AdvanTIG-105: Phase 1b dose-expansion study of ociperlimab plus tislelizumab in patients with metastatic NSCLC

Rajiv Kumar*†,[1] Se Hyun Kim*,[2] Shun Lu,[3] Ying Cheng,[4] Ming Chen,[5] EunKyung Cho,[6] Tim Clay,[7] Gyeong-Won Lee,[8] Meili Sun,[9] Byoung Yong Shim,[10] David R. Spigel,[11] Tsung-Ying Yang,[12] Qiming Wang,[13] Gee-Chen Chang,[14] Guohua Yu,[15] Ruihua Wang,[16] Wei Tan,[16] Hao Zheng,[17] Rang Gao,[16] Hye Ryun Kim‡[18]

[1]New Zealand Clinical Research, Christchurch, New Zealand and Department of Pathology, University of Otago, Dunedin, New Zealand; [2]Division of Hematology and Medical Oncology, Department of Internal Medicine, Seoul National University Bundang Hospital, Seoul National University College of Medicine, Seongnam, Korea; [3]Shanghai Lung Cancer Center, Shanghai Chest Hospital, Shanghai Jiao Tong University, Shanghai, China; [4]Department of Thoracic Oncology, Jilin Cancer Hospital, Changchun, China; [5]Department of Bioinformatics, State Key Laboratory of Plant Physiology and Biochemistry, College of Life Sciences, Zhejiang University Cancer Hospital, Hangzhou, China; [6]Gil Medical Center, Gachon University College of Medicine, Incheon, Korea; [7]Department of Medical Oncology, St John of God Subaico Hospital, Western Australia, Australia; [8]Division of Hematology and Oncology, Department of Internal Medicine, Gyeongsang National University Hospital, Gyeongsang National University College of Medicine, Jinju, Korea; [9]Department of Oncology, Jinan Central Hospital Affiliated to Shandong First Medical University, Jinan, China; [10]Division of Medical Oncology, Department of Internal Medicine, St. Vincent's Hospital, The Catholic University of Korea, Suwon, Korea; [11]Sarah Cannon Research Institute (SCRI)/Tennessee Oncology, PLLC, Nashville, TN, USA; [12]Division of Chest Medicine, Department of Internal Medicine, Taichung Veterans General Hospital, Taichung, Taiwan; [13]Department of Internal Medicine, Affiliated Cancer Hospital of Zhengzhou University, Henan Cancer Hospital, Zhengzhou, China; [14]Division of Pulmonary Medicine, Department of Internal Medicine, Chung Shan Medical University Hospital, Taichung, Taiwan; [15]Oncology Department, Weifang People's Hospital, Weifang Medical University, Weifang, China; [16]BeiGene (Shanghai) Co., Ltd., Shanghai, China; [17]BeiGene USA, Inc., San Mateo, CA, USA; [18]Division of Medical Oncology, Department of Internal Medicine, Yonsei Cancer Centre, Yonsei University College of Medicine, Seoul, Korea.

*Co-first author; †Presenting author; ‡Corresponding author

Poster No: EP08.01-073 presented at WCLC 2022, Vienna, Austria

Conclusions

Ociperlimab plus tislelizumab demonstrated antitumor activity as first-line treatment for patients with metastatic NSCLC with PD-L1 positive tumors (TC ≥1%).

Antitumor activity was observed in patients with tumors with PD-L1 TC 1-49% and PD-L1 TC ≥50%, with a higher response rate in patients with high PD-L1 TC ≥50%.

The combination of ociperlimab plus tislelizumab had an acceptable safety profile, with most TEAEs being grade 1 or 2 in severity.

Background

Programmed cell death protein 1 (PD-1)/programmed death-ligand 1 (PD-L1) inhibitors have improved outcomes for patients with non-small cell lung cancer (NSCLC), however unmet needs remain.[1]

Inhibition of T-cell immunoreceptor with immunoglobulin and immunoreceptor tyrosine-based inhibitory motif domains (TIGIT) in combination with PD-1/PD-L1 inhibition has demonstrated early efficacy in NSCLC.[2-4]

Ociperlimab is a humanized Fc-intact IgG1 monoclonal antibody (mAb) designed to bind to TIGIT with high specificity and affinity.[5] Tislelizumab is an anti-PD-1 mAb approved for the treatment of NSCLC in China.[6]

In the ongoing phase 1/1b, open-label AdvanTIG-105 dose-escalation/-expansion (NCT04047862) study, ociperlimab plus tislelizumab was well tolerated in patients with advanced, unresectable solid tumors.[7]

Methods

- The recommended phase 2 dose was ociperlimab 900 mg intravenously (IV) every three weeks (Q3W) plus tislelizumab 200 mg IV Q3W in the dose-escalation part of the study[7]
- Here we report data from the dose-expansion part (Cohort 3) of the phase 1/1b AdvanTIG-105 study, in patients with metastatic NSCLC (**Figure 1**)

Figure 1. AdvanTIG-105 study design (Cohort 3)

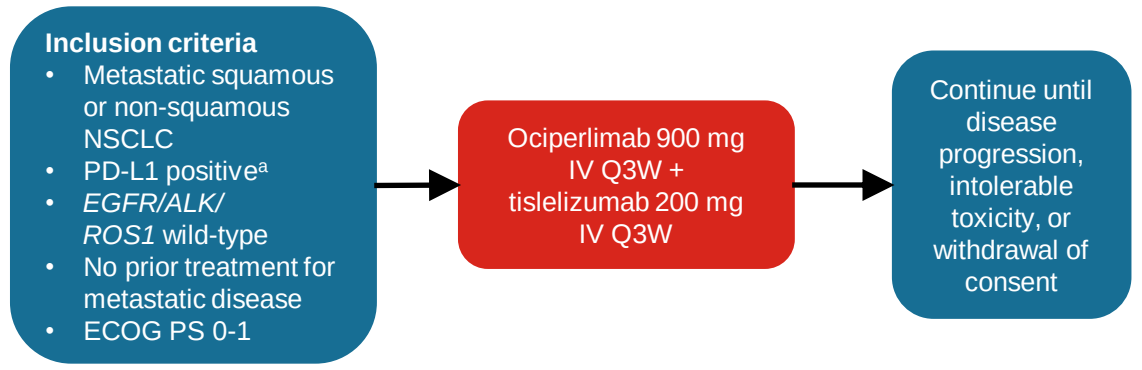
Inclusion criteria
Metastatic squamous or non-squamous NSCLC
PD-L1 positive[a]
EGFR/ALK/ ROS1 wild-type
No prior treatment for metastatic disease
ECOG PS 0-1
Ociperlimab 900 mg IV Q3W + tislelizumab 200 mg IV Q3W
Continue until disease progression, intolerable toxicity, or withdrawal of consent

Primary endpoint:
- Investigator-assessed ORR per RECIST v1.1

Key secondary endpoints:
- Investigator-assessed PFS, DoR, and DCR per RECIST v1.1
- Safety
- Correlation of PD-L1 expression with efficacy endpoints

Key exploratory endpoint:
- OS

[a]TC ≥1% by VENTANA PD-L1 (SP263) assay by central lab.
Abbreviations: ALK, anaplastic lymphoma kinase; DCR, disease control rate; DoR; duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; IV, intravenously; NSCLC, non-small cell lung cancer; ORR, objective response rate; PD-L1, programmed death-ligand 1; PFS, progression-free survival; Q3W, every three weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; ROS1, c-ros oncogene 1; TC, tumor cell.

Results

Baseline characteristics

- As of April 5, 2022, 40 patients were enrolled in Cohort 3 and comprised the safety analysis set, who received at least one dose of the study drug
- The median age was 65.0 years (range 46-81), and 32.5% of patients were female
- In total, 35.9% (14/39) of patients were PD-L1 TC ≥50%
- The median study follow-up was 28.1 weeks (range 3.1-61.7)

Efficacy

- In total, 39 patients were evaluable for efficacy
- The unconfirmed ORR was 53.8% (95% CI: 37.2, 69.9). In patients with PD-L1 TC 1-49% and PD-L1 TC ≥50% subgroups, the unconfirmed ORR was 44.0% and 71.4%, respectively (**Table 1**)
- The median DoR was not evaluable (NE) (**Table 1**), and the median PFS was 5.4 months (95% CI: 4.2, NE), with 5.2 months and 5.6 months in the PD-L1 TC 1-49% and PD-L1 TC ≥50% subgroups, respectively
- The best change in target lesions and the duration of treatment and response are shown in **Figures 2** and **3**, respectively

Safety

- The safety profiles of ociperlimab and tislelizumab are shown in **Table 2**

Table 1. Summary of antitumor activity

	PD-L1 TC 1-49% (n=25)	PD-L1 TC ≥50% (n=14)	Total (N=39)
Unconfirmed ORR, n (%) (95% CI)	11 (44.0) (24.4, 65.1)	10 (71.4) (41.9, 91.6)	21 (53.8) (37.2, 69.9)
Unconfirmed BOR, n (%)[a]			
CR	0 (0)	1 (7.1)	1 (2.6)
PR	11 (44.0)	9 (64.3)	20 (51.3)
SD	11 (44.0)	3 (21.4)	14 (35.9)
PD	2 (8.0)	1 (7.1)	3 (7.7)
DCR, n (%)[b]	22 (88.0)	13 (92.9)	35 (89.7)
Median DoR, months (95% CI)[b]	NE (2.2, NE)	NE	NE (4.2, NE)

[a]One patient in the PD-L1 1-49% group was NE. This patient had symptoms which were assessed as disease progression, but was not considered to have radiological progression; [b]Confirmed.
Abbreviations: BOR, best overall response; CI, confidence interval; CR, complete response; DCR, disease control rate; DoR, duration of response; NE, not evaluable; ORR, objective response rate; PD, progressive disease; PD-L1, programmed-death ligand 1; PR, partial response; SD, stable disease; TC, tumor cell.

Figure 2. Best change in target lesions

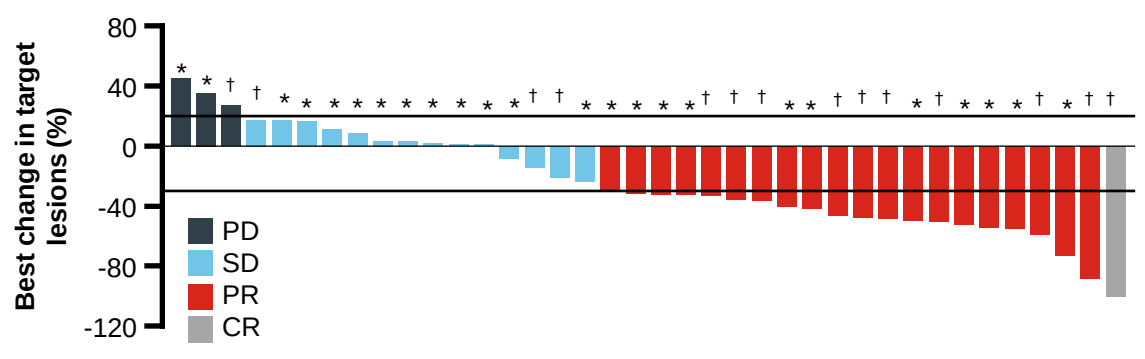
Best change in target lesions (%)
80
40
0
-40
-80
-120
PD
SD
PR
CR

*These patients were PD-L1 TC 1-49%; †These patients were PD-L1 TC ≥50%.
Abbreviations: CR, complete response; PD, progressive disease; PD-L1, programmed death-ligand 1; PR, partial response; SD, stable disease; TC, tumor cell.

Figure 3. Duration of treatment and response

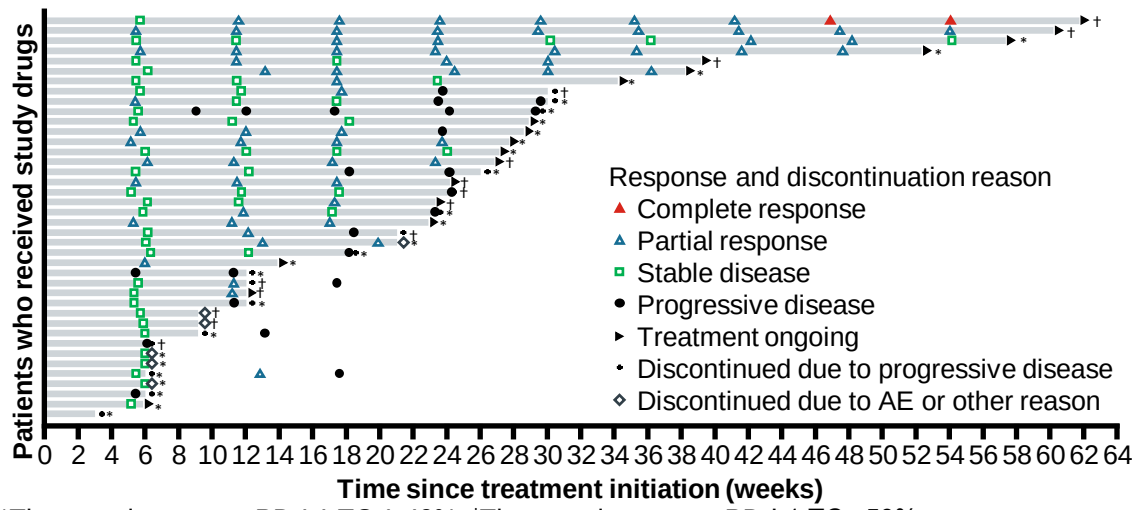
Patients who received study drugs
Time since treatment initiation (weeks)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44 46 48 50 52 54 56 58 60 62 64
Response and discontinuation reason
Complete response
Partial response
Stable disease
Progressive disease
Treatment ongoing
Discontinued due to progressive disease
Discontinued due to AE or other reason

*These patients were PD-L1 TC 1-49%; †These patients were PD-L1 TC ≥50%.
Abbreviations: AE, adverse event; PD-L1, programmed-death ligand 1; TC, tumor cell.

Table 2. Summary of TEAEs and TRAEs (safety analysis set)

Patients, n (%)	TEAEs (N=40)	TRAEs (N=40)
Patients with at least one AE	38 (95.0)[a]	31 (77.5)
≥ Grade 3 AE	11 (27.5)[b]	4 (10.0)[c]
Serious AE	10 (25.0)	4 (10.0)
AE leading to ociperlimab dose modification	17 (42.5)	–
AE leading to tislelizumab dose modification	17 (42.5)	–
AE leading to ociperlimab discontinuation	3 (7.5)	1 (2.5)
AE leading to tislelizumab discontinuation	3 (7.5)	1 (2.5)
AE leading to death	1 (2.5)	0 (0)

[a]The most common TEAEs were pruritus (32.5%), pyrexia (30.0%), decreased appetite (20.0%), rash (20.0%), anemia (17.5%), nausea (17.5%), and dyspnea (17.5%); [b]The most common ≥ grade 3 TEAEs were pneumonia (7.5%) and anemia (5.0%); [c]The TRAEs were hyperglycemia, lipase increased, pruritis, acute kidney injury, and pneumonia.
Abbreviations: AE, adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

References

1. De Giglio A, et al. *Current Onc Rep*. 2021;23:126; 2. Rodriguez-Abreu D, et al. *J Clin Oncol*. 2020 (Abs 9503) [presented at ASCO 2020]; 3. Niu J, et al. *Ann Oncol*. 2020 (Abs 1410P) [presented at ESMO 2020]; 4. Ahn M-J, et al. *Ann Oncol*. 2020 (Abs 1400P) [presented at ESMO 2020]; 5. Chen X, et al. Data presented at AACR 2021. Poster 1854; 6. BeiGene. China NMPA approves tislelizumab as second- or third-line treatment for patients with locally advanced or metastatic non-small cell lung cancer. Available at: https://ir.beigene.com/news-details/?id=3e337eaa-a5f6-4368-95e0-3e0d35a71254. Accessed March 2022; 7. Frentzas S, et al. *J Clin Oncol*. 2021 (Abs 2583) [presented at ASCO 2021].

Acknowledgments

This study was sponsored by BeiGene, Ltd. Medical writing support, under direction of the authors, was provided by Adeline Lum Nde, PhD, of Ashfield MedComms, an Inizio company, and was funded by BeiGene, Ltd.

Disclosures

SHK: Astellas, AstraZeneca, BNS/Ono Pharma, Roche. **SL**: AstraZeneca, BeiGene, BMS, GenomiCare, Hansoh, Heng Rui, Hengrui Therapeutics, Hutchison, InventisBio Co. Ltd., Menarini, Pfizer, Roche, Yuhan Corporation, ZaiLab. **TC**: Abbvie, Amgen, Astellas, AstraZeneca, BeiGene, BMS, Clovis, Daiichi Sankyo, Foundation Medicine, Immutep, Janssen, MSD, Pfizer. **DRS:** Aeglea Biotherapeutics, Agios, Amgen, Apollomics, Arcus, Arrys Therapeutics, Astellas, AstraZeneca, Bayer, BeiGene, BIND Therapeutics, BioNTech RNA Pharmaceuticals, Blueprint Medicine, BMS, Boehringer-Ingelheim, Bristol-Myers Squibb, Calithera, Celgene, Celldex, Clovis, Curio Science, Cyteir Therapeutics, Daiichi Sankyo, Denovo Biopharma, Eisai, Elevation Oncology, EMD Serono, Evelo Biosciences, Evidera, Exelixis, G1 Therapeutics, Genentech/Roche, GlaxoSmithKline, GRAIL, Hutchison MediPharma, ImClone Systems, ImmunoGen, Incyte, Intellisphere, Ipsen, Janssen, Jazz Pharmaceuticals, Kronos Bio, Lilly, Loxo Oncology, MacroGenics, MedImmune, Merck. Molecular Partners, Mirati Therapeutics, Molecular Templates, Neon Therapeutics, Novartis, Novocure, Oncologie, Pfizer, PTC Therapeutics, Puma Biotechnology, PureTech Health, Razor Genomics, Regeneron, Repare Therapeutics, Rgenix, Sanofi-Aventi, Takeda, Tesaro, Tizona Therapeutics, Transgene, UT Southwestern, Verastem. **G-CC**: AstraZeneca, Boehringer-Ingelheim, Bristol-Myers Squibb, Company Oncology, Eli Lilly, F. Hoffmann-La Roche, Ltd, Merck Sharp & Dohme, Novartis, Pfizer. **RW**, **WT**, **HZ** and **RG**: employees of BeiGene. All other authors have declared no competing interests.

***Author contact details:**
Rajiv.Kumar@nzcr.co.nz (Rajiv Kumar)

Poster recording