ASSOCIATION BETWEEN PROGRAMMED DEATH-LIGAND 1 EXPRESSION AND GENE SIGNATURES OF RESPONSE OR RESISTANCE TO TISLELIZUMAB MONOTHERAPY IN HEPATOCELLULAR CARCINOMA

Ming-Mo Hou[1], Kun-Ming Rau[2], Yoon-Koo Kang[3], Jong-Seok Lee[4], Hongming Pan[5], Ying Yuan[6], Cunjing Yu[7], Yun Zhang[7], Xiaopeng Ma[7], Xikun Wu[7], Xin Li[7], Katie Wood[8], Chia-Jui Yen[9]

[1]Chang Gung Memorial Hospital, Linkou, Taiwan; [2]Kaohsiung Chang Gung Memorial Hospital; [3]Asan Medical Center, Seoul, Republic of Korea; [4]Seoul National University Bundang Hospital, Seongnam-si, Gyeonggi-do, Republic of Korea; [5]Sir Run Run Shaw Hospital, Zhejiang University, Hangzhou, China; [6]The Second Affiliated Hospital Zhejiang University School of Medicine, Hangzhou, China; [7]BeiGene (Beijing) Co., Ltd., Beijing, China; [8]BeiGene USA, Inc., San Mateo, CA, USA; [9]National Cheng Kung University Hospital

Poster: 77
Society for Immunotherapy of Cancer
November 9-14, 2020, Virtual Congress

BACKGROUND

- Liver cancer was the sixth most common type of cancer and the fourth leading cause of cancer death worldwide in 2018, with 841,080 new cases worldwide[1,2]
 - Hepatocellular carcinoma (HCC) accounted for 85% to 90% of all reported liver cancer cases[1,2]
- Sorafenib as a treatment option for advanced-stage HCC yields moderate survival benefit[3]
- Recently, treatment with programmed cell death protein-1 (PD-1) and programmed death-ligand 1 (PD-L1) inhibitors in HCC have shown clinical benefit[4,5,6]
 - Response or resistance mechanisms are not yet clear, highlighting an unmet need to identify optimal biomarkers that can predict response
- Exploring immune- and tumor-transcriptomic features and their association with anti-PD-1 efficacy may increase the understanding of the tumor microenvironment in HCC and aid in the identification of potential response/resistance mechanisms
- Tislelizumab, an anti-PD-1 monoclonal antibody, has demonstrated single-agent antitumor activity in patients with advanced, previously treated HCC in two phase 1 studies (BGB-A317-001 [NCT02407990], BGB-A317-102 [NCT04068519])
 - Biomarkers, including PD-L1 expression and gene expression profiles (GEPs), and their association with response and resistance to tislelizumab were explored in these studies

METHODS

Study Design

- Pooled analysis from two clinical trials were used:
 - BGB-A317-001 (NCT02407990): First-in-human, multicenter, phase 1a/1b dose-escalation/indication-expansion study
 - BGB-A317-102 (NCT04068519): Chinese, multicenter, phase 1/2 study
 - Samples from both studies were analyzed from patients with previously treated advanced HCC
- Previous reports have shown increased PD-L1 expression on tumor cells (TCs) after sorafenib treatment[7]
- In this study, sorafenib exposed samples were defined as tissue samples collected after a record of sorafenib treatment and sorafenib non-exposed samples were defined as tissue samples collected prior to record of sorafenib treatment
- PD-L1 expression was evaluated on TCs using the VENTANA PD-L1 (SP263) assay in baseline tumor samples from 62 patients
- Clinical benefit (CB) was defined as the proportion of patients who have complete response, partial response, and stable disease that is ≥24 weeks in duration per Response Evaluation Criteria in Solid Tumors v1.1

Gene Expression Profiling

- Gene expression profiling using the HTG EdgeSeq Precision Immuno-Oncology Panel was performed on baseline tumor samples (formalin-fixed, paraffin-embedded blocks or cut slides)
- Signature scores were calculated using the Gene Set Variation Analysis package with publicly available gene signatures
- Differentially expressed gene signature (DEG) analysis was performed between responders (Rs) and non-responders (NRs) using a Wilcoxon rank-sum test
- Non-responder subgroups were hierarchically clustered by one minus Pearson's correlation with average linkage by columns

Statistical Analysis

- Kruskal-Wallis test was used to detect overall response effect, and pairwise Wilcoxon test was used to detect differences between two response subgroups
 - Bonferroni method was used to adjust multiple comparisons for each signature
- Potential associations with survival were analyzed using a log-rank test and Cox proportional hazards model

RESULTS

Patient Characteristics

- Of the 68 HCC patients enrolled in the BGB-A317-001 and -102 studies, 62/68 (91%) had sample collection and sorafenib treatment records available for analysis
 - Patient baseline characteristics and clinical efficacy are shown in Table 1

Table 1: Baseline Characteristics and Clinical Outcome

Characteristic		ITT (N=68)	GEP (N=62)
Age, n (%)	<65	52 (76.5)	47 (75.8)
	≥65	16 (23.5)	15 (24.2)
Sex, n (%)	Male	56 (82.4)	51 (82.3)
	Female	12 (17.6)	11 (17.7)
Tumor stage, n (%)	Stage III	4 (5.9)	2 (3.2)
	Stage IV	64 (94.1)	60 (96.8)
Histologic grade at baseline, n (%)	Well differentiated	19 (50.0)	17 (47.2)
	Moderately differentiated	12 (31.6)	12 (33.3)
	Poorly differentiated	7 (18.4)	7 (19.4)
ECOG score at baseline, n (%)	0	27 (39.7)	23 (37.1)
	1	41 (60.3)	39 (62.9)
AFP (ng/ml), n (%)	≥400	28 (29.4)	26 (41.9)
	<400	30 (44.1)	26 (41.9)
Objective response rate, % (95% CI)		13.24 (6.23, 23.64)	14.52 (6.86, 25.78)
Median PFS, months (95% CI)		3.33 (2.10, 4.07)	3.57 (2.13, 4.07)
Median OS, months (95% CI)		13.3 (11.13, NE)	12.97 (11.13, NE)
Median follow-up, months (95% CI)		14.47 (8.50, 17.47)	14.03 (8.50, 17.47)

Abbreviations: AFP, alpha-fetoprotein; CI, confidence interval; ECOG, Eastern Cooperative Oncology Group; GEP, gene expression profile; ITT, intent-to-treat; NE; not evaluable; OS, overall survival; PFS, progression-free survival; SD, standard deviation.

Prevalence of PD-L1 TC ≥1% and GEP Analysis Showed Different Patterns in Sorafenib Exposed and Non-Exposed Samples

- Sorafenib-exposed (n=41) samples had a higher prevalence of PD-L1 TC ≥1% compared with non-exposed samples (n=16) (53.7% vs 25%; *P*=0.08; Figure 1A)
- In sorafenib non-exposed samples, multiple immune suppressive factors were enriched, including immune-related (regulatory T cell [Treg] and myeloid-derived suppressor cell) and tumor intrinsic (TGFβ and hypoxia) signatures
- In sorafenib-exposed samples, immune cell activation signatures were enriched along with the co-inhibition signature (Figure 1B)

Figure 1: Prior Sorafenib Exposure May Affect PD-L1 Expression as Well as Tumor Microenvironment-Related Gene Signature

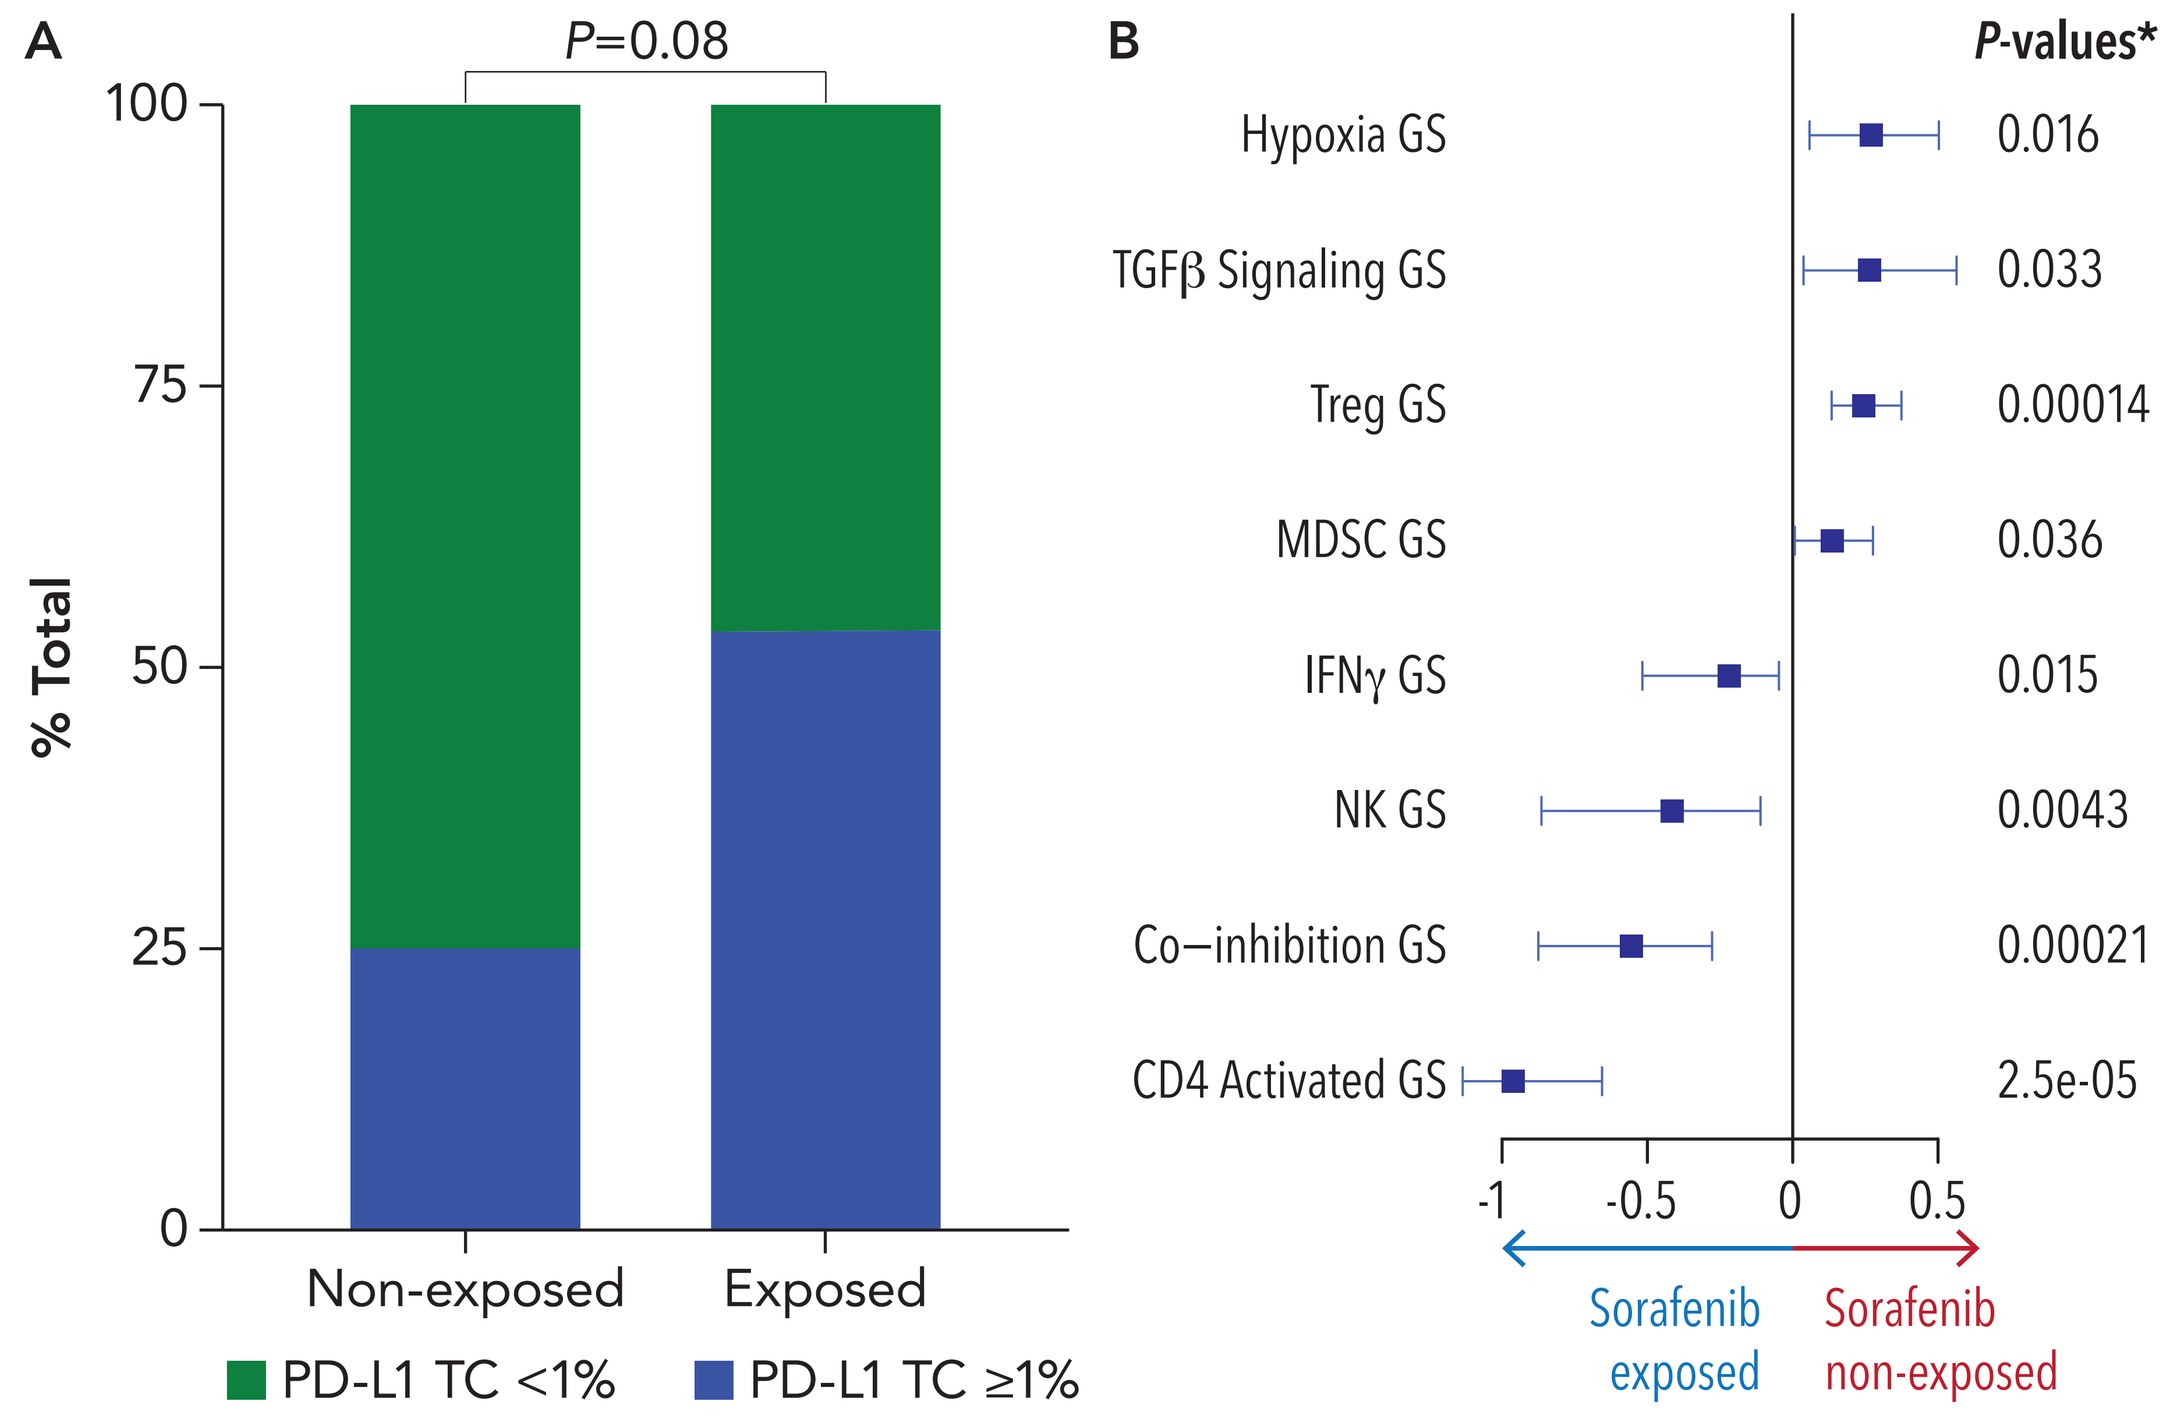
A
P=0.08
% Total
100
75
50
25
0
Non-exposed
Exposed
PD-L1 TC <1%
PD-L1 TC ≥1%
B
P-values*
Hypoxia GS 0.016
TGFβ Signaling GS 0.033
Treg GS 0.00014
MDSC GS 0.036
IFNγ GS 0.015
NK GS 0.0043
Co−inhibition GS 0.00021
CD4 Activated GS 2.5e-05
-1 -0.5 0 0.5
Sorafenib exposed
Sorafenib non-exposed

*Median of the difference (Wilcoxon test) and the nonparametric confidence interval.
Abbreviations: GS, gene signature; IFNγ, interferon gamma; MDSC, myeloid-derived suppressor cell; NK, natural killer; PD-L1; programmed death-ligand 1; TC, tumor cell; TGFβ, transforming growth factor beta; Treg, regulatory T cell.

- In sorafenib-exposed samples, PD-L1 TC expression was significantly higher in patients with CB versus patients with no clinical benefit (non-CB) as revealed by DEG and box-plot analysis (*P*=0.0027; Figure 2A)
- Higher objective response rates (ORRs) were observed in patients with PD-L1 TC ≥1% compared with patients with PD-L1 TC <1% (ORR=23.8% vs 0%; *P*=0.049; Figure 2B)
- A longer progression-free survival (PFS) and overall survival (OS) trend were linked to patients with PD-L1 TC ≥1%, and separation of Kaplan-Meier curves between the 2 arms was observed (Figure 2C and 2D)

Figure 2: PD-L1 Expression Was Positively Correlated With Clinical Benefit and a Trend of Longer PFS in Sorafenib-Exposed Samples

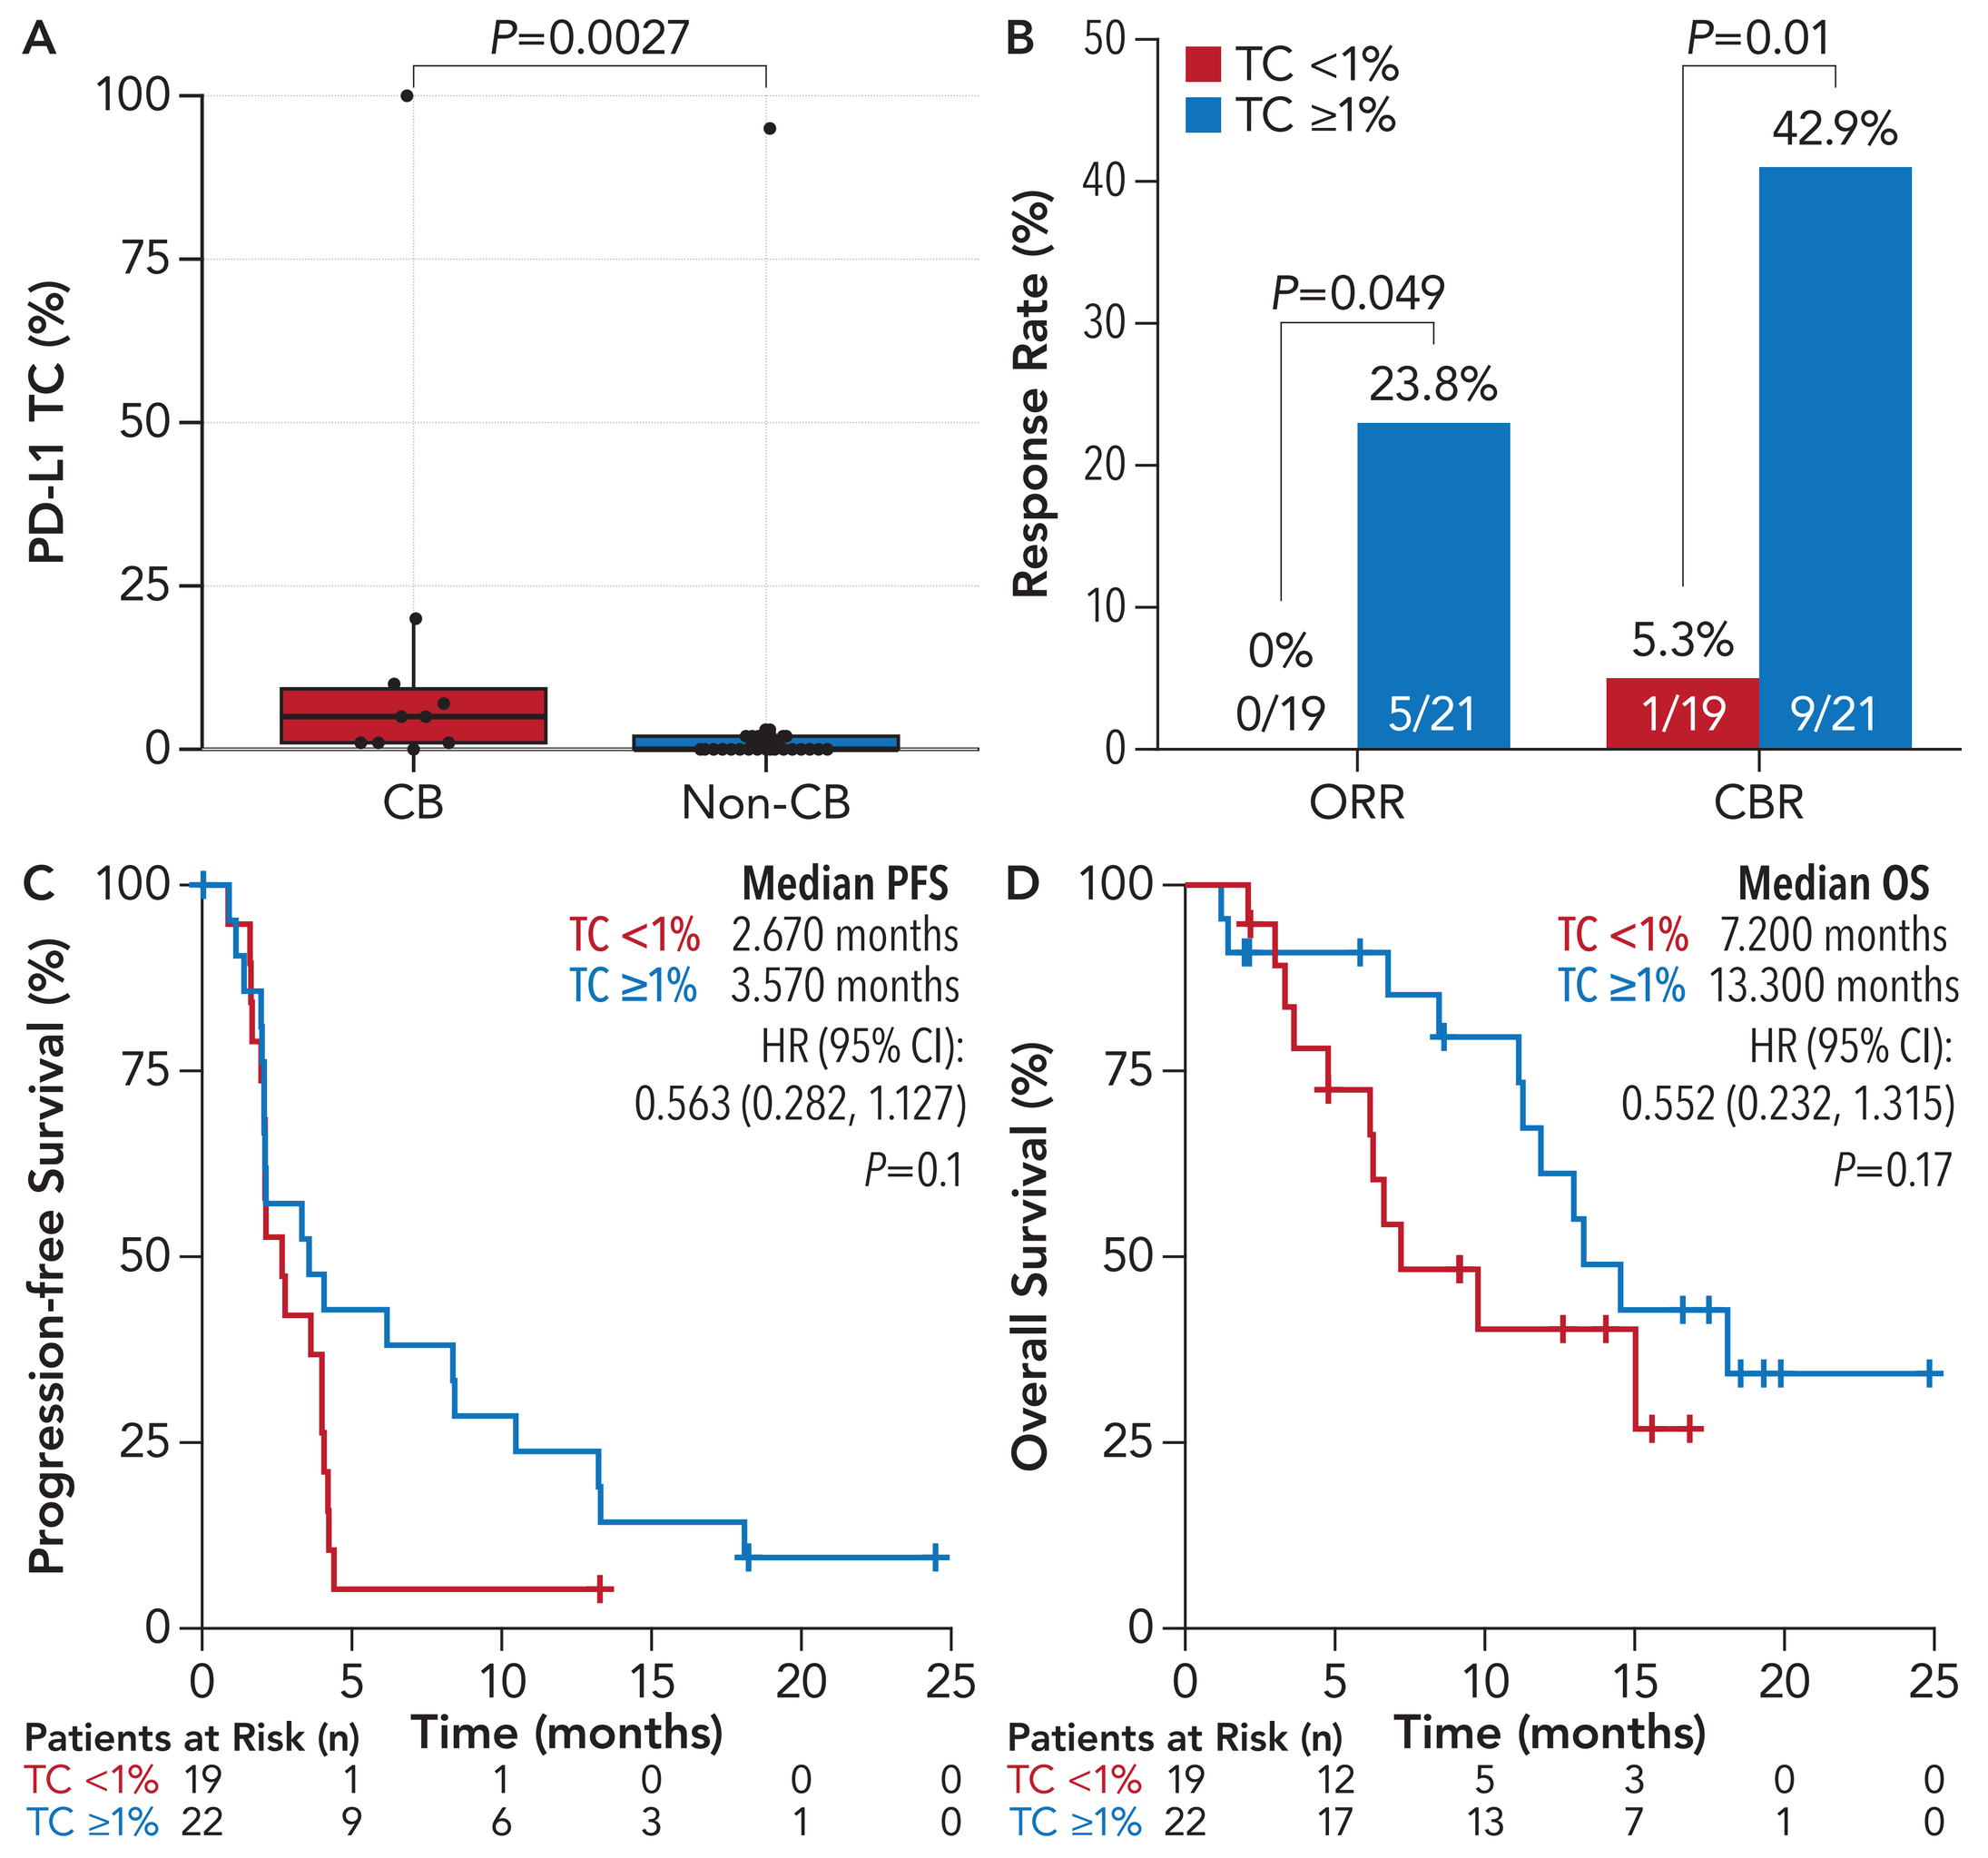
A
P=0.0027
PD-L1 TC (%)
CB
Non-CB
B
TC <1%
TC ≥1%
Response Rate (%)
P=0.049
23.8%
0%
0/19
5/21
ORR
P=0.01
42.9%
5.3%
1/19
9/21
CBR
C
Progression-free Survival (%)
Median PFS
TC <1% 2.670 months
TC ≥1% 3.570 months
HR (95% CI):
0.563 (0.282, 1.127)
P=0.1
Patients at Risk (n)
Time (months)
TC <1% 19 1 1 0 0 0
TC ≥1% 22 9 6 3 1 0
D
Overall Survival (%)
Median OS
TC <1% 7.200 months
TC ≥1% 13.300 months
HR (95% CI):
0.552 (0.232, 1.315)
P=0.17
Patients at Risk (n)
Time (months)
TC <1% 19 12 5 3 0 0
TC ≥1% 22 17 13 7 1 0

Abbreviations: CB, clinical benefit; CBR, clinical benefit rate; CI, confidence interval; HR, hazard ratio; non-CB, no clinical benefit; ORR, objective response rate; OS, overall survival; PD-L1, programmed death-ligand 1; PFS, progression-free survival; TC, tumor cell.

- In sorafenib-exposed samples, the natural killer (NK) cell–mediated cytotoxicity signature was significantly higher in tumors from CB versus non-CB as revealed by DEG and box-plot analysis (*P*=0.03; Figure 3A)
- A trend of improved PFS was observed in patients with NK cytotoxicity-high versus NK cytotoxicity-low signatures (median PFS: 3.57 vs 2.12, *P*=0.062; Figure 3B)
- No significant association between NK cytotoxicity signature and OS was observed (Figure 3C)

Figure 3: In Sorafenib-Exposed Samples, Natural Killer Cell–Mediated Cytotoxicity Is Associated With Clinical Outcomes of Tislelizumab Monotherapy

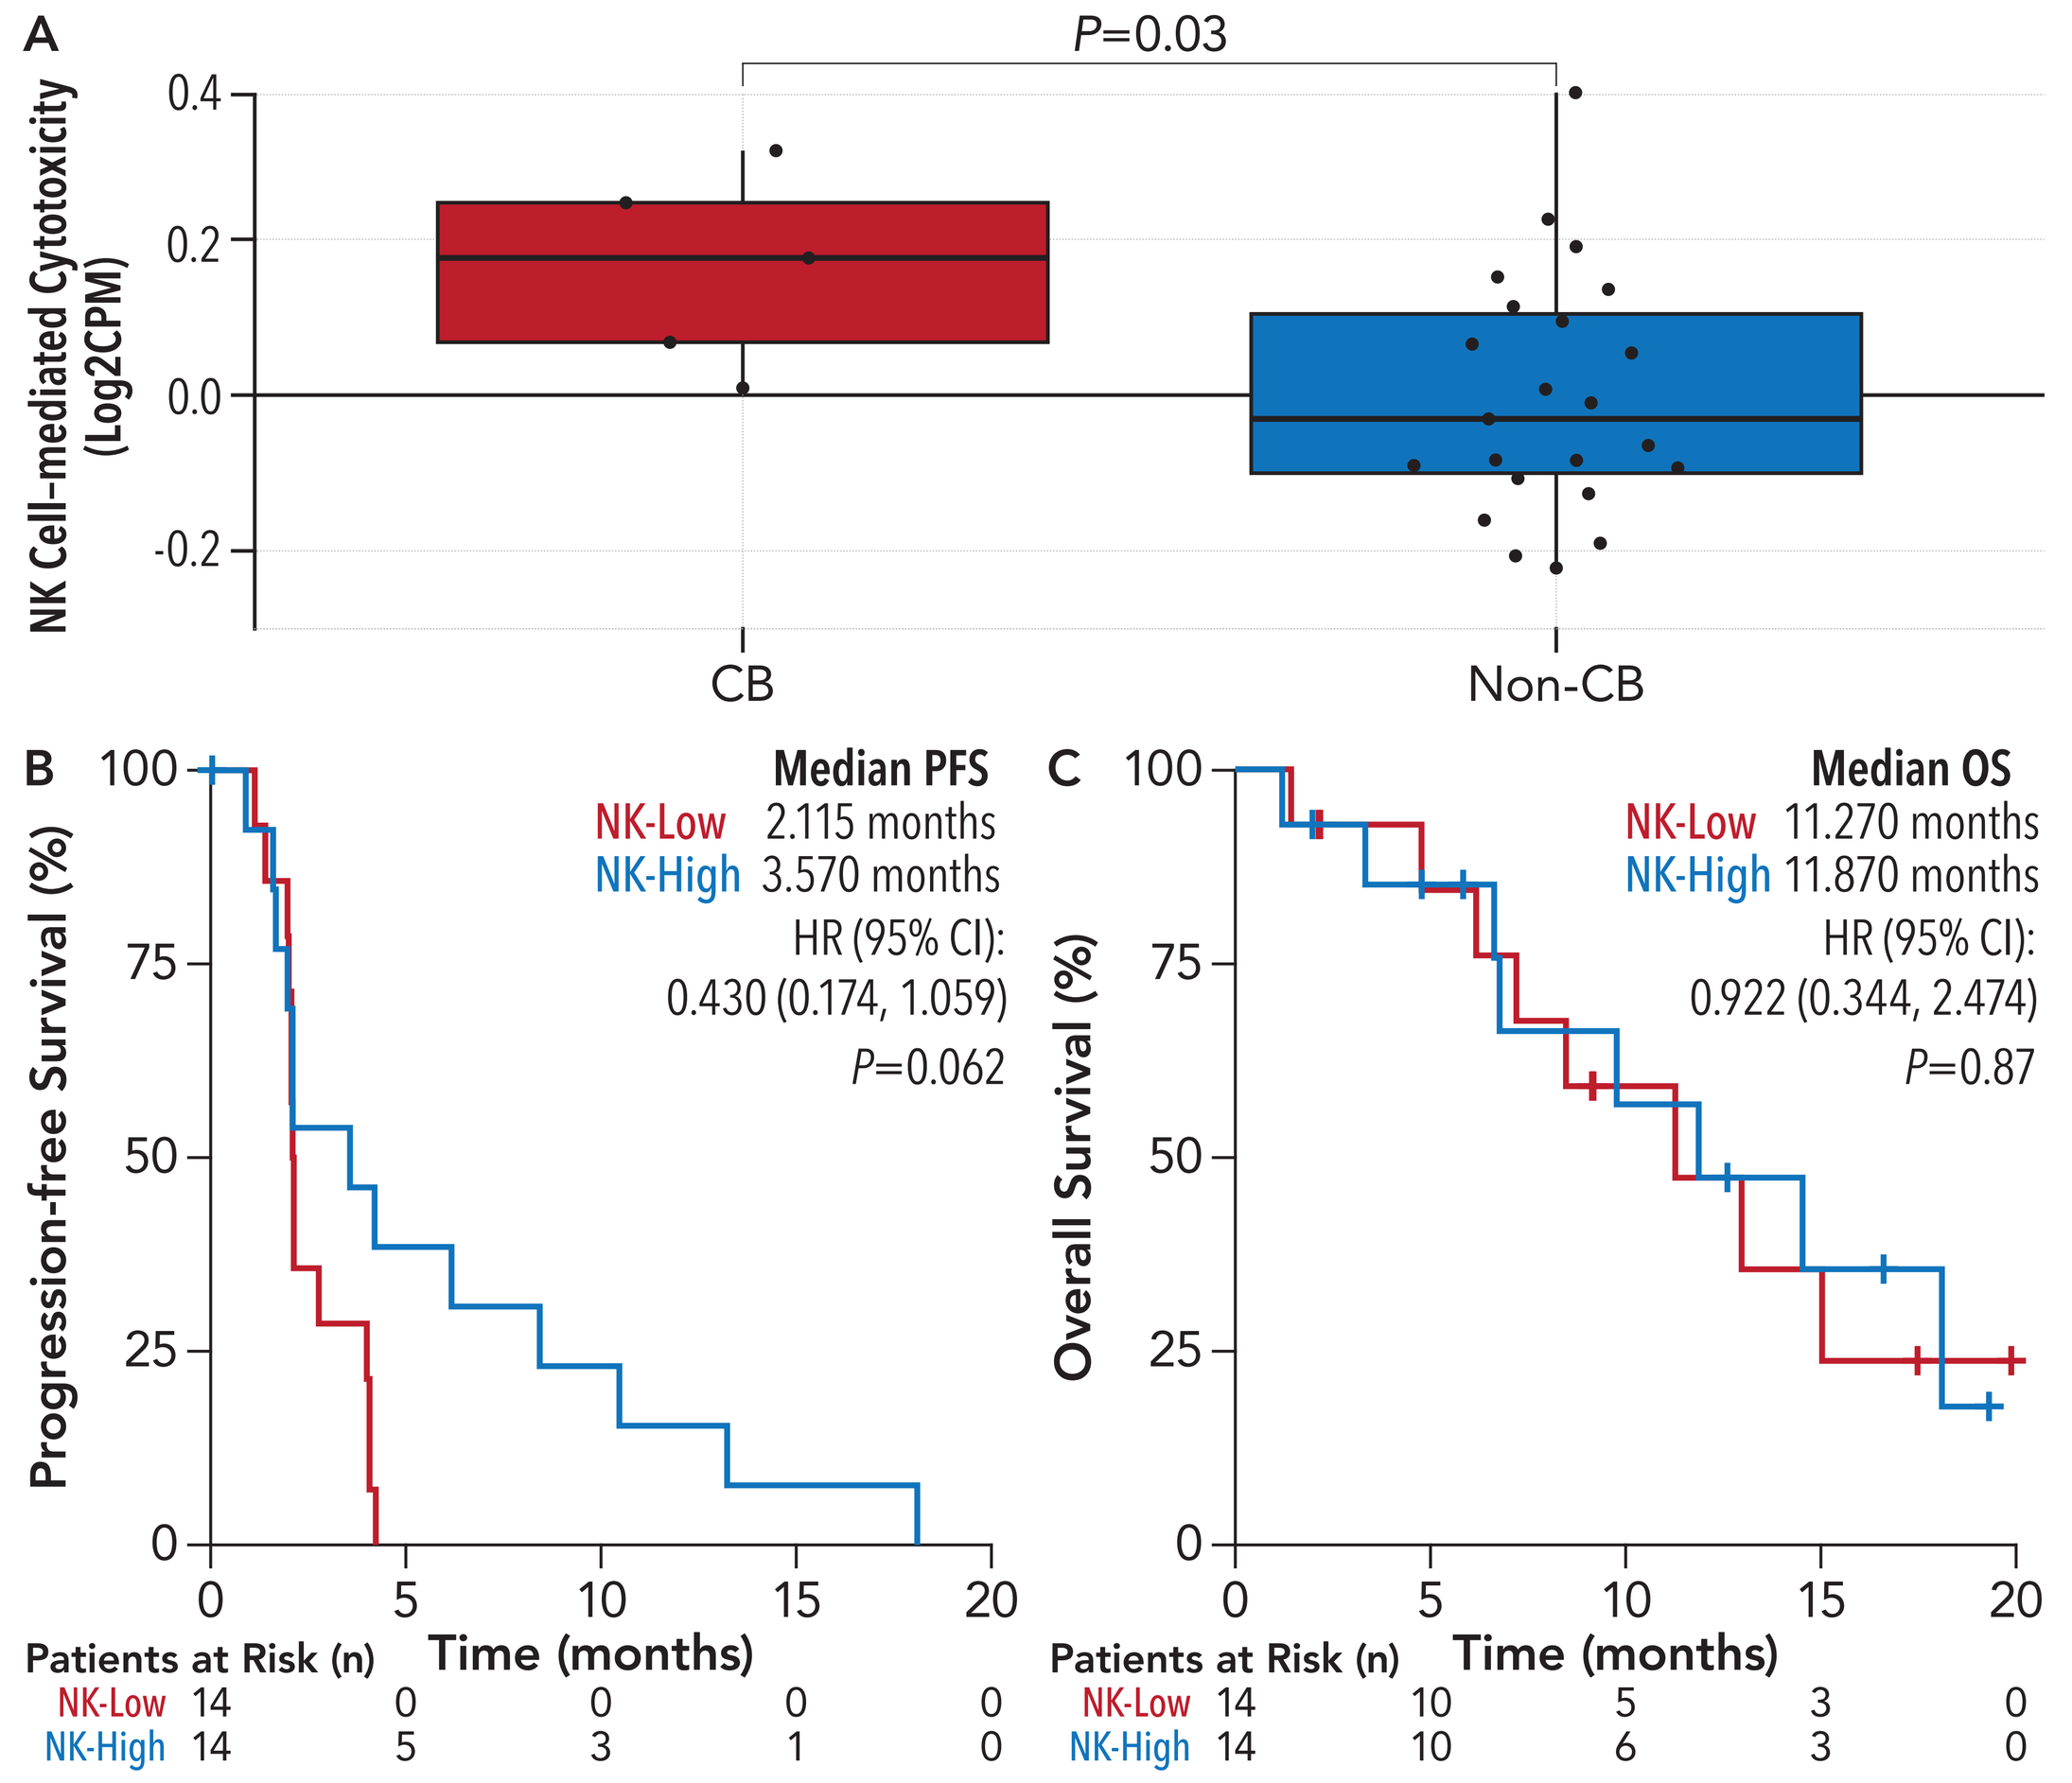
A
P=0.03
NK Cell-mediated Cytotoxicity (Log2CPM)
CB
Non-CB
B
Progression-free Survival (%)
Median PFS
NK-Low 2.115 months
NK-High 3.570 months
HR (95% CI):
0.430 (0.174, 1.059)
P=0.062
Patients at Risk (n)
Time (months)
NK-Low 14 0 0 0 0
NK-High 14 5 3 1 0
C
Overall Survival (%)
Median OS
NK-Low 11.270 months
NK-High 11.870 months
HR (95% CI):
0.922 (0.344, 2.474)
P=0.87
Patients at Risk (n)
Time (months)
NK-Low 14 10 5 3 0
NK-High 14 10 6 3 0

Abbreviations: CB, clinical benefit; CI, confidence interval; HR, hazard ratio; NK, natural killer; non-CB, no clinical benefit; OS, overall survival; PFS, progression-free survival.

- Monotherapy NRs could be clustered into three distinct GEP subgroups according to the immune and gene signatures listed in Table 2

Table 2: Immune and Tumor Gene Signatures Utilized for NR Subgroup Clustering

Antitumor Immune Activity		Immune Cell Population Abundance			Feature of Tumor	
Antigen presentation	IFNγ	CD4/CD8	Mast cell	NK cell	Cell cycle	DNA repair
Cytotoxicity	Immune checkpoint	Exhausted CD8	Dendritic cell	Neutrophil	TGFβ	Angiogenesis
Inflammatory		Macrophage	Treg	MDSC		

Abbreviations: IFNγ, interferon gamma; MDSC, myeloid-derived suppressor cell; NK, natural killer; TGFβ, transforming growth factor beta; Treg, regulatory T cell.

- Differentially expressed gene signatures and PFS/OS among distinct NR subgroups and Rs are shown in Figure 4 and Figure 5
 - NR1 had significantly higher angiogenesis signatures than R (adjusted *P*=0.05) (driven by *TEK*, *KDR*, *HGF*, and *EGR1*, etc.), as well as the lowest median PFS and OS (1.97 months and 6.63 months, respectively; Figure 4A and Figure 5)
 - NR2 had a trend of higher expression of the T-cell inhibition signature than R (adjusted *P*=0.09) (driven by *CD274*, *CTLA4*, *TIGIT*, and *CD96*), with a median PFS of 2.13 months and median OS of 9.77 months (Figure 4B and Figure 5)
 - NR3 had the highest cell cycle signature, which was driven by *E2F7*, *FOXA1*, and *FANCD2*, with a median PFS of 5.17 months and median OS of 15.03 months (Figure 4C and Figure 5)

Figure 4: Tumor-Immune Profiles of Responder and Non-Responder Subgroups of Tislelizumab in HCC

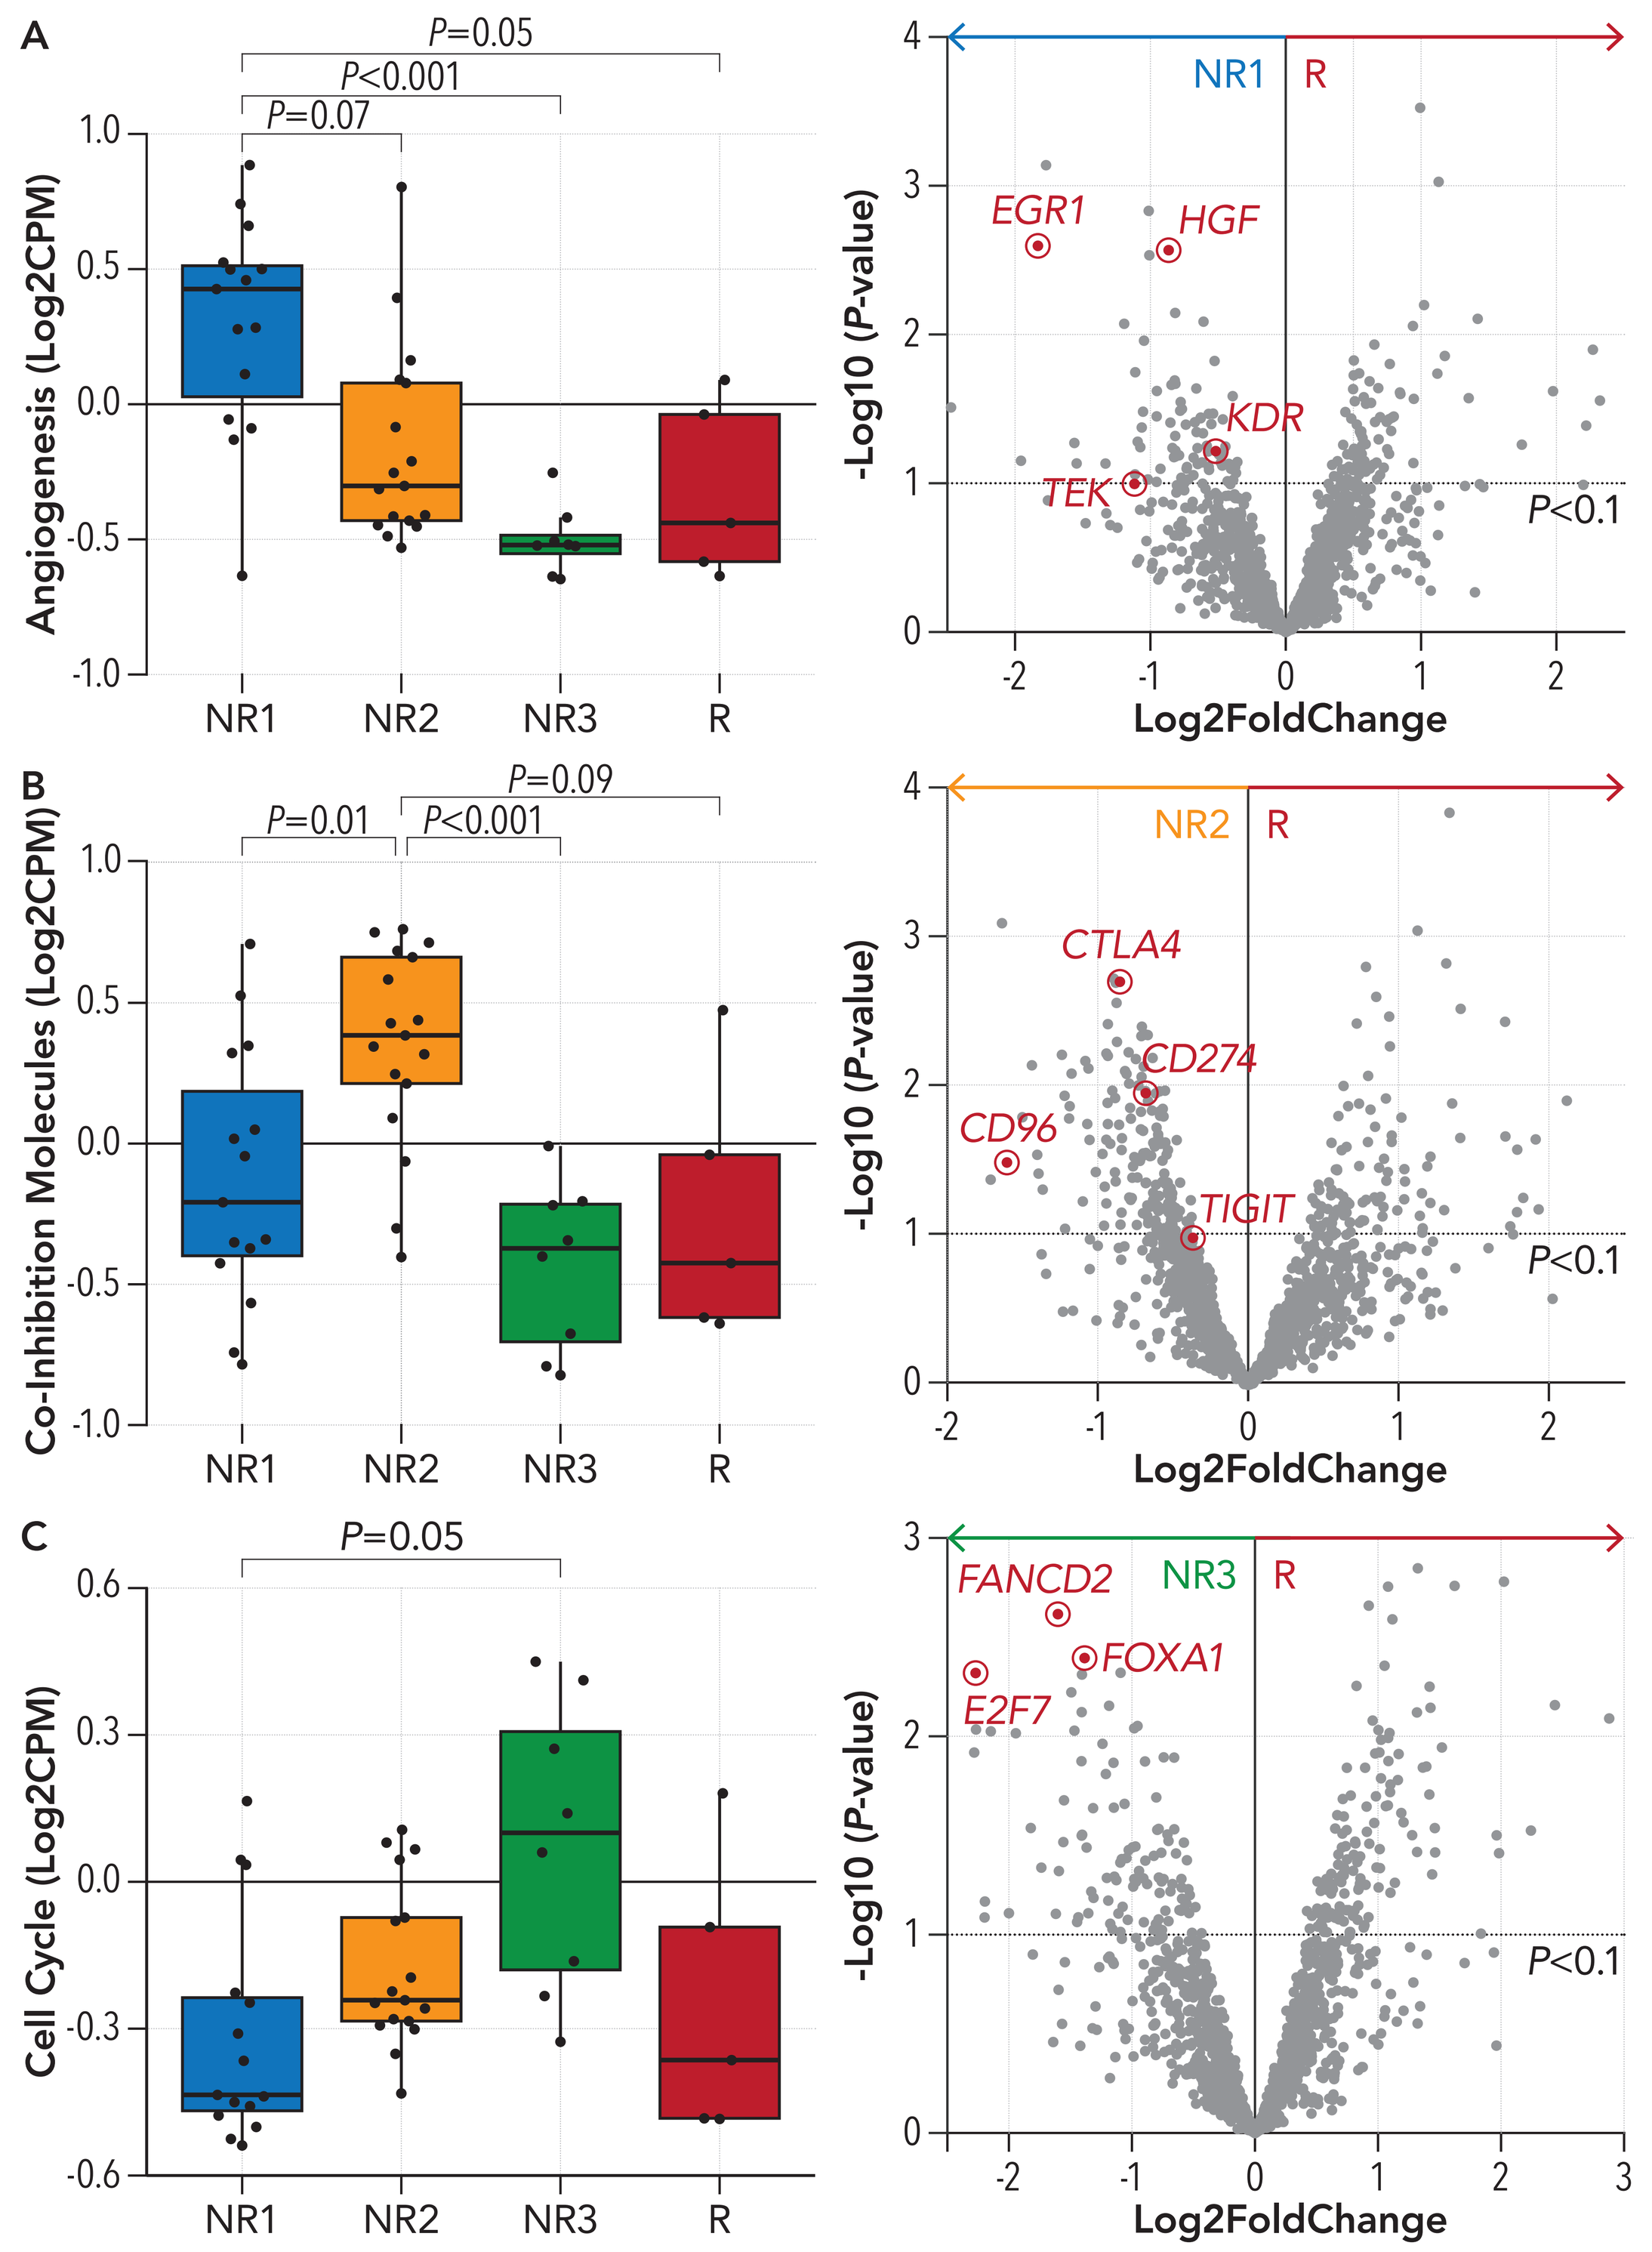
A
P=0.05
P<0.001
P=0.07
Angiogenesis (Log2CPM)
NR1 NR2 NR3 R
NR1
R
EGR1
HGF
KDR
TEK
P<0.1
-Log10 (P-value)
Log2FoldChange
B
P=0.09
P=0.01
P<0.001
Co-Inhibition Molecules (Log2CPM)
NR1 NR2 NR3 R
NR2
R
CTLA4
CD274
CD96
TIGIT
P<0.1
-Log10 (P-value)
Log2FoldChange
C
P=0.05
Cell Cycle (Log2CPM)
NR1 NR2 NR3 R
FANCD2
NR3
R
FOXA1
E2F7
P<0.1
-Log10 (P-value)
Log2FoldChange

Abbreviations: HCC, hepatocellular carcinoma; NR, non-responder; R, responder.

CONCLUSIONS

- Sorafenib exposure may affect the predictive value of PD-L1 expression and tumor microenviroment-related gene signatures to anti-PD-1 therapy
 - In sorafenib exposed tissue samples, there was an association of PD-L1 expression and NK cell-mediated cytotoxicity signatures with clinical outcomes from tislelizumab monotherapy
- Elevated angiogenesis, immune checkpoint expression, and cell cycle signatures were observed in distinct non-responder subgroups of tislelizumab monotherapy, which may indicate resistance to single-agent PD-1 inhibitors and may potentially identify novel combination treatment strategies
- These findings increase our understanding of PD-L1 expression levels and tumor-immune profiles in HCC that are associated with the clinical efficacy of anti-PD-1 monotherapy
 - Further validation will be considered in an ongoing phase 3 study (BGB-A317-301, RATIONALE 301 [NCT03412773])

Figure 5: Progression-Free Survival and Overall Survival of Responder and Non-Responder Subgroups of Tislelizumab in HCC

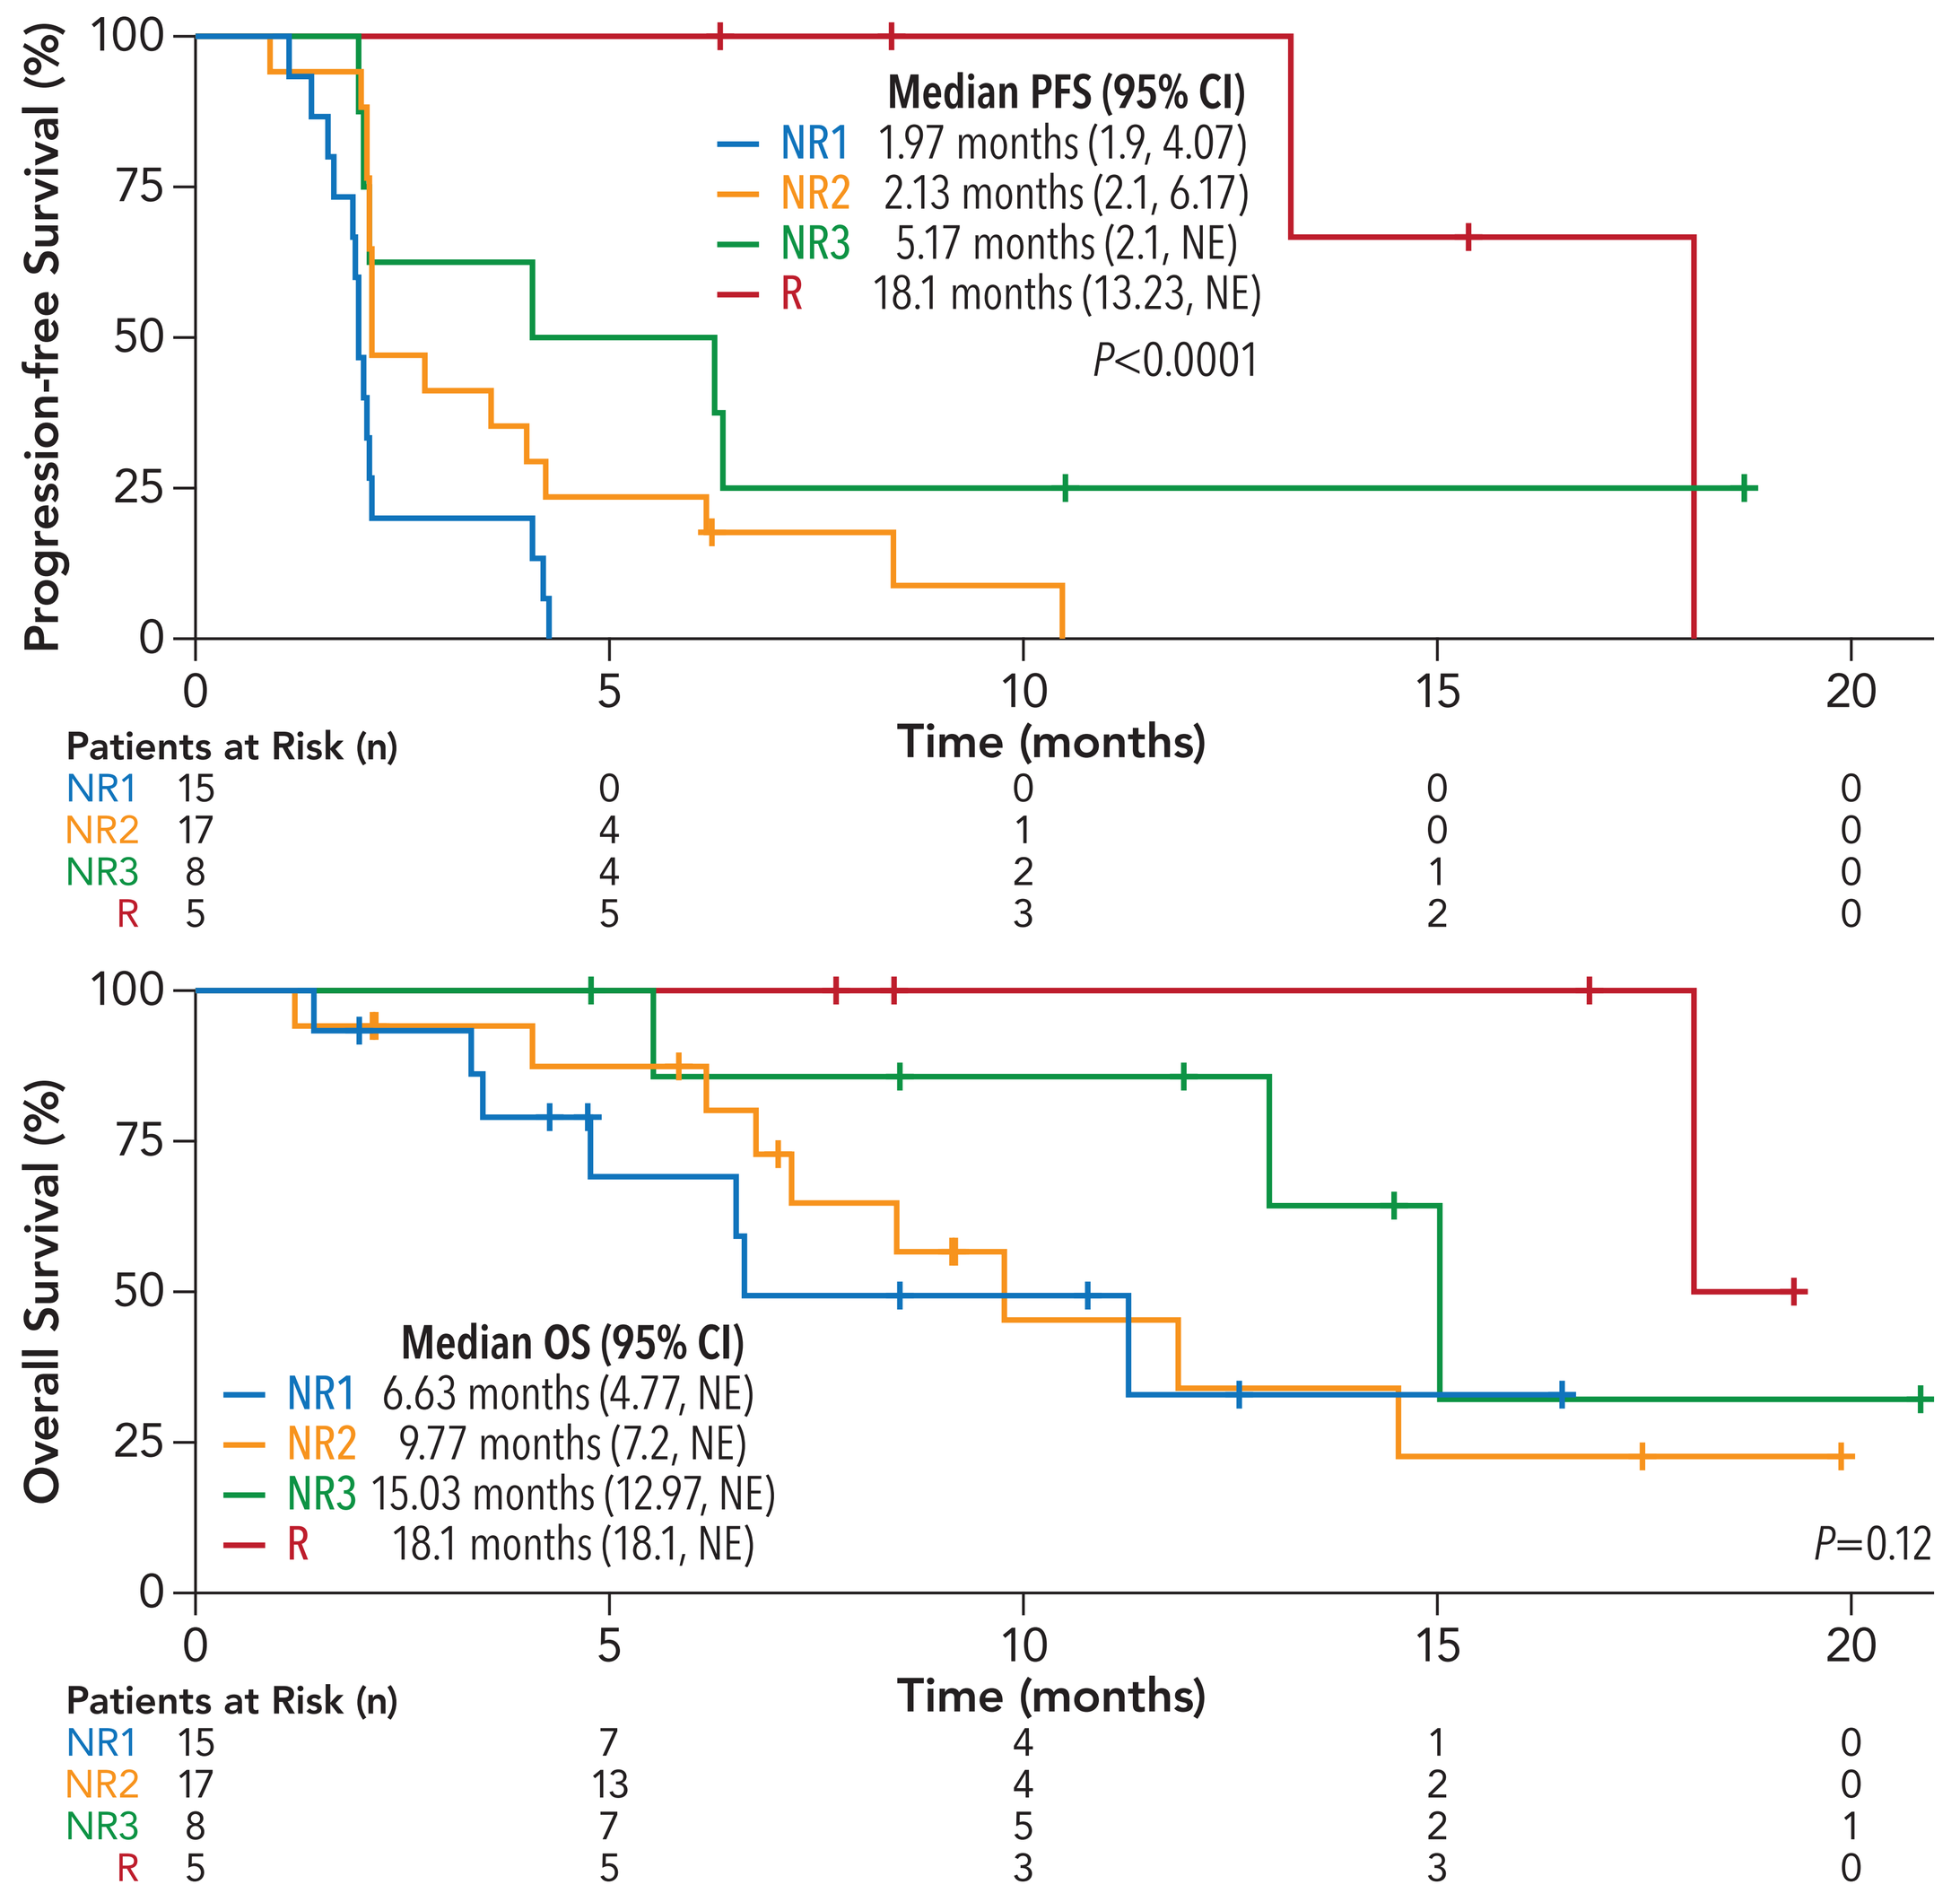
Progression-free Survival (%)
Median PFS (95% CI)
NR1 1.97 months (1.9, 4.07)
NR2 2.13 months (2.1, 6.17)
NR3 5.17 months (2.1, NE)
R 18.1 months (13.23, NE)
P<0.0001
Patients at Risk (n)
Time (months)
NR1 15 0 0 0 0
NR2 17 4 1 0 0
NR3 8 4 2 1 0
R 5 5 3 2 0
Overall Survival (%)
Median OS (95% CI)
NR1 6.63 months (4.77, NE)
NR2 9.77 months (7.2, NE)
NR3 15.03 months (12.97, NE)
R 18.1 months (18.1, NE)
P=0.12
Patients at Risk (n)
Time (months)
NR1 15 7 4 1 0
NR2 17 13 4 2 0
NR3 8 7 5 2 1
R 5 5 3 3 0

Abbreviations: CI, confidence interval; HCC, hepatocellular carcinoma; NE, not evaluable; NR, non-responder; OS, overall survival; PFS, progression-free survival; R, responder.

REFERENCES

1. Iñarrairaegui M, Melero I, Sangro B. Immunotherapy of hepatocellular carcinoma: facts and hopes. *Clin Cancer Res*. 2018;24(7):1518-1524. doi:10.1158/1078-0432.CCR-17-0289
2. Cheng AL, Hsu C, Chan SL, et al. Challenges of combination therapy with immune checkpoint inhibitors for hepatocellular carcinoma. *J Hepatol*. 2020;72(2):307-319. doi:10.1016/j.jhep.2019.09.025
3. Raoul JL, Kudo M, Finn RS, et al. Systemic therapy for intermediate and advanced hepatocellular carcinoma: Sorafenib and beyond. *Cancer Treat Rev*. 2018;68:16-24. doi:10.1016/j.ctrv.2018.05.006
4. Finn RS, Ryoo BY, Merle P, et al. Pembrolizumab as second-line therapy in patients with advanced hepatocellular carcinoma in KEYNOTE-240: A randomized, double-blind, phase III trial. *J Clin Oncol*. 2020;38(3):193-202. doi:10.1200/JCO.19.01307
5. El-Khoueiry AB, Sangro B, Yau T, et al. Nivolumab in patients with advanced hepatocellular carcinoma (CheckMate 040): an open-label, non-comparative, phase 1/2 dose escalation and expansion trial. *Lancet*. 2017;389(10088):2492-2502. doi:10.1016/S0140-6736(17)31046-2.
6. Finn RS, Qin S, Masafumi I, et al. Atezolizumab plus bevacizumab in unresectable hepatocellular carcinoma. *N Engl J Med*. 2020;382 (20):1894-1905. doi:10.1056/NEJMoa1915745.
7. Lu LC, Lee YH, Chang CJ, et al. Increased expression of programmed death-ligand 1 in infiltrating immune cells in hepatocellular carcinoma tissues after sorafenib treatment. *Liver Cancer*. 2019;8(2):110-120. doi:10.1159/000489021.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff, the study patients, and their families. BeiGene, Ltd. provided financial support for this presentation, including editorial assistance by Agnieszka Laskowski, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com