PHASE 2 STUDY OF PAMIPARIB IN CHINESE PATIENTS WITH ADVANCED OVARIAN CANCER

Xiaohua Wu[1], Jianqing Zhu[2], Jing Wang[3], Zhongqiu Lin[4], Beihua Kong[5], Rutie Yin[6], Wei Sun[7], Qi Zhou[8], Songling Zhang[9], Danbo Wang[10], Hong Shi[11], Yunong Gao[12], Yi Huang[13], Guiling Li[14], Xiuli Wang[15], Ying Cheng[16], Ge Lou[17], Li Li[18], Xiyan Mu[19], Miao Li[18]

[1]Department of Gynecologic Oncology, Fudan University Shanghai Cancer Center, Shanghai, China; [2]Department of Gynecologic Oncology, Zhejiang Cancer Hospital, Hangzhou, China; [3]Hunan Cancer Hospital/The Affiliated Cancer Hospital of Xiangya School of Medicine, Central South University, Changsha, China; [4]Department of Gynecological Oncology, Sun Yat-sen Memorial Hospital, Sun Yat-sen University, Guangzhou, China; [5]Department of Obstetrics and Gynecology, Qilu Hospital of Shandong University, Ji'nan, China; [6]Department of Obstetrics and Gynecology, West China Second University Hospital, Sichuan University, Chengdu, China; [7]Department of Gynecologic Oncology, Anhui Provincial Cancer Hospital, The First Affiliated Hospital of USTC, Hefei, China; [8]Department of Gynecologic Oncology, Chongqing University Cancer Hospital & Chongqing Cancer Institute & Chongqing Cancer Hospital, Chongqing, China; [9]Department of Obstetrics and Gynecology, The First Hospital of Jilin University, Changchun, China; [10]Department of Gynecology, Cancer Hospital of China Medical University, Liaoning Cancer Hospital and Institute, Shenyang, Liaoning, China; [11]Department of Obstetrics and Gynecology, First Affiliated Hospital, Dalian Medical University, Dalian, China; [12]Department of Gynecological Oncology, Peking University School of Oncology, Beijing Cancer Hospital, Beijing Institute for Cancer Research, Beijing, China; [13]Department of Gynecological Oncology, Hubei Cancer Hospital, Tongji Medical College, Huazhong University of Science and Technology, Wuhan, China; [14]Cancer Center, Union Hospital, Tongji Medical College, Huazhong University of Science and Technology, Wuhan, China; [15]Department of Oncology and Hematology, the Second Hospital of Jilin University, Changchun, China; [16]Department of Medical Oncology, Jilin Cancer Hospital, Changchun, China; [17]Department of Gynecology, Harbin Medical University Cancer Hospital, Harbin, China; [18]Beigene (Beijing) Co. Ltd., Beijing, China; [19]BeiGene (Shanghai) Co., Ltd., Shanghai, China

Poster: 820P
European Society of Medical Oncology
September 19-21, 2020, Virtual Congress

INTRODUCTION

- Poly (ADP-ribose) polymerase (PARP) proteins are involved in the repair of single- and double-strand DNA breaks[1,2]
- Small-molecule PARP inhibitors are a class of therapeutic agents used to treat various malignancies, including tumors harboring *BRCA1/2* mutations[3,4]
 - PARP inhibition impairs DNA repair and traps PARP proteins on damaged DNA, resulting in cytotoxicity that is exacerbated in homologous recombination deficient cells[3]
- Pamiparib is an investigational, potent, selective, oral PARP1/2 inhibitor[5]
 - In preclinical models, pamiparib demonstrated PARP-DNA complex trapping, inhibition of PARylation, brain penetration, and antitumor activity[5]
- Results of the first-in-human study (BGB-290-AU-002) showed that pamiparib was generally well tolerated and had antitumor activity, notably in patients with epithelial ovarian cancer (OC)[6]
 - The recommended phase 2 dose (RP2D) of pamiparib was established as 60 mg orally (PO) twice daily (BID)
- The RP2D was confirmed in Chinese patients with advanced OC or triple-negative breast cancer in the dose-escalation (phase 1) portion of this phase 1/2 study (BGB-290-102)[7]
- Here we present the preliminary results of the RP2D-expansion in Chinese patients with *BRCA1/2* mutation–positive platinum-sensitive OC (PSOC) or platinum-resistant OC (PROC)

METHODS

Study Design

- This phase 1/2 study is an open-label, multicenter study assessing the safety and antitumor activity of pamiparib in adult (≥18 years) Chinese patients with advanced solid tumors whose disease progressed despite standard therapy or for which there is no standard therapy (Figure 1)
- Pamiparib 60 mg was administered PO BID on Day 1 of Cycle 1 (21-day cycle) and continuously in all subsequent cycles until disease progression, toxicity, or patient withdrawal

Figure 1: Study Design

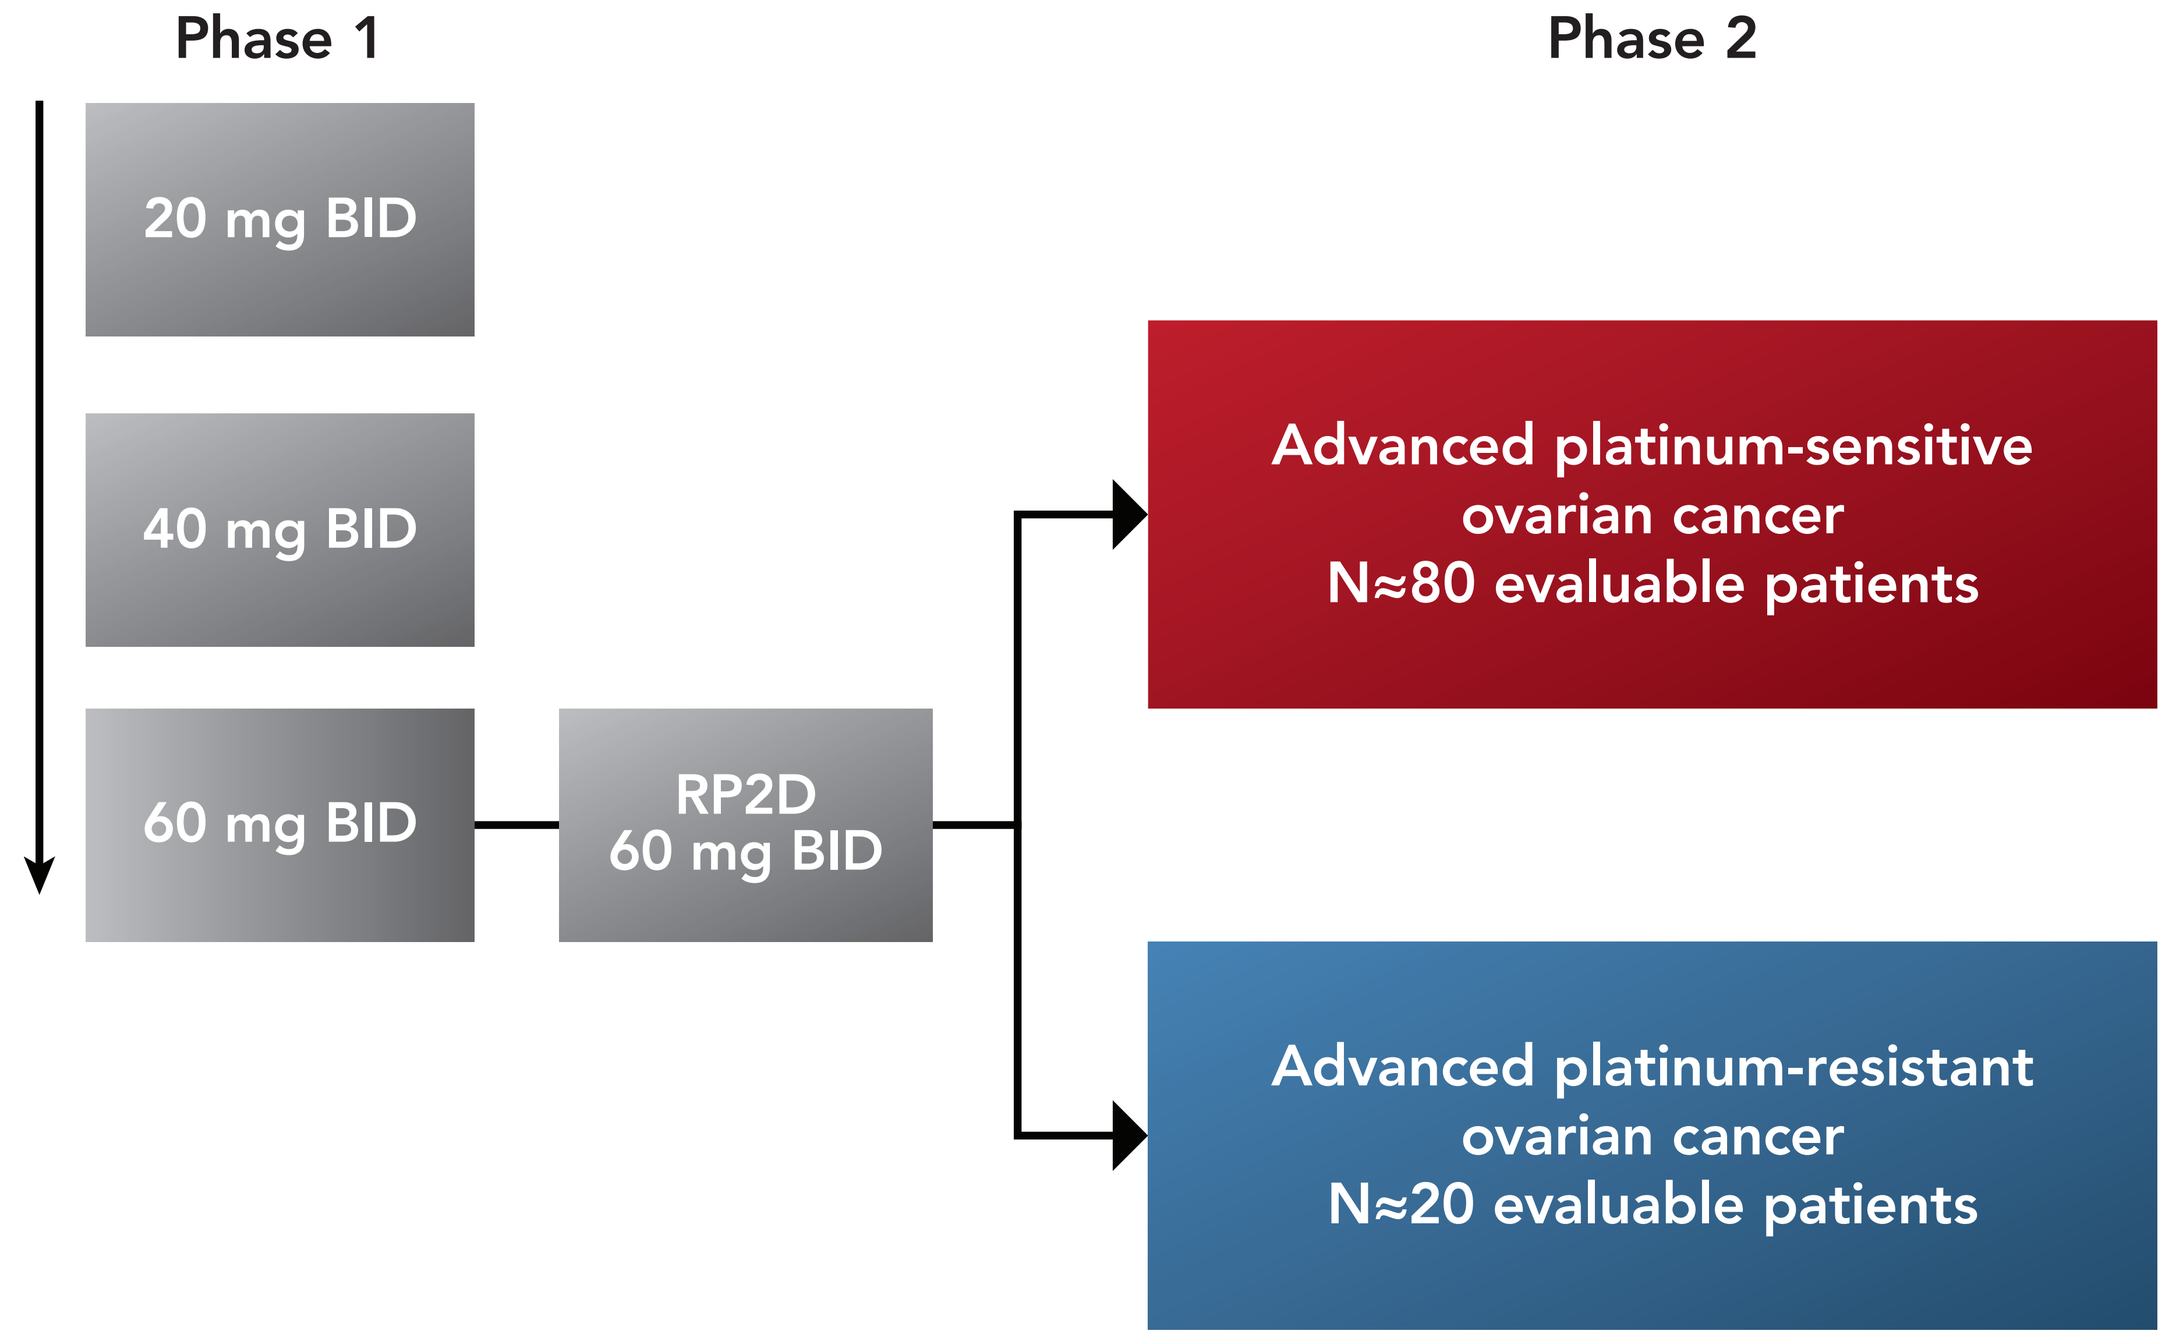

Phase 1
Phase 2
20 mg BID
40 mg BID
60 mg BID
RP2D
60 mg BID
Advanced platinum-sensitive ovarian cancer
N≈80 evaluable patients
Advanced platinum-resistant ovarian cancer
N≈20 evaluable patients

Abbreviations: BID, twice daily; RP2D, recommended phase 2 dose.

Study Population

- Phase 2 enrolled female patients with histologically or cytologically confirmed high-grade, non-mucinous, epithelial OC (including fallopian or primary peritoneal cancer) and an Eastern Cooperative Oncology Group (ECOG) performance status of 0-1; patients had advanced OC that was either platinum-sensitive (Cohort 1) or platinum-resistant (Cohort 2)
 - Platinum-sensitive was defined as disease progression occurring ≥6 months after last platinum treatment
 - Platinum-resistant was defined as disease progression that occurred <6 months after last platinum treatment
- Both cohorts enrolled patients with either known deleterious or suspected deleterious *gBRCA*^{mut} who received at least two lines of standard chemotherapy and were either currently experiencing relapsed disease or had discontinued the most recent standard treatment due to unacceptable toxicity
- Patients were excluded if they had untreated and/or active brain metastases or received chemotherapy, radiotherapy, biologic therapy, immunotherapy, investigational agent, anticancer Chinese medicine, or anticancer herbal remedies within 14 days of initiating study

Endpoints and Assessments

- The primary endpoint was objective response rate (ORR) based on independent review committee (IRC) per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1
- Secondary endpoints included duration of response and progression-free survival (PFS) by IRC and investigator review; disease control rate and clinical benefit rate by IRC and investigator review; ORR by investigator review; overall survival (OS); CA-125 response rate per Gynecologic Cancer Intergroup criteria; and pamiparib safety/tolerability profile
- Tumor imaging and CA-125 testing occurred every 6 weeks after the first dose of pamiparib for the first 18 weeks, every 9 weeks for the remaining period in the first year, and every 12 weeks from the second year onward
- Safety and tolerability assessments were based on monitoring of adverse events (AEs), as well as on vital signs, electrocardiograms, physical examinations, and clinical laboratory results
 - A protocol amendment (PA; protocol v5.0) initiated a more proactive dose modification algorithm and close hematology monitoring; a pre- and post-PA safety analysis was conducted; the post-PA subgroup included patients who signed the first informed consent form under the amended protocol

Statistical Methods

- Antitumor activity per RECIST v1.1 was assessed in all efficacy-evaluable patients
- Safety and tolerability were evaluated in all patients who received ≥1 dose of pamiparib

RESULTS

Patient Demographics and Baseline Disease Characteristics

- As of February 2, 2020, 113 patients who had received multiple lines of therapy (PSOC, n=90; PROC, n=23) with a median age of 54 years were enrolled (Table 1), of which, 74 discontinued from treatment
 - Reasons for treatment discontinuation include progressive disease (n=47), AE (n=14), patient withdrawal (n=10), and investigator's decision (n=3)
- Median study follow-up was 12.2 months (range, 0.2-21.5)

Table 1: Patient Demographics and Baseline Characteristics

		PSOC (N=90)	PROC (N=23)	Total (N=113)
Median age, years (range)		54 (39-79)	54 (34-66)	54 (34-79)
<65 years		80 (88.9)	21 (91.3)	101 (89.4)
ECOG score, n (%)	0	42 (46.7)	10 (43.5)	52 (46.0)
	1	48 (53.3)	13 (56.5)	61 (54.0)
Number of prior lines of systemic chemotherapy, n (%)	2	52 (57.8)	3 (13.0)	55 (48.7)
	3	19 (21.1)	10 (43.5)	29 (25.7)
	4	8 (8.9)	6 (26.1)	14 (12.4)
	5	4 (4.4)	1 (4.3)	5 (4.4)
	≥6	7 (7.8)	3 (13.0)	10 (8.8)
gBRCA status, n (%)	*BRCA1* mutation	79 (87.8)	19 (82.6)	98 (86.7)
	BRCA2 mutation	11 (12.2)	4 (17.4)	15 (13.3)
Years from initial diagnosis, median (range)		3.9 (1.4-13.6)	3.6 (1.1-7.1)	3.9 (1.1-13.6)
FIGO histology, n (%)	Serous epithelial tumors	85 (94.4)	23 (100.0)	108 (95.6)
	Endometrioid epithelial tumors	4 (4.4)	0	4 (3.5)
	Mixed epithelial tumors	1 (1.1)	0	1 (0.9)
Target lesion diameter per IRC at study entry, n (%)	<50 mm	41 (45.6)	8 (34.8)	49 (43.4)
	≥50 mm	41 (45.6)	11 (47.8)	52 (46.0)
	Missing	8 (8.9)	4 (17.4)	12 (10.6)
CA-125 value at study entry, n (%)	<70 kU/L	16 (17.8)	2 (8.7)	18 (15.9)
	≥70 kU/L	74 (82.2)	21 (91.3)	95 (84.1)

Abbreviations: *BRCA*, breast cancer susceptibility gene; ECOG, Eastern Cooperative Oncology Group; FIGO, International Federation of Gynecology and Obstetrics; IRC, independent review committee; PROC, platinum-resistant ovarian cancer; PSOC, platinum-sensitive ovarian cancer.

Antitumor Activity

- Objective response rate and complete response rate per RECIST v1.1 was similar between IRC and investigator assessment (Table 2)
- CA-125 response rate was 79.7% (95% CI, 68.8-88.2) in PSOC patients and 38.1% (95% CI, 18.1-61.6) in PROC patients

Table 2: Tumor Response by Patient Cohort in the Efficacy-Evaluable Population by IRC and Investigator Assessment Based on RECIST v1.1

			PSOC (n=82)	PROC (n=19)
IRC Assessment	BOR, n (%)	Complete response	8 (9.8)	0 (0.0)
		Partial response	45 (54.9)	6 (31.6)
		Stable disease	25 (30.5)	12 (63.2)
		Progressive disease	4 (4.9)	1 (5.3)
		Not estimable	0 (0.0)	0 (0.0)
	ORR, % (95% CI)		64.6 (53.3-74.9)	31.6 (12.6-56.6)
	DCR, % (95% CI)		95.1 (88.0-98.7)	94.7 (74.0-99.9)
	CBR ≥24 weeks, % (95% CI)		74.4 (63.6-83.4)	52.6 (28.9-75.6)
	Median time to response, months (min, max)		1.7 (1.3, 6.3)	1.4 (1.2, 1.4)
Investigator Assessment	BOR, n (%)	Complete response	5 (6.1)	0 (0.0)
		Partial response	46 (56.1)	5 (26.3)
		Stable disease	28 (34.1)	10 (52.6)
		Progressive disease	3 (3.7)	3 (15.8)
		Not estimable	0 (0.0)	1 (5.3)
	ORR, % (95% CI)		62.2 (50.8-72.7)	26.3 (9.1-51.2)
	DCR, % (95% CI)		96.3 (89.7-99.2)	78.9 (54.4-93.9)
	CBR ≥24 weeks, % (95% CI)		72.0 (60.9-81.3)	52.6 (28.9-75.6)
	Median time to response, months (min, max)		2.7 (1.2, 8.3)	1.3 (1.2, 4.2)

CBR=CR+PR+SD ≥24 weeks; DCR=CR+PR+SD; ORR=CR+PR.
Abbreviations: BOR, best overall response; CBR, clinical benefit rate; CI, confidence interval; DCR, disease control rate; IRC, independent review committee; NE, not estimable; OC, ovarian cancer; ORR, objective response rate; PROC, platinum-resistant ovarian cancer; PSOC, platinum-sensitive ovarian cancer; RECIST, Response Evaluation Criteria in Solid Tumors.

- In both cohorts, most patients had a reduction in target lesions from baseline (Figure 2A and 2B)

Figure 2: Best Change in Sum of Target Lesion Diameters by Confirmed Best Overall Response of the Efficacy-Evaluable Population per RECIST v1.1

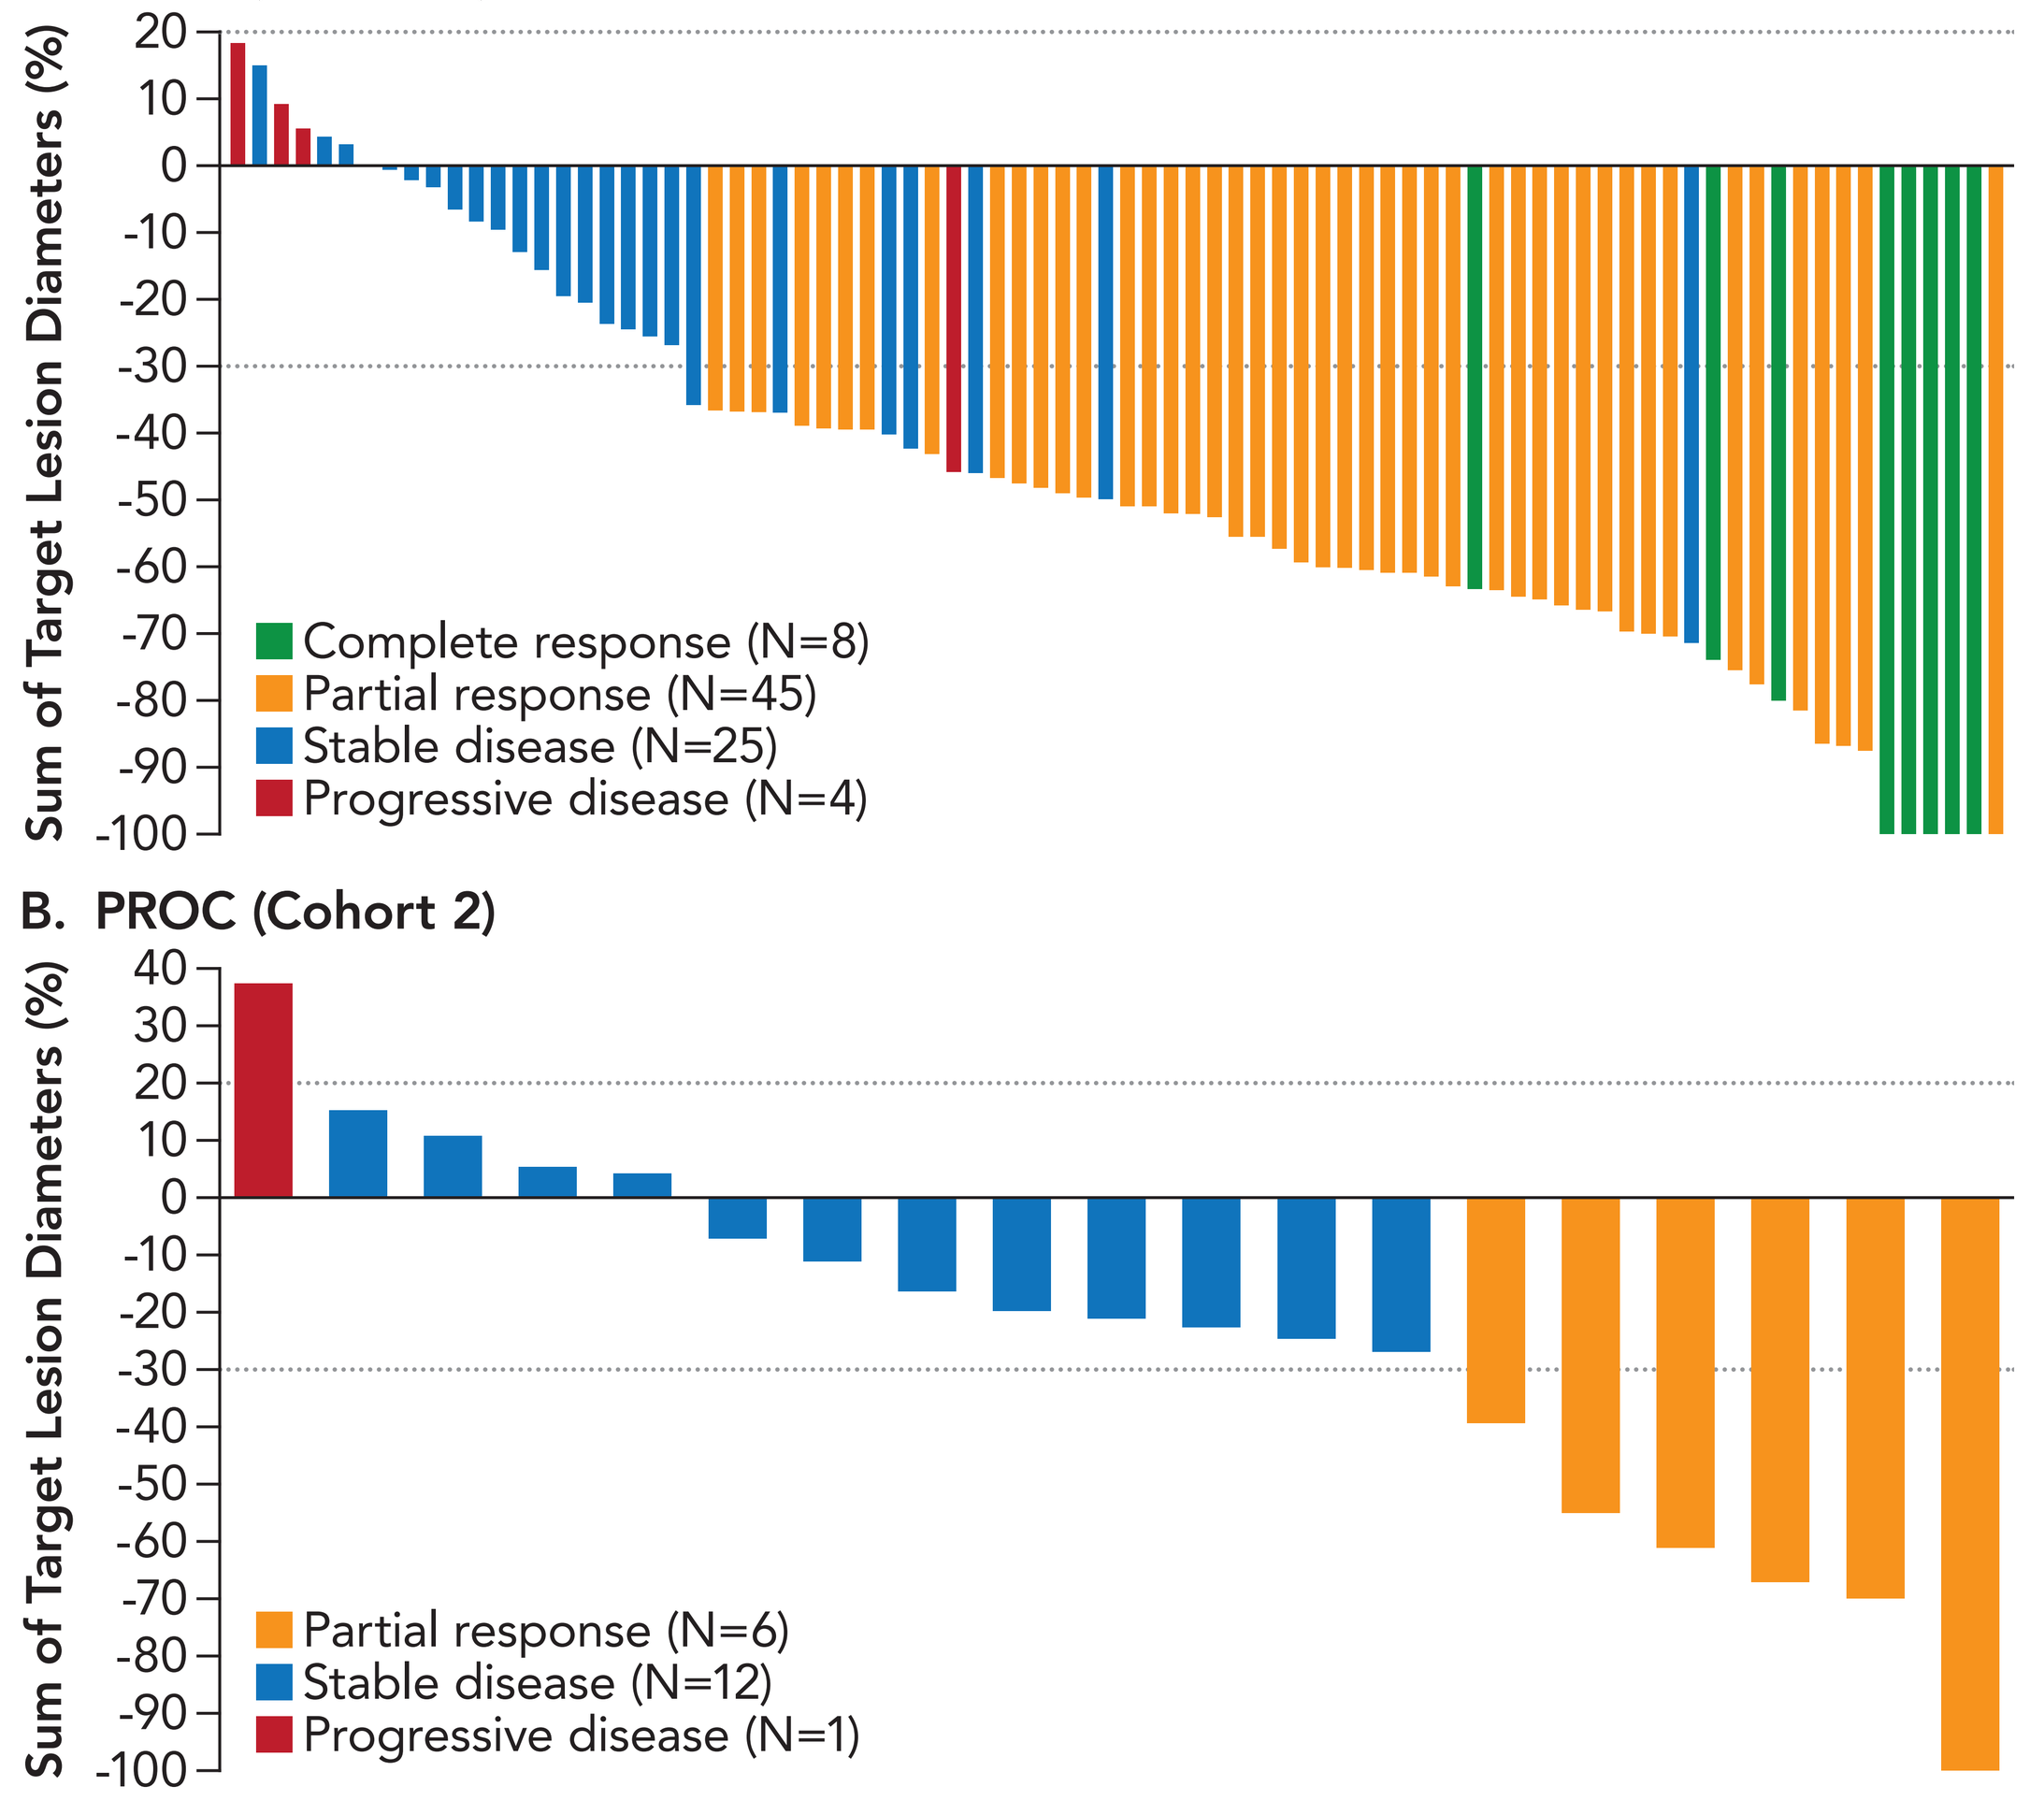

A. PSOC (Cohort 1)
Sum of Target Lesion Diameters (%)
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
Complete response (N=8)
Partial response (N=45)
Stable disease (N=25)
Progressive disease (N=4)
B. PROC (Cohort 2)
Sum of Target Lesion Diameters (%)
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
Partial response (N=6)
Stable disease (N=12)
Progressive disease (N=1)

Patients were considered efficacy-evaluable if they had measurable disease at baseline per RECIST v1.1 and had ≥1 postbaseline tumor assessment, unless treatment had been discontinued due to clinical progression or death prior to tumor assessment.
Abbreviations: PROC, platinum-resistant ovarian cancer; PSOC, platinum-sensitive ovarian cancer; RECIST, Response Evaluation Criteria in Solid Tumors.

- Primary endpoint of ORR in PSOC patients was generally consistent across all subgroups analyzed (Figure 3)

Figure 3: IRC Assessed Objective Response Rates (RECIST v1.1) by Baseline Characteristics in PSOC Patients

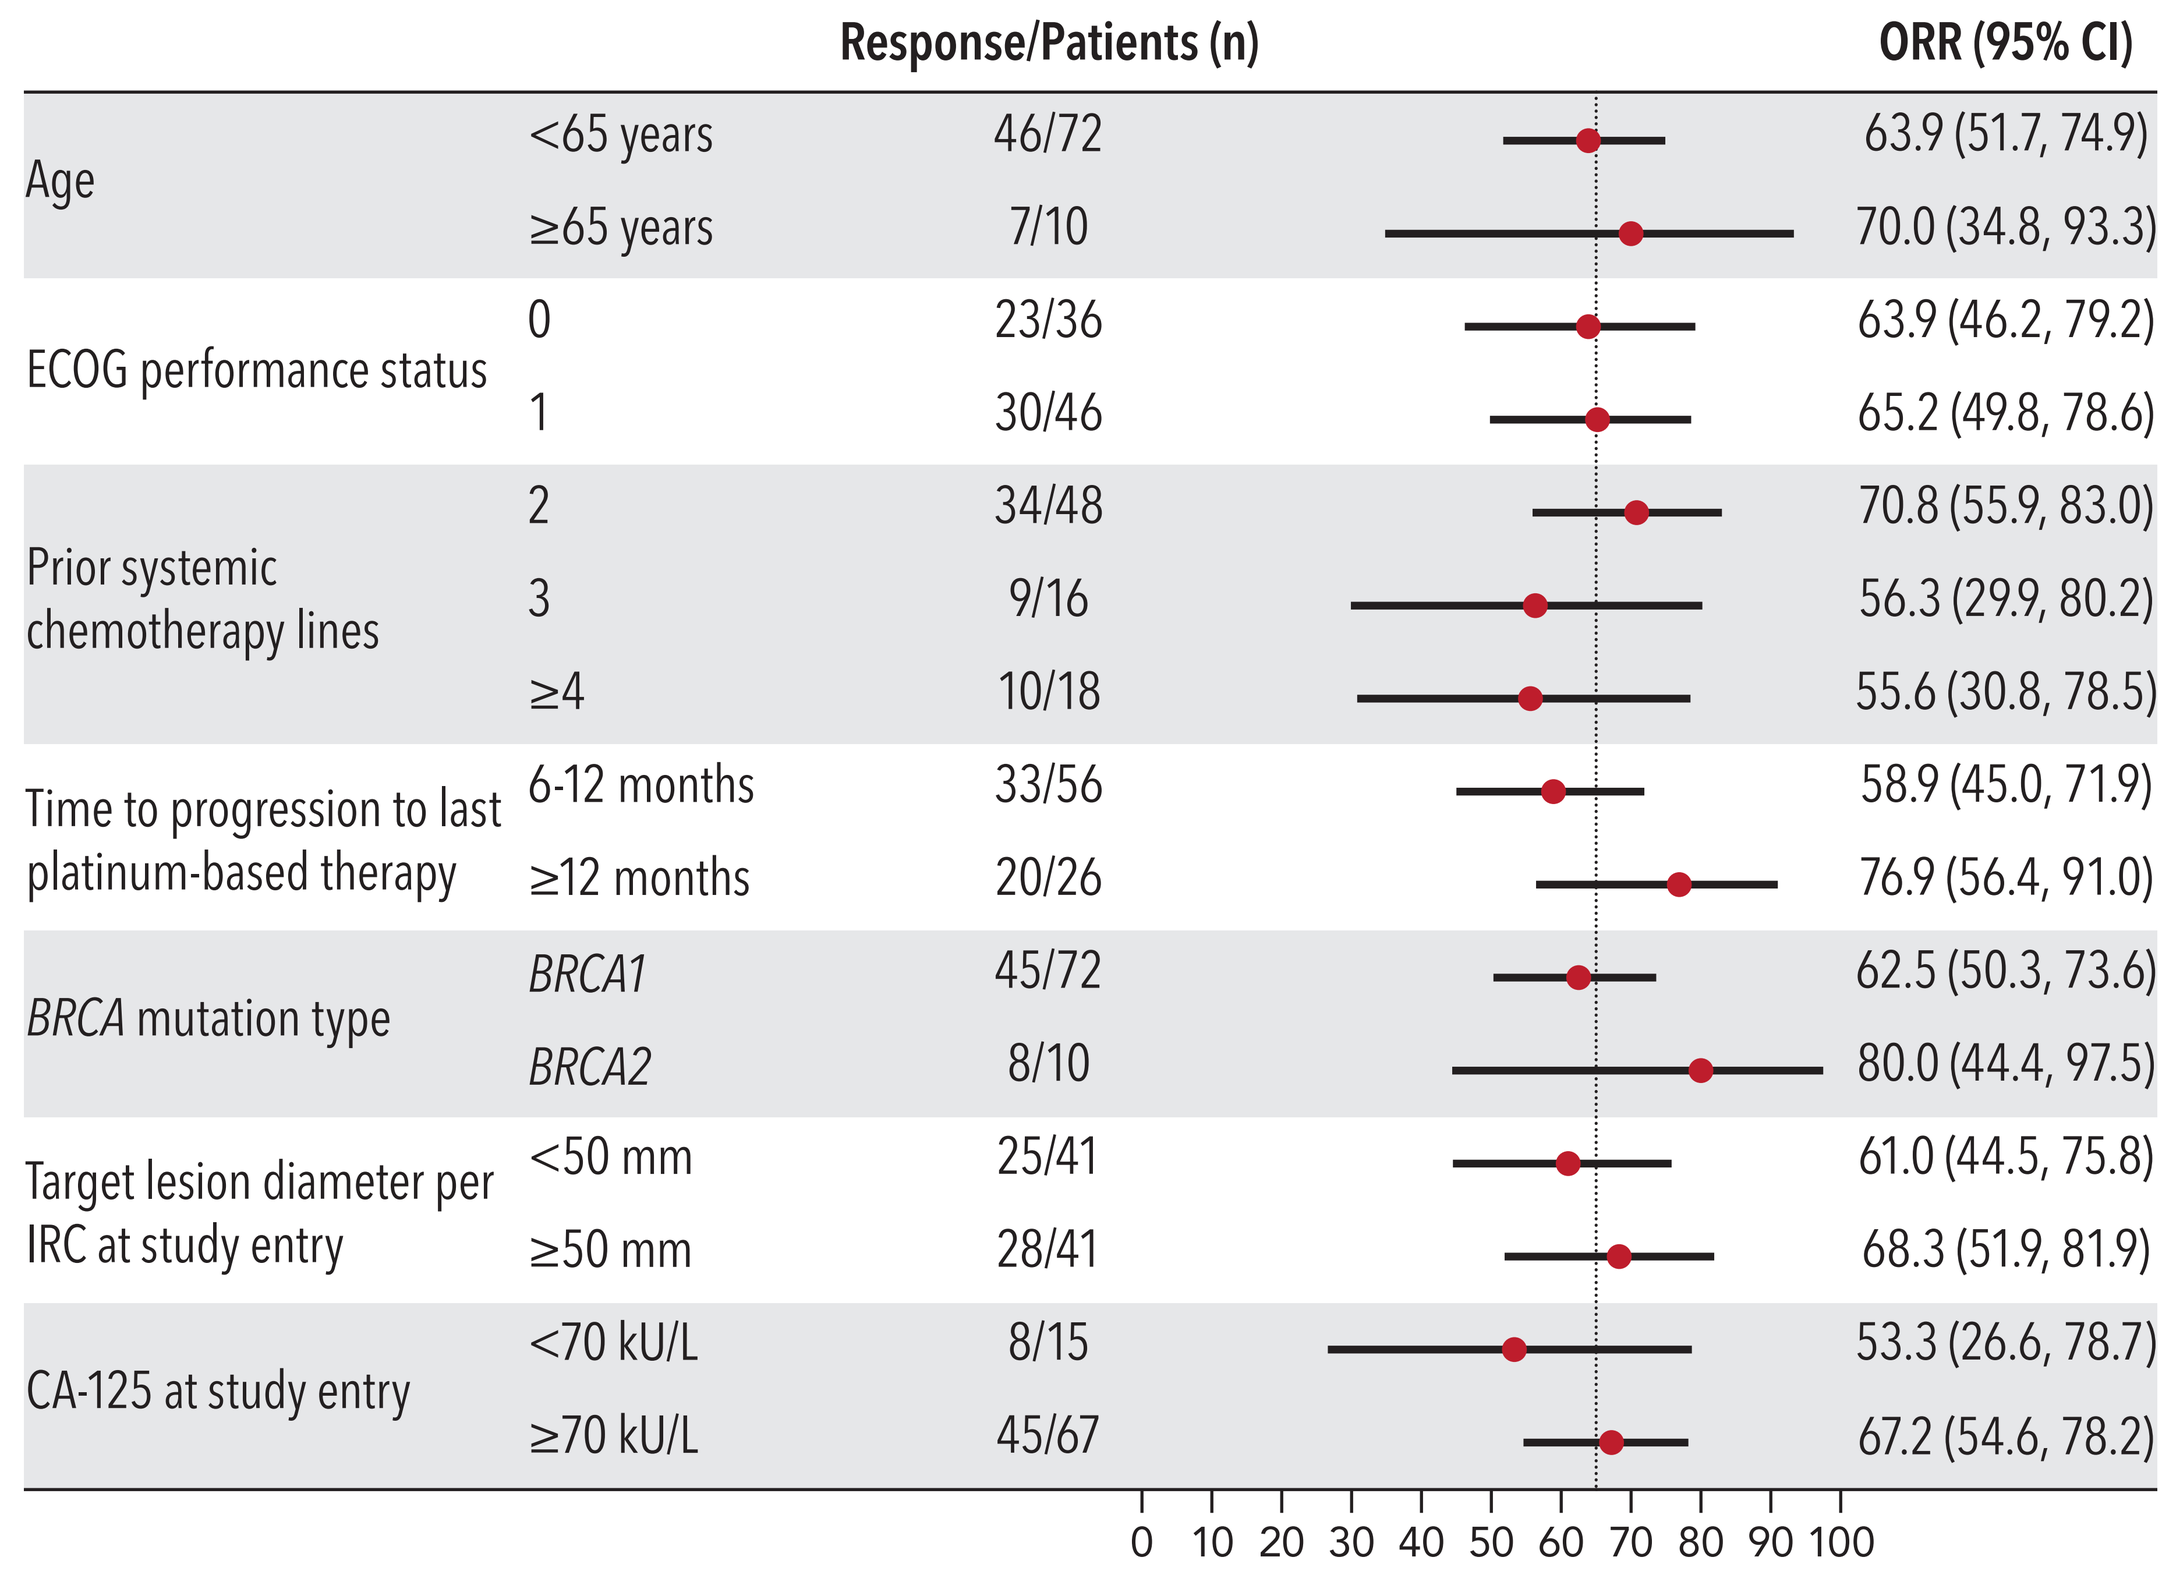

Response/Patients (n)
ORR (95% CI)
Age
<65 years 46/72 63.9 (51.7, 74.9)
≥65 years 7/10 70.0 (34.8, 93.3)
ECOG performance status
0 23/36 63.9 (46.2, 79.2)
1 30/46 65.2 (49.8, 78.6)
Prior systemic chemotherapy lines
2 34/48 70.8 (55.9, 83.0)
3 9/16 56.3 (29.9, 80.2)
≥4 10/18 55.6 (30.8, 78.5)
Time to progression to last platinum-based therapy
6-12 months 33/56 58.9 (45.0, 71.9)
≥12 months 20/26 76.9 (56.4, 91.0)
BRCA mutation type
BRCA1 45/72 62.5 (50.3, 73.6)
BRCA2 8/10 80.0 (44.4, 97.5)
Target lesion diameter per IRC at study entry
<50 mm 25/41 61.0 (44.5, 75.8)
≥50 mm 28/41 68.3 (51.9, 81.9)
CA-125 at study entry
<70 kU/L 8/15 53.3 (26.6, 78.7)
≥70 kU/L 45/67 67.2 (54.6, 78.2)
0 10 20 30 40 50 60 70 80 90 100

Data are presented as ORR (range); the dotted line corresponds to 65% ORR.
Abbreviations: *BRCA*, breast cancer susceptibility gene; CI, confidence interval; ECOG, Eastern Cooperative Oncology Group; IRC, independent review committee; ORR, objective response rate; PSOC, platinum-sensitive ovarian cancer; RECIST, Response Evaluation Criteria in Solid Tumors.

- In PSOC patients, median duration of response was 14.5 months (95% CI, 11.1-NE) (Figure 4A) and median PFS was 15.2 months (95% CI, 10.35-NE) (Figure 4B)

Figure 4: Duration of Response and Progression-Free Survival in PSOC Patients by IRC Assessment per RECIST v1.1

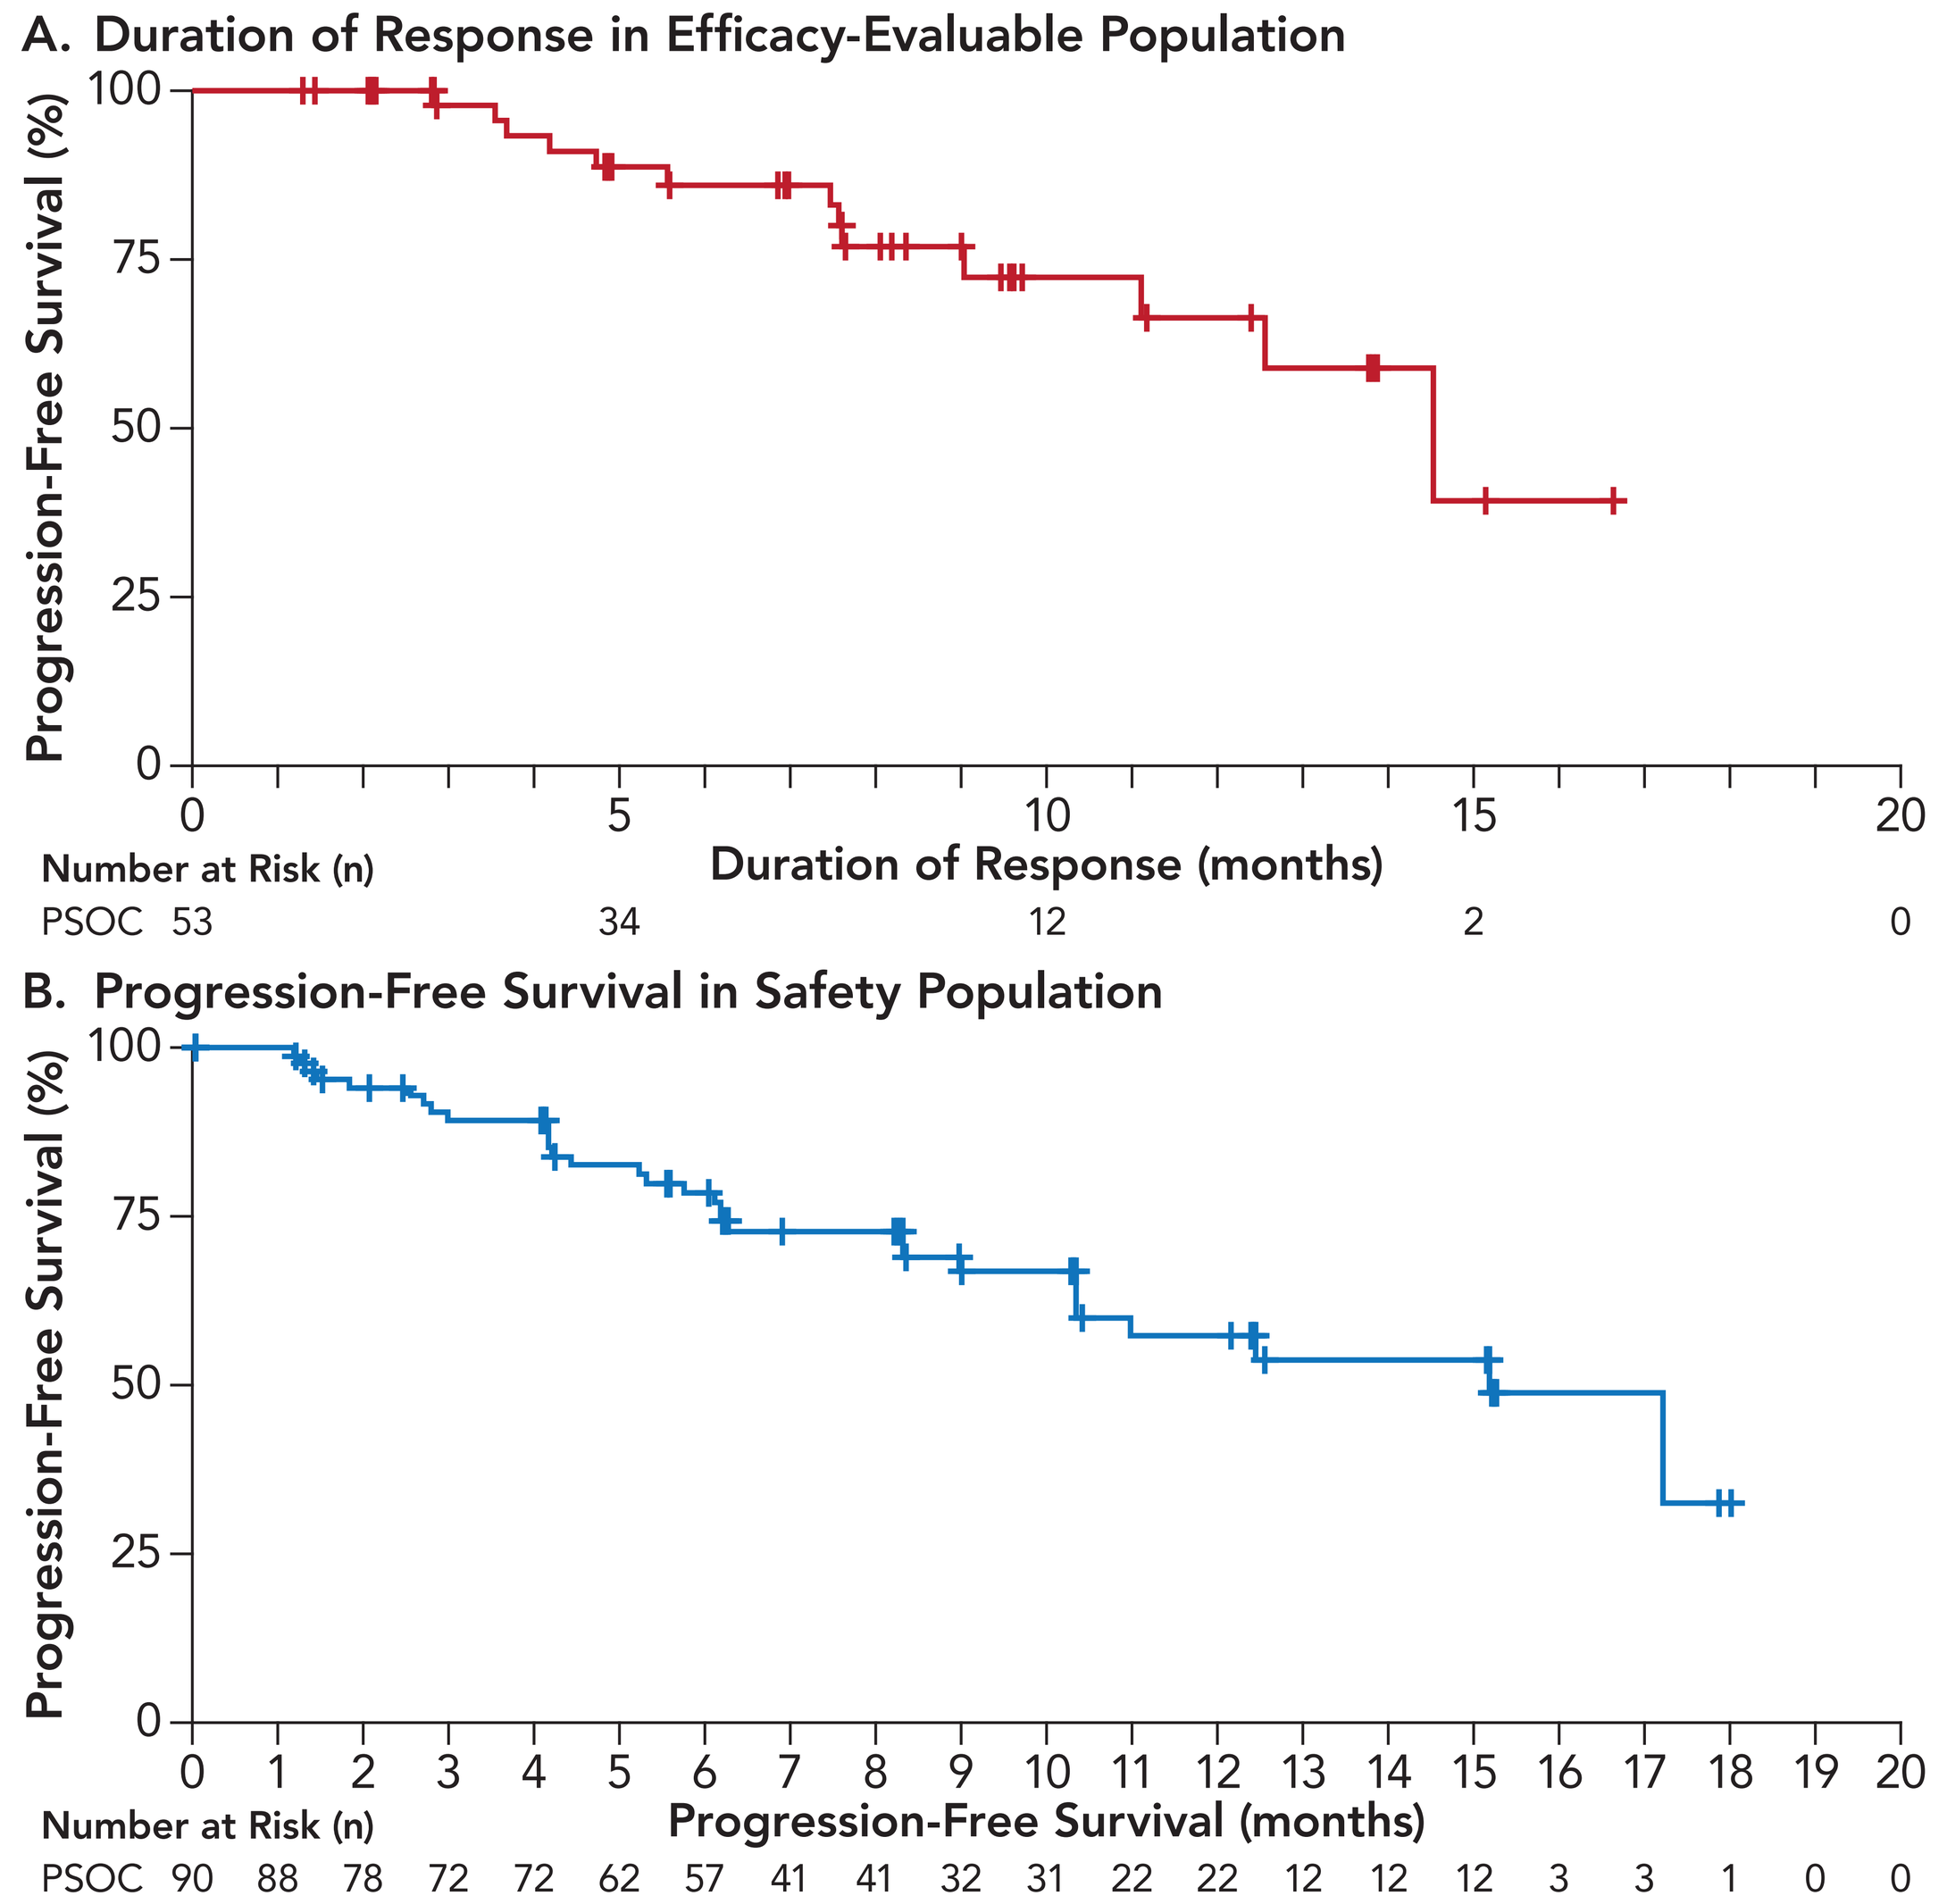

A. Duration of Response in Efficacy-Evaluable Population
Progression-Free Survival (%)
100
75
50
25
0
0 5 10 15 20
Duration of Response (months)
Number at Risk (n)
PSOC 53 34 12 2 0
B. Progression-Free Survival in Safety Population
Progression-Free Survival (%)
100
75
50
25
0
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20
Progression-Free Survival (months)
Number at Risk (n)
PSOC 90 88 78 72 72 62 57 41 41 32 31 22 22 12 12 12 3 3 1 0 0

Abbreviations: PSOC, platinum-sensitive ovarian cancer; RECIST, Response Evaluation Criteria in Solid Tumors.

Safety and Tolerability

- Median treatment duration was 8.3 (range, 0.1-19.3) months in PSOC patients and 4.1 (range, 0.1-19.9) months in PROC patients
- Across both PSOC and PROC cohorts, the most frequently reported AEs of any grade were gastrointestinal disorders and hematologic toxicities (Table 3)

CONCLUSIONS

- Statistical and clinical meaningful and durable response were observed in patients with PSOC; clinical meaningful and durable responses were observed in patients with PROC
- Pamiparib 60 mg PO BID demonstrated a generally tolerated and acceptable safety profile
 - The overall safety profile was generally consistent between patients with PSOC and PROC
- Similar to other PARP inhibitors, hematologic toxicities were the most significant safety events observed
 - The hematological toxicities were manageable and could be better managed with a more proactive modification plan and closer hematologic monitoring
 - No myelodysplastic syndrome reported
 - No significant complications (eg grade ≥3 hemorrhage, fever, or infection) potentially related to hematologic toxicity were reported

- In the post-PA subgroup, the percentage of patients who experienced grade ≥3 hematologic AEs was lower, compared with the pre-PA subgroup (Table 3)
- No patient in the post-PA subgroup experienced a hematologic AE that led to treatment discontinuation

Table 3: Treatment-Emergent Adverse Events of Any Grade (≥20% in the Total Population) and of Grade ≥3

	PSOC (n=90)		PROC (n=23)		Total (N=113)		Pre-PA (n=74)		Post-PA (n=39)	
	All Grades	Grade ≥3	All Grades	Grade ≥3	All Grades	Grade ≥3	All Grades	Grade ≥3	All Grades	Grade ≥3
Anemia	80 (88.9)	34 (37.8)	21 (91.3)	13 (56.5)	101 (89.4)	47 (41.6)	65 (87.8)	37 (50.0)	36 (92.3)	10 (25.6)
Nausea	61 (67.8)	1 (1.1)	16 (69.6)	0 (0.0)	77 (68.1)	1 (0.9)	48 (64.9)	1 (1.4)	29 (74.4)	0 (0.0)
Decreased neutrophil count	56 (62.2)	28 (31.1)	13 (56.5)	10 (43.5)	69 (61.1)	38 (33.6)	46 (62.2)	29 (39.2)	23 (59.0)	9 (23.1)
Decreased white blood cell count	54 (60.0)	17 (18.9)	14 (60.9)	5 (21.7)	68 (60.2)	22 (19.5)	44 (59.5)	16 (21.6)	24 (61.5)	6 (15.4)
Vomiting	46 (51.1)	4 (4.4)	11 (47.8)	1 (4.3)	57 (50.4)	5 (4.4)	42 (56.8)	5 (6.8)	15 (38.5)	0 (0.0)
Decreased platelet count	25 (27.8)	4 (4.4)	10 (43.5)	1 (4.3)	35 (31.0)	5 (4.4)	22 (29.7)	2 (2.7)	13 (33.3)	3 (7.7)
Decreased appetite	29 (32.2)	0 (0.0)	5 (21.7)	0 (0.0)	34 (30.1)	0 (0.0)	21 (28.4)	0 (0.0)	13 (33.3)	0 (0.0)
Asthenia	26 (28.9)	1 (1.1)	6 (26.1)	0 (0.0)	32 (28.3)	1 (0.9)	20 (27.0)	1 (1.4)	12 (30.8)	0 (0.0)
Diarrhea	19 (21.1)	3 (3.3)	6 (26.1)	0 (0.0)	25 (22.1)	3 (2.7)	19 (25.7)	1 (1.4)	6 (15.4)	2 (5.1)
Increased AST	20 (22.2)	0 (0.0)	4 (17.4)	1 (4.3)	24 (21.2)	1 (0.9)	15 (20.3)	1 (1.4)	9 (23.1)	0 (0.0)
Decreased lymphocyte count	19 (21.1)	6 (6.7)	5 (21.7)	2 (8.7)	24 (21.2)	8 (7.1)	19 (25.7)	7 (9.5)	5 (12.8)	1 (2.6)
Increased ALT	18 (20.0)	1 (1.1)	5 (21.7)	0 (0.0)	23 (20.4)	1 (0.9)	15 (20.3)	1 (1.4)	8 (20.5)	0 (0.0)
Leukopenia	20 (22.2)	9 (10.0)	3 (13.0)	3 (13.0)	23 (20.4)	12 (10.6)	17 (23.0)	11 (14.9)	6 (15.4)	1 (2.6)

Data presented as n (%).
Abbreviations: ALT, alanine aminotransferase; AST, aspartate aminotransferase; PA, protocol amendment; PROC, platinum-resistant ovarian cancer; PSOC, platinum-sensitive ovarian cancer.

REFERENCES

1. Thomas C, Tulin AV. Poly-ADP-ribose polymerase: machinery for nuclear processes. *Mol Aspects Med.* 2013;34(6):1124-1137.
2. Dziadkowiec KN, Gasiorowska E, Nowak-Markwitz E, Jankowska A. PARP inhibitors: review of mechanisms of action and BRCA1/2 mutation targeting. *Prz Menopauzalny.* 2016;15(4):215-219.
3. Lord CJ, Ashworth A. PARP inhibitors: Synthetic lethality in the clinic. *Science.* 2017;355(6330):1152-1158.
4. Mateo J, Lord CJ, Serra V, et al. A decade of clinical development of PARP inhibitors in perspective. *Ann Oncol.* 2019;30(9):1437-1447.
5. Xiong Y, Guo Y, Liu Y, et al. Pamiparib is a potent and selective PARP inhibitor with unique potential for the treatment of brain tumor. *Neoplasia.* 2020;22(9):431-440.
6. Lickliter J, Mileshkin L, Voskoboynik M, et al. Dose escalation/expansion study to investigate the safety, pharmacokinetics, food effect, and antitumor activity of BGB-290 in patients with advanced solid tumors. *Ann Oncol.* 2017;28(suppl 5):v123.
7. Xu B, Yin Y, Song Y, et al. A phase I dose escalation study of BGB-290 in Chinese subjects with advanced ovarian, fallopian, and primary peritoneal, or advanced triple-negative breast cancer. *Cancer Res.* 2018;78(suppl 13):Abstract CT050.

CONFLICTS OF INTEREST

LL, **XM**, **ML** are all employees with stock options at BeiGene, Ltd. **All other authors** report drug and research funding support from BeiGene, Ltd., except for **JZ**, **GL**, and **YC** who have nothing to disclose.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff, the study patients, and their families. BeiGene, Ltd. provided financial support for this manuscript, including writing and editorial assistance by Cathy R. Winter, PhD, Regina Switzer, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com