SAFETY, ANTITUMOR ACTIVITY, AND PHARMACOKINETICS OF PAMIPARIB (BGB-290), A PARP1/2 INHIBITOR, IN PATIENTS WITH ADVANCED SOLID TUMORS: UPDATED PHASE 1 DOSE-ESCALATION/EXPANSION RESULTS

Mark Voskoboynik,[1] Linda Mileshkin,[2] Hui Gan,[3] Michael Millward,[4] George Au-Yeung,[2] Tarek M. Meniawy,[4] Ganessan Kichenadasse,[5] Kathy Zhang,[6] Jang Yun,[6] Maggie Zhang,[6] Song Mu,[6] Jason D. Lickliter[1]

[1]Oncology, Nucleus Network, Melbourne, VIC, Australia; [2]Department of Medical Oncology, Peter MacCallum Cancer Centre, Melbourne, VIC, Australia; [3]Austin Hospital, Heidelberg, VIC, Australia; [4]Oncology, Linear Clinical Research, Nedlands, WA, Australia; [5]Flinders Centre for Innovation in Cancer, Flinders Medical Centre, Bedford Park, SA, Australia; [6]BeiGene USA, Inc., Emeryville, CA, USA

POSTER 452PD
European Society for Medical Oncology Congress
27 September–1 October, 2019, Barcelona, Spain

BACKGROUND

- Poly(ADP-ribose) polymerase (PARP) proteins play a key role in the repair of single-strand (ss) and double-strand (ds) DNA breaks[1,2]
- Normal cells repair DNA breaks using base-excision repair (BER) and homologous recombination (HR) pathways; cancer cells that are HR deficient (HRD$^+$) are unable to repair dsDNA breaks
- PARP inhibition impairs DNA repair and traps PARP proteins on damaged DNA, resulting in cytotoxicity that is exacerbated in HRD$^+$ cells (synthetic lethality)[3–9]
- Pamiparib is an investigational PARP1/2 inhibitor that has demonstrated brain penetration and PARP-DNA complex–trapping capabilities in preclinical studies[10]
- In the phase 1, dose-escalation/expansion study of patients with advanced solid tumors, pamiparib was generally well-tolerated and showed preliminary antitumor activity[11]
- Here, we report updated safety data from the study and updated efficacy data from the ovarian and associated cancer cohort

METHODS

Study Design

- This is a two-stage dose-escalation/expansion study (Figure 1)
 - The dose-escalation component established the safety, tolerability, maximum tolerated dose (MTD), recommended phase 2 dose (RP2D), and pharmacokinetic (PK) profile of pamiparib
 - MTD was identified as 80 mg PO BID and RP2D was established as 60 mg PO BID
 - The dose-expansion component was conducted in patients with ovarian and associated cancers and other solid tumors

Figure 1: Study Design

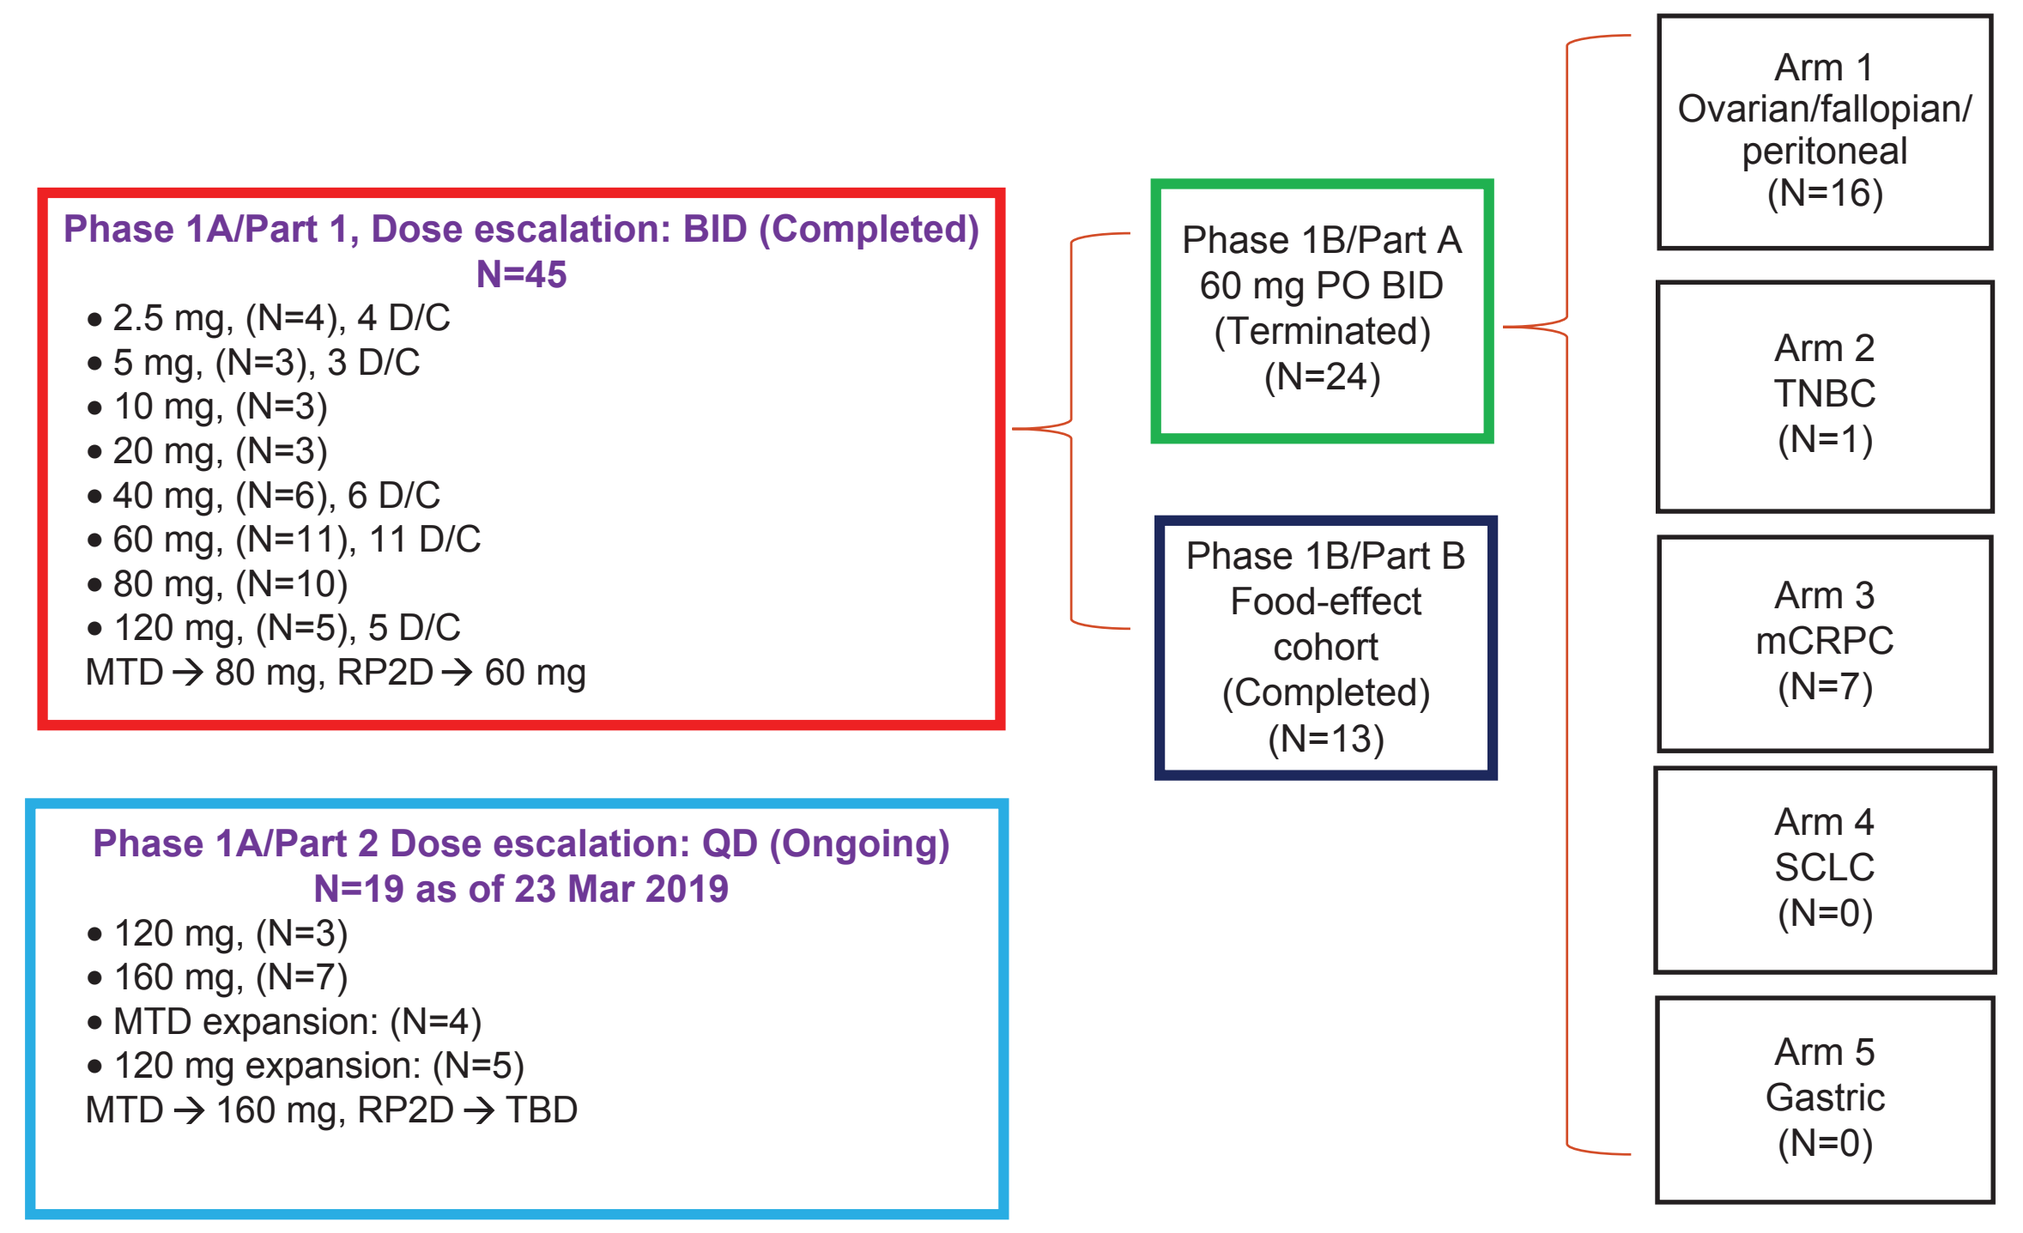
Phase 1A/Part 1, Dose escalation: BID (Completed)
N=45
• 2.5 mg, (N=4), 4 D/C
• 5 mg, (N=3), 3 D/C
• 10 mg, (N=3)
• 20 mg, (N=3)
• 40 mg, (N=6), 6 D/C
• 60 mg, (N=11), 11 D/C
• 80 mg, (N=10)
• 120 mg, (N=5), 5 D/C
MTD → 80 mg, RP2D → 60 mg
Phase 1A/Part 2 Dose escalation: QD (Ongoing)
N=19 as of 23 Mar 2019
• 120 mg, (N=3)
• 160 mg, (N=7)
• MTD expansion: (N=4)
• 120 mg expansion: (N=5)
MTD → 160 mg, RP2D → TBD
Phase 1B/Part A
60 mg PO BID
(Terminated)
(N=24)
Phase 1B/Part B
Food-effect
cohort
(Completed)
(N=13)
Arm 1
Ovarian/fallopian/
peritoneal
(N=16)
Arm 2
TNBC
(N=1)
Arm 3
mCRPC
(N=7)
Arm 4
SCLC
(N=0)
Arm 5
Gastric
(N=0)

Abbreviations: BID=twice daily, D/C=discontinued, mCRPC=metastatic castration-resistant prostate cancer, MTD=maximum tolerated dose, QD=once daily, PO=per orem, RP2D=recommended phase 2 dose, SCLC=small cell lung cancer, TBD=to be determined, TNBC=triple-negative breast cancer

Study Assessments and Analyses

- Antitumor activity was assessed in all evaluable patients based on RECIST v1.1 criteria
- Safety and tolerability were evaluated in all patients who received ≥1 dose of pamiparib
- Safety and tolerability assessments were based on monitoring of treatment-emergent adverse events (TEAEs), physical examinations, and clinical laboratory results

RESULTS

- As of 1 June 2019, 101 patients (64 dose-escalation, 37 dose-expansion; median age, 60 years; ECOG PS of 0, 1, or 2 [36.6%, 62.4%, and 1%, respectively]) were enrolled (Table 1). Of 101 enrolled patients, 63 patients had ovarian, fallopian, or peritoneal cancer; 28 of the 63 patients received pamiparib 60 mg PO BID (the RP2D)

Table 1: Patient Disposition, Demographics, and Baseline Characteristics

Baseline characteristic	Phase 1A (n=64)	Phase 1B (n=37)	Total (N=101)
Age, mean, years (SD)	60.1 (10.08)	60.0 (11.06)	60.1 (10.40)
Gender, n (%)			
Female	51 (79.7)	29 (78.4)	80 (79.2)
Male	13 (20.3)	8 (21.6)	21 (20.8)
Race, n (%)			
Caucasian	57 (89.1)	34 (91.9)	91 (90.1)
Asian	6 (9.4)	2 (5.4)	8 (7.9)
Not reported	1 (1.6)	1 (2.7)	2 (2.0)
ECOG PS, n (%)			
0	22 (34.4)	15 (40.5)	37 (36.6)
1	41 (64.1)	22 (59.5)	63 (62.4)
2	1 (1.6)	0 (0)	1 (1.0)
Median number of prior therapies (min, max)	3.5 (1, 15)	3.0 (1, 7)	3.0 (1, 15)

Abbreviations: ECOG PS=Eastern Cooperative Oncology Group performance status, SD=standard deviation

Pharmacokinetics

- Plasma exposure increased near proportionally with increase in dose
- Reduction of 13% in AUC with a high-fat meal was not considered to be clinically relevant; patients may take pamiparib without regard to food

Figure 2: Pamiparib Plasma Concentration-Time Profiles

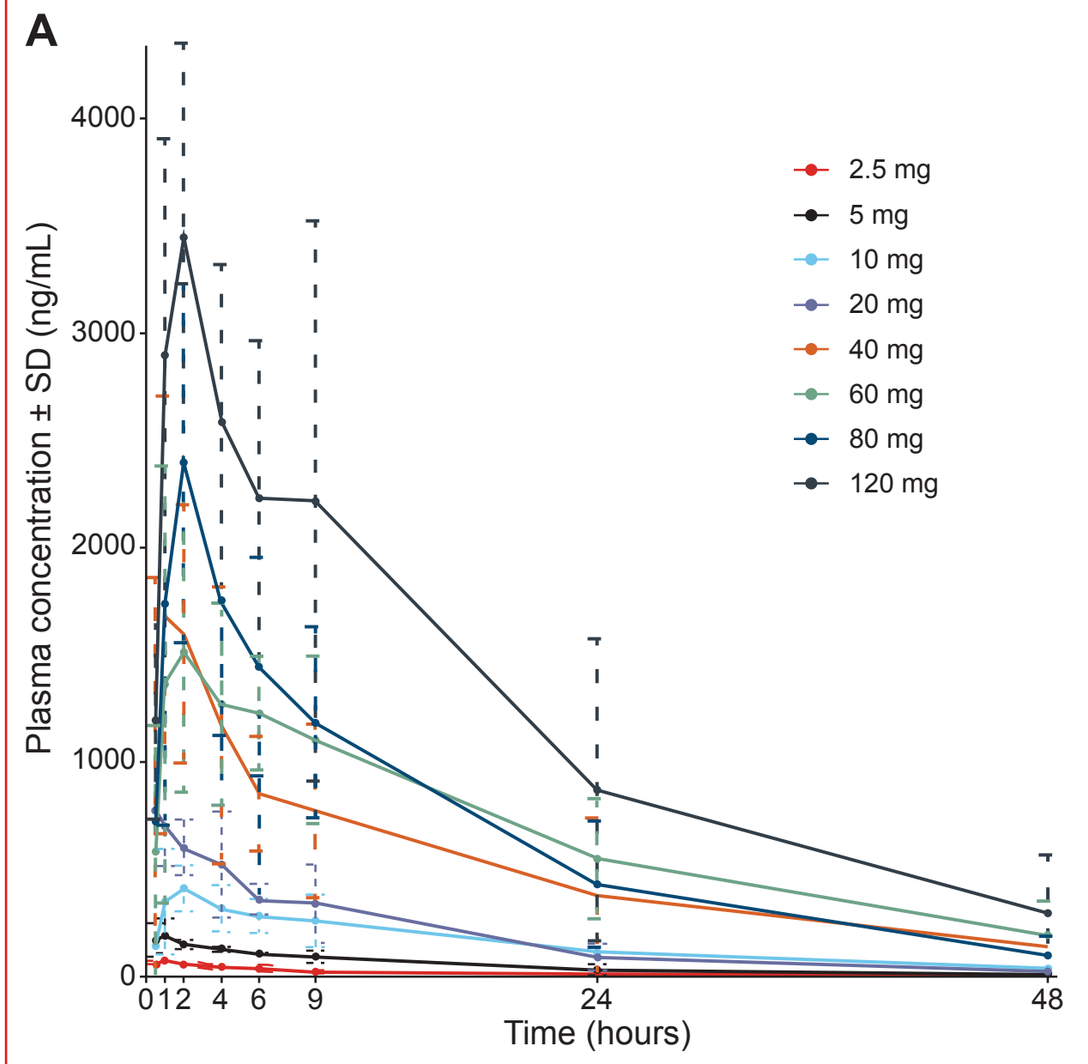
A
Plasma concentration ± SD (ng/mL)
4000
3000
2000
1000
0
0 1 2 4 6 9 24 48
Time (hours)
2.5 mg
5 mg
10 mg
20 mg
40 mg
60 mg
80 mg
120 mg

Summary PK parameters after a single-dose administration

Dose level (PO BID)	N	CYCLE 1 DAY 1: C_{max} (ng/mL)	T_{max} (h)	AUC_{inf} (ng/mL*h)	AUC_{0-9h} (ng/mL*h)	$t_{1/2}$ (h)
2.5	3	70 (29)	1.0 (0.5–1.0)	587 (35)	346 (31)	8.7 (7.5–9.5)
5	3	193 (38)	1.0 (0.6–2.0)	2431 (50)	1099 (15)	11.0 (8.3–17.3)
10	3	412 (31)	2.0 (1.1–2.0)	7371 (36)	2576 (39)	14.3 (13.5–15.3)
20	3	783 (9)	0.5 (0.5–1.0)	7843 (59)	4135 (30)	8.6 (5.8–12.0)
40	6	1755 (44)	1.0 (0.9–2.0)	21934 (86)	8955 (44)	11.9 (6.3–25.3)
60	11	1881 (27)	2.0 (1.0–9.0)	31976 (55)	10404 (29)	13.5 (7.3–24.6)
80	10	2318 (43)	2.0 (1.0–2.0)	28695 (56)	13213 (41)	9.5 (6.2–14.3)
120	5	3587 (26)	1.9 (1.0–2.0)	51815 (85)	21101 (33)	10.6 (5.5–16.9)

PK parameters presented as geometric mean and %CV except for T_{max} (median, range) and $t_{1/2}$ (geomean, range)

Summary PK parameters for the food-effect cohort

Parameter	Fed (n=13)	Fasted (n=13)	Treatment comparison GMR (90% CI)
AUC_{inf} [ng*h/mL], Geo mean (CV, %)	24860 (61)	29449 (62)	0.87 (0.76–1.00)
C_{max} [ng/mL], Geo mean (CV, %)	1185 (32)	2013 (32)	0.59 (0.53–0.66)
T_{max} [h], median (range)	7.0 (2.0–7.1)	2.0 (1.0–4.1)	
$t_{1/2}$ [h], Geo mean (range)	12.6 (5–22)	12.4 (5–23)	

Abbreviations: AUC=area under the curve, BID=twice daily, CI=confidence interval, C_{max}=maximum concentration, CV=coefficient of variation, GMR=geometric mean ratio, PK=pharmacokinetic, PO=per orem, T_{max}=time to maximum concentration, $t_{1/2}$=half-life

Efficacy in the Ovarian and Associated Cancer Cohort

- A total of 58 patients with ovarian and associated cancer were efficacy evaluable per RECIST v1.1 criteria (≥1 postbaseline tumor assessment)
 - 23 of the 58 (39.7%) patients achieved a confirmed objective response (complete response [CR], n=4; partial response [PR], n=19) (Table 2)

Table 2: Best Overall Response in Ovarian and Associated Cancer Patients

Best overall response, n (%)	Total (N=58)
Overall response rate per RECIST v1.1 (CR + PR)	**23 (39.7)**
CR	4 (6.9)
PR	19 (32.8)
SD	29 (50.0)
PD	2 (3.4)
Clinical benefit rate (CR + PR + SD with ≥24 weeks duration)	**31 (53.4)**

Abbreviations: CR=complete response, PD=progressive disease, PR=partial response, SD=stable disease

Figure 3: Best Percentage Change From Baseline in Target Lesions in Ovarian and Associated Cancer Patients

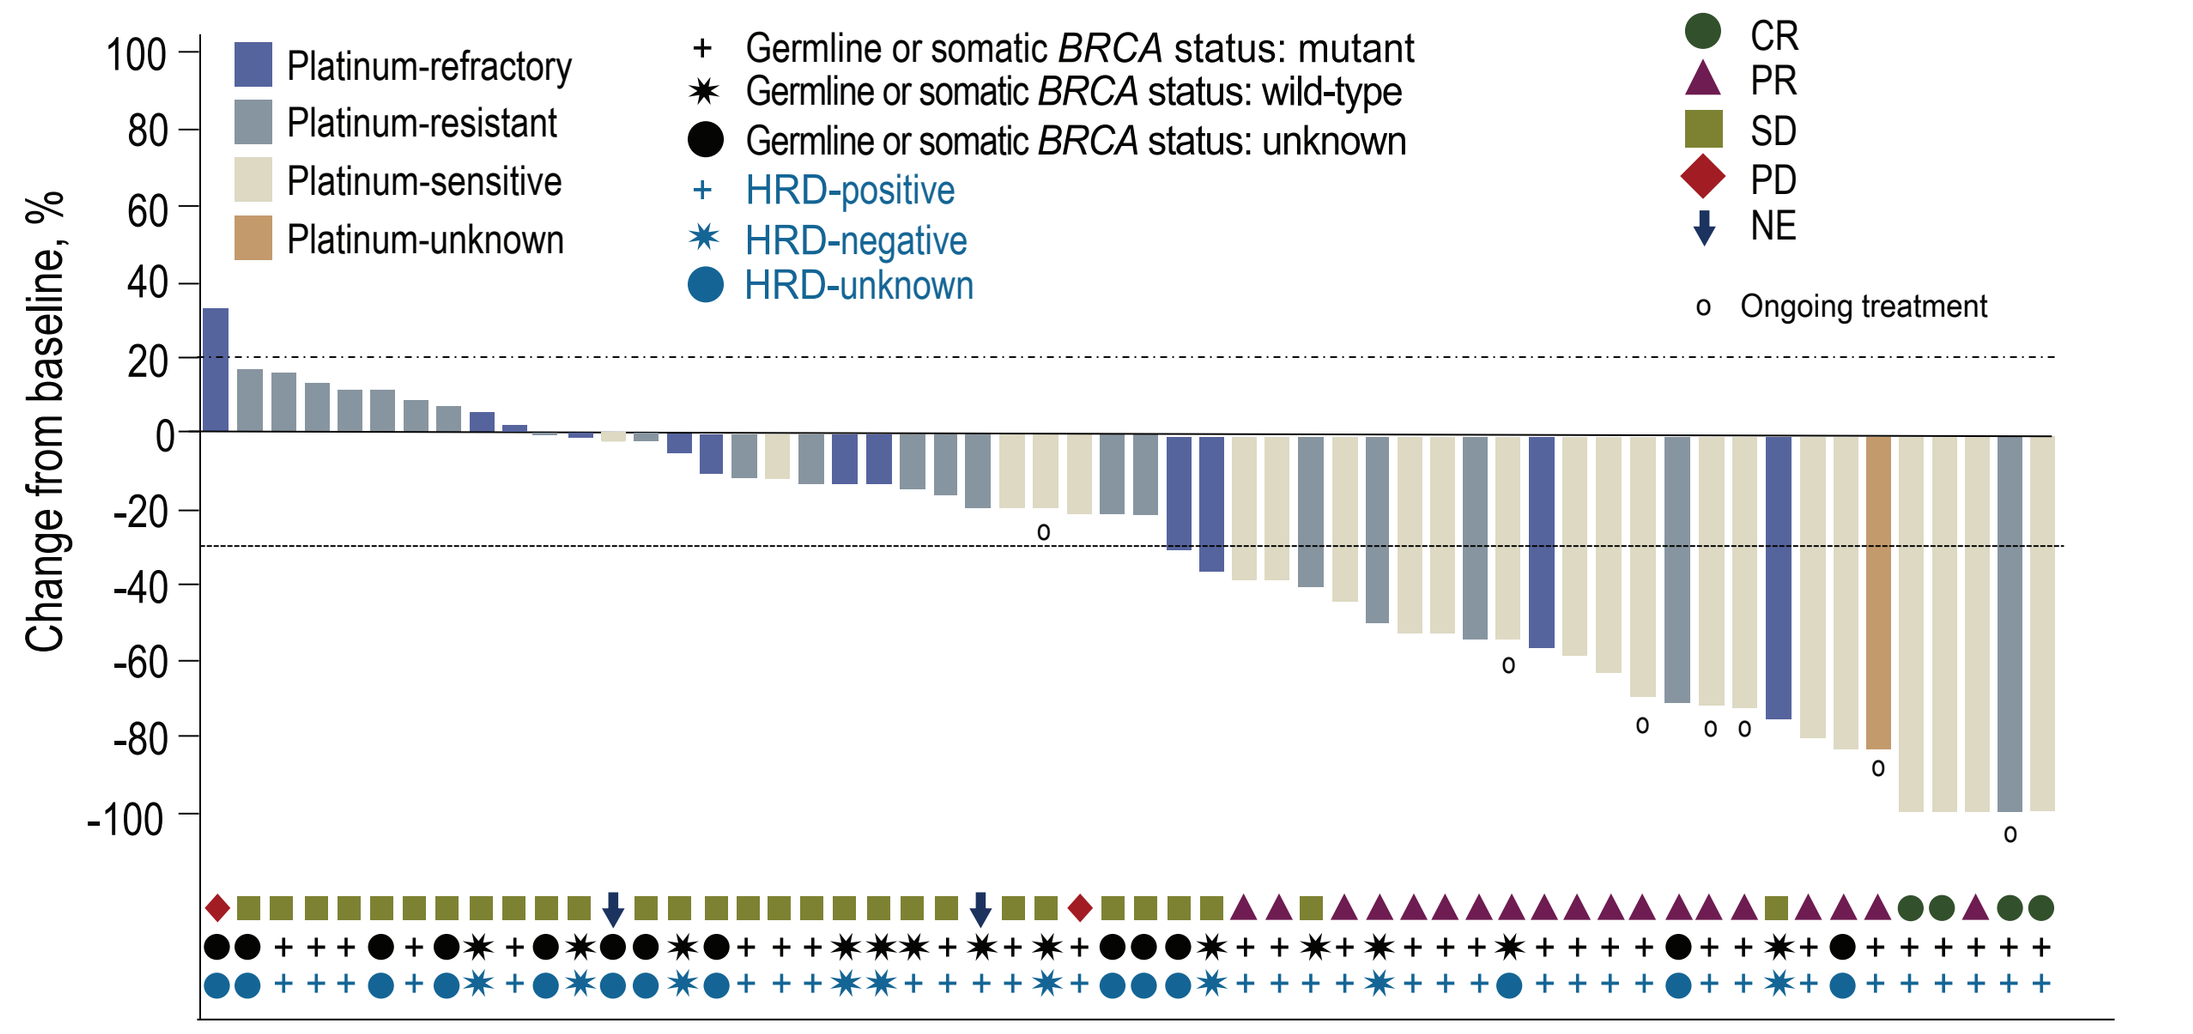
Change from baseline, %
100
80
60
40
20
0
-20
-40
-60
-80
-100
Platinum-refractory
Platinum-resistant
Platinum-sensitive
Platinum-unknown
Germline or somatic BRCA status: mutant
Germline or somatic BRCA status: wild-type
Germline or somatic BRCA status: unknown
HRD-positive
HRD-negative
HRD-unknown
CR
PR
SD
PD
NE
Ongoing treatment

Abbreviations: *BRCA*=breast cancer susceptibility gene, CR=complete response, HRD=homologous recombination deficiency, NR=not reported, ORR=objective response rate, PD=progressive disease, PR=partial response, SD=stable disease

Figure 4: Duration of Treatment in Ovarian and Associated Cancer Patients

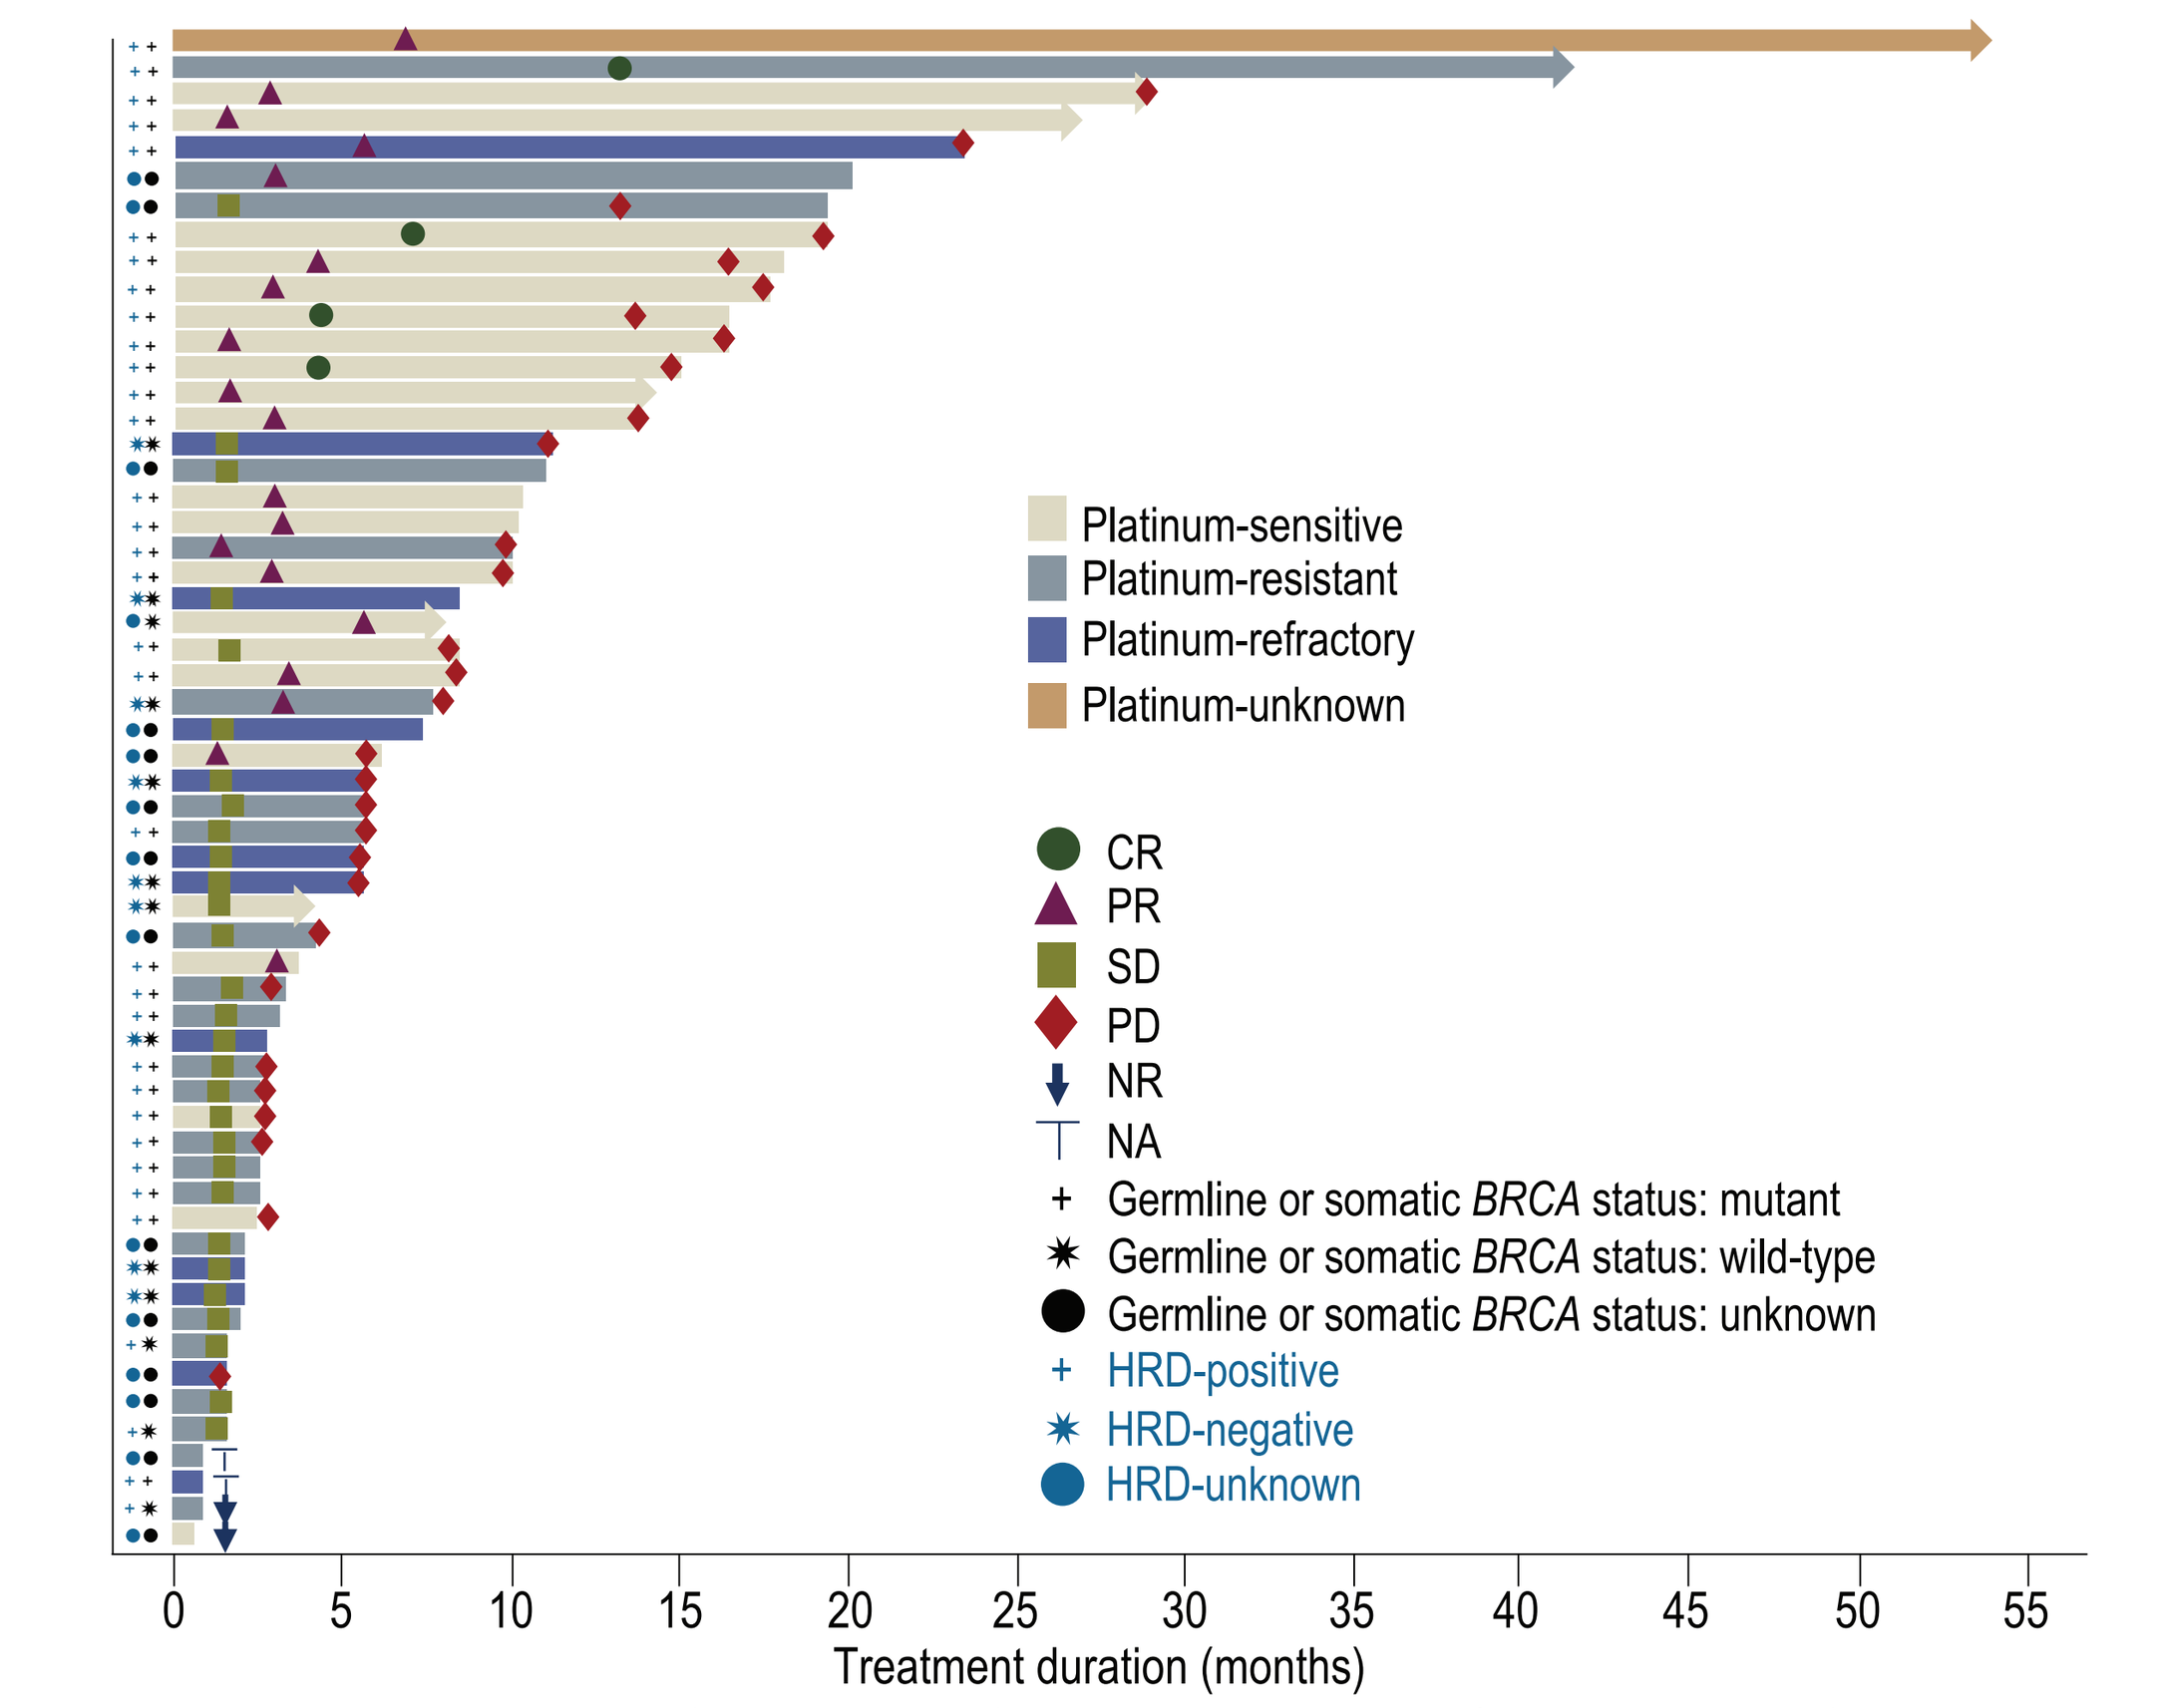
Platinum-sensitive
Platinum-resistant
Platinum-refractory
Platinum-unknown
CR
PR
SD
PD
NR
NA
Germline or somatic BRCA status: mutant
Germline or somatic BRCA status: wild-type
Germline or somatic BRCA status: unknown
HRD-positive
HRD-negative
HRD-unknown
0
5
10
15
20
25
30
35
40
45
50
55
Treatment duration (months)

Abbreviations: *BRCA*=breast cancer susceptibility gene, CR=complete response, DOR=duration of response, HRD=homologous recombination deficiency, NA=not assessed, NR=not reported, PD=progressive disease, PR=partial response, SD=stable disease

- 31 patients had a germline or somatic *BRCA* mutation (g/s *BRCA*$^{mut+}$), 27 patients were germline or somatic *BRCA* wild-type (g/s *BRCA*^{wt}) or unknown (*BRCA*^{unk})
 - The objective response rate (ORR) by g/s *BRCA* mutation status was 61.3% (19 of 31) in the g/s *BRCA*$^{mut+}$ population and 14.8% (4 of 27) in the g/s *BRCA*^{wt} or *BRCA*^{unk} population
 - CR, PR, and stable disease (SD) with tumor reductions were observed regardless of platinum-sensitivity, *BRCA* mutation, and HRD status (Figure 3)
 - Confirmed CR was achieved by 4 patients who received pamiparib 20–80 mg PO BID; PR was achieved by 19 patients (17 patients who received pamiparib 2.5–120 mg PO BID, and 2 patients who received 160 mg PO QD); 29 patients achieved SD (Figures 3 and 4)
- The clinical benefit rate was 53.4% (Table 2)
- The median duration of response was 14.9 months (range, 11.0–17.9)
- 22 patients were platinum-sensitive, 23 patients were platinum-resistant, and 12 patients were platinum-refractory (Table 3)
 - ORR by platinum-sensitivity status was 77.3% (17 of 22) in the platinum-sensitive population, 17.4% (4 of 23) in the platinum-resistant population, and 8.3% (1 of 12) in the platinum-refractory population
 - In platinum-sensitive patients, ORR was 83.3% (15 of 18) in the *BRCA*$^{mut+}$ population and 50.0% (2 of 4) in the *BRCA*^{wt}/*BRCA*^{unk} population
 - In platinum-resistant patients, ORR was 20.0% (2 of 10) in the *BRCA*$^{mut+}$ population and 15.4% (2 of 13) in the *BRCA*^{wt}/*BRCA*^{unk} population

Table 3: Objective Response Rates for Patients With Ovarian and Associated Cancer by Germline or Somatic *BRCA*/HRD Status vs. Platinum-Sensitivity Status

	Platinum-sensitive	Platinum-resistant	Platinum-refractory	Total
G/s *BRCA* status: mutant	15/18 (83.3%)	2/10 (20.0%)	1/2 (50.0%)	19/31* (61.3%)
G/s *BRCA* status: wild-type	1/2 (50.0%)	1/4 (25.0%)	0/7 (0.0%)	2/13 (15.4%)
G/s *BRCA* status: unknown	1/2 (50.0%)	1/9 (11.1%)	0/3 (0.0%)	2/14 (14.3%)
HRD status: positive†	15/18 (83.3%)	2/13 (15.4%)	1/2 (50.0%)	19/34* (55.9%)
HRD status: negative	0/1 (0.0%)	1/1 (100.0%)	0/7 (0.0%)	1/9 (11.1%)
HRD status: unknown	2/3 (66.7%)	1/9 (11.1%)	0/3 (0.0%)	3/15 (20.0%)
Total	17/22 (77.3%)	4/23 (17.4%)	1/12 (8.3%)	

*One patient has unknown platinum-sensitivity status.
†Three patients (best overall response: stable disease [n=2]; non-evaluable [n=1]) have HRD+ and g/s *BRCA*^{wt} or *BRCA*^{unk} status.
Abbreviations: *BRCA*=breast cancer susceptibility gene, g/s=germline/somatic, HRD=homologous recombination deficiency

Safety

- In the safety population (n=101), TEAEs in ≥10% of patients were nausea, fatigue, anemia, diarrhea, vomiting, decreased appetite, constipation, abdominal pain, urinary tract infection, headache, alanine aminotransferase (ALT) increase, and upper respiratory tract infection (Table 4)
- TEAEs led to treatment discontinuation in 6.9% of patients
- At the 60-mg BID dose, TEAEs led to dose interruption in 70.8% of patients with dose reduction in 12.5% of patients
- As of 01 Jun 2019, 10.9% of patients (n=8, dose escalation; and n=3, dose expansion) remained on treatment

Table 4: Summary of Treatment-Emergent Adverse Events Across the Study

Event, n (%)	Phase 1A (n=64)	Phase 1B (n=37)	Total (N=101)
Patients reporting ≥1 TEAE	64 (100.0)	37 (100.0)	101 (100.0)
Patients reporting ≥1 treatment-related TEAE	52 (81.3)	31 (83.8)	83 (82.2)
Patients reporting ≥1 serious TEAE	31 (48.4)	13 (35.1)	44 (43.6)
Patients who experienced ≥1 DLT	5 (7.8)	0 (0)	5 (5.0)
TEAE leading to treatment discontinuation	6 (9.4)	1 (2.7)	7* (6.9)
TEAE leading to dose modification	40 (62.5)	26 (70.3)	66 (65.3)
TEAE leading to dose interruption	40 (62.5)	26 (70.3)	66 (65.3)
TEAE leading to dose reduction	5 (7.8)	6 (16.2)	11 (10.9)
TEAE leading to death	4 (6.3)	1 (7.7)	5† (5.0)
TEAE occurring in ≥10% (all grades), n (%)	**Grade 1 or 2**	**Grade ≥3‡**	**Total (N=101)**
Nausea	66 (65.3)	4 (4.0)	70 (69.3)
Fatigue	46 (45.5)	3 (3.0)	49 (48.5)
Anemia	11 (10.9)	25 (24.8)	36 (35.6)
Diarrhea	32 (31.7)	2 (2.0)	34 (33.7)
Vomiting	31 (30.7)	1 (1.0)	32 (31.7)
Decreased appetite	23 (22.8)	0 (0)	23 (22.8)
Constipation	22 (21.8)	0 (0)	22 (21.8)
Abdominal pain	16 (15.8)	1 (1.0)	17 (16.8)
Urinary tract infection	13 (12.9)	1 (1.0)	14 (13.9)
Headache	12 (11.9)	0 (0)	12 (11.9)
ALT increase	7 (6.9)	5 (5.0)	12 (11.9)
Upper respiratory tract infection§	9 (8.9)	0 (0)	11 (10.9)

*Treatment-related AEs led to treatment discontinuation in 6 (5.9%) patients.
†No TEAEs leading to death were treatment-related.
‡None were Grade 4 nor 5.
§n=2 missing.
Abbreviations: ALT=alanine aminotransferase, DLT=dose-limiting toxicity, TEAE=treatment-emergent adverse event

CONCLUSIONS

- Pamiparib continued to be generally well-tolerated in this update of an ongoing, phase 1 dose-escalation/expansion study in patients with advanced solid tumors
 - Anemia was the most frequent Grade ≥3 TEAE
 - As of 01 Jun 2019, 11 patients remained on treatment
- Linear pharmacokinetics with a terminal half-life of approximately 13 hours; pamiparib can be administered without regard to food
- Pamiparib continued to demonstrate promising antitumor activity in patients with ovarian and associated cancer
 - Confirmed complete or partial responses were observed in 23 of 58 (39.7%) evaluable patients
 - The median duration of response for all patients was 14.9 months (range 11.0–17.9)
- Pamiparib treatment showed higher ORR in g/s *BRCA*$^{mut+}$ vs. g/s *BRCA*^{wt}/*BRCA*^{unk} patients (61.3% vs. 14.8%)
 - In the platinum-sensitive population, higher ORR was achieved in g/s *BRCA*$^{mut+}$ patients vs. *BRCA*^{wt}/*BRCA*^{unk} patients (83.3% vs. 50.0%)

REFERENCES

1. Thomas C, Tulin AV. Poly-ADP-ribose polymerase: machinery for nuclear processes. *Mol Aspects Med.* 2013;34(6):1124-1137.
2. Coleman RL, Sill MW, Bell-McGuinn K, et al. A phase II evaluation of the potent, highly selective PARP inhibitor veliparib in the treatment of persistent or recurrent epithelial ovarian, fallopian tube, or primary peritoneal cancer in patients who carry a germline BRCA1 or BRCA2 mutation—An NRG Oncology/Gynecologic Oncology Group study. *Gynecol Oncol.* 2015;137(3):386-391.
3. Pommier Y, O'Connor MJ, de Bono J. Laying a trap to kill cancer cells: PARP inhibitors and their mechanisms of action. *Sci Transl Med.* 2016;8(362):362ps317.
4. Kubota E, Williamson CT, Ye R, et al. Low ATM protein expression and depletion of p53 correlates with olaparib sensitivity in gastric cancer cell lines. *Cell Cycle.* 2014;13(13):2129-2137.
5. Ledermann J, Harter P, Gourley C, et al. Olaparib maintenance therapy in platinum-sensitive relapsed ovarian cancer. *N Engl J Med.* 2012;366(15):1382-1392.
6. Owonikoko TK, Dahlberg SE, Khan SA, et al. A phase 1 safety study of veliparib combined with cisplatin and etoposide in extensive stage small cell lung cancer: A trial of the ECOG-ACRIN Cancer Research Group (E2511). *Lung Cancer.* 2015;89(1):66-70.
7. Robinson D, Van Allen EM, Wu YM, et al. Integrative clinical genomics of advanced prostate cancer. *Cell.* 2015;161(5):1215-1228.
8. Tutt A, Robson M, Garber JE, et al. Oral poly(ADP-ribose) polymerase inhibitor olaparib in patients with *BRCA1* or *BRCA2* mutations and advanced breast cancer: A proof-of concept trial. *Lancet.* 2010;376(9737):235-244.
9. Murai J, Huang S-Y, Das B, et al. Trapping of PARP1 and PARP2 by clinical PARP inhibitors. *Cancer Res.* 2012;72(21):5588-5599.
10. Tang Z, Jiang B, Shi Z, et al. BGB-290, a novel PARP inhibitor with unique brain penetration ability, demonstrated strong synergism with temozolomide in subcutaneous and intracranial xenograft models. *Cancer Res.* 2015;75(suppl 15):Abstract 1651.
11. Lickliter J, Mileshkin L, Voskoboynikm M, et al. Dose escalation/expansion study to investigate the safety, pharmacokinetics, food effect, and antitumor activity of BGB-290 in patients with advanced solid tumors. *Ann Oncol.* 2017;28(suppl 5):v122-v141.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff, the study patients, and their families. BeiGene, Ltd. provided financial support for this presentation, including writing and editorial assistance by Ira Mills, PhD, and Erin Spohr of Ashfield Healthcare Communications.

AUTHOR DISCLOSURE

M. Voskoboynik reports honoraria from AstraZeneca and MSD Oncology. Other author disclosures detailed in the Quick Response (QR) Code.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com