PHASE 1/2 STUDY OF SINGLE-AGENT ZANUBRUTINIB IN PATIENTS WITH RELAPSED/REFRACTORY MARGINAL ZONE LYMPHOMA

Abstract 2804

Alessandra Tedeschi, MD[1]; Judith Trotman, MBChB, FRACP, FRCPA[2,3]; Constantine Tam, MBBS, MD, FRACP, FRCPA[4-7]; David Simpson, MBChB, FRACP, FRCPA[8,9]; Hyeon-Seok Eom, MD, PhD[10]; Rebecca Elstrom, MD[9]; Ziwen Tan, MSc[11]; Jennifer C. Stern, PhD[9]; Siminder Atwal, PhD[9]; William Novotny, MD[9]; Jane Huang, MD[9]; and Stephen Opat, MBBS, FRACP, FRCPA[12,13]

[1]ASST Grande Osperdale Metropolitano Niguarda, Milan, Italy; [2]Concord Repatriation General Hospital, Concord, New South Wales, Australia; [3]University of Sydney, Concord, New South Wales, Australia; [4]Peter MacCallum Cancer Centre, Melbourne, Victoria, Australia; [5]St Vincent's Hospital, Fitzroy, Victoria, Australia; [6]University of Melbourne, Parkville, Victoria, Australia; [7]Royal Melbourne Hospital, Parkville, Victoria, Australia; [8]North Shore Hospital, Auckland, New Zealand; [9]BeiGene USA, Inc., San Mateo, CA, USA; [10]National Cancer Center – Korea, Goyang-si Gyeonggi-do, Republic of Korea; [11]BeiGene (Beijing) Co., Ltd., Beijing, China; [12]Monash Health, Clayton, Victoria, Australia; and [13]Monash University, Clayton, Victoria, Australia

INTRODUCTION

- Marginal zone lymphoma (MZL) is the third most common lymphoma and represents approximately 5%-15% of all non-Hodgkin lymphomas[1]
- B-cell receptor (BCR) signaling is a key activation pathway in MZL, and Bruton tyrosine kinase (BTK), a target of BCR, has been shown to mediate B-cell proliferation, migration, and cell survival[2-4]
 - Inhibitors of BTK have shown improved efficacy and tolerable toxicity profiles compared with chemotherapy-based approaches in MZL[5]
- Zanubrutinib, a potent, selective, and irreversible BTK inhibitor, was designed to maximize BTK occupancy and minimize off-target inhibition of TEC, ITK, and EGFR-family kinases[6]
- Zanubrutinib was investigated in a first-in-human phase 1/2 study (AU-003), designed to evaluate the safety, pharmacokinetics, and antitumor activity of zanubrutinib in patients with B-cell malignancies
 - Study includes disease-specific cohorts, including relapsed/refractory (R/R) MZL
 - Enrollment is complete, and a total of 384 patients have received treatment in this study, including 20 patients with MZL
 - At a median follow-up of **27.1 months**, we report safety and efficacy data for the 20 patients with MZL treated with single-agent zanubrutinib

OBJECTIVES

- Evaluate safety and preliminary efficacy of zanubrutinib monotherapy in patients with R/R MZL

METHODS

- AU-003 is a first-in-human, open-label, multicenter, phase 1/2 study of zanubrutinib in patients with B-cell malignancies (NCT02343120; **Figure 1**)

Figure 1: AU-003 Study Schema
Indication-Specific Expansion Cohorts

DOSE ESCALATION

Dose	All Dosed (MZL)
40 mg qd	3 (0)
80 mg qd	4 (0)
160 mg qd	5 (0)
320 mg qd	1 (0)
160 mg bid	4 (0)

→ **RP2D[a]** 320 mg qd or 160 mg bid →

DOSE EXPANSION

Pop	RP2D Dose	Disease	All Dosed (MZL)
R/R	**qd**	**All B-cell**	**18 (3)**
R/R	**bid**	**All B-cell**	**21 (3)**
R/R	bid	Non-GCB DLBCL	38
R/R	bid	CLL/SLL	71
R/R	bid	WM	21
R/R	qd	CLL/SLL	20
Any	Any	WM	50
R/R	Any	MCL	20
TN	Any	CLL/SLL	21
TN	Any	MCL	20
R/R	Any	HCL	11
R/R	**bid**	**iNHL**	**39 (14)**
R/R	bid	Richter transformation	15
R/R	bid	All B-cell (prior BTKi)	3

Key Eligibility
- WHO-defined B-cell malignancy
- >1 prior therapy (relapsed cohorts only)
- No available higher-priority treatment
- ECOG PS 0-2
- ANC >1000/µL, platelets >100000/µL[b]
- Adequate renal and hepatic function; no significant cardiac disease[c]

Cohorts containing MZL pts (n=20) in blue

ANC, absolute neutrophil count; bid, twice daily; BTKi, Bruton tyrosine kinase inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; GCB-DLBCL, germinal center B-cell–like diffuse large B-cell lymphoma; HCL, hairy cell leukemia; iNHL, indolent non-Hodgkin lymphoma; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; qd, every day; R/R, relapsed/refractory; RP2D, recommended phase 2 dose; TN, treatment naïve; WM, Waldenström macroglobulinemia.
[a]Both doses RP2D but as of protocol v.6, all patients were encouraged to switch to 160 mg bid.
[b]Growth factor/transfusion allowed.
[c]Anticoagulation allowed.

Figure 2: Disposition for Patients With R/R MZL

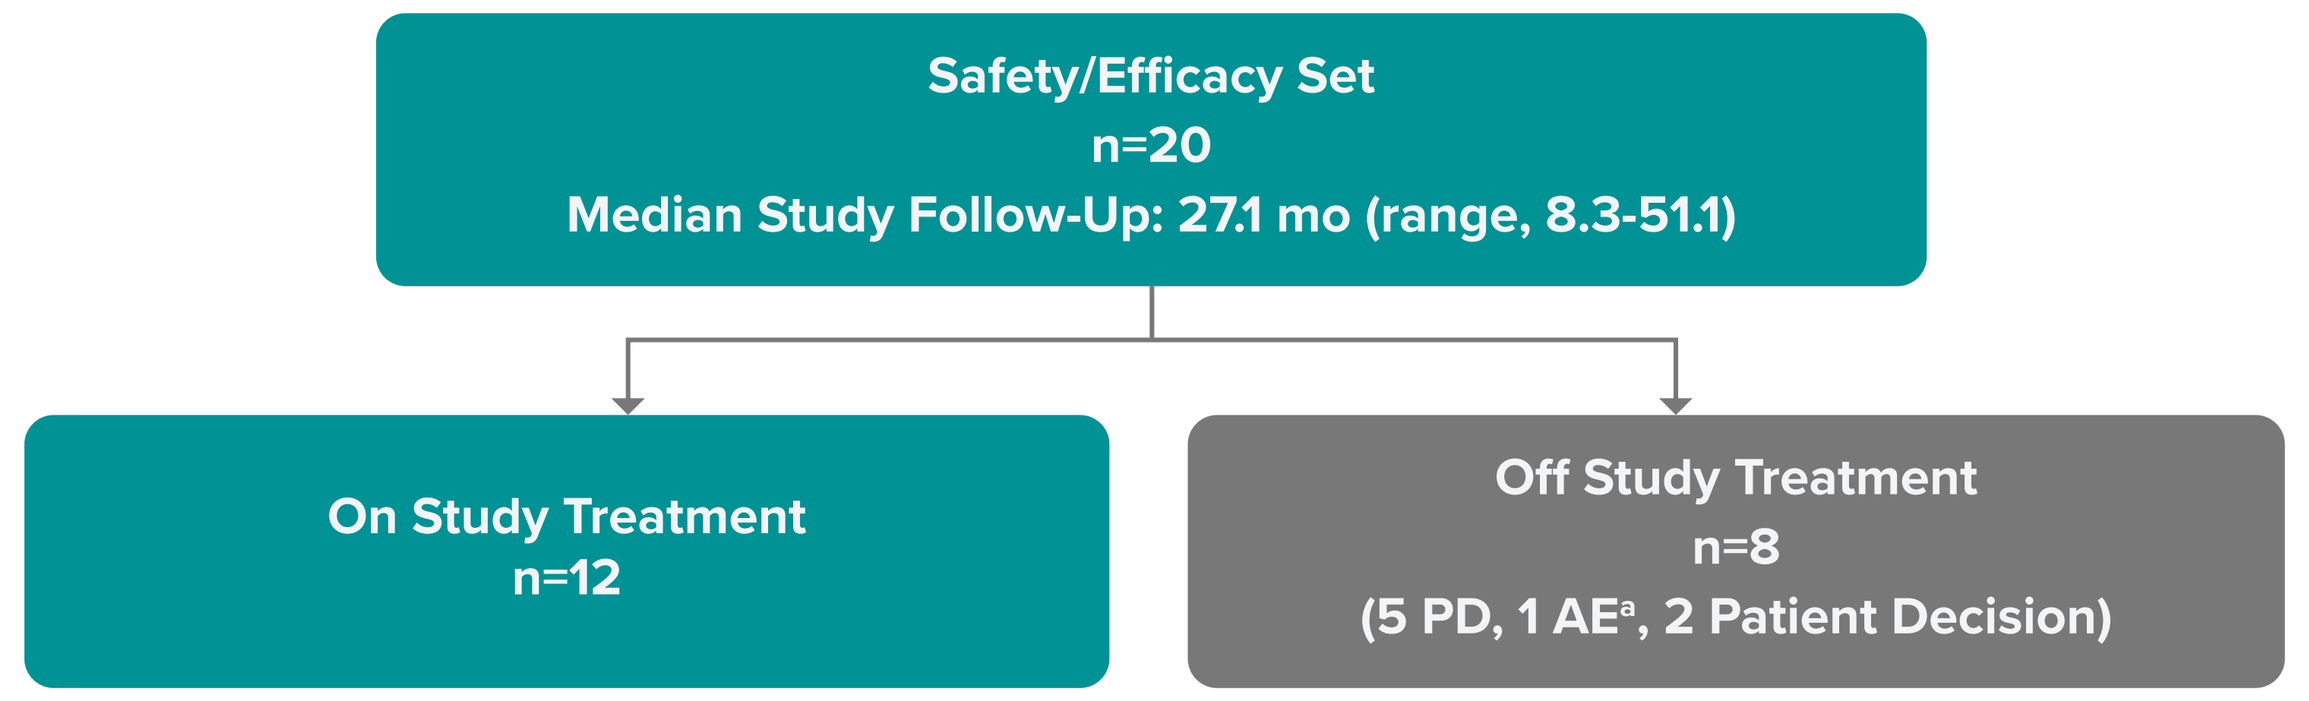
Safety/Efficacy Set
n=20
Median Study Follow-Up: 27.1 mo (range, 8.3-51.1)
On Study Treatment
n=12
Off Study Treatment
n=8
(5 PD, 1 AE[a], 2 Patient Decision)

AE, adverse event; PD, progressive disease.
[a]Grade 3 diarrhea.

Table 1. Patient and Disease Characteristics

Characteristic	R/R MZL (N=20)
Age, y	
Median (range)	69.5 (52-85)
>75 y, n (%)	3 (15)
ECOG PS, n (%)	
0/1	18 (90)
2	2 (10)
Bulky disease, n (%)	
LDi >5 cm	5 (25)
Bone marrow involvement, n (%)[a]	13 (65)
Extra nodal disease, n (%)[b]	20 (100)
No. of prior systemic therapies, median (range)	2 (1-5)
Subtype, n (%)	
Extranodal	9 (45)
Nodal	5 (25)
Splenic	6 (30)

ECOG PS, Eastern Cooperative Oncology Group performance status; LDi, longest diameter; MZL, marginal zone lymphoma; R/R, relapsed/refractory.
[a]Derived from baseline tumor biopsy/aspiration per investigator assessment.
[b]Extranodal disease is defined as patients with extranodal baseline target or nontarget lesions, or bone marrow involvement by biopsy per investigator assessment.

Table 2. Best Overall Response by Investigator Assessment

Best Response	R/R MZL (N=20)
ORR[a] n (%) [95% CI]	16 (80) [56.3, 94.3]
CR, n (%)	3 (15)
PR, n (%)	13 (65)
SD, n (%)	3 (15)
PD, n (%)	1 (5)
Median (range) TTR (≥PR), mo	2.8 (2.6-39.6)
Median (range) study follow-up, mo	27.1 (8.3-51.1)
18-mo DOR, % [95% CI]	66.2 [32.4, 86]
24-mo PFS, % [95% CI]	59.4 [33, 79]
24-mo OS, % [95% CI]	83.2 [56, 94]

CR, complete response; DOR, duration of response; MZL, marginal zone lymphoma; ORR, overall response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; R/R, relapsed/refractory; SD, stable disease; TTR, time to response.
[a]ORR includes PR and CR.

Table 3. Best Overall Response by MZL Subtype

Best Response, n (%)[a]	Extranodal (n=9)	Nodal (n=5)	Splenic (n=6)
ORR [95% CI]	7 (77.8) [40.0, 97.2]	5 (100) [47.8, 100.0]	4 (66.7) [22.3, 95.7]
CR	1 (11.1)	2 (40)	0 (0)
PR	6 (66.7)	3 (60)	4 (67)
SD	2 (22.2)	0 (0)	1 (16.7)
PD	0 (0)	0 (0)	1 (16.7)

CR, complete response; ORR, overall response rate; PD, progressive disease; PR, partial response; SD, stable disease.
[a]ORR includes PR and CR.

Figure 3: SPD Change From Baseline by MZL Subtype

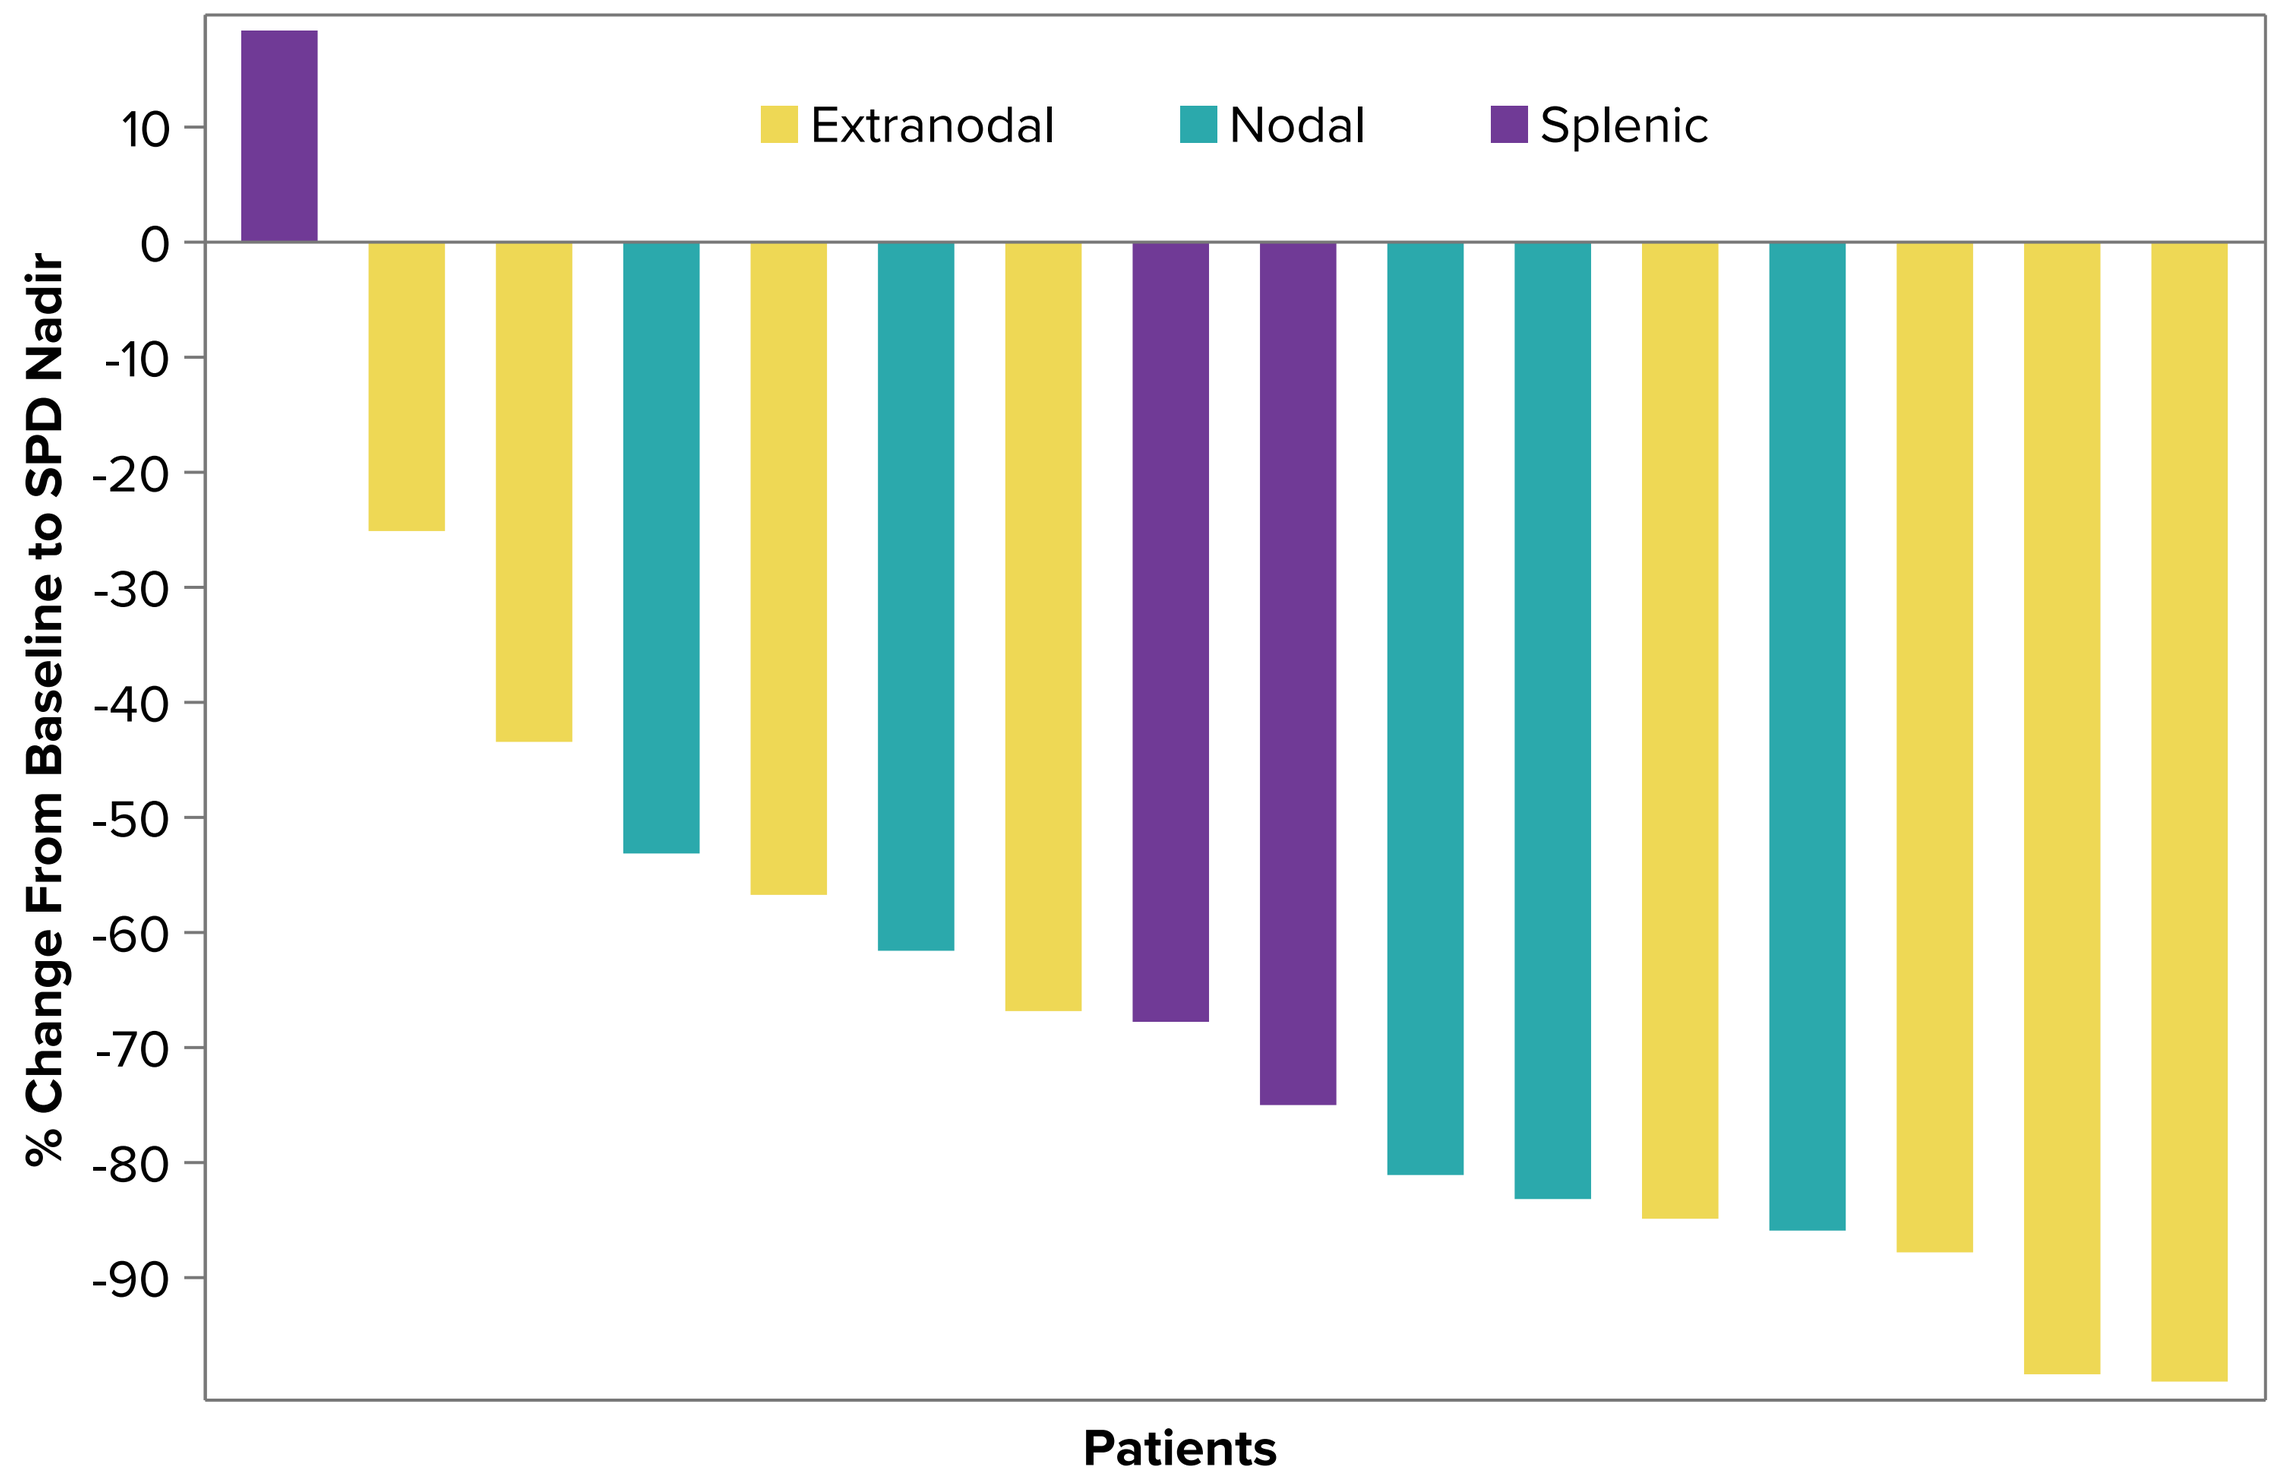
Extranodal
Nodal
Splenic
% Change From Baseline to SPD Nadir
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
Patients

Data cutoff: January 29, 2020. Note: 1 patient had inconsistent lymph nodes reported at baseline and post baseline and was not included in the analysis.
MZL, marginal zone lymphoma; SPD, sum of the products of diameters.

Figure 4: Duration of Response in Patients With MZL

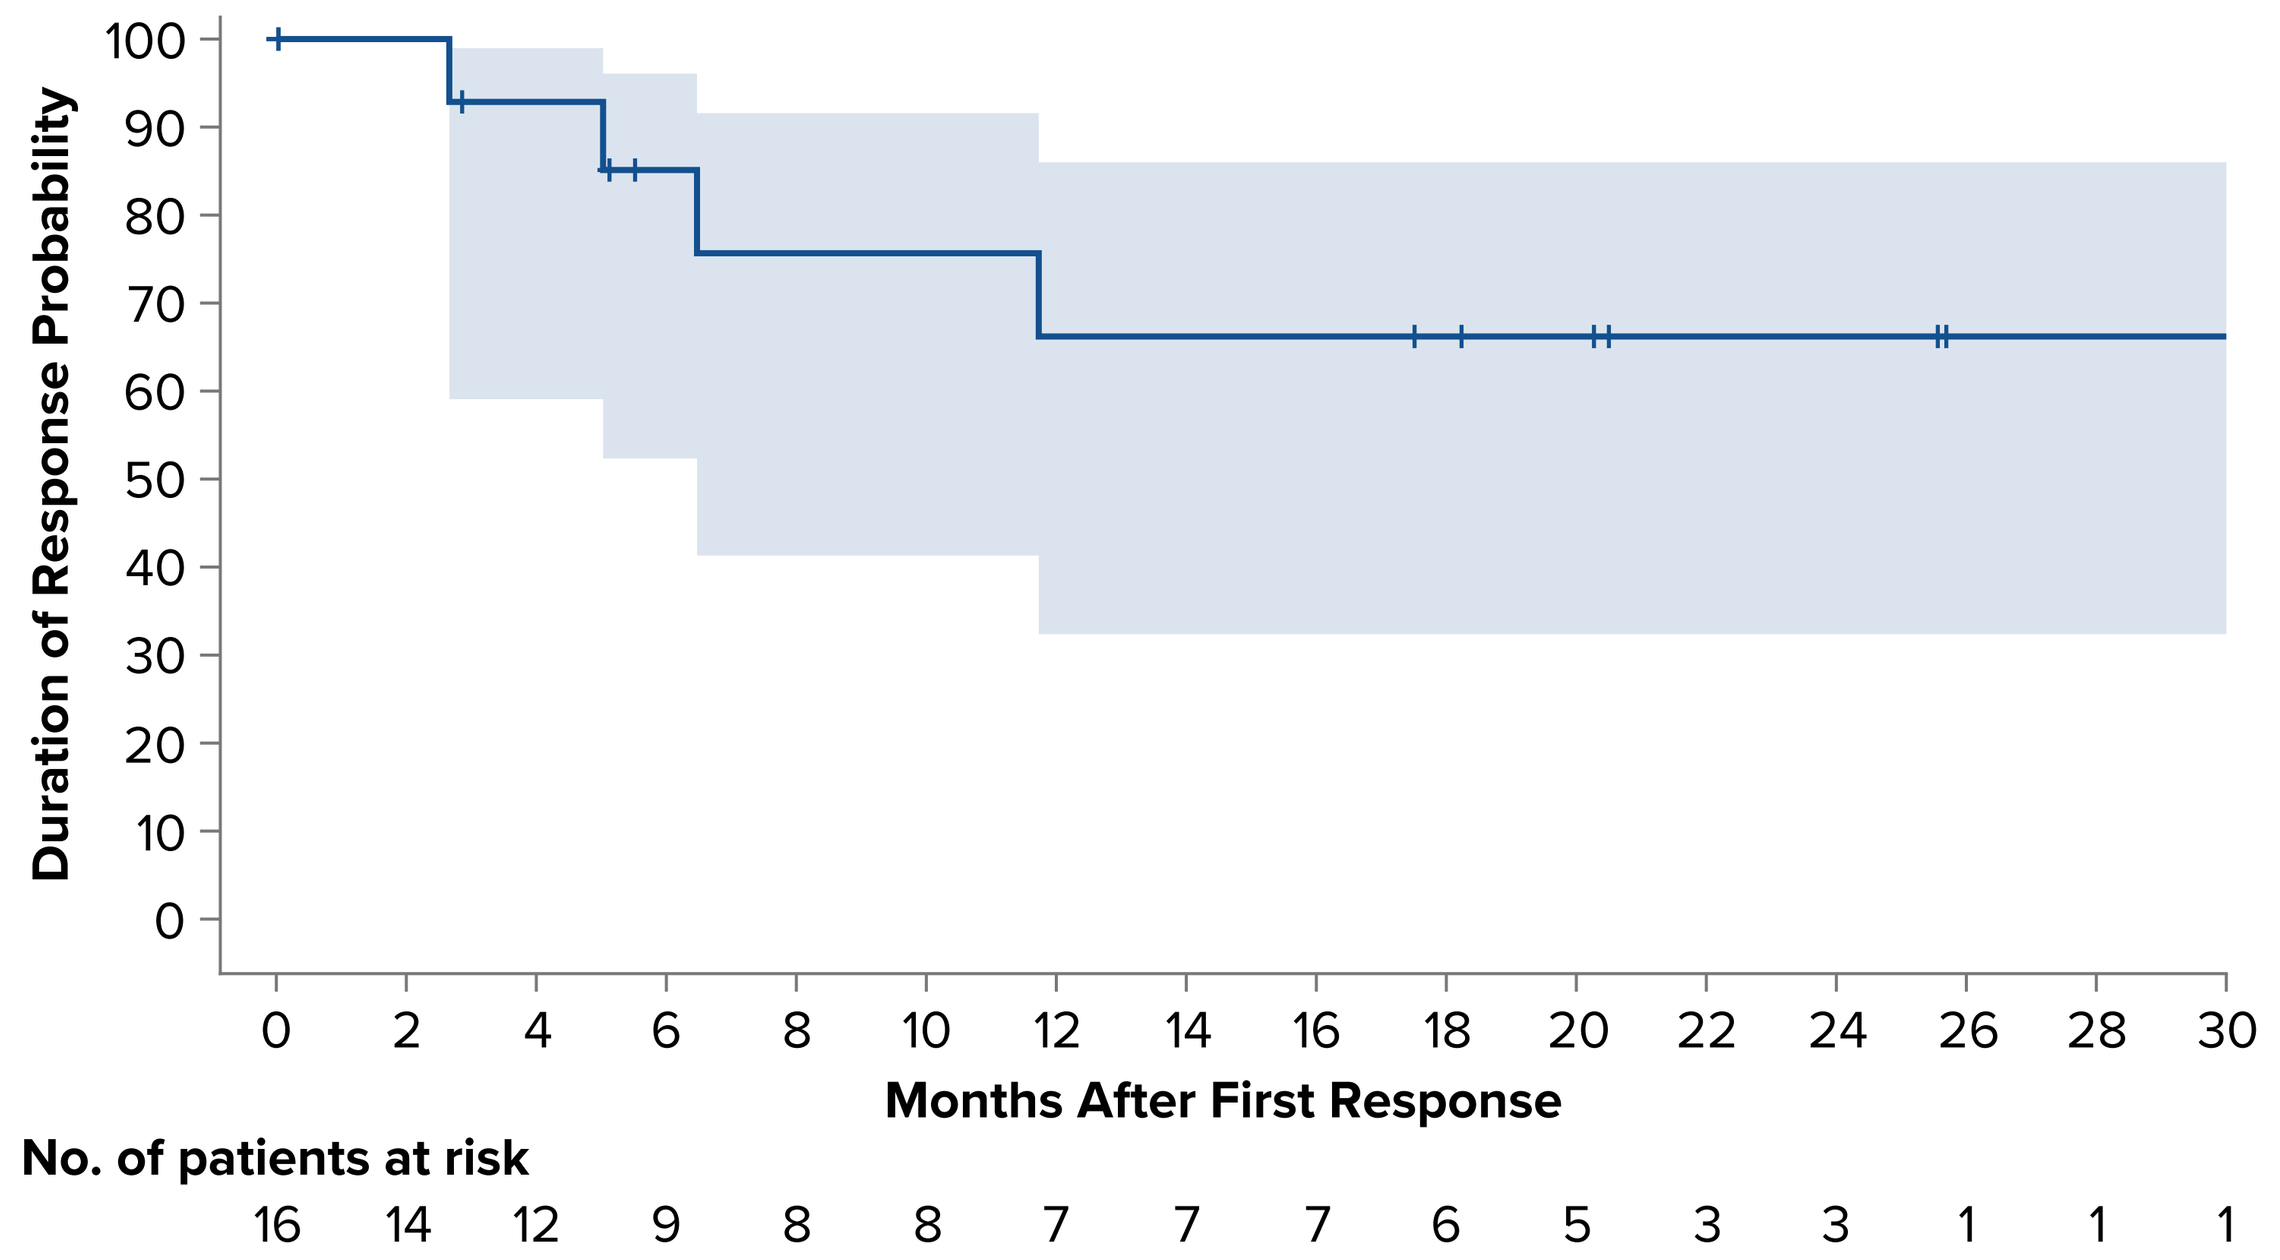
Duration of Response Probability
100
90
80
70
60
50
40
30
20
10
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Months After First Response
No. of patients at risk
16 14 12 9 8 8 7 7 7 6 5 3 3 1 1 1

Data cutoff: January 29, 2020. Note: Shaded area shows the 95% CI.
MZL, marginal zone lymphoma.

Figure 5. Progression-Free Survival in Patients With MZL

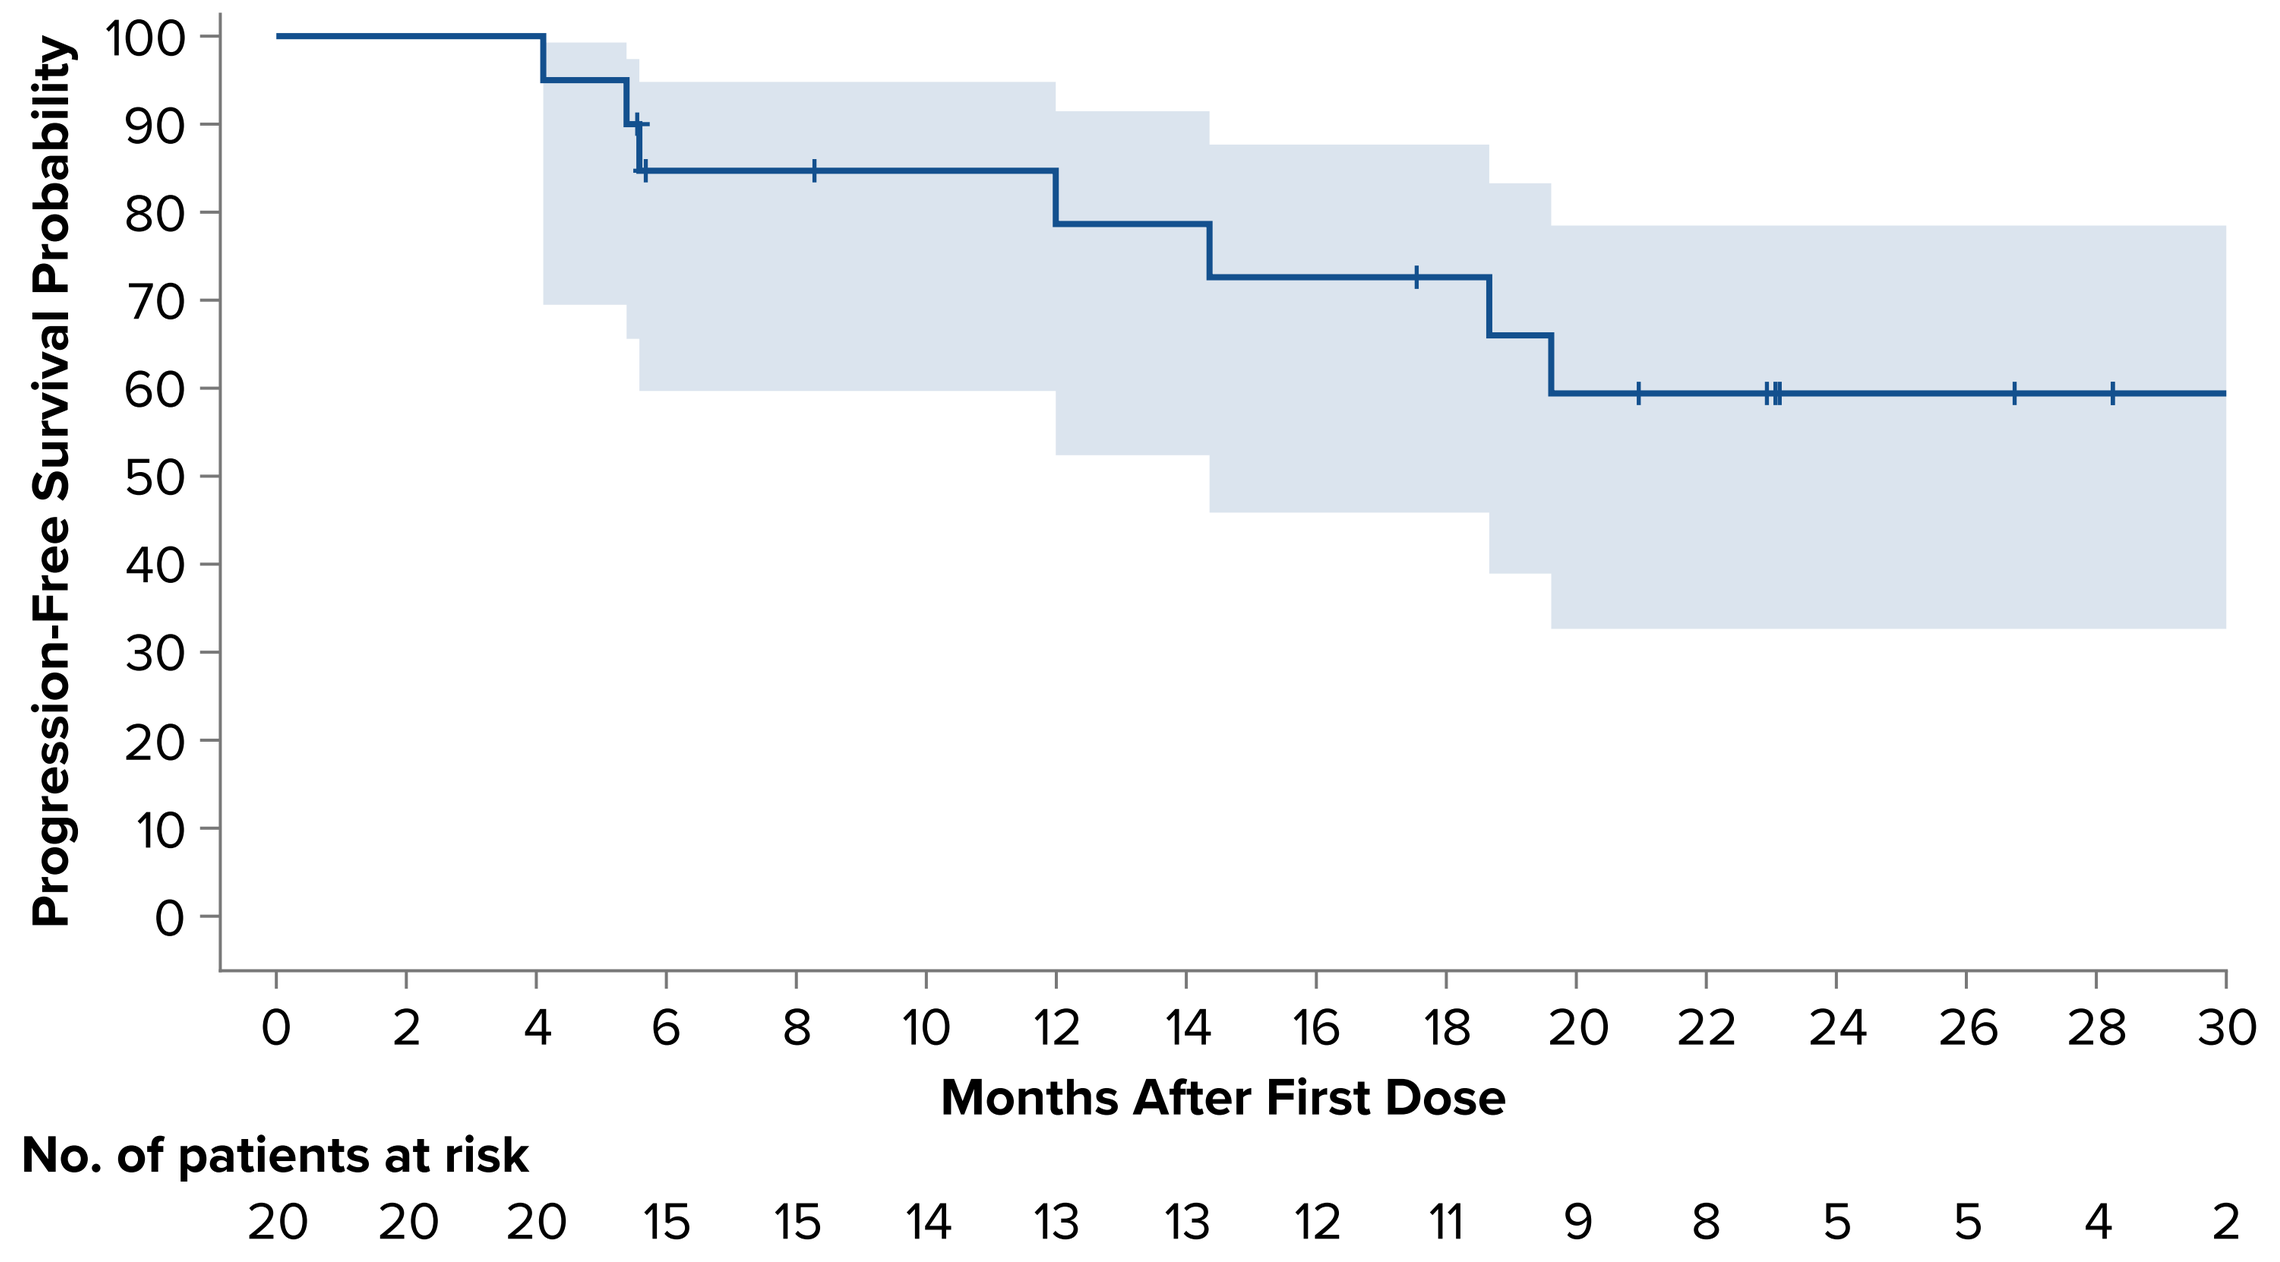
Progression-Free Survival Probability
100
90
80
70
60
50
40
30
20
10
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Months After First Dose
No. of patients at risk
20 20 20 15 15 14 13 13 12 11 9 8 5 5 4 2

Data cutoff: January 29, 2020. Note: Shaded area shows the 95% CI.
MZL, marginal zone lymphoma.

Table 4: Safety Summary

Event, n (%)	R/R MZL (N=20)
Any AE	20 (100)
Grade ≥3 AE	10 (50)
Serious AE	9 (45)
AEs leading to treatment discontinuation	1 (5)[a]
AEs leading to death	0 (0)

Data cutoff: January 29, 2020
AE, adverse event; MZL, marginal zone lymphoma; R/R, relapsed/refractory.
[a]Diarrhea (grade 3).

Figure 6: Common Adverse Events Regardless of Causality

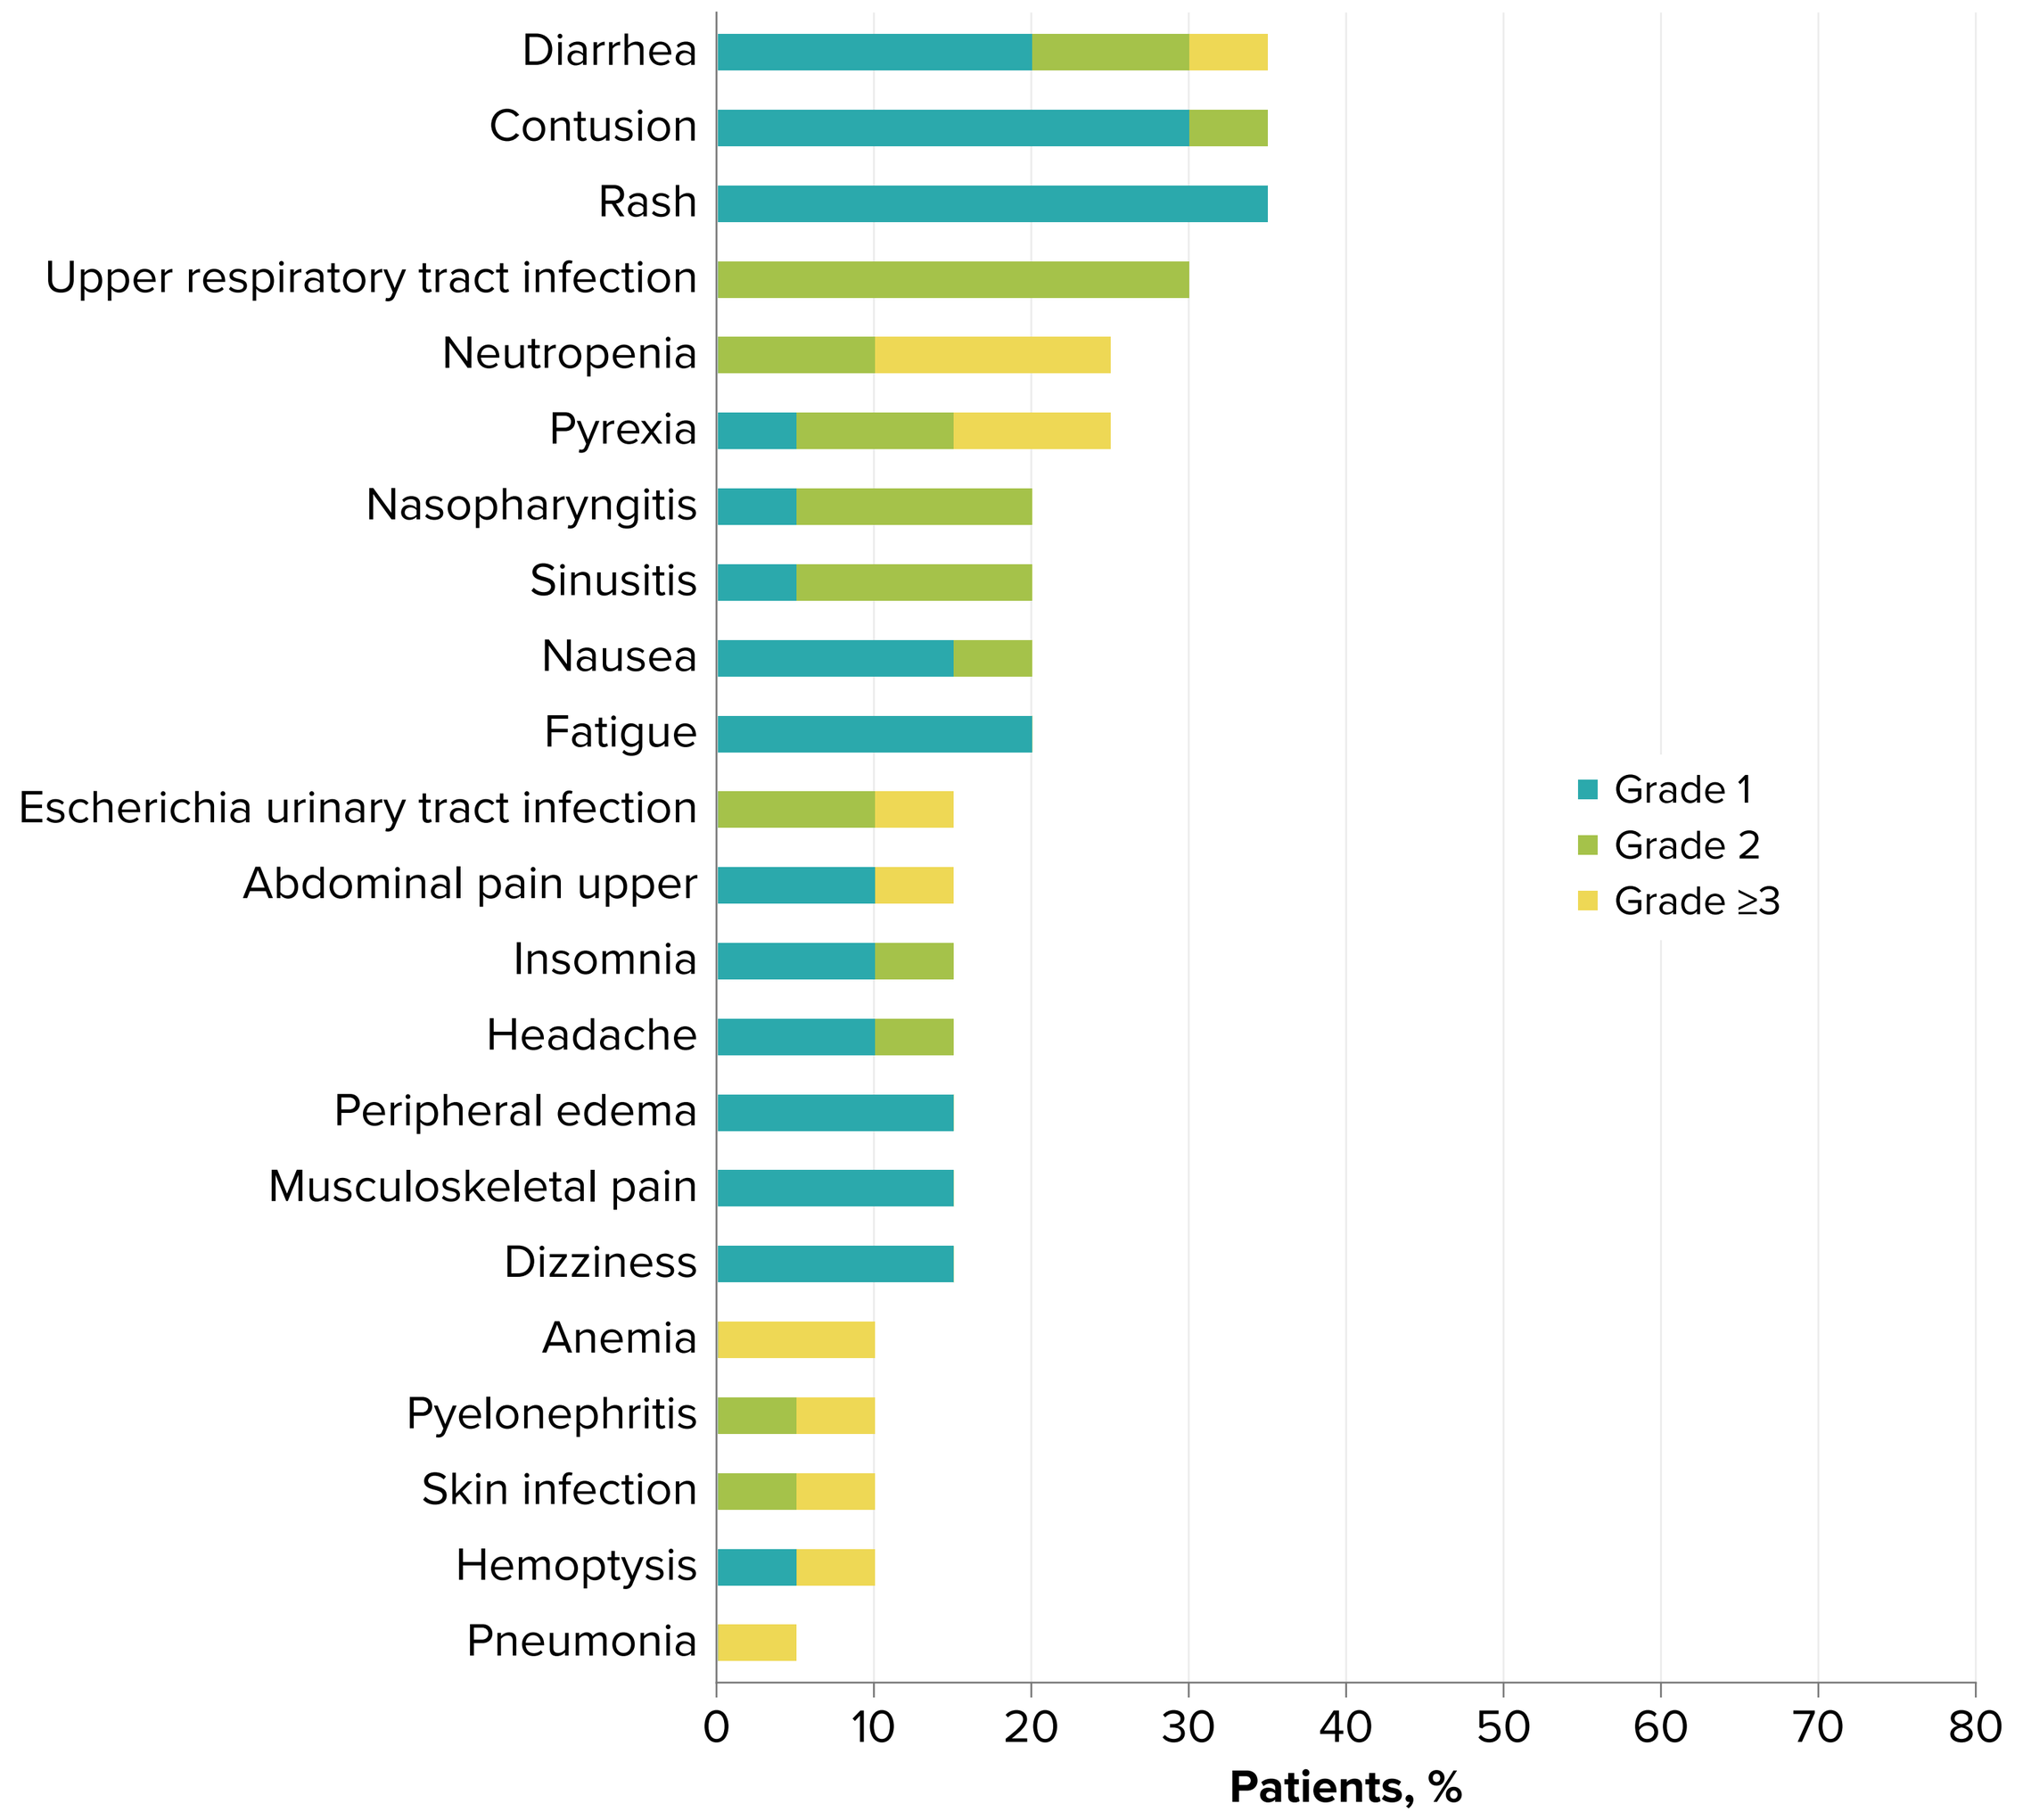
Diarrhea
Contusion
Rash
Upper respiratory tract infection
Neutropenia
Pyrexia
Nasopharyngitis
Sinusitis
Nausea
Fatigue
Escherichia urinary tract infection
Abdominal pain upper
Insomnia
Headache
Peripheral edema
Musculoskeletal pain
Dizziness
Anemia
Pyelonephritis
Skin infection
Hemoptysis
Pneumonia
Grade 1
Grade 2
Grade ≥3
0 10 20 30 40 50 60 70 80
Patients, %

Data cutoff: January 29, 2020. Adverse events by Preferred Term. Note: all grade >10% of patients; grade ≥3 in ≥2% of patients.

Figure 7: Adverse Events of Interest[a]

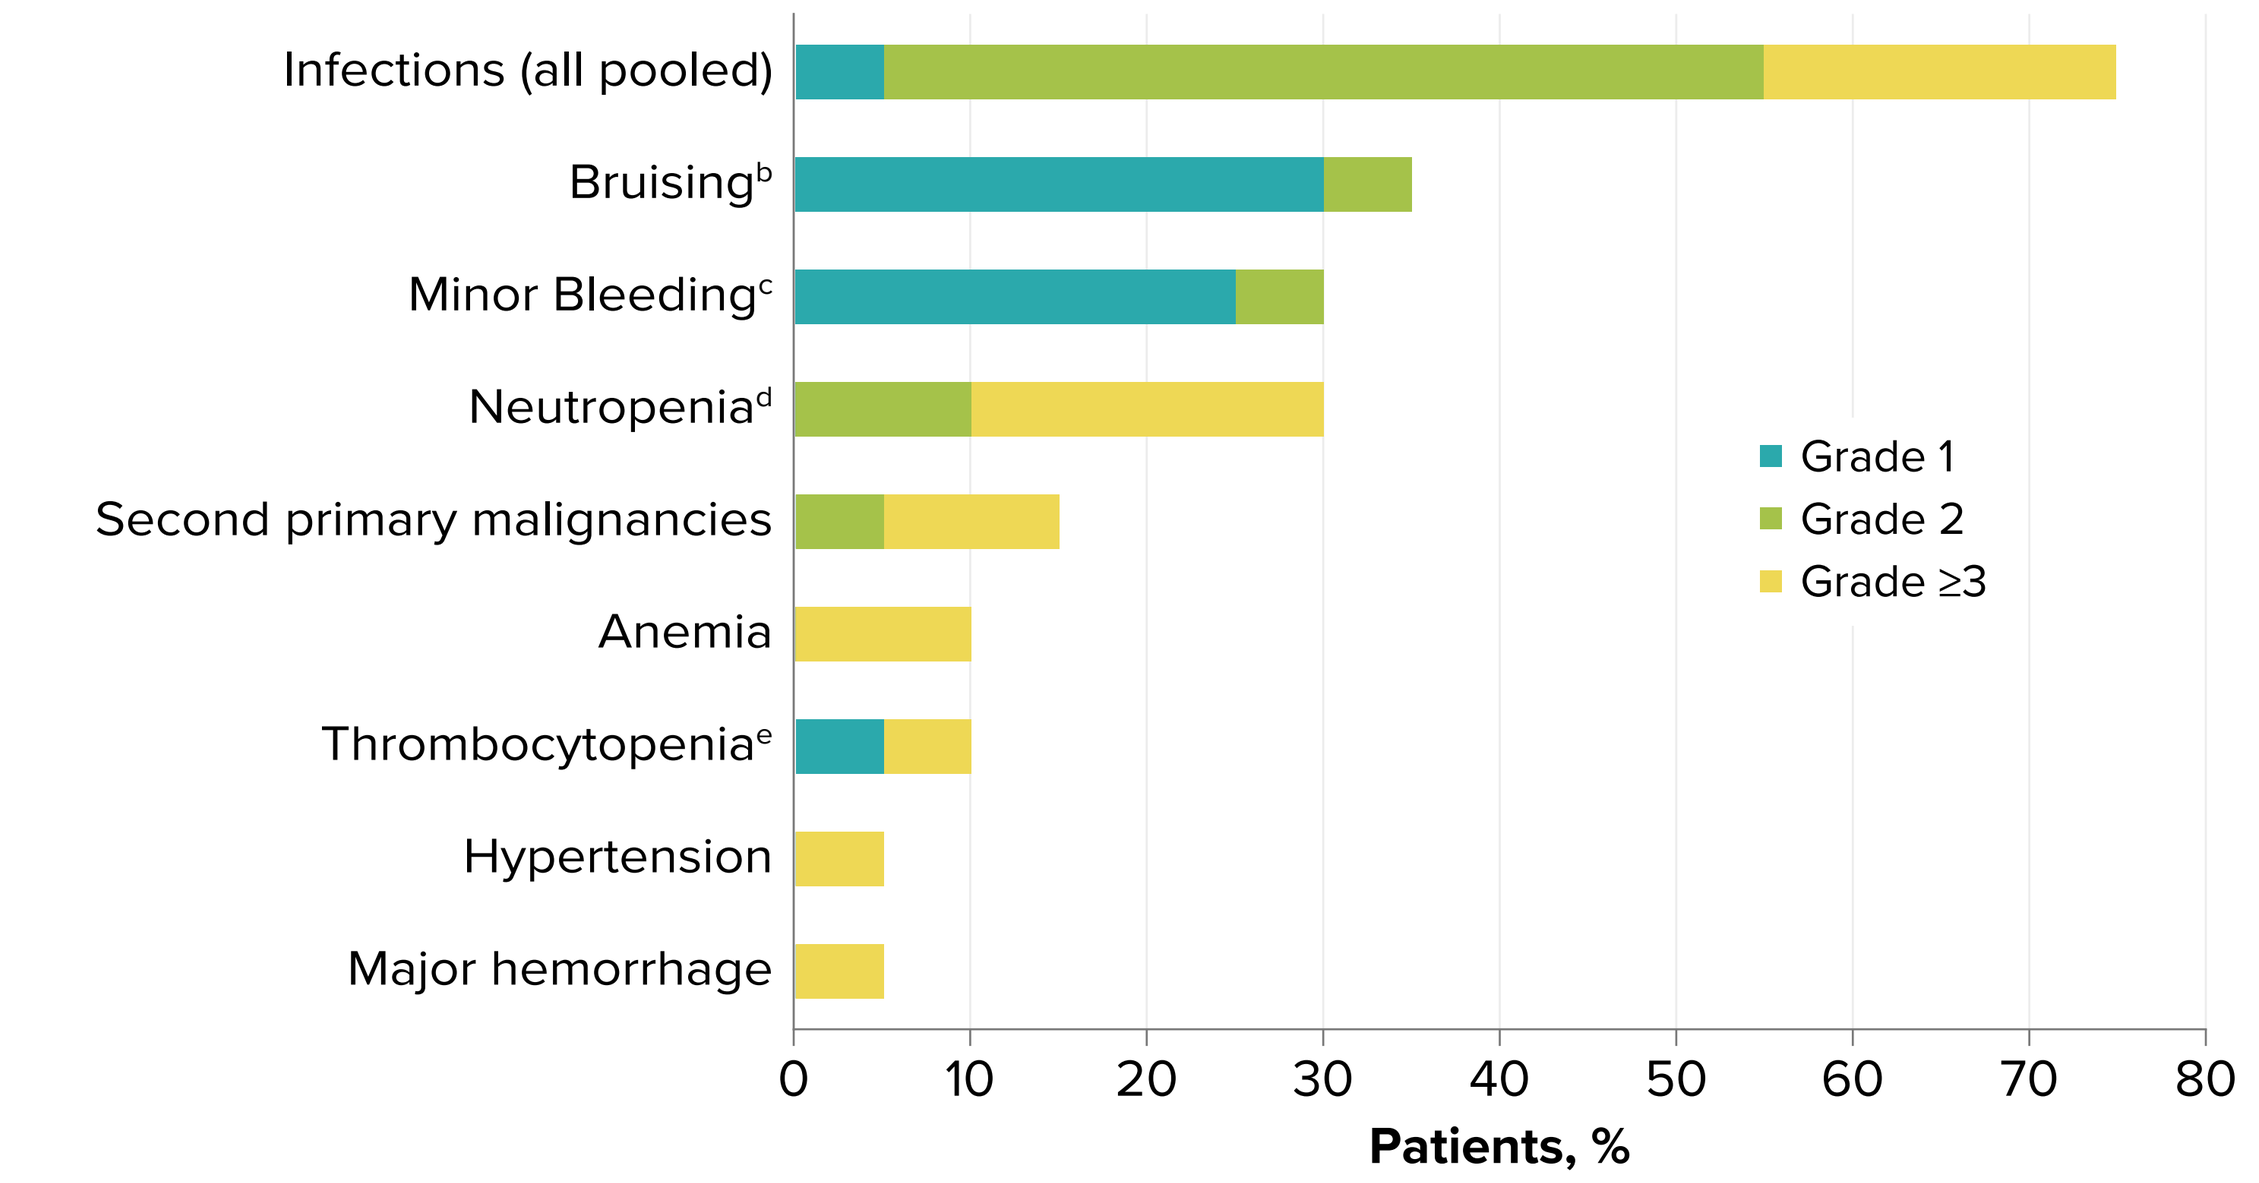
Infections (all pooled)
Bruising[b]
Minor Bleeding[c]
Neutropenia[d]
Second primary malignancies
Anemia
Thrombocytopenia[e]
Hypertension
Major hemorrhage
Grade 1
Grade 2
Grade ≥3
0 10 20 30 40 50 60 70 80
Patients, %

Data cutoff: Jan 29, 2020. Note: no atrial fibrillation/flutter was reported.
[a]Pooled terms where appropriate.
[b]Contusion, purpura, increased tendency to bruise, and ecchymosis.
[c]Pooled bleeding terms.
[d]Neutropenia, neutrophil count decreased, or febrile neutropenia.
[e]Thrombocytopenia or platelet count decreased.
Note: no atrial fibrillation/flutter was reported.

CONCLUSIONS

- Zanubrutinib therapy demonstrated a favorable safety profile in patients with R/R MZL and resulted in durable responses
- Responses were observed in all MZL subtypes
- At a median follow-up of 27.1 months:
 - ORR was 80%, with a CR rate of 15%
 - All 5 patients with nodal MZL subtype achieved a response (ORR: 100%)
 - Median time to first response was 2.8 months
 - 66% of responders were still responding to treatment at 18 months
 - PFS rate at 24 months was 59.4%
 - One patient discontinued treatment due to an AE
 - No patients with MZL experienced grade 5 AEs
 - No patients with MZL experienced atrial fibrillation/flutter

REFERENCES

1. Zucca E, et al. *Ann Oncol*. 2020;31:17-21.
2. Rickert RC. *Nat Rev Immunol*. 2013;13:578-591.
3. Choe H, Ruan J. *Oncology (Williston Park)*. 2016;30:847-858.
4. Aalipour A, Advani RH. *Br J Haematol*. 2013;163:436-443.
5. Noy A, et al. *Blood*. 2017;129:2224-2232.
6. Tam CS, et al. *Blood*. 2019;134:851-859.

CORRESPONDENCE

alessandra.tedeschi@ospedaleniguarda.it

DISCLOSURES

AT: Honoraria from AbbVie, AstraZeneca, and Janssen; consulting/advisory role with AbbVie, AstraZeneca, and Janssen.

JT: Research funding from BeiGene, Celgene, A Bristol-Myers Squibb Company, Pharmacyclics, Roche, and Takeda.

CST: Research funding from AbbVie, BeiGene, Janssen, Pharmacyclics, TG Therapeutics; consulting/advisory role with AbbVie, BeiGene, Janssen, LOXO, and Roche.

DS: Employment and stock option with BeiGene; honoraria from AbbVie, Janssen, and Roche; research funding from AbbVie, Acerta, Amgen, BeiGene, Celgene, A Bristol-Myers Squibb Company, GSK, MSD, Pharmacyclics, and Sanofi; travel expenses from AbbVie.

H-SE: has nothing to disclose.

RE, ZT, SA, and **WN:** Employment and stock options with BeiGene.

JCC: Employment and stock options with BeiGene and consulting/advisory role with Acadia Pharmaceuticals.

JH: Employment, leadership role, and stock options with BeiGene.

SO: Honoraria from and consulting/advisory role with AbbVie, AstraZeneca, Merck, Gilead, Janssen, Novartis, and Roche; research funding from Amgen, AstraZeneca, BeiGene, Epizyme, Janssen, and Roche; travel expenses from Roche.

ACKNOWLEDGMENTS

We thank the investigators, site support staff, and especially the patients for participating in this study.

This study was sponsored by BeiGene; editorial support was provided by Bio Connections, LLC and funded by BeiGene.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.

Presented at the 25th Congress of the European Hematology Association, June 11-14, 2020; Virtual Format