PRELIMINARY RESULTS WITH TISLELIZUMAB IN CHINESE PATIENTS WITH NON-SMALL CELL LUNG CANCER

Yi-Long Wu[1], Jun Zhao[2], Qingyuan Zhang[3], Hongming Pan[4], Jie Wang[5], Yuxian Bai[3], Jian Li[6], Zuobai Wang[6], Chenlu Wei[6], Xin Li[6], Qing Zhou[1]

[1]Guangdong Lung Cancer Institute, Guangdong General Hospital and Guangdong Academy of Medical Sciences, Guangzhou, China; [2]Peking University Cancer Hospital & Institute, Beijing, China; [3]Harbin Medical University Cancer Hospital, Harbin, China; [4]Sir Run Shaw Hospital, Zhejiang University, Hangzhou, China; [5]Cancer Institute and Hospital, Chinese Academy of Medical Sciences, Beijing, China; [6]BeiGene (Beijing) Co., Ltd., Beijing, China

Poster Number: P2.04-29

World Congress on Lung Cancer
September 23–26, 2018, Toronto, Canada

INTRODUCTION

- Non-small cell lung cancer (NSCLC) accounts for 80–85% of all lung cancers[1]
- The prognosis for patients with NSCLC is relatively poor, particularly when it is diagnosed in later stages where 5-year survival rates are approximately 36% for Stage IIIA, 26% for Stage IIIB, and 1% for Stage IV[2]
- Recent studies of immune checkpoint inhibitors have shown efficacy in patients with PD-L1-positive (PD-L1+) advanced NSCLC[3–5] as well as patients with PD-L1-negative (PD-L1–) NSCLC[5]
- Tislelizumab is a humanized IgG4 monoclonal antibody with high affinity and specificity for PD-1 that was specifically engineered to minimize FcγR binding on macrophages, thereby abrogating antibody-dependent phagocytosis, a potential mechanism of T-cell clearance
- In a first-in-human, phase 1A/1B study (NCT02407990), single-agent tislelizumab was generally well tolerated and demonstrated evidence of antitumor activity in patients with solid tumors, including NSCLC[6]
 - A recommended dose of 200 mg administered intravenously (IV) every 3 weeks (Q3W) was identified for tislelizumab
- The safety and efficacy of tislelizumab as a single agent or in combination with chemotherapy are currently being evaluated in phase 3 studies in patients with NSCLC (NCT03358875 and NCT03594747)
- A dose-verification and indication-expansion study of tislelizumab was conducted in Chinese patients with advanced malignant solid tumors, with the purpose of determining the safety, tolerability, pharmacokinetic characteristics, and preliminary efficacy of tislelizumab in Chinese patients with advanced solid tumors (CTR20160872)
 - Data presented here are the efficacy and safety results from patients with PD-L1+ (Arm 2) and PD-L1– (Arm 3) NSCLC in the indication-expansion phase of the study

METHODS

- The overall design of the study is detailed in Figure 1
- In the dose-verification phase 1 study, patients were administered tislelizumab at 200 mg Q3W IV until they demonstrated no evidence of continued clinical benefit, unacceptable toxicity in the opinion of the investigator, or until they chose to withdraw informed consent
- Once the recommended phase 2 dose (RP2D) of tislelizumab was confirmed to be 200 mg Q3W IV, patients were enrolled into 11 groups of tumor types in the indication-expansion phase 2 study and were administered tislelizumab at this dose level
- Disease assessment by radiographic imaging (enhanced CT or MRI) was performed approximately every 9 weeks during first 12 months and approximately every 12 weeks thereafter; response was assessed by the investigator according to RECIST v1.1 criteria
- Vital signs, weight, physical examinations, Eastern Cooperative Oncology Group (ECOG) performance status, electrocardiograms and laboratory safety tests (eg, hematology, serum chemistry, urinalysis, coagulation, anti-tislelizumab antibodies, thyroid function, viral antigen reactions) were assessed locally throughout the study
- Adverse events (AEs) were graded and recorded throughout the study according to NCI-CTCAE, version 4.03

Figure 1: Study Design

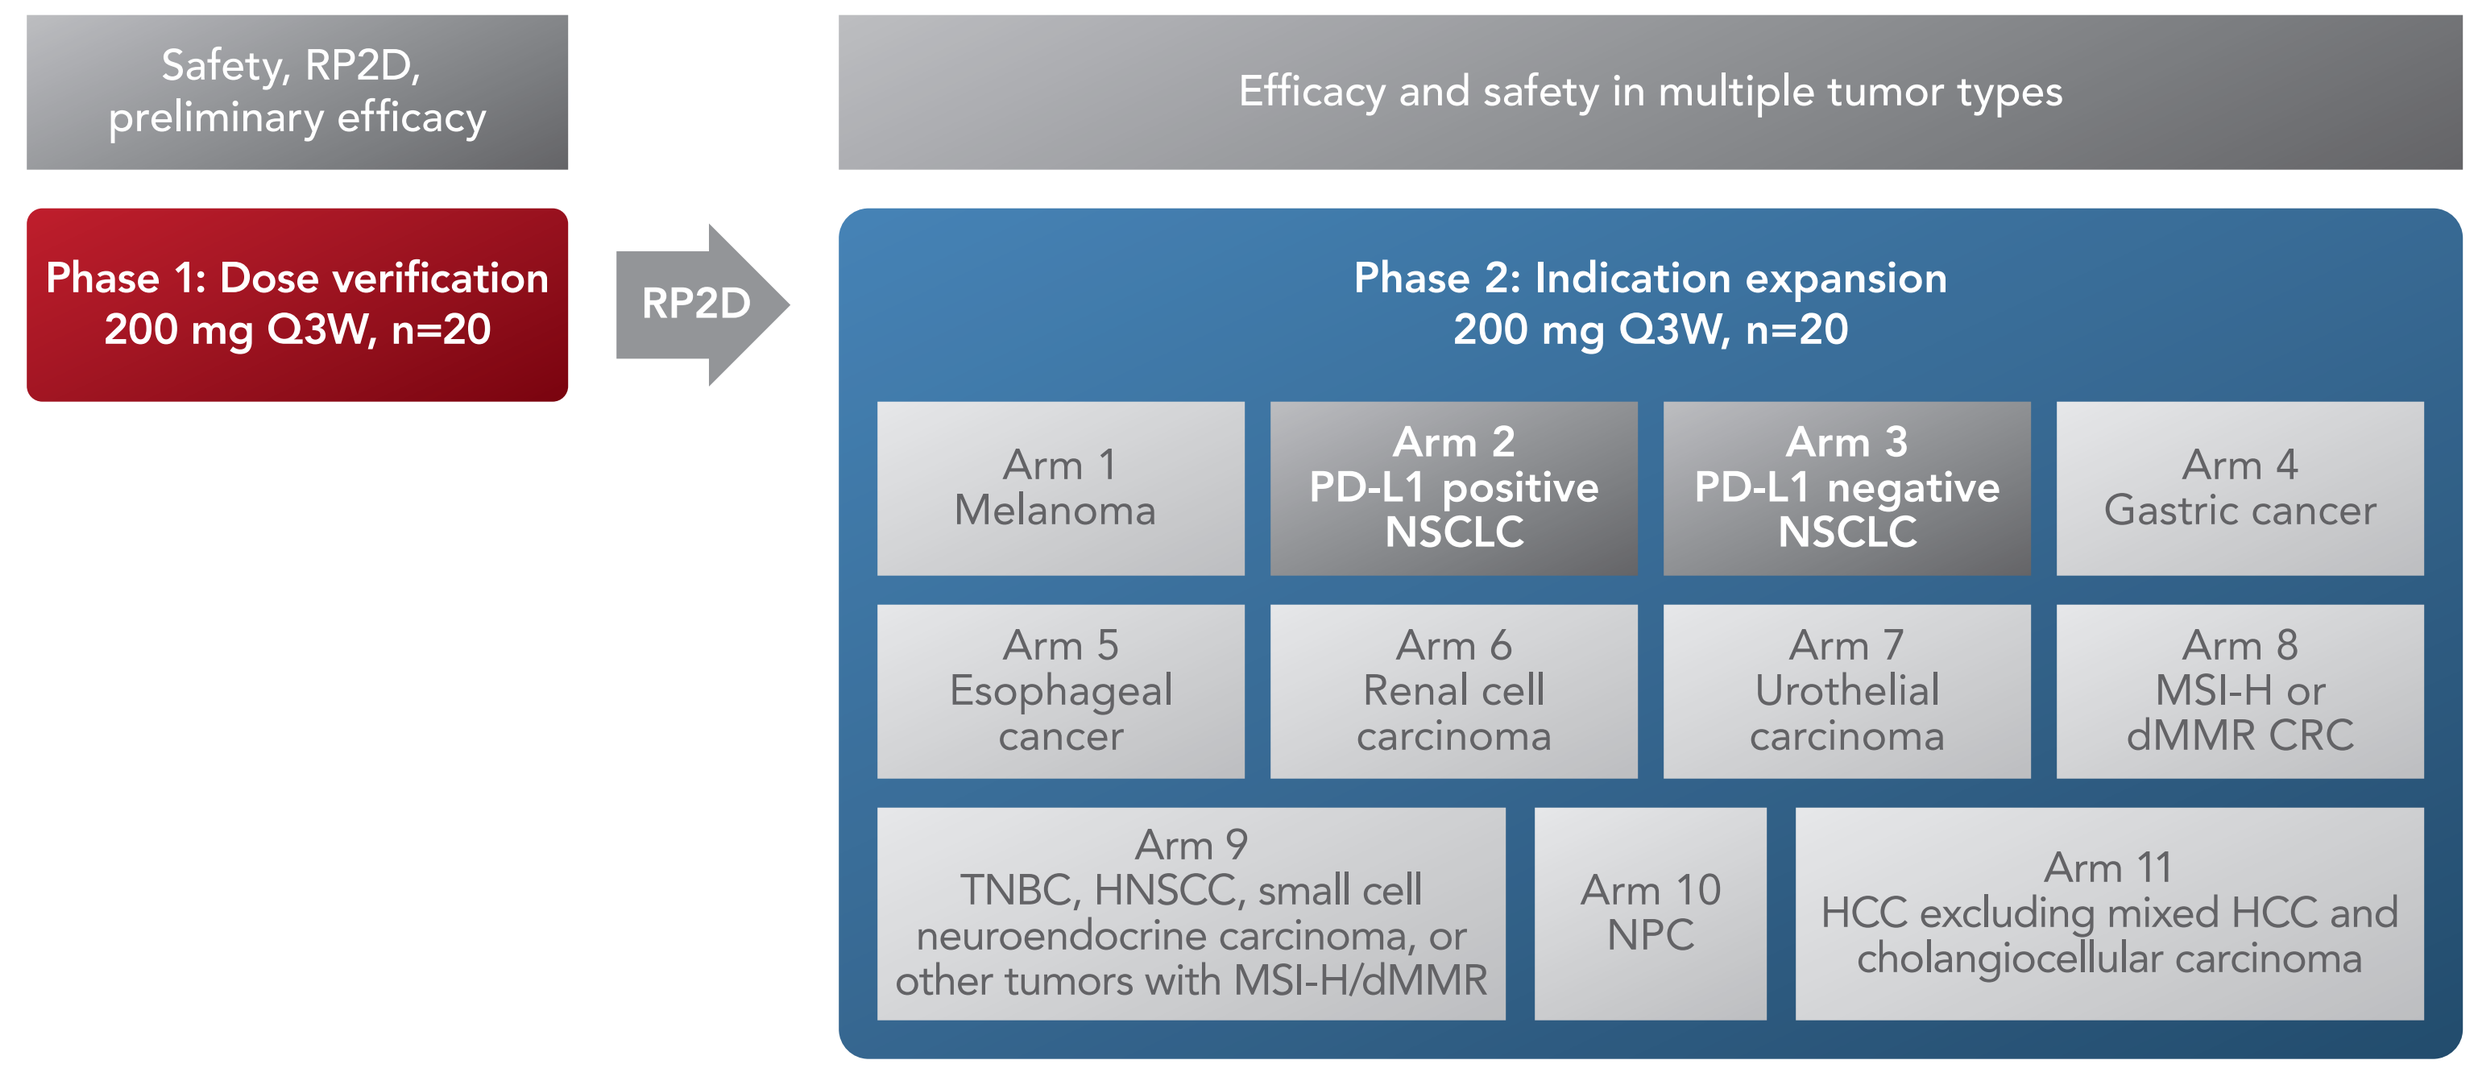
Safety, RP2D, preliminary efficacy
Efficacy and safety in multiple tumor types
Phase 1: Dose verification 200 mg Q3W, n=20
RP2D
Phase 2: Indication expansion 200 mg Q3W, n=20
Arm 1 Melanoma
Arm 2 PD-L1 positive NSCLC
Arm 3 PD-L1 negative NSCLC
Arm 4 Gastric cancer
Arm 5 Esophageal cancer
Arm 6 Renal cell carcinoma
Arm 7 Urothelial carcinoma
Arm 8 MSI-H or dMMR CRC
Arm 9 TNBC, HNSCC, small cell neuroendocrine carcinoma, or other tumors with MSI-H/dMMR
Arm 10 NPC
Arm 11 HCC excluding mixed HCC and cholangiocellular carcinoma

The two red boxes in the phase 2 indication expansion grey box above highlight the data presented in this poster.

Abbreviations: CRC, colorectal carcinoma; dMMR, mismatch repair deficient; HCC, hepatocellular carcinoma; HNSCC, head and neck squamous cell carcinoma; MSI-H, microsatellite instability-high; NPC, nasopharyngeal carcinoma; NSCLC, non-small cell lung cancer; PD-L1, programmed cell death ligand-1; Q3W, every three weeks; RP2D, recommended phase 2 dose; TNBC, triple-negative breast cancer.

Non-Small Cell Lung Cancer Study Population

- Adult patients (aged ≥18 years) with histologically or cytologically confirmed advanced/metastatic (unresectable) NSCLC, who had progressed on or were unable to tolerate standard antitumor treatment, and had no access to standard treatment or had refused standard therapy, were enrolled to receive tislelizumab 200 mg Q3W administered by IV infusion
 - PD-L1 expression status was tested prospectively at the central laboratory and determined by PD-L1 membranous staining using Ventana PD-L1 protocol (SP263 antibody)
 - PD-L1+ (Arm 2) was defined as expression of PD-L1 in ≥10% tumor cells; otherwise tumors were considered PD-L1– (Arm 3)
- Patients were excluded if they had known NSCLC with *EGFR* mutation or ALK rearrangement, a history of severe hypersensitivity reactions to other monoclonal antibodies, if they had received prior therapies targeting PD-1 or PD-L1, or if they had a prior malignancy active within the previous two years (except for NSCLC and locally curable cancers that have apparently been cured (eg, basal or squamous cell skin cancer, superficial bladder cancer, or carcinoma in situ of the cervix or breast)
- Patients were also excluded if they had a history of interstitial lung disease, non-infectious pneumonitis or uncontrolled systemic diseases, including diabetes, hypertension, pulmonary fibrosis, acute lung diseases, or significant cardiovascular disease
- Patients with severe chronic or active infections requiring systemic antibacterial, antifungal, or antiviral therapy, including tuberculosis infections, were also excluded

Phase 2 Study Assessments

- Objective response rate (ORR) was the primary endpoint for these indication-expansion cohorts
- Disease control rate (DCR), clinical benefit rate (CBR), progression-free survival (PFS), overall survival (OS), and duration of response (DoR) were secondary endpoints (PFS and OS results were not mature enough at data cut-off to be included)
- Evaluation of the safety/tolerability profile of tislelizumab was a secondary objective

RESULTS

Patient Disposition

- As of May 11, 2018, 46 patients with NSCLC (n=21, PD-L1+; n=25, PD-L1–) were enrolled in the study (Table 1); 15 remained on treatment and 31 discontinued tislelizumab (reasons for discontinuation included AE [n=1], disease progression [n=23], death [n=2], withdrawal of consent [n=4], and other [n=1])
- The median duration of tislelizumab treatment was 4.14 months (0.2–11.27)
 - In Arm 2 (PD-L1+, n=21), the median duration of tislelizumab treatment was 3.45 months (0.2–11.2) and median follow-up duration was 6.28 months (0.2–11.83)
 - In Arm 3 (PD-L1–, n=25), the median duration of tislelizumab treatment was 4.37 months (0.69–11.27) and median follow-up duration was 8.61 months (2.0–10.84)
- The majority of patients were male (n=32; 69.6%), former smokers (n=26; 56.5%), and had received ≥1 prior anticancer therapy (n=44; 95.6%)
 - In Arm 2 (PD-L1+, n=21), 81% of patients were male, 71.4% were former smokers, and all had received ≥1 prior anticancer therapy
 - In Arm 3 (PD-L1–, n=25), 60% of patients were male, 44% were former smokers, and 92% had received ≥1 prior anticancer therapy

Table 1: Patient Demographics and Disease Characteristics in Non-small Cell Lung Cancer

		Arm 2: PD-L1+ (n=21)	Arm 3: PD-L1– (n=25)	Overall (N=46)
Median age, years (min, max)		58.0 (26, 72)	54.0 (37, 72)	55.5 (26, 72)
Sex, n (%)	Male	17 (81.0)	15 (60.0)	32 (69.6)
	Female	4 (19.0)	10 (40.0)	14 (30.4)
Smoking status, n (%)	Former	15 (71.4)	11 (44.0)	26 (56.5)
	Current	0	1 (4.0)	1 (2.2)
	Non-smoker	6 (28.6)	13 (52.0)	19 (41.3)
Prior lines of anticancer therapy regimens, n (%)	0	0	2 (8.0)	2 (4.3)
	1	5 (23.8)	9 (36.0)	14 (30.4)
	2	8 (38.1)	7 (28.0)	15 (32.6)
	≥3	8 (38.1)	7 (28.0)	15 (32.6)
Prior anticancer radiotherapy, n (%)		9 (42.9)	7 (28.0)	16 (34.8)

Preliminary Antitumor Activity

- At total of 42 patients were evaluable, defined as any patient who had measurable disease at baseline and at least one postbaseline tumor assessment
- A total of seven patients (n=2, PD-L1+; n=5, PD-L1–) achieved a confirmed partial response (PR); 17 patients achieved stable disease (SD) (Table 2)
- The objective response rates (ORR=CR+PR) for Arm 2 [PD-L1+, n=17] and Arm 3 (PD-L1–, n=25) were 11.8% (95% CI: 1.46–36.44) and 20% (95% CI: 6.83–40.7), respectively
- Across the two arms, clinical benefit and disease control rates were 28.6% and 57.1%, respectively
 - The clinical benefit rate (CR+PR+SD ≥24 weeks) was similar between PD-L1+ (29.4% [95% CI: 10.31–55.96]) and PD-L1– (28% [95% CI: 12.07–49.39]) NSCLC patients
 - The disease control rate (CR+PR+SD) was high (>45%) in both PD-L1+ (47.1% [95% CI: 22.98–72.19]) and PD-L1– (64.0% [95% CI: 42.52–82.03]) NSCLC patients
- The antitumor activity of tislelizumab is presented in Figures 2–4

Figure 2: Maximum Tumor Reduction in Evaluable Patients With PD-L1+ NSCLC (A) or PD-L1– NSCLC (B)

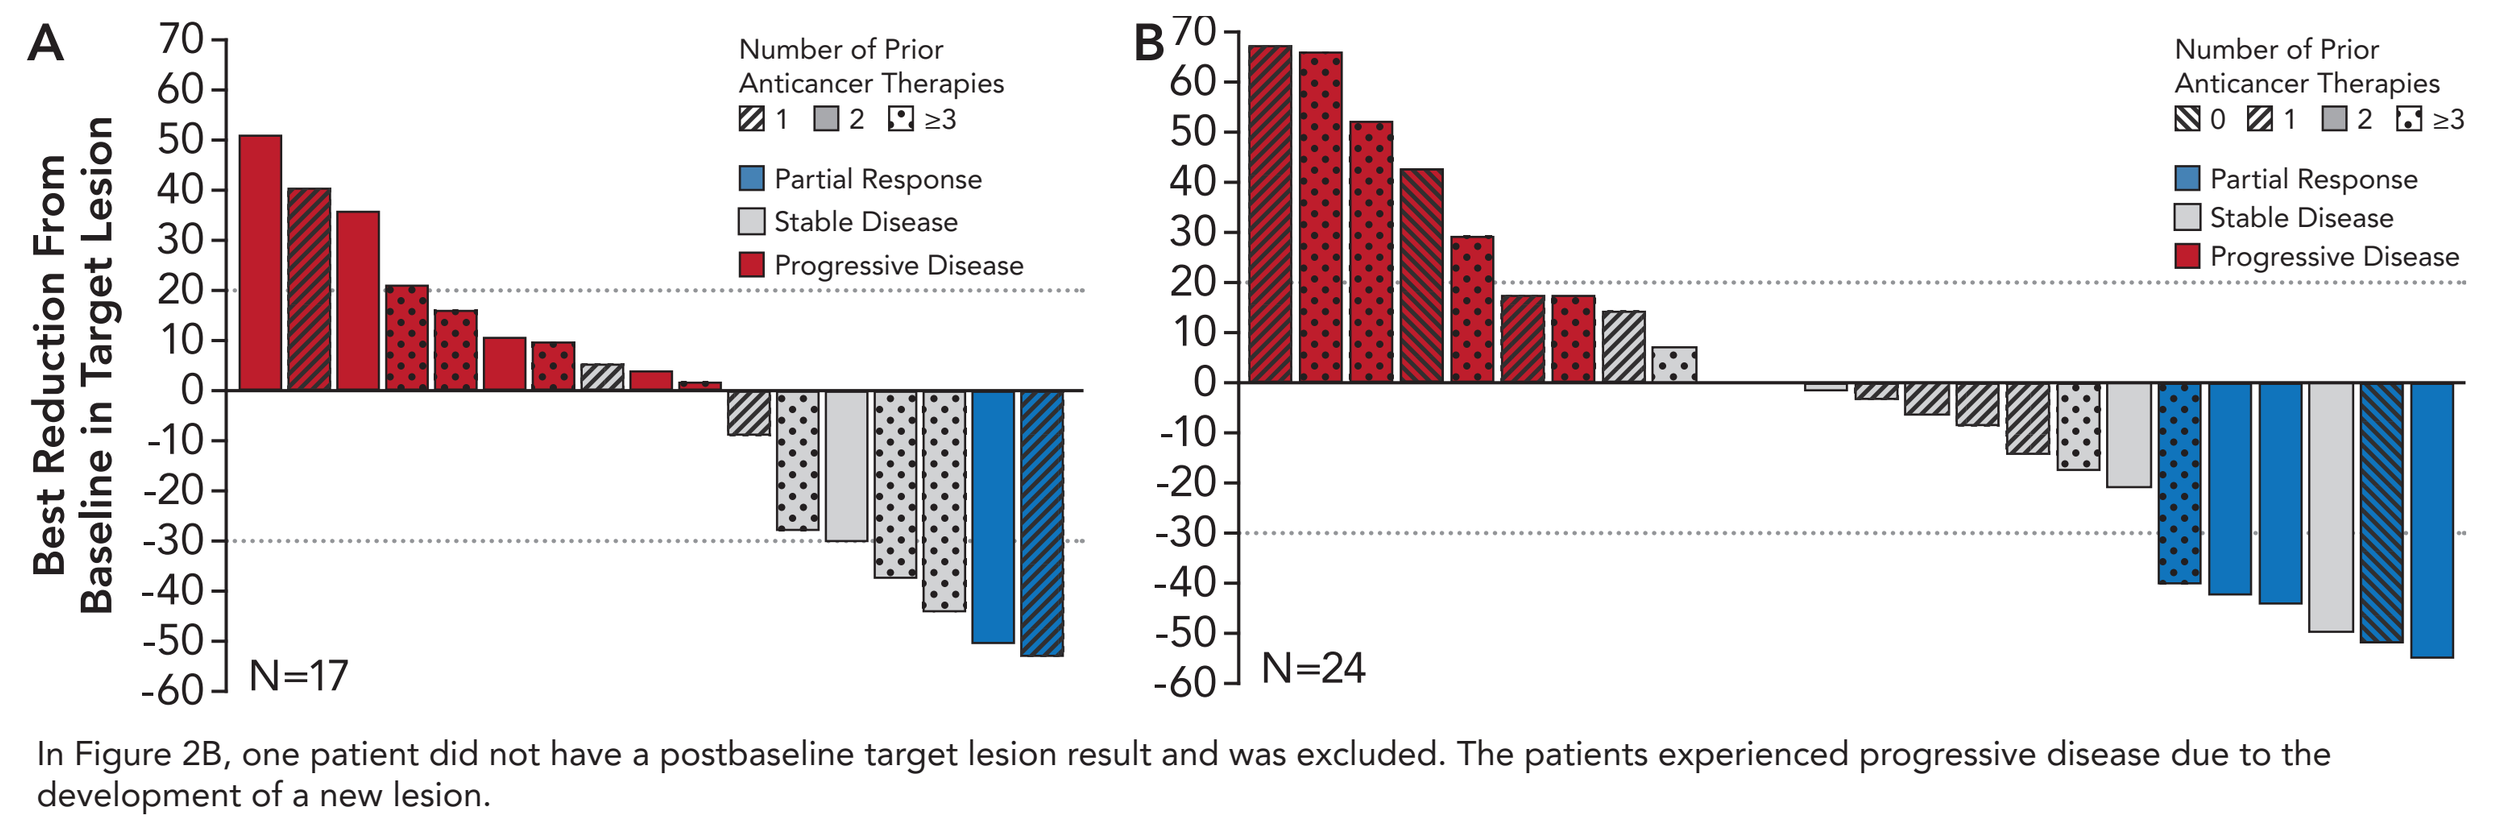
A
B
Best Reduction From Baseline in Target Lesion
70
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
N=17
N=24
Number of Prior Anticancer Therapies
0
1
2
≥3
Partial Response
Stable Disease
Progressive Disease

In Figure 2B, one patient did not have a postbaseline target lesion result and was excluded. The patients experienced progressive disease due to the development of a new lesion.

Figure 3: Duration of Treatment and Response in Evaluable Patients With PD-L1+ NSCLC (A) or PD-L1– NSCLC (B)

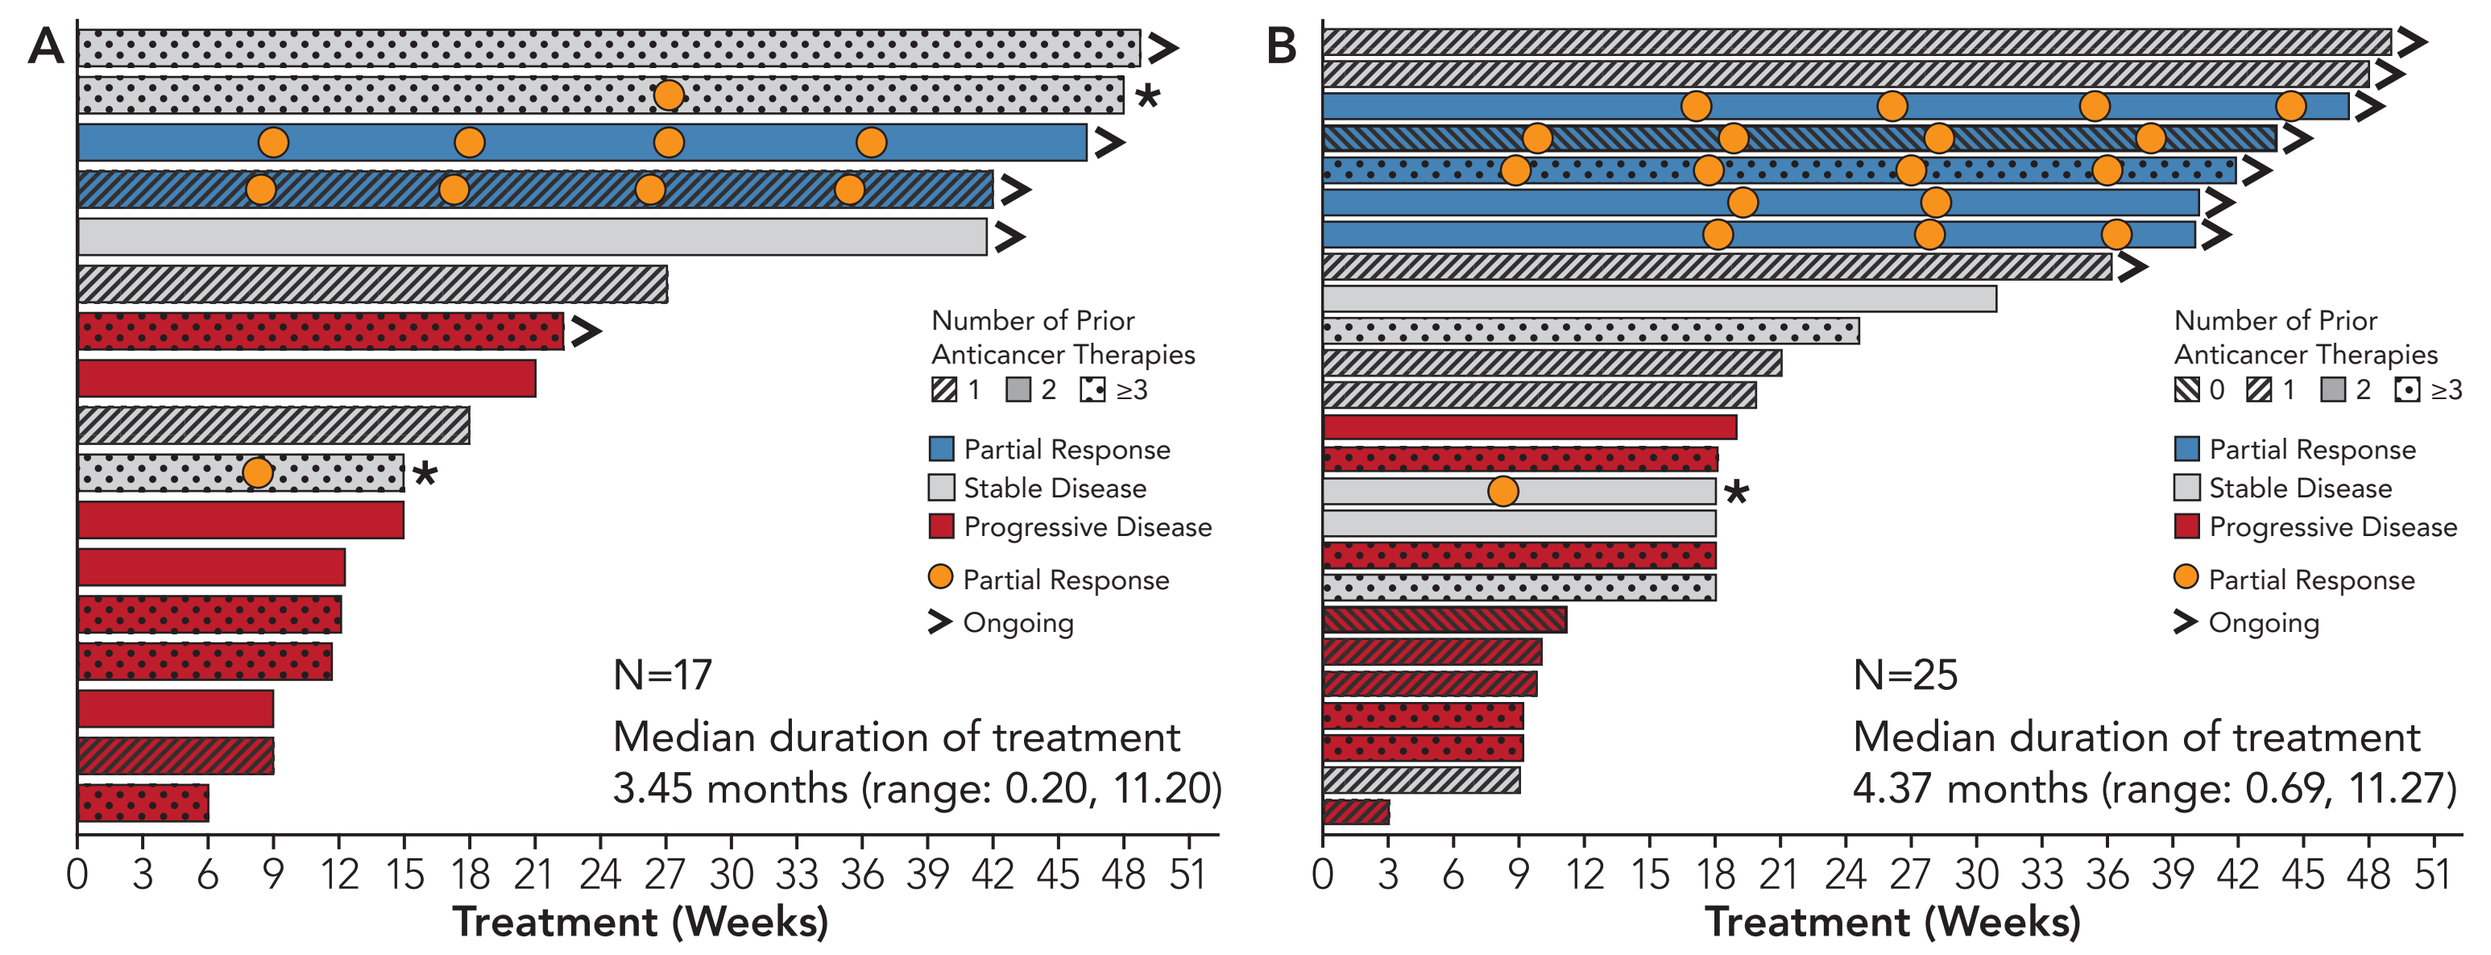
A
B
Number of Prior Anticancer Therapies
0
1
2
≥3
Partial Response
Stable Disease
Progressive Disease
Partial Response
Ongoing
N=17
Median duration of treatment 3.45 months (range: 0.20, 11.20)
N=25
Median duration of treatment 4.37 months (range: 0.69, 11.27)
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51
Treatment (Weeks)

*Unconfirmed response.

Figure 4: Radiographic Images of a Patients with PD-L1+ (A) and PD-L1– (B) NSCLC

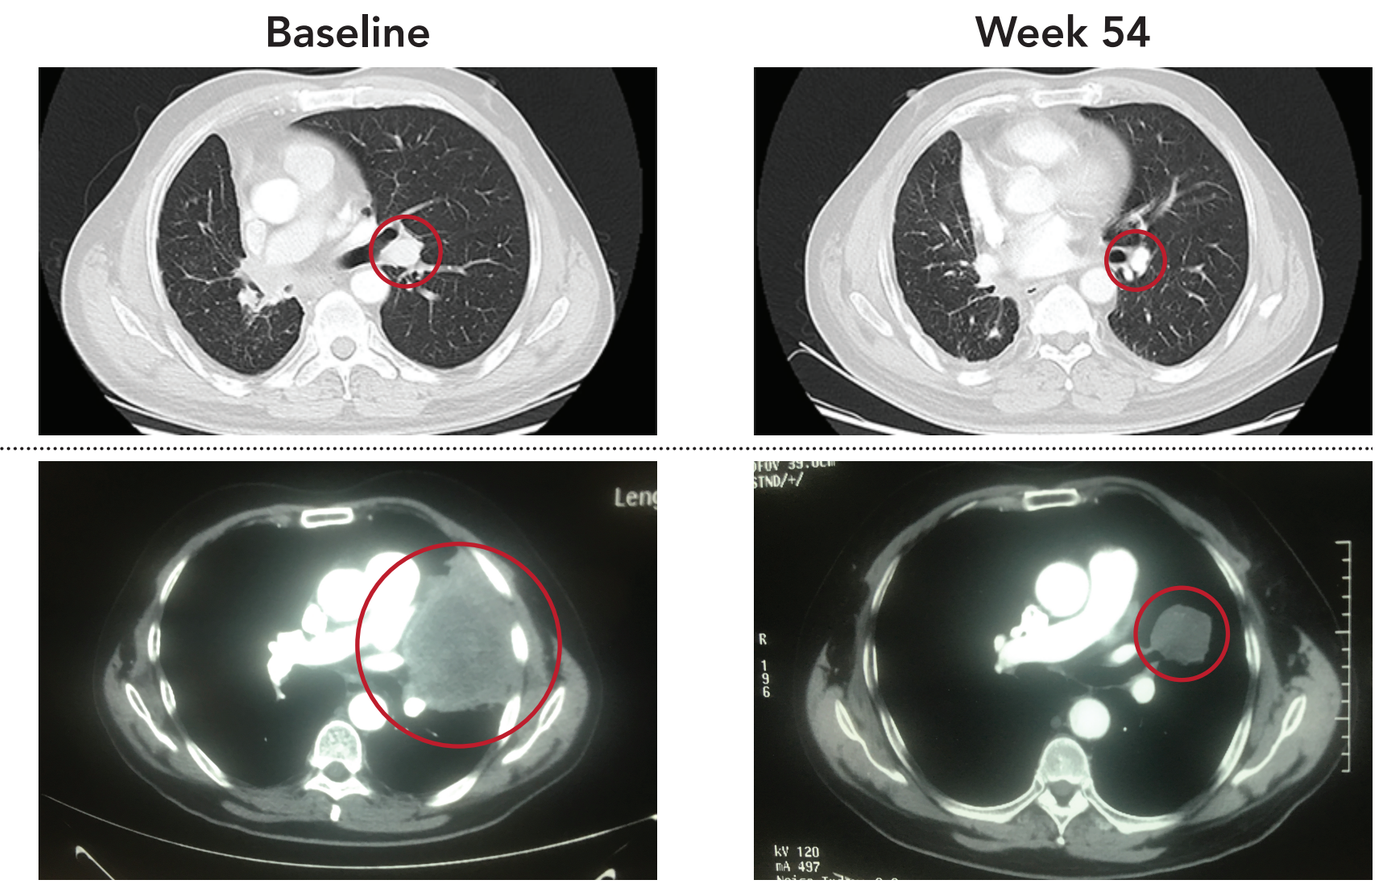
Baseline
Week 54

A Male, 62-year old former smoker with PD-L1+ squamous NSCLC and LN metastases who previously received 1 prior line of anticancer therapy

B Male, 56-year old former smoker with PD-L1– NSCLC of adenocarcinoma histology and LN, liver, and adrenal gland metastases who previously received 2 prior lines of anticancer therapy

Abbreviations: LN, lymph node; NSCLC, non-small cell lung cancer; PD-L1, programmed cell death ligand-1.

Table 2: Best Overall Response Observed with Tislelizumab Evaluable Patients with PD-L1+ NSCLC or PD-L1– NSCLC

		Arm 2: PD-L1+ (n=17)	Arm 3: PD-L1– (n=25)	Overall (N=42)
BOR per RECIST 1.1 (confirmed), n (%)	CR	0	0	0
	PR	2 (11.8)	5 (20.0)	7 (16.7)
	SD	6 (35.3)	11 (44.0)	17 (40.5)
	PD	9 (52.9)	9 (36.0)	18 (42.9)
ORR (CR+PR), % (95% CI)		11.8 (1.46, 36.44)	20.0 (6.83, 40.70)	16.7 (6.97, 31.36)
CBR (CR+PR+ durable SD*), % (95% CI)		29.4 (10.31, 55.96)	28.0 (12.07, 49.39)	28.6 (15.72, 44.58)
DCR (CR+PR+SD), % (95% CI)		47.1 (22.98, 72.19)	64.0 (42.52, 82.03)	57.1 (40.96, 72.28)

*Durable SD: stable disease for ≥24 weeks.

Abbreviations: BOR, best overall response; CBR, clinical benefit rate; CI, confidence interval; CR, complete response; DCR, disease control rate; NE, not evaluable; ORR, objective response rate; PD, progressive disease; PD-L1, programmed cell death ligand-1; PR, partial response; RECIST, response evaluation criteria in solid tumors; SD, stable disease.

Safety and Tolerability

- Adverse events considered at least possibly related to tislelizumab were reported in 30 out of a total 46 patients with NSCLC (Table 3)
- Across the two NSCLC populations, the most commonly reported treatment-related AEs (TRAEs) considered were increases in AST (n=11)/ALT (n=10), rash (n=5), and hypothyroidism (n=5)
 - The majority of the TRAEs were grade ≤2 in severity; five patients experienced grade ≥3 TRAEs (increased AST, n=3; increased ALT, n=2; increased gamma-glutamyltransferase (GGT), n=1; maculo-papular rash, n=1; hyperglycemia, n=1)
- A total of 26 patients experienced immune-related AEs (irAEs)
 - The most common irAEs were increased AST (n=11), increased ALT (n=10), hypothyroidism (n=5), and rash (n=5)
- One patient discontinued due to a Grade 3 AE (hypotension) considered not related to treatment
- Fourteen patients had serious AEs; three patients experienced serious TRAEs (nausea and vomiting, n=1; increased AST, n=1; hyperglycemia, n=1)
- Across these two study arms, three patients had a TEAE with a fatal outcome (multiple organ dysfunction syndrome, n=1; central nervous system metastases, n=1; hypotension, n=1); none were determined to be related to treatment

Table 3: Treatment-Related Adverse Events Occurring in ≥2 Patients in Either Treatment Arm

	Arm 2: PD-L1+		Arm 3: PD-L1–		Overall	
	All grades (n=21)	Grade ≥3 (n=21)	All grades (n=25)	Grade ≥3 (n=25)	All grades (N=46)	Grade ≥3 (N=46)
Patients who had ≥1 TRAE	15 (71.4)	0	15 (60.0)	5 (20.0)	30 (65.2)	5 (10.9)
Transaminases increased	5 (23.8)	0	7 (28.0)	3 (12.0)	12 (26.1)	3 (6.5)
Increased AST	5 (23.8)	0	6 (24.0)	3 (12.0)	11 (23.9)	3 (6.5)
Increased ALT	4 (19.0)	0	6 (24.0)	2 (8.0)	10 (21.7)	2 (4.3)
Rash	2 (9.5)	0	3 (12.0)	0	5 (10.9)	0
Hypothyroidism	3 (14.3)	0	2 (8.0)	0	5 (10.9)	0
Increased bilirubin	3 (14.3)	0	1 (4.0)	0	4 (8.7)	0
Increased GGT	3 (14.3)	0	1 (4.0)	1 (4.0)	4 (8.7)	1 (2.2)
Nausea	2 (9.5)	0	2 (8.0)	0	4 (8.7)	0
Pyrexia	2 (9.5)	0	2 (8.0)	0	4 (8.7)	0
Decreased blood TSH	0	0	3 (12.0)	0	3 (6.5)	0
Increased blood TSH	0	0	3 (12.0)	0	3 (6.5)	0
Vomiting	1 (4.8)	0	2 (8.0)	0	3 (6.5)	0
Increased free thyroxin	0	0	2 (8.0)	0	2 (4.3)	0
Decreased weight	0	0	2 (8.0)	0	2 (4.3)	0
Anemia	2 (9.5)	0	0	0	2 (4.3)	0

Data presented as n, (%).

Abbreviations: ALT, alanine aminotransferase; AST, aspartate aminotransferase; GGT, gamma-glutamyltransferase; PD-L1, programmed cell death ligand-1; TRAE, treatment-related adverse event; TSH, thyroid stimulating hormone.

CONCLUSIONS

- Preliminary data suggest treatment with tislelizumab was generally well tolerated and has antitumor activity in patients with advanced NSCLC
 - With a median follow-up of 8.4 months, 15 (32.6%) patients remain on study
 - The rate of treatment discontinuation due to an AE was low (n=1/46)
- Partial responses were observed in seven patients (n=2/17 [PD-L1+]; n=5/25 [PD-L1–])
- Disease control rate was ≥45% in both NSCLC expansion cohorts
- There was no clear relationship between PD-L1 status and clinical efficacy measures based on the small sample size
- Adverse events reported in the two cohorts of PD-L1+ and PD-L1– were consistent with the overall safety profile of tislelizumab observed in previous studies with other tumor types and were generally of low severity
- The preliminary safety profile and antitumor activity support continued development of tislelizumab in patients with advanced NSCLC; phase 3 studies of tislelizumab as treatment for NSCLC have been initiated (NCT03358875 and NCT03594747)

REFERENCES

1. PDQ Adult Treatment Editorial Board. Small cell lung cancer treatment (PDQ®): Health professional version. 2017 Jan 20. In: PDQ cancer information summaries [Internet]. Bethesda (MD): National Cancer Institute (US); 2002. Available from: https://www.ncbi.nlm.nih.gov/books/NBK65909/. Accessed April 2018.
2. Non-small cell lung cancer survival rates, by stage. American Cancer Society. 2017. Available from: https://www.cancer.org/content/cancer/en/cancer/nonsmall-cell-lung-cancer/detection-diagnosis-staging/survival-rates.html. Accessed May 2018.
3. Gettinger SN, Horn L, Gandhi L, et al. Overall survival and long-term safety of nivolumab (anti-programmed death 1 antibody, BMS-936558, ONO-4538) in patients with previously treated advanced non-small-cell lung cancer. *J Clin Oncol.* 2015;33(18):2004–2012.
4. Herbst RS, Bass P, Kim DW, et al. Pembrolizumab versus docetaxel for previously treated, PD-L1-positive, advanced non-small-cell lung cancer (KEYNOTE-010): a randomised controlled trial. *Lancet.* 2016;387(10027):1540–1550.
5. Rizvi NA, Mazières J, Planchard D, et al. Activity and safety of nivolumab, an anti-PD-1 immune checkpoint inhibitor, for patients with advanced, refractory squamous non-small-cell lung cancer (CheckMate 063): a phase 2, single-arm trial. *Lancet Oncol.* 2015;16(3):257–265.
6. Desai J, Markman B, Sandhu SK, et al. Updated safety, efficacy, and pharmacokinetics (PK) results from the phase I study of BGB-A317, an anti-programmed death-1 (PD-1) mAb in patients with advanced solid tumors. *J Immunother Cancer.* 2016;4(suppl 1):154.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff and study patients, and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. BeiGene, Ltd. provided financial support for this presentation, including writing and editorial assistance by SuccinctChoice Medical Communications, Chicago, IL.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com