EFFICACY AND SAFETY DATA FROM A PHASE 1/2 TRIAL OF TISLELIZUMAB IN CHINESE PATIENTS WITH NON-SMALL CELL LUNG CANCER

Qing Zhou[1], Jun Zhao[2], Jie Wang[3], Qingyuan Zhang[4], Hongming Pan[5], Lin Shen[2], Ying Yuan[6], Yuxian Bai[4], Tianshu Liu[7], Yujuan Gao[8], Yanjun Li[8], Juan Zhang[8], Yi-Long Wu[1]

[1]Guangdong Lung Cancer Institute, Guangdong Provincial People's Hospital and Guangdong Academy of Medical Sciences, Guangzhou, China; [2]Beijing Cancer Hospital, Beijing, China; [3]Tumor Hospital of Chinese Medical Science Institute, Beijing, China; [4]Harbin Medical University Cancer Hospital, Harbin, China; [5]Sir Run Run Shaw Hospital, Zhejiang University, School of Medicine, Hangzhou, China; [6]The Second Affiliated Hospital Zhejiang University School of Medicine, Hangzhou, China; [7]Zhongshan Hospital Fudan University, Shanghai, China; [8]BeiGene (Beijing) Co., Ltd., Beijing, China

RATIONALE TRIAL PROGRAM

North America Conference on Lung Cancer
October 16-17, 2020, Virtual Congress

BACKGROUND

- The programmed cell death protein-1/programmed death-ligand 1 (PD-1/PD-L1) axis plays a central role in suppressing antitumor immunity; dysregulation of the axis can be used by cancer cells to evade the immune system[1]
 - Monoclonal antibodies against PD-1 have demonstrated antitumor activity in a multitude of tumor types[2]
- Tislelizumab, an anti-PD-1 antibody, was engineered to minimize binding to FcγR on macrophages to abrogate antibody-dependent phagocytosis, a potential mechanism of T-cell clearance and resistance to anti-PD-1 therapy[3,4]
- Tislelizumab 200 mg every 3 weeks (Q3W) has been approved in China for patients with previously treated relapsed/refractory classical Hodgkin lymphoma and previously treated locally advanced or metastatic urothelial carcinoma with PD-L1 tumor cell (TC) expression ≥10%
 - Two supplemental new drug applications were accepted for review of tislelizumab plus chemotherapy as first-line treatment for advanced squamous (April 2020) and nonsquamous (June 2020) non-small cell lung cancer (NSCLC)
- This phase 1/2 dose-verification/indication-expansion study (BGB-A317-102; NCT04068519) was the first trial of tislelizumab monotherapy for patients with advanced solid tumors, including NSCLC, conducted in China[5]
 - At data cutoff (01 December 2018), patients with NSCLC (n=56) had a confirmed objective response rate (ORR) of 18% (95% CI: 8.9, 30.4) and responses were seen in patients regardless of PD-L1 TC expression
 - Median overall survival (OS) was not reached with a median study follow-up of 9 months for patients with NSCLC
- Here, we present updated clinical data with longer follow-up time from patients with advanced NSCLC who received tislelizumab during the BGB-A317-102 study

METHODS

Overall Design and Study Objectives

- A full description of the design, patient population, and treatment administration for this study is presented in the primary publication[5]
- Briefly, adult patients (aged ≥18 years) with histologically or cytologically confirmed advanced/metastatic disease, who experienced progression or intolerability to treatment since their last standard antitumor treatment (or had no standard treatment or refused standard therapy), received tislelizumab 200 mg administered intravenously Q3W until there was no evidence of continued clinical benefit, or until unacceptable toxicity or withdrawal of consent
 - During phase 2 (indication-expansion), PD-L1 expression and *EGFR* mutation status (for those with unknown *EGFR* mutation status) were prospectively tested at a central laboratory; patients with known *EGFR* mutations or *ALK* rearrangements were ineligible, but these requirements were not applicable to phase 1 (dose-verification)
- Disease assessment by radiographic imaging (enhanced CT or MRI) was performed approximately every 9 weeks during the first 12 months and approximately every 12 weeks thereafter according to Response Evaluation Criteria in Solid Tumors v1.1
- Adverse events (AEs) were graded and recorded throughout the study according to National Cancer Institute Common Terminology Criteria for Adverse Events v.4.03
- PD-L1 expression on TC membranes was assessed by the central laboratory using the VENTANA PD-L1 (SP263) assay

RESULTS

Demographics and Baseline Disease Characteristics of Patients With NSCLC

- As of the data cutoff of 31 May 2020, 56 patients with NSCLC were enrolled in phase 1 and phase 2 of the BGB-A317-102 study (nonsquamous, n=31 [55%]; squamous, n=25 [45%])
 - Eight patients remained on treatment; reasons for discontinuation included disease progression (n=36; 64.3%), consent withdrawal (n=4; 7.1%), death (n=3; 5.4%), AEs (n=3; 5.4%), and other (n=2; 3.6%)
- In the 56 patients with NSCLC, the median age was 58 years, 59% were current/former smokers, and 64% had received ≥2 lines of prior systemic therapy (Table 1)
 - Median duration of study follow-up was 19.6 months

Table 1: Demographics and Baseline Characteristics

		Patients With NSCLC[a] (N=56)
Median age, years (range)		58 (26, 72)
Sex, n (%)	Male	40 (71.4)
	Female	16 (28.6)
ECOG status, n (%)	0	14 (25.0)
	1	42 (75.0)
Smoking status, n (%)	Current/former	33 (58.9)
	Never	23 (41.1)
Histological type, n (%)	Nonsquamous NSCLC	31 (55.4)
	Squamous NSCLC	25 (44.6)
Prior lines of systemic therapies, n (%)	0	1 (1.8)
	1	19 (33.9)
	2	20 (35.7)
	≥3	16 (28.6)
PD-L1 expression[b]	PD-L1 ≥10%	24 (42.9)
	PD-L1 <10%	31 (55.4)
	Unknown	1 (1.8)

[a]One patient had an *EGFR* mutation, four patients had unknown *EGFR* status, and the remaining patients had no *EGFR* mutation; one patient had a known *ALK* rearrangement.
[b]PD-L1 expression represents the percent of tumor cells with PD-L1 membrane staining at any intensity.
Abbreviations: ECOG, Eastern Cooperative Oncology Group; NSCLC, non-small cell lung cancer; PD-L1, programmed death-ligand 1.

Antitumor Activity

- Across the 56 patients with advanced NSCLC, 10 achieved a partial response (ORR=17.9%) (Table 2)
 - Responses were observed in patients with PD-L1 TC expression ≥10% and <10%
- With a median follow-up time of 28.3 months, the median duration of response (n=10) was 30.4 months (95% CI: 8.3, 30.4)

Table 2: Confirmed Best Overall Response by PD-L1 Expression (Safety Analysis Set)

		PD-L1 ≥10%[a] (n=24)	PD-L1 <10% (n=31)	Total (N=56)[b]
Best overall response, n (%)	CR	0	0	0
	PR	4 (16.7)	6 (19.4)	10 (17.9)
	SD	8 (33.3)	12 (38.7)	21 (37.5)
	PD	9 (37.5)	12 (38.7)	21 (37.5)
	Missing	3 (12.5)	1 (3.2)	4 (7.1)
ORR (CR+PR), % (95% CI)		16.7 (4.7, 37.4)	19.4 (7.5, 37.5)	17.9 (8.9, 30.4)
DCR (CR+PR+SD), % (95% CI)		50.0 (29.1, 70.9)	58.1 (39.1, 75.5)	55.4 (41.5, 68.7)
CBR (CR+PR+SD ≥16 weeks), % (95% CI)		45.8 (25.6, 67.2)	54.8 (36.0, 72.7)	51.8 (38.0, 65.3)
Median duration of response, months (95% CI)		21.8 (8.3, 30.4)	NE (11.1, NE)	30.4 (8.3, 30.4)

[a]PD-L1 expression represents the percent of tumor cells with PD-L1 membrane staining at any intensity.
[b]One patient had missing/unknown PD-L1 status.
Abbreviations: CBR, clinical benefit rate; CI, confidence interval; CR, complete response; DCR, disease control rate; NE, not estimable; ORR, objective response rate; PD, progressive disease; PD-L1, programmed death-ligand 1; PR, partial response; SD, stable disease.

- Tumor reduction was observed in both nonsquamous and squamous NSCLC, regardless of PD-L1 expression or number of lines of prior systemic therapy (Figure 1)

Figure 1: Maximum Tumor Reduction in Patients With NSCLC Treated With Tislelizumab

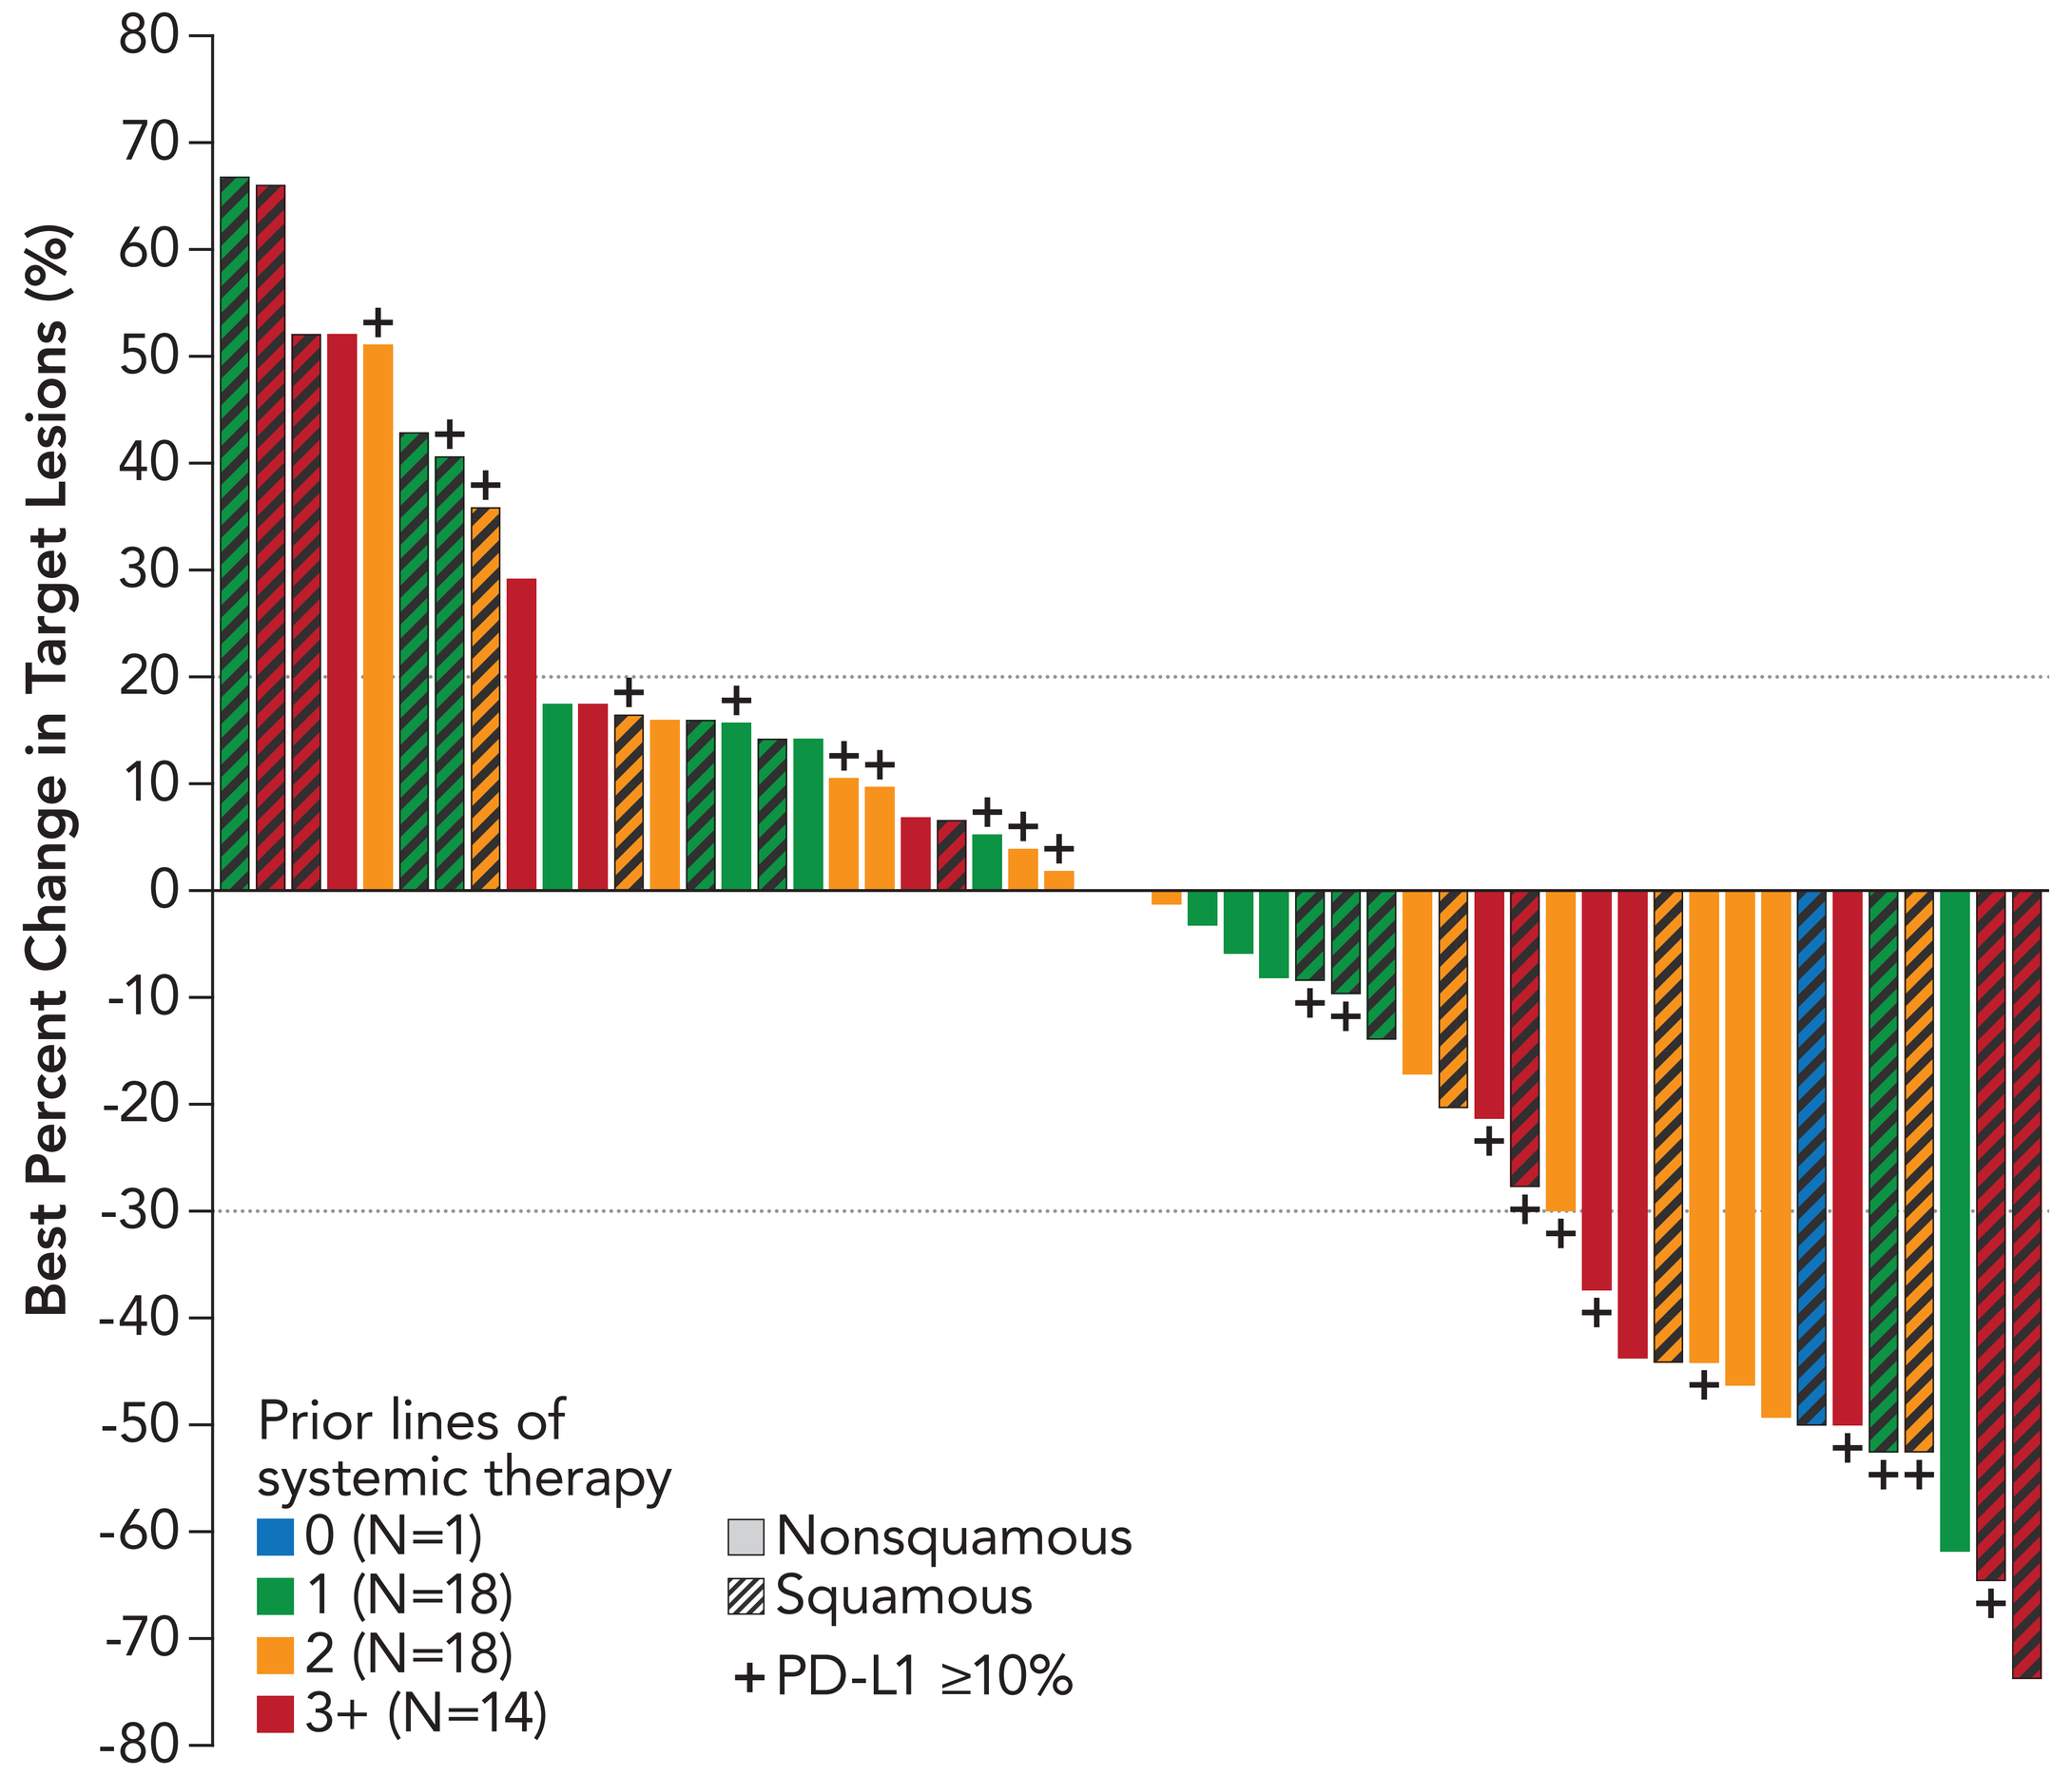
Best Percent Change in Target Lesions (%)
80
70
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
Prior lines of systemic therapy
0 (N=1)
1 (N=18)
2 (N=18)
3+ (N=14)
Nonsquamous
Squamous
+ PD-L1 ≥10%

+Denotes patients with ≥10% of tumor cells with PD-L1 membrane staining at any intensity.
Abbreviations: NSCLC, non-small cell lung cancer; PD-L1, programmed death-ligand 1.

- With a median survival follow-up of 31.4 months (95% CI: 26.8, 34.2), median OS was 22.1 months (95% CI: 10.1, 33.5) among patients with NSCLC (Figure 2)
 - The median survival follow-up as well as 1- and 2-year OS rates are included in Table 3

Figure 2: Overall Survival in Patients With NSCLC

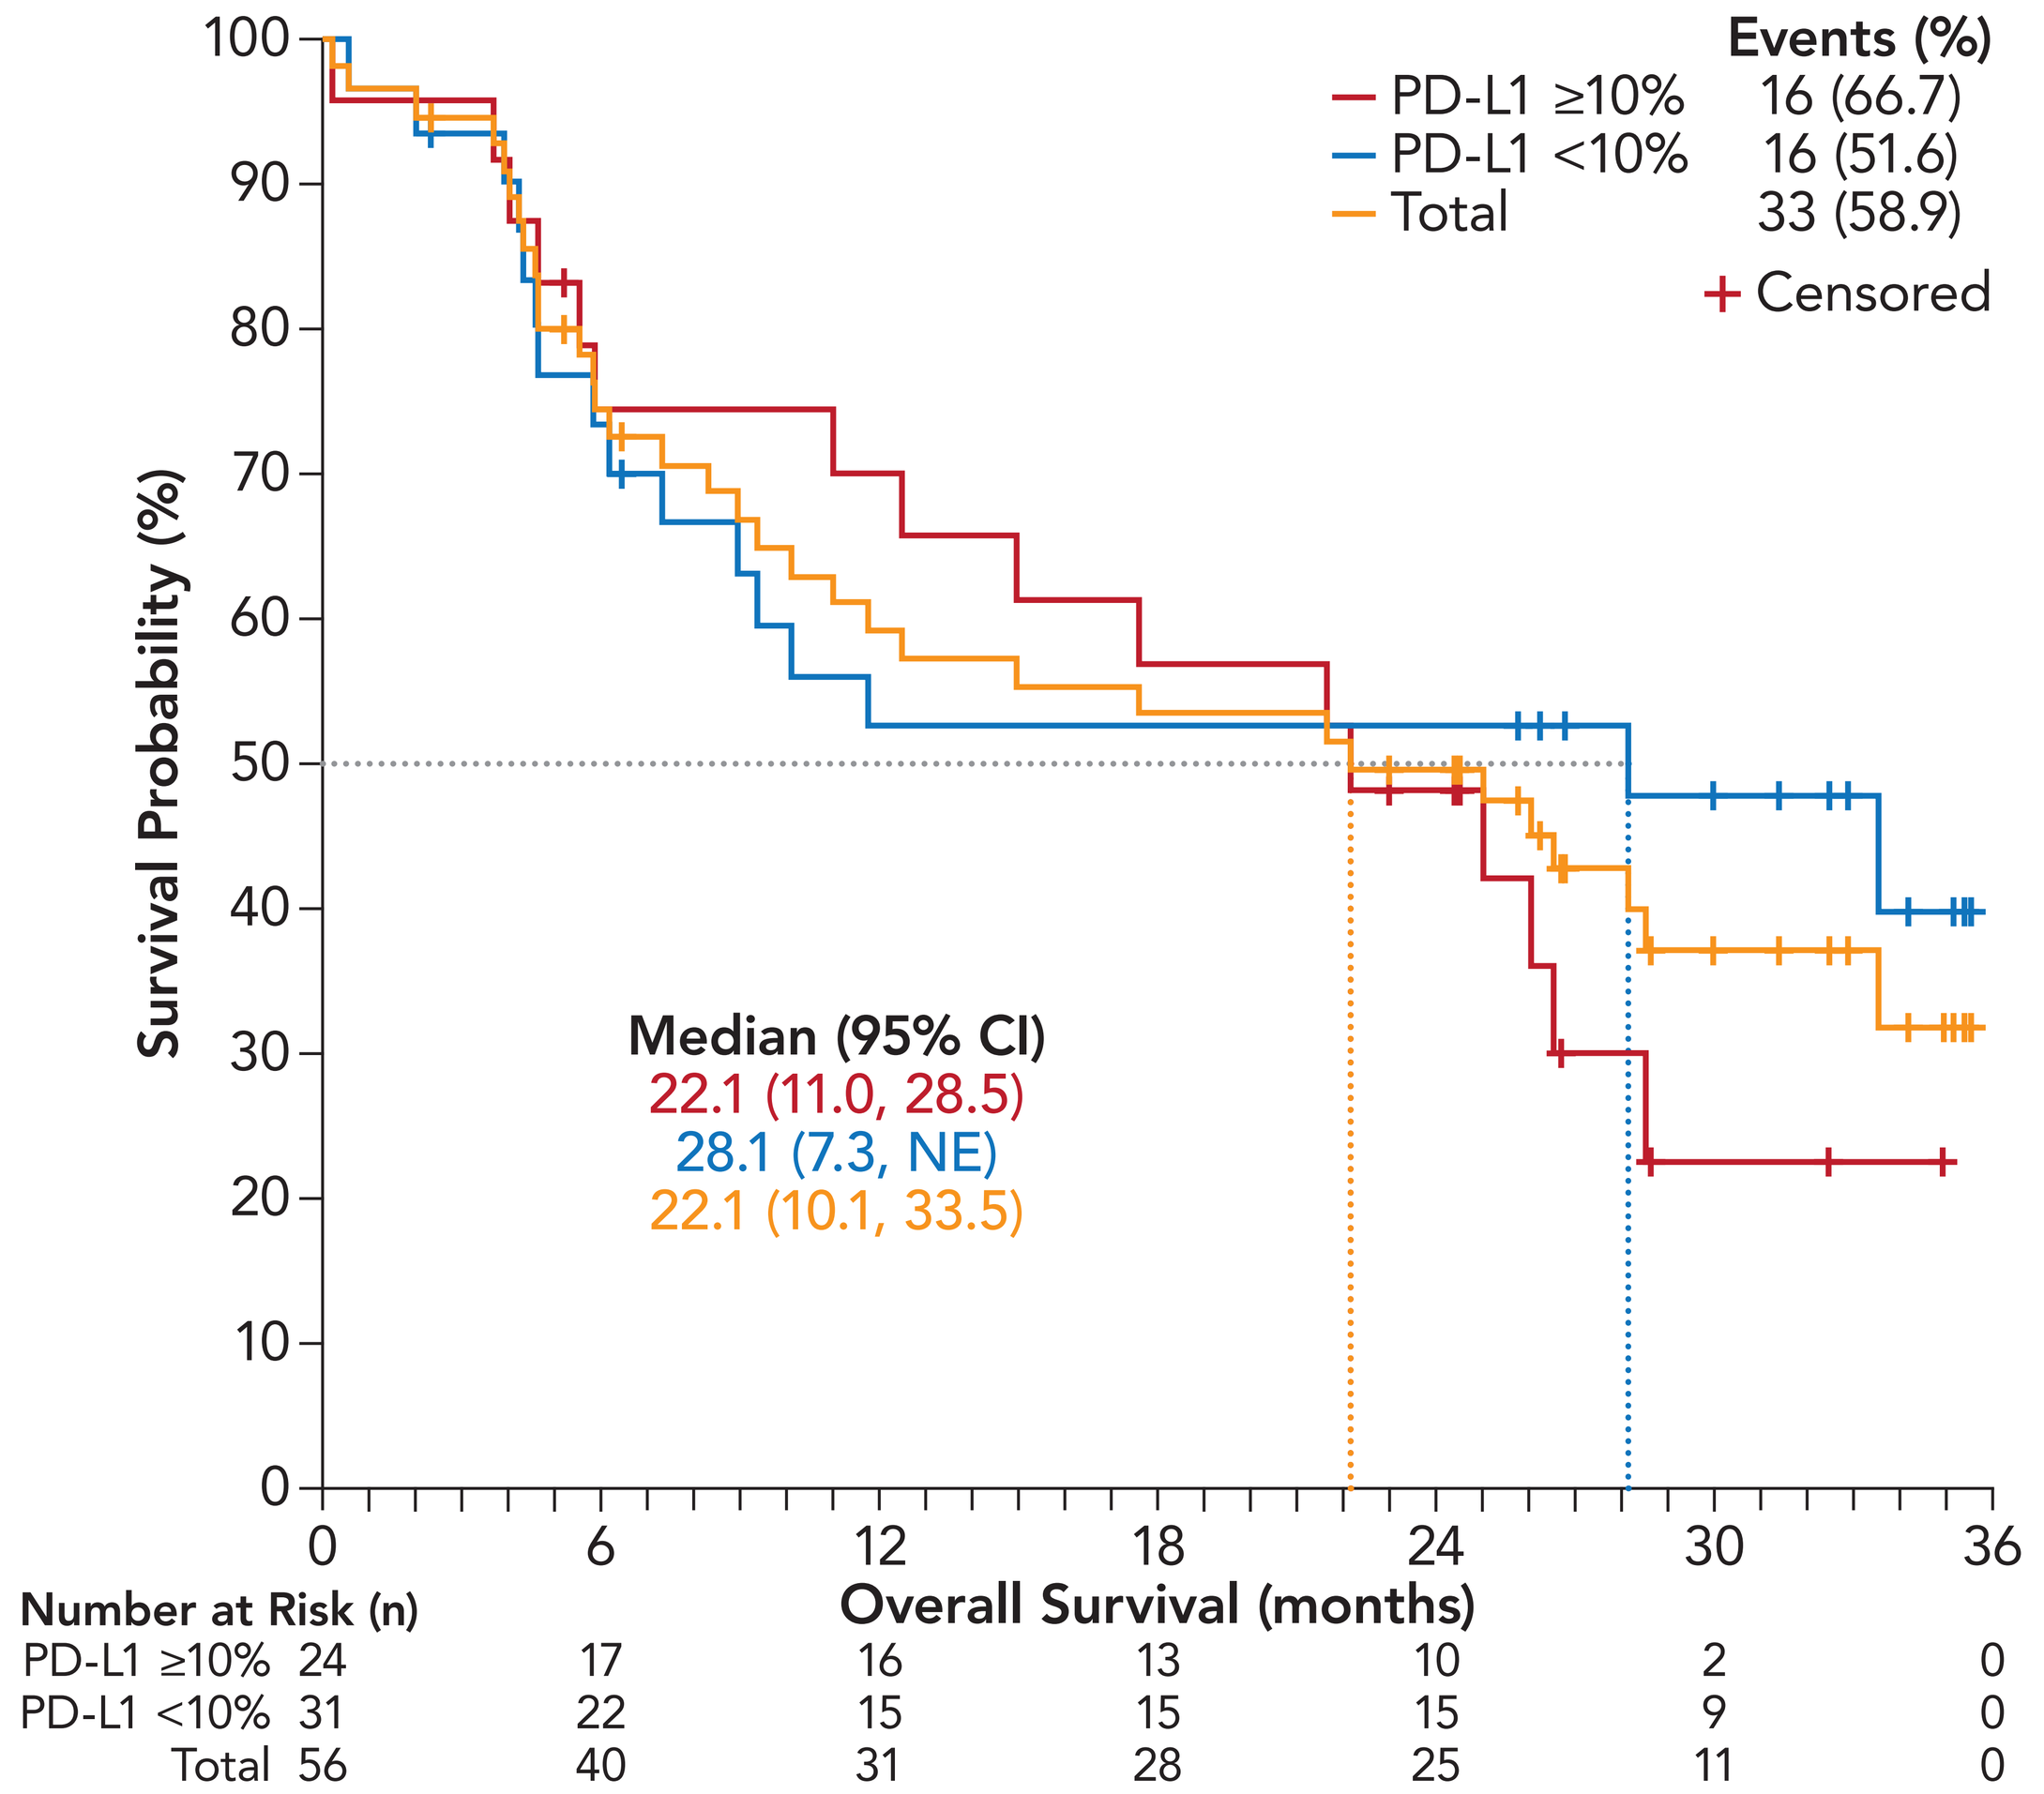
Survival Probability (%)
100
90
80
70
60
50
40
30
20
10
0
Events (%)
PD-L1 ≥10% 16 (66.7)
PD-L1 <10% 16 (51.6)
Total 33 (58.9)
+ Censored
Median (95% CI)
22.1 (11.0, 28.5)
28.1 (7.3, NE)
22.1 (10.1, 33.5)
0 6 12 18 24 30 36
Overall Survival (months)
Number at Risk (n)
PD-L1 ≥10% 24 17 16 13 10 2 0
PD-L1 <10% 31 22 15 15 15 9 0
Total 56 40 31 28 25 11 0

Abbreviations: CI, confidence interval; NE, not estimable; NSCLC, non-small cell lung cancer; PD-L1, programmed death-ligand 1.

Table 3: Overall Survival by PD-L1 Expression

	PD-L1 ≥10%[a] (n=24)	PD-L1 <10% (n=31)	Total (N=56)[b]
Median follow-up, months (95% CI)	28.6 (24.4, 34.9)	32.5 (26.8, 35.1)	31.4 (26.8, 34.2)
1-year overall survival rate, months (95% CI)	0.7 (0.47, 0.85)	0.5 (0.33, 0.69)	0.6 (0.45, 0.71)
2-year overall survival rate, months (95% CI)	0.5 (0.27, 0.67)	0.5 (0.33, 0.69)	0.5 (0.36, 0.62)

[a]PD-L1 expression represents the percent of tumor cells with PD-L1 membrane staining at any intensity.
[b]One patient had missing/unknown PD-L1 status.
Abbreviations: CI, confidence interval; PD-L1, programmed death-ligand 1.

Safety and Tolerability

- Median duration of treatment was 4.3 months
- Treatment-related AEs (TRAEs) across the entire study were mostly of grade ≤2 severity (Table 4)
 - The most common TRAEs were increased aspartate aminotransferase (AST; n=16; 29%) and increased alanine aminotransferase (ALT; n=14; 25%)
 - The only grade ≥3 TRAEs occurring in two or more patients were increased AST (n=3; 5%) and increased ALT (n=2; 4%)
- One patient with lung adenocarcinoma and a history of coronary atherosclerotic heart disease (since 2016) and atrial fibrillation (since 2016) had a TRAE (immune-mediated myocarditis) that led to treatment discontinuation
 - The grade 3 serious AE (immune myocarditis) started on Day 48 and resolved on Day 51; this patient remains on survival follow-up

Table 4: Treatment-Related Adverse Events in ≥5% of Patients With NSCLC

Preferred Term, n (%)[a]	Patients With NSCLC (N=56)	
	Any Grade	Grade ≥3
Any treatment-related adverse event	42 (75.0)	10 (17.9)
AST increased	16 (28.6)	3 (5.4)
ALT increased	14 (25.0)	2 (3.6)
Hypothyroidism	7 (12.5)	0
Rash	7 (12.5)	0
Blood bilirubin increased	6 (10.7)	0
Anemia	5 (8.9)	0
Bilirubin conjugated increased	5 (8.9)	0
Blood TSH increased	5 (8.9)	0
GGT increased	5 (8.9)	1 (1.8)
Nausea	4 (7.1)	0
Pyrexia	4 (7.1)	0
Blood bilirubin unconjugated increased	3 (5.4)	0
Blood TSH decreased	3 (5.4)	0
Diarrhea	3 (5.4)	0
Malaise	3 (5.4)	0
Pruritus	3 (5.4)	0
Vomiting	3 (5.4)	0
Weight decreased	3 (5.4)	0

[a]Patients may have had more than one treatment-related adverse event. Patients with multiple events for a given preferred term were counted only once at the maximum severity for the preferred term.
Abbreviations: ALT, alanine aminotransferase; AST, aspartate aminotransferase; GGT, gamma-glutamyltransferase; NSCLC, non-small cell lung cancer; TSH, thyroid stimulating hormone.

- Sponsor-adjudicated immune-mediated AEs were reported in 15 patients (26.8%) and were generally of low severity (Table 5)
 - Five patients (8.9%) experienced grade ≥3 immune-mediated AEs
 - One patient developed grade ≥3 immune-mediated pneumonitis that led to a dose delay and resolved 19 days after onset; treatment was resumed until the patient discontinued due to disease progression
- No patients with NSCLC reported a TRAE leading to death

Table 5: Sponsor-adjudicated Immune-mediated Adverse Events in Patients With NSCLC

Category, n (%)[a]	Patients With NSCLC (N=56)	
	Any Grade	Grade ≥3
Any immune-mediated adverse event	15 (26.8)	5 (8.9)
Immune-mediated hypothyroidism	5 (8.9)	0
Immune-mediated skin adverse reaction	5 (8.9)	1 (1.8)
Immune-mediated hepatitis	2 (3.6)	2 (3.6)
Immune-mediated pneumonitis	2 (3.6)	1 (1.8)
Immune-mediated hyperthyroidism	1 (1.8)	0
Immune-mediated myocarditis	1 (1.8)	1 (1.8)

[a]Patients may have had more than one immune-mediated adverse event. Patients with multiple events for a given preferred term were counted only once at the maximum severity for the preferred term.
Abbreviation: NSCLC, non-small cell lung cancer.

CONCLUSIONS

- Single-agent tislelizumab was generally well tolerated when administered in patients with advanced NSCLC and observed AEs were consistent with prior reports for tislelizumab monotherapy[5]
- Tislelizumab demonstrated antitumor activity in patients with advanced NSCLC
 - With a median survival follow-up of 31.4 months, median OS was 22.1 months among patients with NSCLC
 - The ORR was 17.9%, and responses were observed regardless of PD-L1 expression
- A phase 3 study of tislelizumab as treatment for second-line NSCLC (RATIONALE 303; NCT03358875) is ongoing

REFERENCES

1. Mahoney KM, Freeman GJ, McDermott DF. The next immune-checkpoint inhibitors: PD-1/PD-L1 blockade in melanoma. *Clin Ther.* 2015;37(4):764-782.
2. Naidoo J, Page DB, Li BT, et al. Toxicities of the anti-PD-1 and anti-PD-L1 immune checkpoint antibodies. *Ann Oncol.* 2015;26(12):2375-2391.
3. Dahan R, Sega E, Engelhardt J, Selby M, Korman AJ, Ravetch JV. FcγRs modulate the anti-tumor activity of antibodies targeting the PD-1/PD-L1 axis. *Cancer Cell.* 2015;28(3):285-295.
4. Zhang T, Song X, Xu L, et al. The binding of an anti-PD-1 antibody to FcγRI has a profound impact on its biological functions. *Cancer Immunol Immunother.* 2018;67(7):1079-1090.
5. Shen L, Guo J, Zhang Q, et al. Tislelizumab in Chinese patients with advanced solid tumors: an open-label, non-comparative, phase 1/2 study. *J Immunother Cancer.* 2020;8(1).

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative centers' study staff and study patients, and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. This study was sponsored by BeiGene, Ltd. Writing and editorial assistance was provided by Stephan Lindsey, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL), and funded by the study sponsor.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com