TISLELIZUMAB VERSUS PLACEBO IN COMBINATION WITH CONCURRENT CHEMORADIOTHERAPY IN PATIENTS WITH LOCALIZED ESOPHAGEAL SQUAMOUS CELL CARCINOMA: A PHASE 3 TRIAL IN PROGRESS

Weihu Wang[1], Jiancheng Li[2], Tao Li[3], Kuaile Zhao[4], Rong Yu[1], Wenqing Wang[5], Long Liang[3], Liyun Li[6], Wentao Yu[6], Yidi Wang[6], Wei Shen[6], Zhe Wu[6], Ginny Paton[7], Zefen Xiao[5]

[1]Peking University Cancer Hospital, Beijing, China; [2]Fujian Cancer Hospital, Fuzhou, China; [3]Sichuan Cancer Hospital, Chengdu, China; [4]Fudan University Shanghai Cancer Center, Shanghai, China; [5]Cancer Hospital and Institute, Chinese Academy of Medical Sciences, Beijing, China; [6]BeiGene (Beijing) Co., Ltd., Beijing, China; [7]BeiGene USA, Inc., San Mateo, CA, USA

Abstract: TPS475; Board: L19

American Society for Clinical Oncology: Gastrointestinal Cancers Symposium

January 23-25, 2020, San Francisco, CA

BACKGROUND

- Esophageal squamous cell carcinoma (ESCC) is a common cancer type in China that is associated with a poor prognosis[1]
 - At initial diagnosis, half of the patients present with locally advanced disease and many are unfit for surgery[2]
- An accepted alternative to surgery is concurrent chemoradiotherapy (cCRT)[3]; however, many patients experience local failure or distant metastasis after cCRT and innovative therapies are needed
- New therapeutic models for the treatment of gastrointestinal tumors have focused on targeting the immune system, including the programmed cell death protein-1/programmed death-ligand 1 (PD-1/PD-L1) axis[4]
- The PD-1/PD-L1 axis plays a central role in suppressing antitumor immunity; dysregulation of the axis can be used by cancer cells to evade the immune system[5,6]
 - Antibodies targeting PD-1 have demonstrated antitumor activity in patients with advanced esophageal cancers[7,8]
- PD-1 inhibition in combination with chemoradiotherapy has demonstrated synergistic antitumor activity in both preclinical models[9] and in clinical trials[10-12]
- Tislelizumab is a monoclonal antibody with high affinity and specificity for PD-1
- Tislelizumab was specifically engineered to minimize binding to FcγR on macrophages to abrogate antibody-dependent phagocytosis, a mechanism of T-cell clearance and potential resistance to anti-PD-1 therapy (Figure 1)[5,13]

Figure 1: Lack of FcγR Binding May Help Prevent Macrophage-Mediated T-Cell Clearance

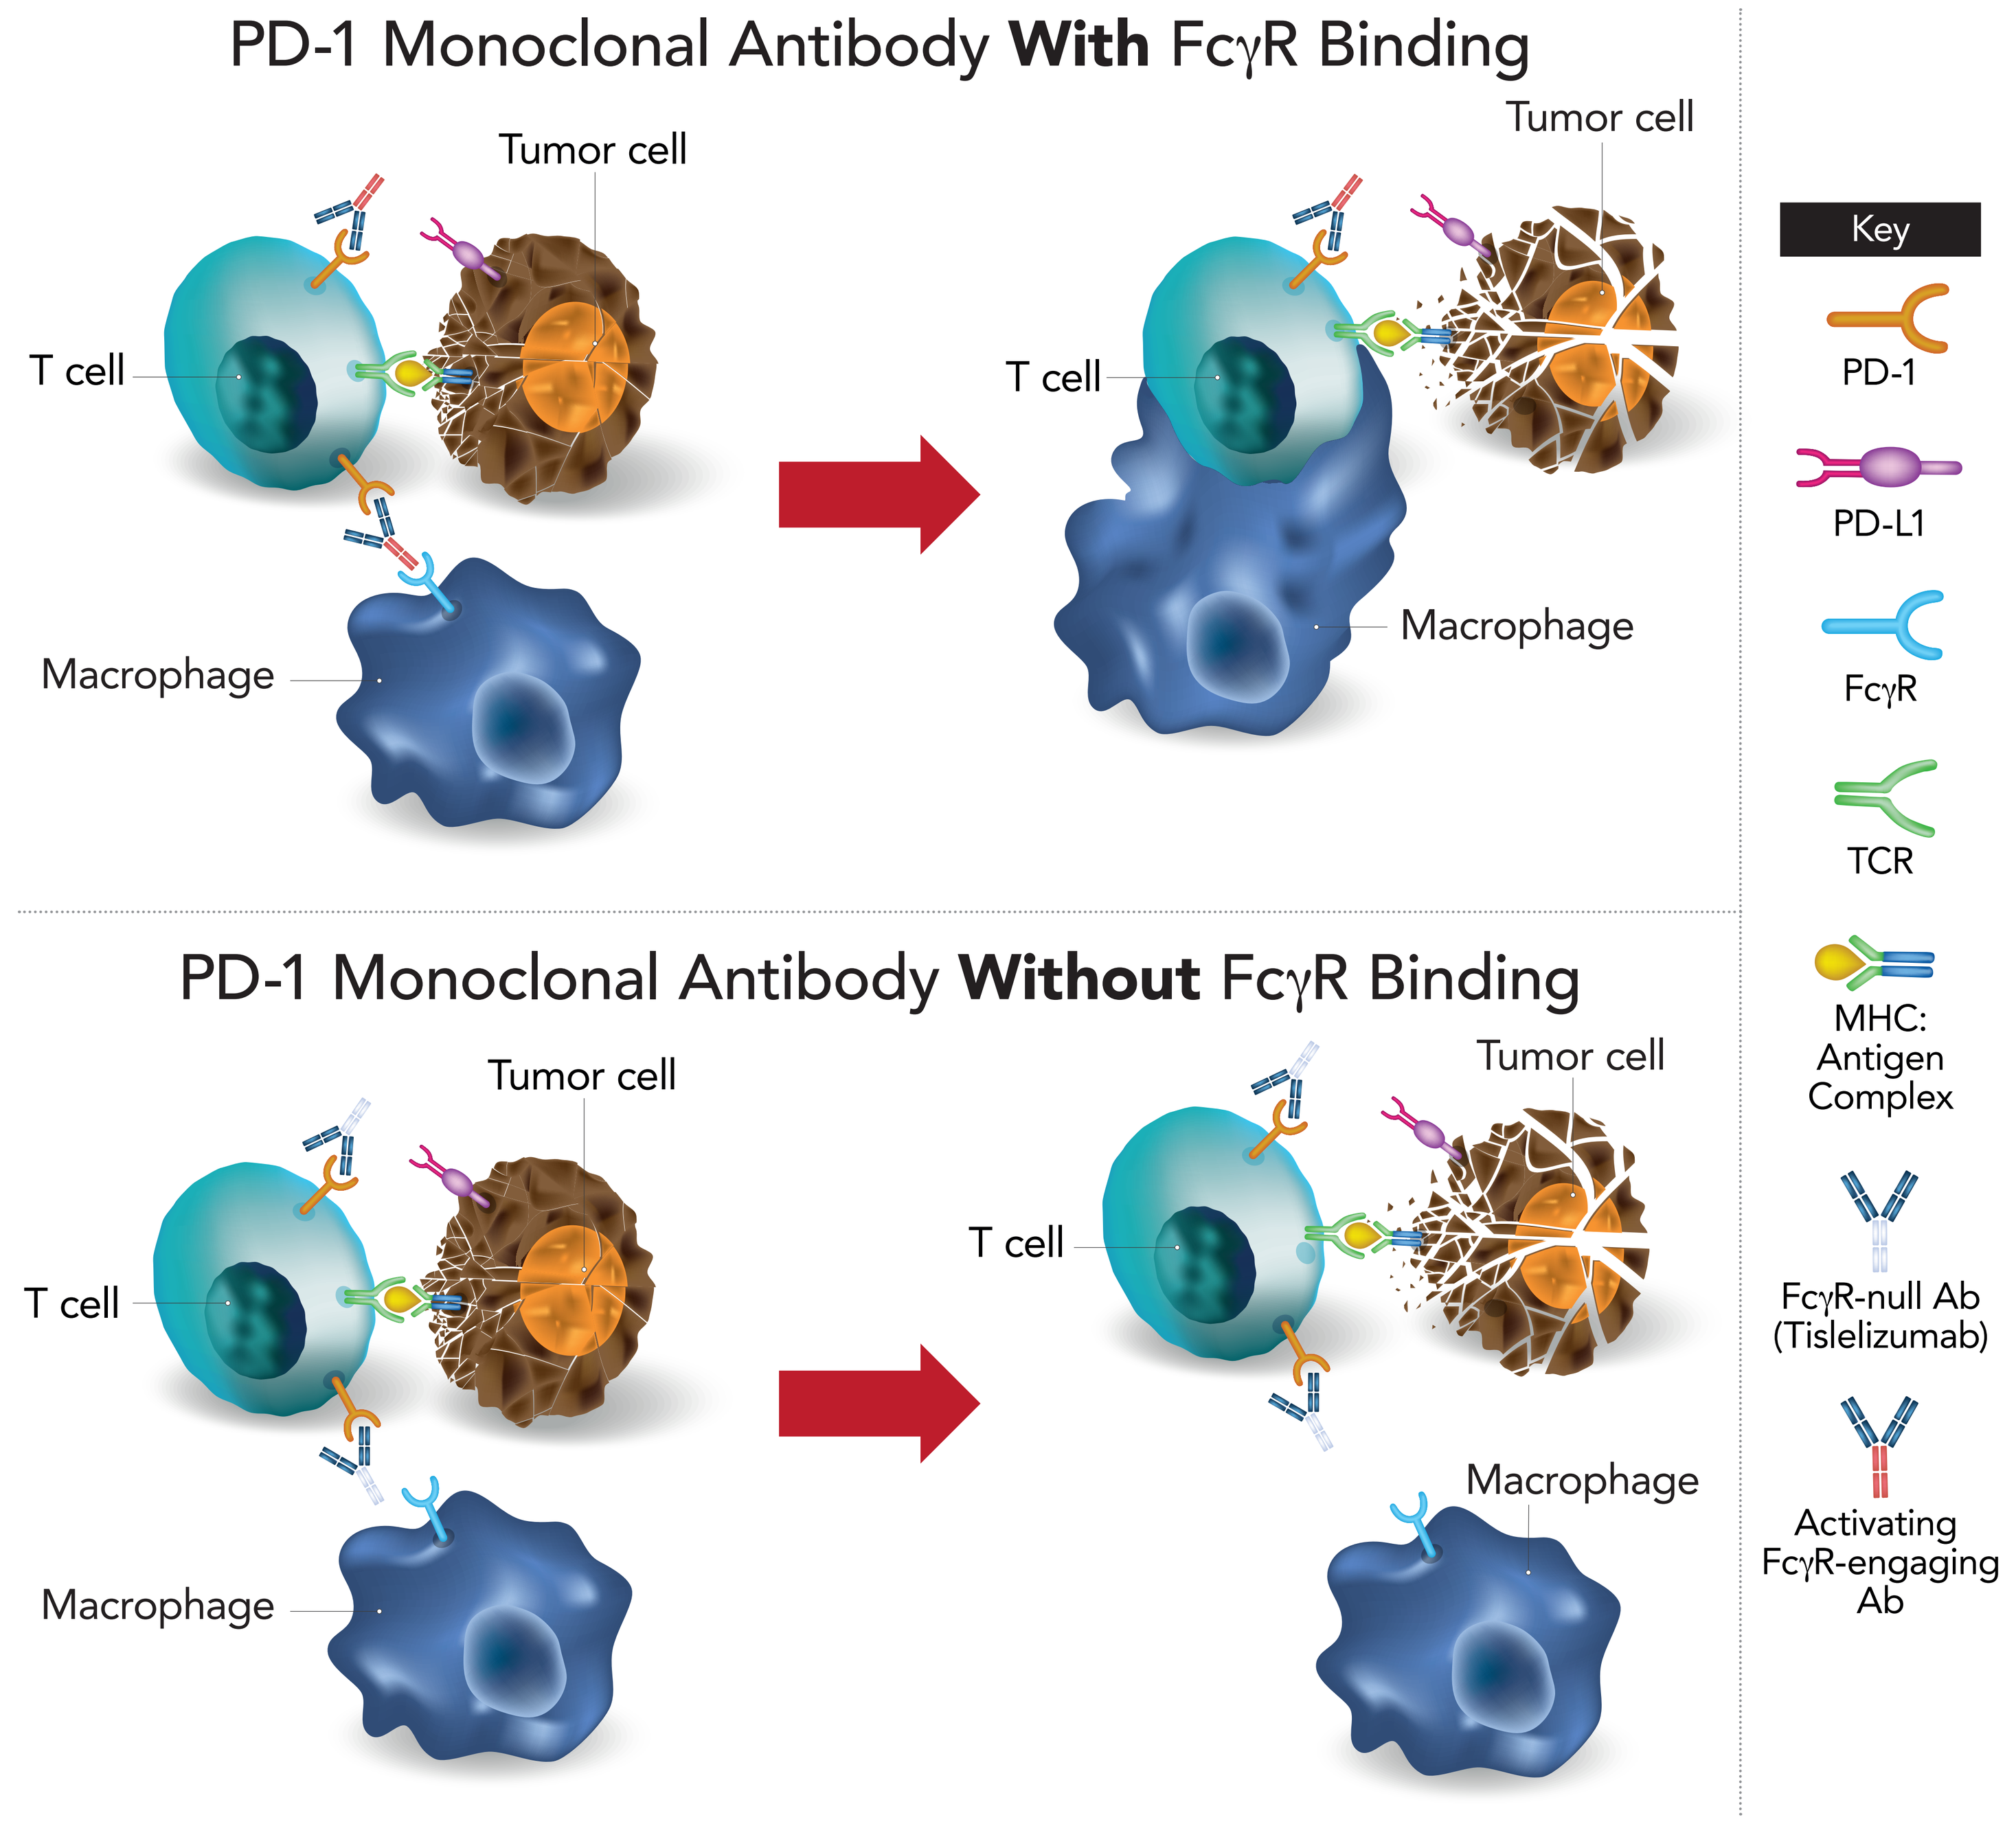
PD-1 Monoclonal Antibody **With** FcγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
PD-1 Monoclonal Antibody **Without** FcγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
Key
PD-1
PD-L1
FcγR
TCR
MHC: Antigen Complex
FcγR-null Ab (Tislelizumab)
Activating FcγR-engaging Ab

Abbreviations: Ab, antibody; MHC, major histocompatibility complex; PD-1, programmed death-1 receptor; PD-L1, programmed death-ligand 1; TCR, T-cell receptor.

- Tislelizumab has shown higher affinity to PD-1 compared with pembrolizumab and nivolumab with an ~100- and 50-fold slower off-rate, respectively[14]
 - Tislelizumab has a different binding orientation to PD-1 compared with pembrolizumab and nivolumab; the binding surface on PD-1 for tislelizumab partially overlaps with that for pembrolizumab, but differs significantly from that for nivolumab (Figure 2)

Figure 2: Tislelizumab Binds to PD-1 in an Orientation Different From Pembrolizumab (A) and Nivolumab (B)

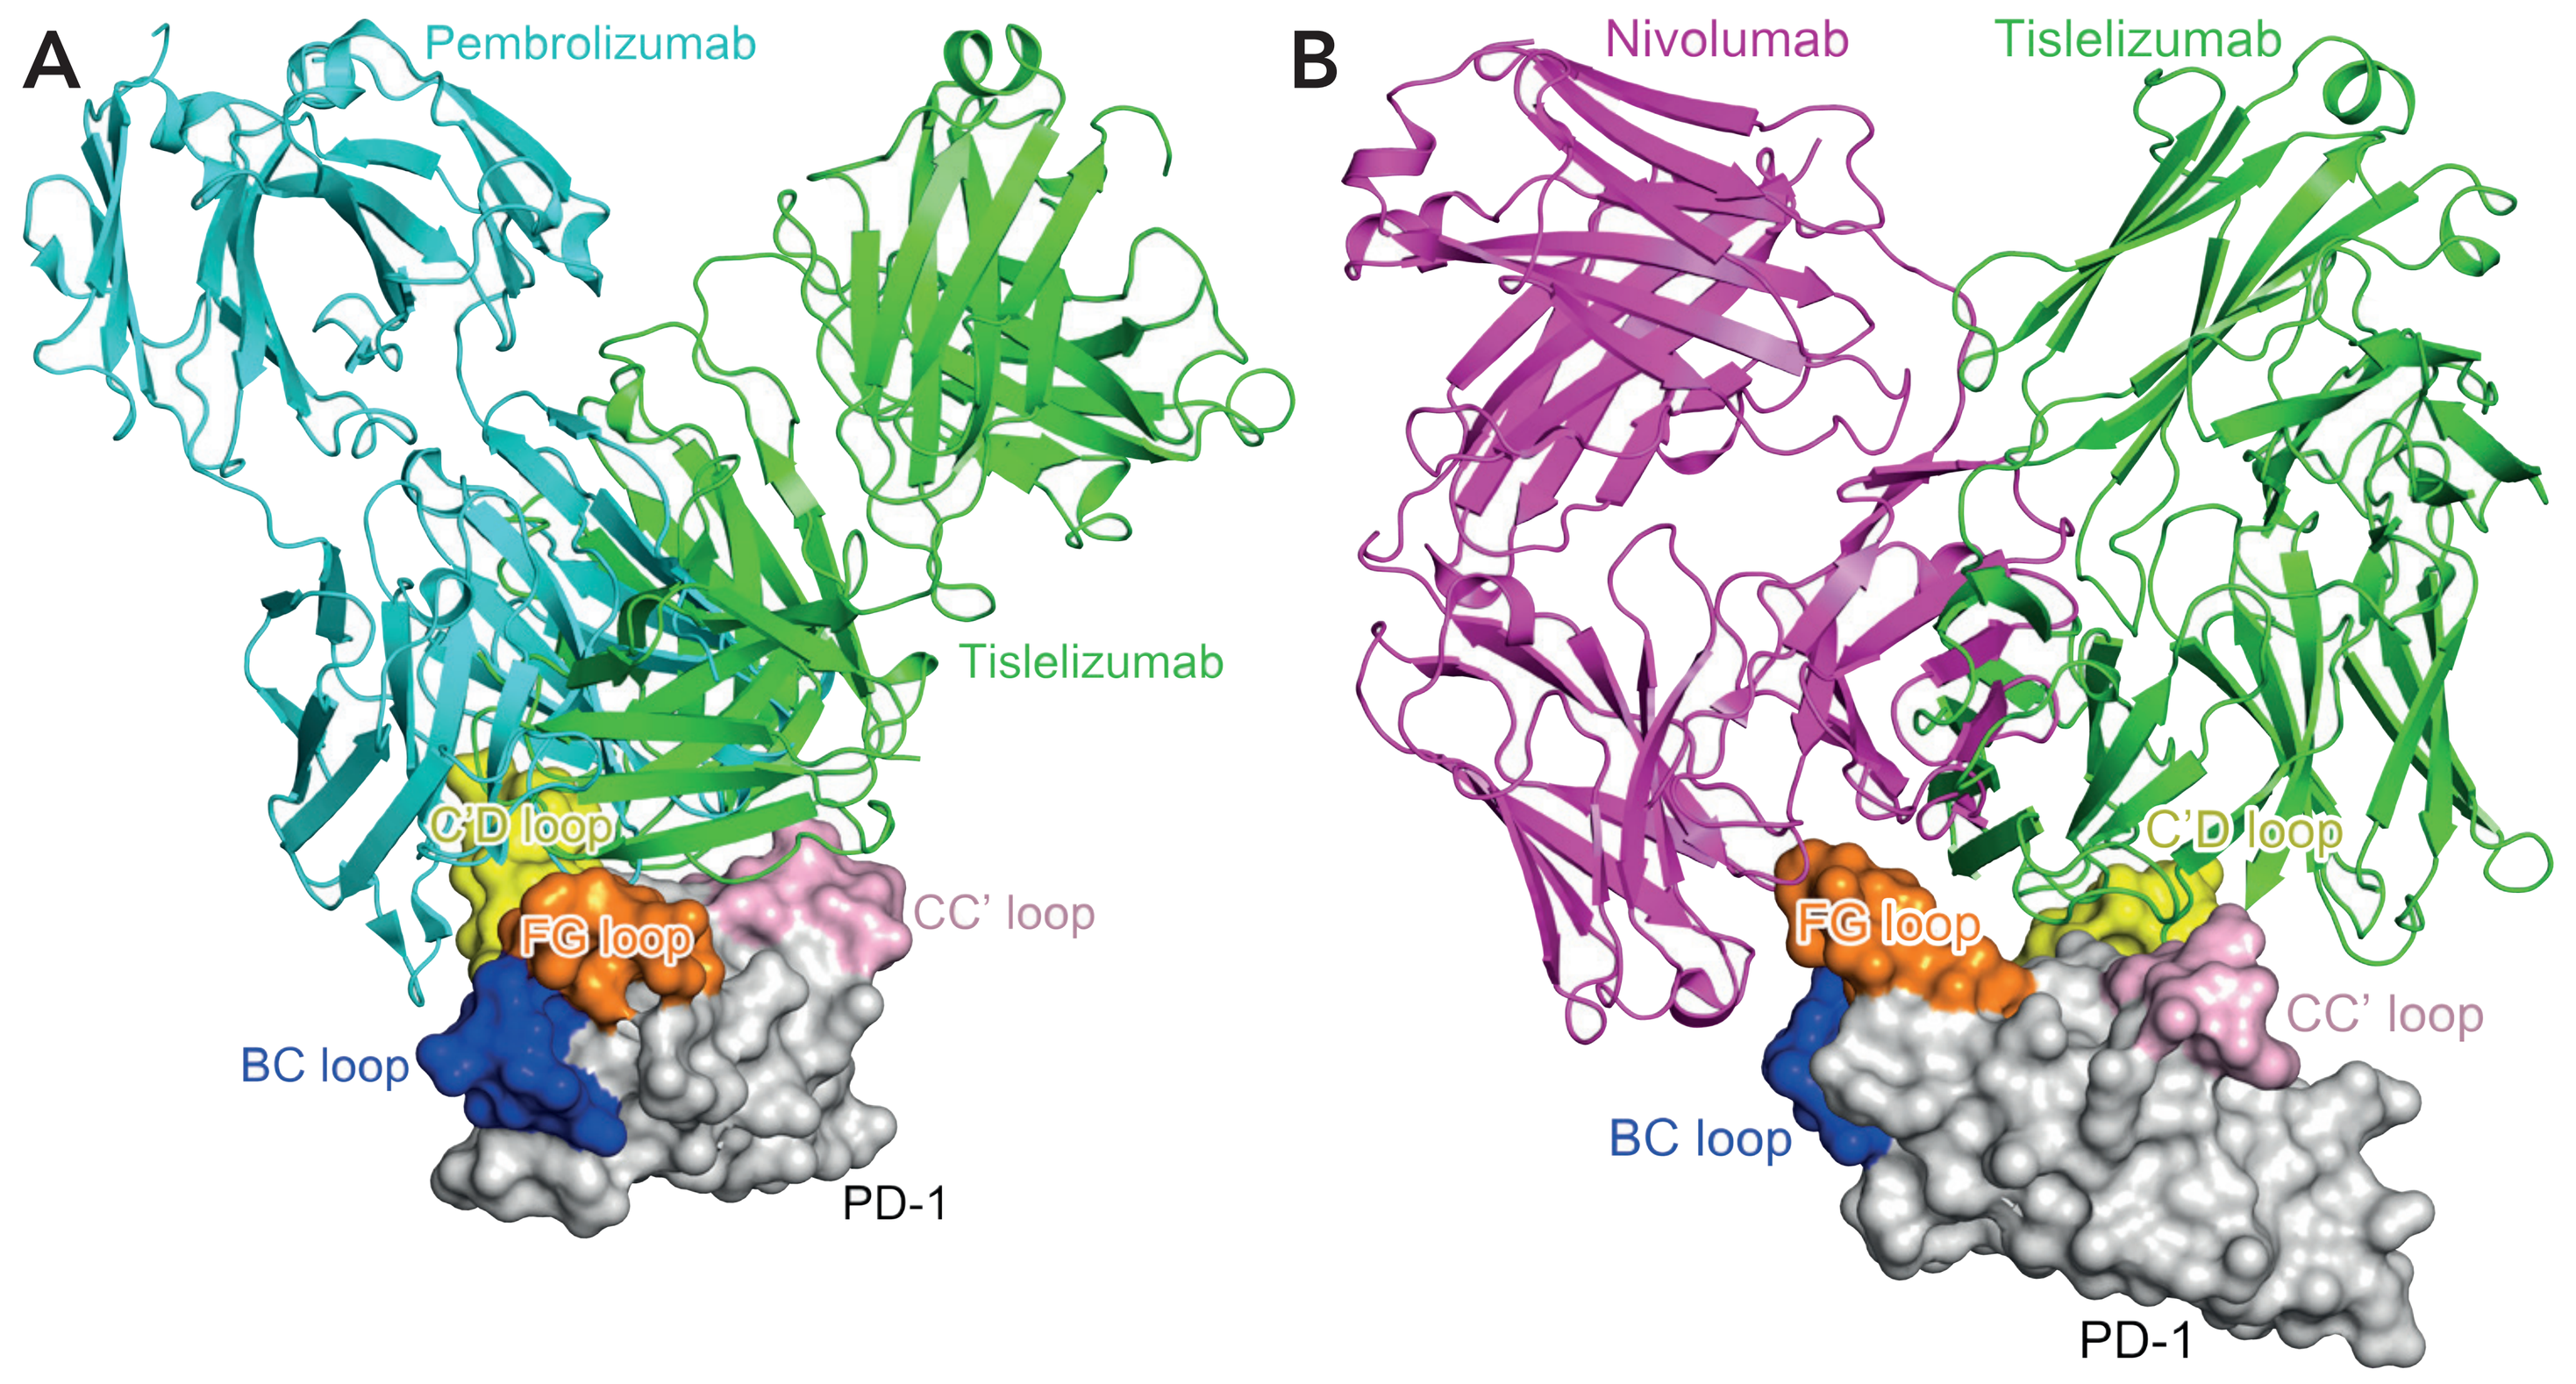
A
Pembrolizumab
Tislelizumab
C'D loop
FG loop
CC' loop
BC loop
PD-1
B
Nivolumab
Tislelizumab
C'D loop
FG loop
CC' loop
BC loop
PD-1

PD-1, tislelizumab, pembrolizumab, and nivolumab are colored in gray, green, cyan, and magenta, respectively. The BC, CC′, C′D, and FG loops of PD-1 are colored in blue, pink, yellow, and orange, respectively.
Abbreviation: PD-1, programmed death-1 receptor.

- Early phase studies (NCT02407990; NCT03469557) have reported that tislelizumab, as a single agent and in combination with platinum-containing chemotherapy, was generally well tolerated and demonstrated preliminary antitumor activity in patients with ESCC[15,16]

METHODS

Overall Design and Study Objectives

- This phase 3, randomized, double-blind, placebo-controlled study (NCT03957590) is being conducted in approximately 316 patients from 35 centers; the study is designed to compare the efficacy of tislelizumab versus placebo in combination with cCRT (Figure 3)
- The primary objective is to compare progression-free survival (PFS), assessed by a Blinded Independent Review Committee (BIRC) per the Response Evaluation Criteria in Solid Tumors (RECIST) v1.1, between tislelizumab and placebo in combination with cCRT in the intent-to-treat (ITT) population
 - An Independent Data Monitoring Committee (IDMC) will review data on a predefined schedule
- Secondary objectives include:
 - Evaluation of the overall response rate (ORR) and duration of response (DoR), assessed by the BIRC per RECIST v1.1, in patients treated with tislelizumab or placebo in combination with cCRT in the ITT population
 - Comparison of the overall survival (OS) between tislelizumab plus cCRT and placebo plus cCRT in the ITT population
 - Comparison of patient-reported outcomes of health-related quality of life (HRQoL) between tislelizumab and placebo in combination with cCRT
 - Evaluation of the safety/tolerability profile of tislelizumab in combination with cCRT

Figure 3: Study Design

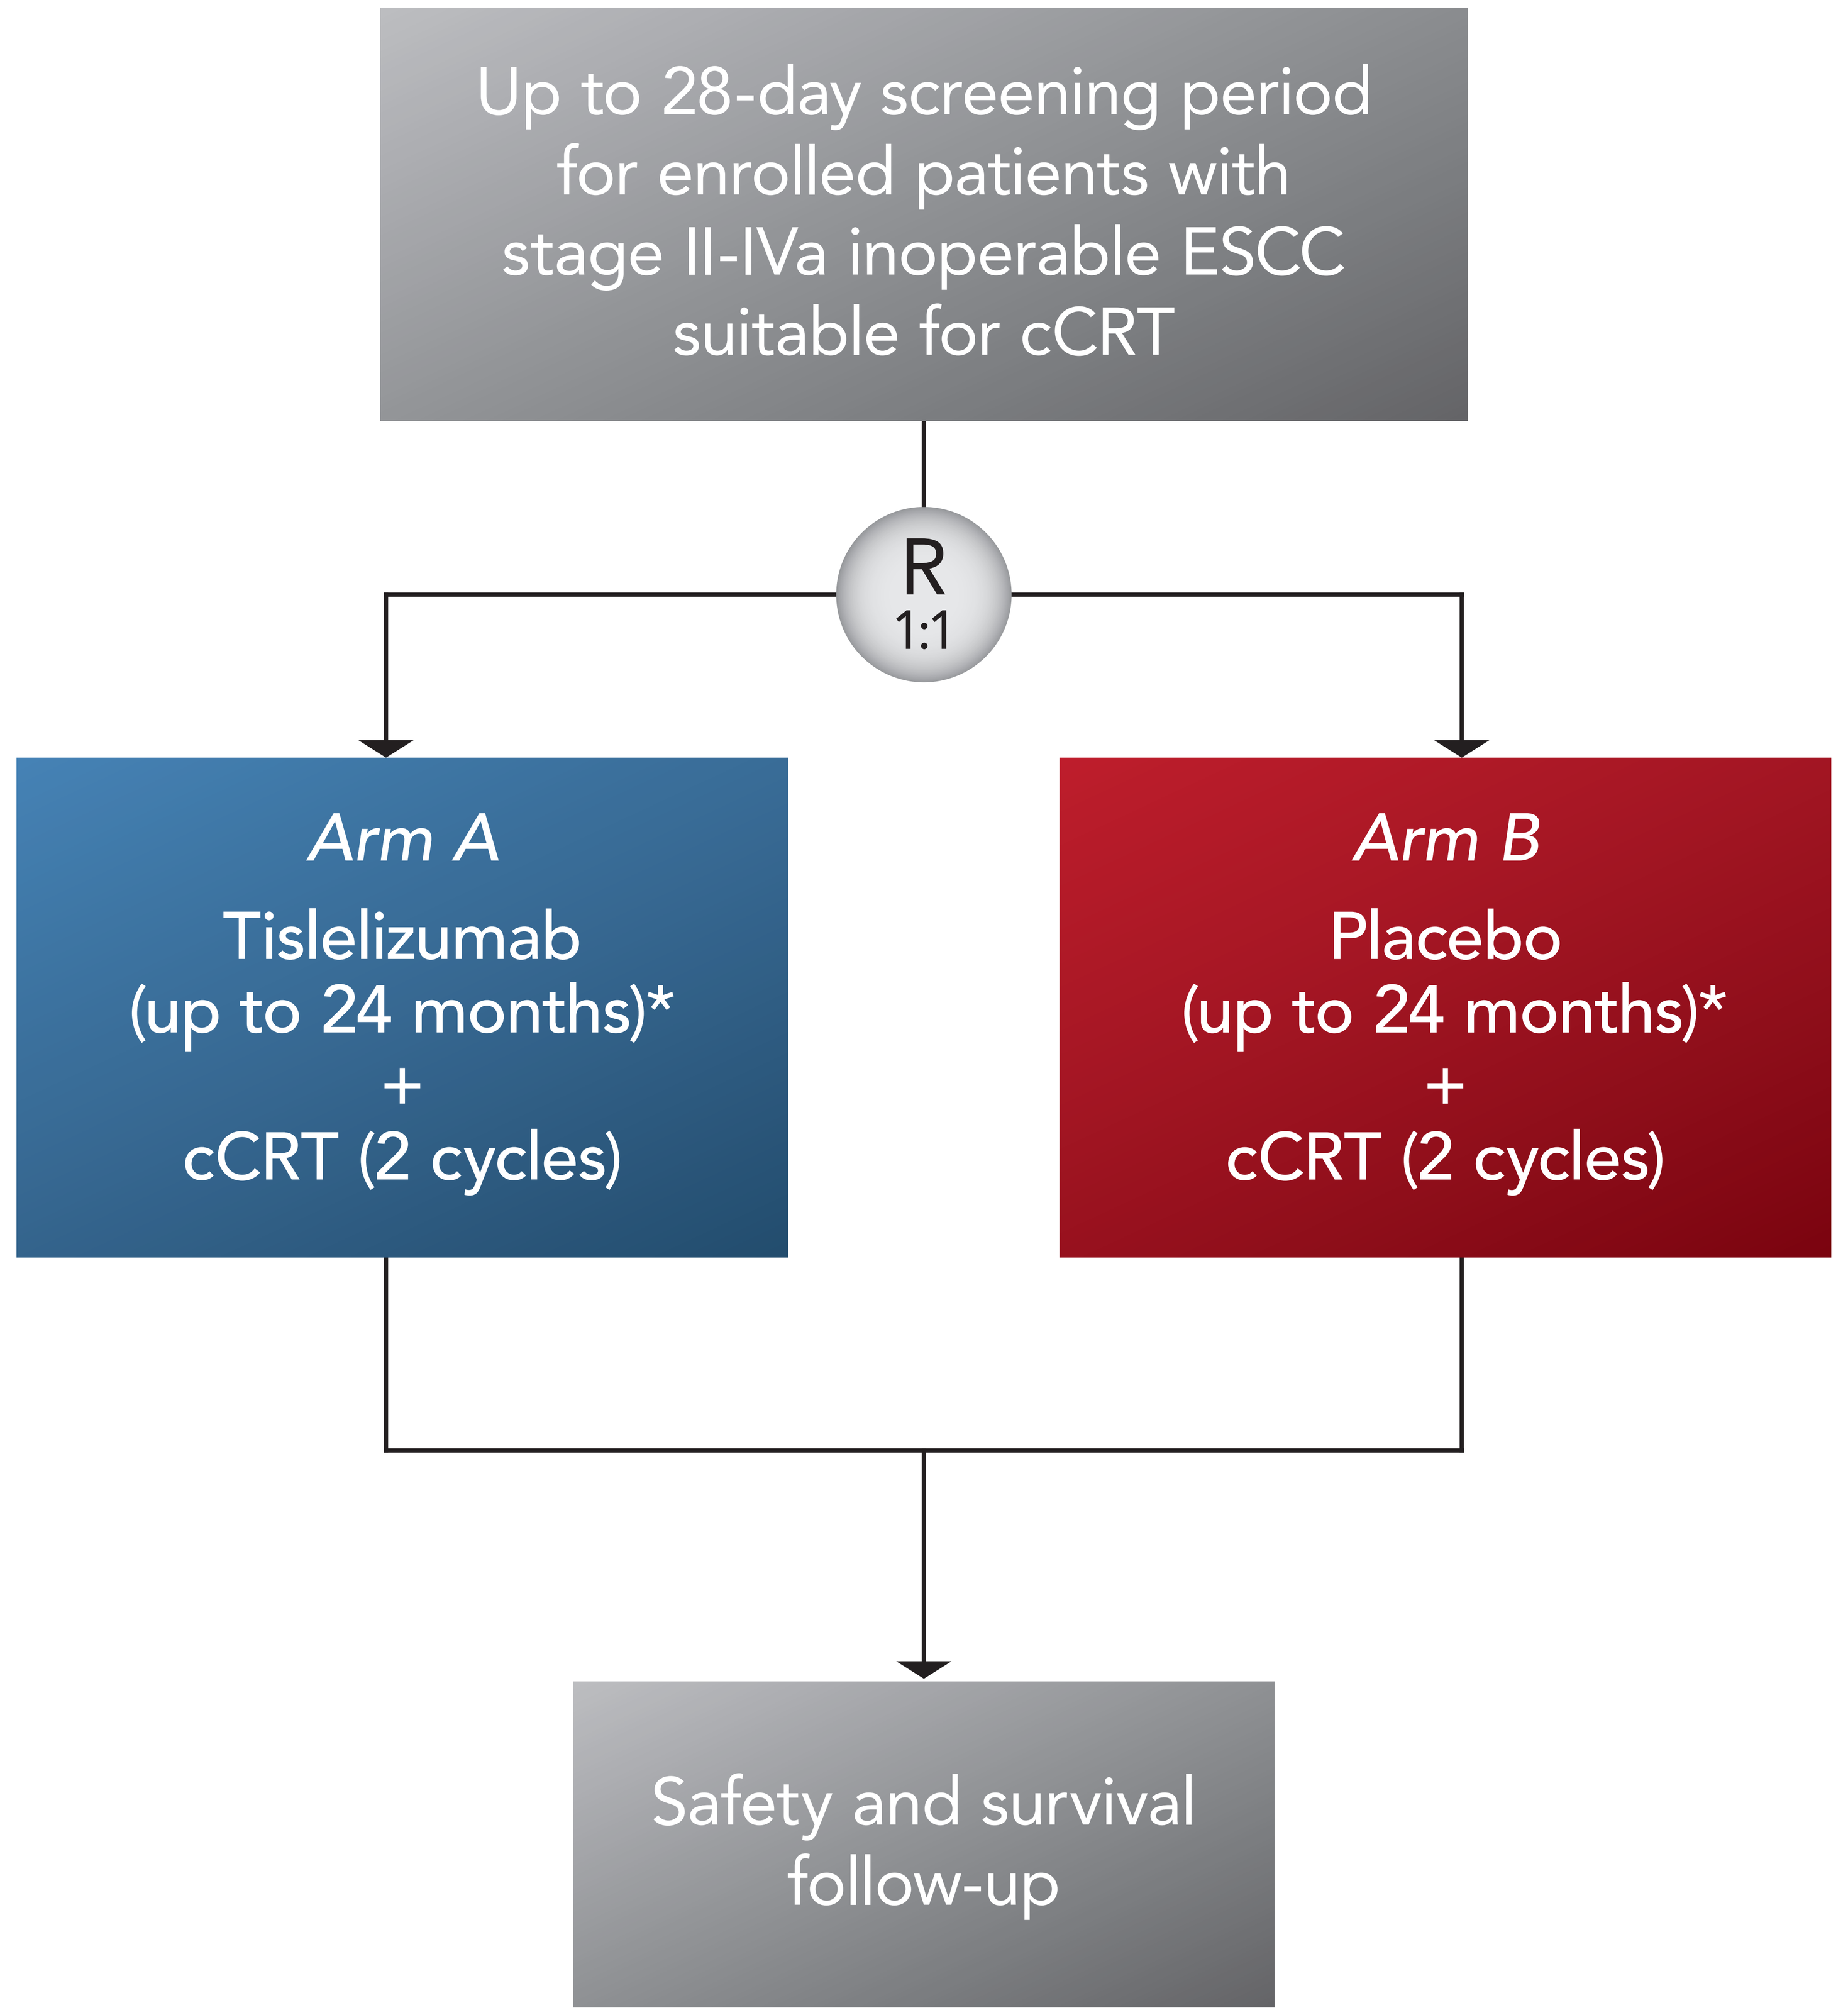
Up to 28-day screening period for enrolled patients with stage II-IVa inoperable ESCC suitable for cCRT
R
1:1
Arm A
Tislelizumab
(up to 24 months)*
+
cCRT (2 cycles)
Arm B
Placebo
(up to 24 months)*
+
cCRT (2 cycles)
Safety and survival follow-up

*Or until investigator-assessed disease progression, unacceptable toxicity, or withdrawal for other reasons.
Abbreviations: cCRT, concurrent chemoradiotherapy; ESCC, esophageal squamous cell carcinoma; R, randomized.

Study Population

- Adult patients (aged 18-75 years) with histologically confirmed localized ESCC with an Eastern Cooperative Oncology Group (ECOG) performance status ≤1 for whom cCRT is suitable and surgery is unsuitable/declined are being enrolled
 - Patients with stage II-IVa inoperable ESCC (medically unsuitable for surgery or refuses surgical intervention) are eligible
 - Patients who received prior chemotherapy (no more than three cycles) without radiotherapy can be enrolled
- Patients will be excluded if:
 - History of surgery for esophageal cancer
 - History of fistula due to primary tumor invasion, a high risk of fistula, or sign of perforation
 - Evidence of distant metastases
 - Intolerable or resistant to protocol-specified chemotherapy, or have received prior radiotherapy or therapies targeting PD-1, PD-L1, PD-L2, or other immune-oncology therapies

Treatment

- Patients will be randomized 1:1 to receive either tislelizumab 200 mg IV every 3 weeks (Q3W; Day 1 of each 21-day cycle) plus cCRT (*Arm A*) or placebo IV Q3W plus cCRT (*Arm B*)
- Chemotherapy, consisting of cisplatin 25 mg/m^2 IV on Days 1-3 of each 21-day cycle plus paclitaxel 135 mg/m^2 IV on Day 1 of every 21-day cycle, will be administered for two cycles; radiotherapy will be delivered with the total dose of 50.4 Gy in 28 fractions
- Duration of treatment will be up to 24 months (~35 cycles) for tislelizumab (*Arm A*) or placebo (*Arm B*), including about 6 weeks (two cycles) for concurrent administration of chemotherapy and radiotherapy
- Survival follow-up information will be collected every 3 months after the safety follow-up visit until death, unacceptable toxicity, or another treatment discontinuation criterion is met

Study Assessments and Statistical Analysis

- Tumor assessments will occur at baseline, every 9 weeks for the first 54 weeks, and every 12 weeks thereafter until radiographic disease progression or death
- The primary endpoint, PFS per BIRC assessment, will be compared between tislelizumab in combination with cCRT (*Arm A*) and placebo with cCRT (*Arm B*)
- Response endpoints (eg, ORR, DoR) will be assessed by the BIRC per RECIST v1.1, OS will be estimated using the Kaplan-Meier method
- Safety/tolerability of tislelizumab or placebo in combination with cCRT will be assessed by the incidence and severity of treatment-emergent adverse events according to National Cancer Institute Common Terminology Criteria for Adverse Events v5.0 criteria

REFERENCES

1. Wang AH, Liu Y, Wang B, et al. Epidemiological studies of esophageal cancer in the era of genome-wide association studies. *World J Gastrointest Pathophysiol.* 2014;5:335-343.
2. Jia R, Yin W, Li S, et al. Chemoradiation versus oesophagectomy for locally advanced oesophageal cancer in Chinese patients: study protocol for a randomised controlled trial. *Trials.* 2019; 2019;20:206.
3. Stahl M, Budach W. Definitive chemoradiotherapy. *J Thorac Dis.* 2017;9:S792-S798.
4. Stein A, Moehler M, Trojan J, et al. Immuno-oncology in GI tumours: Clinical evidence and emerging trials of PD-1/PD-L1 antagonists. *Crit Rev Oncol Hematol.* 2018;130:13-26.
5. Dahan R, Sega E, Engelhardt J, et al. FcγRs modulate the anti-tumor activity of antibodies targeting the PD-1/PD-L1 axis. *Cancer Cell.* 2015;28:285-295.
6. Mahoney KM, Freeman GJ, McDermott DF. The next immune-checkpoint inhibitors: PD-1/PD-L1 blockade in melanoma. *Clin Ther.* 2015;37:764-782.
7. Kudo T, Hamamoto Y, Kato K, et al. Nivolumab treatment for oesophageal squamous-cell carcinoma: an open-label, multicentre, phase 2 trial. *Lancet Oncol.* 2017;18:631-639.
8. Doi T, Piha-Paul SA, Jalal SI, et al. Safety and antitumor activity of the anti-programmed death-1 antibody pembrolizumab in patients with advanced esophageal carcinoma. *J Clin Oncol.* 2018;36:61-67.
9. Oh P, Du KL, Leichman L, et al. PD-1 blockade enhances the efficacy of chemoradiation in a mouse model of esophageal cancer. *Int J Radiat Oncol Biol Phys.* 2016;96:S127-S128.
10. Shaverdian N, Lisberg AE, Bornazyan K, et al. Previous radiotherapy and the clinical activity and toxicity of pembrolizumab in the treatment of non-small-cell lung cancer: a secondary analysis of the KEYNOTE-001 phase 1 trial. *Lancet Oncol.* 2017;18:895-903.
11. Powell SF, Gitau MM, Sumey CJ, et al. Safety of pembrolizumab with chemoradiation (CRT) in locally advanced squamous cell carcinoma of the head and neck (LA-SCCHN). *J Clin Oncol.* 2017;35(suppl 15):6011.
12. Antonia SJ, Villegas A, Daniel D, et al. Durvalumab after chemotherapy in stage III non-small-cell lung cancer. *New Engl J Med.* 2017;377:1919-1929.
13. Zhang T, Song X, Xu L, et al. Durvalumab after chemoradiotherapy in stage III non-small-cell lung cancer. *Cancer Immunol Immunother.* 2018;67:1079-1090.
14. Feng Y HY, Sun H, Zhang B, et al. The molecular binding mechanism of tislelizumab, an investigational anti-PD-1 antibody, is differentiated from pembrolizumab and nivolumab. In: *Proceedings of the 110th Annual Meeting of the American Association for Cancer Research.* March 29-April 3, 2019; Atlanta, Georgia.
15. Deva S, Lee J-S, Lin C-C, et al. A phase 1A/1B trial of tislelizumab, an anti-PD-1 antibody, in patients with advanced solid tumors. Presented at: ESMO Immuno Oncology Congress. December 13-16, 2018; Geneva, Switzerland.
16. Xu N, Yuan X, Wang B, et al. Tislelizumab in combination with chemotherapy for the treatment of Chinese patients (pts) with esophageal squamous cell carcinoma (ESCC): Results from one cohort of an ongoing phase 2 study. *J Clin Oncol.* 2019;37:14-14.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative centers' study staff and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. This study was sponsored by BeiGene, Ltd. Writing and editorial assistance was provided by Regina Switzer, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL), and funded by the study sponsor.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com