Tislelizumab Combined With Chemotherapy as First-line Therapy for Locally Advanced or Metastatic Non-squamous Non-small Cell Lung Cancer (nsq-NSCLC): Programmed Death-Ligand 1 (PD-L1) Expression ≥50% Subgroup Analysis of the Randomized, Phase 3 RATIONALE-304 Trial

Yan Yu,[1*] Mengzhao Wang,[2] Jie Wang,[3] Xinmin Yu,[4] Yanping Hu,[5] Liao Wangjun,[6] Xingya Li,[7] Yuepeng Liu,[8] Weidong Li,[9] Yuanyuan Bao,[10] Shun Lu[11]

[1]Department of Thoracic Oncology, Harbin Medical University Cancer Hospital, Harbin, China; [2]Department of Pulmonary Medicine, Peking Union Medical College Hospital, Beijing, China; [3]Department of Medical Oncology, Cancer Hospital Chinese Academy of Medical Sciences, Beijing, China; [4]Department of Thoracic Oncology, Zhejiang Cancer Hospital, Hangzhou, China; [5]Department of Thoracic Oncology, Hubei Cancer Hospital, Wuhan, China; [6]Department of Oncology, Nanfang Hospital, Southern Medical University, Guangzhou, China; [7]Department of Medical Oncology, The First Affiliated Hospital of Zhengzhou University/Henan Cancer Hospital, Zhengzhou, China; [8]Department of Medical Oncology, The First Hospital of China Medical University, Shenyang, China; [9]Department of Medical Oncology, Cancer Center of Guangzhou Medical University, Guangzhou, China; [10]Global Statistics and Data Science, BeiGene (Shanghai) Co., Ltd., Shanghai, China; [11]Department of Oncology, Shanghai Chest Hospital, Shanghai Jiao Tong University, School of Medicine, Shanghai, China. *Corresponding and presenting author.

Poster No: 37P presented at ELCC, Paris, France, March 26-29, 2025

Conclusions

In patients with advanced nsq-NSCLC and tumor PD-L1 expression ≥50%, first-line tislelizumab plus chemotherapy demonstrated clinically meaningful improvements in progression-free survival (PFS), overall survival (OS), objective response rate (ORR), and duration of response (DoR) and a manageable safety profile compared with chemotherapy alone.

Long-term follow-up data demonstrated durable clinical improvement with tislelizumab plus chemotherapy.

These findings support tislelizumab plus chemotherapy as a treatment option in first-line nsq-NSCLC patients with high PD-L1 expression.

Background

Lung cancer remains a leading cause of cancer-related mortality worldwide.[1] In advanced NSCLC, approximately 22%-30% of patients exhibit high PD-L1 expression (defined as ≥50% of tumor cells expressing PD-L1).[2,3] Higher PD-L1 expression levels in some tumor types have been associated with improved responses to immune checkpoint inhibitors.[4]

The phase 3 RATIONALE-304 trial (NCT03663205) demonstrated that first-line tislelizumab (BGB-A317, an anti-programmed death-1 inhibitor) plus chemotherapy improved efficacy outcomes compared with chemotherapy alone in Chinese patients with advanced nsq-NSCLC.[5] This exploratory analysis specifically examines the outcomes in patients from RATIONALE-304 whose tumors exhibited high PD-L1 expression (≥50%).

Methods

- Patients aged 18-75 years with treatment-naive, stage IIIB (not amenable to curative surgery/radiotherapy) or stage IV nsq-NSCLC were enrolled[6]
- Patients were randomized (2:1) to open-label:
 - Tislelizumab 200 mg intravenously every 3 weeks plus platinum-based chemotherapy for 4-6 cycles, followed by maintenance tislelizumab plus pemetrexed; or
 - Platinum-based chemotherapy alone for 4-6 cycles, followed by maintenance pemetrexed
- Endpoints in the PD-L1 ≥50% population:
 - Primary: PFS assessed by independent review committee (IRC)
 - Secondary: OS, ORR assessed by IRC using Response Evaluation Criteria in Solid Tumors version 1.1, DoR, and safety
- PD-L1 expression was assessed during screening using the VENTANA PD-L1 (SP263) Assay (Roche Diagnostics, Indianapolis, IN, USA) at a central laboratory

Results

Patient Disposition and Baseline Characteristics

- Of 334 randomized patients, 110 (32.9%) had PD-L1 expression ≥50% (tislelizumab plus chemotherapy: n=74; chemotherapy alone: n=36)
- Baseline patient characteristics were similar between arms and consistent with the ITT population[5]; median age was 62 years, 71.8% were male, and 81.8% had stage IV disease
- Median study follow-up was 16.5 months (data cutoff: October 26, 2020) and 23.4 months (updated OS analysis; data cutoff: April 26, 2023)
- Subsequent systemic therapy was received by 36.5% (27/74) of patients in the tislelizumab plus chemotherapy arm vs 66.7% (24/36) in the chemotherapy alone arm, with 8.1% (6/74) and 41.7% (15/36) receiving subsequent immunotherapy, respectively; 36.1% (13/36) patients from the chemotherapy alone arm crossed over to tislelizumab
- Treatment discontinuation occurred in 60.8% (45/74) of patients in the tislelizumab plus chemotherapy arm and 97.2% (35/36) in the chemotherapy alone arm

Efficacy

- At the final analysis, tislelizumab plus chemotherapy improved PFS_{IRC} vs chemotherapy alone (median PFS_{IRC} 14.6 vs 4.6 months; stratified HR=0.31; 95% CI: 0.18, 0.55) in the PD-L1 ≥50% population (**Figure 1A**)
 - PFS_{IRC} benefit for tislelizumab plus chemotherapy was consistent across various patient subgroups (**Figure 1B**)
- Tislelizumab plus chemotherapy showed higher confirmed ORR_{IRC} (70.3% vs 30.6%), greater complete response (CR) rate (9.5% vs 0%), and longer DoR (not estimable [NE] vs 8.5 months) vs chemotherapy alone (**Table 1**)
- OS was improved with tislelizumab plus chemotherapy vs chemotherapy alone (**Table 1**):
 - Final analysis: median OS was NE vs 13.1 months (HR=0.39; 95% CI: 0.22, 0.71)
 - Updated analysis (with an additional 6.9 months of median follow-up): median OS was 41.9 vs 13.1 months (HR=0.38; 95% CI: 0.24, 0.63) (**Figure 2A**)
 - OS results from subgroup analyses were variable due to limited sample sizes (**Figure 2B**)
- Patients with long-term tislelizumab treatment (≥35 cycles, n=22) demonstrated 100% ORR_{IRC} (36.4% CR), median DoR NE (95% CI: 29.6 months, NE), and 4-year OS rate of 90.5% (95% CI: 67.0, 97.5); 63.6% remained progression-free without subsequent therapy

Figure 1. (A) Kaplan–Meier Curves of PFS_{IRC} and (B) Prespecified Subgroup Analysis of PFS_{IRC} (PD-L1 ≥50% Population)

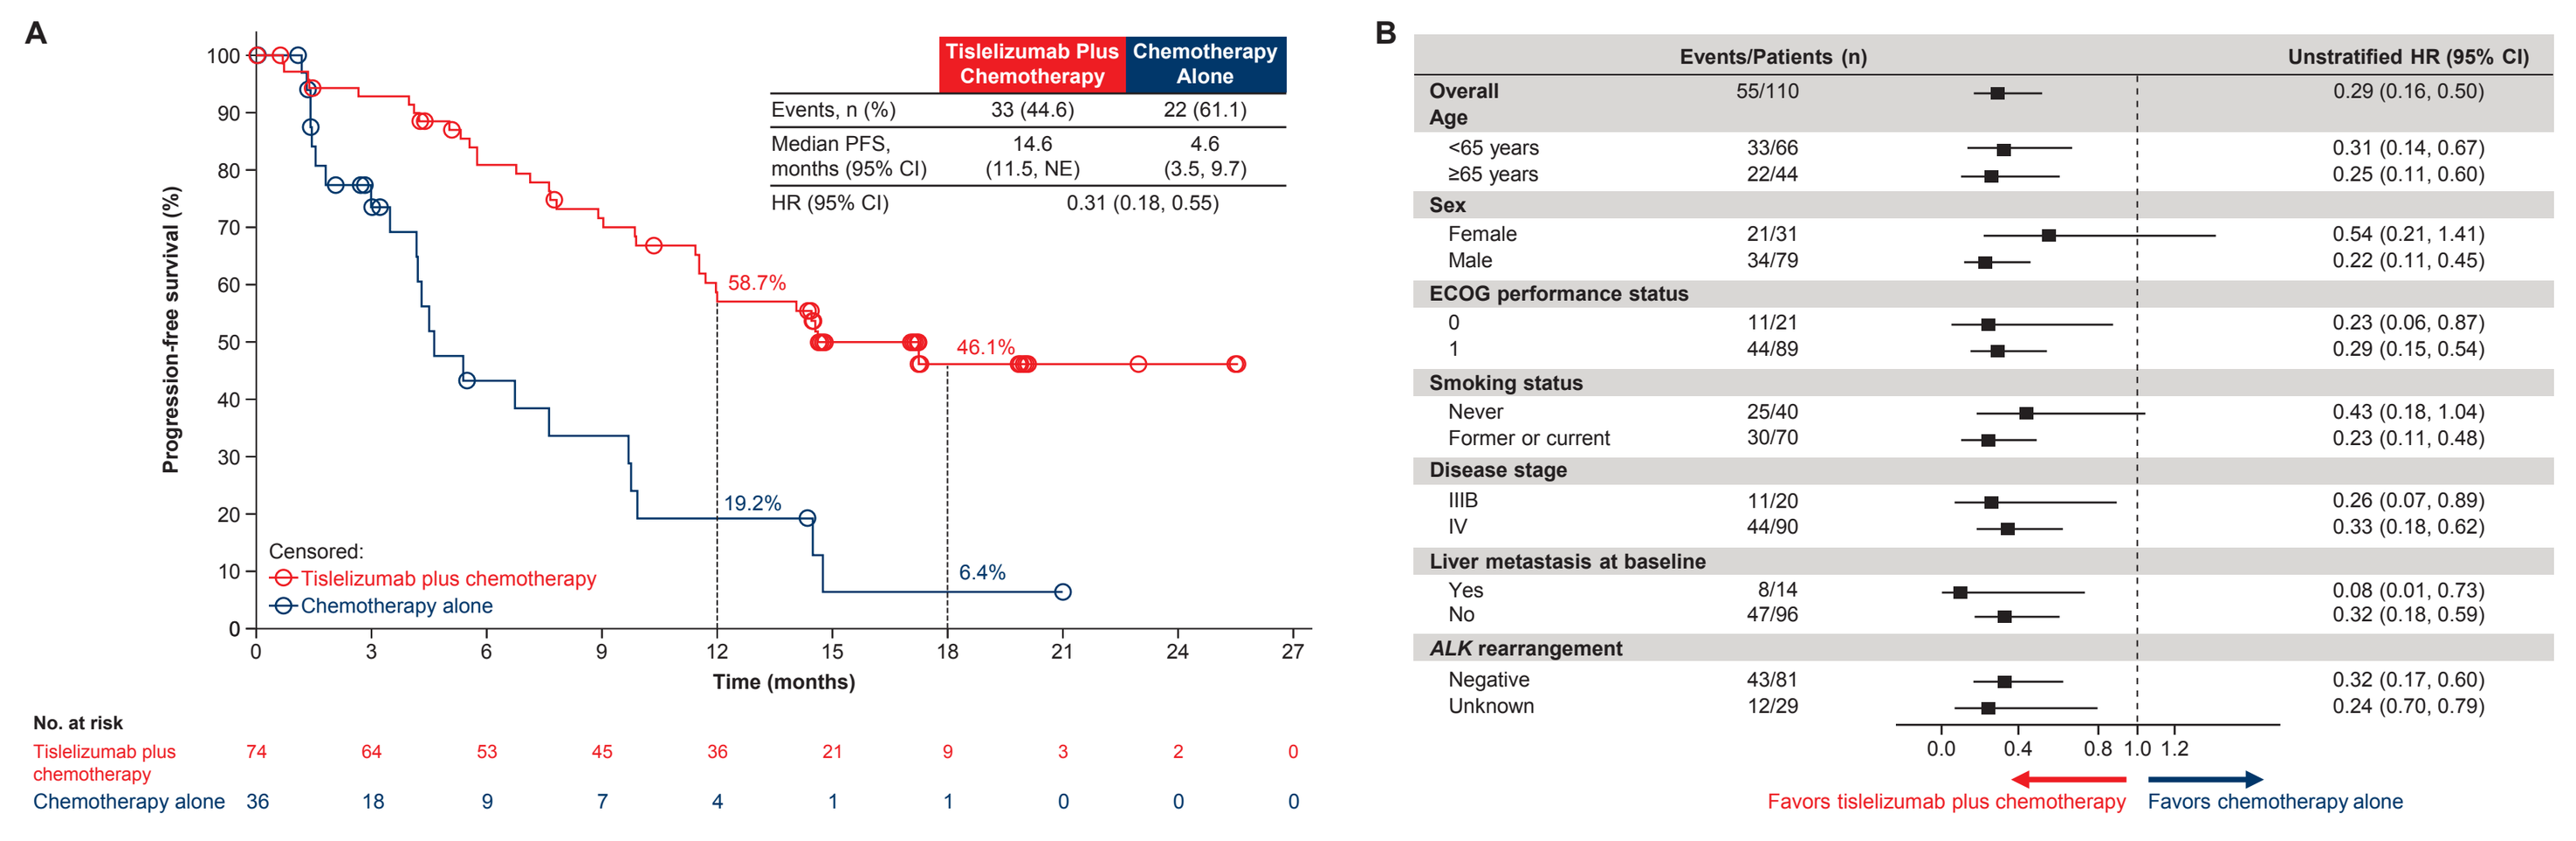
A
Tislelizumab Plus Chemotherapy
Chemotherapy Alone
Events, n (%) 33 (44.6) 22 (61.1)
Median PFS, months (95% CI) 14.6 (11.5, NE) 4.6 (3.5, 9.7)
HR (95% CI) 0.31 (0.18, 0.55)
Progression-free survival (%)
58.7%
46.1%
19.2%
6.4%
Censored:
Tislelizumab plus chemotherapy
Chemotherapy alone
Time (months)
0 3 6 9 12 15 18 21 24 27
No. at risk
Tislelizumab plus chemotherapy 74 64 53 45 36 21 9 3 2 0
Chemotherapy alone 36 18 9 7 4 1 1 0 0 0
B
Events/Patients (n)
Unstratified HR (95% CI)
Overall 55/110 0.29 (0.16, 0.50)
Age
<65 years 33/66 0.31 (0.14, 0.67)
≥65 years 22/44 0.25 (0.11, 0.60)
Sex
Female 21/31 0.54 (0.21, 1.41)
Male 34/79 0.22 (0.11, 0.45)
ECOG performance status
0 11/21 0.23 (0.06, 0.87)
1 44/89 0.29 (0.15, 0.54)
Smoking status
Never 25/40 0.43 (0.18, 1.04)
Former or current 30/70 0.23 (0.11, 0.48)
Disease stage
IIIB 11/20 0.26 (0.07, 0.89)
IV 44/90 0.33 (0.18, 0.62)
Liver metastasis at baseline
Yes 8/14 0.08 (0.01, 0.73)
No 47/96 0.32 (0.18, 0.59)
ALK rearrangement
Negative 43/81 0.32 (0.17, 0.60)
Unknown 12/29 0.24 (0.70, 0.79)
0.0 0.4 0.8 1.0 1.2
Favors tislelizumab plus chemotherapy
Favors chemotherapy alone

Data cutoff: October 26, 2020. (A) HRs and associated 95% CIs were calculated using a Cox proportional-hazards model stratified by stage IIIB vs IV disease. (B) HRs and associated 95% CIs were calculated using an unstratified Cox proportional-hazards model.

Abbreviations: ALK, anaplastic lymphoma kinase; CI, confidence interval; ECOG, Eastern Cooperative Oncology Group; HR, hazard ratio; IRC, independent review committee; NE, not estimable; PD-L1, programmed death-ligand 1; PFS, progression-free survival.

Table 1. Secondary Efficacy Endpoints (PD-L1 ≥50% Population)

	Tislelizumab Plus Chemotherapy (n=74)	Chemotherapy Alone (n=36)	HR (95% CI)
ORR_{IRC}, % (95% CI)[a]	70.3 (58.5, 80.3)	30.6 (16.3, 48.1)	-
ORR difference, % (95% CI)[b]	39.5 (21.2, 57.9)		-
Median DoR, months (95% CI)[c]	NE (13.2, NE)	8.5 (3.3, NE)	0.32 (0.13, 0.82)
Median OS (final analysis), months (95% CI)[c]	NE (NE, NE)	13.1 (5.6, NE)	0.39 (0.22, 0.71)[d]
Median OS (updated analysis), months (95% CI)[c,e]	41.9 (24.1, NE)	13.1 (5.6, 19.4)	0.38 (0.24, 0.63)

Data cutoff: October 26, 2020. The chemotherapy alone arm was the reference group.

[a] 95% CI for ORR was calculated using the Clopper–Pearson method. [b] ORR difference was calculated using the Cochran–Mantel–Haenszel chi-square test with actual stratification factors as strata. [c] Medians were estimated by Kaplan–Meier methodology, with 95% CIs estimated using the method of Brookmeyer and Crowley. [d] HR and 95% CI were calculated using a Cox proportional-hazards model stratified by stage IIIB vs IV disease. [e] Updated OS analysis data cutoff: April 26, 2023.

Abbreviations: CI, confidence interval; DoR, duration of response; HR, hazard ratio; NE, not evaluable; ORR_{IRC}, objective response rate by independent review committee; OS, overall survival; PD-L1, programmed death-ligand 1.

Figure 2. (A) Kaplan–Meier Curves of OS and (B) Prespecified Subgroup Analysis of OS (PD-L1 ≥50% Population)

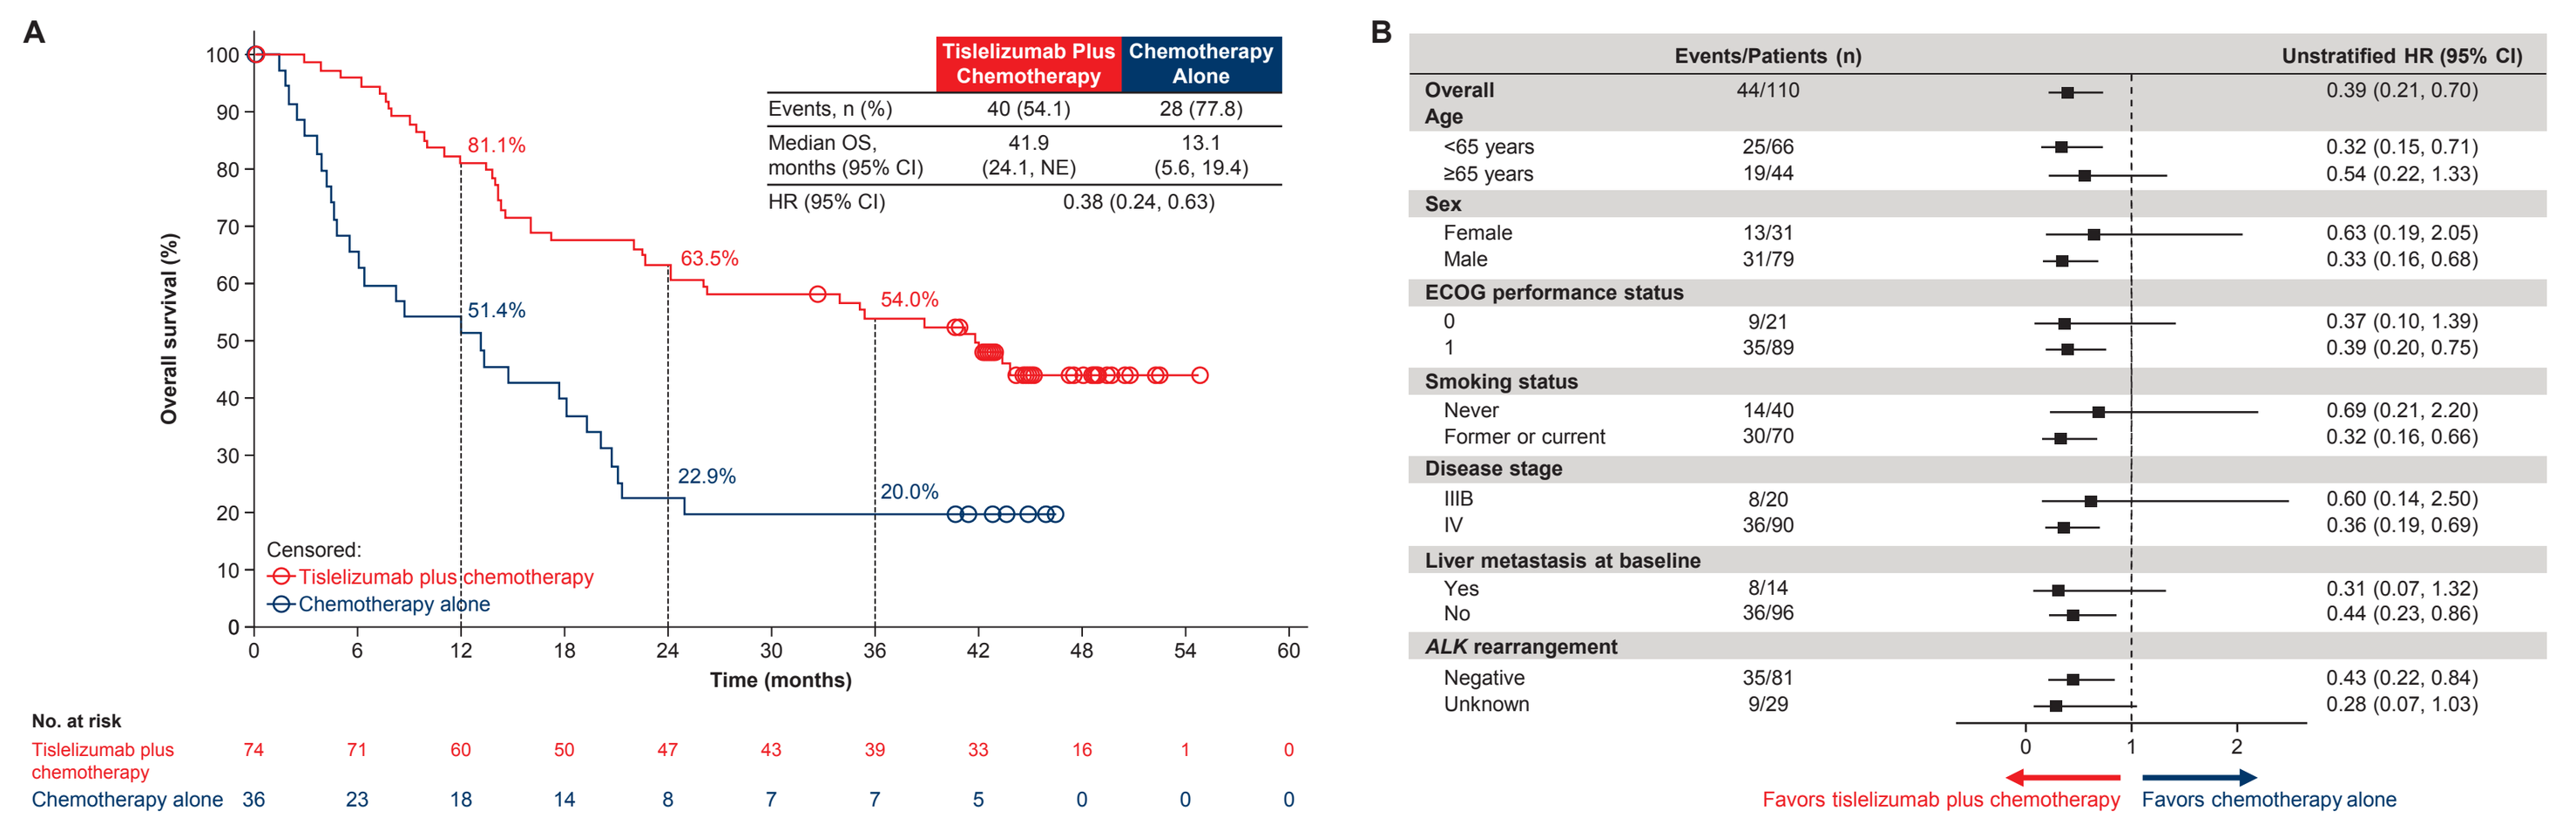
A
Tislelizumab Plus Chemotherapy
Chemotherapy Alone
Events, n (%) 40 (54.1) 28 (77.8)
Median OS, months (95% CI) 41.9 (24.1, NE) 13.1 (5.6, 19.4)
HR (95% CI) 0.38 (0.24, 0.63)
Overall survival (%)
81.1%
63.5%
54.0%
51.4%
22.9%
20.0%
Censored:
Tislelizumab plus chemotherapy
Chemotherapy alone
Time (months)
0 6 12 18 24 30 36 42 48 54 60
No. at risk
Tislelizumab plus chemotherapy 74 71 60 50 47 43 39 33 16 1 0
Chemotherapy alone 36 23 18 14 8 7 7 5 0 0 0
B
Events/Patients (n)
Unstratified HR (95% CI)
Overall 44/110 0.39 (0.21, 0.70)
Age
<65 years 25/66 0.32 (0.15, 0.71)
≥65 years 19/44 0.54 (0.22, 1.33)
Sex
Female 13/31 0.63 (0.19, 2.05)
Male 31/79 0.33 (0.16, 0.68)
ECOG performance status
0 9/21 0.37 (0.10, 1.39)
1 35/89 0.39 (0.20, 0.75)
Smoking status
Never 14/40 0.69 (0.21, 2.20)
Former or current 30/70 0.32 (0.16, 0.66)
Disease stage
IIIB 8/20 0.60 (0.14, 2.50)
IV 36/90 0.36 (0.19, 0.69)
Liver metastasis at baseline
Yes 8/14 0.31 (0.07, 1.32)
No 36/96 0.44 (0.23, 0.86)
ALK rearrangement
Negative 35/81 0.43 (0.22, 0.84)
Unknown 9/29 0.28 (0.07, 1.03)
0 1 2
Favors tislelizumab plus chemotherapy
Favors chemotherapy alone

(A) Data cutoff: April 26, 2023. HRs and associated 95% CIs were estimated from an unstratified Cox proportional-hazards model with the chemotherapy alone arm as a reference group. Event-free rates were estimated by Kaplan–Meier method with 95% CIs estimated using the Greenwood's formula. (B) Data cutoff: October 26, 2020. HRs and associated 95% CIs were calculated using an unstratified Cox proportional-hazards model.

Abbreviations: ALK, anaplastic lymphoma kinase; CI, confidence interval; ECOG, Eastern Cooperative Oncology Group; HR, hazard ratio; NE, not estimable; OS, overall survival; PD-L1, programmed death-ligand 1.

Safety

- The incidences of grade ≥3 treatment-emergent adverse events (TEAEs) (75.7% vs 48.6%) and serious TEAEs (43.2% vs 28.6%) were higher in the tislelizumab plus chemotherapy arm compared with the chemotherapy alone arm (**Table 2**)
- The most frequently reported grade ≥3 TEAEs in both arms were neutropenia, thrombocytopenia, leukopenia, and anemia
- Immune-mediated adverse events (imAEs) of all grades were reported in 39.2% of patients in the tislelizumab plus chemotherapy arm vs 5.7% in the chemotherapy alone arm (**Table 2**)
- The most frequently reported grade ≥3 imAEs in the tislelizumab plus chemotherapy arm were pneumonitis, endocrinopathies (diabetes mellitus), and myocarditis/pericarditis

Table 2. Summary of TEAEs (PD-L1 ≥50% Population; Safety Analysis Set)

	Tislelizumab Plus Chemotherapy (n=74)	Chemotherapy Alone (n=35)
Patients with ≥1 TEAE, n (%)	74 (100.0)	35 (100.0)
Grade ≥3	56 (75.7)	17 (48.6)
Serious	32 (43.2)	10 (28.6)
Leading to death	3 (4.1)	1 (2.9)
Patients with ≥1 TEAE leading to discontinuation, n (%)	21 (28.4)	2 (5.7)
Patients with ≥1 TEAE leading to treatment modification,[a,b] n (%)	61 (82.4)	20 (57.1)
Patients with ≥1 imAE, n (%)	29 (39.2)	2 (5.7)
Grade ≥3	7 (9.5)	1 (2.9)

Data cutoff: October 26, 2020. The safety analysis set includes all enrolled patients who received ≥1 dose of study drug. Adverse event grades were evaluated based on National Cancer Institute – Common Terminology Criteria for Adverse Events (version 5.0).

[a] Treatment modification of tislelizumab included dose delay, infusion interruption, and infusion rate decrease. [b] Treatment modification of chemotherapy included dose reduction, infusion interruption, dose delay, and infusion rate decrease.

Abbreviations: imAE, immune-mediated adverse event; PD-L1, programmed death-ligand 1; TEAE, treatment-emergent adverse event.

References

1. Sung H, et al. *CA Cancer J Clin*. 2021;71:209-249.
2. Dietel M, et al. *Lung Cancer*. 2019;134:174-179.
3. Skov BG, et al. *Mod Pathol*. 2020;33:109-117.
4. Tang Q, et al. *Front Immunol*. 2022;13:964442.
5. Lu S, et al. *ESMO Open*. 2024;9:103728.
6. Lu S, et al. *J Thorac Oncol*. 2021;16:1512-1522.

Acknowledgements

We would like to thank the investigators, the site support staff, and especially the patients for participating in this study. This study was sponsored by BeiGene. Medical writing support was provided Izabela Bombik, PhD, of Parexel, with funding provided by BeiGene.

Disclosures

Yan Yu reports no conflicts of interest.

*Author contact details: gpyuyan@163.com (Yan Yu)

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

FPO

GS4566_287831-L3-2250-CR03_BeiGene ELCC 2025 304 Poster_190w x90h cm S03.indd 1

3/14/25 9:50 AM