Abstract Number: 445

2018 Genitourinary Cancers Symposium
February 8–10, 2018
San Francisco, CA, USA

BeiGene

PRELIMINARY RESULTS FROM PATIENTS WITH UROTHELIAL CARCINOMA IN A PHASE 1A/1B STUDY OF TISLELIZUMAB (BGB-A317), AN ANTI-PD-1 MONOCLONAL ANTIBODY

Shahneen Sandhu[1], Andrew Hill[2], Hui Gan[3], Michael Friedlander[4], Mark Voskoboynik[5], Paula Barlow[6], Amanda Townsend[7], James Song[8], Yun Zhang[9], Zhirong Shen[9], Qinzhou Qi[9], Jayesh Desai[1]

[1]Peter MacCallum Cancer Centre, Melbourne, Australia; [2]Tasman Oncology Research Ltd, Queensland, Australia; [3]Austin Health and Olivia Newton-John Cancer Research Institute, Melbourne, Australia; [4]The Prince of Wales Hospital, Randwick, Australia; [5]Nucleus Network, Melbourne, Australia; [6]Auckland City Hospital, Auckland, New Zealand; [7]The Queen Elizabeth Hospital, Woodville, Australia; [8]BeiGene USA, Inc., Fort Lee, NJ, USA; [9]BeiGene (Beijing) Co. Ltd., Beijing, China

INTRODUCTION

- Advances in treatment of metastatic urothelial carcinoma (UC) have stagnated over the past 3 decades; however, the monoclonal antibodies (mAbs) against immune checkpoint inhibitory receptors, like programmed cell death-1 (PD-1), have demonstrated promising antitumor activity across multiple malignancies,[1] including UC[2–7]
- PD-1 is relatively overexpressed on $CD8^+$ effector, tumor-infiltrated T lymphocytes (TILs); anti-PD-1 antibodies induce an increase in $CD8^+$ T cell percentages within the tumor microenvironment[8]
- In vivo evidence has shown that anti-PD-1 antibodies demonstrate reduced tumor cytotoxicity when the Fc domain of the antibody engages with Fc-gamma receptors (FcγRs)[9]
 - FcγR engagement results in preferential depletion of these $CD8^+$ TILs;[8] this decrease may correlate with the dampening of anti-PD-1 tumor activity
- Tislelizumab (BGB-A317) is a humanized IgG4 mAb with high affinity and binding specificity against PD-1
- Tislelizumab was specifically engineered to minimize FcγR binding on macrophages and myeloid-derived suppressor cells; abrogation of FcγR binding may improve clinical activity by preserving activated T cells (**Figure 1**)

Figure 1: Lack of FcγR Binding Prevents Macrophage-Mediated T-Cell Clearance

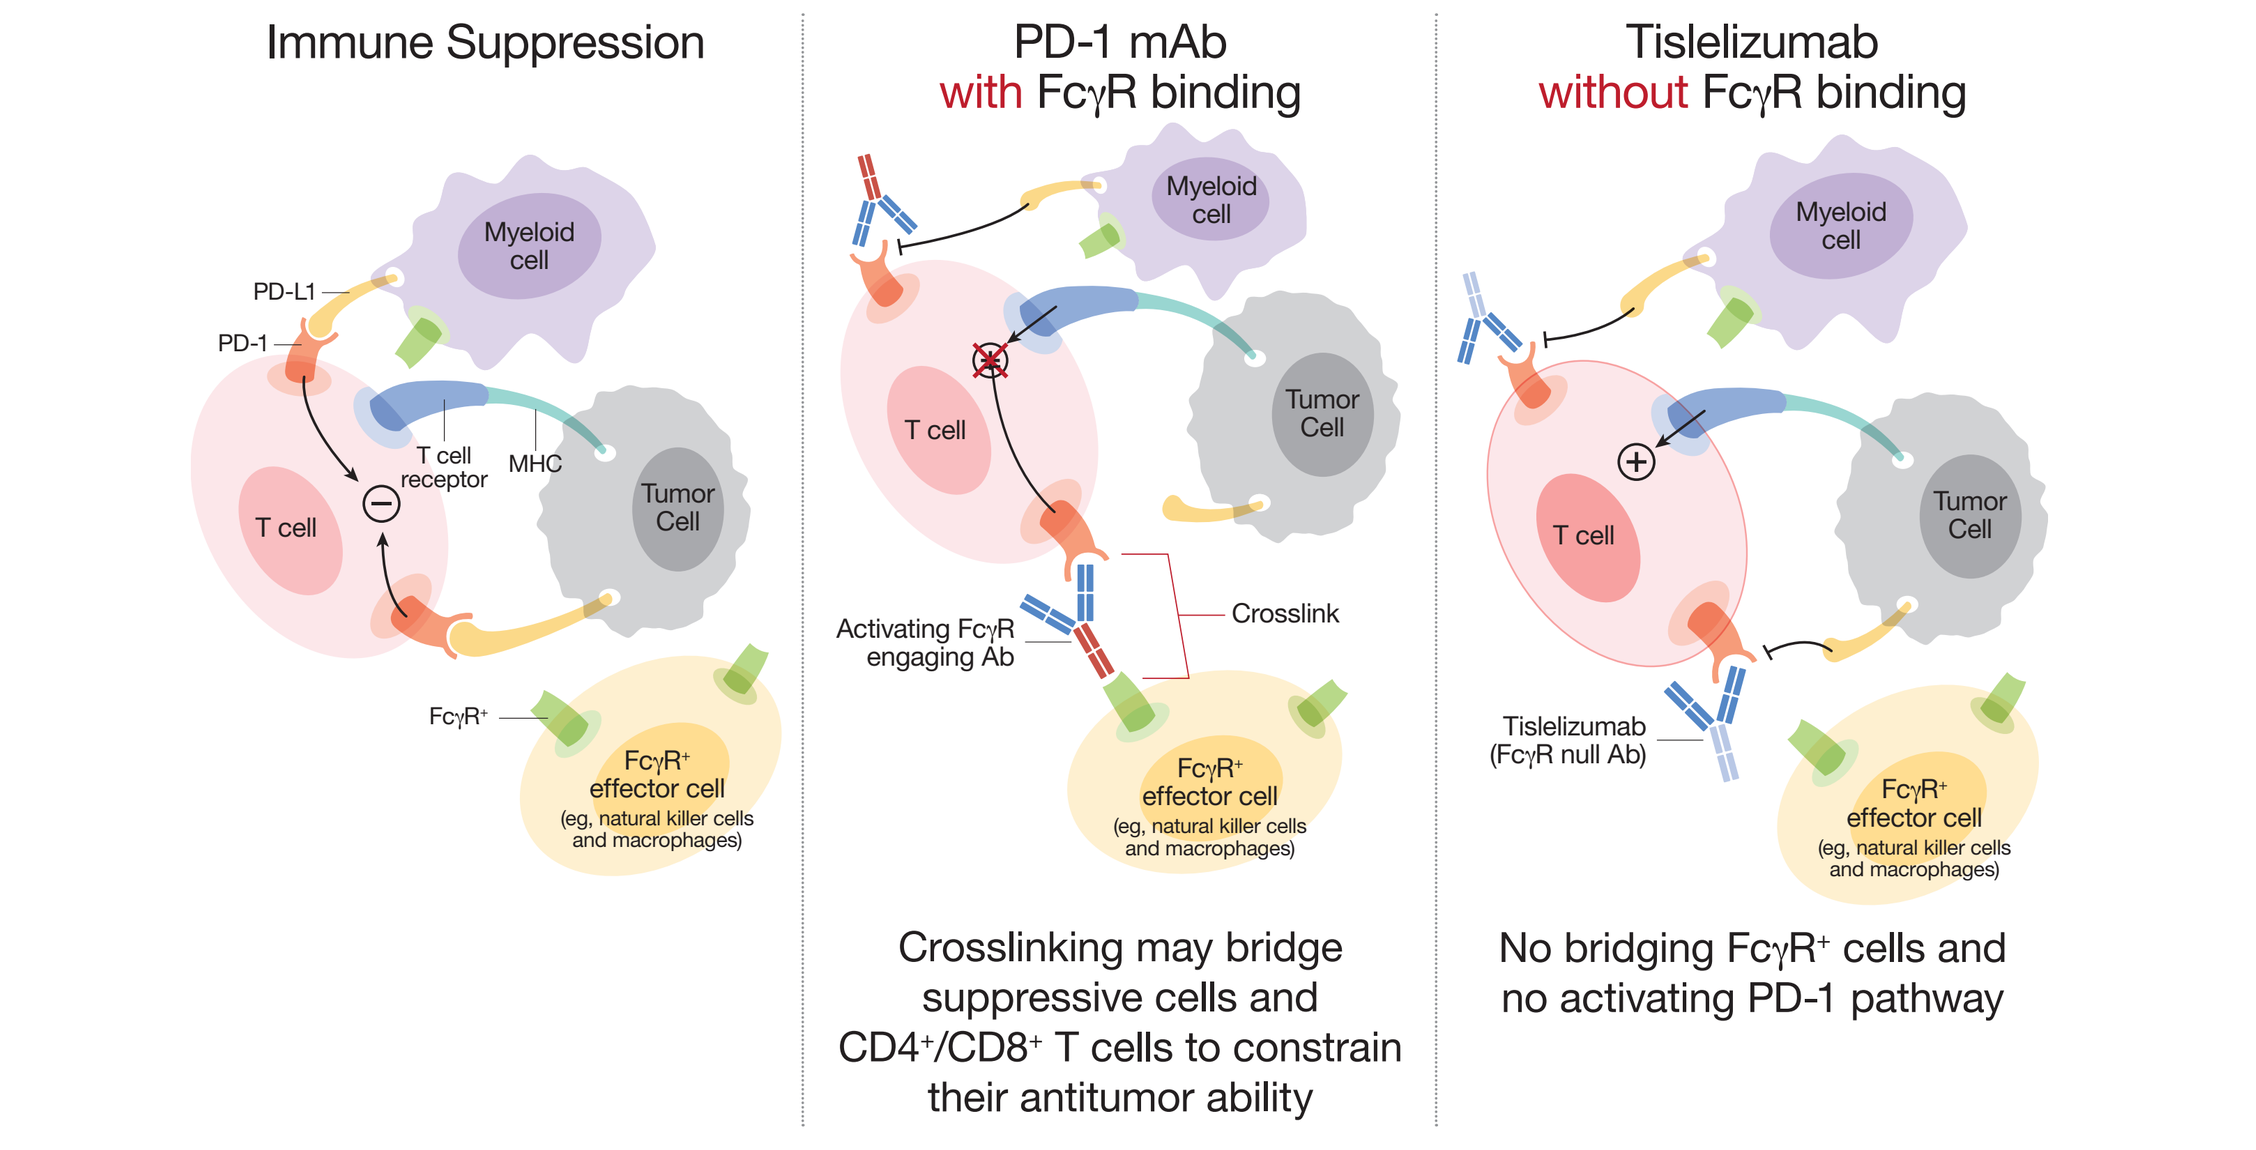

Immune Suppression
PD-1 mAb with FcγR binding
Tislelizumab without FcγR binding
Myeloid cell
PD-L1
PD-1
T cell
T cell receptor
MHC
Tumor Cell
FcγR+
FcγR+ effector cell (eg, natural killer cells and macrophages)
Activating FcγR engaging Ab
Crosslink
Tislelizumab (FcγR null Ab)
Crosslinking may bridge suppressive cells and CD4+/CD8+ T cells to constrain their antitumor ability
No bridging FcγR+ cells and no activating PD-1 pathway

Abbreviations: Ab, antibody; CD, cluster of differentiation; FcγR, Fc-gamma receptor; mAb, monoclonal antibody; MHC, major histocompatibility complex; PD-1, programmed cell death-1; PD-L1, programmed death-ligand 1.

- Previous reports from an ongoing Phase 1A/1B study (NCT02407990)[10] of tislelizumab in patients with advanced solid tumors suggested that tislelizumab is generally well tolerated
- In addition, the toxicity profile of tislelizumab demonstrates that adverse events (AEs) are generally of low-to-moderate severity, manageable, and reversible[11]
- Here, we present the preliminary results of patients with UC enrolled in this Phase 1A/1B study; the trial is ongoing to collect more mature safety and antitumor activity data

METHODS

- The study design is detailed in **Figure 2**
 - In Phase 1A, 10 mg/kg once every 2 weeks (Q2W) was the maximum administered dosage of tislelizumab; the maximum tolerated dose (MTD) was not reached
 - All patients in Phase 1B received tislelizumab as a 5 mg/kg intravenous (IV) infusion once every 3 weeks (Q3W)
 - Radiographic assessment was performed every 8 or 9 weeks per Response Evaluation Criteria In Solid Tumors guidelines version 1.1 (RECIST v1.1)

Key Eligibility Criteria of the UC Subset

- Adult patients (aged ≥18 years) with histologically or cytologically confirmed UC who have at least one measurable lesion, as defined per RECIST v1.1; who have received standard therapy but no prior anti-PD-1 or programmed death-ligand 1 (PD-L1) treatment; and an Eastern Cooperative Oncology Group (ECOG) performance status of ≤1 were enrolled
- Patients were excluded if they had a history of severe hypersensitivity reactions to other mAbs
- Patients who had prior malignancy active within the previous 2 years except for UC, and locally curable cancers that have been apparently cured, such as basal or squamous cell skin cancer, superficial bladder cancer, or carcinoma in situ of the cervix or breast, were excluded

Figure 2: Study Design

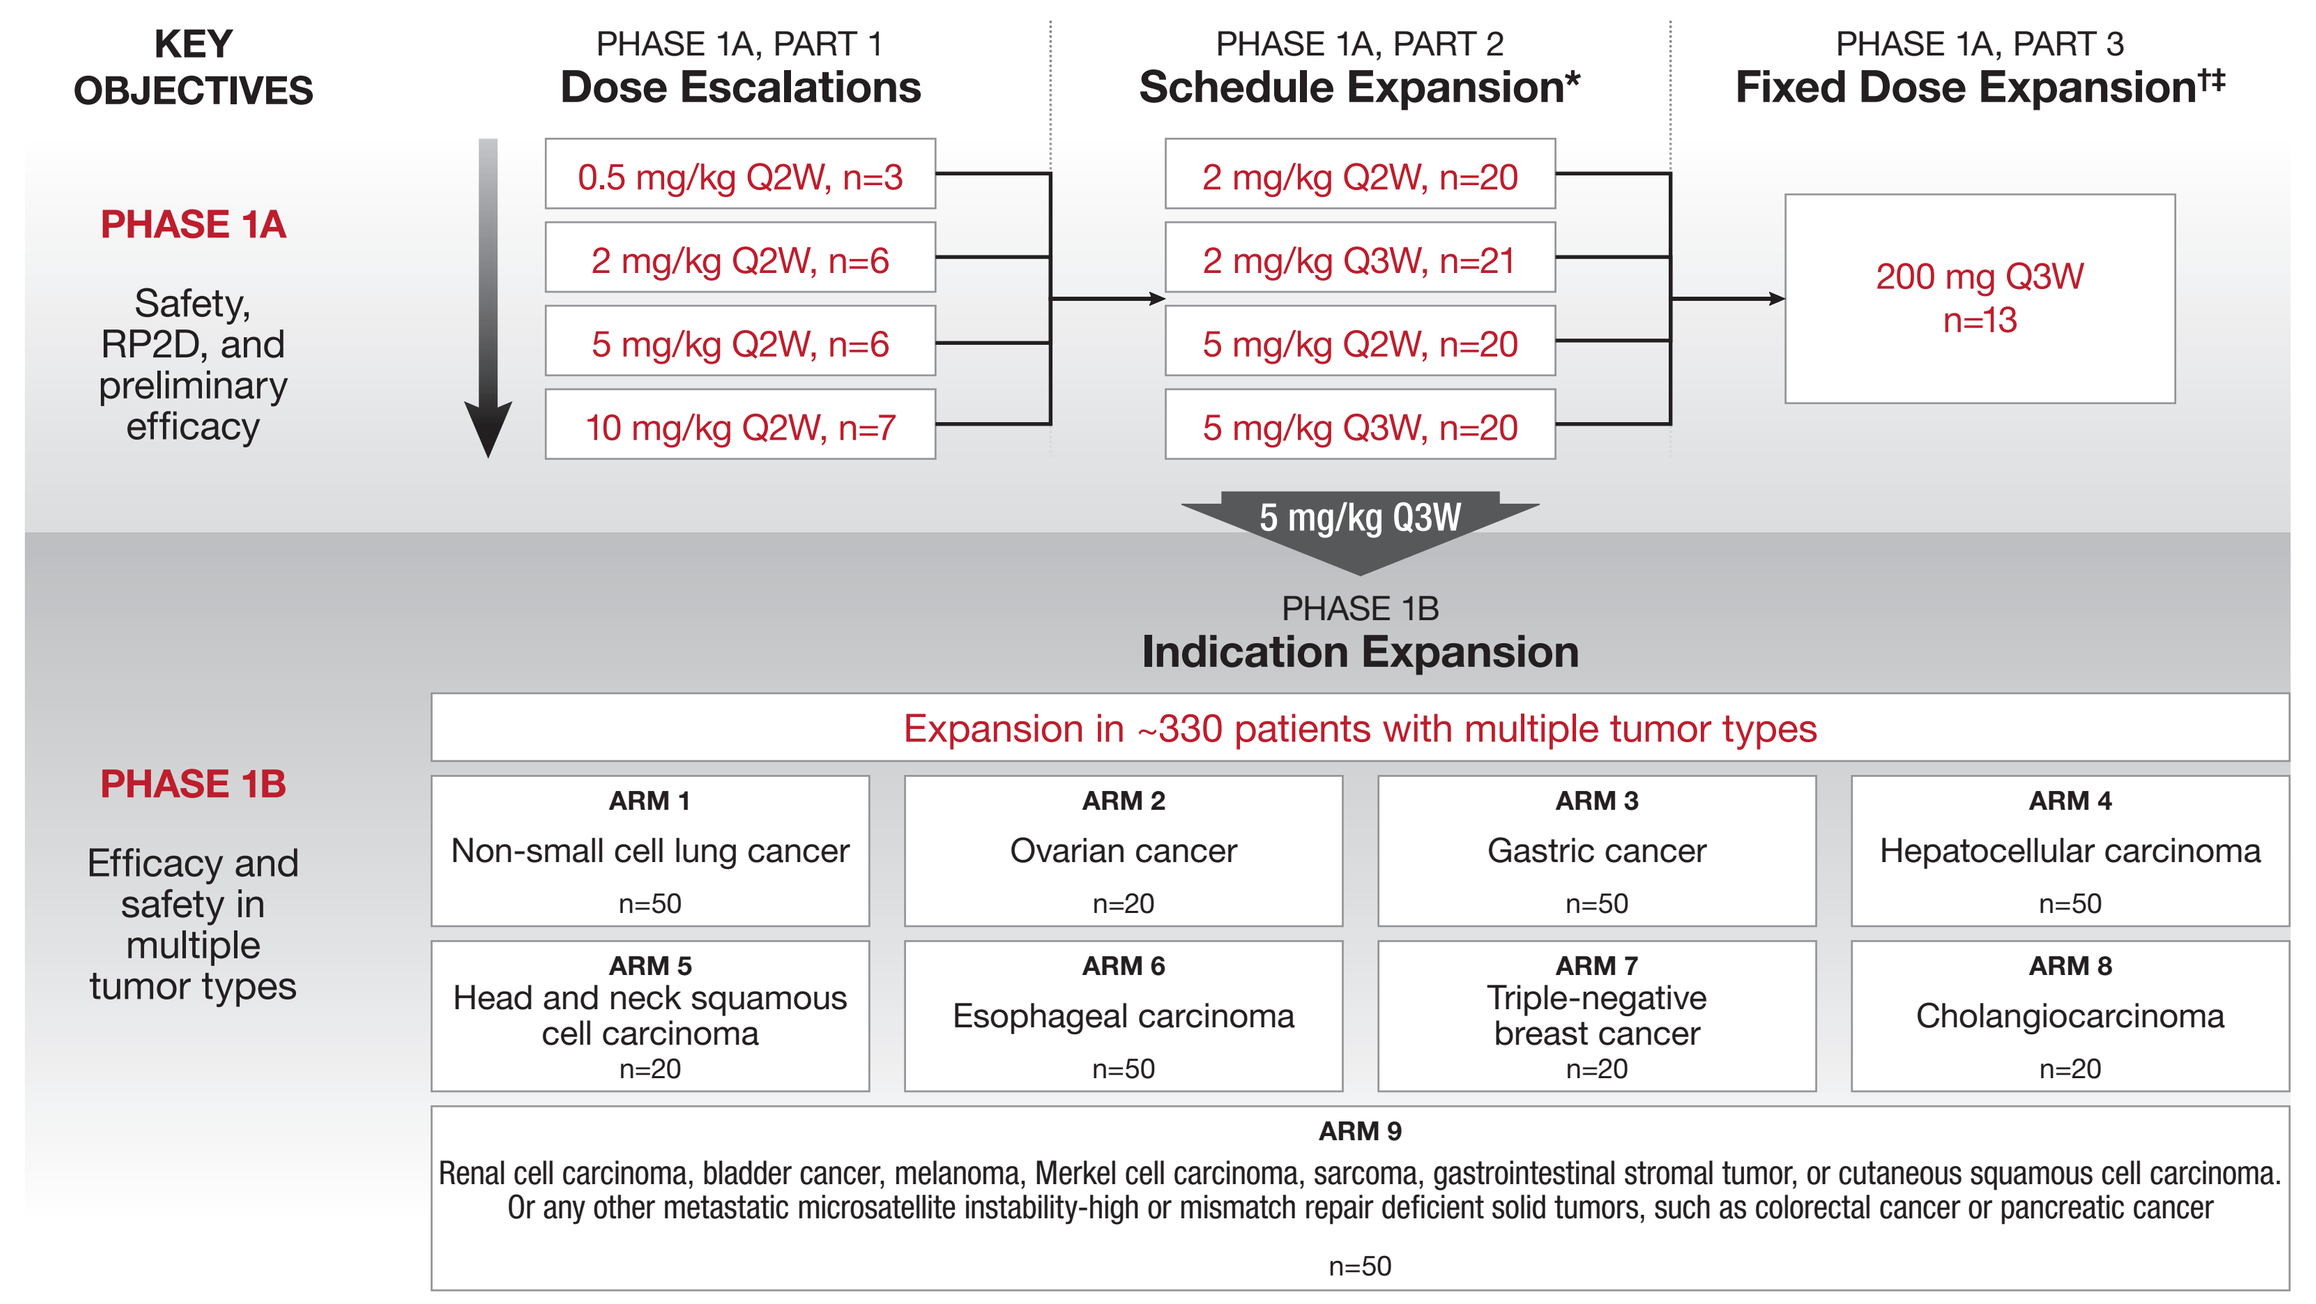

KEY OBJECTIVES
PHASE 1A, PART 1
Dose Escalations
PHASE 1A, PART 2
Schedule Expansion*
PHASE 1A, PART 3
Fixed Dose Expansion†‡
PHASE 1A
Safety, RP2D, and preliminary efficacy
0.5 mg/kg Q2W, n=3
2 mg/kg Q2W, n=6
5 mg/kg Q2W, n=6
10 mg/kg Q2W, n=7
2 mg/kg Q2W, n=20
2 mg/kg Q3W, n=21
5 mg/kg Q2W, n=20
5 mg/kg Q3W, n=20
200 mg Q3W n=13
5 mg/kg Q3W
PHASE 1B
Indication Expansion
PHASE 1B
Efficacy and safety in multiple tumor types
Expansion in ~330 patients with multiple tumor types
ARM 1 Non-small cell lung cancer n=50
ARM 2 Ovarian cancer n=20
ARM 3 Gastric cancer n=50
ARM 4 Hepatocellular carcinoma n=50
ARM 5 Head and neck squamous cell carcinoma n=20
ARM 6 Esophageal carcinoma n=50
ARM 7 Triple-negative breast cancer n=20
ARM 8 Cholangiocarcinoma n=20
ARM 9
Renal cell carcinoma, bladder cancer, melanoma, Merkel cell carcinoma, sarcoma, gastrointestinal stromal tumor, or cutaneous squamous cell carcinoma.
Or any other metastatic microsatellite instability-high or mismatch repair deficient solid tumors, such as colorectal cancer or pancreatic cancer
n=50

*In select tumors for RP2D determination and preliminary differentiation, †In select tumors at fixed doses that do not exceed the exposure of maximum tolerated dose, ‡Conducted in parallel with Phase 1B.

Abbreviations: Q2W, once every 2 weeks; Q3W, once every 3 weeks; RP2D, recommended Phase 2 dose.

- Pretreatment tumor samples were evaluated for PD-L1 membrane expression by immunohistochemistry performed on an automated platform (VENTANA PD-L1 [SP263] assay)
- PD-L1 expression status of any intensity was assessed on tumor cells (TC) and tumor-associated immune cells (IC) and tissue was scored as follows:
 - PD-L1 was defined as high if either ≥25% TC or ≥25% IC express membranous PD-L1
 - PD-L1 was defined as low/negative if both TC and IC had <25% PD-L1 membrane staining

RESULTS

Patient Disposition

- As of 28 August 2017, 16 patients with UC had enrolled in this study (**Table 1**); of these, 12 patients had ≥1 prior systemic anticancer therapy and five patients had prior radiotherapy
 - A total of six patients remain on treatment

Table 1: Patient Demographics and Disease Characteristics of Patients with UC

		UC Population (N=16)
Median age, years (min, max)		71.5 (39, 79)
Sex, n	Male	13
	Female	3
Race, n	White	16
Median treatment duration, months (min, max)		4.3 (0.7, 18.3)
Prior systemic anticancer therapy regimens,* n	1	6
	2	3
	≥3	3

*Four patients received prior treatment in the adjuvant and/or neoadjuvant setting.

Abbreviations: max, maximum; min, minimum; UC, urothelial carcinoma.

Preliminary Antitumor Activity

- As of 28 August 2017, 15 patients with UC were evaluable (defined as having a measurable baseline tumor assessment and at least one evaluable post-baseline tumor response assessment, or had progressed or died prior to the initial tumor assessment)
 - One patient achieved a confirmed complete response (CR), four achieved a confirmed partial response (PR) and three achieved stable disease (SD)
 - Disease control rate (DCR=CR+PR+SD) was 53.3% (n=8/15)
 - Median treatment duration was 4.3 months (range: 0.7–18.3 months)
 - Tumor responses did not appear to be dose dependent
- Antitumor activity of tislelizumab is presented in **Figures 3–5**

Figure 3: Duration of Treatment and Response in Patients with UC (N=16)

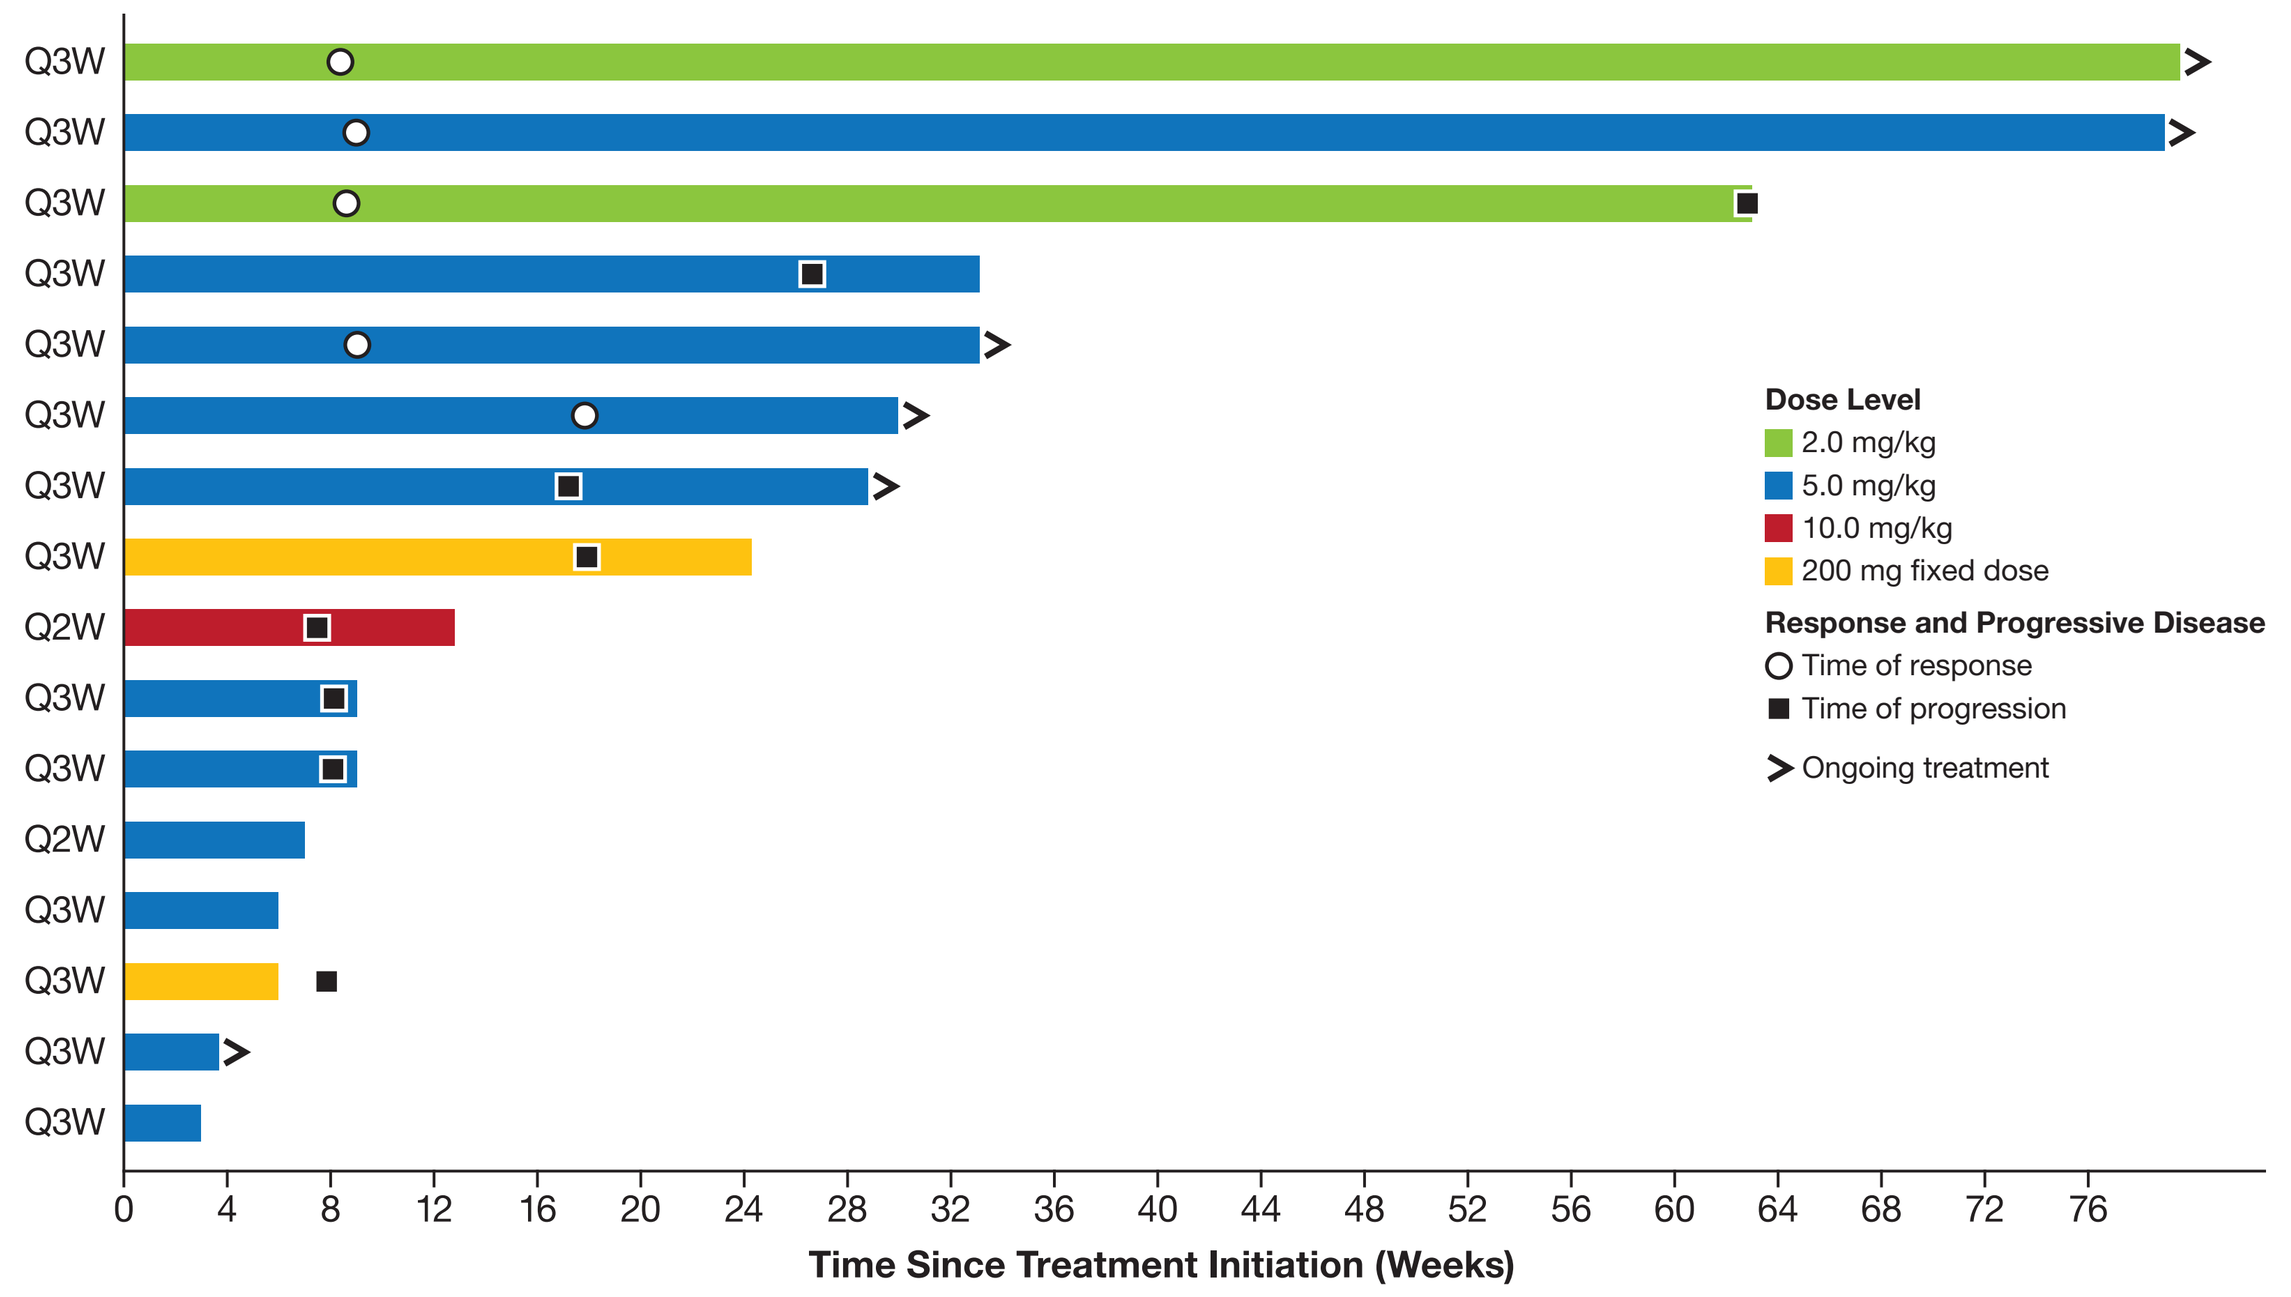

Q3W
Q3W
Q3W
Q3W
Q3W
Q3W
Q3W
Q3W
Q2W
Q3W
Q3W
Q2W
Q3W
Q3W
Q3W
Q3W
Dose Level
2.0 mg/kg
5.0 mg/kg
10.0 mg/kg
200 mg fixed dose
Response and Progressive Disease
Time of response
Time of progression
Ongoing treatment
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76
Time Since Treatment Initiation (Weeks)

Abbreviations: Q2W, once every 2 weeks; Q3W, once every 3 weeks; UC, urothelial carcinoma.

Figure 4: Maximum Tumor Reduction in Evaluable Patients with UC (N=12)*

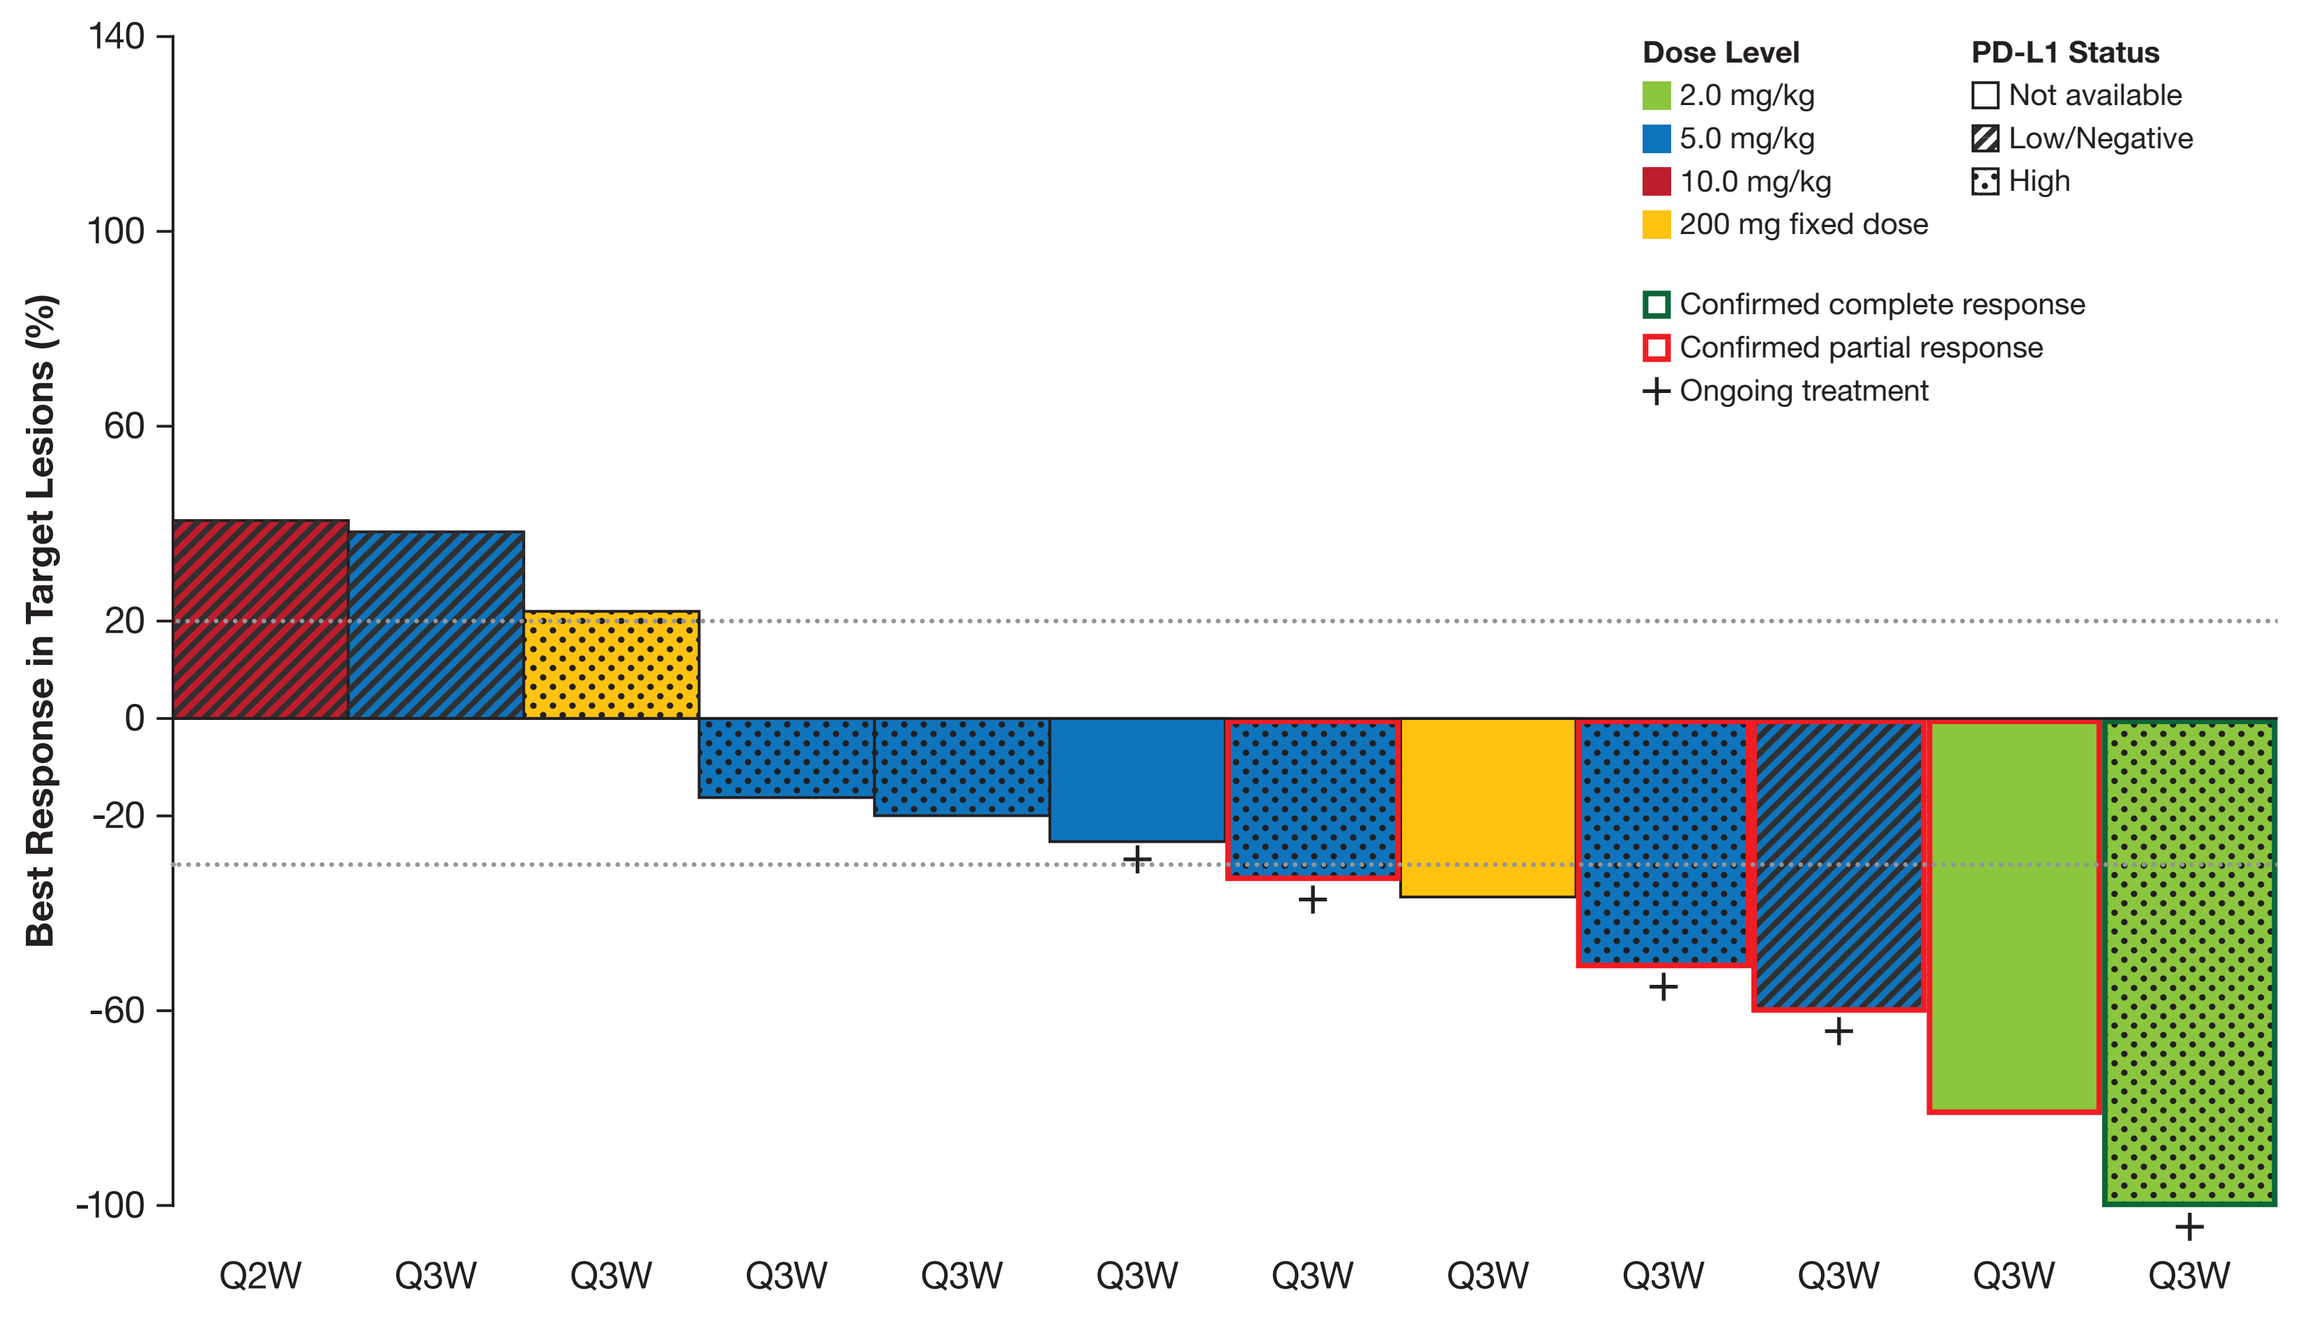

Best Response in Target Lesions (%)
140
100
60
20
0
-20
-60
-100
Dose Level
2.0 mg/kg
5.0 mg/kg
10.0 mg/kg
200 mg fixed dose
PD-L1 Status
Not available
Low/Negative
High
Confirmed complete response
Confirmed partial response
Ongoing treatment
Q2W Q3W Q3W Q3W Q3W Q3W Q3W Q3W Q3W Q3W Q3W Q3W

*Population does not include patients who had progressed or died prior to the initial tumor assessment.

Abbreviations: Q2W, once every 2 weeks; Q3W, once every 3 weeks; UC, urothelial carcinoma.

Figure 5: Change in Target Lesions over Time in Evaluable Patients with UC (N=12)*

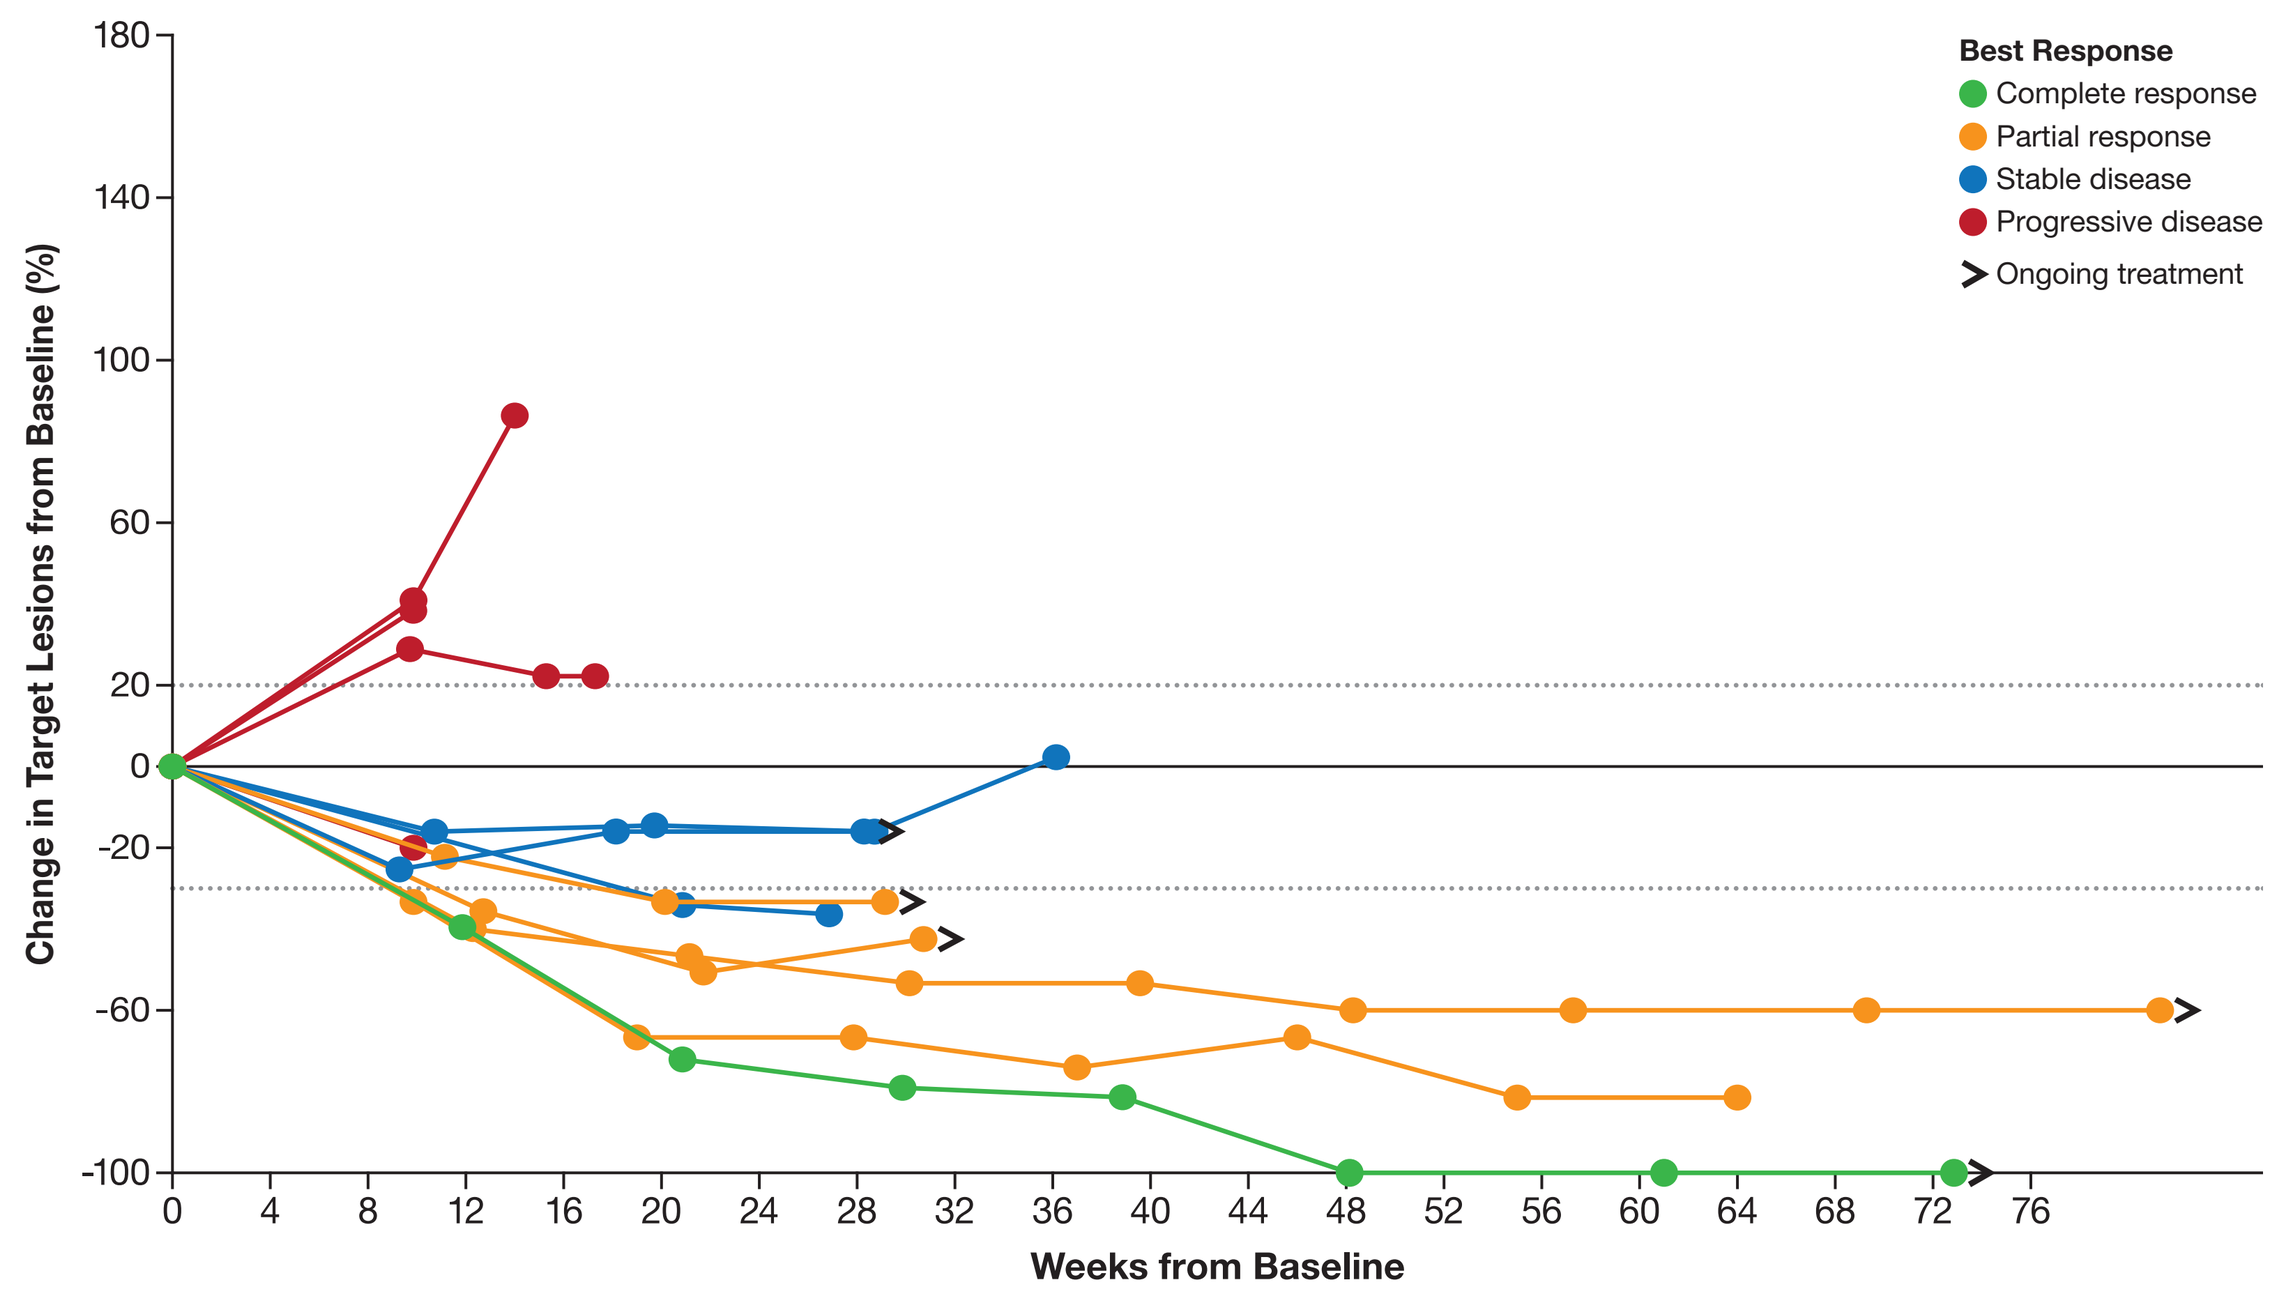

Change in Target Lesions from Baseline (%)
180
140
100
60
20
0
-20
-60
-100
Best Response
Complete response
Partial response
Stable disease
Progressive disease
Ongoing treatment
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76
Weeks from Baseline

*Population does not include patients who had progressed or died prior to the initial tumor assessment.

Abbreviations: UC, urothelial carcinoma.

Response by PD-L1 Status

- As of 4 December 2017, a total of nine patients were evaluable for both PD-L1 status and clinical response
- Clinical responses were observed in patients with both PD-L1 high and PD-L1 low expression (**Table 2** and **Figure 4**)
- Evaluation of PD-L1 expression in tislelizumab-treated patients with UC is ongoing

Table 2: Confirmed Best Overall Response for Each Evaluable Patient by PD-L1 Status

Patient Number	Treatment Start	Treatment End	PD-L1 Status	Confirmed Response	Treatment Ongoing
1	19 Feb 2016	—	High	CR	Yes
2	24 Jan 2017	15 May 2017	High	PD	No
3	29 Nov 2016	31 Jan 2017	High	PD	No
4	9 Jan 2017	—	High	PR	Yes
5	31 Jan 2017	—	High	PR	Yes
6	30 Jun 2016	8 Mar 2017	High	SD	No
7	23 Feb 2016	—	Low/negative	PR	Yes
8	23 Feb 2016	20 Apr 2016	Low/negative	PD	No
9	11 Dec 2015	3 Mar 2016	Low/negative	PD	No
10	25 Nov 2015	8 Feb 2017	Not available*	PR	No
11	8 Feb 2017	—	Not available	SD	Yes
12	14 Feb 2017	2 Aug 2017	Not available	SD	No

*PD-L1 status was not evaluable due to insufficient tumor tissue. The category of PD-L1 high is defined as ≥25% tumor cells (TC) or ≥25% tumor-associated immune cells (IC); samples were categorized as PD-L1 low/negative if both TC and IC had <25% PD-L1 staining.

Abbreviations: CR, complete response; PD, progressive disease; PD-L1, programmed cell death ligand-1; PR, partial response; SD, stable disease.

Safety and Tolerability

- Treatment with tislelizumab was generally well tolerated in pretreated patients with UC
- Treatment-related AEs (TRAEs) occurred in 14 of the 16 patients with UC (**Table 3**)
 - Fatigue (n=1), hyperglycemia (n=1), and type 1 diabetes mellitus (n=1), were the only AEs considered related to treatment that were Grade ≥3 in severity
- One patient discontinued treatment due to an infusion-related reaction considered related to tislelizumab
- One adverse event of muscle weakness, which was associated with disease progression and occurred more than 1 month after last dose of study drug, had a fatal outcome; this event was considered not related to treatment

Table 3: Treatment-Related AEs Occurring in ≥2 Patients with UC

	UC Population (N=16)	
	All grades, n	Grade ≥3, n
Fatigue	5	1
Rash	3	0
Infusion-related reaction	2	0
Nausea	2	0
Pain in extremity	2	0
Proteinuria	2	0

Abbreviations: UC, urothelial carcinoma.

CONCLUSIONS

- Treatment with tislelizumab was generally well tolerated in pretreated patients with UC
- As of 28 August 2017, six (37.5%) patients remained on treatment; the median treatment duration was 4.3 months (range: 0.7–18.3 months)
- AEs reported in patients with UC were consistent with the overall safety profile observed in the study and were generally of low or moderate severity and manageable
- Of the 15 evaluable patients, tumor reduction meeting the definition of CR was observed in one patient, PRs were observed in four patients, and three patients achieved a confirmed best overall response of SD
 - Objective responses were observed at a higher rate in PD-L1$^+$ disease compared with PD-L1$^-$ disease
- The preliminary safety profile and antitumor activity from this ongoing study support continued development of tislelizumab in patients with UC
 - Tislelizumab is currently being investigated in China as monotherapy for patients with PD-L1$^+$ UC (CTR20170071)

REFERENCES

1. Topalian SL, et al. *N Engl J Med.* 2012; 366: 2443–2454.
2. Plimack ER, et al. *J Clin Oncol.* 2015; 33(suppl; abstr 4502).
3. Bellmunt J, et al. Abstract 470. Presented at 31st Society for Immunotherapy of Cancer Annual Meeting, November 9–13, 2016, National Harbor, MD, USA.
4. Balar A, et al. *Ann Oncol.* 2016; 27: A32 Abstract LB.
5. Sharma P, et al. *J Clin Oncol.* 2016; 34(suppl; abstr 4501).
6. Galsky MD, et al. *J Clin Oncol.* 2016; LB: A31.
7. Sharma P, et al. *Lancet Oncol.* 2016; 17: 1590–1598.
8. Ahmadzadeh M, et al. *Blood.* 2009;114: 1537–1544.
9. Dahan R, et al. *Cancer Cell.* 2015; 28: 285–295.
10. ClinicalTrials.gov. Study of the safety, pharmacokinetics and antitumor activities of BGB-A317 in subjects with advanced tumors. Available at: https://clinicaltrials.gov/ct2/show/NCT02407990 (last accessed December 2017).
11. Desai J, et al. *J Immunother Cancer.* 2016; 4(Suppl 1): P154.

ACKNOWLEDGMENTS

This study was supported by BeiGene, Ltd (Emeryville, CA, USA). Financial support for this presentation, including writing and editorial assistance by Aarati Rai, PhD MBA (SuccinctChoice Medical Communications, Chicago, IL, USA), was provided by BeiGene, Ltd.

Presented at the 2018 Genitourinary Cancers Symposium in San Francisco, CA, USA, February 8–10, 2018

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com.