COST-EFFECTIVENESS OF ZANUBRUTINIB VERSUS IBRUTINIB IN ADULT PATIENTS WITH WALDENSTRÖM MACROGLOBULINEMIA

Jorge J. Castillo[1], Keri Yang[2], Rongzhe Liu[3], Yu Wang[4], Aileen Cohen[2], Todd Zimmerman[2], Qian Zhao[3], Xin Gao[3], Boxiong Tang[2]

[1]Dana-Farber Cancer Institute, Boston, MA; [2]BeiGene USA, San Mateo, CA; [3]OPEN Health, Bethesda, MD; [4]BeiGene (Shanghai) Co., Ltd

Introduction

- Waldenström macroglobulinemia (WM) is a rare, incurable non-Hodgkin lymphoma
- On August 31, 2021, the United States (US) Food and Drug Administration approved zanubrutinib (Brukinsa®), a Bruton tyrosine kinase inhibitor, for adult patients with WM[1]
- The efficacy of zanubrutinib and ibrutinib was examined in the randomized phase 3 ASPEN trial (NCT03053440) in adult patients with treatment-naïve and relapsed and refractory WM[2]

Objective

- This study aimed to assess the cost-effectiveness of zanubrutinib versus ibrutinib in WM from a US payer perspective

Methods

- A three-state (pre-progression, post-progression, and death) partitioned survival model was used to estimate the life years (LYs), quality-adjusted life years (QALYs), and costs for each treatment over a 20-year lifetime horizon
- Overall survival (OS), progression-free survival (PFS), and time-to-discontinuation (TTD) were based on the ASPEN trial data
- OS, PFS, and TTD curves were fitted using six parametric distributions (exponential, Weibull, Gompertz, gamma, log-logistic, and log-normal) to extrapolate long-term survival outcomes
- Selection of the parametric models for each outcome and treatment was based on assessments of 1) the proportional hazard assumption, 2) goodness-of-fit, and 3) clinical plausibility of extrapolated mean OS and associated hazard patterns (based on literature and US clinical expert input) and the alignment between PFS and TTD
- The OS and PFS curves were applied to determine the proportion of patients in each health state (pre-progression, post-progression, and death). The TTD curve was used to estimate the total drug costs
- Background US mortality was accounted for in the model such that the mortality rates for the modeled population should not be lower than the mortality rates for the US general population
- Costs included drug acquisition, adverse event (AE) management, routine care, and terminal care, reported in 2020 US dollars. Drug wholesale acquisition costs were obtained from RED BOOK Online.[3] Unit costs for the management of AEs and resource use were obtained from the Healthcare Cost and Utilization Project (HCUPnet) database.[4] The terminal care cost was obtained from published literature (Table 1)
- Utilities were based on the ASPEN EQ-5D data and assumptions (Table 1). Incidences of AEs were obtained from the ASPEN trial

Table 1. Key model inputs

Parameter	Value	Reference
Key cost inputs		
Drug acquisition, zanubrutinib	$12,935.00 per 120 80 mg capsules	RED BOOK Online (2020)[3]
Drug acquisition, ibrutinib	$12,966.10 per 28 420 mg tablets	RED BOOK Online (2020)[3]
Routine care, pre-progression (per month)	$3,155.74	Iyengar et al. (2019)[5]
Routine care, post-progression (per month)	$6,490.76	Iyengar et al. (2019)[5]
Terminal care (one-time cost)	$102,517.29	Chastek et al. (2012)[6]
Utility inputs		
Progression-free survival	0.791	ASPEN trial EQ-5D data
Post-progression survival	0.691	Assuming disutility of 0.100 due to progression

- All outcomes were discounted at 3% annually
- Sensitivity analyses were conducted to evaluate the impact of parameter uncertainty or structural uncertainty on model outcomes, including one-way sensitivity analyses (OWSA), probabilistic sensitivity analyses (PSA) with 1000 runs, and scenario analyses

Results

- In the base case analyses using the dependent exponential model for all outcomes over a 20-year time horizon, zanubrutinib led to 0.86 LY and 0.77 QALY gained with an additional total cost of $4,924, leading to an incremental cost-effectiveness ratio (ICER) of $6,419 per QALY gained (**Table 2**)

Table 2. Cost-effectiveness model results

	Zanubrutinib	Ibrutinib	Incremental
Total LYs	10.68	9.82	0.86
Total QALYs	8.25	7.48	0.77
Total costs	$1,475,663	$1,470,739	$ 4,924
Incremental cost per QALY (ICER)			**$6,419**

Abbreviations: LYs, life years; QALYs, quality-adjusted life years

- The additional cost was primarily driven by patients staying on zanubrutinib treatment longer as zanubrutinib has longer time to treatment failure. However, this is partially offset by zanubrutinib's lower monthly drug acquisition, reduced cost of disease management (–$1,665) and other direct costs (–$4,732) compared to ibrutinib **(Figure 1)**

Figure 1. Total discounted costs for zanubrutinib and ibrutinib by cost category

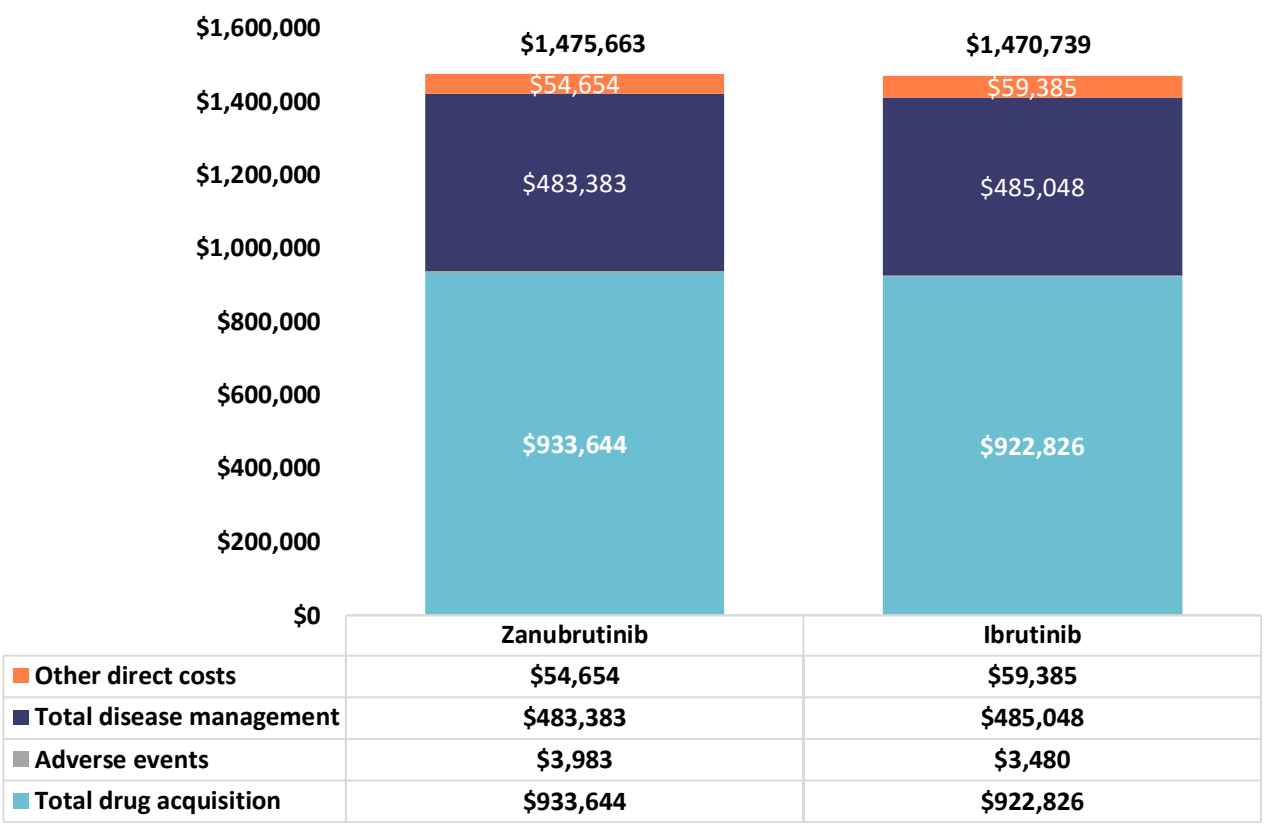
$1,600,000
$1,400,000
$1,200,000
$1,000,000
$800,000
$600,000
$400,000
$200,000
$0
$1,475,663
$54,654
$483,383
$933,644
$1,470,739
$59,385
$485,048
$922,826
Zanubrutinib
Ibrutinib
Other direct costs $54,654 $59,385
Total disease management $483,383 $485,048
Adverse events $3,983 $3,480
Total drug acquisition $933,644 $922,826

Table 3. Cost-effectiveness model, sensitivity analyses

Outcome	Incremental LYs	Incremental QALYs	Incremental costs	ICER
Base case	0.86	0.77	$4,924	$6,419
5 years	0.14	0.13	-$28,238	Dominant
10 years	0.44	0.39	-$18,444	Dominant
15 years	0.71	0.63	$3,886	Dominant
30 years	0.94	0.84	$11,132	$13,205

Abbreviations: LYs, life years; QALYs, quality-adjusted life years; ICER, incremental cost-effectiveness ratio

Conclusion

- The cost-effectiveness analysis demonstrated zanubrutinib is cost-effective compared with ibrutinib for the treatment of patients with WM in the US

- Results of OWSA showed that ICER was most sensitive to drug acquisition costs of both zanubrutinib and ibrutinib. The PSA showed that the mean probabilistic incremental cost was $10,726 with a mean probabilistic incremental QALY of 0.73 at a willingness-to-pay threshold of $100,000 per QALY gained
- Varying the time horizon consistently led to zanubrutinib being dominant (ie, greater QALYs but lower costs) **(Table 3)**

Discussion

- The main limitation of the economic analysis lies with the immaturity of the survival data from the relatively short follow-up in the ASPEN trial, which raises uncertainties as to the results of long-term survival extrapolation. To mitigate the limitation, clinical experts were consulted as to the clinical plausibility of the extrapolated survival
- An updated analysis is warranted upon availability of long-term survival data

References

1. U.S. Food & Drug Administration. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-zanubrutinib-waldenstroms-macroglobulinemia. Accessed September 5, 2021.
2. Tam CS, et al. A randomized phase 3 trial of zanubrutinib vs ibrutinib in symptomatic Waldenström macroglobulinemia: the ASPEN study. Blood. 2020;136(18):2038-2050.
3. IBM Micromedex. RED BOOK Online. https://www.micromedexsolutions.com. Accessed January 12, 2021.
4. Agency for Healthcare Research and Quality. Healthcare Cost and Utilization Project (HCUP). 2015. https://hcupnet.ahrq.gov. Accessed May 9, 2021.
5. Iyengar R, et al. Comparison of healthcare resource utilization and costs for Waldenström's macroglobulinemia (WM) patients treated with ibrutinib or chemoimmunotherapy. Value Health. 2019;22(suppl 1):S76.
6. Chastek B, et al. Health care costs for patients with cancer at the end of life. J Oncol Pract. 2012;8(6):75s-80s.

This study was funded by BeiGene, Ltd
Correspondence: keri.yang@beigene.com

Presented at the AMCP Nexus 2021, Denver, Colorado, October 18–21, 2021