Zanidatamab, an anti-HER2 bispecific antibody, plus chemotherapy with/without tislelizumab as first-line treatment for patients with advanced HER2-positive breast cancer or gastric/gastroesophageal junction adenocarcinoma: A Phase 1B/2 trial-in-progress

Keun-Wook Lee,[1] Young-Hyuck Im,[2] Keun Seok Lee,[3] Jae Yong Cho,[4] Do-Youn Oh,[5] Hyun Cheol Chung,[4] Yee Chao,[6] Li-Yuan Bai,[7] Chia-Jui Yen,[8] In-Ho Kim,[9] Sang Cheul Oh,[10] Chia-Chi Lin,[11] Jin Wang,[12] Xin Wang,[12] Huiyan Li,[12] Yen-Yang Chen,[13] Yoon-Koo Kang[14]

[1]Seoul National University Bundang Hospital, Seoul National University College of Medicine, Seongnam, South Korea; [2]Samsung Medical Center, Sungkyunkwan University, School of Medicine, Seoul, South Korea; [3]National Cancer Center, Goyang, South Korea; [4]Yonsei Cancer Center, Severance Hospital, Yonsei University College of Medicine, Seoul, South Korea; [5]Seoul National University Hospital, Cancer Research Institute, Seoul National University College of Medicine, Seoul, South Korea; [6]Department of Oncology, Taipei Veterans General Hospital, Taipei City, Taiwan; [7]China Medical University Hospital, China Medical University, Taichung, Taiwan; [8]National Cheng Kung University Hospital, Tainan City, Taiwan; [9]The Catholic University of Korea, Seoul St. Mary's Hospital, Seoul, South Korea; [10]Korea University Guro Hospital, Seoul, South Korea; [11]National Taiwan University Hospital, Taipei, Taiwan; [12]BeiGene (Shanghai) Co, Ltd., Shanghai, China; [13]Kaohsiung Chang Gung Memorial Hospital, Kaohsiung City, Taiwan; [14]Asan Medical Centre, University of Uslan College of Medicine, Seoul, South Korea

Poster No. TPS2656

Introduction

- Human epidermal growth factor receptor 2 (HER2) overexpression has been correlated with aggressive tumors characterized by poor therapeutic response to conventional therapies and short survival time[1,2]
- Despite the advances in treatment and outcomes that have been introduced by HER2-targeted agents over the last decade in breast cancer and gastric/gastroesophageal junction adenocarcinoma (GC/GEJC), many patients develop resistance and/or relapse[3,4]
- Zanidatamab (ZW25) is a novel Azymetric™ bispecific antibody that simultaneously binds to two distinct sites on HER2 extracellular domain (ECD) (**Figure 1**)[5]
 - This unique binding configuration drives novel mechanisms of action (**Figure 2**)
- In a Phase 1 dose-escalation and expansion study, single-agent zanidatamab was generally well tolerated and showed antitumor activity in patients with advanced HER2-positive cancers; patients with gastroesophageal cancer and/or breast cancer had overall response rates (ORR) that ranged from 33% to 39%[6-8]
- Combining HER2-targeted agents with chemotherapy has resulted in improved survival and the highly immunogenic nature of HER2 tumors has led to the development of therapies combining anti-HER2 therapies with immune checkpoint blockade[9]
- Tislelizumab was specifically engineered to minimize binding to FcγR on macrophages in order to abrogate antibody-dependent phagocytosis, a mechanism of T-cell clearance and potential resistance to anti PD-1 therapy (**Figure 3**)[10,11]
 - Previous reports indicate that tislelizumab was generally well tolerated and had antitumor activity alone and in combination with chemotherapy in patients with advanced solid tumors[12,13]
- This open label, 2-cohort Phase 1B/2 study (NCT04276493) is designed to evaluate zanidatamab as a first-line therapy with chemotherapy in patients with HER2-positive unresectable locally advanced or metastatic breast cancer, and zanidatamab as a first-line therapy with chemotherapy and with tislelizumab in patients with HER2-positive unresectable locally advanced or metastatic gastric/gastroesophageal junction adenocarcinoma

Figure 1. Structure of zanidatamab (ZW25)[5]

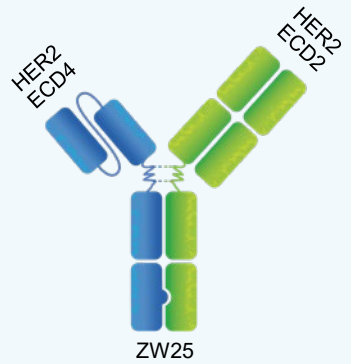

HER2 ECD4
HER2 ECD2
ZW25

- Designed using the Azymetric™ bispecific platform
- Biparatopic — simultaneously binds to two distinct sites on HER2: ECD4 and ECD2
- Unique binding configuration results in multiple mechanisms of action that lead to:[5,14]
 - Improved binding, clustering, and receptor internalization & downregulation
 - Inhibition of ligand-dependent and -independent proliferation
 - Potent activation of antibody-dependent cellular cytotoxicity & phagocytosis
 - Potent complement-dependent cytotoxicity

ECD2, extracellular domain 2; ECD4, extracellular domain 4; HER2, human epidermal growth factor receptor 2; ZW25, zanidatamab

Figure 2. Unique binding configuration of zanidatamab drives novel mechanisms of action: Enhanced tumor cell binding and internalization relative to trastuzumab[5,8]

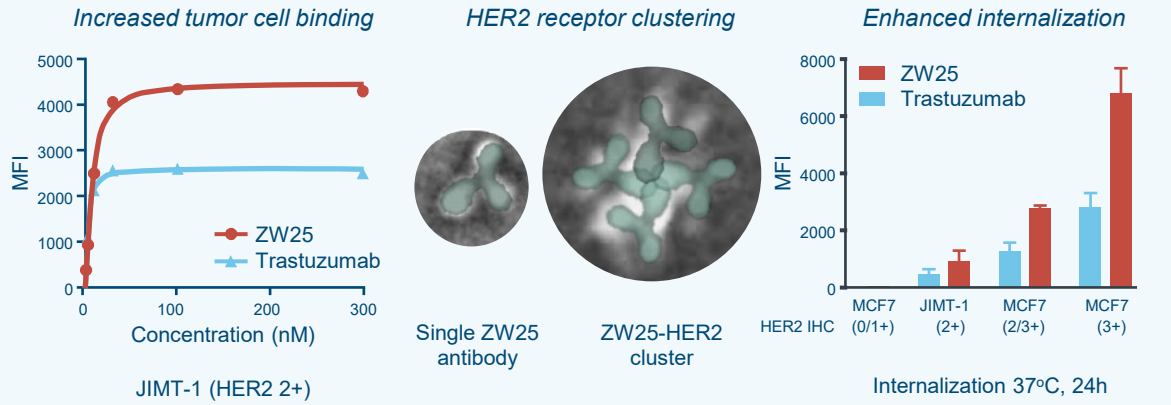

Increased tumor cell binding
HER2 receptor clustering
Enhanced internalization
MFI
5000
4000
3000
2000
1000
0
0
100
200
300
ZW25
Trastuzumab
Concentration (nM)
JIMT-1 (HER2 2+)
Single ZW25 antibody
ZW25-HER2 cluster
8000
6000
4000
2000
0
HER2 IHC
MCF7 (0/1+)
JIMT-1 (2+)
MCF7 (2/3+)
MCF7 (3+)
Internalization 37°C, 24h

h, hour; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; MFI, mean fluorescence intensity; nM, nanomolar; ZW25, zanidatamab

Conclusions

BGB-A317-ZW25 is an **ongoing Phase 1B/2 study** evaluating **zanidatamab** as a **first-line therapy with chemotherapy** in patients with **HER2-positive, unresectable locally advanced** or **metastatic breast cancer**, or **with chemotherapy plus tislelizumab** in patients with **HER2-positive unresectable locally advanced** or **metastatic gastric/gastroesophageal junction adenocarcinoma**

Figure 3. Lack of FcγR binding may prevent macrophage-mediated T-cell clearance[15]

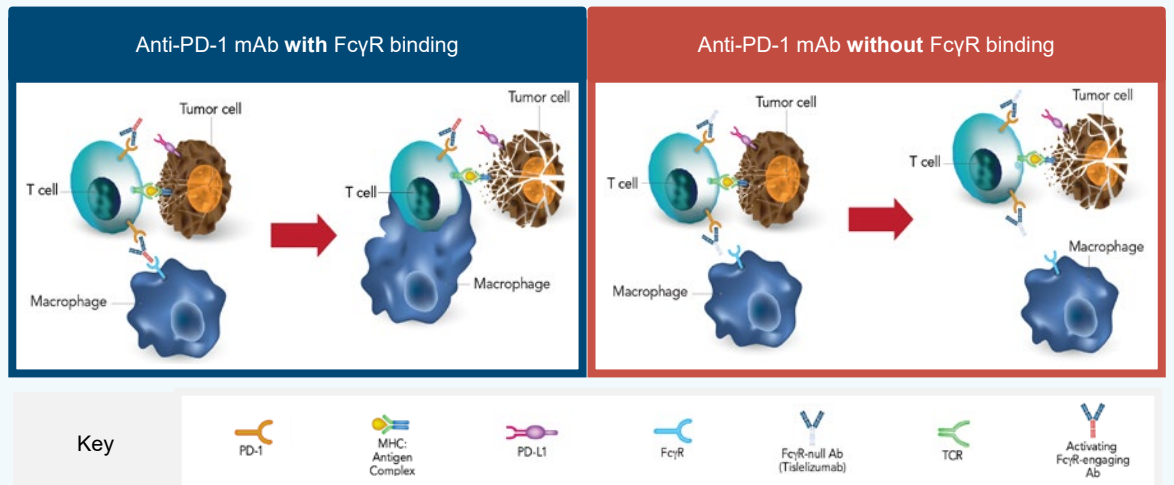

Anti-PD-1 mAb with FcγR binding
Anti-PD-1 mAb without FcγR binding
Tumor cell
T cell
Macrophage
Key
PD-1
MHC: Antigen Complex
PD-L1
FcγR
FcγR-null Ab (Tislelizumab)
TCR
Activating FcγR-engaging Ab

Ab, antibody; FcγR, Fc gamma receptor; mAb, monoclonal antibody; MHC, major histocompatibility complex; PD-1, programmed cell death protein 1; PD-L1, programmed death-ligand 1; TCR, T-cell receptor

Methods

Study design and objectives

- The study design is summarized in **Figure 4**
- Approximately 50 patients across 12 centers in Asian countries will be recruited
- The primary objectives are to assess the safety/tolerability and preliminary antitumor activity (as measured by ORR) of zanidatamab in combination with docetaxel in Cohorts 1a and 1b, and zanidatamab in combination with tislelizumab and CAPOX (a multi-agent chemotherapy regimen consisting of capecitabine and oxaliplatin) in Cohorts 2a and 2b
- Secondary objectives are to further evaluate the preliminary antitumor activity in each cohort as measured by duration of response (DoR), time to response, progression-free survival (PFS), disease control rate (DCR), and overall survival (OS), as well as to characterize the pharmacokinetics and immunogenicity of zanidatamab

Study population

- All eligible patients will be ≥ 18 years, have an Eastern Cooperative Oncology Group performance status score ≤ 1, and ≥ 1 measurable lesion per Response Evaluation Criteria in Solid Tumors (RECSIT) v1.1
- Cohort 1: Female patients with histologically or cytologically confirmed unresectable, locally advanced or recurrent or metastatic HER2-positive breast cancer. Patients will not have received previous systemic anticancer therapy for locally advanced unresectable or metastatic disease
- Cohort 2: Patients with histologically or cytologically confirmed unresectable, locally advanced or recurrent or metastatic HER2-positive GC/GEJC. Patients will not have received previous systemic anticancer therapy for locally advanced unresectable or metastatic disease, or any anti-HER2 treatment
- HER2-positive disease by overexpression and/or gene amplification was determined by investigators or central laboratory, using archival tumor tissue or fresh biopsy sample

Figure 4. Study design and treatment dosing and administration

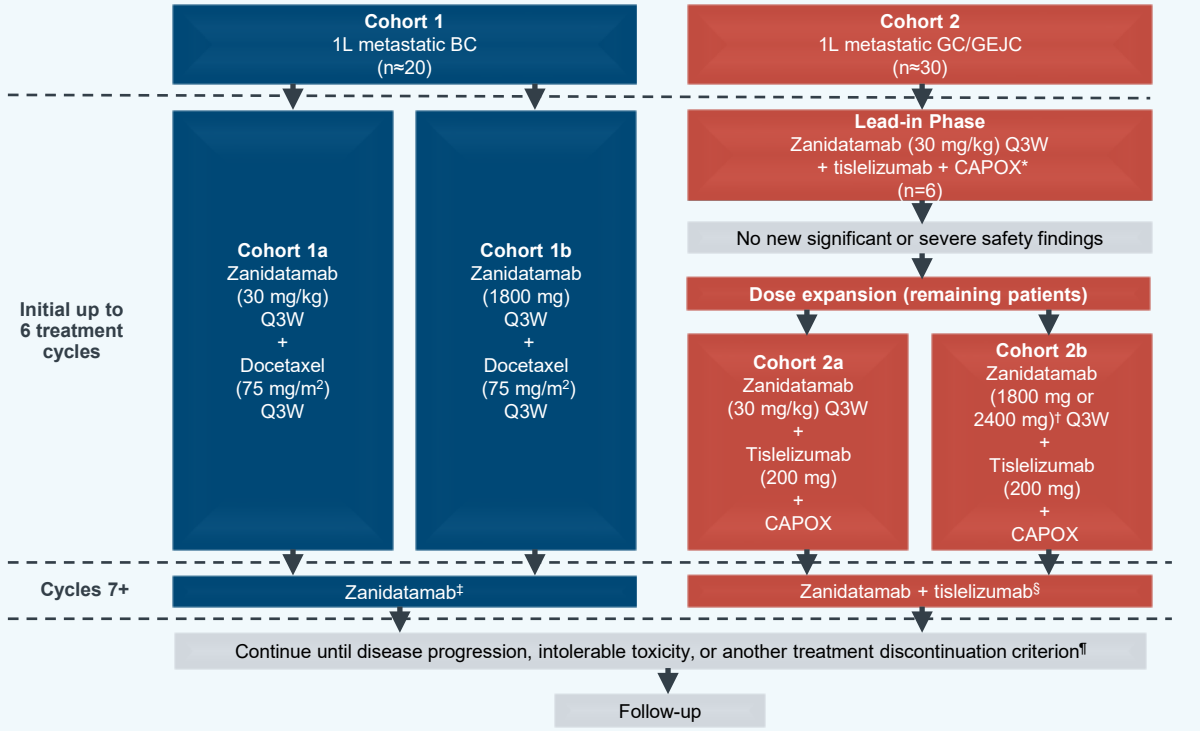

Cohort 1
1L metastatic BC
(n≈20)
Cohort 2
1L metastatic GC/GEJC
(n≈30)
Lead-in Phase
Zanidatamab (30 mg/kg) Q3W
+ tislelizumab + CAPOX*
(n=6)
No new significant or severe safety findings
Dose expansion (remaining patients)
Initial up to 6 treatment cycles
Cohort 1a
Zanidatamab
(30 mg/kg)
Q3W
+
Docetaxel
(75 mg/m²)
Q3W
Cohort 1b
Zanidatamab
(1800 mg)
Q3W
+
Docetaxel
(75 mg/m²)
Q3W
Cohort 2a
Zanidatamab
(30 mg/kg) Q3W
+
Tislelizumab
(200 mg)
+
CAPOX
Cohort 2b
Zanidatamab
(1800 mg or
2400 mg)† Q3W
+
Tislelizumab
(200 mg)
+
CAPOX
Cycles 7+
Zanidatamab‡
Zanidatamab + tislelizumab§
Continue until disease progression, intolerable toxicity, or another treatment discontinuation criterion¶
Follow-up

**CAPOX is a multi-agent chemotherapy regimen consisting of capecitabine and oxaliplatin. †Patients < 70 kg treated with 1800 mg, patients ≥ 70 kg treated with 2400 mg. ‡For Cohort 1, continuation of docetaxel treatment is at the discretion of the investigator after Cycle 6. §For Cohort 2, continuation of capecitabine as maintenance treatment is at the discretion of the investigator after Cycle 6. ¶Treatment beyond initial investigator-assessed RECIST v1.1-defined progression is only permitted if the tislelizumab-treated patient in Cohort 2 potentially has "pseudoprogression" and after discussion with sponsor and re-consent of patient*

1L, first-line; BC, breast cancer; Q3W, every 3 weeks

Treatment

- Treatment dosing and administration is summarized in **Figure 4**
- Maintenance therapy with zanidatamab (Cohort 1) and zanidatamab plus tislelizumab (Cohort 2) will be administered until disease progression, intolerable toxicity, or discontinuation

Study assessments

- Safety/tolerability will be assessed by the type, frequency and severity of adverse events according to the National Cancer Institute Common Terminology Criteria for Adverse Events v5.0 and results from physical examinations, vital signs, electrocardiogram, and laboratory tests
- ORR, the primary endpoint, will be assessed by investigator (INV) per RECIST v1.1
- Tumor assessments will occur at baseline, every 6 weeks (± 7 days) for 36 weeks, and every 12 weeks (± 7 days) thereafter until disease progression, withdrawal of consent, death, or the start of a new anticancer therapy
- Secondary efficacy endpoints including DoR, time to response, PFS, and DCR will be assessed by INV per RECIST v1.1
- Two-sided 95% confidence intervals will be calculated for ORR and DCR in each cohort, and the Kaplan-Meier method will be used to estimate DoR, time to response, PFS, and OS

References

1. Wang J and Xu B. Signal Transduct Target Ther 2019;4:34
2. Van Cutsem E, et al. Lancet 2016;388:2654–64
3. Ayoub NM, Al-Shami KM and Yaghan RJ. NM, et al. Breast Cancer 2019;11:53-69
4. Mitani S and Kawakami H. Cancers 2020;12;400
5. Weisser N, et al. AACR 2017. April 1-5. Washington DC, USA
6. Oh D-Y, et al. Presented at ESMO Asia 2019
7. Meric-Bernstam F, et al. Presented at ASCO-GI 2021
8. Meric-Bernstam F, et al. Presented as ESMO 2019
9. Pernas S and Tolaney SM. Ther Adv Med Oncol 2019;11:1758835919833519
10. Zhang T, et al. Cancer Immunol Immunother 2018;67:1079–90
11. Dahan R, et al. Cancer Cell 2015;28:285–95
12. Deva S, et al. Ann Oncol 2018;29
13. Bai Y, et al. J Clin Oncol 2019;37:11
14. Weisser N, et al. Presented at AACR Annual Meeting 2021
15. Figure taken from D-Y Oh, et al. Presented at ASCO 2020

Acknowledgements

The study was sponsored by BeiGene, Ltd. Medical writing support for the development of this poster, under the direction of the authors, was provided by Shannon Galgani, MSci, of Ashfield MedComms, an Ashfield Health company, and funded by BeiGene, Ltd.

Author contact details: imdoctor@snu.ac.kr (Keun-Wook Lee)

Abstract 331839, Poster Discussion Session, Developmental Therapeutics—Immunotherapy
2021 American Society of Clinical Oncology: ASCO 2021 Virtual Conference, June 4–8, 2021