Abstract #664 BGB-10188, a Highly Selective PI3Kδ Inhibitor with Improved Safety Profile and Superior Anti-Tumor Activities in vivo

Xiao Yang, Xiaolong Yang, Xinxin Cui, Dan Su, Yue Wu, Xuebing Sun, Jingyuan Wang, Huichen Bai, Wei Wei, Jing Li, Xi Yuan, Ye Liu, Fan Wang, Zhiwei Wang, Lai Wang, Xuesong Liu, Xiaomin Song*

*Authors' Affiliations: BeiGene Global Research, Beijing 102206, P.R. China; *Correspondence: xiaomin.song@beigene.com*

BeiGene

Abstract

Phosphoinositide 3-kinases (PI3Ks) are a family of enzymes capable of phosphorylating phosphatidylinositol to phosphoinositides, which are important secondary messengers involved in various cell signaling and functions. PI3Kδ is one of four isoforms (PI3Kα, β, δ and γ) of the PI3K class I family. It is restrictively expressed in leukocytes. PI3Kδ is a key signal transduction component for normal and malignant B cells and also important for the homeostasis and function of T-regulatory cells (Treg), making it a promising target for treatment of both hematologic malignancies and solid tumors.

BGB-10188 is a highly selective inhibitor of PI3Kδ, showing no significant inhibition over 376 protein kinases and 17 lipid kinases, and more than three-thousand folds selectivity over PI3Kα, PI3Kβ, and PI3Kγ. BGB-10188 potently inhibited PI3Kδ in biochemical, cellular and human whole blood assays with IC50s ranging from 1.7-16 nM. It also showed a long-lasting and strong target inhibition activity in mouse pharmacodynamics (PD) studies at doses as low as 10mg/kg. The elimination half-life (t1/2) of BGB-10188 in plasma was 12.6 hours and 10.4 hours in rats and dogs, respectively.

BGB-10188 showed significant antitumor effects in both B cell Lymphoma xenograft and solid tumor models as single agent or in combination with PD-1 antibodies. The liver toxicities of BGB-10188 were evaluated in mice and significantly improved safety profile was observed for BGB-10188 in comparison with other PI3Kδ inhibitors.

In summary, BGB-10188 is a novel PI3Kδ inhibitor with high selectivity, potency and improved safety profile shown in preclinical studies, which is promising and warrants the testing of the compound in human.

Biochemical and Cellular Potency

Table 1: Selectivity and potency of BGB-10188

Assay	BGB-10188	Idelalisib
Enzymatic assay IC50 (nM)		
PI3K δ	1.7	2.3
Selectivity (Folds)		
PI3K α	4900	430
PI3K β	5100	360
PI3K γ	3800	52
Cellular assay IC50 (nM)		
Raji (Anti-IgM)	16	12
Human whole blood assay IC50 (nM)		
Farage	9.2	189

Pharmacokinetics Profile

Figure 1: PK profiles of BGB-10188 in dog

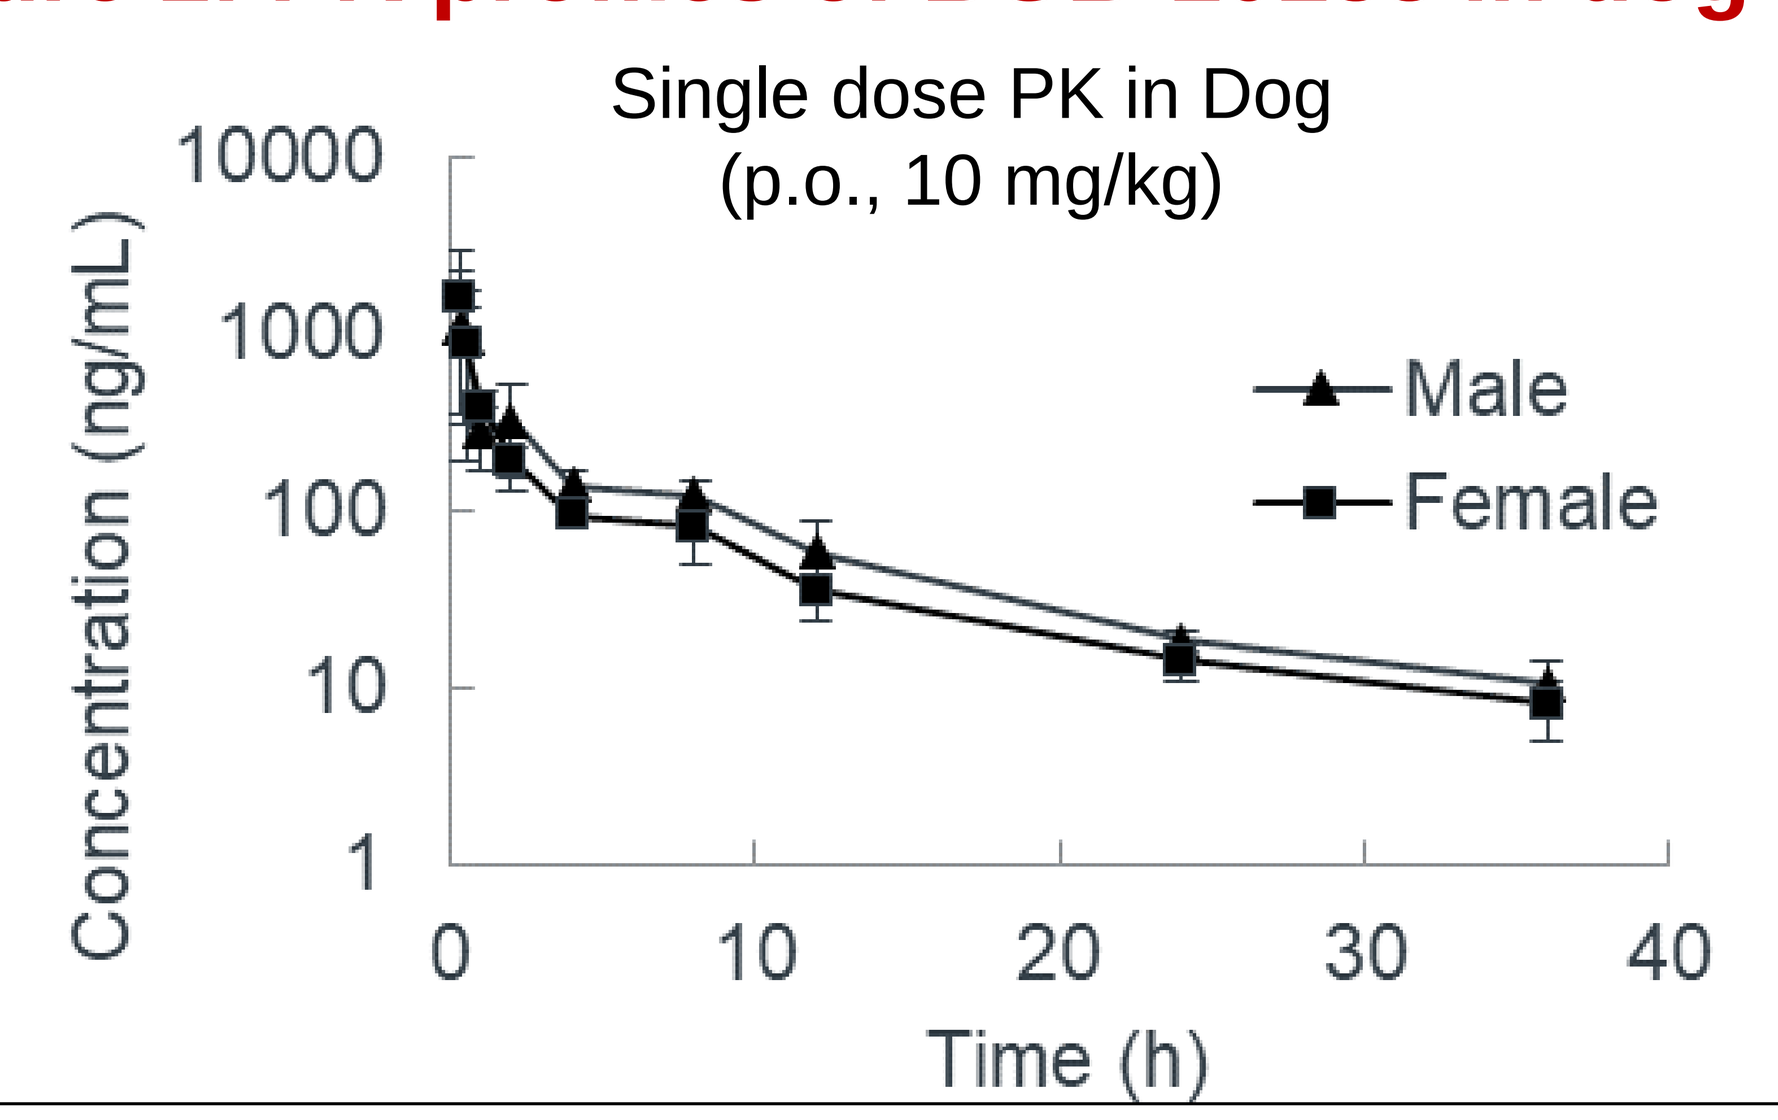
Single dose PK in Dog
(p.o., 10 mg/kg)
Concentration (ng/mL)
Time (h)
Male
Female

Parameters			
CL (ml/min/kg)	38.9	$T_{1/2}$ (h)	10.4
Vd (l/kg)	25.5	F%	54.3

PD Activity in Mouse

Figure 2: PD activity in mouse peripheral blood B cells

BALB/c mice were treated with 10 and 30 mg/kg of BGB-10188 and euthanized at different time points after dosing as indicated. Whole blood was collected and stimulated by anti-mouse IgD antibody for activating B-cell receptors. The level of pAKT in B cells was measured by flow cytometry.

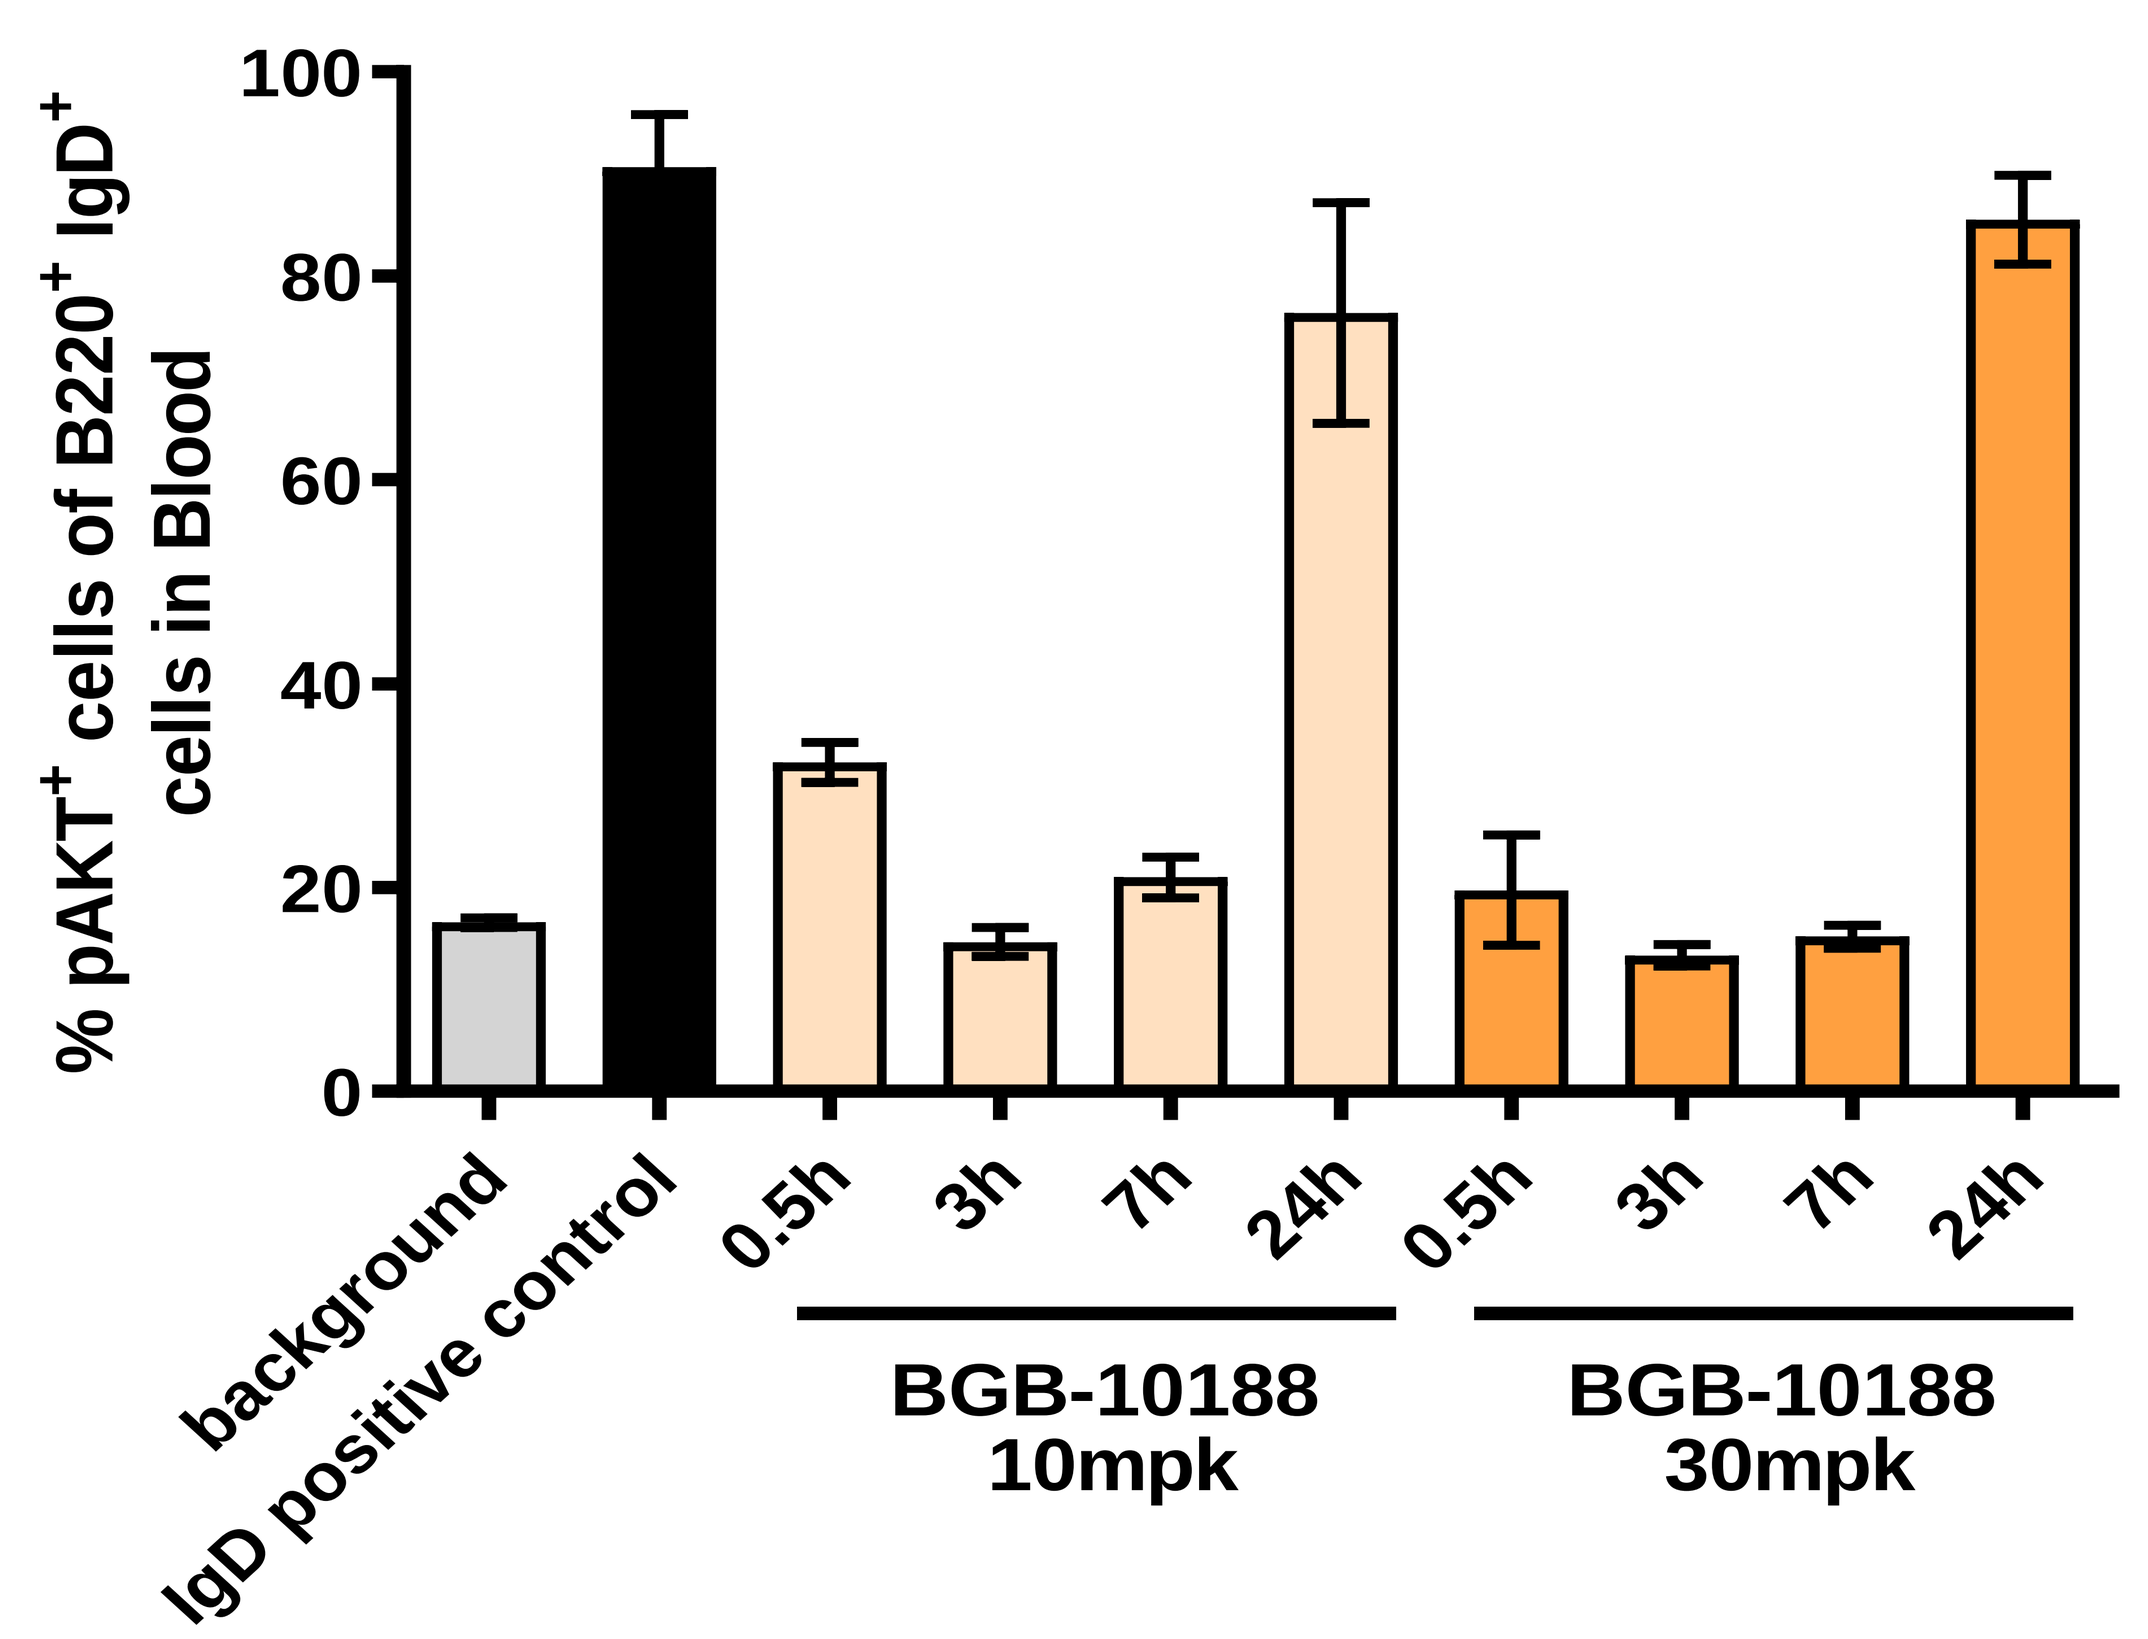
% pAKT+ cells of B220+ IgD+ cells in Blood
background
IgD positive control
0.5h
3h
7h
24h
BGB-10188 10mpk
BGB-10188 30mpk

✓ **BGB-10188 showed strong and sustained inhibition on pAKT in B cells in peripheral blood at doses as low as 10 mg/kg in mice.**

Efficacy in B cell Lymphoma Model

Figure 3. Efficacy of BGB-10188 in Farage subcutaneous model

Farage tumor cells (3×10⁶) alone (A) or together with hPBMC (6×10⁵) (B) were implanted subcutaneously in female NCG mice.

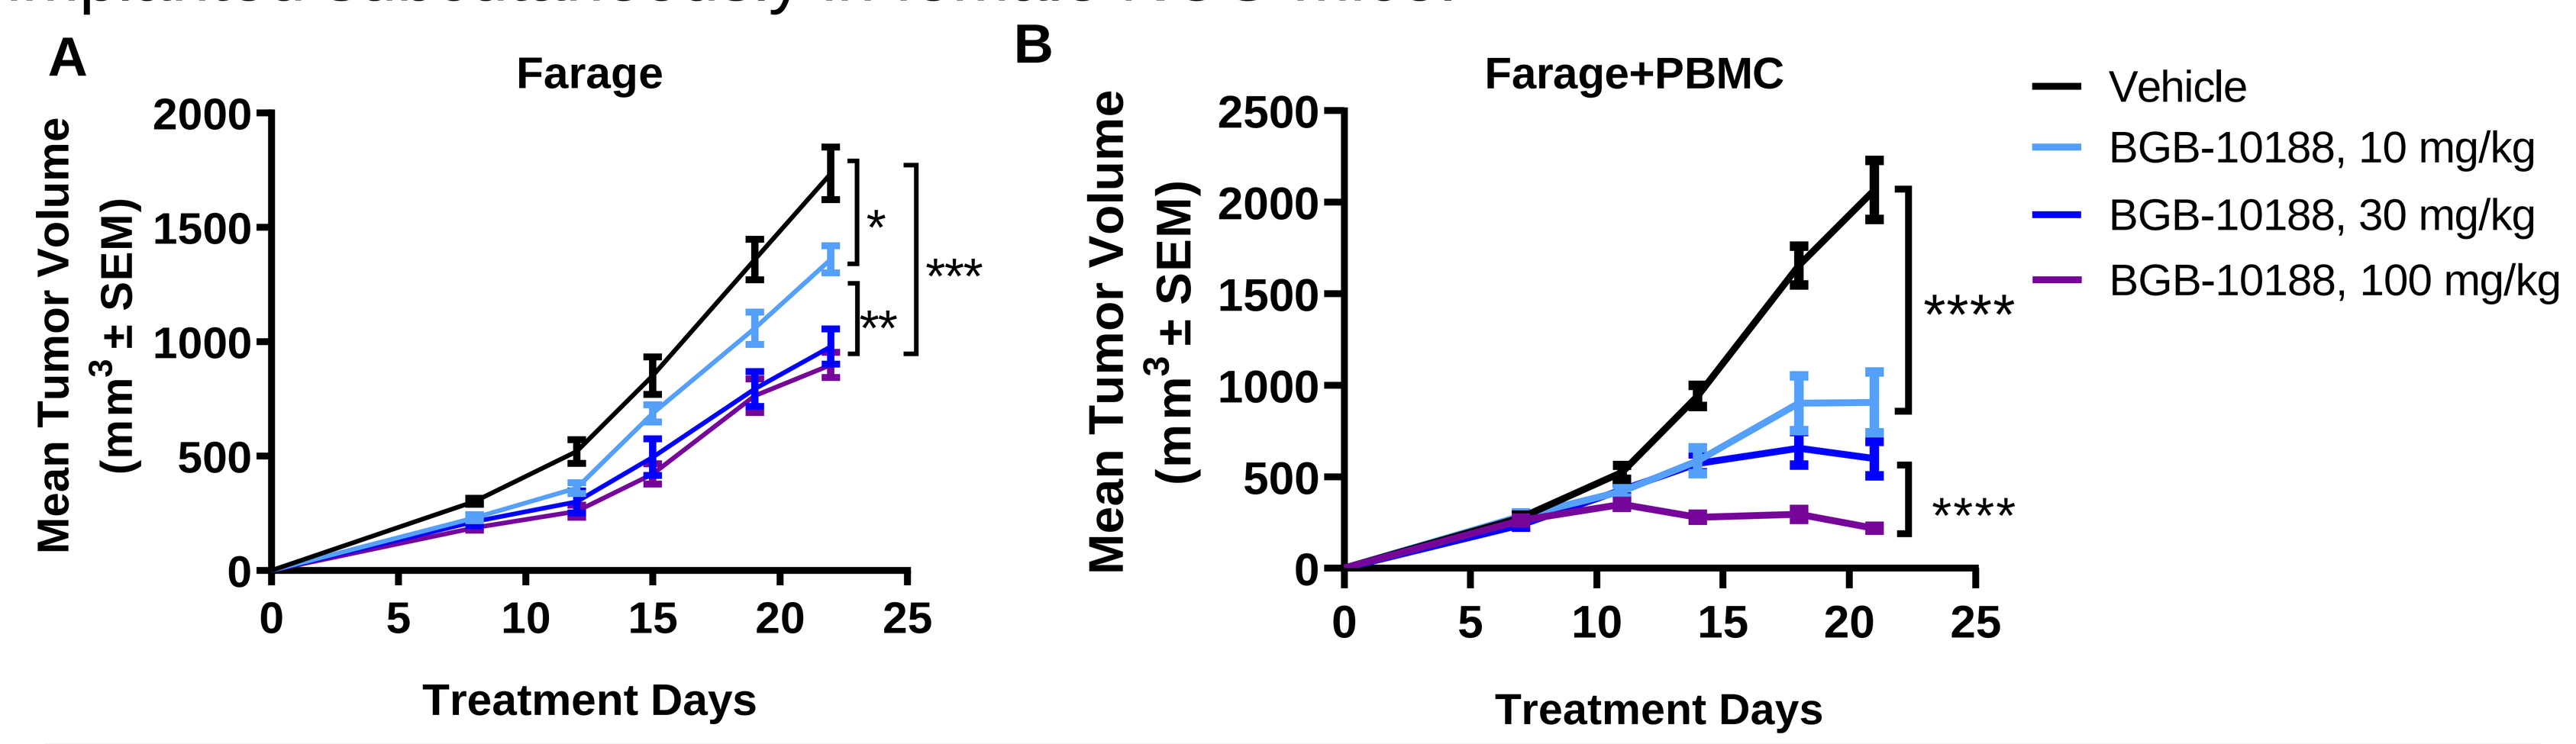
A
Farage
B
Farage+PBMC
Mean Tumor Volume (mm3 ± SEM)
Treatment Days
Vehicle
BGB-10188, 10 mg/kg
BGB-10188, 30 mg/kg
BGB-10188, 100 mg/kg

Test Article	Dose (mg/kg, BID)	N	Farage TGI (Day 22)	Farage Mean Tumor Volume on Day 22 (mm³)	Farage+PBMC TGI (Day 22)	Farage+PBMC Mean Tumor Volume on Day 22 (mm³)
Vehicle	-	10	-	1736.4	-	2066.7
BGB-10188	10	10	22%	1360.6	56%	905.6
	30	10	44%	979.2	71%	600.4
	100	10	48%	899.4	89%	217.8

✓ **BGB-10188 showed dose-dependent anti-tumor activity in Farage (DLBCL) xenograft model;**

✓ **In the humanized model with hPBMC transplantation, BGB-10188 showed much better anti-tumor activity than in the xenograft model, indicating that BGB-10188 has duel effect on both tumor and immune cells and could achieve more significant anti-tumor activity with the existence of immune system.**

Combination with PD-1 Ab

Figure 4. Efficacy of the combination of BGB-10188 and anti-mouse PD-1 antibody Ch15mt in CT26WT syngeneic subcutaneous model

CT26WT cells (3×10⁴) were implanted subcutaneously in BALB/c mice. mice were euthanized using carbon dioxide once their body weight loss was over 20% or their tumor volume reached ≥ 2000 mm³ or the tumor was ulcerated.

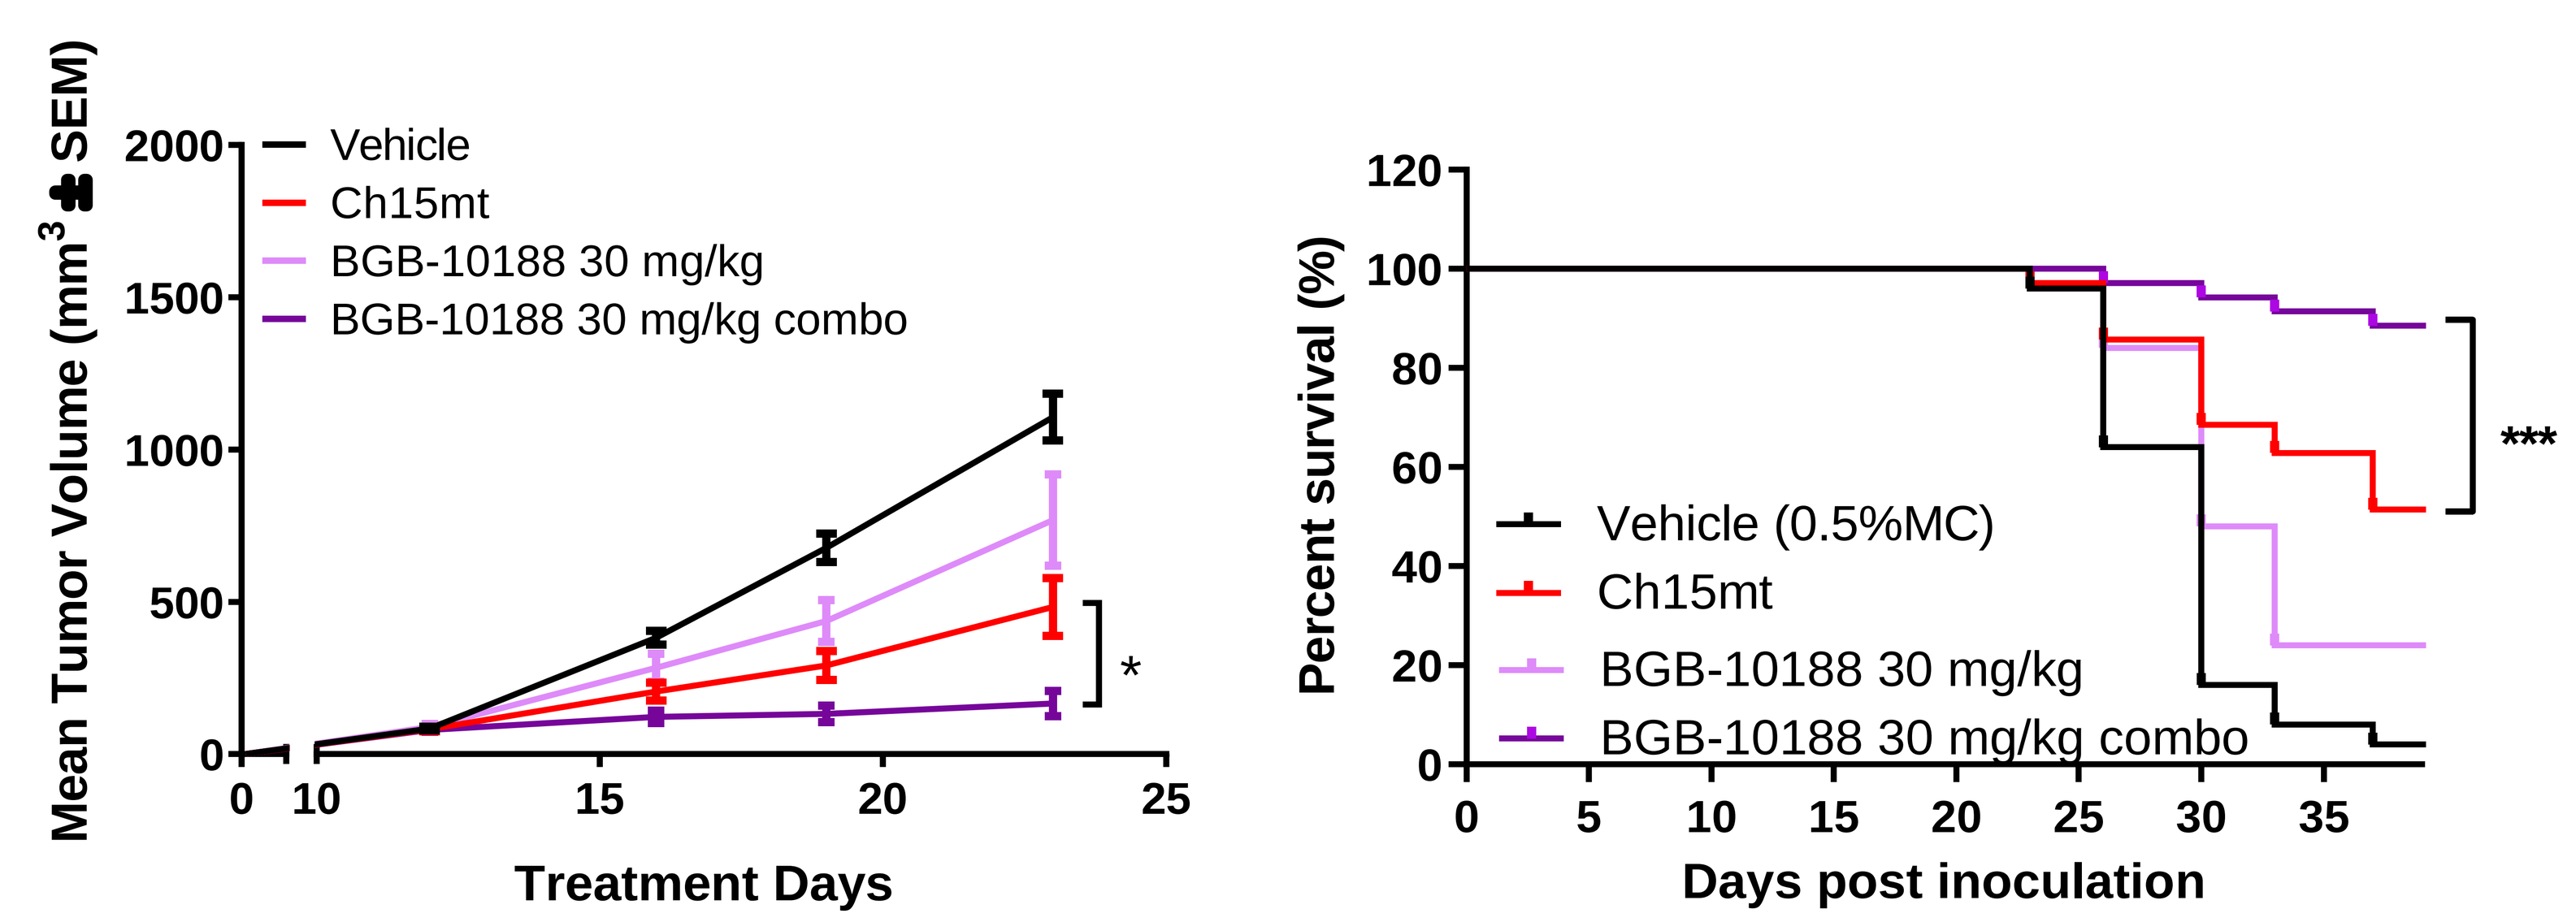
Mean Tumor Volume (mm3 ± SEM)
Treatment Days
Vehicle
Ch15mt
BGB-10188 30 mg/kg
BGB-10188 30 mg/kg combo
Percent survival (%)
Days post inoculation
Vehicle (0.5%MC)

Test Article	Dose (mg/kg, BID)	N	TGI (Day 23)	Mean Tumor Volume on Day 23 (mm³)	TF
Vehicle	-	25	-	-	0
Ch15mt	3	35	56%	483.4	13
BGB-10188	30	25	31%	768.4	1
Ch15mt+BGB-10188	3+30	35	85%	166.2	19

Note: TF: Tumor Free

✓ **Significantly improved antitumor activity and prolonged survival were observed in the combination treatment group.**

Immune Regulatory Activity in vivo

Figure 5. Inhibitory activity of BGB-10188 on Treg

BALB/c mice with CT26WT tumors were orally treated with BGB-10188 at different doses when tumor volume reached around 80 mm³. The mice were sacrificed 14 days after drug administration to collect spleen, blood and tumor for immune cell profiling by flow cytometry.

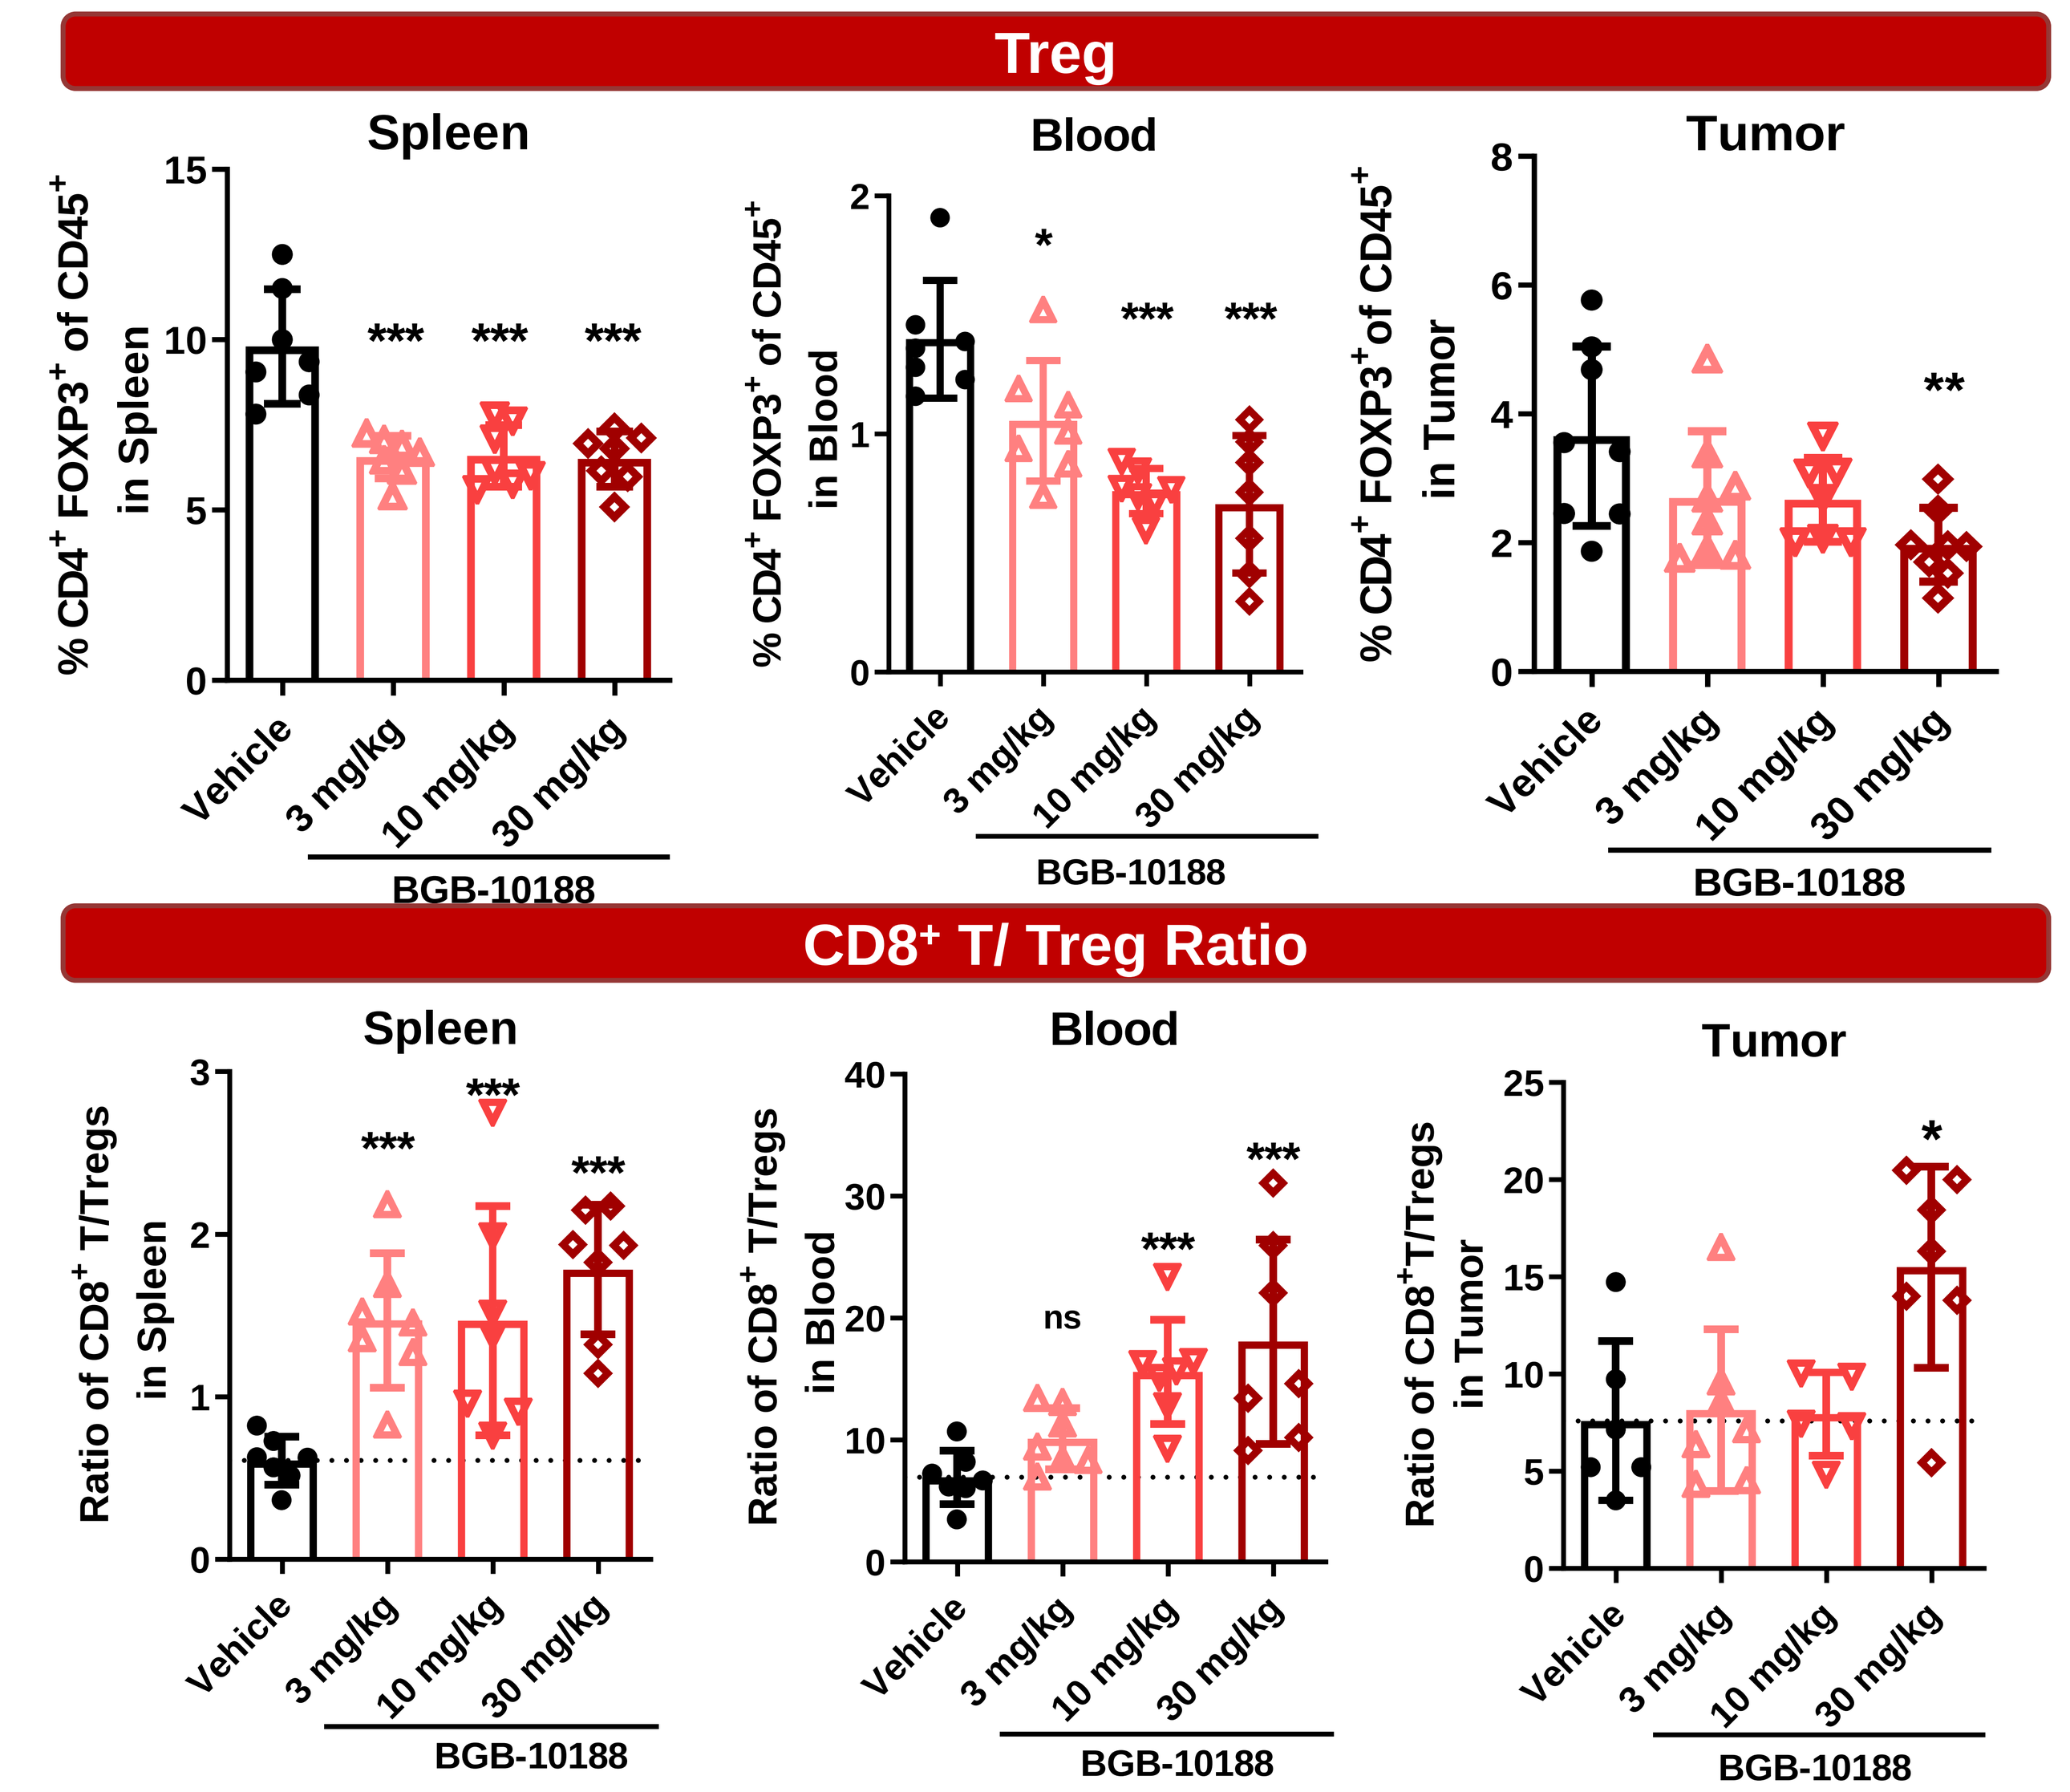
Treg
Spleen
Blood
Tumor
% CD4+ FOXP3+ of CD45+ in Spleen
% CD4+ FOXP3+ of CD45+ in Blood
% CD4+ FOXP3+ of CD45+ in Tumor
CD8+ T/ Treg Ratio
Ratio of CD8+ T/Tregs in Spleen
Ratio of CD8+ T/Tregs in Blood
Ratio of CD8+ T/Tregs in Tumor
Vehicle
3 mg/kg
10 mg/kg
30 mg/kg
BGB-10188

✓ **BGB-10188 at doses ≥ 3 mg/kg showed inhibitory activity on Treg in blood, spleen and tumor tissues.**

Liver Toxicity Evaluation

Figure 6: ALT and AST level in mice with the treatment of idelalisib and BGB-10188

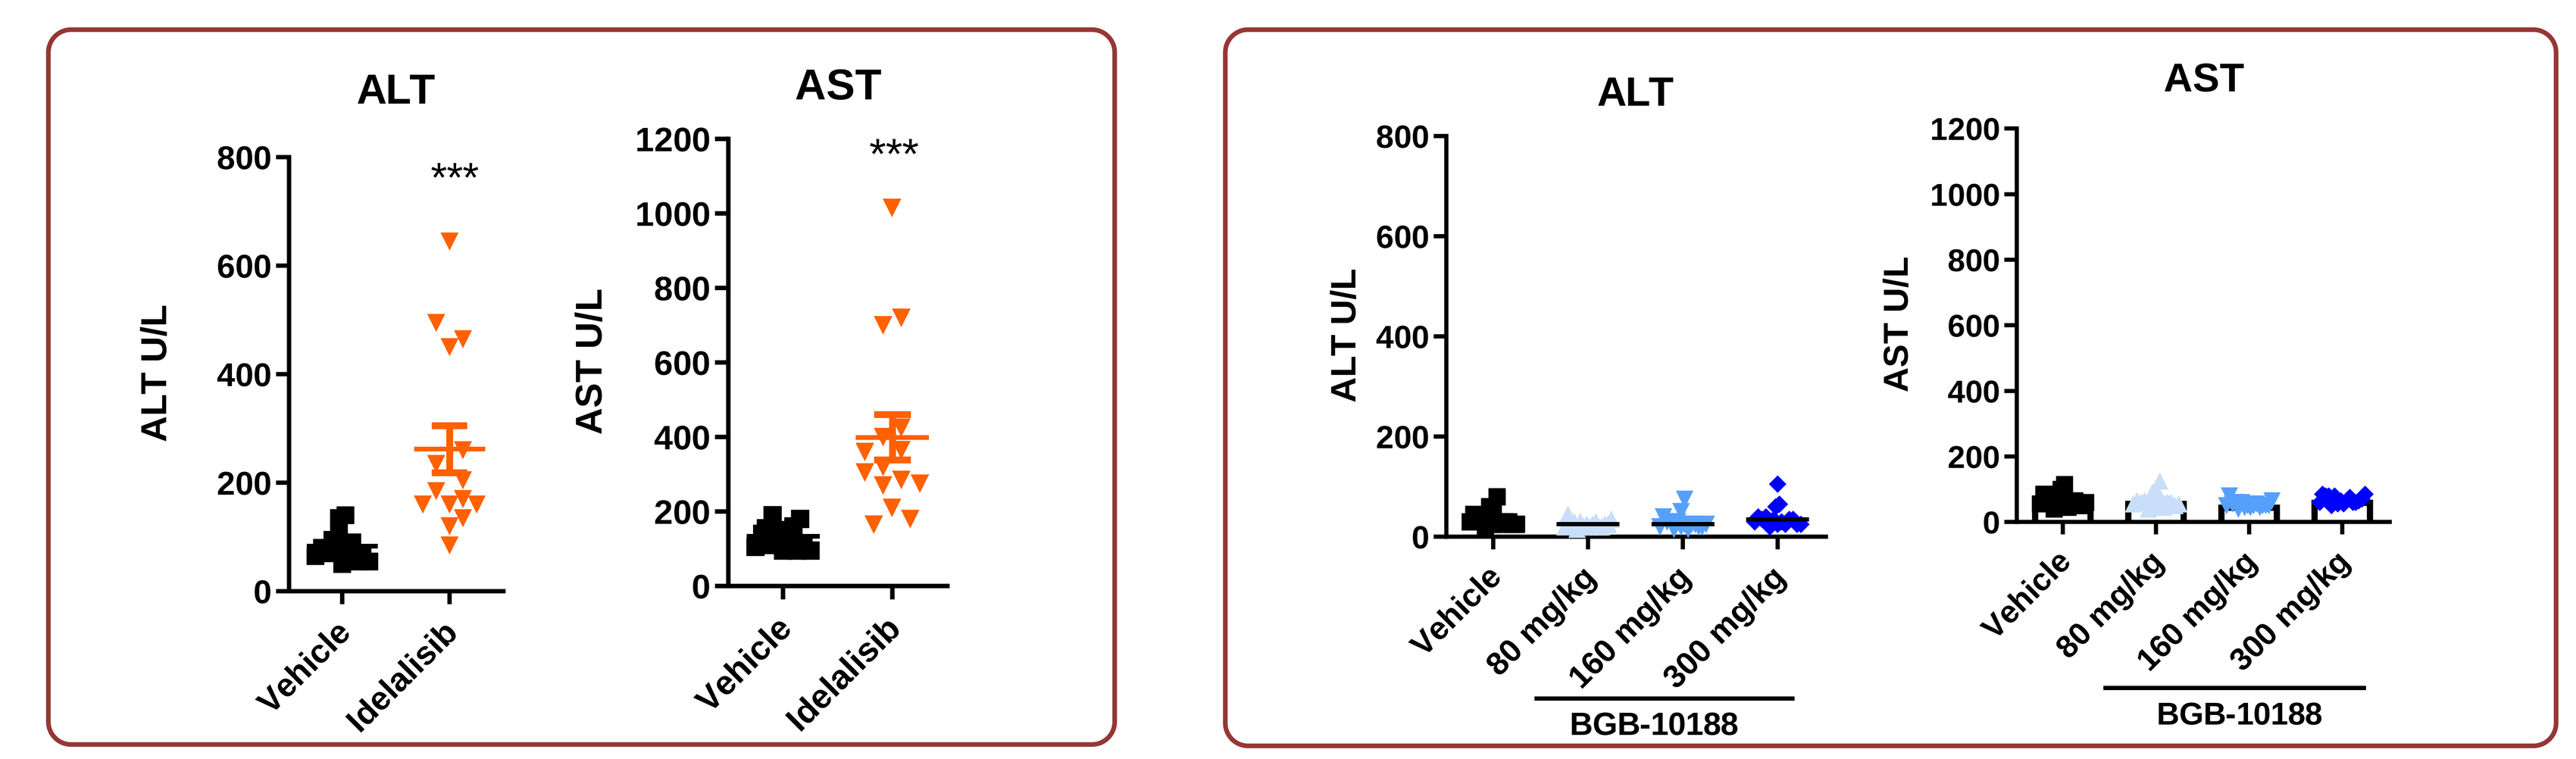
ALT
AST
ALT U/L
AST U/L
Vehicle
Idelalisib
80 mg/kg
160 mg/kg
300 mg/kg
BGB-10188

BALB/c mice were orally treated with 20 mg/kg of idelalisib or 80, 160 and 300 mg/kg of BGB-10188 twice a day for 20 days.

✓ **BGB-10188 didn't Induce transaminitis in mice at doses up to 300 mg/kg.**

Conclusion

- ✓ BGB-10188 is a potent and highly selective PI3Kδ inhibitor.
- ✓ BGB-10188 showed long half life in rat and dog.
- ✓ BGB-10188 showed dose-dependent anti-tumor activities on B cell malignant tumors in both xenograft and humanized models.
- ✓ BGB-10188 showed inhibitory activity on Treg in blood, spleen and tumor tissues and increased anti-tumor activity with PD-1 Abs in CT26WT syngeneic model.
- ✓ BGB-10188 has improved safety profile regarding to liver toxicities in mice compared to idelalisib.