TISLELIZUMAB PLUS CHEMOTHERAPY AS FIRST-LINE TREATMENT FOR UNRESECTABLE, LOCALLY ADVANCED RECURRENT/METASTATIC ESOPHAGEAL SQUAMOUS CELL CARCINOMA

Harry Yoon[1], Ken Kato[2], Richard Hubner[3], Eric Raymond[4], Aiyang Tao[5], Sumei Liu[6], Ibrahim Qazi[5], Jianming Xu[7]

[1]Mayo Clinic, Rochester, MN, USA; [2]National Cancer Center Hospital, Tokyo, Japan; [3]Christie NHS Foundation Trust, Manchester, UK; [4]Centre Hospitalier Paris Saint-Joseph, Paris, France; [5]BeiGene USA, Inc., San Mateo, CA, USA; [6]BeiGene (Beijing) Co., Ltd., Beijing, China; [7]The Fifth Medical Center, People's Liberation Army General Hospital, Beijing, China

RATIONALE TRIAL PROGRAM

Abstract: TPS462; Board: L6

American Society for Clinical Oncology: Gastrointestinal Cancers Symposium

January 23-25, 2020, San Francisco, CA

BACKGROUND

- Esophageal cancer is the seventh most common cancer worldwide and the sixth most common cause of cancer-related deaths[1]
- Esophageal squamous cell cancer (ESCC) remains the predominant histological subtype and accounts for most deaths from esophageal cancer[2]
- Monoclonal antibodies against immune checkpoint receptors, such as programmed cell death protein-1 (PD-1), have demonstrated promising antitumor activity and manageable safety in patients with advanced unresectable or metastatic ESCC[3,4]
- Tislelizumab is a monoclonal antibody with high affinity and specificity for PD-1
 - Tislelizumab shows higher affinity to PD-1 than pembrolizumab and nivolumab with ~100- and 50-fold slower off-rates, respectively[5]
- Tislelizumab has a different binding orientation to PD-1 compared with pembrolizumab and nivolumab; the binding surface on PD-1 for tislelizumab partially overlaps with that for pembrolizumab, but differs significantly from that for nivolumab (**Figure 1**)[5]

Figure 1: Tislelizumab Binds to PD-1 in an Orientation Different From Pembrolizumab (A) and Nivolumab (B)

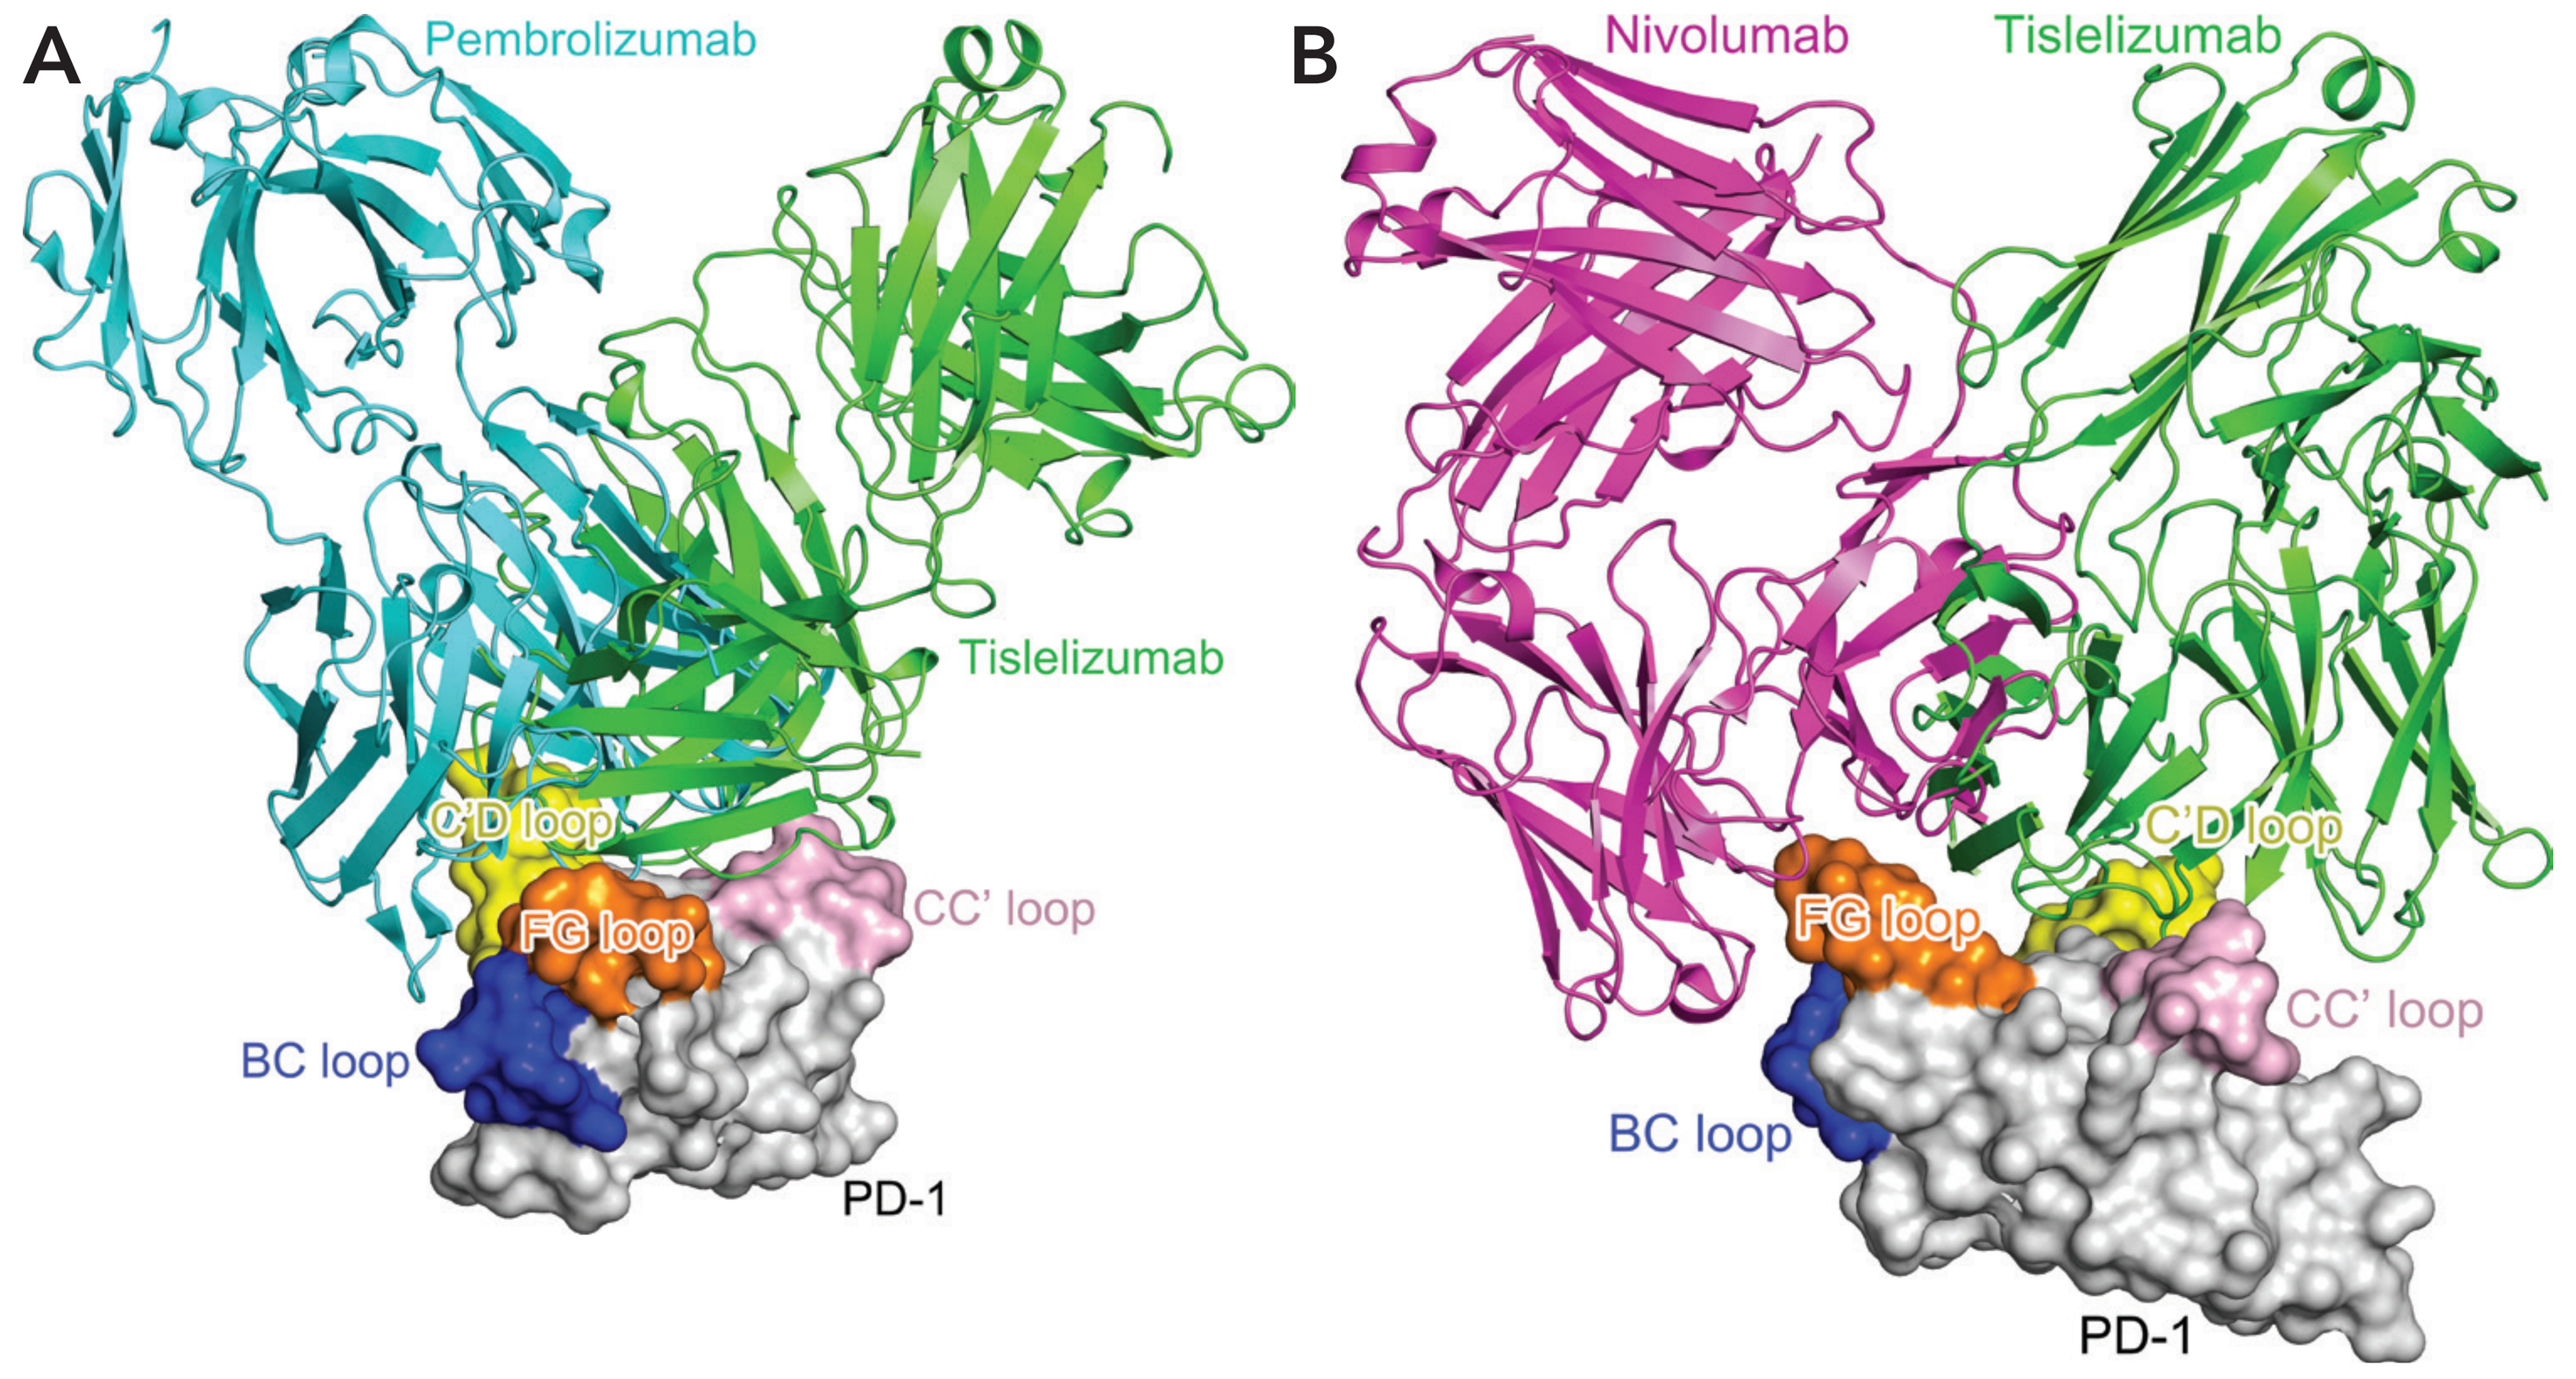

A
Pembrolizumab
Tislelizumab
C'D loop
FG loop
CC' loop
BC loop
PD-1
B
Nivolumab
Tislelizumab
FG loop
C'D loop
CC' loop
BC loop
PD-1

PD-1, tislelizumab, pembrolizumab, and nivolumab are colored in gray, green, cyan, and magenta, respectively. The BC, CC', C'D, and FG loops of PD-1 are colored in blue, pink, yellow, and orange, respectively.
Abbreviation: PD-1, programmed death-1 receptor.

- Tislelizumab was specifically engineered to minimize binding to Fc gamma receptors (FcγRs) on macrophages in order to abrogate antibody-dependent phagocytosis, a mechanism of T-cell clearance and potential resistance to anti-PD-1 therapy (**Figure 2**)[6,7]
- Results from early phase clinical studies suggest tislelizumab, as a single agent or in combination with chemotherapy, was generally well tolerated and had antitumor activity in patients with advanced solid tumors, including ESCC[8-10]

Figure 2: Lack of FcγR Binding May Help Prevent Macrophage-Mediated T-Cell Clearance

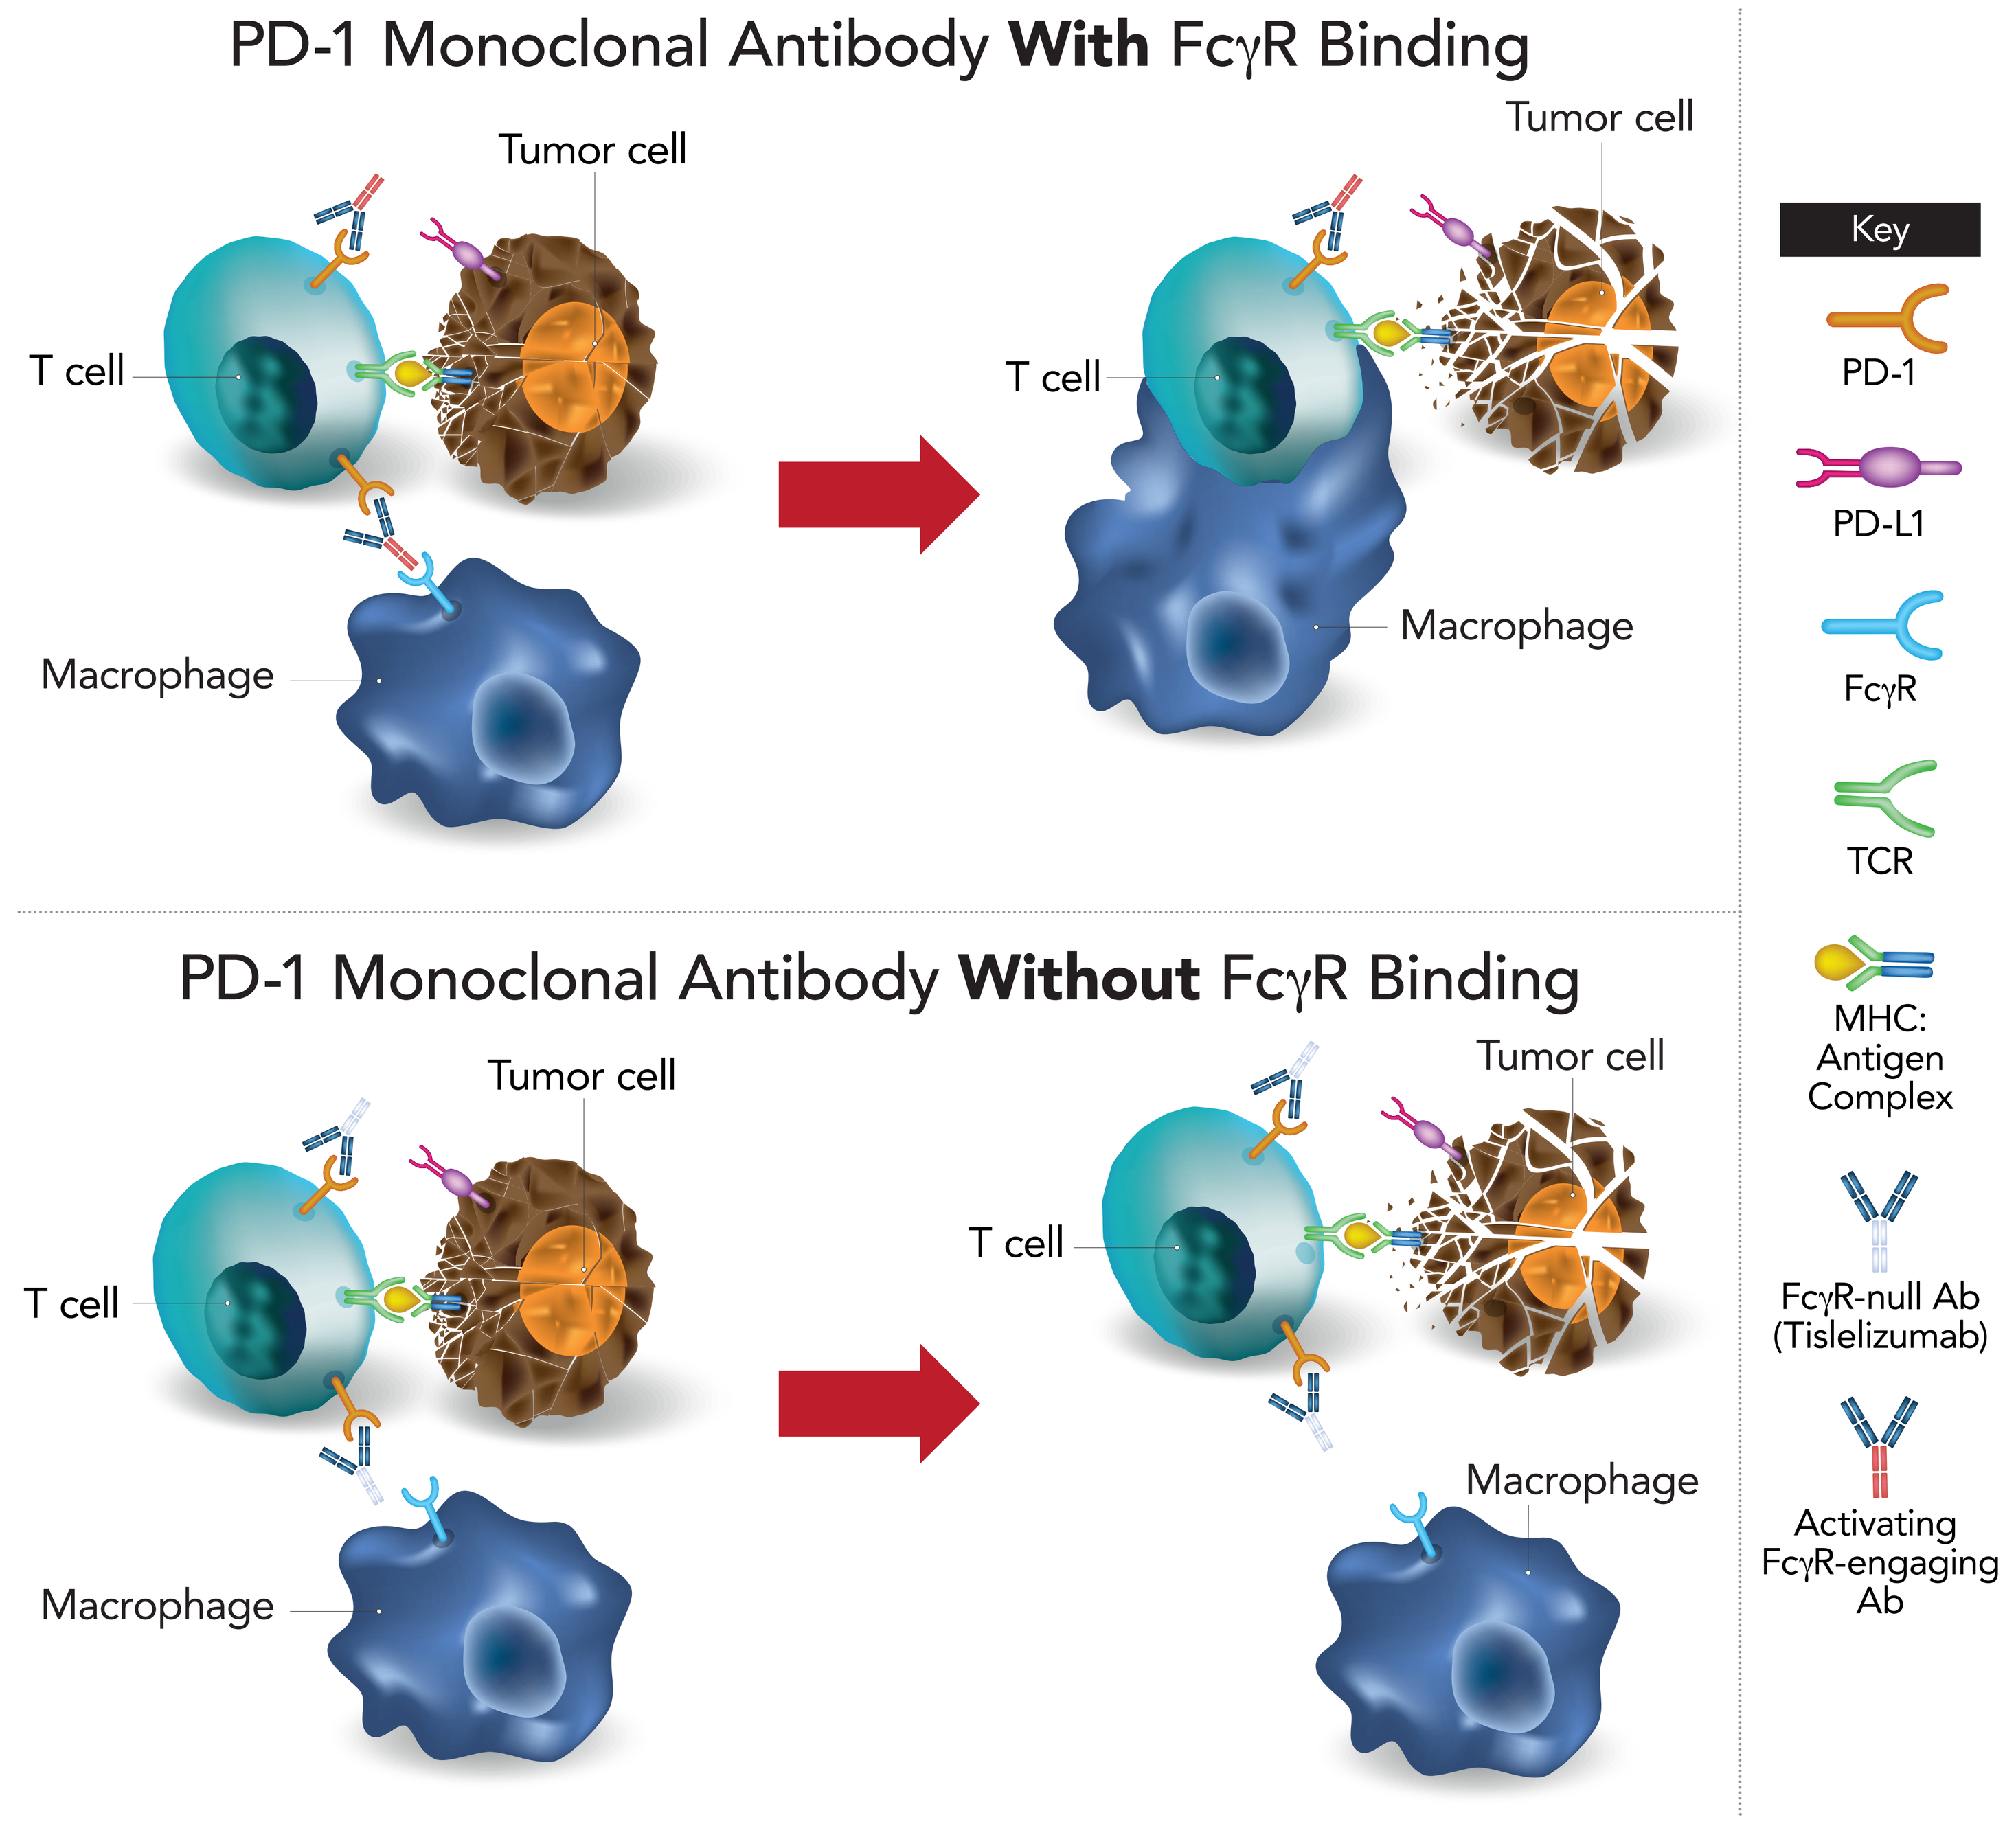

PD-1 Monoclonal Antibody **With** FcγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
PD-1 Monoclonal Antibody **Without** FcγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
Key
PD-1
PD-L1
FcγR
TCR
MHC: Antigen Complex
FcγR-null Ab (Tislelizumab)
Activating FcγR-engaging Ab

Abbreviations: Ab, antibody; MHC, major histocompatibility complex; PD-1, programmed death-1 receptor; PD-L1, programmed death-ligand 1; TCR, T-cell receptor.

METHODS

Overall Design and Study Objectives

- RATIONALE 306 (NCT03783442) is a global, double-blind, placebo-controlled, randomized, phase 3 study that is being conducted in approximately 160 study centers in Asia, Europe, North America, and Australia with approximately 480 patients and is designed to evaluate tislelizumab plus chemotherapy as first-line treatment of unresectable, locally advanced recurrent or metastatic ESCC (**Figure 3**)
- The primary objective is to compare survival (progression-free [PFS] and overall [OS]) of patients treated with tislelizumab plus investigator-chosen chemotherapy (ICC) versus patients treated with placebo plus ICC
- Secondary endpoints will include the safety of combination therapy, objective response rate (ORR), and duration of response (DoR), as assessed by blinded independent review committee per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1 criteria, as well as health-related quality of life measures (European Organization for Research and Treatment of Cancer [EORTC] Quality of Life Questionnaire [QLQ]-C30 index, the EORTC QLQ esophageal cancer module OES18, and EQ-5D-5L)

Figure 3: Study Design

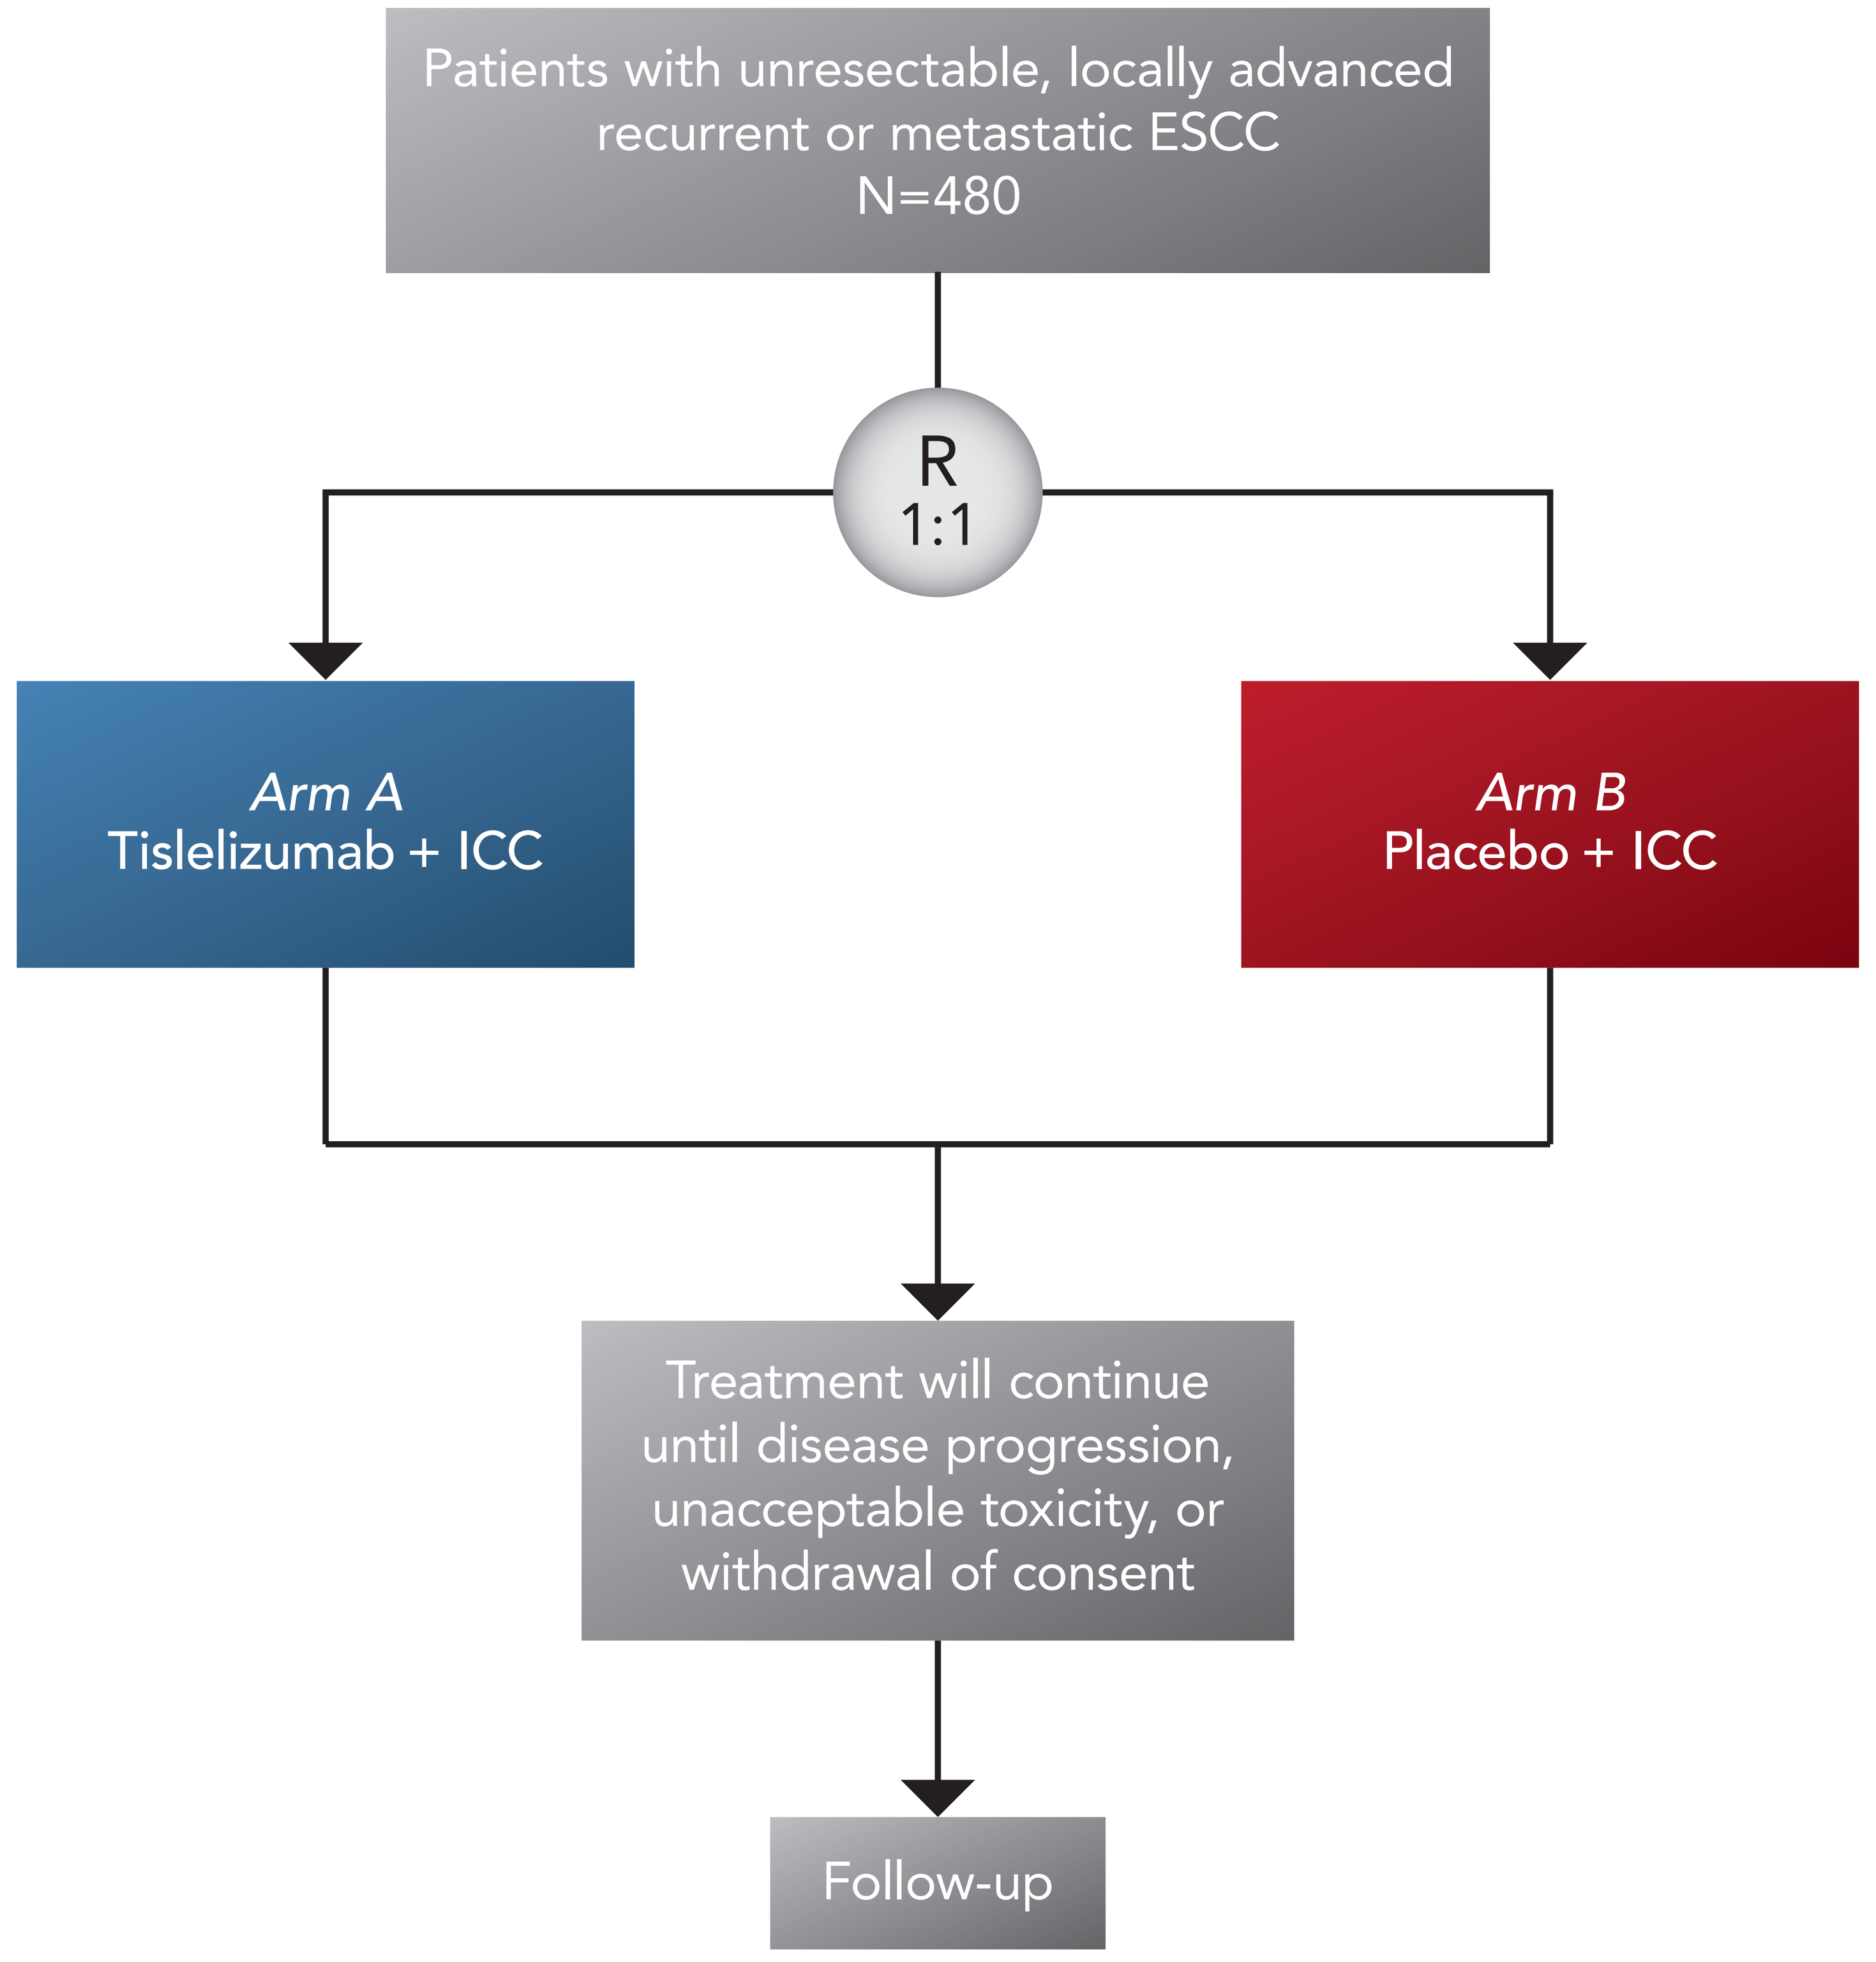

Patients with unresectable, locally advanced recurrent or metastatic ESCC
N=480
R
1:1
Arm A
Tislelizumab + ICC
Arm B
Placebo + ICC
Treatment will continue until disease progression, unacceptable toxicity, or withdrawal of consent
Follow-up

Abbreviations: ESCC, esophageal cell carcinoma; ICC, investigator-chosen chemotherapy; R, randomized.

Study Population

- Adult patients with histologically confirmed unresectable stage IV ESCC at first diagnosis (or locally advanced recurrent/metastatic disease with a treatment-free interval of at least 6 months), an Eastern Cooperative Oncology Group performance status score of ≤1, and at least one measurable or evaluable lesion as assessed per RECIST v1.1 are eligible
 - Patients must not have received prior PD-1 or programmed death-ligand 1 or -2 (PD-L1 or PD-L2) therapy, systemic therapy for unresectable locally advanced recurrent or metastatic ESCC, or palliative radiation treatment ≤4 weeks before treatment
- Patients with evidence of fistula (either esophageal/bronchial or esophageal/aorta) or uncontrollable pleural effusion, pericardial effusion, or ascites requiring frequent drainage will be excluded from the study

Treatment

- Patients will be randomized 1:1 to receive either tislelizumab 200 mg by IV infusion every 3 weeks (Q3W; Day 1 of each 21-day cycle) plus ICC (*Arm A*) or placebo plus ICC (*Arm B*)
- Investigators can choose from the following chemotherapy options:
 - Cisplatin 60-80 mg/m^2 (oxaliplatin 130 mg/m^2 outside of China or Japan) by IV infusion on Day 1 Q3W + 5-FU 750-800 mg/m^2 by continuous IV infusion over 24 hours on Days 1-5 Q3W; **or**
 - Cisplatin/oxaliplatin on Day 1 Q3W + oral capecitabine 1000 mg/m^2 twice daily on Days 1-14 Q3W; **or**
 - Cisplatin/oxaliplatin on Day 1 Q3W + paclitaxel 175 mg/m^2 IV on Day 1 Q3W (cisplatin may be given on an alternate schedule based on local guidelines)

Study Assessments and Statistical Analysis

- The primary endpoints, PFS and OS of tislelizumab + ICC versus placebo + ICC, will be assessed using the Kaplan-Meier method
- Safety will be assessed by the incidence and severity of adverse events (AEs) according to National Cancer Institute Common Terminology Criteria for AEs, v4.03 criteria, physical examinations, vital signs, electrocardiograms, and laboratory test results up to 30 days after the last dose of study drug
 - Safety will be assessed in the safety analysis set, which will consist of all subjects who receive ≥1 dose of the assigned study drug
- Quality of life assessments will occur at baseline, on Day 1 of every cycle through Cycle 6, then every other cycle thereafter until the safety follow-up visit

REFERENCES

1. Bray F, Ferlay J, Soerjomataram I, et al. Global cancer statistics 2018: Globocan estimates of incidence and mortality worldwide for 36 cancers in 185 countries. *CA Cancer J Clin.* 2018;68:394-424.
2. Abbas G, Krasna M. Overview of esophageal cancer. *Ann Cardiothorac Surg.* 2017;6:131-136.
3. Doi T, Piha-Paul SA, Jalal SI, et al. Safety and antitumor activity of the anti-programmed death-1 antibody pembrolizumab in patients with advanced esophageal carcinoma. *J Clin Oncol.* 2018;36:61-67.
4. Kudo T, Hamamoto Y, Kato K, et al. Nivolumab treatment for oesophageal squamous-cell carcinoma: An open-label, multicentre, phase 2 trial. *Lancet Oncol.* 2017;18:631-639.
5. Feng Y, Hong Y, Sun H, Zhang B, et al. The molecular binding mechanism of tislelizumab, an investigational anti-PD-1 antibody, is differentiated from pembrolizumab and nivolumab. In: *Proceedings of the 110th Annual Meeting of the American Association for Cancer Research.* Atlanta, GA: American Association of Cancer Research; 2019. Abstract 4048.
6. Zhang T, Song X, Xu L, et al. The binding of an anti-PD-1 antibody to FcγRI has a profound impact on its biological functions. *Cancer Immunol Immunother.* 2018;67:1079-1090.
7. Dahan R, Sega E, Engelhardt J, et al. FcγRs modulate the anti-tumor activity of antibodies targeting the PD-1/PD-L1 axis. *Cancer Cell.* 2015;28:543.
8. Desai J, Markman B, Sandhu SK, et al. A phase I dose-escalation study of BGB-A317, an anti-programmed death-1 (PD-1) mAb in patients with advanced solid tumors. *J Immunother Cancer.* 2016;4(suppl 1):154.
9. Desai J, Millward M, Chao Y, et al. Preliminary results from subsets of patients (pts) with advanced gastric cancer (GC) and esophageal carcinoma (EC) in a dose-escalation/expansion study of BGB-A317, an anti-PD-1 monoclonal antibody (mAb). *Ann Oncol.* 2017;28(suppl 5):v122-v141.
10. Xu N, Yuan X, Wang B, et al. Tislelizumab in combination with chemotherapy for the treatment of Chinese patients (pts) with esophageal squamous cell carcinoma (ESCC): Results from one cohort of an ongoing phase 2 study. *J Clin Oncol.* 2019;37(suppl 4):14.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative centers' study staff and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. This study was sponsored by BeiGene, Ltd. Writing and editorial assistance was provided by Regina Switzer, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago. IL), and funded by the study sponsor.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com