UPDATED RESULTS OF THE PARP1/2 INHIBITOR PAMIPARIB IN COMBINATION WITH LOW-DOSE TEMOZOLOMIDE IN PATIENTS WITH LOCALLY ADVANCED OR METASTATIC SOLID TUMORS

Agostina Stradella[1], Melissa Johnson[2], Sanjay Goel[3], Sreenivasa R. Chandana[4], Matthew D. Galsky[5], Emiliano Calvo[6], Victor Moreno[7], Haeseong Park[8], Tobias Arkenau[9], Andrés Cervantes[10], Lorena Fariñas-Madrid[11], Linda Mileshkin[12], Ruth Plummer[13], Jeff Evans[14], Lisa Horvath[15], Amy Prawira[16], Robert Pelham[17], Song Mu[18], Claudia Andreu-Vieyra[19], and Minal Barve[20]

[1]Institut Catala, Barcelona, Spain; [2]Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN, USA; [3]Medical Oncology, Montefiore Medical Center, Bronx, NY, USA; [4]START Midwest, Grand Rapids, Michigan; [5]Hematology and Medical Oncology, Icahn School of Medicine at Mount Sinai, New York, NY, USA; [6]START Madrid - CIOCC, Centro Integral Oncológico Clara Campal, Hospital Madrid Norte Sanchinarro, Madrid, Spain; [7]START Madrid - FJD, Hospital Fundación Jiménez Díaz, Madrid, Spain; [8]Medical Oncology, Washington University, St. Louis, MO, USA; [9]Sarah Cannon Research Institute, London, UK; [10]Medical Oncology, Biomedical Research Institute, Incliva, Valencia, Spain; [11]Vall d'Hebron, Barcelona, Spain; [12]Peter MacCallum Cancer Centre, Melbourne, Australia; [13]Sir Bobby Robson Cancer Trials Research Centre, Northern Institute for Cancer Research, Newcastle University, Newcastle UK; [14]Beatson West of Scotland, Glasgow, UK; [15]Chris O'Brien Lifehouse, Sydney, Australia; [16]St Vincent's Hospital, Sydney, Australia; [17]CDx and Biomarker Development, BeiGene USA, Inc., San Mateo, CA, USA; [18]Clinical Pharmacology, BeiGene USA, Inc., San Mateo, CA, USA; [19]Clinical Science, BeiGene USA, Inc., San Mateo, CA, USA; [20]Medical Oncology, Mary Crowley Cancer Research, Dallas, TX, USA

POSTER 451PD
European Society for Medical Oncology Congress
27 September–1 October, 2019, Barcelona, Spain

BACKGROUND

- Poly (ADP-ribose) polymerase (PARP) proteins play a key role in the repair of single strand (ss) and double strand (ds) DNA breaks[1,2]
- Normal cells repair DNA breaks using the base excision repair (BER) and the homologous recombination (HR) pathways; cancer cells that are HR deficient (HRD$^+$) are unable to repair dsDNA breaks
- PARP inhibition impairs DNA repair and traps PARP proteins on damaged DNA, resulting in cytotoxicity that is exacerbated in HRD$^+$ cells (synthetic lethality)[3–9]
- Pamiparib is an investigational PARP1/2 inhibitor that has demonstrated brain penetration and PARP-DNA complex–trapping capabilities in preclinical studies and promising antitumor activity in ovarian cancer[10]
- Temozolomide (TMZ) methylates DNA bases, creating adducts that are repaired by the BER pathway in a PARP-dependent fashion; PARP inhibition results in the accumulation of highly cytotoxic adducts, leading to cell death (Figure 1)
- Low-dose (ld) TMZ (11.5 mg/m^2 to 69 mg/m^2) has been shown to cause DNA damage in tissues and peripheral blood cells in preclinical *in vivo* studies[11]
- We hypothesize that DNA damage caused by ld TMZ may synergize with PARP inhibition, and that this synergy will result in increased antitumor activity
- This phase 1b study (NCT03150810) evaluates the safety and preliminary antitumor activity of pamiparib in combination with ld TMZ in patients with locally advanced and metastatic tumors. We have previously shown that pamiparib in combination with ld TMZ was generally well-tolerated and has antitumor activity.[12] Here, we present updated results that include the recommended dose (RP2D) and schedule as well as data from patients enrolled in the gastric/gastroesophageal junction (G/GEJ) cancer and the extensive-stage small cell lung cancer (ES SCLC) expansion cohorts

Figure 1: Rationale for Combining Pamiparib With Low-Dose Temozolomide

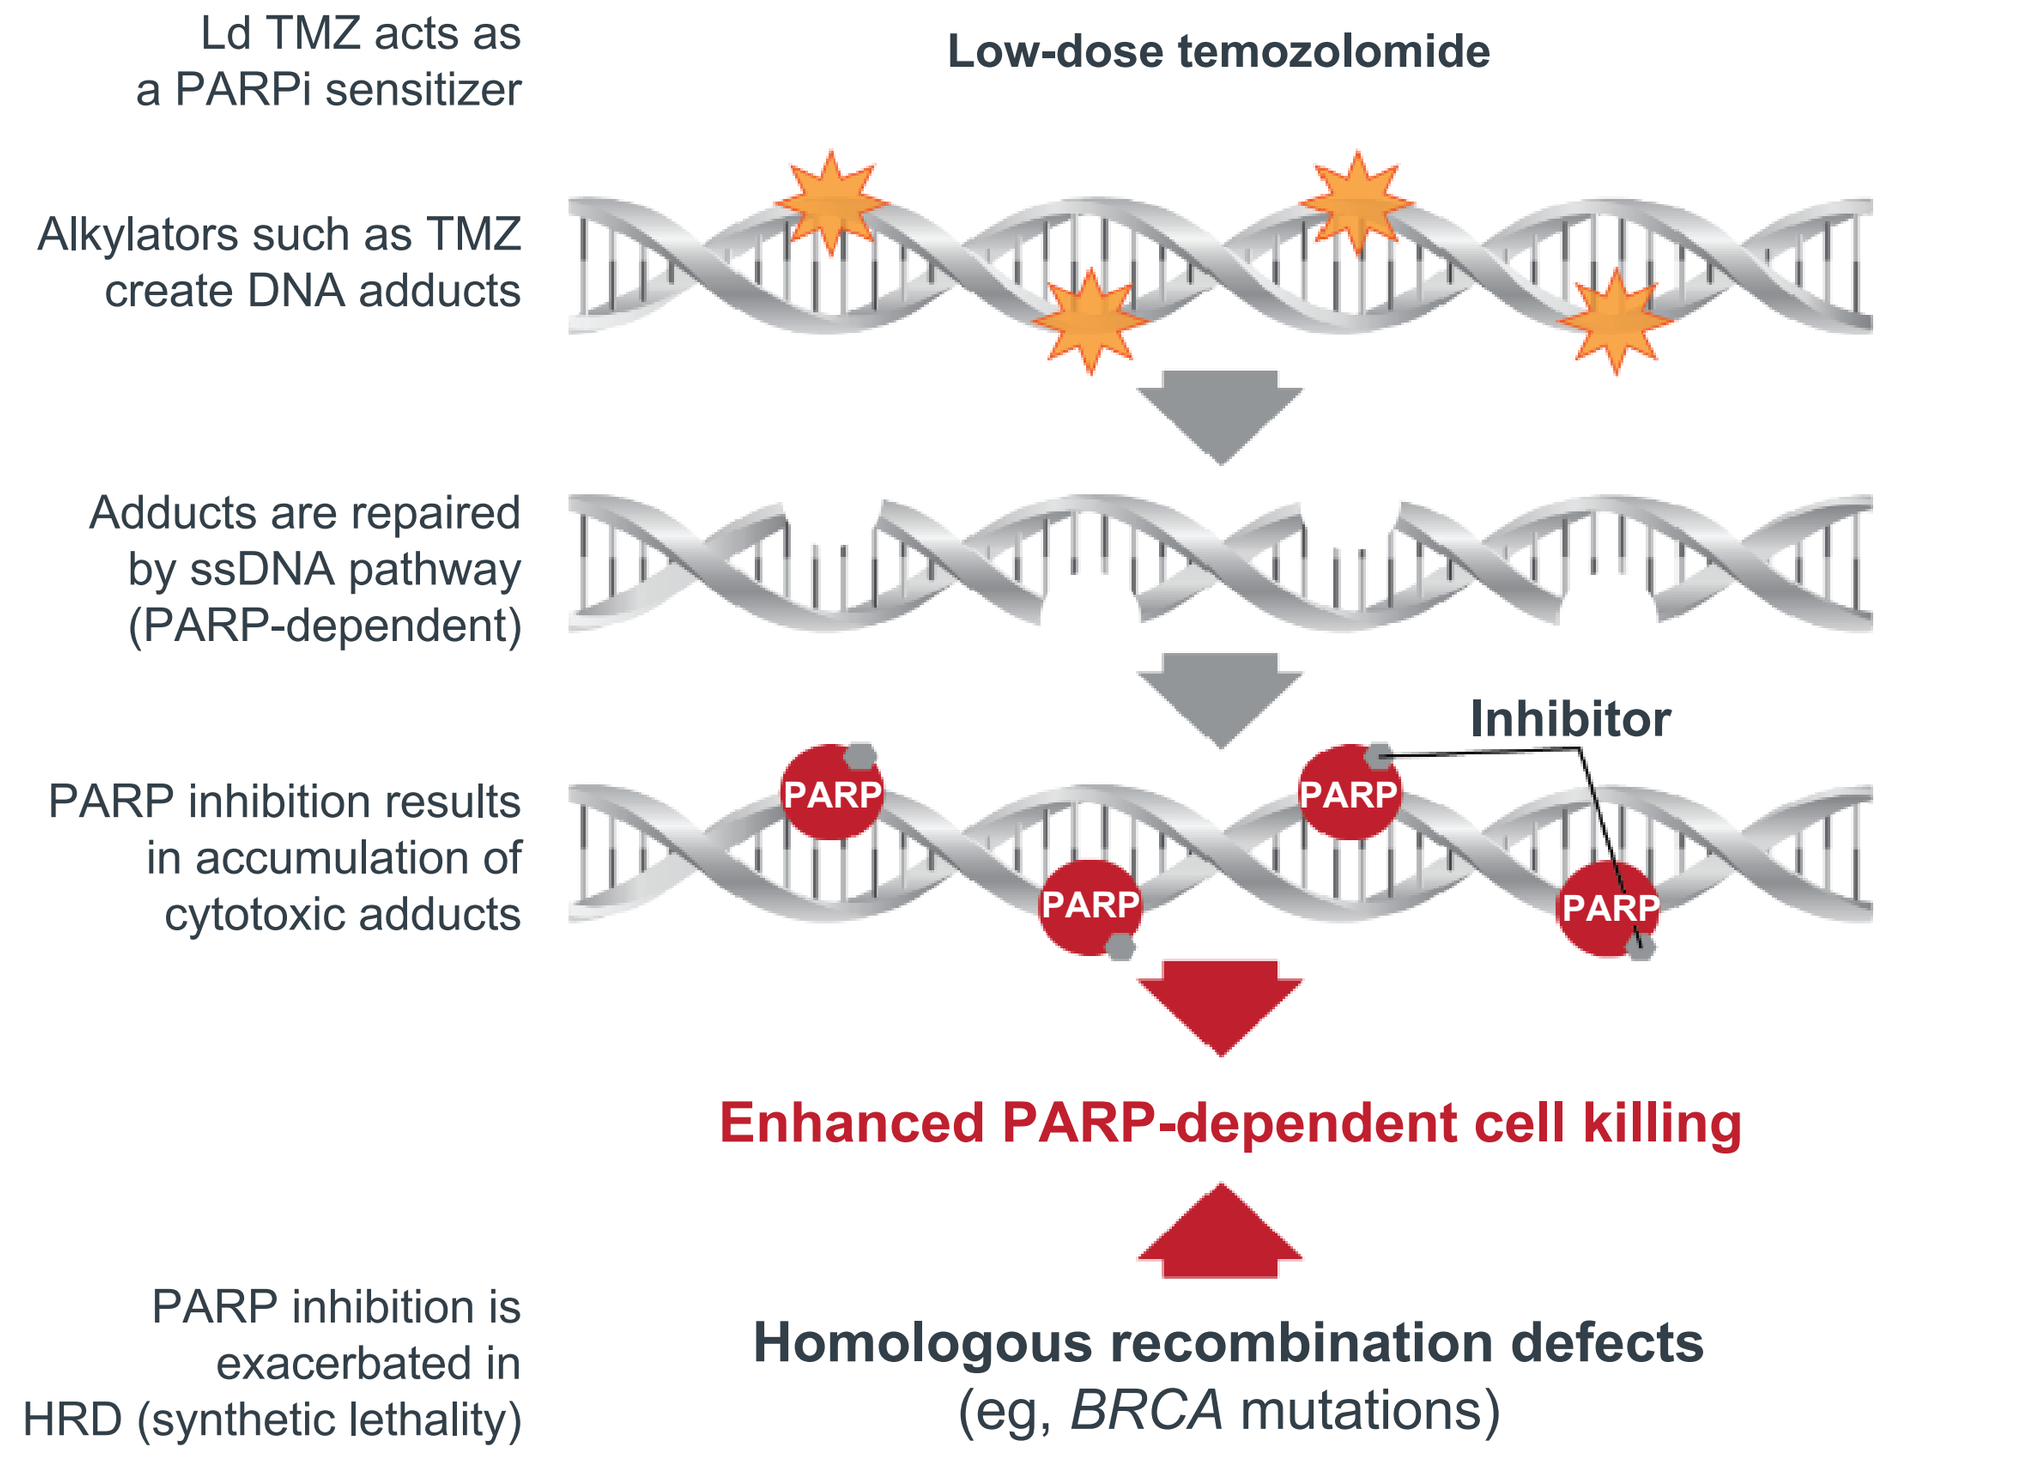

Ld TMZ acts as a PARPi sensitizer
Low-dose temozolomide
Alkylators such as TMZ create DNA adducts
Adducts are repaired by ssDNA pathway (PARP-dependent)
Inhibitor
PARP inhibition results in accumulation of cytotoxic adducts
PARP
Enhanced PARP-dependent cell killing
PARP inhibition is exacerbated in HRD (synthetic lethality)
Homologous recombination defects (eg, BRCA mutations)

Abbreviations: *BRCA*=breast cancer susceptibility gene, DNA=deoxyribonucleic acid, HRD=homologous recombination deficiency, Ld=low dose, PARP=poly(ADP-ribose) polymerase, PARPi=PARP inhibitor, ssDNA=single-strand DNA,TMZ=temozolomide

METHODS

- The study design is detailed in Figure 2
 - The design allowed for exploration of intermediate doses and alternate administration schedules based on emerging safety and pharmacokinetic data
- Radiographic assessments are being conducted every 8 weeks; efficacy set was defined as patients with ≥1 post-baseline tumor assessments

Key Eligibility

- Adult patients (≥18 years old) with a confirmed malignancy that has progressed to advanced or metastatic stage with no option for effective standard therapy
- ECOG PS ≤1
- Disease that is measurable or evaluable per RECIST v1.1; prostate cancer patients were evaluated using Prostate Cancer Working Group 2 (PCWG2) criteria
- Patients were excluded if they had hypersensitivity to TMZ or dacarbazine, or had prior treatment with a PARP inhibitor
- ≤2 prior lines of chemotherapy in the advanced setting (for patients enrolled in the ES SCLC and G/GEJ cancer expansion cohorts only)

Figure 2: Study Design

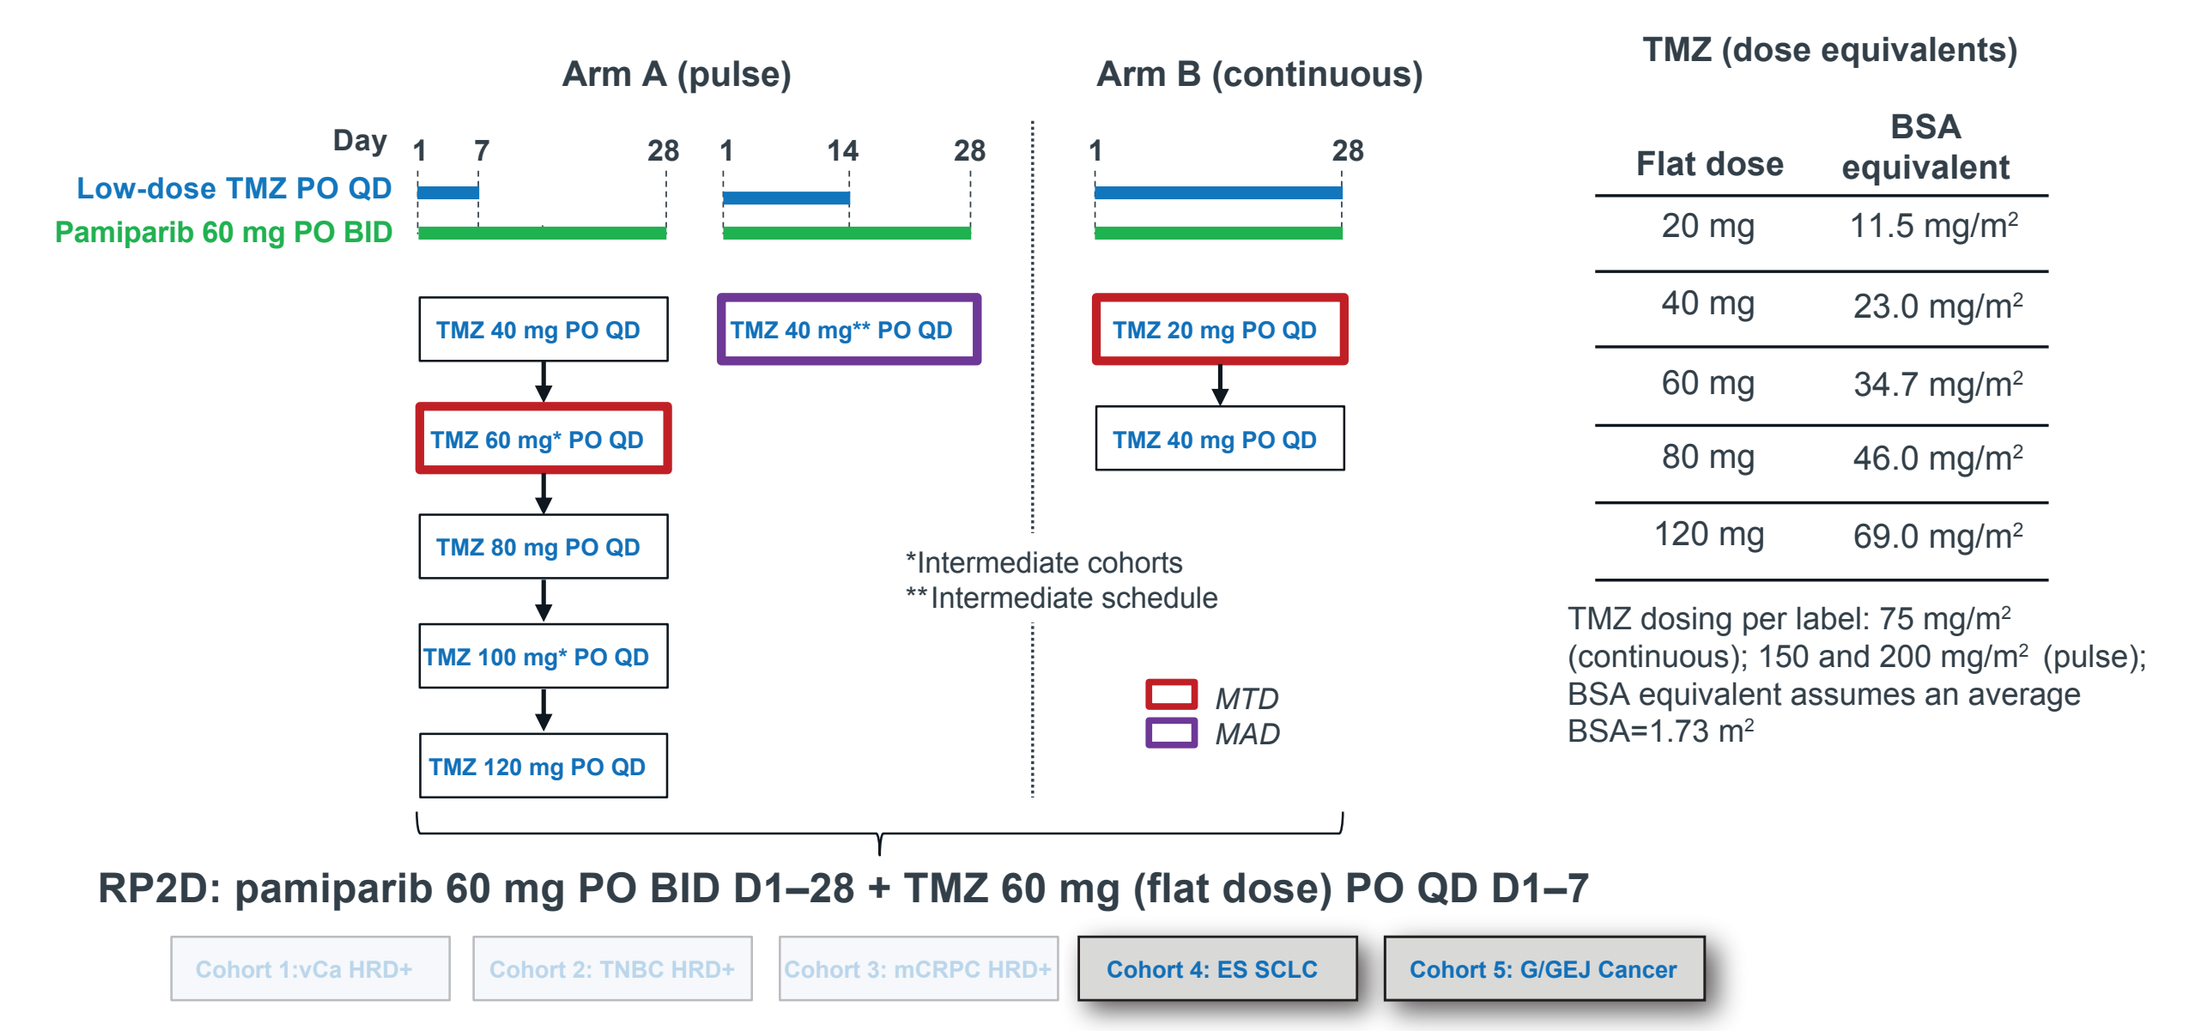

Arm A (pulse)
Arm B (continuous)
Day 1 7 28 1 14 28 1 28
Low-dose TMZ PO QD
Pamiparib 60 mg PO BID
TMZ 40 mg PO QD
TMZ 60 mg* PO QD
TMZ 80 mg PO QD
TMZ 100 mg* PO QD
TMZ 120 mg PO QD
TMZ 40 mg** PO QD
TMZ 20 mg PO QD
TMZ 40 mg PO QD
*Intermediate cohorts
**Intermediate schedule
MTD
MAD
TMZ (dose equivalents)
Flat dose | BSA equivalent
20 mg | 11.5 mg/m²
40 mg | 23.0 mg/m²
60 mg | 34.7 mg/m²
80 mg | 46.0 mg/m²
120 mg | 69.0 mg/m²
TMZ dosing per label: 75 mg/m² (continuous); 150 and 200 mg/m² (pulse); BSA equivalent assumes an average BSA=1.73 m²
RP2D: pamiparib 60 mg PO BID D1–28 + TMZ 60 mg (flat dose) PO QD D1–7
Cohort 1:vCa HRD+
Cohort 2: TNBC HRD+
Cohort 3: mCRPC HRD+
Cohort 4: ES SCLC
Cohort 5: G/GEJ Cancer

Abbreviations: BID=twice daily, BSA=body surface area, D=day, ES SCLC=extensive-stage small cell lung cancer, G/GEJ=gastric/gastroesophageal junction, HRD=homologous recombination deficiency, MAD=maximum administered dose, mCRPC=metastatic castration-resistant prostate cancer, MTD=maximum tolerated dose, OvCa=ovarian cancer, PO=per orem, QD=once daily, RP2D=recommended phase 2 dose, TMZ=temozolomide, TNBC=triple-negative breast cancer

RESULTS

Patient Disposition

- As of 29 July 2019, 113 patients with solid tumors have been enrolled in the study (Table 1); median treatment duration was 2.4 months (range: 0.2–18.1)
- A total of 17 patients (15%) remain on pamiparib and ld TMZ treatment

Table 1: Patient Demographics

		Dose escalation (n=66)	Dose expansion (n=47)	Overall (N=113)
Median age, years (range)		66 (38–85)	61.0 (26–77)	64.0 (26–85)
Sex, n (%)	Male	33 (50.0)	26 (55.3)	59 (52.2)
	Female	33 (50.0)	21 (44.7)	54 (47.8)
Race, n (%)	Caucasian	46 (69.7)	38 (80.9)	84 (74.3)
	Black/African American	9 (13.6)	1 (2.1)	10 (8.8)
	Asian	1 (1.5)	1 (2.1)	2 (1.8)
	Other	5 (7.6)	5 (10.6)	10 (8.8)
	Unknown	5 (7.6)	2 (4.3)	7 (6.2)
ECOG PS, n (%)	0	16 (24.2)	14 (29.8)	30 (26.5)
	1	50 (75.8)	33 (70.2)	83 (73.5)
Median prior cancer therapies, n (range)		3.0 (1–10)	2.0 (1–5)	3.0 (1–10)
Prior anticancer therapies, n (%)	≤3	34 (51.5)	44 (93.6)	78 (69.0)
	4–6	22 (33.3)	2 (4.3)	24 (21.2)
	≥7	10 (15.2)	0 (0)	10 (8.8)
	Missing	0	1 (2)	1 (0.9)

Abbreviations: ECOG PS=Eastern Cooperative Oncology Group performance status

Recommended Phase 2 Dose and Schedule

- Enrollment in the dose-escalation phase was completed in September 2018; the RP2D was determined to be pamiparib 60 mg PO BID D1–28 in combination with TMZ 60 mg PO QD D1–7, based on tolerability

Pharmacokinetics

- Pamiparib plasma exposure in patients treated with the combination regimen (Table 2) was similar to that of single-agent pamiparib

Table 2: Pamiparib Pharmacokinetics

Time point	Geometric Mean Pamiparib Concentration (%CV)			
	Pamiparib + ld TMZ (N=51)		Pamiparib monotherapy (N=11)	
	Single dose	Steady state	Single dose	Steady state
Predose	0 ng/mL (0)	2005 ng/mL (69)	0 ng/mL (0)	2327 ng/mL (35)
2h postdose	1744 ng/mL (32)	3450 ng/mL (40)	1881 ng/mL (27)	3891 ng/mL (47)

Abbreviations: CV=coefficient of variation, ld=low dose, TMZ=temozolomide

Dose Escalation: Preliminary Antitumor Activity

Figure 3: Maximum Tumor Reduction in Evaluable Patients (A) and Biomarker Assessment (B)

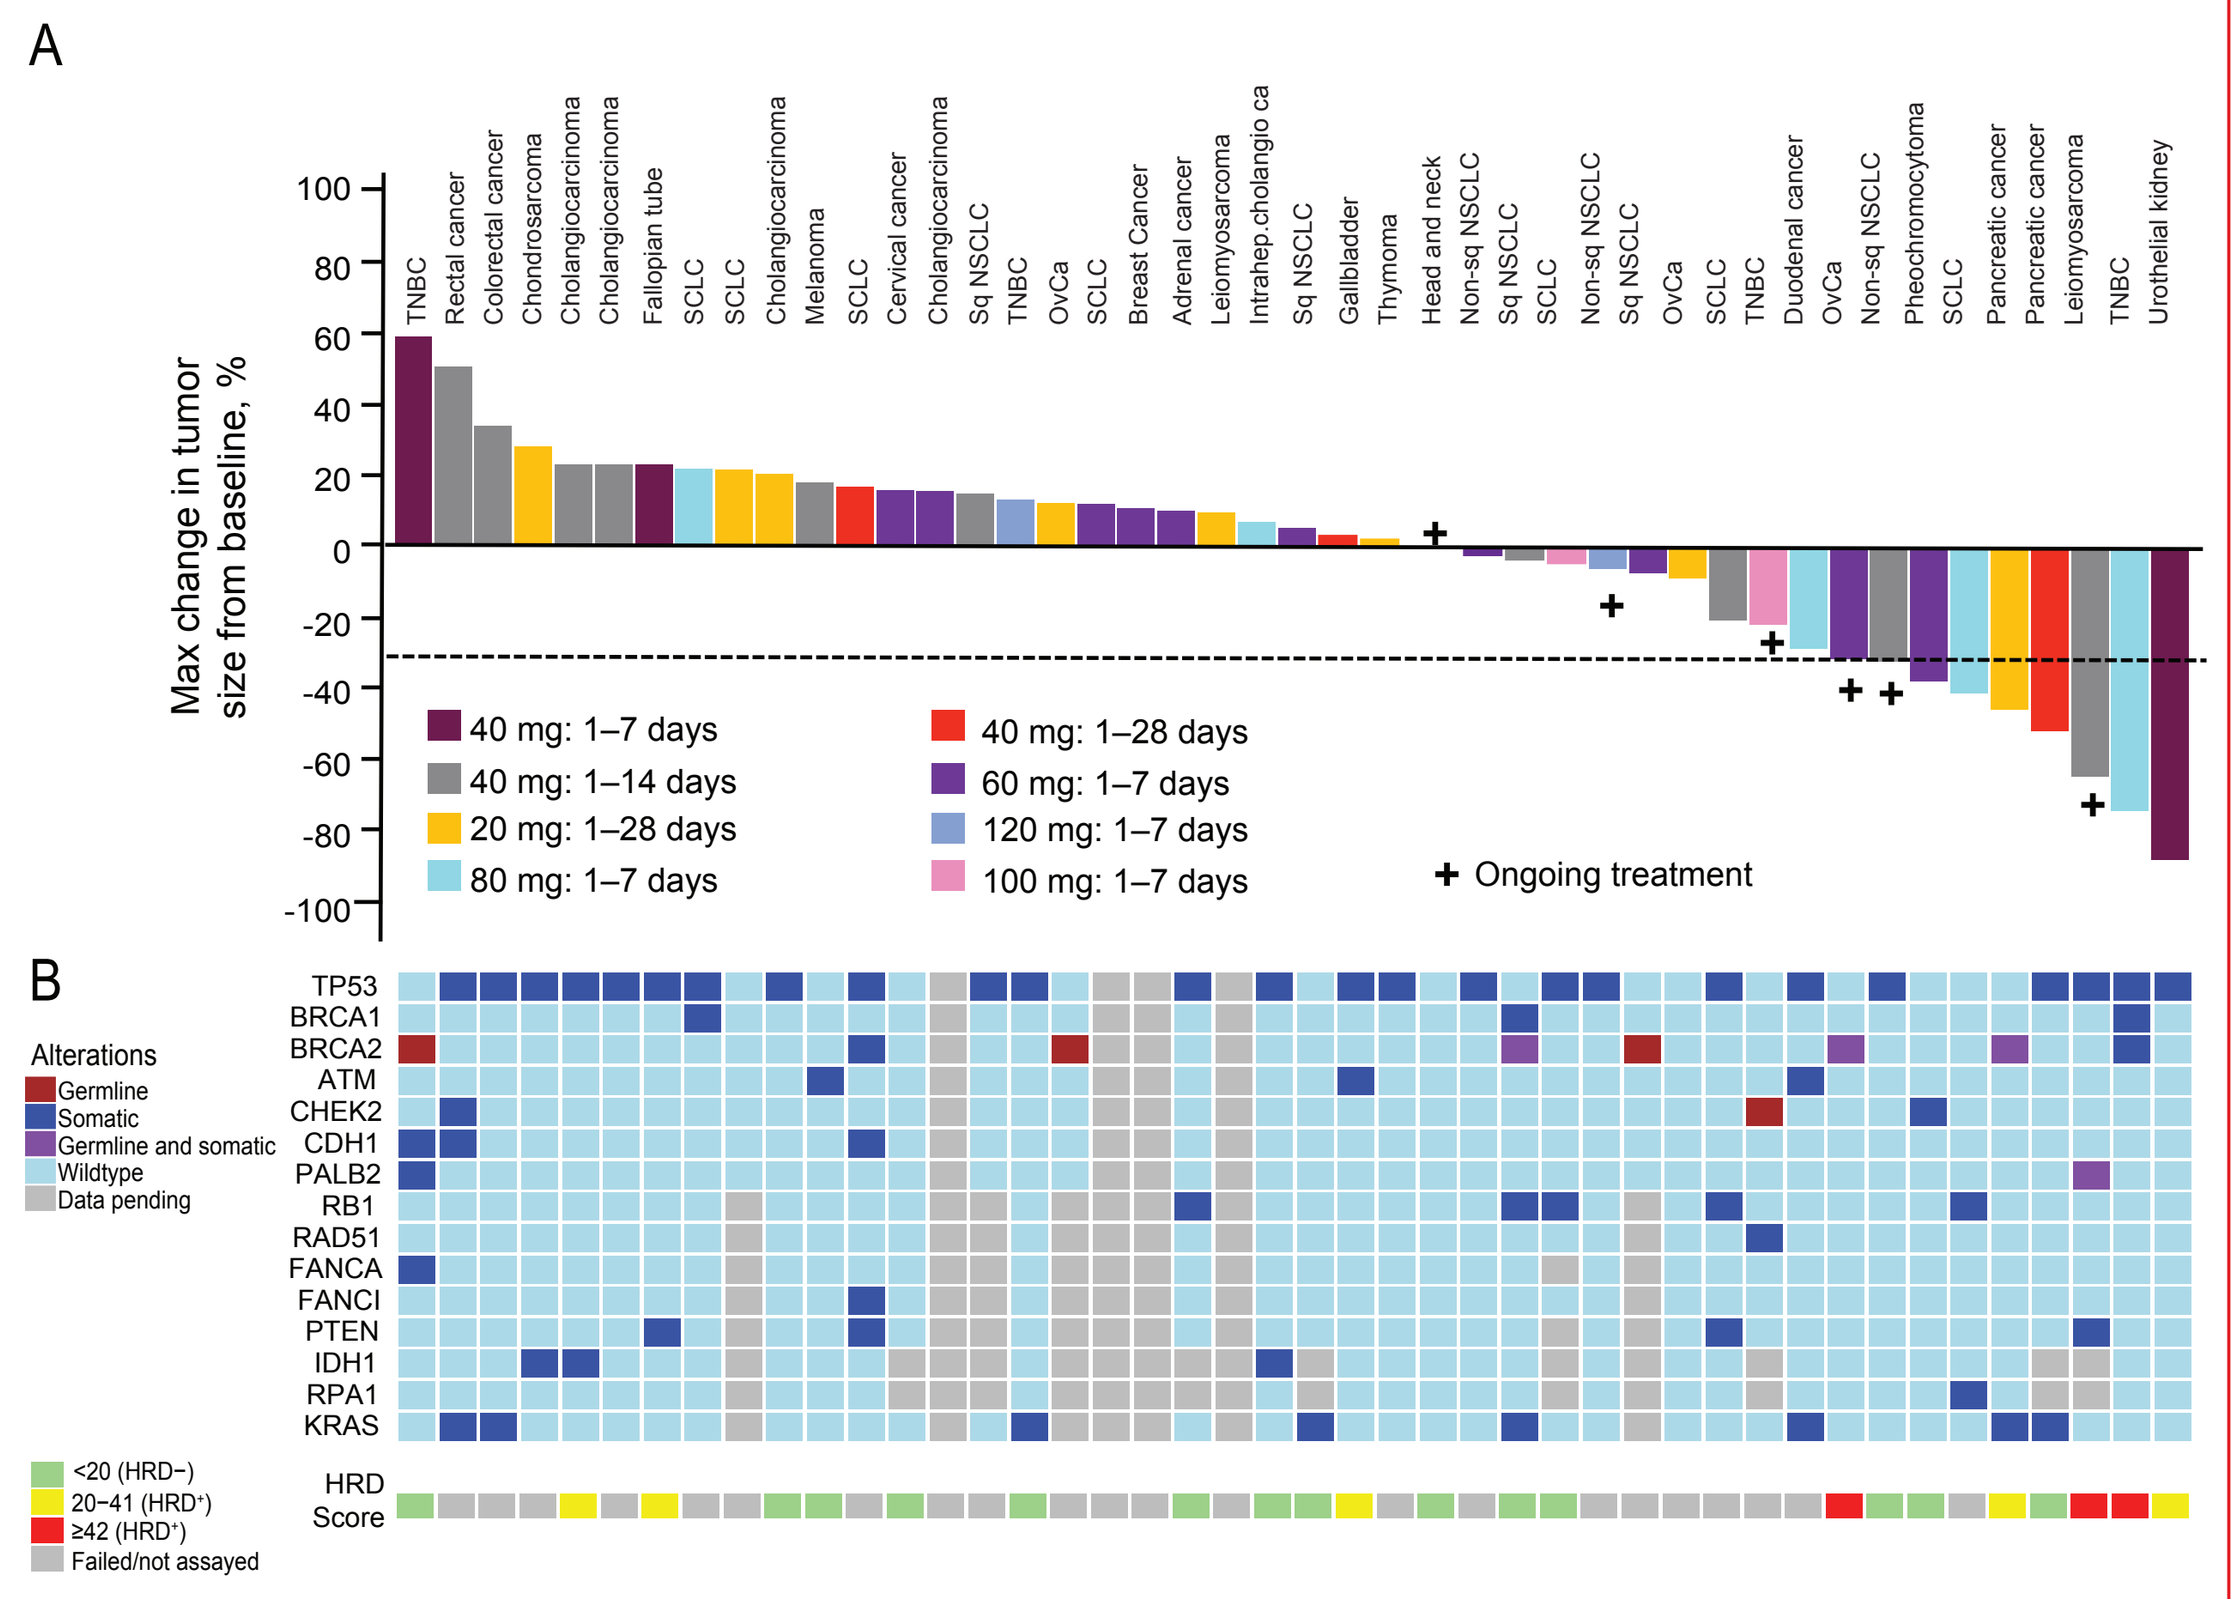

A
Max change in tumor size from baseline, %
TNBC
Rectal cancer
Colorectal cancer
Chondrosarcoma
Cholangiocarcinoma
Cholangiocarcinoma
Fallopian tube
SCLC
SCLC
Cholangiocarcinoma
Melanoma
SCLC
Cervical cancer
Cholangiocarcinoma
Sq NSCLC
TNBC
OvCa
SCLC
Breast Cancer
Adrenal cancer
Leiomyosarcoma
Intrahep.cholangio ca
Sq NSCLC
Gallbladder
Thymoma
Head and neck
Non-sq NSCLC
Sq NSCLC
SCLC
Non-sq NSCLC
Sq NSCLC
OvCa
SCLC
TNBC
Duodenal cancer
OvCa
Non-sq NSCLC
Pheochromocytoma
SCLC
Pancreatic cancer
Pancreatic cancer
Leiomyosarcoma
TNBC
Urothelial kidney
40 mg: 1–7 days
40 mg: 1–14 days
20 mg: 1–28 days
80 mg: 1–7 days
40 mg: 1–28 days
60 mg: 1–7 days
120 mg: 1–7 days
100 mg: 1–7 days
+ Ongoing treatment
B
Alterations
Germline
Somatic
Germline and somatic
Wildtype
Data pending
TP53
BRCA1
BRCA2
ATM
CHEK2
CDH1
PALB2
RB1
RAD51
FANCA
FANCI
PTEN
IDH1
RPA1
KRAS
<20 (HRD−)
20–41 (HRD+)
≥42 (HRD+)
Failed/not assayed
HRD Score

Biomarker analysis: Biomarker data were generated from ctDNA (C1D1), gDNA (C1D1) and/or tissue samples. NGS of ctDNA was performed using a 600-gene panel (PredicineATLAS). Somatic *BRCA1* and *BRCA2* status, HRD score and mutational status of selected genes were determined using NGS of DNA extracted from FFPE tissues (Myriad myChoice HRD). The mutational status of germline *BRCA1*, *BRCA2* status, and selected genes were determined using NGS of gDNA (Color Genomics). Only pathogenic mutations are shown (ClinVar) annotated as germline, somatic, or both.

Abbreviations: *BRCA*=breast cancer susceptibility gene, ctDNA=circulating tumor DNA, DNA=deoxyribonucleic acid, FFPE=formalin-fixed/parafin embedded, gDNA=genomic DNA, HRD=homologous recombination deficiency, NGS=next-generation sequencing, Non-sq NSCLC=non-squamous non-small cell lung cancer, OvCa=ovarian cancer, PCWG2=Prostate Cancer Working Group 2, PSA=prostate-specific antigen, SCLC=small cell lung cancer, Sq NSCLC=squamous non-small cell lung cancer, TNBC=triple-negative breast cancer

- As of 29 July 2019, 57/66 patients enrolled in the dose-escalation phase were evaluable for response; 52/66 patients had measurable disease and were evaluated by either RECIST (Figure 3) or PCWG2 (prostate cancer)
 - Overall response rate (ORR) was 19.3% (11 PR); 8/11 responses were confirmed. In addition, an unconfirmed PSA response was observed in 1 prostate cancer patient
 - Disease control rate (DCR) was 64.9% (95% CI, 51.1–77.1)
 - Median duration of response was 6.4 months (95% CI, 2.1–7.7)
 - Median treatment duration was 3.7 months (range 0.2–18.1)
- Biomarker analyses showed an HRD$^+$ score rate of 32.2% (N=22; HRD$^+$=8/22)
 - 62.5% of patients (5/8) with HRD$^+$ scores showed a response
- Not all HRD$^+$ patients harbored mutations in *BRCA1*, *BRCA2*, or *PALB2*

G/GEJ Cancer Expansion Cohort: Preliminary Antitumor Activity (≤2 Prior Lines of Chemotherapy)

Figure 4: Treatment Duration in Evaluable Patients

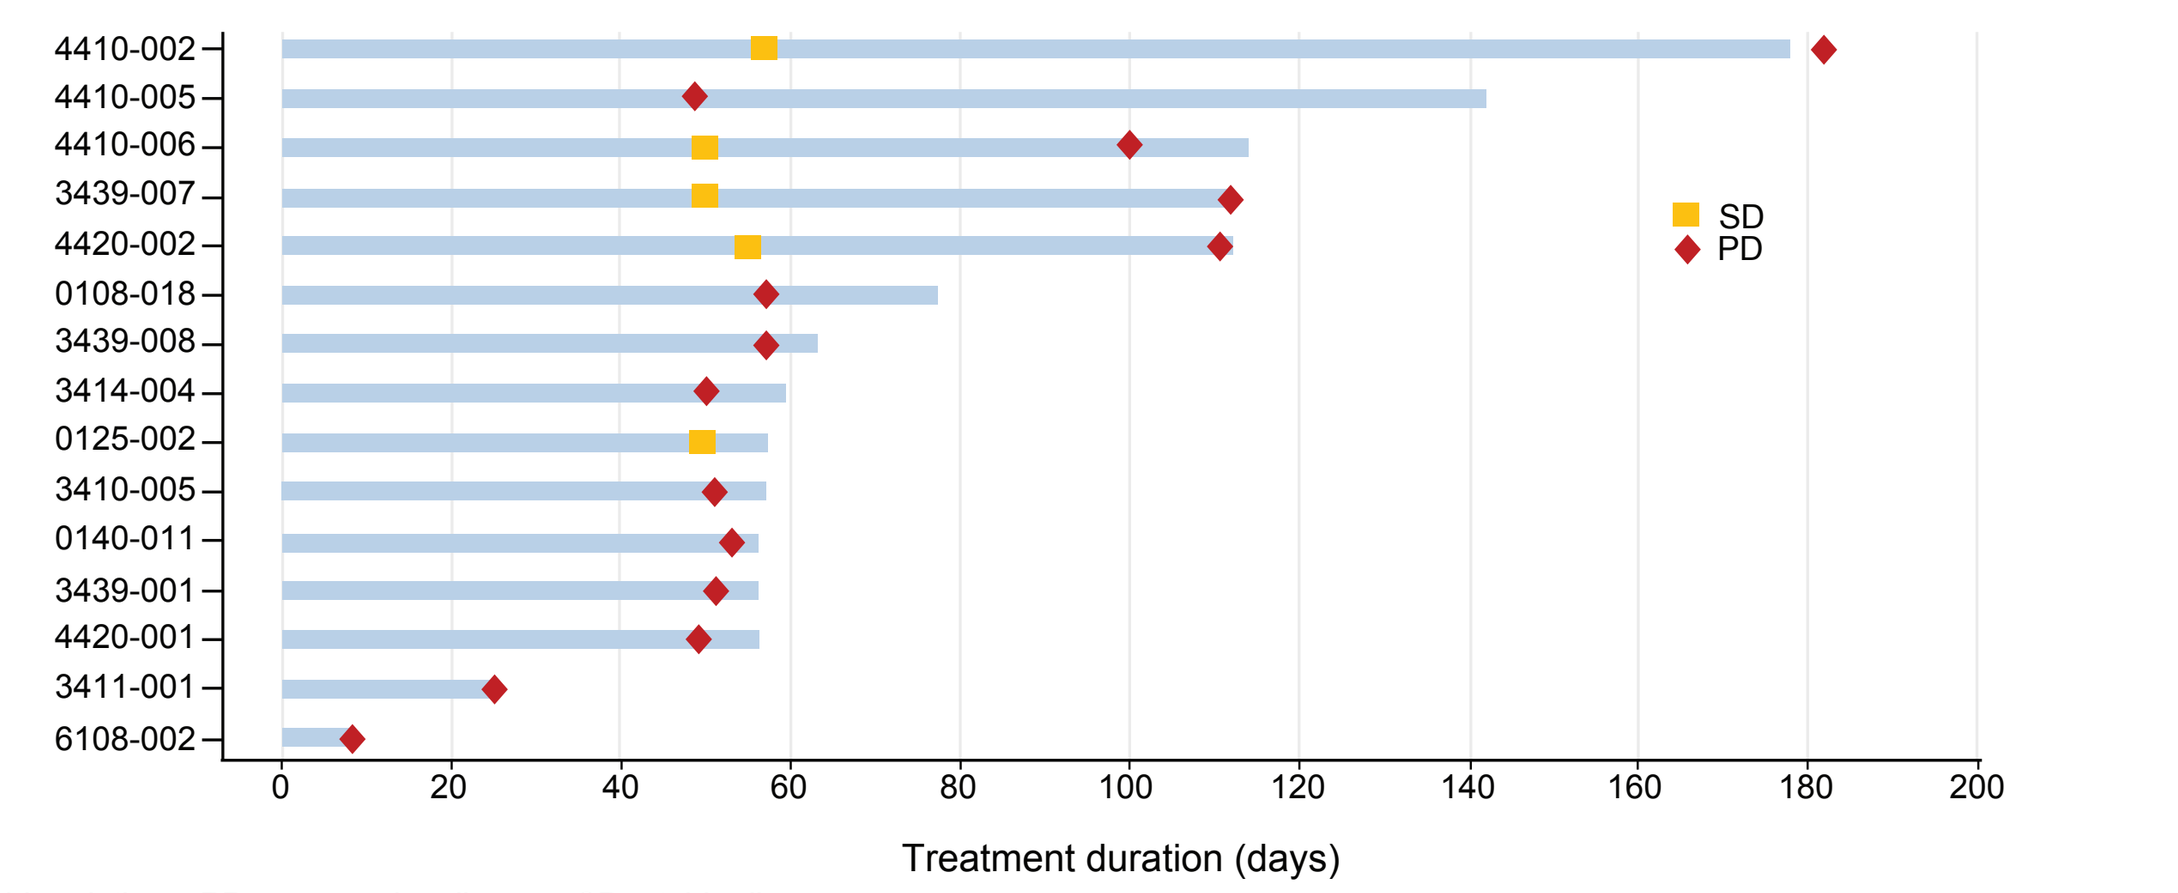

4410-002
4410-005
4410-006
3439-007
4420-002
0108-018
3439-008
3414-004
0125-002
3410-005
0140-011
3439-001
4420-001
3411-001
6108-002
SD
PD
0 20 40 60 80 100 120 140 160 180 200
Treatment duration (days)

Abbreviations: PD=progressive disease, SD=stable disease

- At the time of data cut off, 20 G/GEJ cancer patients had been enrolled in the dose-expansion phase, and 15 were evaluable for response. The median follow-up time was 4 months (range 0.4–7.4)
 - 70% of patients had an ECOG PS of 1
 - ORR was 0%; DCR was 33.3% (95% CI, 11.8–61.6)
 - Median treatment duration was 1.94 months (range 0.3–5.8); 2 patients are still ongoing (Figure 4)

ES SCLC Expansion Cohort: Preliminary Antitumor Activity (≤2 Prior Lines of Chemotherapy)

Figure 5: Maximum Tumor Reduction in Evaluable Patients (A) and Treatment Duration (B)

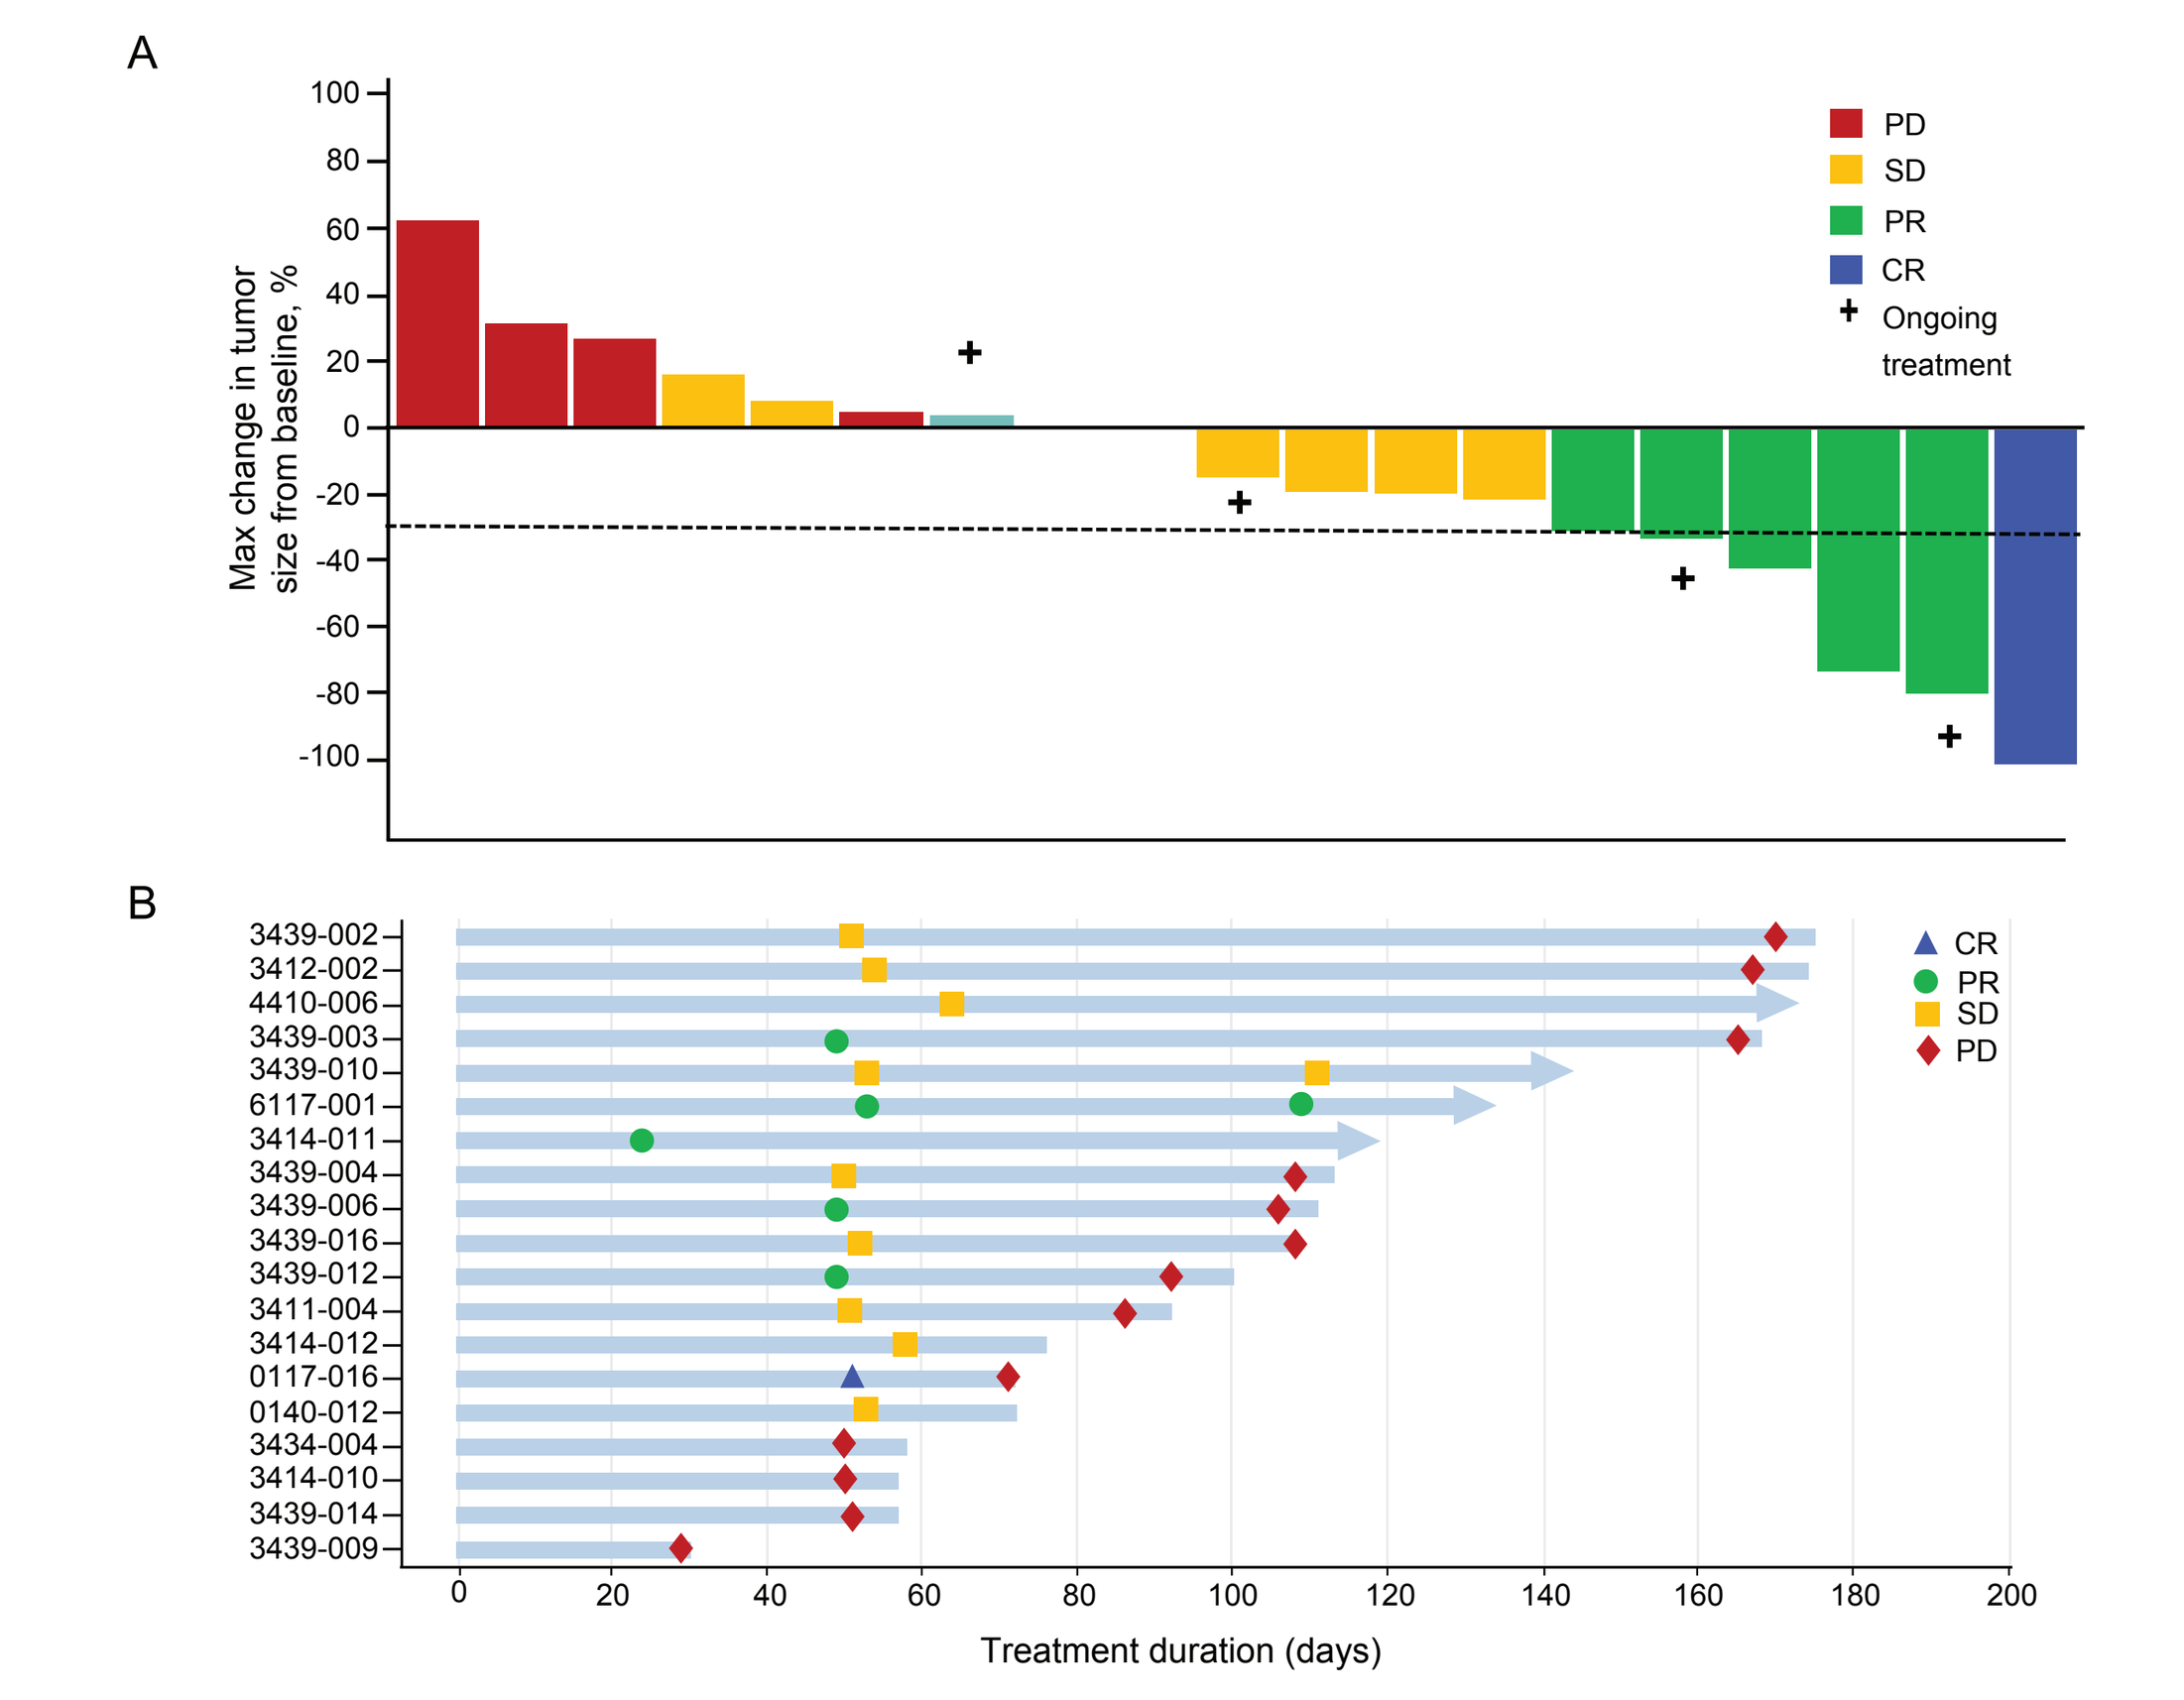

A
Max change in tumor size from baseline, %
PD
SD
PR
CR
+ Ongoing treatment
B
3439-002
3412-002
4410-006
3439-003
3439-010
6117-001
3414-011
3439-004
3439-006
3439-016
3439-012
3411-004
3414-012
0117-016
0140-012
3434-004
3414-010
3439-014
3439-009
CR
PR
SD
PD
0 20 40 60 80 100 120 140 160 180 200
Treatment duration (days)

Abbreviations: CR=complete response, PD=progressive disease, PR=partial response, SD=stable disease

- As of 29 July 2019, 22 ES SCLC patients had been enrolled in the dose-expansion phase and 19 were evaluable for response (Figure 5). The median follow-up time was 4 months (range 0.5–6.4)
 - 50% of patients had a chemotherapy-free interval ≥90 days from their last platinum regimen and 86.4% of patients had an ECOG PS of 1
 - ORR was 31.6% (5 PR and 1 CR); DCR was 78.9% (95% CI, 54.4–93.9) (Figure 5A)
 - Median treatment duration was 3.6 months (range 1.0–5.7); 5 patients are still ongoing (Figure 4)

Safety and Tolerability

- 112 patients had ≥1 treatment-emergent adverse event (TEAE); the most common TEAEs (all grades) were anemia, nausea, fatigue, decreased appetite, neutropenia, thrombocytopenia, and vomiting (Table 3)
- 63 (55.8%) patients experienced Grade ≥3 TEAEs related to the study drug (Table 4)
 - The most common Grade ≥3 adverse events (AEs) were cytopenias (anemia, neutropenia, and thrombocytopenia), all of which were manageable and reversible
 - Related Grade 4 AEs included neutropenia (9.7%), decreased neutrophil counts (8.8%), thrombocytopenia (11.5%), decreased platelet counts (7.1%), decreased WBC (2.7%); and anemia, aspartate aminotransferase (AST) elevation and alanine aminotransferase (ALT) elevation (0.9% each)
 - No related Grade 5 AEs were reported
- 3 (2.7%) patients experienced AEs related to the study drug that resulted in treatment discontinuation
- Serious AEs related to study treatment were reported in 11 (9.7%) patients
- 4 (3.5%) patients experienced AEs with fatal outcome, all of which were deemed unrelated to the study drug

Table 3: TEAEs Occurring in ≥9% of Patients Regardless of Relationship (All Grades)

TEAEs	Dose escalation (n=66)	Dose expansion (n=47)	All patients (N=113)
Anemia	36 (54.5)	29 (61.7)	65 (57.5)
Nausea	35 (53.0)	26 (55.3)	61 (54.0)
Fatigue	30 (45.5)	25 (53.2)	55 (48.7)
Decreased appetite	20 (30.3)	19 (40.4)	39 (34.5)
Neutropenia	22 (33.3)	15 (31.9)	37 (32.7)
Thrombocytopenia	23 (34.8)	13 (27.7)	36 (31.9)
Vomiting	15 (22.7)	18 (38.3)	33 (29.2)
Platelet count decrease	12 (18.2)	14 (29.8)	26 (23.0)
Constipation	7 (10.6)	15 (31.9)	22 (19.5)
Diarrhea	14 (21.2)	8 (17.0)	22 (19.5)
Neutrophil count decrease	12 (18.2)	10 (21.3)	22 (19.5)
Abdominal pain	11 (16.7)	7 (14.9)	18 (15.9)
Cough	9 (13.6)	6 (12.8)	15 (13.3)
Dyspnea	12 (18.2)	3 (6.4)	15 (13.3)
Dizziness	10 (15.2)	4 (8.5)	14 (12.4)
Aspartate aminotransferase increase	9 (13.6)	4 (8.5)	13 (11.5)
Headache	7 (10.6)	5 (10.6)	12 (10.6)
White blood cell count decrease	8 (12.1)	4 (8.5)	12 (10.6)
Blood bilirubin increase	7 (10.6)	3 (6.4)	10 (8.8)
Hypokalemia	8 (12.1)	2 (4.3)	10 (8.8)

Abbreviation: TEAE=treatment-emergent adverse event

CONCLUSIONS

- The RP2D and schedule of the combination was determined to be pamiparib 60 mg PO BID D1–28 and TMZ 60 mg PO QD D1–7
- The combination of pamiparib and ld TMZ shows:
 - a manageable safety profile, with cytopenias being the most frequent Grade ≥3 events
 - promising preliminary efficacy in patients with ES SCLC and various other tumor types
- Dose-escalation patients with HRD$^+$ tumors, irrespective of *BRCA* mutational status, showed a high response rate
 - HRD status may be a promising biomarker for sensitivity to the pamiparib + ld TMZ combination, regardless of tumor type
- Further biomarker exploration is underway

Table 4: TEAEs Grade ≥3 Reported as Related to Pamiparib and/or Temozolomide Occurring in More Than 2 Patients

TEAEs	Dose escalation (n=66)	Dose expansion (n=47)	All patients (N=113)
Anemia	21 (31.8)	16 (34.0)	37 (32.7)
Neutropenia	18 (27.3)	11 (23.4)	29 (25.7)
Thrombocytopenia	13 (19.7)	9 (19.1)	22 (19.5)
Neutrophil count decrease	11 (16.7)	9 (19.1)	20 (17.7)
Platelet count decreased	7 (10.6)	8 (17.0)	15 (13.3)
WBC count decreased	7 (10.6)	4 (8.5)	11 (9.7)
Asthenia	0 (0)	3 (6.4)	3 (2.7)

Abbreviations: TEAE=treatment-emergent adverse event, WBC=white blood cell

REFERENCES

1. Thomas C, Tulin AV. Poly-ADP-ribose polymerase: Machinery for nuclear processes. *Mol Aspects Med.* 2013;34(6): 1124-1137.
2. Coleman RL, Sill MW, Bell-McGuinn K, et al. A phase II evaluation of the potent, highly selective PARP inhibitor veliparib in the treatment of persistent or recurrent epithelial ovarian, fallopian tube, or primary peritoneal cancer in patients who carry a germline BRCA1 or BRCA2 mutation - An NRG Oncology/Gynecologic Oncology Group study. *Gynecol Oncol.* 2015;137(3):386-391.
3. Pommier Y, O'Connor MJ, de Bono J. Laying a trap to kill cancer cells: PARP inhibitors and their mechanisms of action. *Sci Transl Med.* 2016;8(362):362ps317.
4. Kubota E, Williamson CT, Ye R, et al. Low ATM protein expression and depletion of p53 correlates with olaparib sensitivity in gastric cancer cell lines. *Cell Cycle.* 2014;13(13):2129-2137.
5. Ledermann J, Harter P, Gourley C, et al. Olaparib maintenance therapy in platinum-sensitive relapsed ovarian cancer. *N Engl J Med.* 2012;366(15):1382-1392.
6. Owonikoko TK, Dahlberg SE, Khan SA, et al. A phase 1 safety study of veliparib combined with cisplatin and etoposide in extensive stage small cell lung cancer: A trial of the ECOG-ACRIN Cancer Research Group (E2511). *Lung Cancer.* 2015;89(1):66-70.
7. Robinson D, Van Allen EM, Wu YM, et al. Integrative clinical genomics of advanced prostate cancer. *Cell.* 2015;161(5): 1215-1228.
8. Tutt A, Robson M, Garber JE, et al. Oral poly(ADP-ribose) polymerase inhibitor olaparib in patients with *BRCA*1 or *BRCA*2 mutations and advanced breast cancer: a proof-of concept trial. *Lancet.* 2010;376(9737):235-244.
9. Murai J, Huang S-Y, Das B, et al. Trapping of PARP1 and PARP2 by clinical PARP inhibitors. *Cancer Res.* 2012;72(21): 5588-5599.
10. Lickliter J, Mileshkin L, Voskoboynikm M, et al. Dose escalation/expansion study to investigate the safety, pharmacokinetics, food effect, and antitumor activity of BGB-290 in patients with advanced solid tumors. *Ann Oncol.* 2017; 28 (suppl 5): v122-v141.
11. Guerard M, Johnson G, Dertinger S, et al. Dose-response relationship of temozolomide, determined by the Pig-a, comet, and micronucleous assay. *Arch Toxicol.* 2017; 91:2443-2453.
12. Johnson M, Galsky M, Barve M, et al. Preliminary results of pampirarib, a PARP1/2 inhibitor, in combination with temozolomide in patients with locally advanced or metastatic solid tumors. Poster number 421P. Presented at ESMO. October 19–23 2018. Munich, Germany.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff, the study patients, and their families. BeiGene, Ltd. provided financial support for this presentation, including writing and editorial assistance by Ira Mills, PhD, and Erin Spohr of Ashfield Healthcare Communications.

AUTHOR DISCLOSURE

A Stradella served as a consultant for Celgene, Eisai, and Roche and received travel support from Celgene, Eisai, Pfizer, and Roche. Other author disclosures detailed in the Quick Response (QR) Code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com