ZANUBRUTINIB IN PATIENTS WITH RELAPSED/REFRACTORY MANTLE CELL LYMPHOMA: LONG-TERM EFFICACY AND SAFETY RESULTS FROM A PHASE 2 STUDY

Abstract EP789

Yuqin Song, MD, PhD[1]; Keshu Zhou, MD[2]; Dehui Zou, MD[3]; Jianfeng Zhou, PhD[4]; Jianda Hu, PhD[5]; Haiyan Yang, PhD[6]; Huilai Zhang, PhD[7]; Jie Ji, MD[8]; Wei Xu, MD, PhD[9]; Jie Jin, PhD[10]; Fangfang Lv, MD[11]; Ru Feng, MD[12]; Sujun Gao, PhD[13]; Haiyi Guo, MD[14]; Lei Zhou, MD, PhD[14]; Jane Huang, MD[14]; William Novotny, MD[14]; Rachel Wei, PhD[14]; and Jun Zhu, MD, PhD[1]

[1]Department of Lymphoma, Peking University Cancer Hospital & Institute (Beijing Cancer Hospital), Beijing, China; [2]Department of Hematology, Affiliated Cancer Hospital of Zhengzhou University, Henan Cancer Hospital, Zhengzhou, China; [3]State Key Laboratory of Experimental Hematology, Institute of Hematology & Blood Diseases Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Tianjin, China; [4]Department of Hematology, Tongji Hospital, Tongji Medical College, Wuhan, China; [5]Fujian Institute of Hematology, Fujian Provincial Key Laboratory on Hematology, Fujian Medical University Union Hospital, Fuzhou, China; [6]Department of Oncology, Zhejiang Cancer Hospital, Hangzhou, China; [7]Department of Lymphoma, Tianjin Medical University Cancer Institute and Hospital, National Clinical Research Center for Cancer, Key Laboratory of Cancer Prevention and Therapy, Tianjin's Clinical Research Center for Cancer, Tianjin, China; [8]Department of Hematology, West China Hospital of Sichuan University, Chengdu, China; [9]Department of Hematology, the First Affiliated Hospital of Nanjing Medical University, Jiangsu Province Hospital, Nanjing, China; [10]Department of Hematology, the First Affiliated Hospital, Zhejiang University College of Medicine, Hangzhou, China; [11]Department of Medical Oncology, Fudan University Shanghai Cancer Center, Shanghai, China; [12]Department of Hematology, Nanfang Hospital of Southern Medical University, Guangzhou, China; [13]Department of Hematology, Cancer Center, The First Hospital of Jilin University, Changchun, China; and [14]BeiGene (Beijing) Co., Ltd., Beijing, China, BeiGene (Shanghai) Co., Ltd., China, and BeiGene USA, Inc., San Mateo, CA, USA

INTRODUCTION

- BTK (Bruton tyrosine kinase) inhibitors are effective therapies for mantle cell lymphoma (MCL)[1]
- Zanubrutinib is a highly selective, potent and irreversible BTK inhibitor approved for treatment of adult patients with MCL who have received ≥ 1 prior therapy[2]
- Zanubrutinib was designed to maximize BTK occupancy and minimize off-target inhibition of TEC- and epidermal growth factor receptor-family kinases with potentially advantageous pharmacokinetic/pharmacodynamic properties[3,4]
- High activity of zanubrutinib monotherapy in patients with relapsed/refractory (R/R) MCL was demonstrated in a phase 2 study (BGB-3111-206, NCT03206970) after 18.4 months of follow-up[5]
- We present updated efficacy and safety results from the BGB-3111-206 study with ~3 years of follow-up

METHODS

- BGB-3111-206 was a multicenter, open-label phase 2 study of zanubrutinib monotherapy in patients with R/R MCL (**Figure 1**)
- Key inclusion criteria are 18-75 years of age; diagnosis of R/R MCL confirmed by central histological review; ≥1 to <5 prior therapies; ECOG performance status 0-2; measurable node disease by CT/MRI
- Long-term efficacy was assessed by investigator using PET-based imaging according to the Lugano Classification[6]

Figure 1. BGB-3111-206 Study Design

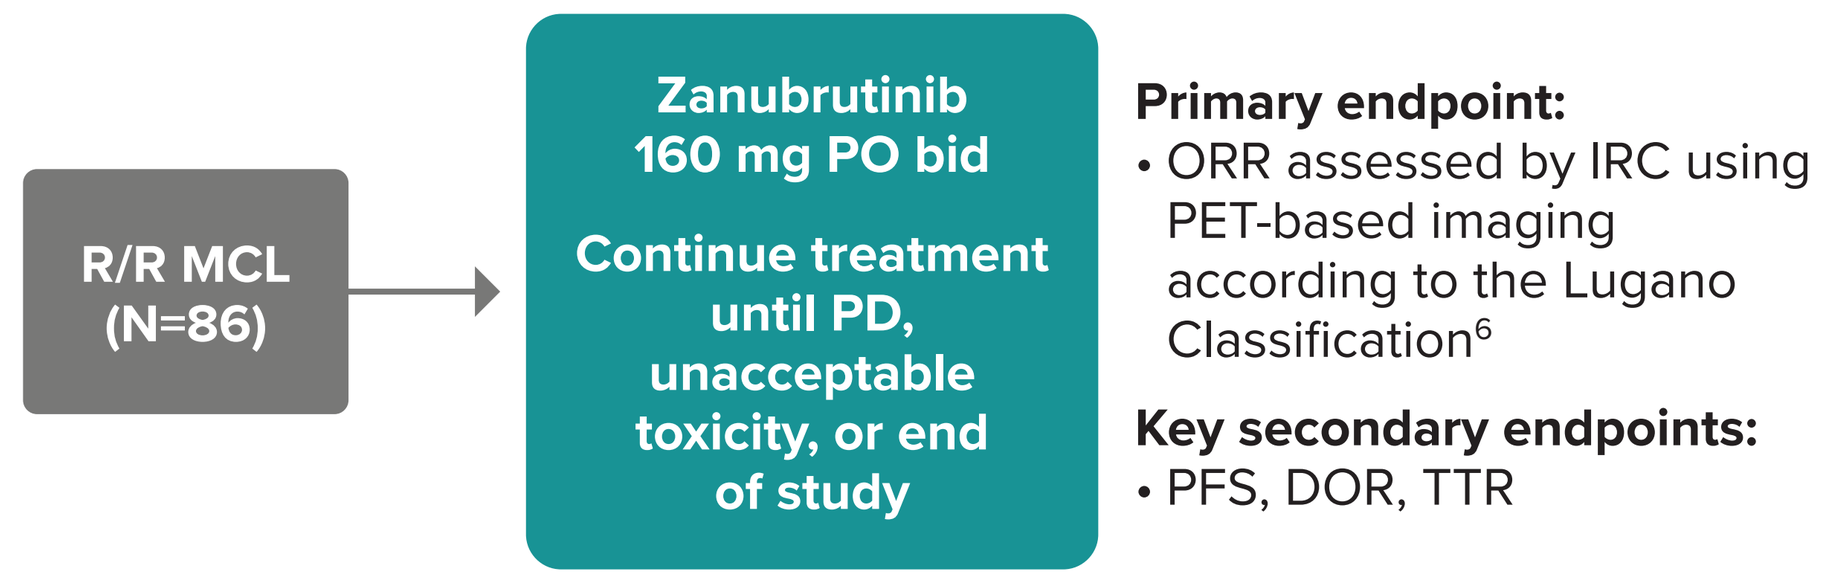
R/R MCL
(N=86)
Zanubrutinib
160 mg PO bid
Continue treatment until PD, unacceptable toxicity, or end of study
Primary endpoint:
• ORR assessed by IRC using PET-based imaging according to the Lugano Classification[6]
Key secondary endpoints:
• PFS, DOR, TTR

bid, twice daily; DOR, duration of response; IRC, independent review committee; MCL, mantle cell lymphoma; ORR, overall response rate; PD, progressive disease; PET, positron emission tomography; PFS, progression-free survival; PO, orally; R/R, relapsed/refractory; TTR, time to response.

RESULTS

Patients

- A total of 86 patients with R/R MCL were enrolled across 13 centers in China
- Demographics and baseline characteristics were published previously[5]
 - Median age was 60.5 years (range, 34-75); 83.7% of patients had intermediate-/high-risk disease per MCL International Prognostic Index Combined Biologic Index (MIPI-b)
 - Median prior lines of therapy were 2 (range, 1-4); 52.3% of patients had refractory disease
- As of the cutoff date at risk (8 September 2020), 39 patients (45.3%) remain on zanubrutinib (**Table 1**)

Table 1. Patient Disposition and Treatment Exposure

	All Patients (N=86)
Study follow-up, median (range), months	35.3 (0.3-41.6)
Duration of exposure, median (range), months	27.6 (0.2-41.6)
Relative dose intensity, median (range), %	99.87 (46.0-100.6)
Patients on treatment, n (%)	39 (45.3)
Reason for treatment discontinuation, n (%)	
Progressive disease	37 (43.0)
Adverse event	8 (9.3)
Investigator's discretion	1 (1.2)
Withdrawal of consent[a]	1 (1.2)

[a]Patient achieved a complete response and withdrew consent.

RESULTS *(CONTINUED)*

Efficacy

- The overall response rate was 83.7% (95% CI: 74.2-90.8%) with 77.9% complete response (CR)
 - Investigator-assessed best response rate did not change with longer follow-up (**Table 2**)
- Most patients achieved a response by the first efficacy assessment; median time to response was 2.73 months
- Median duration of response was not reached; the estimated 30-month event-free (progressive disease/death) rate was 57.3% (95% CI: 44.9-67.9%)

Table 2. Investigator-Assessed Best Response and Duration of Response

	N=86
ORR (CR + PR), % (95% CI)	83.7 (74.2-90.8)
Best response, n (%)	
CR	67 (77.9)
PR	5 (5.8)
SD	1 (1.2)
PD	8 (9.3)
Discontinued prior to first assessment	5 (5.8)
Median time to response, months (range)	2.73 (2.5-3.0)
Median time to CR, months (range)	2.79 (2.5-16.7)
Median DOR, months (95% CI)	NE (24.9-NE)
Event-at risk free rate at 30 months, % (95% CI)	57.3 (44.9-67.9)

CR, complete response; DOR, duration of response; NE, not estimable; ORR, overall response rate; PD, progressive disease; PR, partial response; SD, stable disease.

- Median progression-free survival (PFS) was 33.0 months (95% CI: 19.4-NE) and median overall survival (OS) was not reached (**Figure 2A and B**)
- Favorable survival was observed in patients who achieved CR (**Figure 2C and D**)
 - Median PFS was not reached (95% CI: 27.8-NE [not estimable]), 16.6 months (95% CI: 5.3-NE), and 2.6 months (95% CI: 0.8-2.9) for patients who achieved CR, PR (partial response), or nonresponders (stable disease/progressive disease), respectively

Figure 2. Overall PFS (A) and OS (B) and PFS (C) and OS (D) by Investigator-Assessed Best Response

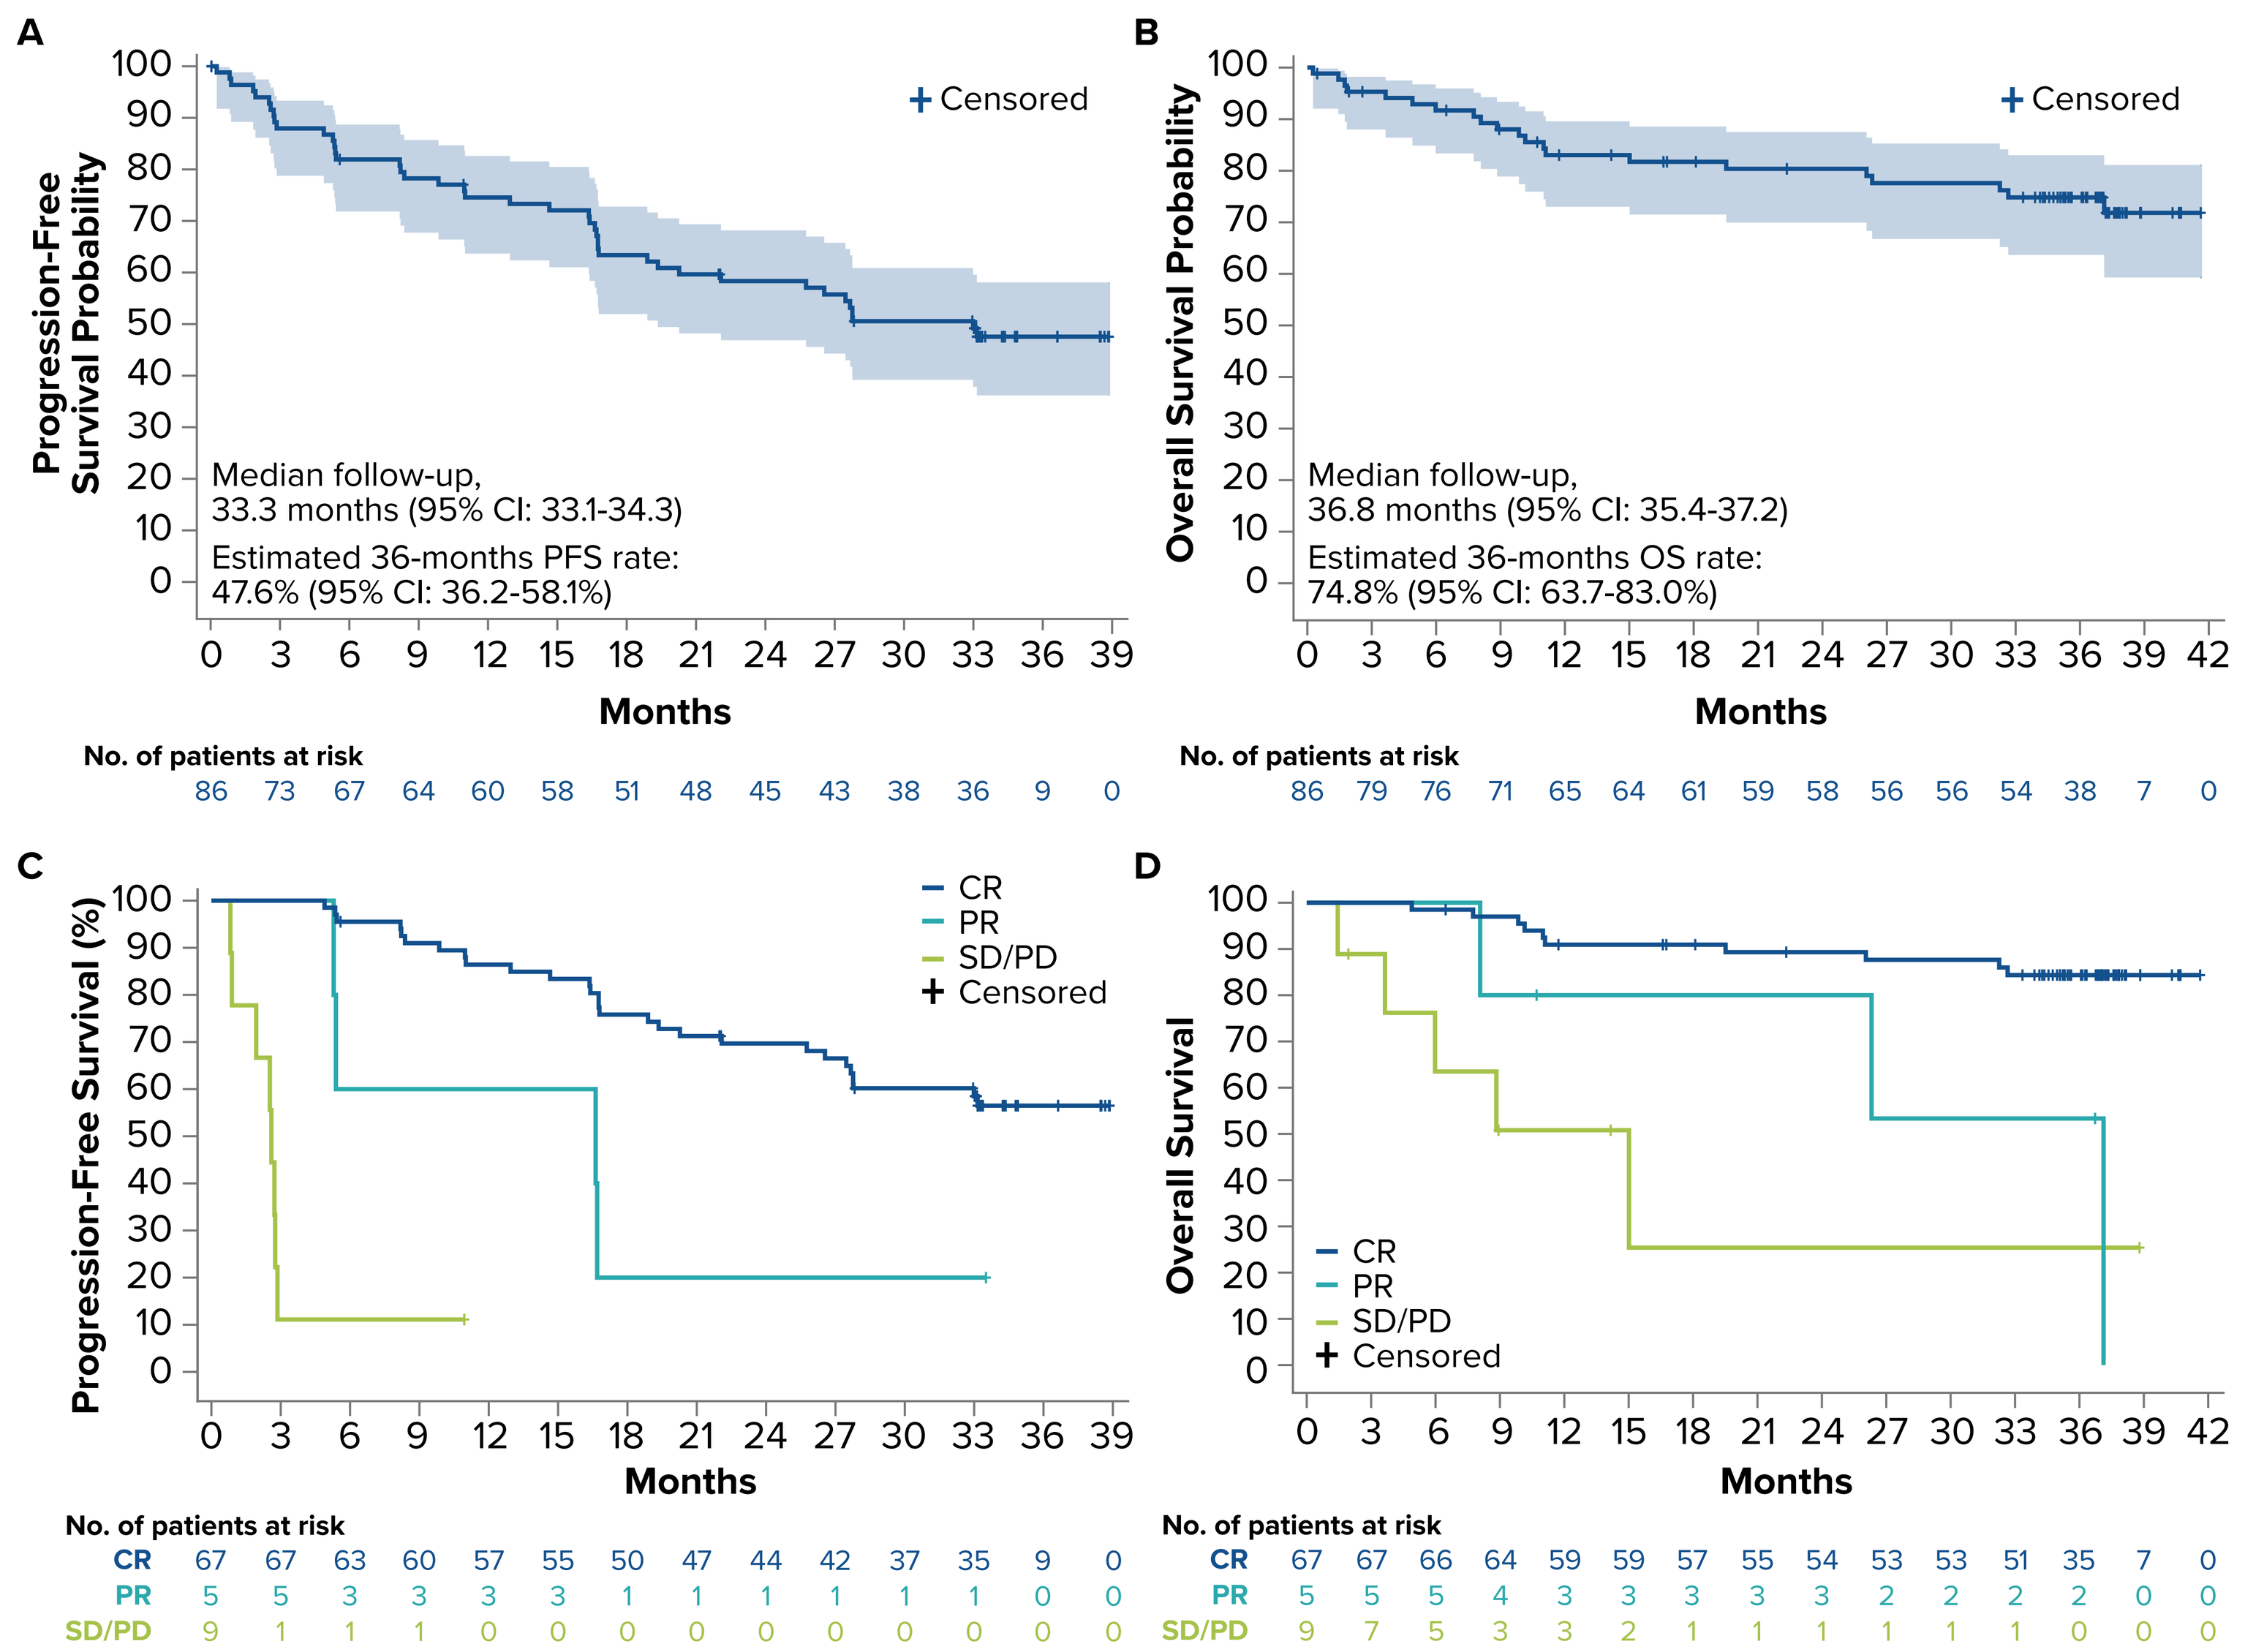
A
Progression-Free Survival Probability
+ Censored
Median follow-up,
33.3 months (95% CI: 33.1-34.3)
Estimated 36-months PFS rate:
47.6% (95% CI: 36.2-58.1%)
Months
No. of patients at risk
86 73 67 64 60 58 51 48 45 43 38 36 9 0
B
Overall Survival Probability
+ Censored
Median follow-up,
36.8 months (95% CI: 35.4-37.2)
Estimated 36-months OS rate:
74.8% (95% CI: 63.7-83.0%)
Months
No. of patients at risk
86 79 76 71 65 64 61 59 58 56 56 54 38 7 0
C
Progression-Free Survival (%)
CR
PR
SD/PD
+ Censored
Months
No. of patients at risk
CR 67 67 63 60 57 55 50 47 44 42 37 35 9 0
PR 5 5 3 3 3 3 1 1 1 1 1 1 0 0
SD/PD 9 1 1 1 0 0 0 0 0 0 0 0 0 0
D
Overall Survival
CR
PR
SD/PD
+ Censored
Months
No. of patients at risk
CR 67 67 66 64 59 59 57 55 54 53 53 51 35 7 0
PR 5 5 5 4 3 3 3 3 3 2 2 2 2 0 0
SD/PD 9 7 5 3 3 2 1 1 1 1 1 1 0 0 0

CR, complete response; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; SD, stable disease.

- Response was generally consistent across all subgroups analyzed (**Table 3**)
 - PFS was similar in patients with or without refractory disease and blastoid histology
 - Treatment benefit was observed in high-risk patients, though patients with low MIPI-b score, <3 prior lines of therapy or wild-type *TP53* had longer PFS

Table 3. Investigator-Assessed Subgroup Analysis of ORR and PFS

Subgroup	Patients, n	ORR % (95% CI)	PFS Median, (95% CI), months
Blastoid histology			
Yes	12	66.7 (34.9-90.1)	25.0 (2.5-NE)
No	68	86.8 (76.4-93.8)	27.8 (16.8-NE)
MIPI-b			
Low/intermediate	51	94.1 (83.8-98.8)	NE (27.8-NE)
High	33	69.7 (51.3-84.4)	9.1 (5.3-26.5)
Prior line of therapy			
< 3	57	89.5 (78.5-96.0)	NE (19.4-NE)
≥ 3	29	72.4 (52.8-87.3)	22.1 (5.4-33.1)
Refractory disease			
Yes	45	84.4 (70.5-93.5)	NE (16.8-NE)
No	41	82.9 (67.9-92.8)	27.7 (16.6-NE)
***TP53* mutation**			
Wild-type	39	89.7 (75.8-97.1)	NE (19.4-NE)
Mutated	15	80.0 (51.9-95.7)	14.7 (2.9-NE)

MIPI-b, Mantle Cell Lymphoma International Prognostic Index Combined Biologic Index; NE, not estimable; ORR, overall response rate; PFS, progression-free survival.

Safety

- The safety profile was largely unchanged with longer follow-up
 - No new treatment-emergent adverse events (TEAEs) led to death, treatment discontinuation, or dose reduction during the longer follow-up time (**Table 4**)
 - Most adverse events (AEs) occurred during the early stage of zanubrutinib treatment
 - Four new patients had grade ≥3 TEAEs of any infections, and no new patient had grade ≥3 TEAEs of hypertension or major hemorrhage during the longer follow-up period (**Figure 3**)
 - No cases of atrial fibrillation/flutter, grade ≥3 cardiac AEs, second primary malignancies, or tumor lysis syndrome were reported throughout this study

Table 4. Treatment-Emergent Adverse Events (TEAEs)

	All Patients (N=86)	
	18.4 months follow-up, n (%)	35.3 months follow-up, n (%)
Grade ≥3 TEAEs[a]	36 (41.9)	43 (50.0)
Serious TEAEs	21 (24.4)	25 (29.1)
TEAEs leading to study drug discontinuation	8 (9.3)	8 (9.3)
TEAEs leading to study drug interruption	13 (15.1)	16 (18.6)
TEAEs leading to study drug reduction	2 (2.3)	2 (2.3)
Death due to TEAE[b]	5 (5.8)[c]	5 (5.8)[c]

[a]Adverse event grades are evaluated based on National Cancer Institute Common Terminology Criteria for Adverse Events (NCI-CTCAE, version 4.03). [b]Death within 30 days of the last dose of zanubrutinib. [c]The 5 deaths due to TEAE included pneumonia, cerebral hemorrhage, traffic accident, and 2 deaths with unknown reason.

Figure 3. TEAEs of Special Interest (Pooled Terms)

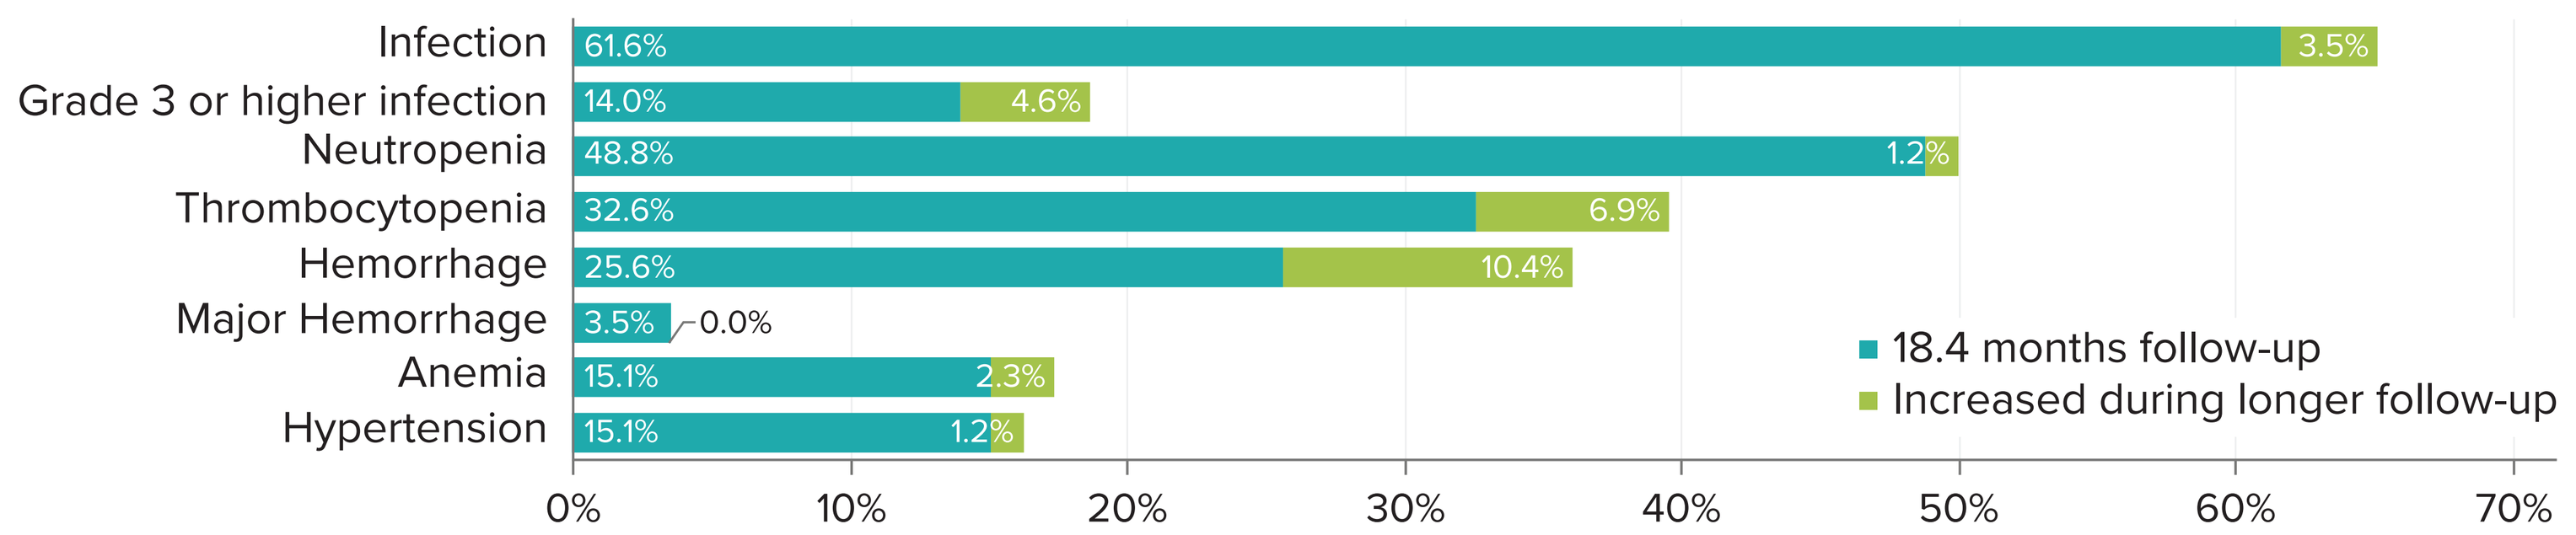
Infection 61.6% 3.5%
Grade 3 or higher infection 14.0% 4.6%
Neutropenia 48.8% 1.2%
Thrombocytopenia 32.6% 6.9%
Hemorrhage 25.6% 10.4%
Major Hemorrhage 3.5% 0.0%
Anemia 15.1% 2.3%
Hypertension 15.1% 1.2%
18.4 months follow-up
Increased during longer follow-up
0% 10% 20% 30% 40% 50% 60% 70%

AE, adverse event; TEAE, treatment-emergent AE.
Note: AE grades are evaluated based on National Cancer Institute Common Terminology Criteria for Adverse Events (NCI-CTCAE, version 4.03).

- With 35.3 months of follow-up, the most common (≥20%) TEAEs observed were decreased neutrophil count (46.5%), upper respiratory tract infection (38.4%), rash (36.0%), decreased white blood cell count (33.7%), and decreased platelet count (32.6%); most were grade 1/2 events (**Figure 4**)
- The prevalence of neutropenia with any grade or grade ≥3 decreased after the first year; no ≥ grade 3 neutropenia occurred after 18 months

Figure 4. TEAEs in ≥10% of Patients Regardless of Causality

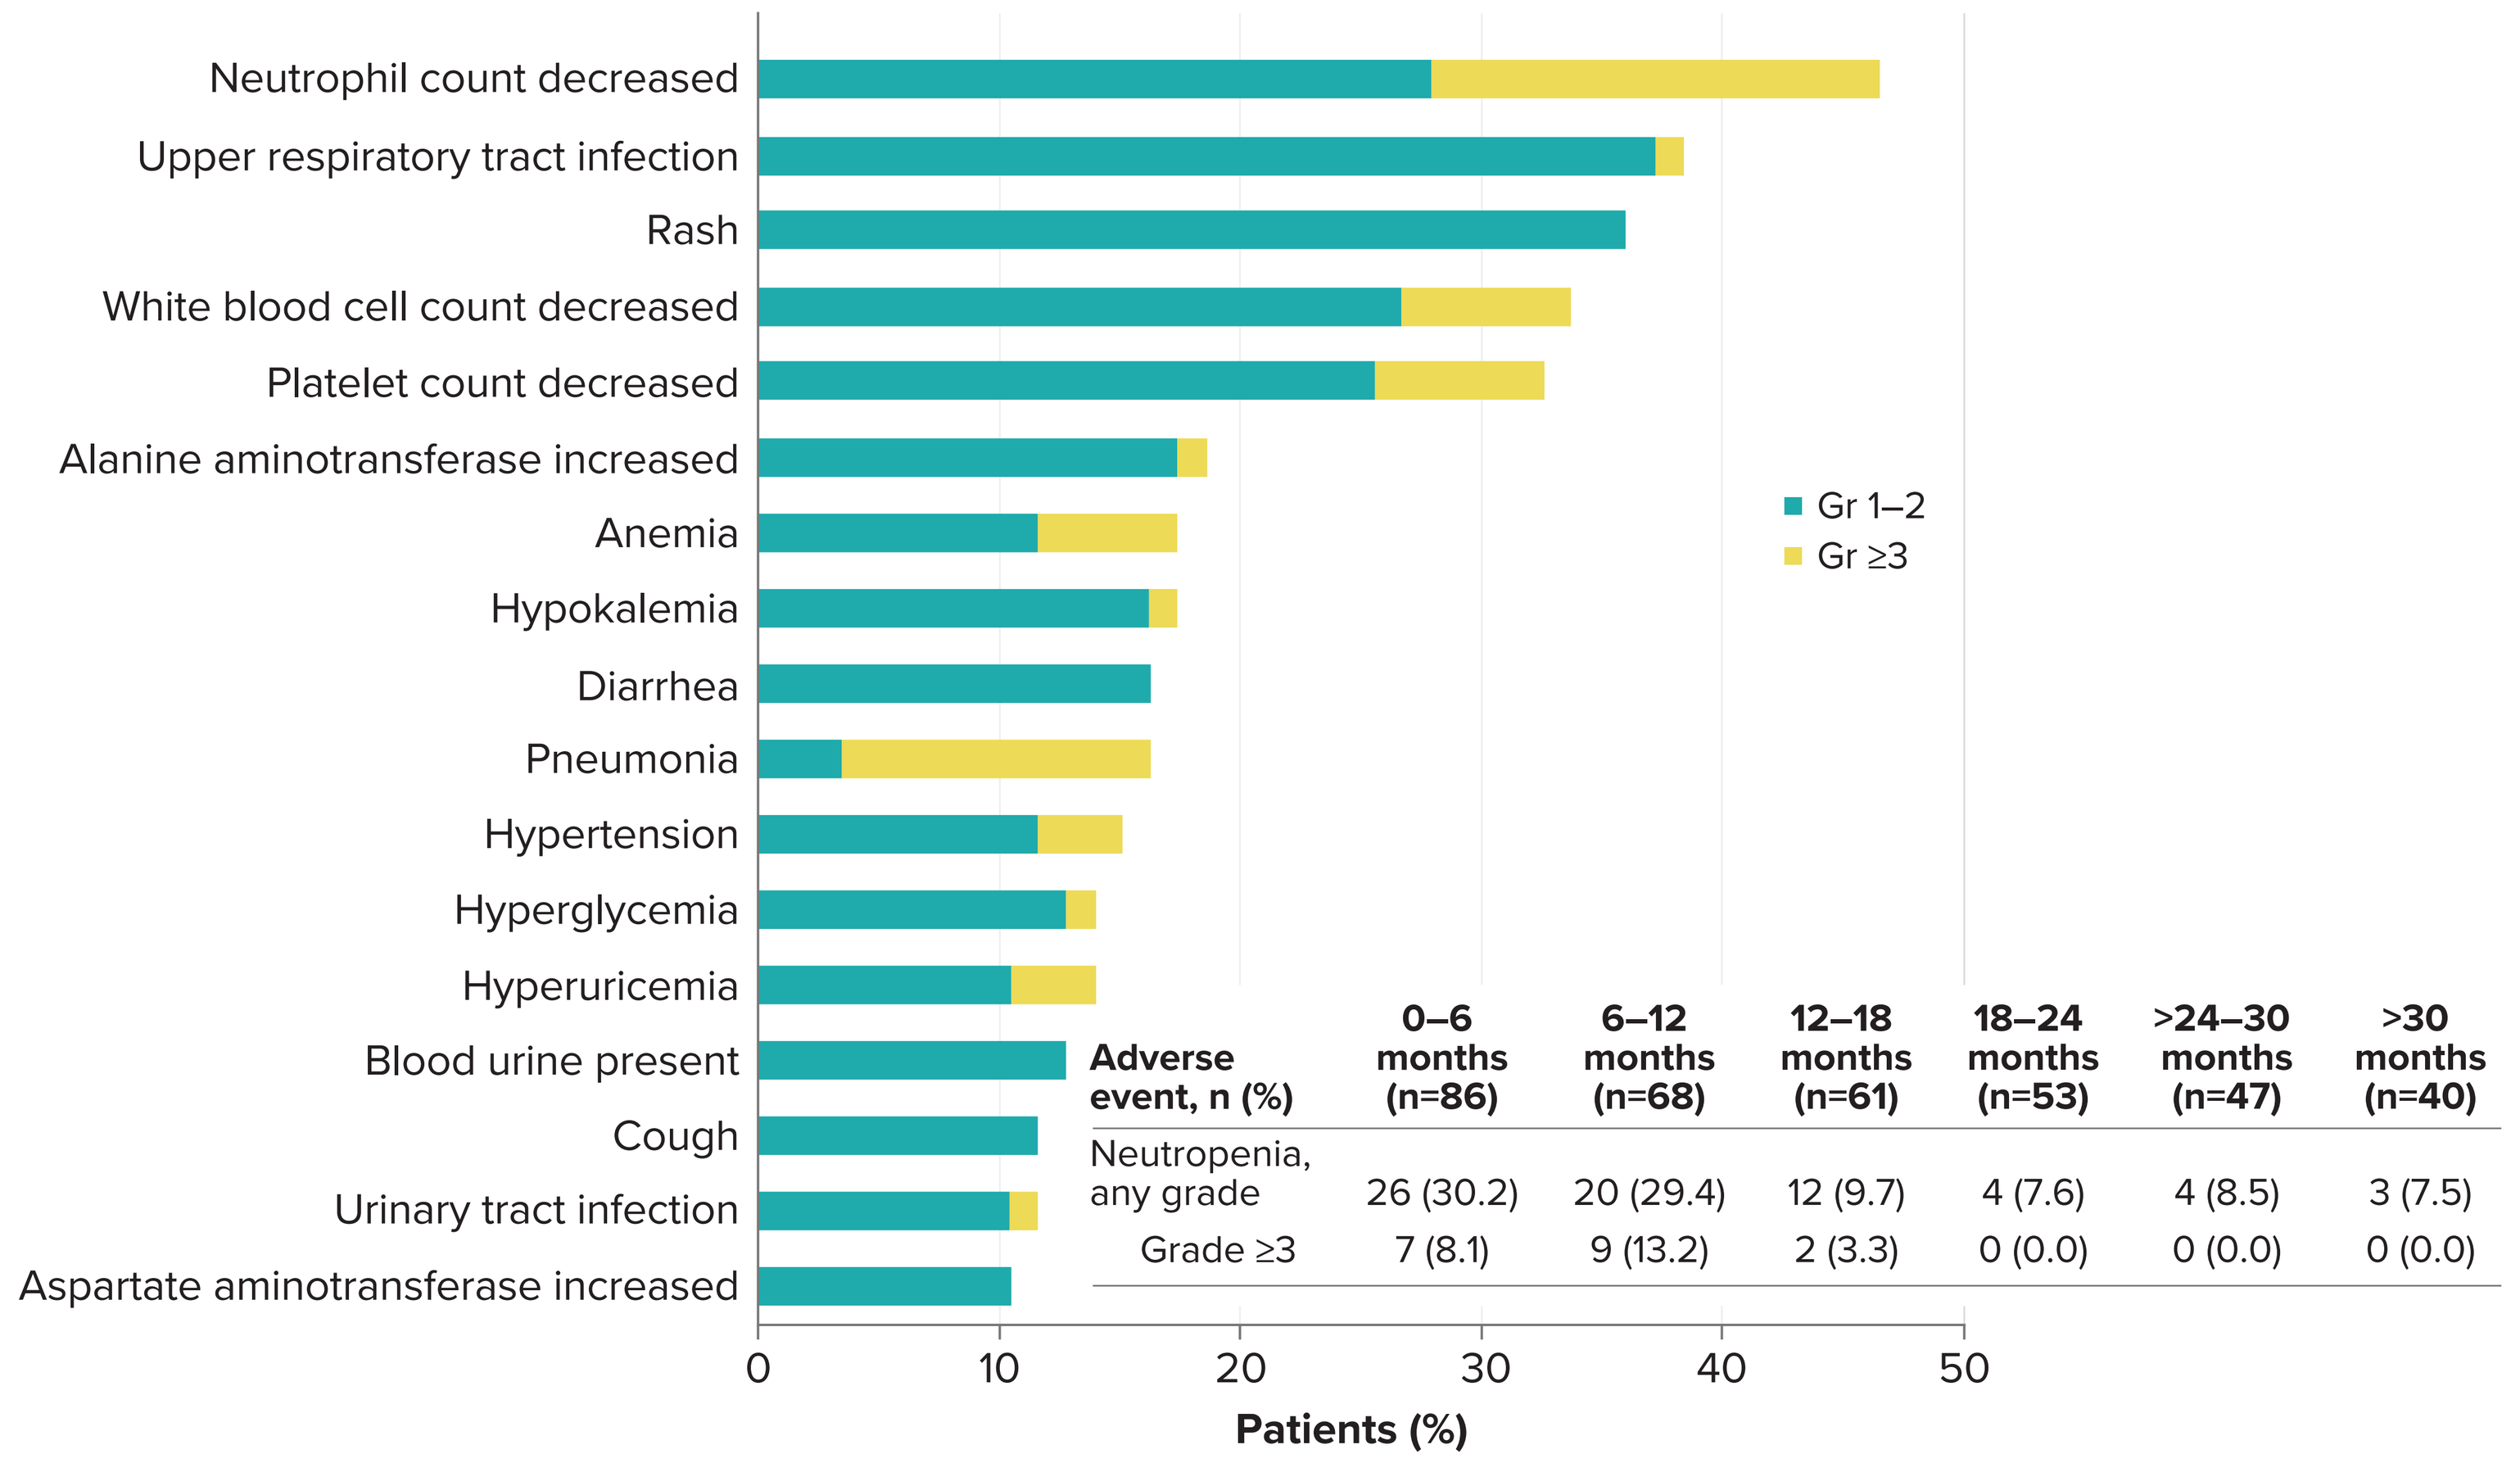
Neutrophil count decreased
Upper respiratory tract infection
Rash
White blood cell count decreased
Platelet count decreased
Alanine aminotransferase increased
Anemia
Hypokalemia
Diarrhea
Pneumonia
Hypertension
Hyperglycemia
Hyperuricemia
Blood urine present
Cough
Urinary tract infection
Aspartate aminotransferase increased
Gr 1–2
Gr ≥3
0 10 20 30 40 50
Patients (%)

Adverse event, n (%)	0–6 months (n=86)	6–12 months (n=68)	12–18 months (n=61)	18–24 months (n=53)	>24–30 months (n=47)	>30 months (n=40)
Neutropenia, any grade	26 (30.2)	20 (29.4)	12 (9.7)	4 (7.6)	4 (8.5)	3 (7.5)
Grade ≥3	7 (8.1)	9 (13.2)	2 (3.3)	0 (0.0)	0 (0.0)	0 (0.0)

Note: AE grades are evaluated based on NCI-CTCAE (version 4.03). AEs were coded using MedDRA Version 23.0. Preferred terms are listed.
AE, adverse event; TEAE, treatment-emergent AE.

CONCLUSIONS

- With 35.3 months follow-up, zanubrutinib continuously showed high, deep, and sustained efficacy in patients with R/R MCL
- The new TEAEs reported during the follow-up period were limited, and there were no new safety concerns
- These results further establish long-term benefit and tolerability for continuous zanubrutinib treatment in patients with R/R MCL

REFERENCES

1. Wang ML, et al. *N Engl J Med*. 2013;369:507-516.
2. BRUKINSA® (zanubrutinib) [United States prescribing information]. San Mateo, CA, USA: BeiGene USA, Inc.; 2019.
3. Guo Y, et al. *J Med Chem*. 2019;62:7923-7940.
4. Tam CS, et al. *Blood*. 2019;134:851-859.
5. Song YQ, et al. *Clin Cancer Res*. 2020;26:4216-4224.
6. Cheson BD, et al. *J Clin Oncol*. 2014;32:3059-3067.

DISCLOSURES

YS, KZ, DZ, JZ, JH, HY, HZ, JJ, WX, JJ, FL, RF, SG and **JZ** have nothing to disclose
HG is an employee of and has equity ownership in BeiGene
LZ is an employee of and has equity ownership in BeiGene
JH is an employee of, has a leadership role, equity ownership in, patents, and has received travel expenses from BeiGene
WN is an employee of and has equity ownership in BeiGene
RW is an employee of and has equity ownership in BeiGene

ACKNOWLEDGMENTS

We thank the investigators, site support staff, and especially the patients and their caregivers for participating in the BGB-3111-206 study
This study was sponsored by BeiGene. Editorial support was provided by Bio Connections, LLC and funded by BeiGene
Contact Yuqin Song, MD, PhD: songyuqin622@163.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.

Connecting Hematology For Clinical and Research Excellence
EHA2021 VIRTUAL JUNE 9 - 17, 2021