ASSOCIATION BETWEEN IMMUNE AND TUMOR GENE SIGNATURES WITH RESPONSE OR RESISTANCE TO TISLELIZUMAB MONOTHERAPY OR IN COMBINATION WITH CHEMOTHERAPY IN GASTROESOPHAGEAL ADENOCARCINOMA

Jianming Xu[1], Tianshu Liu[2], Jong-Seok Lee[3], Yuxian Bai[4], Yee Chao[5], Silu Yang[6], Yun Zhang[6], Xiaopeng Ma[6], Pei Zhang[6], Jianfei Zheng[6], Yanyan Li[6], Jin Wang[6], Lin Shen[7]

[1]The Fifth Medical Center, General Hospital of Chinese People's Liberation Army, Beijing, China; [2]Zhongshan Hospital Fudan University, Shanghai, China; [3]Seoul National University Bundang Hospital, Seongnam-si, Gyeonggi-do, Republic of Korea; [4]Harbin Medical University Cancer Hospital, Harbin, China; [5]Taipei Veterans General Hospital, Taipei, Taiwan; [6]BeiGene (Beijing) Co., Ltd., Beijing, China; [7]Peking University Cancer Hospital & Institute, Beijing, China

Abstract: 3115
American Society of Clinical Oncology
May 29-31, 2020, Chicago, IL

BACKGROUND

- Gastroesophageal adenocarcinoma (GEA), including gastric, gastroesophageal junction (GEJ), and esophageal adenocarcinoma (EAC), has been associated with high mortality and has a low 5-year overall survival rate[1,2] when diagnosed in advanced stage
 - While recently approved PD-1 inhibitors have shown moderate clinical benefit, identification of biomarkers that can predict response is of urgent need
- Exploring immune- and tumor-transcriptomic features and their association with anti-PD(L)1 efficacy may increase the understanding of the tumor microenvironment in GEA and aid in the identification of potential response/resistance mechanisms
- Tislelizumab, an anti–PD-1 monoclonal antibody, has been approved by the China National Medical Products Administration (NMPA) as a treatment for patients with classical Hodgkin's lymphoma and for patients with locally advanced or metastatic urothelial carcinoma (UC) with PD-L1 high expression
- Tislelizumab also demonstrated clinical benefit in patients with GEA as a single agent (NCT02407990 and NCT04068519) and in combination with chemotherapy (NCT03469557)[3,4,5]
- Here we present the retrospective association analysis of immune and tumor gene signatures with clinical efficacy from these studies

METHODS

Study Design

- Pooled analysis from three clinical trials
 - BGB-A317-001 (NCT02407990): first-in-human, multicenter, phase1a/1b dose-escalation/indication-expansion study
 - Samples were analyzed from patients with advanced or metastatic gastric/GEJ adenocarcinoma and EAC
 - BGB-A317-102 (NCT04068519): Chinese, multicenter, phase1/2 study
 - Samples were analyzed from patients with previously treated/untreated advanced or metastatic gastric/GEJ adenocarcinoma
 - BGB-A317-205 (NCT03469557): phase 2 first-line study of tislelizumab plus oxaliplatin and capecitabine in Chinese patients
 - Samples were analyzed from patients with locally advanced or metastatic HER2-negative gastric/GEJ adenocarcinoma

Gene Expression Profiling

- Gene expression profiling (GEP) using the HTG EdgeSeq Precision Immuno-Oncology Panel was performed on baseline tumor samples (formalin-fixed, paraffin-embedded blocks or cut slides)
- Signature scores were calculated using the Gene Set Variation Analysis (GSVA) package with publicly available gene signatures
- Differentially expressed gene signature (DEG) analysis was performed between responders and nonresponders using a Wilcoxon rank-sum test
- Nonresponder subgroups were hierarchically clustered by Euclidean distance metrics with average linkage by columns

Statistical Analysis

- Statistical significance was tested using a two-sided Wilcoxon test
- Potential associations with survival were analyzed using a log-rank test and Cox proportional hazards model

RESULTS

Patient Characteristics

- Of 120 enrolled patients, 87 had samples evaluable for GEP analysis (Table 1)

Table 1: Baseline Characteristics and Clinical Outcome

Characteristic		Monotherapy GEP (n=75)	Monotherapy Overall (n=105)	Combination Therapy GEP (n=12)	Combination Therapy Overall (n=15)
Age, n (%)	<65	51 (68)	66 (62.9)	9 (75)	10 (66.7)
	≥65	24 (32)	39 (37.1)	3 (25)	5 (33.3)
Sex, n (%)	Male	51 (68)	72 (68.6)	10 (83.3)	11 (73.3)
	Female	24 (32)	33 (31.4)	2 (16.7)	4 (26.7)
Histologic grade at baseline, n (%)	Poorly differentiated	32 (42.7)	49 (46.7)	8 (66.7)	9 (60)
	Moderately differentiated	31 (41.3)	39 (37.1)	1 (8.3)	2 (13.3)
	Well differentiated	2 (2.7)	2 (1.9)	1 (8.3)	1 (6.7)
	Unknown	10 (13.3)	15 (14.3)	2 (16.7)	3 (20)
Tumor type, n (%)	Gastric/GEJ adenocarcinoma	53 (70.7)	93 (77.5)	12 (100)	15 (100)
	EAC	22 (29.3)	27 (22.5)	0 (0)	0 (0)
Histological type, n (%)	Adenocarcinoma	65 (86.7)	87 (82.9)	0 (0)	0 (0)
	Signet ring cell carcinoma	3 (4)	4 (3.8)	0 (0)	0 (0)
	Others	7 (9.3)	14 (13.3)	12 (100)	15 (100)
Tumor stage, n (%)	III	4 (5.3)	5 (4.8)	1 (8.3)	1 (6.7)
	IV	71 (94.7)	100 (95.2)	11 (91.7)	14 (93.3)
Response, n (%)	PR	7 (9.3)	12 (11.4)	6 (50)	7 (46.7)
	SD	12 (16)	20 (19)	2 (16.7)	3 (20)
	PD	41 (54.7)	55 (52.4)	1 (8.3)	1 (6.7)
	Non-CR/Non-PD	0 (0)	0 (0)	2 (16.7)	2 (13.3)
	NA	15 (20)	18 (17.1)	1 (8.3)	2 (13.3)
ORR,% (95% CI)		**9.3 (3.8, 18.3)**	**11.4 (6.0, 19.1)**	**50 (21.1, 78.9)**	**46.7 (21.3, 73.4)**
Median PFS, months (95% CI)		**1.97 (1.68, 2.10)**	**2.05 (1.94, 2.14)**	**6.11 (2.76, NR)**	**6.11 (3.78, NR)**
Median OS, months (95% CI)		**5.26 (3.78, 7.29)**	**5.65 (4.27, 8.64)**	**NR (5.88, NR)**	**NR (7.03, NR)**
Median follow up, months (95% CI)		**14.4 (13.9, 21.2)**	**14.5 (13.9, 18.2)**	**15.5 (14.7, 17.2)**	**15.4 (14.7, 17.2)**

Abbreviations: CI, confidence interval; CR, complete response; EAC, esophageal adenocarcinoma; GEJ, gastroesophageal junction; GEP, gene expression profiling; NA, not available; NR, not reached; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PR, partial response; SD, stable disease.

Association of IFNγ-related Gene Signature With Clinical Outcomes of Tislezumab Monotherapy

- A 6-gene IFNγ signature (*IFNγ*, *CXCL9*, *CXCL10*, *IDO1*, *STAT1* and *HLA-DRA*)[6] was significantly increased in tumors from responders (R) versus non-responders (NR) as revealed by DEG and Box-plot analysis (LogFC=0.35, *P*=0.041, Figure 1A)
- With a median IFNγ signature cutoff at -0.076, higher objective response rates (ORR) were observed in patients with IFNγ-high versus -low (ORR=16.2% vs 2.6%; Figure 1B); moreover, 9/11 patients with durable progression-free survival (PFS) (≥6 months) were IFNγ-high (Figure 1C)

Figure 1: IFNγ-related Gene Signatures Were Enriched in Responders to Tislelizumab Monotherapy

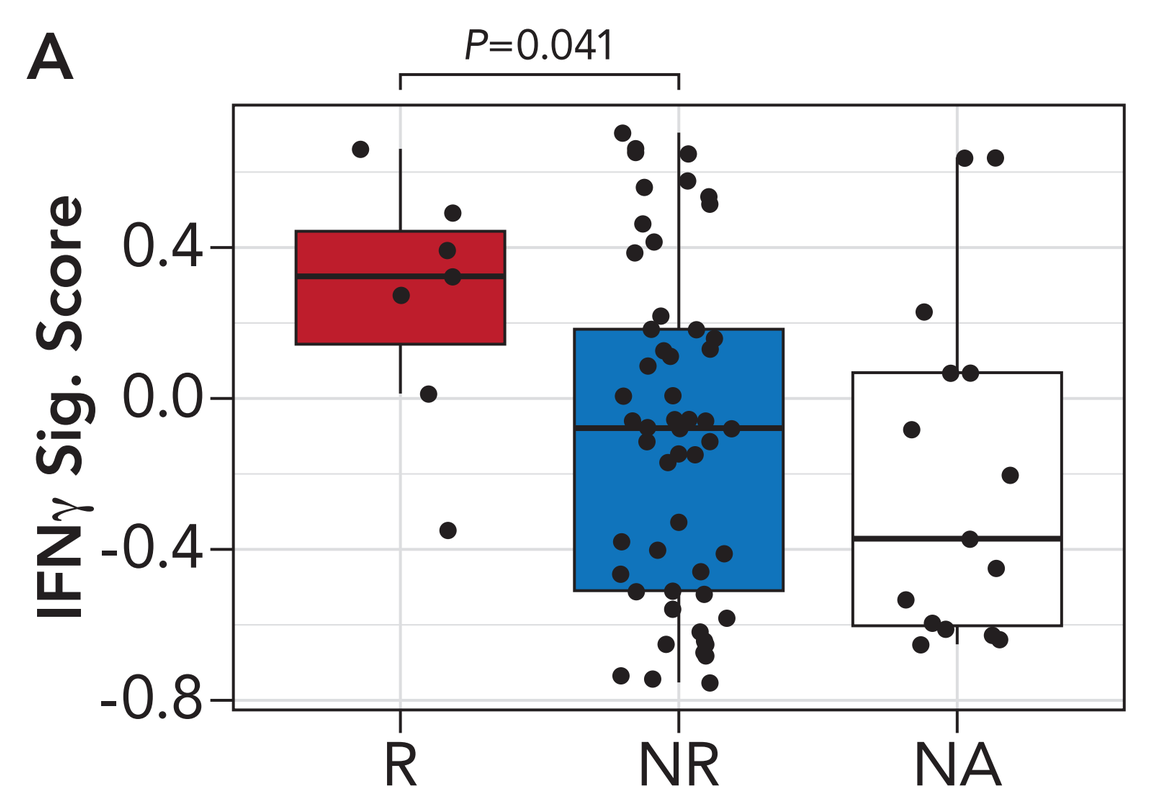
A
P=0.041
IFNγ Sig. Score
0.4
0.0
-0.4
-0.8
R
NR
NA

B

	IFNγ-high (n=37)	IFNγ-low (n=38)	BEP (n=75)
ORR	**16.2%**	**2.6%**	**9.3%**
DCR	**32.4%**	**18.4%**	**25.3%**
PR	16.2%	2.6%	9.3%
SD	16.2%	15.8%	16.0%
PD	54.1%	55.3%	54.7%

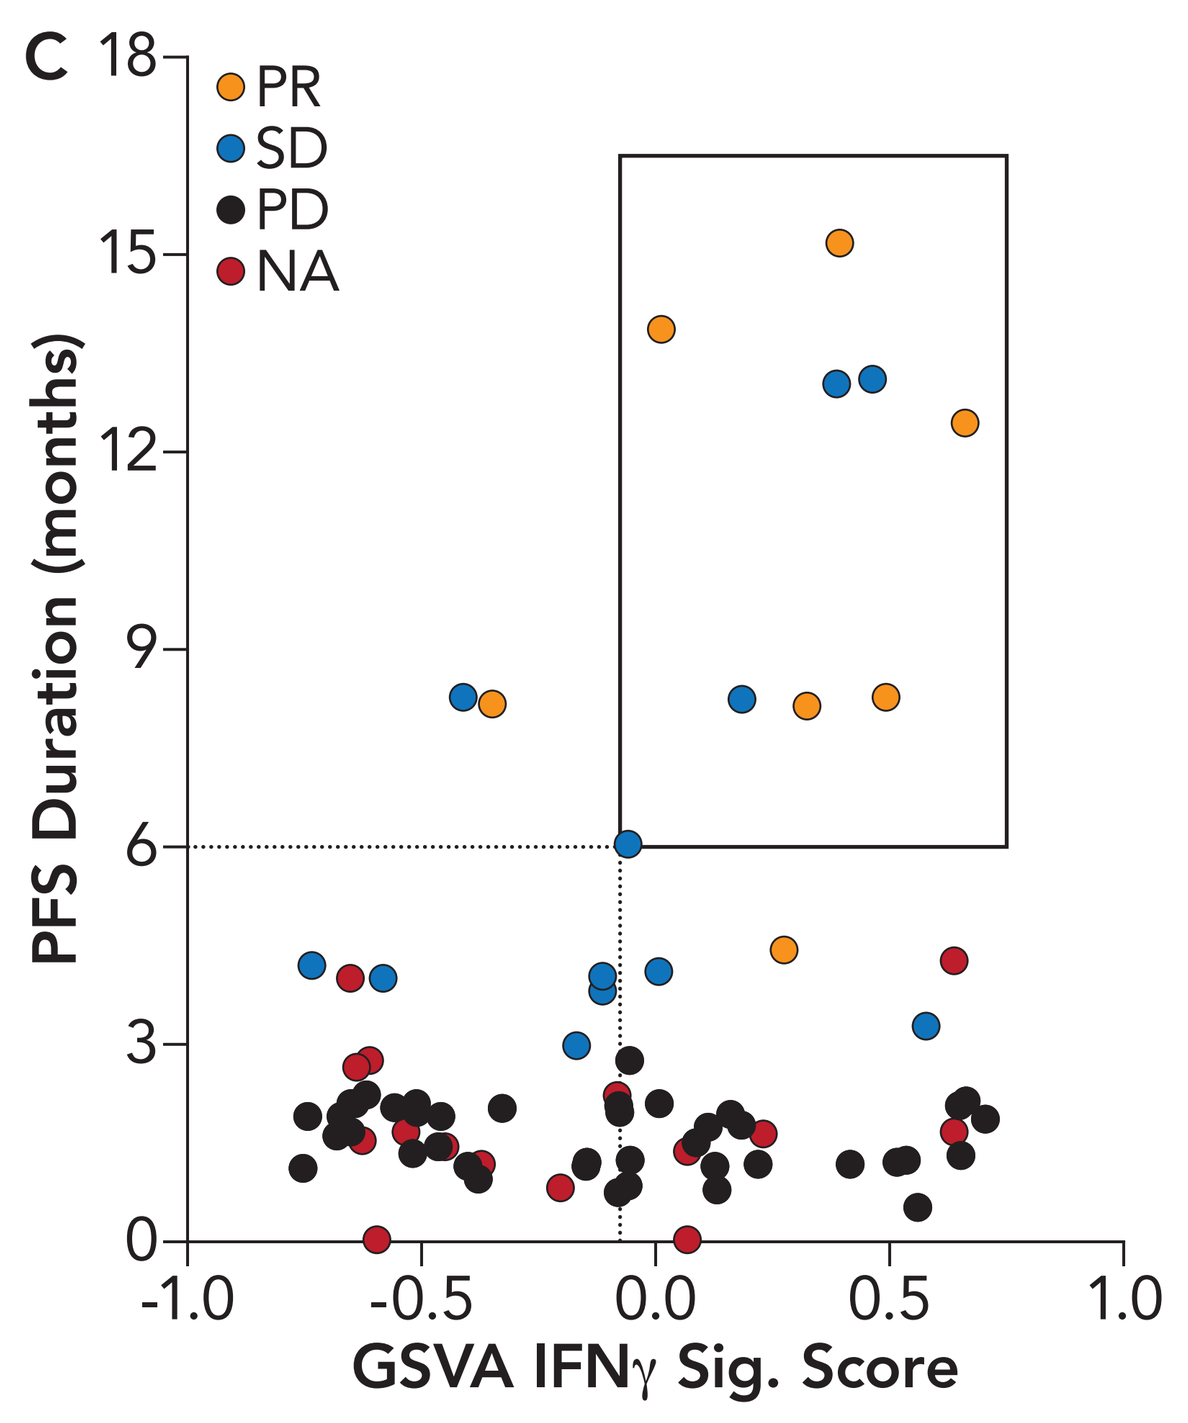
C
PR
SD
PD
NA
PFS Duration (months)
18
15
12
9
6
3
0
-1.0
-0.5
0.0
0.5
1.0
GSVA IFNγ Sig. Score

Abbreviations: BEP, bleomycin, etoposide, and cis-platinum; DCR, disease control rate; NA, not available, NR, nonresponders; ORR, objective response rate; PD, progressive disease; PFS, progression-free survival; PR, partial response; R, responders; SD, stable disease.

- A longer PFS trend was linked to patients with a high IFNγ signature (Figure 2A)
- Significant improvement of OS was observed in patients with IFNγ-high versus IFNγ-low signatures (median OS: 9.13 vs 3.82, *P*=0.003) (Figure 2B)

Figure 2: Kaplan-Meier Estimates for PFS (A) and OS (B) by IFNγ Signature Status (High vs Low)

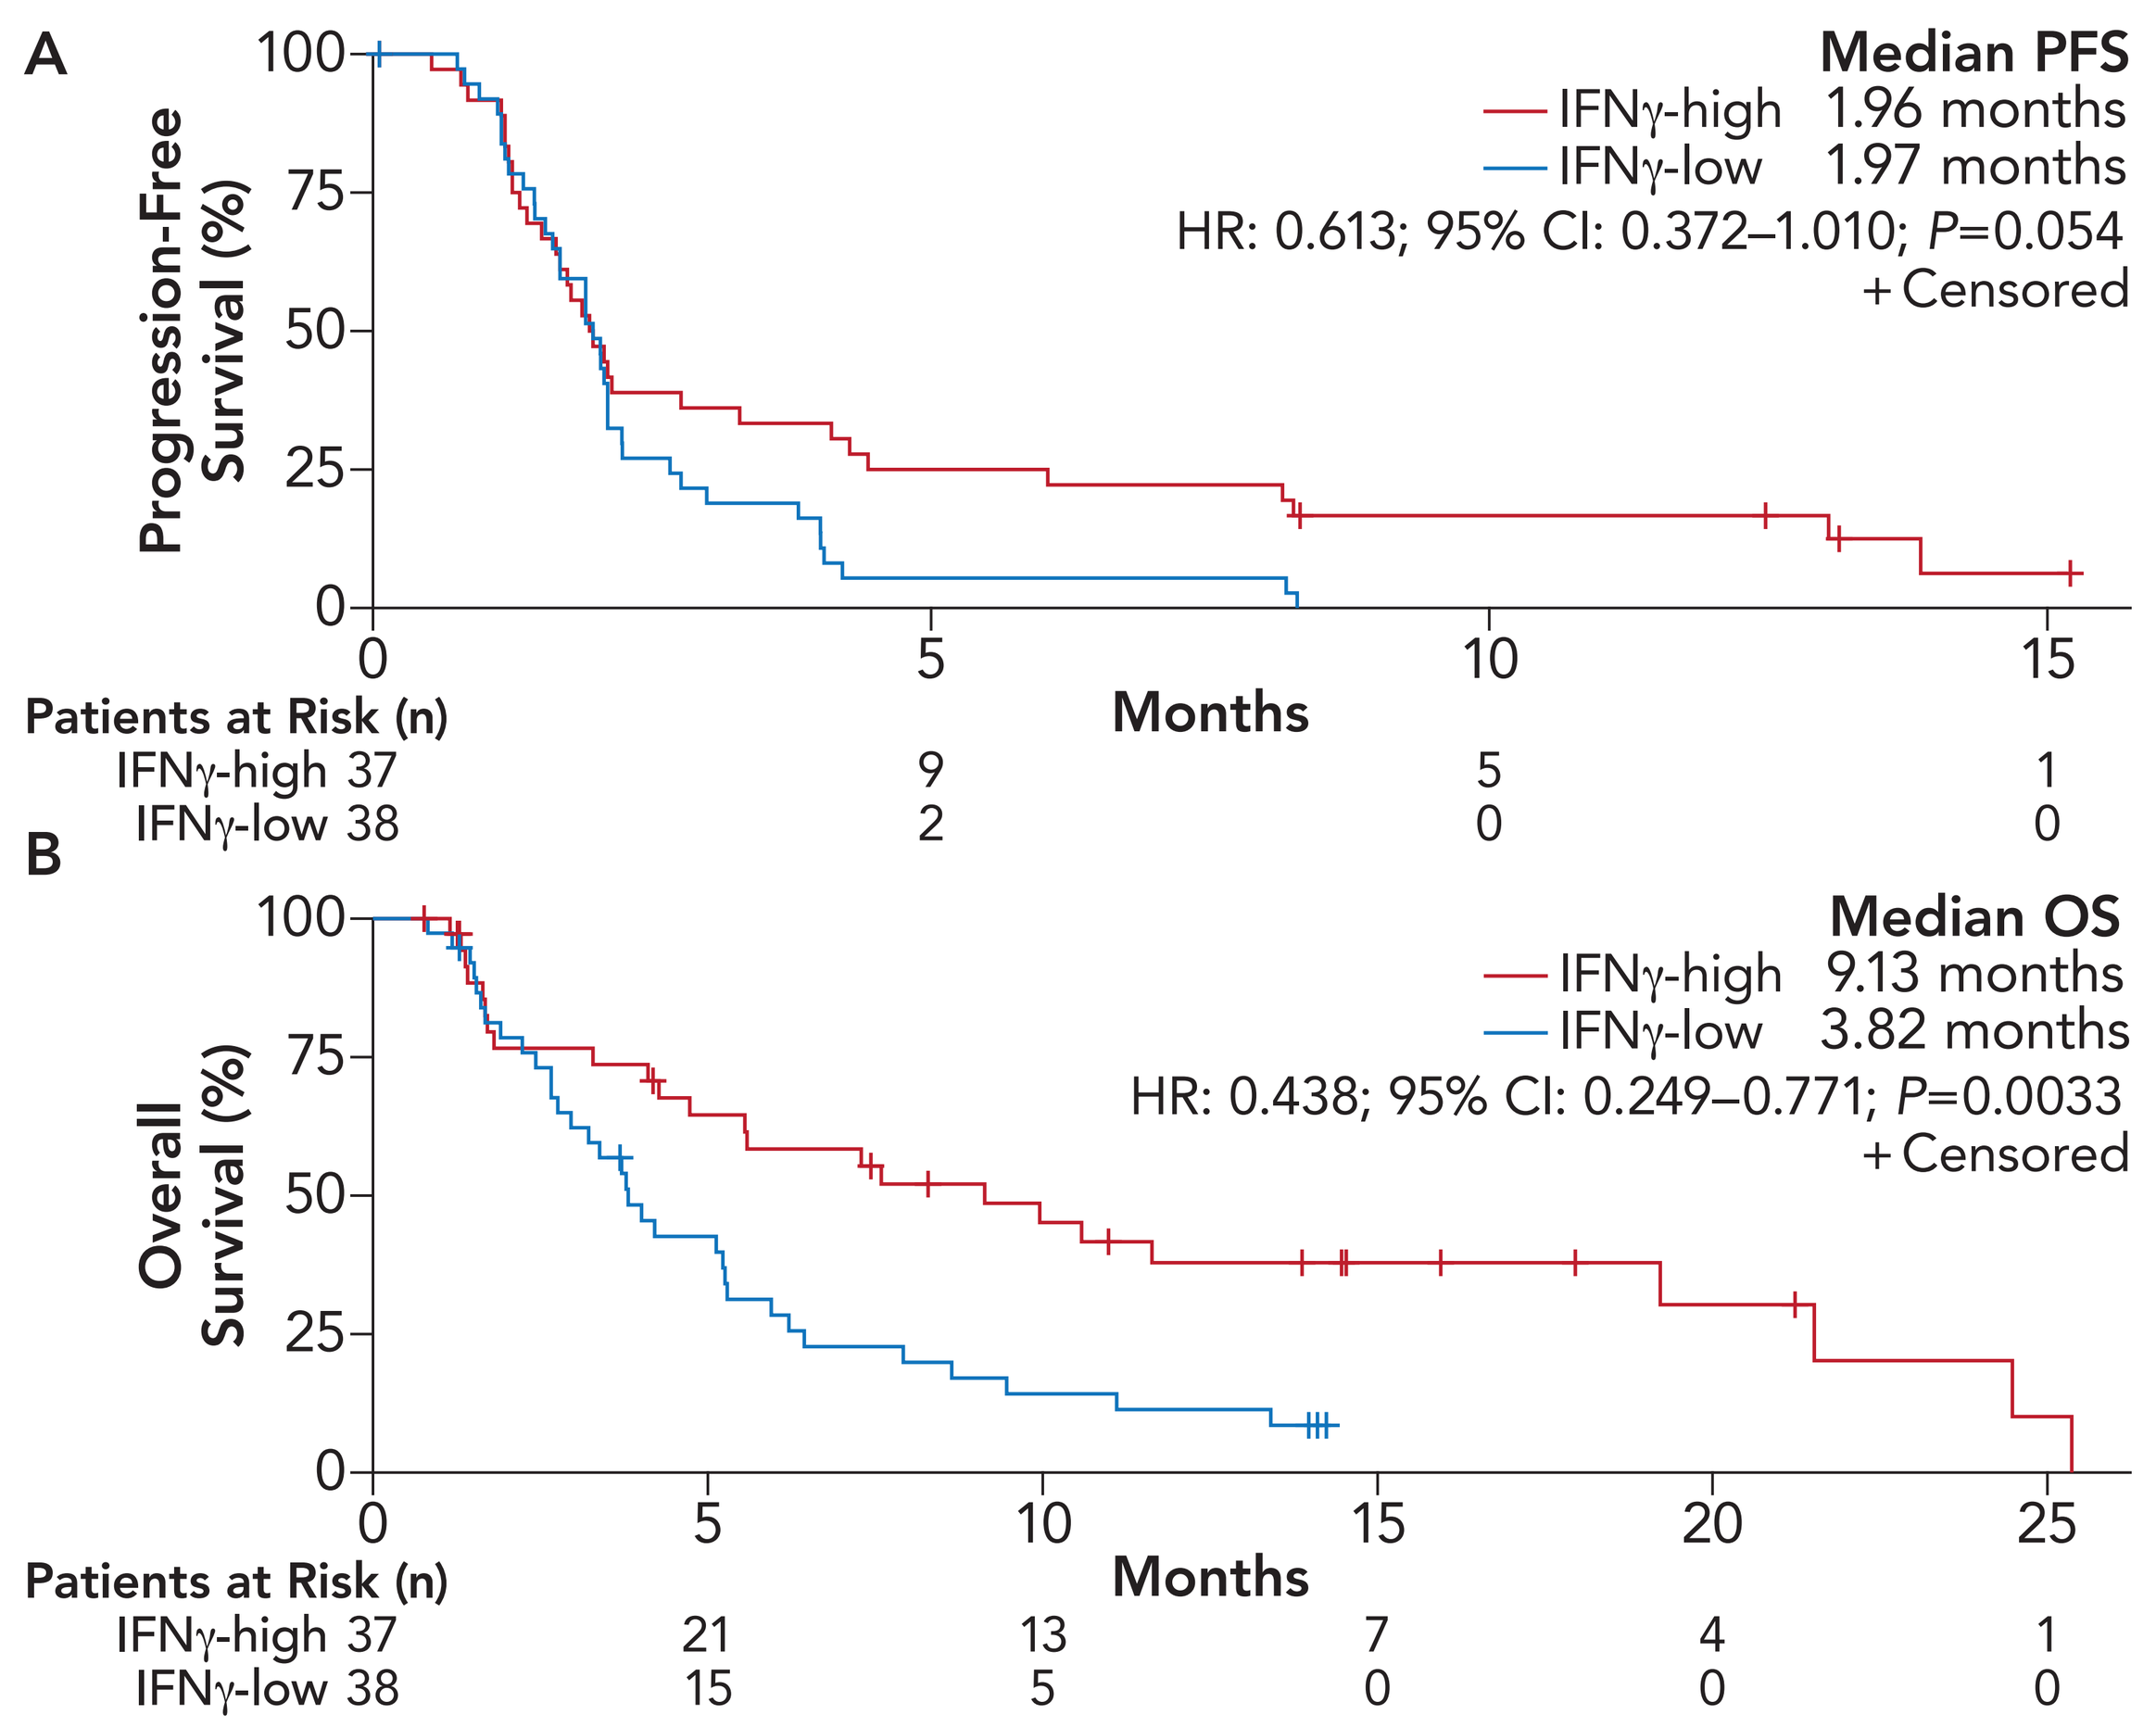
A
Progression-Free Survival (%)
Median PFS
IFNγ-high 1.96 months
IFNγ-low 1.97 months
HR: 0.613; 95% CI: 0.372–1.010; P=0.054
+ Censored
Months
Patients at Risk (n)
IFNγ-high 37 9 5 1
IFNγ-low 38 2 0 0
B
Overall Survival (%)
Median OS
IFNγ-high 9.13 months
IFNγ-low 3.82 months
HR: 0.438; 95% CI: 0.249–0.771; P=0.0033
+ Censored
Months
Patients at Risk (n)
IFNγ-high 37 21 13 7 4 1
IFNγ-low 38 15 5 0 0 0

Abbreviations: CI, confidence interval; HR, hazard ratio; OS, overall survival; PFS, progression-free survival.

- There was no significant association between an IFNγ signature and the clinicopathologic characteristics listed in Table 2

Table 2: Association of IFNγ Signature With Clinicopathologic Characteristics

Characteristics		Sample Size, N	IFNγ-high, n (%)	IFNγ-low, n (%)	*P*-value*
All patients		75	37 (49.3)	38 (50.7)	–
Age	<65	51	22 (43.1)	29 (56.9)	0.188
	≥65	24	15 (62.5)	9 (37.5)	
Sex	Male	51	26 (51)	25 (49)	0.866
	Female	24	11 (45.8)	13 (54.2)	
Histologic grade at baseline	Poorly differentiated	32	19 (59.4)	13 (40.6)	0.191
	Moderately differentiated	31	15 (48.4)	16 (51.6)	
	Well differentiated	2	1 (50)	1 (50)	
	Unknown	10	2 (20)	8 (80)	
Tumor site	Gastric/GEJ adenocarcinoma	53	25 (47.2)	28 (52.8)	0.743
	EAC	22	12 (54.5)	10 (45.5)	
Histological type	Adenocarcinoma	65	32 (49.2)	33 (50.8)	0.117
	Signet ring cell carcinoma	3	0 (0)	3 (100)	
	Unknown	7	5 (71.4)	2 (28.6)	
ECOG peformance score	0	26	14 (53.8)	12 (46.2)	0.744
	≥1	49	23 (46.9)	26 (53.1)	
Race	Asian	34	18 (52.9)	16 (47.1)	0.736
	Non-Asian	41	19 (46.3)	22 (53.7)	
Tumor stage	III	4	1 (25)	3 (75)	0.627
	IV	71	36 (50.7)	35 (49.3)	
Number of lines of prior systemic therapy	0	9	5 (55.6)	4 (44.4)	0.756
	1	28	13 (46.4)	15 (53.6)	
	2	19	11 (57.9)	8 (42.1)	
	≥3	19	8 (42.1)	11 (57.9)	

*Chi-square test.
Abbreviations: EAC, esophageal adenocarcinoma; ECOG PS, Eastern Cooperative Oncology Group; GEJ, gastroesophageal junction.

- Monotherapy NRs could be clustered into four distinct GEP subgroups according to immune and gene signatures listed in Table 3

Table 3: Immune and Tumor Gene Signatures Utilized for NR Subgroup Clustering

Tumor Immunogenicity	Tumor Sensitivity to Immune Attack	Antitumor Immune Activity	Immune Cell Population Abundance		Feature of Tumor
Antigen presentation	DNA damage repair	INFγ	CD8	MDSC	EMT
Cancer antigen	Tumor proliferation	Cytotoxicity	Exhausted CD8	T-reg	Cell adhesion
	Apoptosis	Inflammatory	Macrophage	NK cell	Angiogenesis
			Neutrophil	CD45	Hypoxia
			Mast cell	B cell	NF-kB
			Dendritic cell		

Abbreviations: EMT, epithelial-mesenchymal transition; MDSC, myeloid derived suppressor cell.

- The OS and differentiated expressed gene signatures among distinct NR subgroups and R are shown in Figure 3A (Log-rank *P*<0.0001) and Figure 3B, respectively
 - **NR-A** and **NR-B** had significantly lower IFNγ signatures[6] (*P*=0.013 and *P*=0.0007, respectively) than the responder group; compared with **NR-A** (mOS=9.46 months), **NR-B** had significantly decreased CD45+[7] and NK cell[7] (*P*=0.001 and *P*=0.0001) signatures, and the lowest median OS (3.81 months)
 - **NR-C** had a decreased NK cell signature (*P*=0.030) and trend toward an elevated cell cycle signature[8] (*P*=0.094), as well as relatively low median OS (5.55 months), despite having a comparable IFNγ signature level with responders
 - **NR-D** had the highest angiogenesis[9] (*P*=0.022) and macrophage[7] (*P*=0.035) signatures, with a median OS of 9.13 months

Figure 3: Tumor-immune Profiles of Responder and Nonresponder Subgroups of Tislelizumab Monotherapy

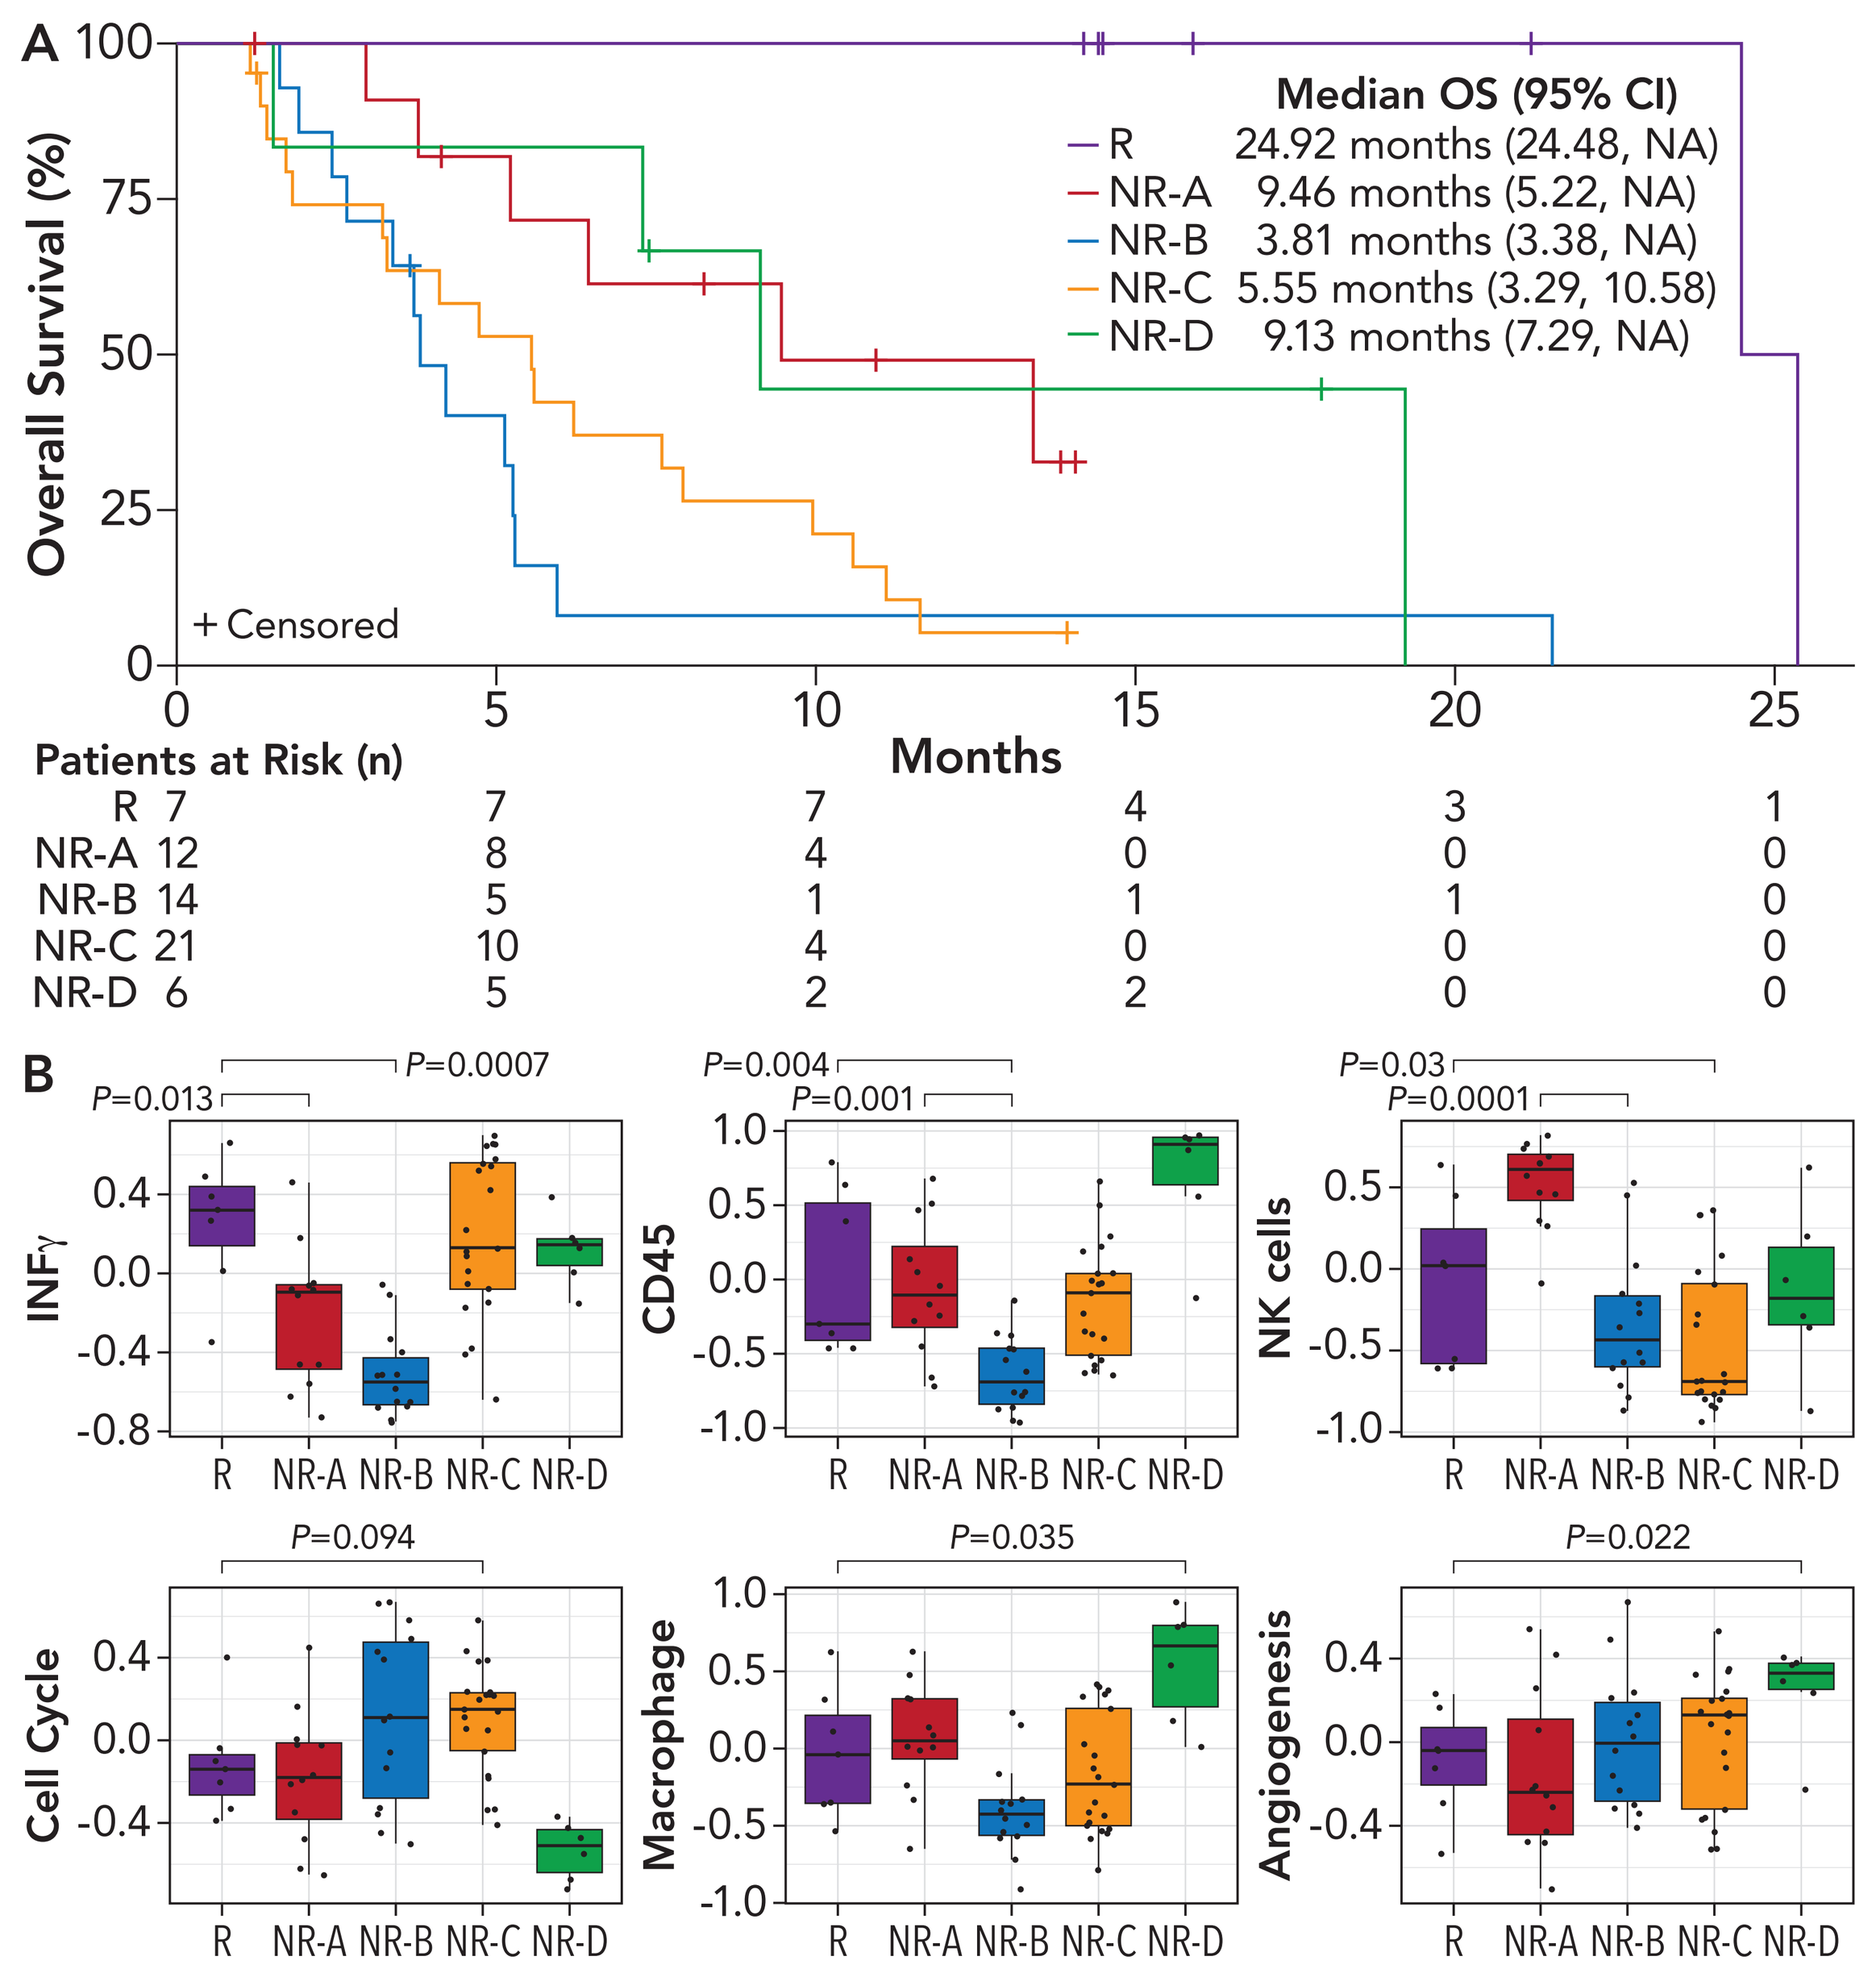
A
Overall Survival (%)
Median OS (95% CI)
R 24.92 months (24.48, NA)
NR-A 9.46 months (5.22, NA)
NR-B 3.81 months (3.38, NA)
NR-C 5.55 months (3.29, 10.58)
NR-D 9.13 months (7.29, NA)
+ Censored
Months
Patients at Risk (n)
R 7 7 7 4 3 1
NR-A 12 8 4 0 0 0
NR-B 14 5 1 1 1 0
NR-C 21 10 4 0 0 0
NR-D 6 5 2 2 0 0
B
P=0.013
P=0.0007
INFγ
P=0.004
P=0.001
CD45
P=0.03
P=0.0001
NK cells
P=0.094
Cell Cycle
P=0.035
Macrophage
P=0.022
Angiogenesis
R NR-A NR-B NR-C NR-D

Abbreviations: CI, confidence interval; GEP, gene expression profiling; NA, not available; NR, nonresponders; OS, overall survival; R, responders.

CONCLUSIONS

- These findings increase the understanding of tumor-immune profiles in GEA and their association with clinical efficacy of anti-PD1 monotherapy by identifying a 6-gene IFNγ signature as a potential biomarker of response, and multiple gene signatures that may indicate resistance:
 - Higher IFNγ signatures were associated with favorable clinical benefit in GEA patients receiving tislelizumab monotherapy
 - Compared with responders, elevated angiogenesis, macrophage, cell cycle, or decreased NK signatures were observed in distinct nonresponder subgroups of tislelizumab monotherapy, respectively
- The association between these tumor-immune profiles and clinical efficacy of tislelizumab plus chemotherapy varied from monotherapy
- Further validation will be considered in an ongoing phase 3 study designed to compare tislelizumab plus platinum/fluoropyrimidine versus placebo plus platinum/fluoropyrimidine as first-line therapy for patients with locally advanced or metastatic Gastric/GEJ adenocarcinoma (NCT03777657)

- Unlike patients receiving monotherapy, responders to combination therapy showed high cell cycle gene expression signatures (Figure 4A)
- Non-responders had numerically higher angiogenesis signatures versus responders; single gene DEG analysis revealed *VEGFA* was highly expressed in nonresponders, suggesting angiogenesis may potentially be associated with resistance to tislelizumab/chemotherapy (Figure 4B)

Figure 4: Tumor-Immune Profiles of Responders and Nonresponders to Tislelizumab Combination Therapy

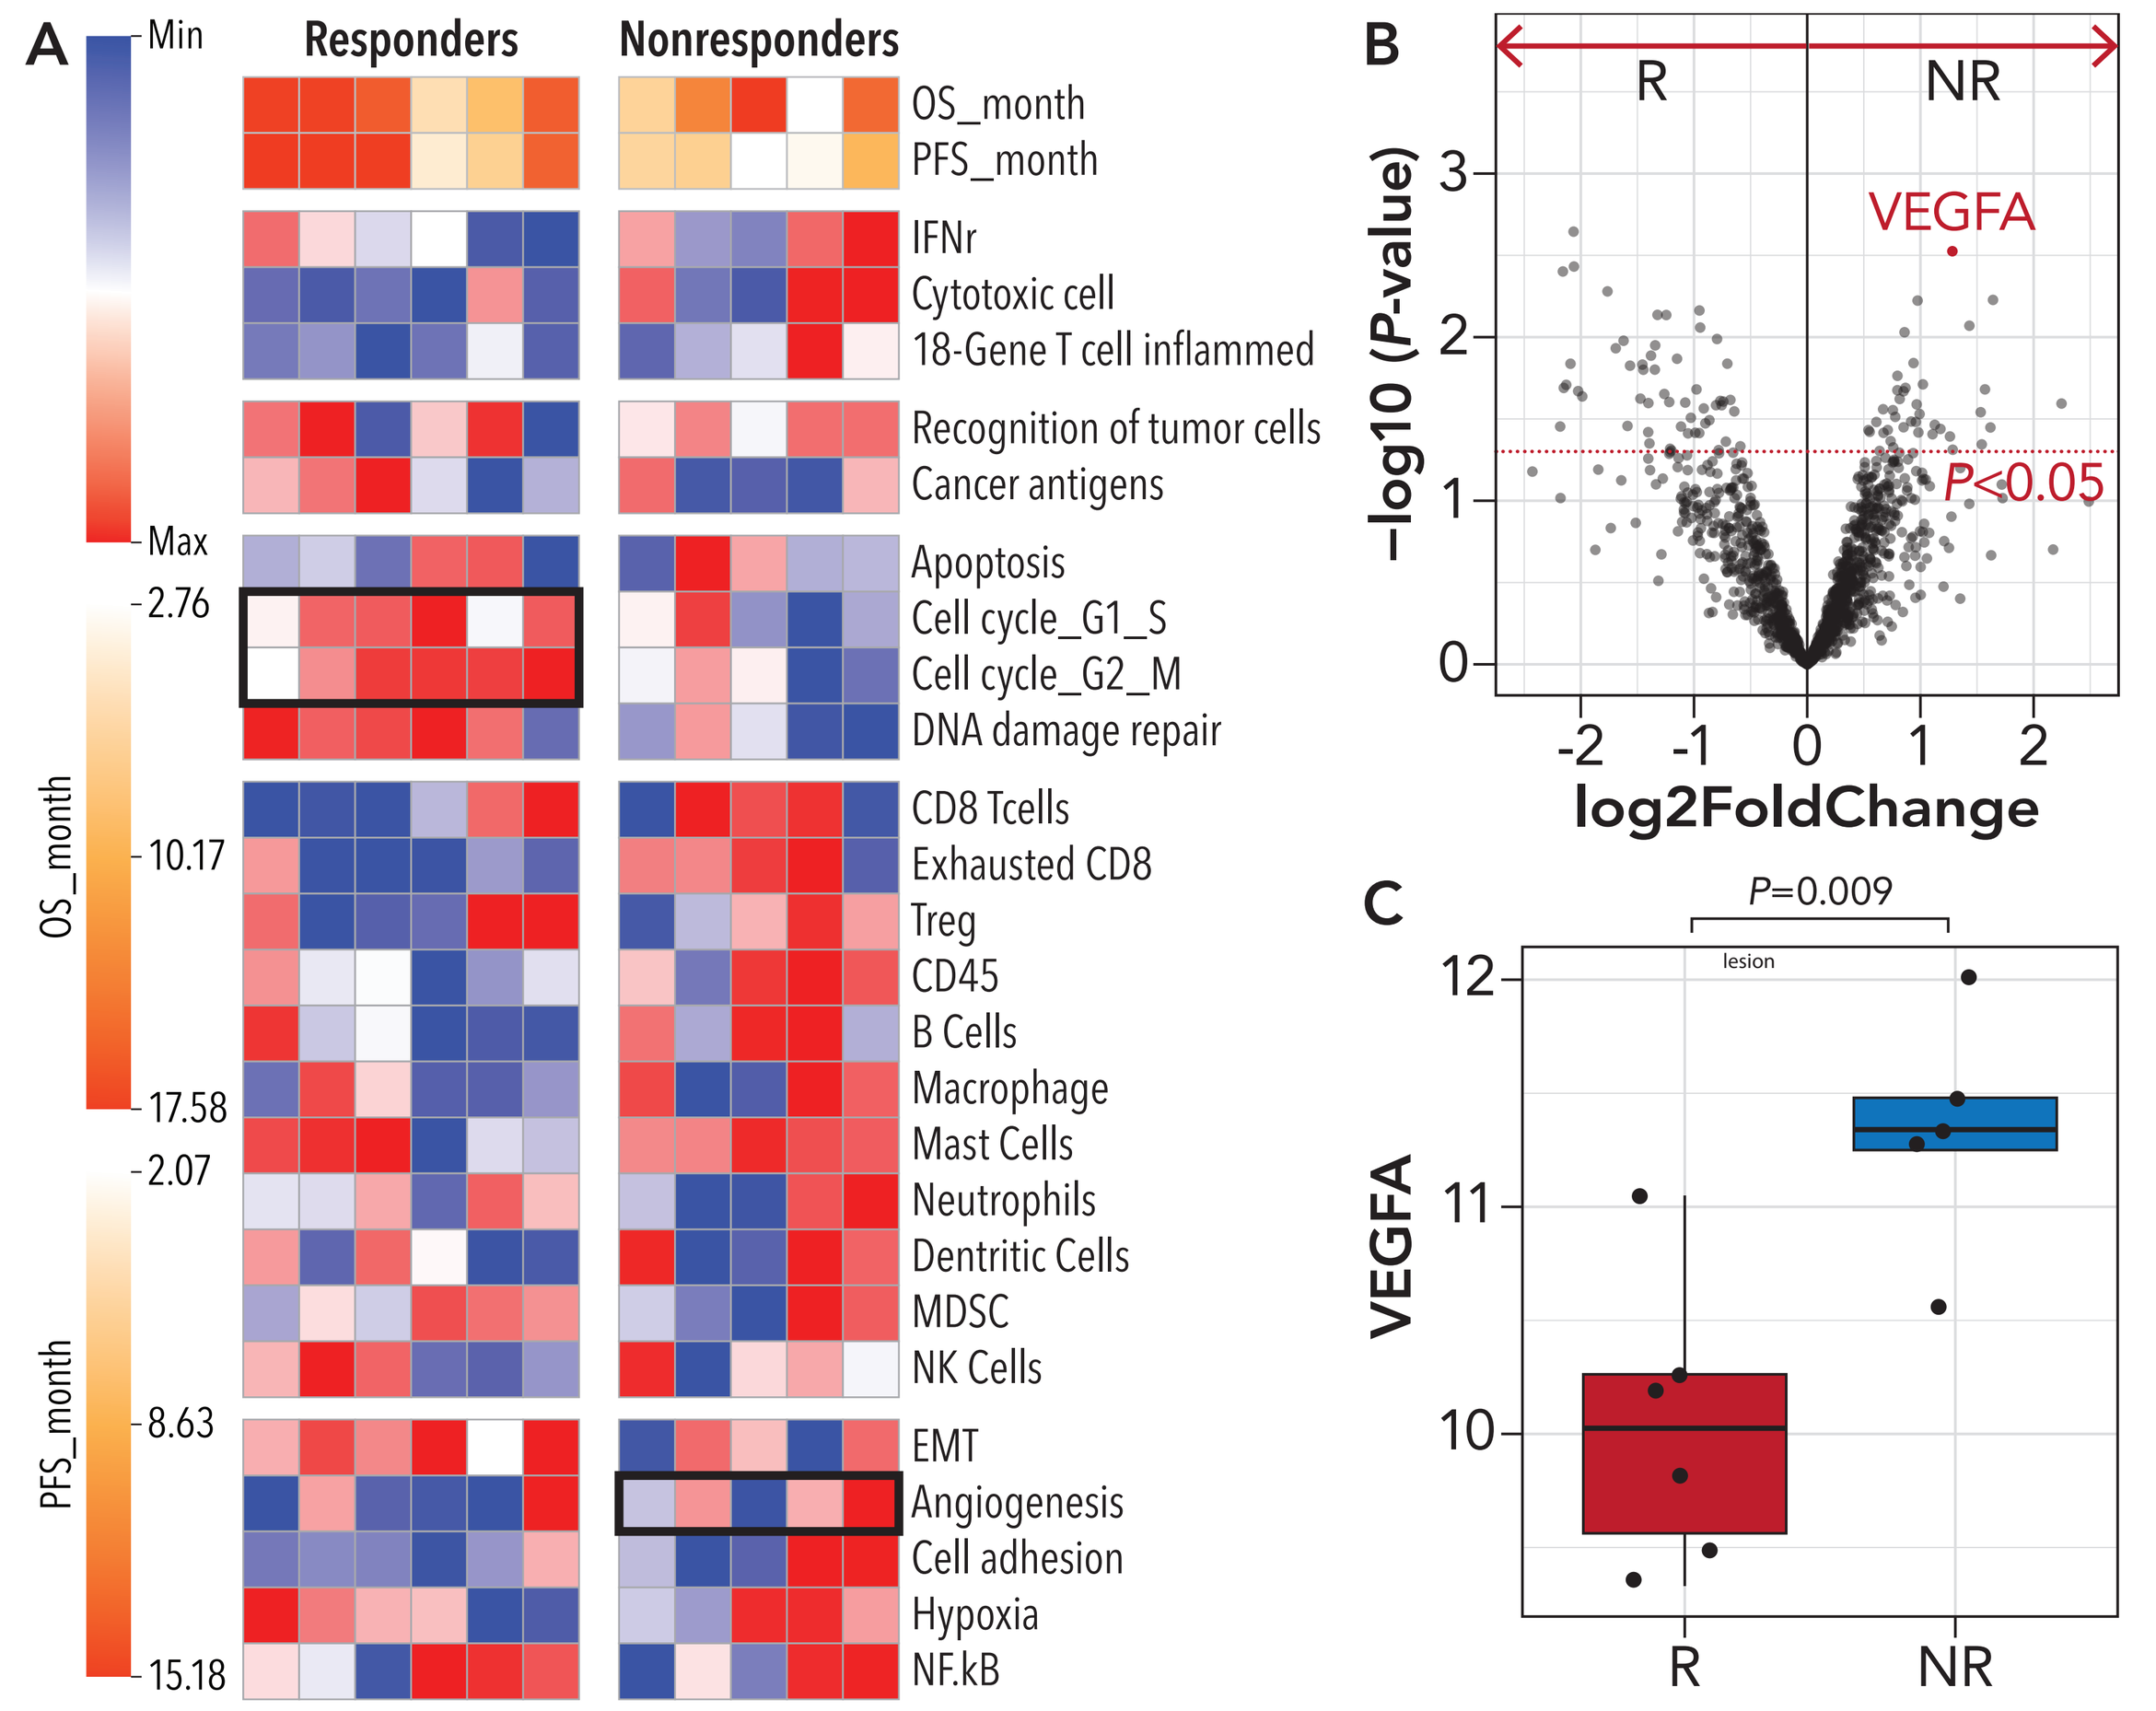
A
Min
Max
Responders
Nonresponders
OS_month
PFS_month
IFNr
Cytotoxic cell
18-Gene T cell inflammed
Recognition of tumor cells
Cancer antigens
Apoptosis
Cell cycle_G1_S
Cell cycle_G2_M
DNA damage repair
CD8 Tcells
Exhausted CD8
Treg
CD45
B Cells
Macrophage
Mast Cells
Neutrophils
Dentritic Cells
MDSC
NK Cells
EMT
Angiogenesis
Cell adhesion
Hypoxia
NF.kB
OS_month
2.76
10.17
17.58
PFS_month
2.07
8.63
15.18
B
R
NR
VEGFA
P<0.05
−log10 (P-value)
log2FoldChange
C
P=0.009
VEGFA
R
NR

Abbreviations: NR, nonresponders; OS, overall survival; PFS, progression-free survival; R, responders.

REFERENCES

1. Ajani, J., Lee, J., Sano, T. et al. *Nat Rev Dis Primers.* 2017; 3:1703.
2. Rubenstein JH, Shaheen NJ. *Gastroenterology.* 2015;149(2):302-317.e1.
3. Desai J, Millward M, Chao Y, et al. *Ann Oncol.* 2017;28(suppl 5): mdx367.021.
4. Shen L, Yuan Y, Bai Y, et al. Presented at: 22nd Annual Chinese Society for Clinical Oncology Congress; September 18-22, 2019; Xiamen, China
5. Xu R, Arkenau T, Bang Y, et al. *J Clin Oncol.* 2019;37(suppl):Abstract TPS2655.
6. Seiwert T, Burtness B, Mehra R, et al. *Lancet Oncol.* 2016;17(7):956-965.
7. Danaher P, Warren S, Rongze L, et al. *J ImmunoTher Cancer.* 2018;6:63.
8. Tirosh I, Izar B, Prakadan SM, et al. Science. 2016;352(6282):189-196.
9. Patwardhan P, Ivy K, Musi E, et al. *Oncotarget.* 2016;7:4093-4109.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative centers' study staff and study patients, and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. This study was sponsored by BeiGene, Ltd. Writing and editorial assistance was provided by Stephan Lindsey, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL), and funded by the study sponsor.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com