BTK-T474I with enhanced kinase activity confers growth advantage over BTK-L528W with kinase deficiency in B malignant cells

BeiGene

Wenjing Zhang, Xinxin Zhu, Qiming Zhou, Ziwei Zhang, Jiayun Song, Yang Li, Bo Li, Sijia Zhai, Peng Chi, Ming Jiang, Xiaoliang Chen, Teiko Sumiyoshi, Yangbo Yue, Si Mou, Pei Zhang, Wei Jin #, Zhirong Shen#

Authors' Affiliation: BeiGene Global Research, P.R. China; #Correspondence: zhirong.shen@beigene.com; wei.jin @beigene.com

#abstract No. 5600

BACKGROUND

Covalent BTKi (cBTKi) have transformed the therapeutic landscape for chronic lymphocytic leukemia (CLL)[1,2]. However, acquired mutations in BTK, often occurring at C481, lead to drug resistance and disease progression[3]. While non-covalent BTKi (ncBTKi) has demonstrated promising efficacy in CLL patients with BTK C481X mediated resistance, non-C481 mutations within BTK kinase domain confer ineffectiveness to cBTKi and ncBTKi as well[4,5]. Those BTK mutations most commonly involve T474I and L528W[5,6]. To better understand their role in supporting B malignant cell growth, we developed clonal competition models by co-culturing isogenic B malignant cells harboring BTK-T474I and BTK-L528W and monitor the clonal dynamics in long term culture. Additionally, we conducted mechanistic analysis utilizing proteomics and phospho-proteomics.

METHODS

T474I and L528W were introduced to BTK allele of B malignant cell line TMD8. BTK downstream signaling in BTK wildtype or mutant cells with or without anti-IgM stimulation was assessed by western blot assay. *In vitro* and *in vivo* models containing TMD8 wildtype and mutant cells were employed to investigate the clonal competition. The dynamics of individual clone were monitored by measuring BTK variant allele frequency (VAF) via amplicon sequencing. TMD8 cells expressing wildtype BTK, BTK-T474I (ENU clone), or BTK-L528W (KI #1) were cultured in medium with 2% FBS for 14 days. Cells were treated with 10 µg/ml anti-IgM or not. Samples were subjected to total proteomics and phospho-proteomics analysis.

RESULTS

Hyperactivation of phosphorylated BTK Y223 in TMD8 BTK-T474I cell and minimal BTK Y223 phosphorylation in TMD8 BTK-L528W upon IgM stimulation was observed. Enhanced BTK kinase activity indicated by downstream signaling (pPLCγ2, pERK and pAKT) were noted in TMD8 BTK-T474I cells compared to BTK-L528W cells. Those findings suggested that BTK-T474I encodes a kinase with enhanced activity while BTK-L528W results in a catalytically inactive form. Clonal competition was evaluated using co-culture models containing TMD8 BTK-T474I, BTK-L528W and wildtype cells in medium with various FBS concentration. BTK T474I clones were significantly expanded while BTK L528W clones were declined in 4 weeks culture period. An increase in BTK T474I VAF and a decrease in BTK L528W VAF were observed during tumor growth in subcutaneous models incorporating TMD8 wildtype and mutant cells. Total proteomics analysis revealed an enhanced cell cycle progression in BTK-T474I cells compared to BTK-L528W cells, as indicated by upregulation of cell cycle-related proteins. In addition, increased kinase activity of key components within BCR pathway was demonstrated through phospho-proteomics.

CONCLUSIONS

Our *in vitro* and *in vivo* clonal competition assays provided novel insights into the impact of BTK-T474I and BTK-L528W on B malignant cell growth fitness. These findings suggested that BTK-T474I, characterized by enhanced kinase activity, confers growth advantage over the kinase deficient form BTK-L528W, and provided evidence of alternative signaling mechanism between BTK mutants with enhanced or impaired kinase activity.

Figure 1. Enhanced BTK kinase activity in TMD8 BTK-T474I than TMD8 BTK-L528W cells

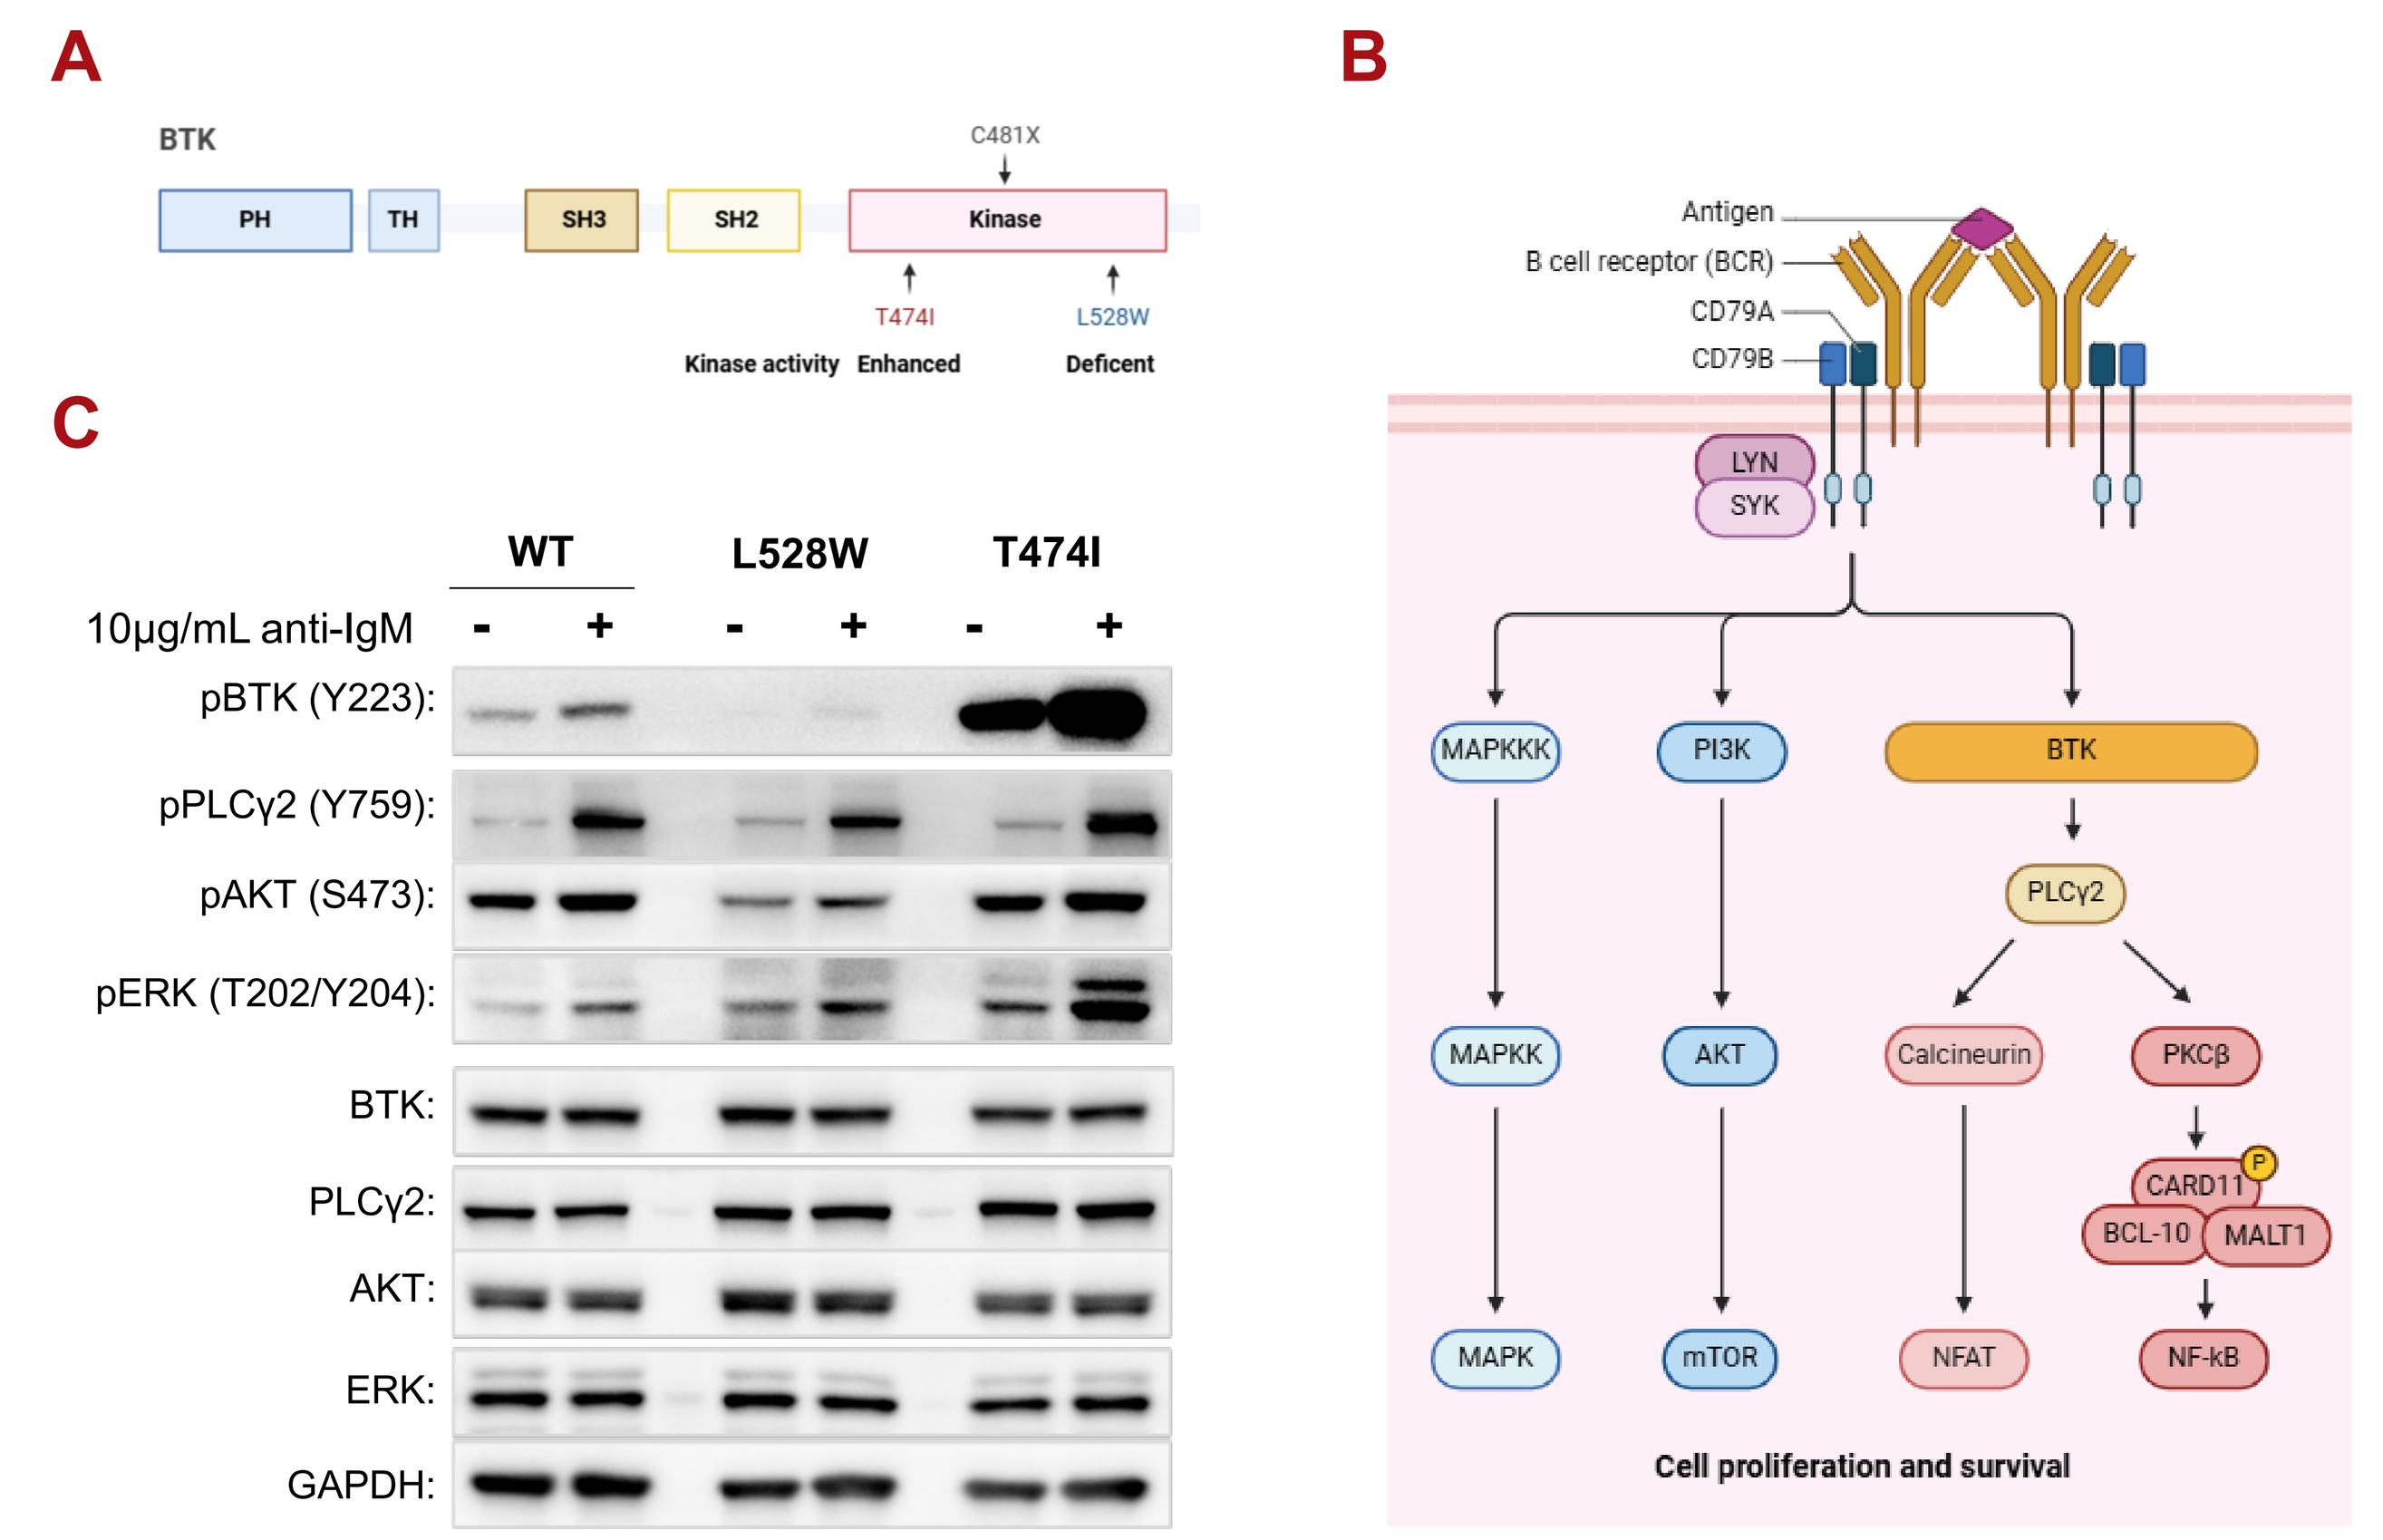
A
BTK
PH
TH
SH3
SH2
Kinase
C481X
T474I
L528W
Kinase activity
Enhanced
Deficent
B
Antigen
B cell receptor (BCR)
CD79A
CD79B
LYN
SYK
MAPKKK
PI3K
BTK
PLCγ2
MAPKK
AKT
Calcineurin
PKCβ
CARD11
BCL-10
MALT1
MAPK
mTOR
NFAT
NF-kB
Cell proliferation and survival
C
WT
L528W
T474I
10µg/mL anti-IgM
pBTK (Y223):
pPLCγ2 (Y759):
pAKT (S473):
pERK (T202/Y204):
BTK:
PLCγ2:
AKT:
ERK:
GAPDH:

(A) Diagram of covalent or non-covalent BTK inhibitor resistant mutation C481X, T474I, L528W in BTK kinase domain. (B) Schematic representation of BCR/BTK signaling pathway. (C) TMD8 cells expressing BTK wildtype (WT), T474I or L528W were incubated with 10µg/ml anti-IgM for BCR signaling stimulation. Phosphorylation of BTK Y223, PLCγ2 Y759, AKT S473, and ERK T202/Y204 were examined by western blot assay. Hyperactivation of BTK Y223 in TMD8 BTK-T474I cell whereas minimal BTK Y223 phosphorylation in TMD8 BTK-L528W cells were noted. Enhanced BTK kinase activity, as indicated by pPLCγ2, pERK, and pAKT levels, was observed in TMD8 BTK-T474I cells compared to TMD8 BTK-L528W cells.

Figure 2. TMD8 BTK-T474I cells derived from ENU mutagenesis show higher expansion potential over TMD8 BTK-L528W cells in *in vitro* clonal competition assay

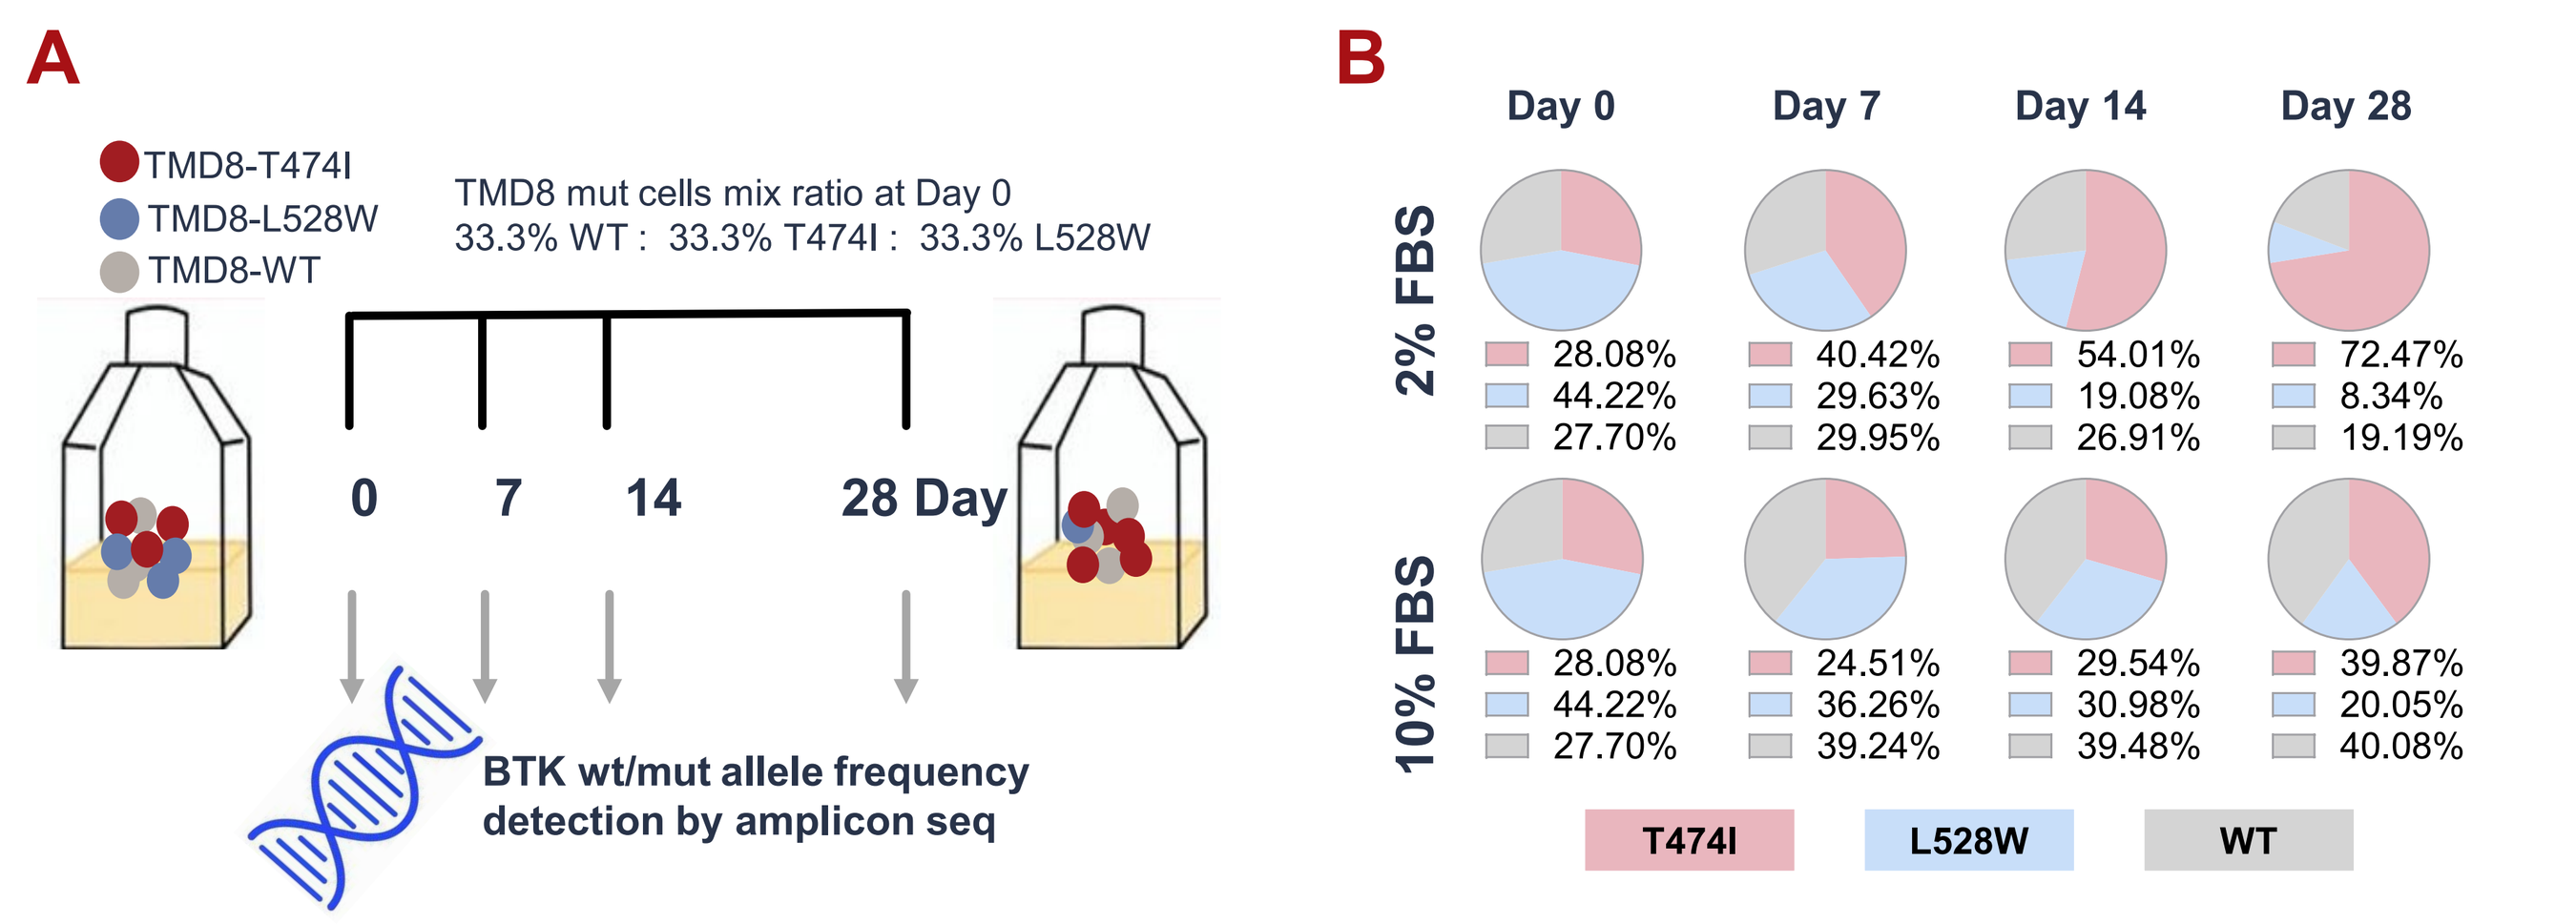
A
TMD8-T474I
TMD8-L528W
TMD8-WT
TMD8 mut cells mix ratio at Day 0
33.3% WT : 33.3% T474I : 33.3% L528W
0
7
14
28 Day
BTK wt/mut allele frequency detection by amplicon seq
B
Day 0
Day 7
Day 14
Day 28
2% FBS
28.08% 44.22% 27.70%
40.42% 29.63% 29.95%
54.01% 19.08% 26.91%
72.47% 8.34% 19.19%
10% FBS
28.08% 44.22% 27.70%
24.51% 36.26% 39.24%
29.54% 30.98% 39.48%
39.87% 20.05% 40.08%
T474I
L528W
WT

(A) Workflow of *in vitro* clonal competition assay. TMD8 wildtype cells, TMD8 BTK-T474I cells derived from ENU (N-ethyl-N-nitrosourea) mutagenesis or TMD8 BTK-L528W cells (knock-in clone #1) were mixed in equal proportion on day 0 and cultured in medium with 2% or 10% of FBS. Cell pellets were collected at specified time points for amplicon sequencing to determine the allele frequency of BTK wildtype or mutants. Results are illustrated in (B). An increase in T474I variant allele frequency (VAF) and a decrease in L528W VAF were observed following 4 weeks of cell culture. The growth advantage of TMD8 BTK-T474I over TMD8 BTK-L528W was particularly pronounced when cultured in medium with lower FBS concentration, which simulates relatively harsh condition for cell proliferation.

Figure 3. TMD8 BTK-T474I cells derived from ENU mutagenesis exhibit outgrowth advantage over TMD8 BTK-L528W cells in *in vivo* clonal competition assay

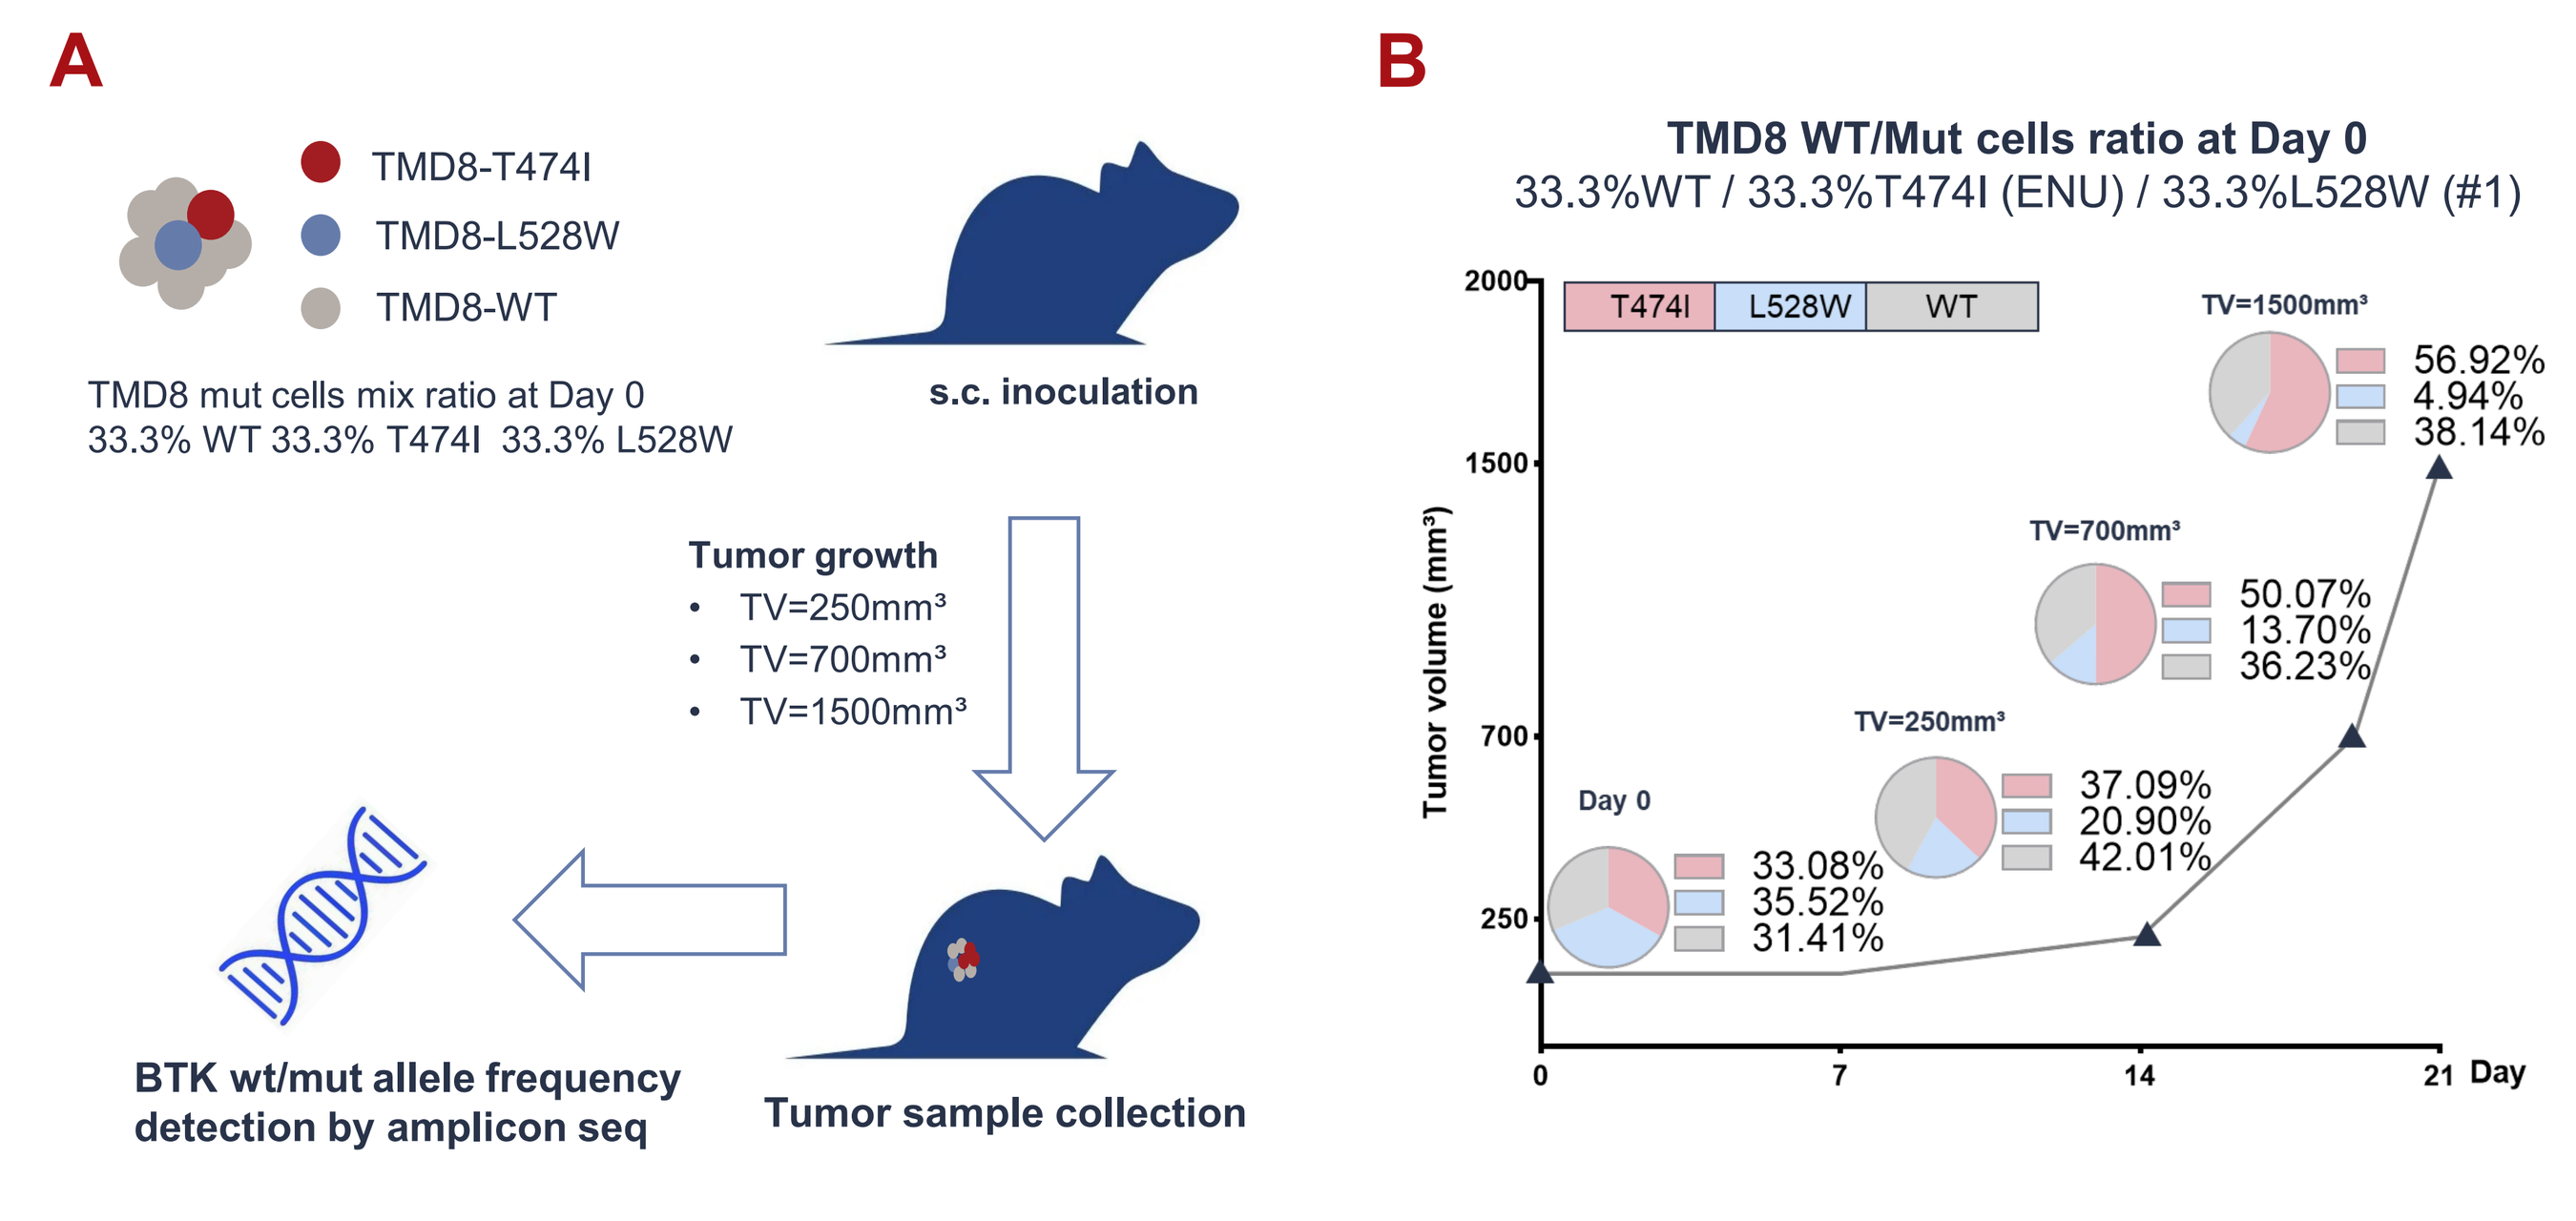
A
TMD8-T474I
TMD8-L528W
TMD8-WT
TMD8 mut cells mix ratio at Day 0
33.3% WT 33.3% T474I 33.3% L528W
s.c. inoculation
Tumor growth
TV=250mm³
TV=700mm³
TV=1500mm³
BTK wt/mut allele frequency detection by amplicon seq
Tumor sample collection
B
TMD8 WT/Mut cells ratio at Day 0
33.3%WT / 33.3%T474I (ENU) / 33.3%L528W (#1)
T474I
L528W
WT
Tumor volume (mm³)
Day 0
33.08% 35.52% 31.41%
TV=250mm³
37.09% 20.90% 42.01%
TV=700mm³
50.07% 13.70% 36.23%
TV=1500mm³
56.92% 4.94% 38.14%
Day

(A) Workflow of *in vivo* clonal competition assay. Cells with wildtype BTK, BTK-T474I and BTK-L528W were mixed in equal proportion and injected into mice on day 0 subcutaneously. Cell mixture pellets from day 0 and tumors at average sizes of 250 mm³, 700 mm³, and 1500 mm³ (N=3) were collected for amplicon sequencing to determine the allele frequencies of BTK wildtype and mutants. The dynamic changes of clone composition for tumors over time were depicted in (B). A significant increase in TMD8 BTK-T474I cells and a corresponding decrease in TMD8 BTK-L528W cells were observed.

Figure 4. BTK-T474I conferring *in vitro* and *in vivo* growth advantage over BTK-L528W was confirmed by independent TMD8 BTK-T474I and BTK-L528W clones

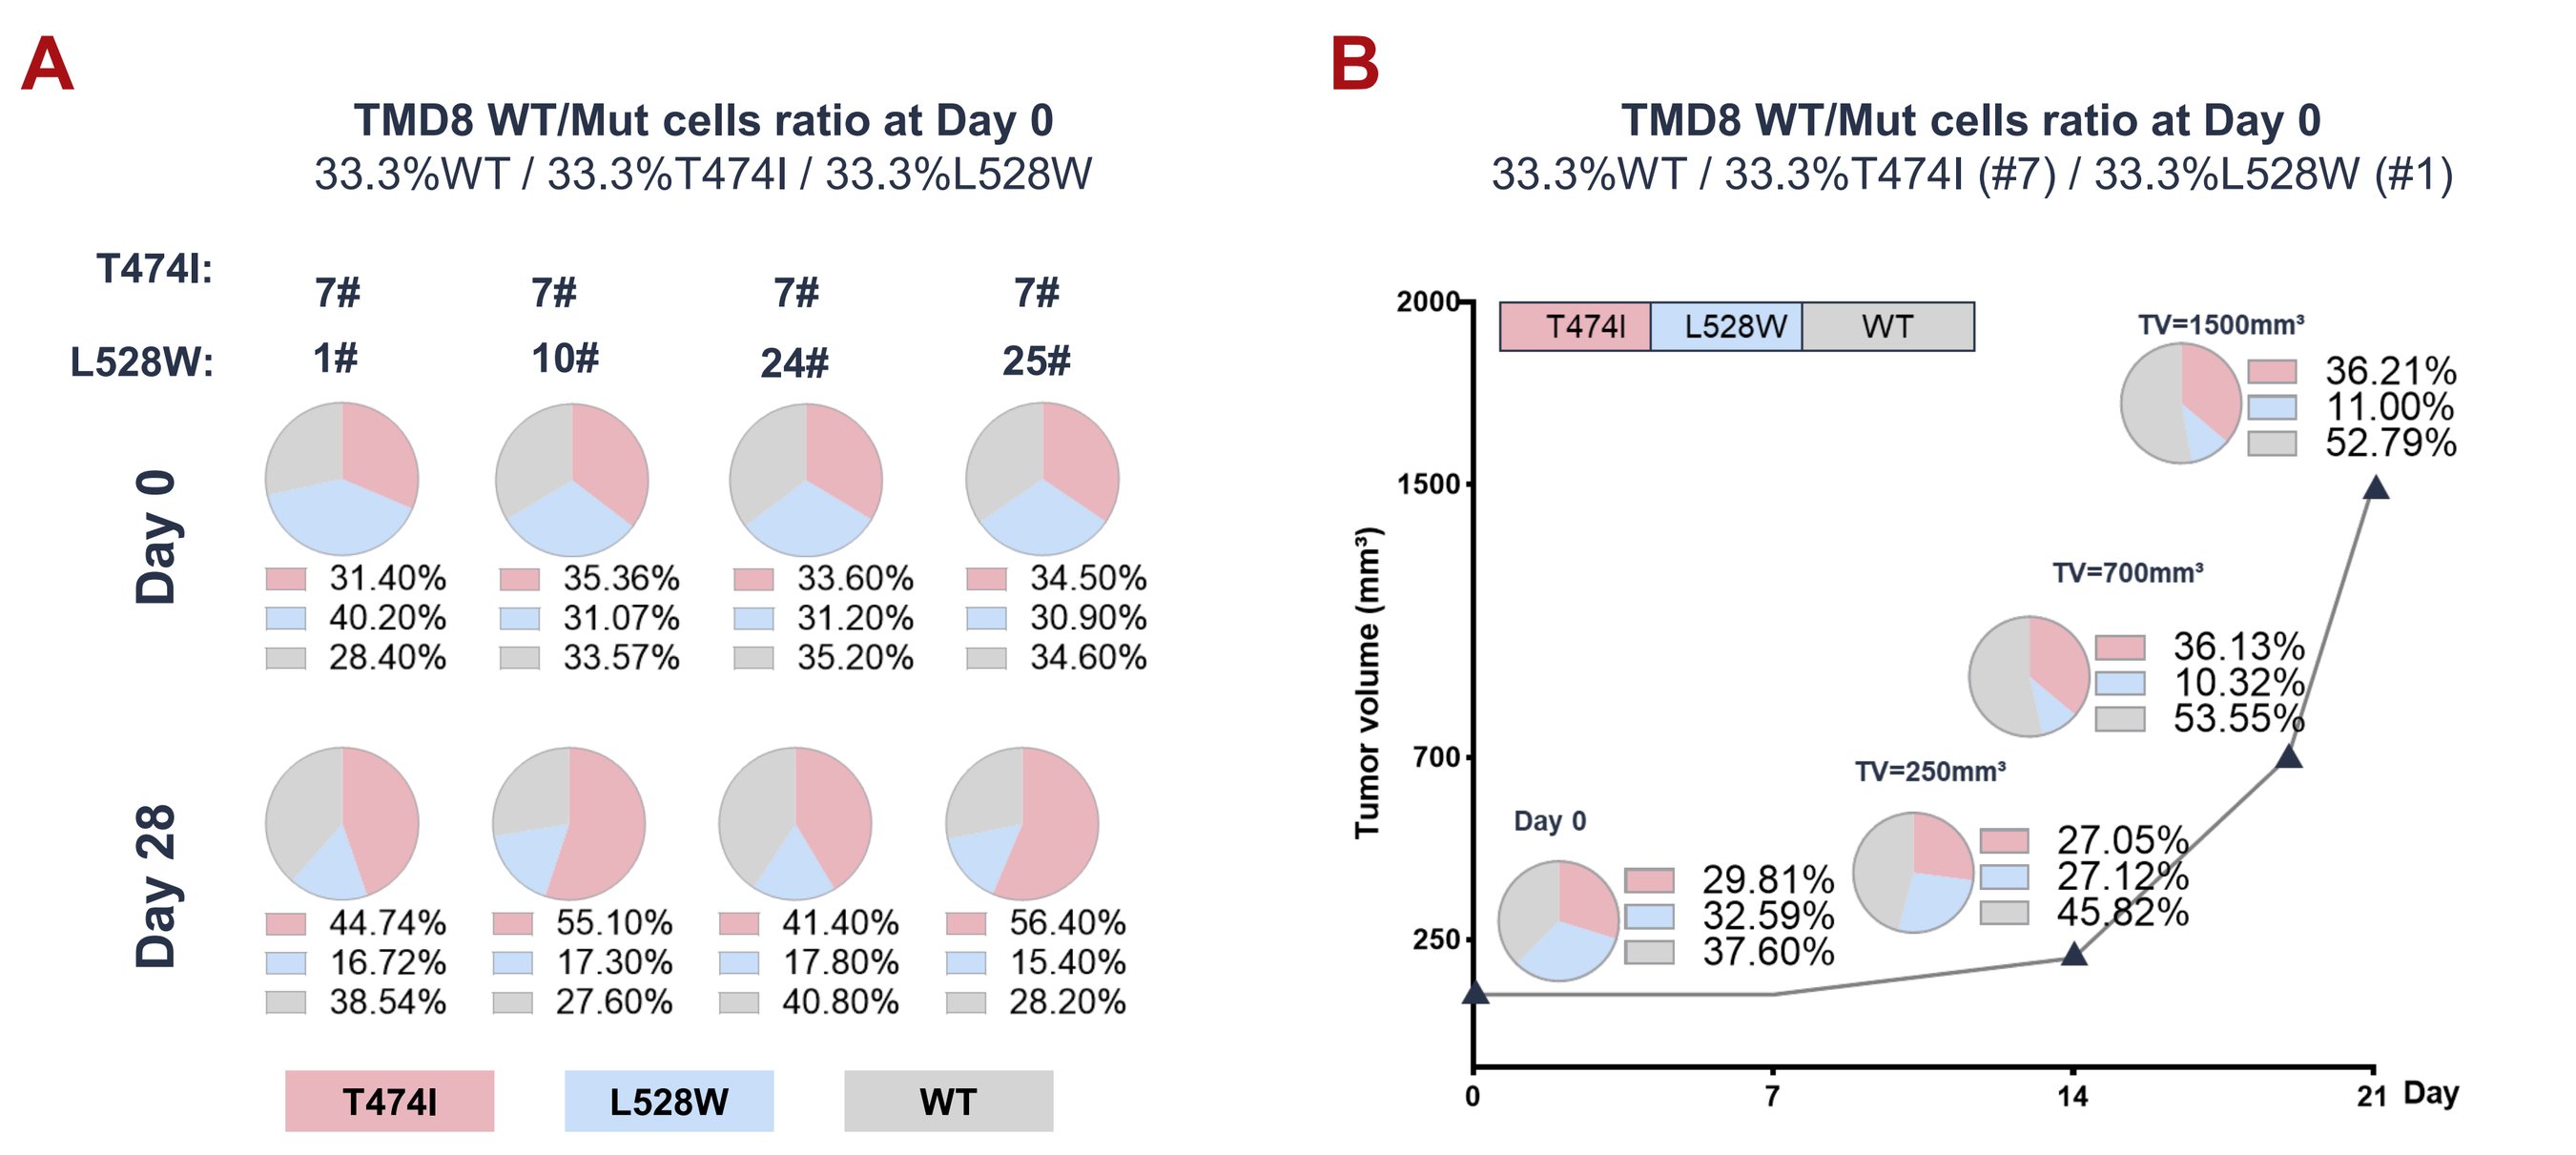
A
TMD8 WT/Mut cells ratio at Day 0
33.3%WT / 33.3%T474I / 33.3%L528W
T474I: 7# 7# 7# 7#
L528W: 1# 10# 24# 25#
Day 0
31.40% 40.20% 28.40%
35.36% 31.07% 33.57%
33.60% 31.20% 35.20%
34.50% 30.90% 34.60%
Day 28
44.74% 16.72% 38.54%
55.10% 17.30% 27.60%
41.40% 17.80% 40.80%
56.40% 15.40% 28.20%
T474I
L528W
WT
B
TMD8 WT/Mut cells ratio at Day 0
33.3%WT / 33.3%T474I (#7) / 33.3%L528W (#1)
Tumor volume (mm³)
Day 0
29.81% 32.59% 37.60%
TV=250mm³
27.05% 27.12% 45.82%
TV=700mm³
36.13% 10.32% 53.55%
TV=1500mm³
36.21% 11.00% 52.79%
Day

The growth advantage of cells harboring BTK-T474I compared to those with BTK-L528W was confirmed through *in vitro* and *in vivo* clonal competition assays, using an independent BTK-T474I knock-in clone (#7) and multiple BTK-L528W knock-in clones. (A) TMD8 cells expressing wildtype BTK, BTK-T474I (KI #7), and various BTK-L528W clones (KI #1, #2, #10, #25) were mixed in equal proportion on day 0 and cultured in medium supplemented with 2% FBS. Cell pellets were collected on day 28 for amplicon sequencing to assess the allele frequency of BTK variants. An increase in variant allele frequency (VAF) of BTK-T474I and a corresponding decrease in VAF for BTK-L528W were observed across four pairwise comparisons. (B) Enhanced outgrowth potential of TMD8 cells carrying BTK-T474I (KI #7) relative to those with BTK-L528W (KI #1) was demonstrated in *in vivo* clonal competition assay.

Figure 5. Enhanced cell cycle progression in TMD8 BTK-T474I than TMD8 BTK-L528W cells revealed through proteomics analysis

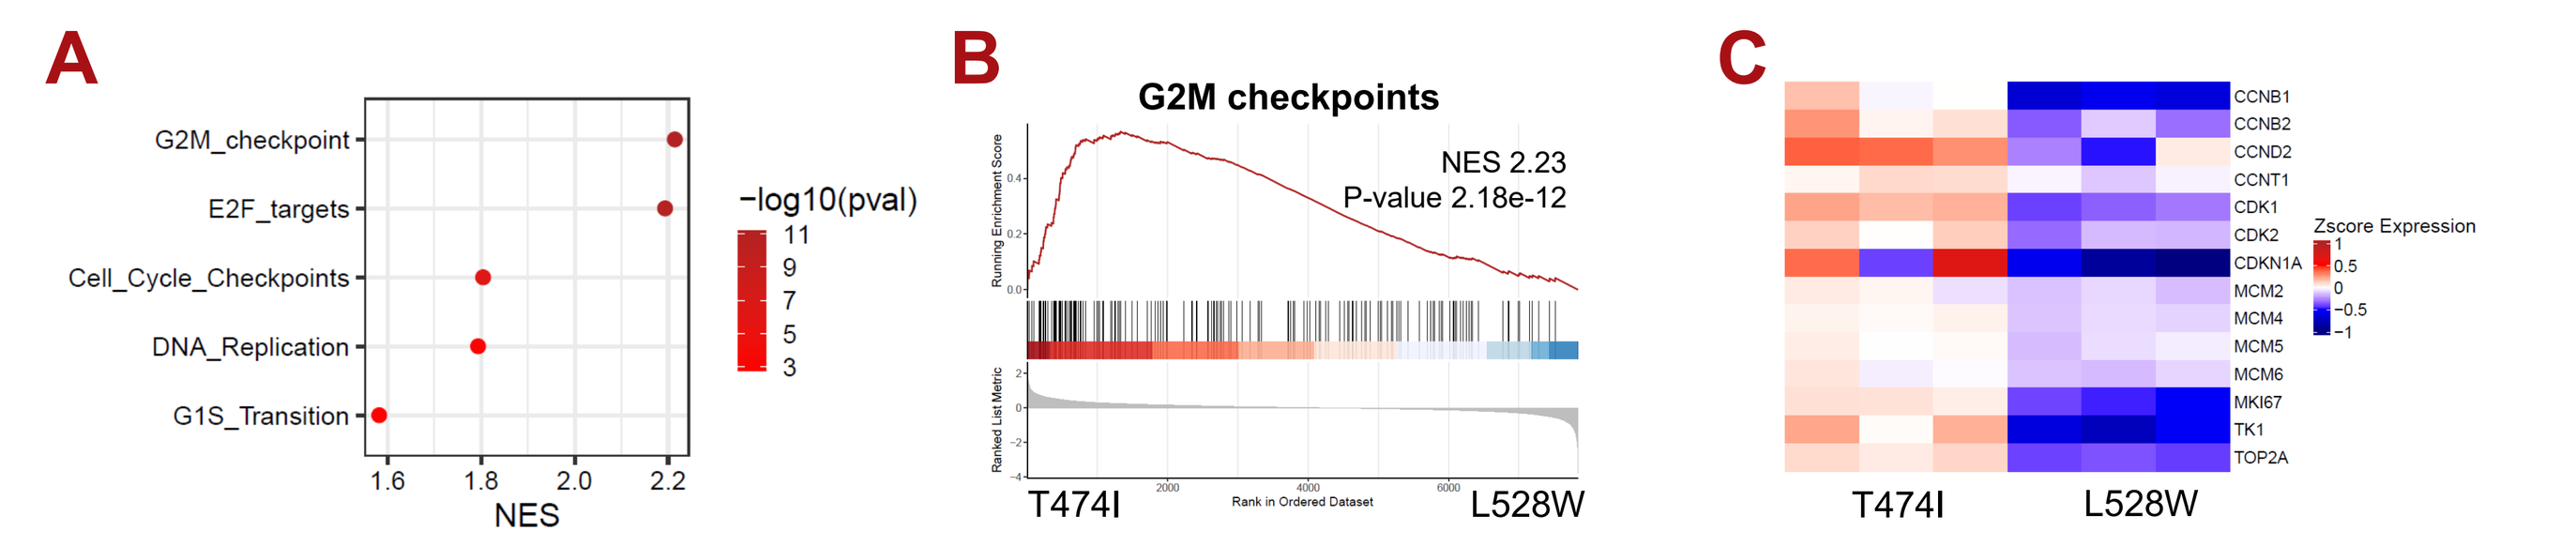
A
G2M_checkpoint
E2F_targets
Cell_Cycle_Checkpoints
DNA_Replication
G1S_Transition
−log10(pval)
NES
B
G2M checkpoints
NES 2.23
P-value 2.18e-12
T474I
L528W
C
CCNB1
CCNB2
CCND2
CCNT1
CDK1
CDK2
CDKN1A
MCM2
MCM4
MCM5
MCM6
MKI67
TK1
TOP2A
Zscore Expression
T474I
L528W

TMD8 cells expressing BTK-T474I (ENU clone), BTK-L528W (KI #1) were cultured in 2% of FBS for 14 days and subjected to proteomics analysis. GSEA top up-regulated pathways in TMD8 BTK-T474I vs BTK-L528W cells (A) and GSEA of G2M checkpoints (B) showed that the differentially expressed proteins were mainly enriched in cell cycle related pathways. (C) Heatmap illustrated the expression level of cell cycle associated proteins in TMD8 BTK-T474I and BTK-L528W cells.

Figure 6. TMD8-BKT T474I showed stronger BCR signal than BTK-L528W cells by phospho-proteomics analysis

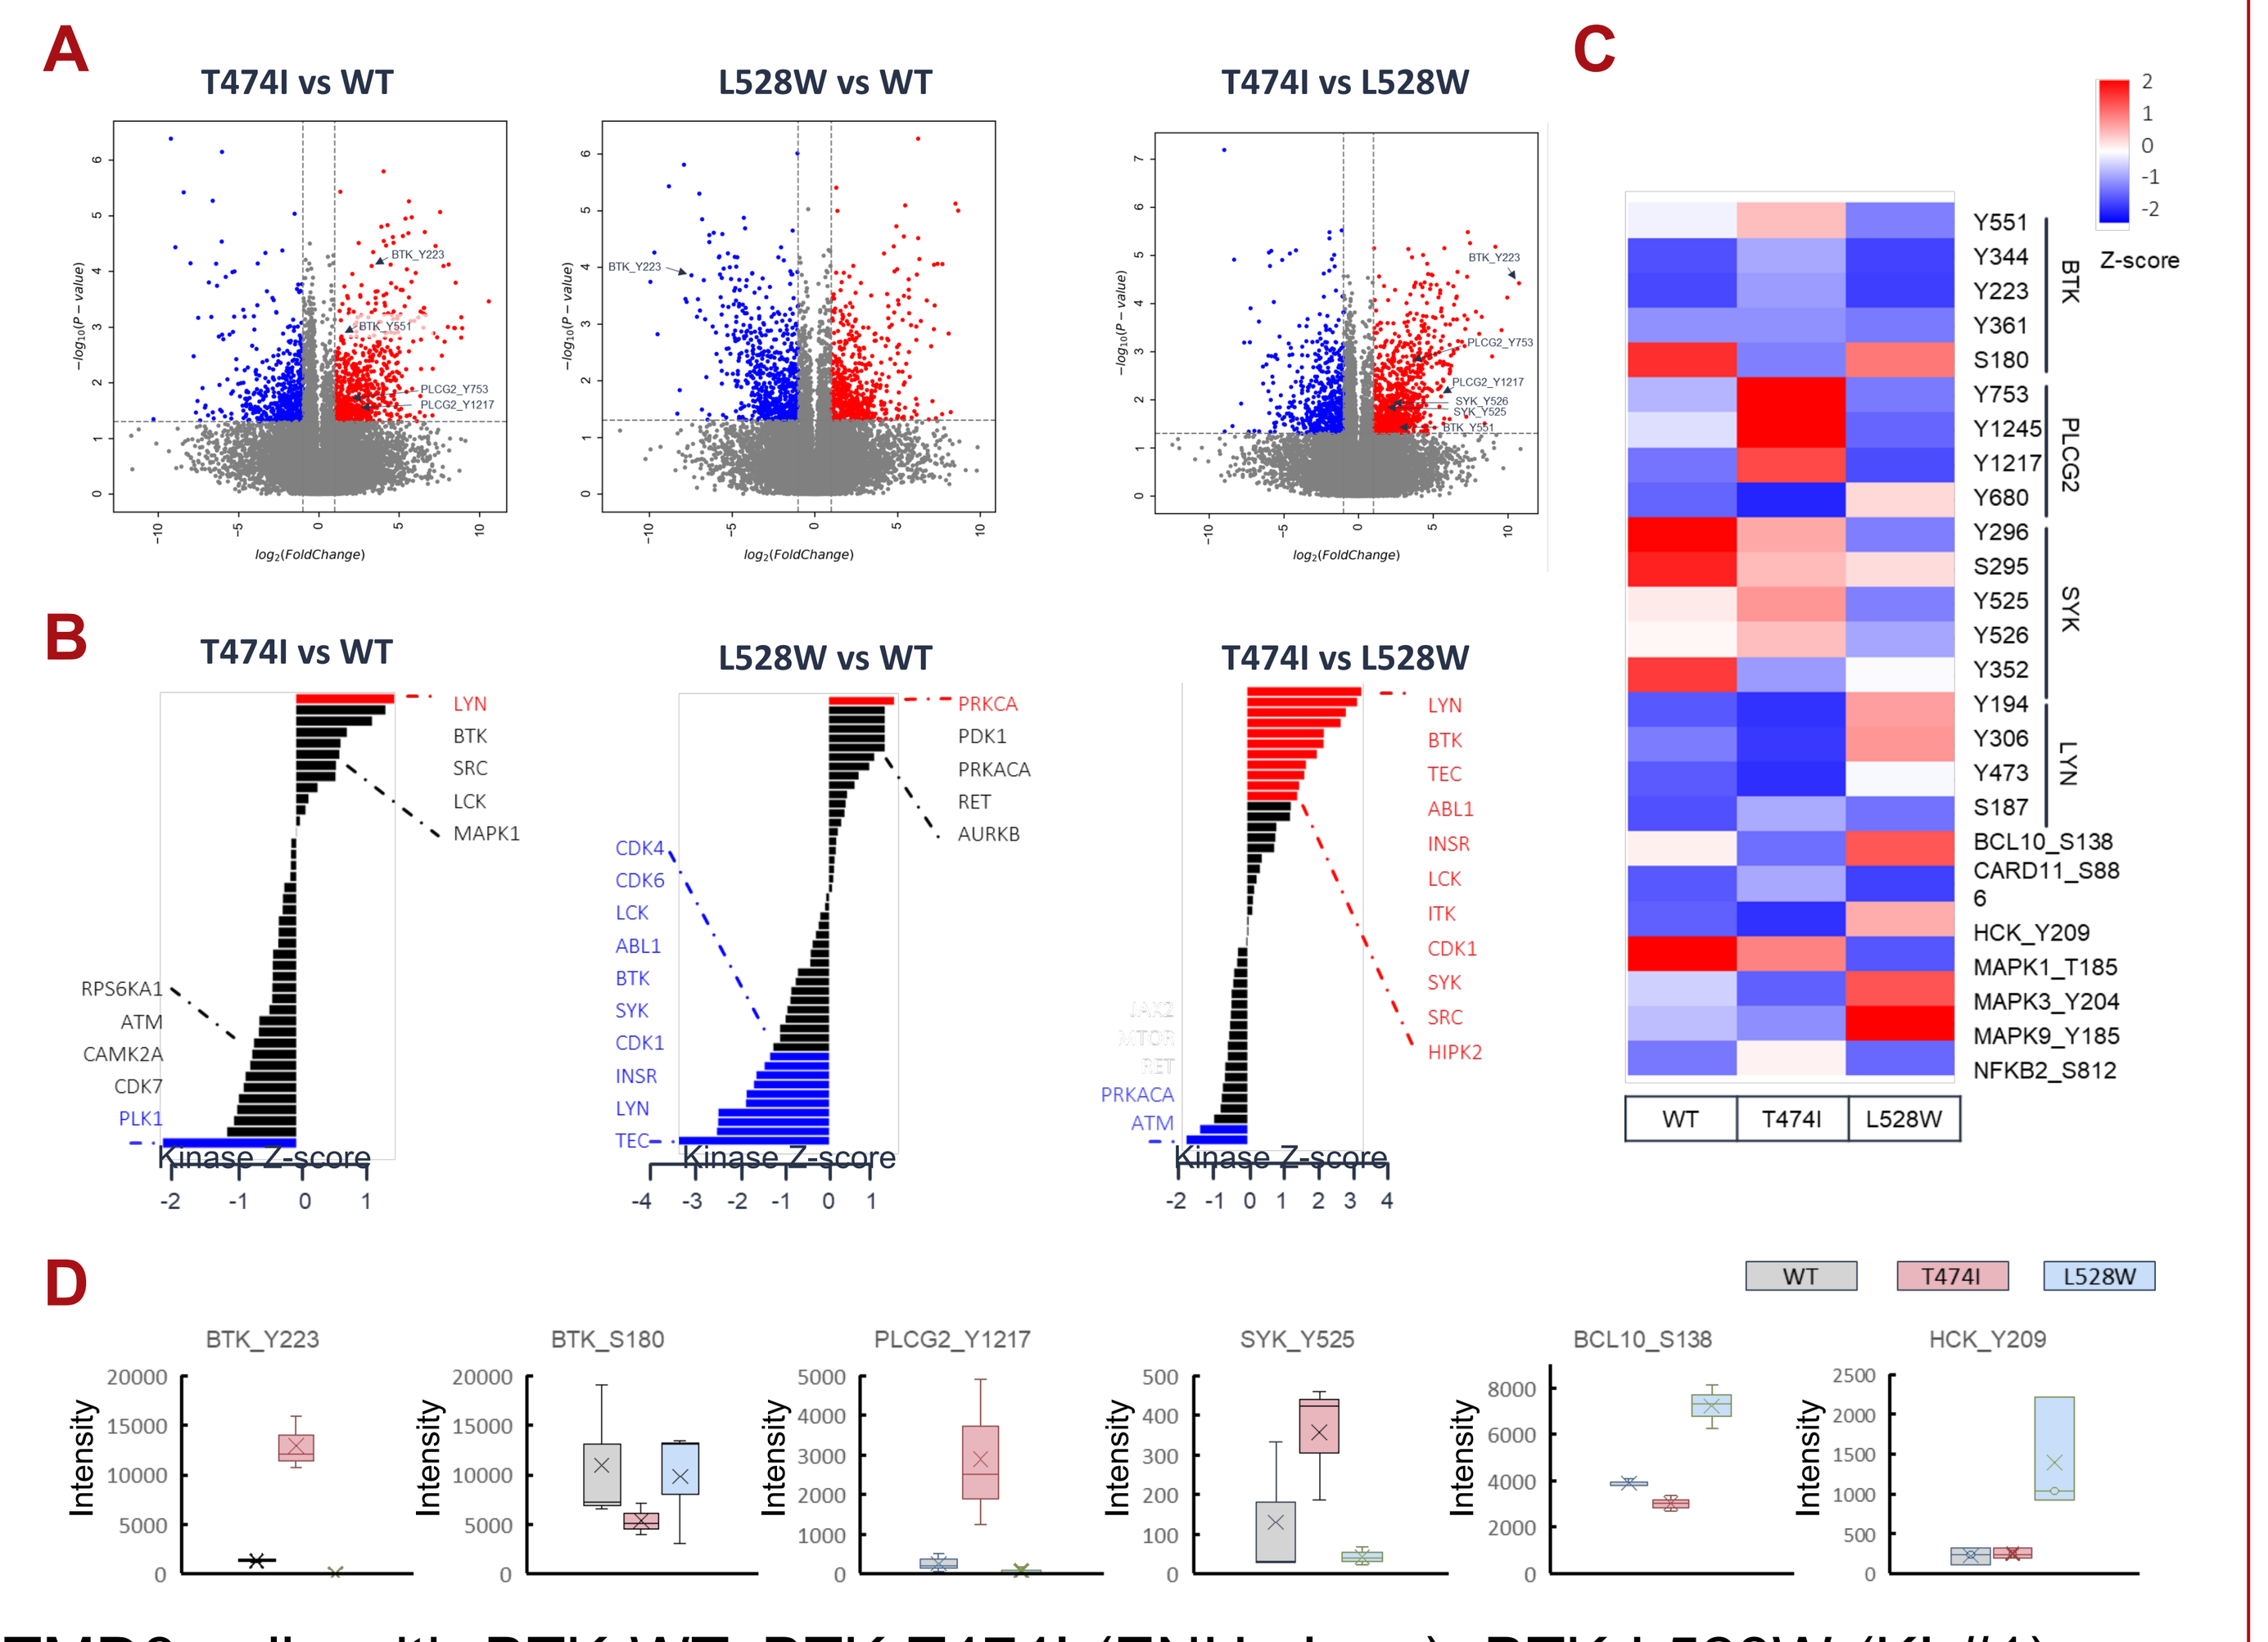
A
T474I vs WT
L528W vs WT
T474I vs L528W
B
T474I vs WT
LYN
BTK
SRC
LCK
MAPK1
RPS6KA1
ATM
CAMK2A
CDK7
PLK1
Kinase Z-score
L528W vs WT
PRKCA
PDK1
PRKACA
RET
AURKB
CDK4
CDK6
LCK
ABL1
BTK
SYK
CDK1
INSR
LYN
TEC
T474I vs L528W
LYN
BTK
TEC
ABL1
INSR
LCK
ITK
CDK1
SYK
SRC
HIPK2
PRKACA
ATM
C
Z-score
BTK
Y551
Y344
Y223
Y361
S180
PLCG2
Y753
Y1245
Y1217
Y680
SYK
Y296
S295
Y525
Y526
Y352
LYN
Y194
Y306
Y473
S187
BCL10_S138
CARD11_S886
HCK_Y209
MAPK1_T185
MAPK3_Y204
MAPK9_Y185
NFKB2_S812
WT
T474I
L528W
D
BTK_Y223
BTK_S180
PLCG2_Y1217
SYK_Y525
BCL10_S138
HCK_Y209
Intensity
WT
T474I
L528W

TMD8 cells with BTK WT, BTK-T474I (ENU clone), BTK-L528W (KI #1) were treated with 10 µg/ml anti-IgM and then subjected to phospho-proteomics analysis. (A) Volcano plots showed differentially phosphorylation sites between groups. Fold change of phosphorylation level for BTK, PLCγ2, SYK are labelled. (B) Kinase substrate enrichment analysis predicted kinase activity difference between groups by calculating hypo/hyper phosphorylated substrates. Significantly active (red) and inactive (blue) kinase between groups are highlighted. (C) Heatmap illustrated the phosphorylation status of key components of BCR pathway and its downstream effectors at indicated residues upon anti-IgM stimulation among groups. (D) Phosphorylation levels of proteins in BCR signalosome among groups. Elevated kinase activity for proteins within BCR pathway, including BTK, SYK, and PLCγ2, was observed in TMD8 BTK-T474I cells compared to BTK-L528W cells. Upregulation of phosphorylation sites indicative of enhanced kinase activity, along with decrease in phosphorylation sites that negatively regulate kinase activity (BTK S180), was evident in TMD8 BTK-T474I relative to BTK-L528W cells.

Reference: 1. N Engl J Med 2013;369:32-42; 2. N Engl J Med. 2023 Jan 26;388(4):319-332; 3. N Engl J Med. 2014 Jun 12;370(24):2286-94; 4. N Engl J Med. 2023 Jul 6;389(1):33-44; 5. N Engl J Med. 2022 Feb 24;386(8):735-743;6. Blood (2023) 142 (Supplement 1): 326.

AACR Annual Meeting 2025 April 25-30, 2025. Chicago, Illinois

AACR 2025