA, Inc; 2019.
13. Dimopoulos M, et al. *Blood*. 2014;124:1404-1411.
14. Albitar A, et al. *Int J Lab Hematol*. 2016;38:133-140.
15. Ngo VN, et al. *Nature*. 2011;470:115-119.
16. National Comprehensive Cancer Network (NCCN). NCCN Guidelines: Lymphoplasmacytic Lymphoma/Waldenström's Macroglobulinemia. 2015; version 2.
17. Owen RG, et al. *Br J Haematol*. 2013;160:171-176.

CORRESPONDENCE

mdimop@med.uoa.gr

DISCLOSURES

MAD: honoraria from and consulting/advisory role with Celgene, Amgen, BMS, Takeda, and Janssen. **RGS:** honoraria from Janssen, Takeda, and Amgen; consulting/advisory role with Janssen; research funding from Gilead Sciences and Incyte; and travel expenses from Janssen and Takeda. **HL:** stock options with CSL and honoraria and advisory board with Janssen and MSD. **MT:** employment with Charles University General Hospital; honoraria from Janssen, Gilead Sciences, Takeda, Bristol-Myers Squibb, Amgen, AbbVie, Roche, MorphoSys, and Incyte and consulting/advisory role with Takeda, Bristol-Myers Squibb, Incyte, AbbVie, Amgen, Roche, Gilead Sciences, Janssen, Celgene, and MorphoSys. **MV:** consulting/advisory role with Janssen and Roche and travel expenses from AbbVie and Janssen. **SO:** honoraria from Roche, Janssen, AbbVie, Gilead, Merck, AstraZeneca, and Novartis; consulting/advisory role with Roche, AstraZeneca, Janssen, AbbVie, Gilead, Merck, and Novartis; research funding from BeiGene, Roche, AstraZeneca, Janssen, Merck, Amgen, and Epizyme; and travel expenses from Roche. **SD:** research funding from Janssen and BeiGene; speakers' bureau with Amgen; and travel expenses with Janssen. **RGO:** honoraria from Janssen and Celgene; consulting/advisory role with Janssen; and travel expenses from Janssen and Celgene. **GC:** research funding from BeiGene, Acerta, and GlycoMimetics. **SM, JC, EA, SG, SR, and JK:** nothing to disclose. **JC:** consulting/advisory role with BeiGene, Janssen, Kymera, and Pharmacyclics and research funding from AbbVie, BeiGene, Janssen, Pharmacyclics, and TG Therapeutics. **MM:** consulting/advisory role with Roche and Janssen. **TS:** consulting/advisory role with AstraZeneca, Kite Pharma, Juno Therapeutics, BeiGene; research funding with PCYC, Juno, BeiGene, AstraZeneca, TG Therapeutics, and Celgene; and speakers' bureau with PCYC, Jannsen, AstraZeneca, and Seattle Genetics. **MGM:** speakers' bureau with Celgene and travel expenses from Janssen, Celgene, Amgen, and Takeda. **MGG:** consulting/advisory role with Celgene and Janssen and travel expenses from Janssen. **DT:** research funding and honoraria from Amgen, Janssen, Roche, and Takeda; advisory board membership for Roche, Janssen, and Amgen. **PLZ:** consulting/advisory role with Verastem, Celltrion, Gilead, Janssen-Cilag, BMS, Servier, Sandoz, MSD, Immune Design, Celgene, Portola, Roche, Eusapharma, Sanofi, and Kyowa Kirin and speakers' bureau with Verastem, Celltrion, Gilead, Janssen-Cilag, BMS, Servier, MSD, Immune Design, Celgene, Portola, Roche, Eusapharma, and Kyowa Kirin. **AO:** consulting/advisory role with Celgene and Amgen; speakers' bureau with Celgene, Amgen, and Janssen. **AT:** honoraria with Janssen, AstraZeneca, and AbbVie and consulting/advisory role with Janssen spa, AstraZeneca, and AbbVie. **CB:** honoraria with Roche, Janssen, Celltrion, and BeiGene; consulting/advisory role with Roche, Janssen, Celltrion, and BeiGene; research funding from Roche, Janssen, and BeiGene; and speakers' bureau with Roche, Janssen, Celltrion, and BeiGene. **VL:** honoraria from AstraZeneca, Roche, Janssen, Gilead, AbbVie, and Amgen; consulting/advisory role with AstraZeneca, Roche, Janssen, and AbbVie; speaker's bureau for AbbVie and Janssen; and travel expenses from AbbVie, Roche, and Janssen. **WYC:** employment with BeiGene and equity ownership with BeiGene and Bristol Myers Squibb. **JS:** employment and equity ownership with BeiGene. **SR:** employment with BeiGene and equity ownership with BeiGene and Amgen. **AC:** employment with, equity ownership with, and travel expenses from BeiGene. **JH:** employment, leadership role, and equity ownership with BeiGene. **CST:** honoraria from BeiGene, Janssen, AbbVie, Novartis, and Roche and research funding from Janssen and AbbVie.

ACKNOWLEDGMENTS

We thank the investigators, site support staff, and especially the patients for participating in this study. This study was sponsored by BeiGene. Editorial support was provided by Bio Connections, LLC and funded by BeiGene.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.

Presented at the 25th Congress of the European Hematology Association, June 11-14, 2020; Virtual Format