POPULATION PHARMACOKINETICS OF TISLELIZUMAB IN PATIENTS WITH ADVANCED TUMORS

C.-Y. Wu[1], Z. Tang[1], L. Liu[2], Y. Gao[2], Y. Ben[3], S. Sahasranaman[1]

[1]Clinical Pharmacology, BeiGene USA, Inc., San Mateo, United States of America; [2]Drug Development Consulting Service, Shanghai Qiangshi Information Technology Co., Ltd., Shanghai, China; [3]Clinical Development, BeiGene USA, Inc., San Mateo, United States of America

483P
European Society for Medical Oncology Congress
September 27-October 1, 2019, Barcelona, Spain

BACKGROUND

- Tislelizumab, an investigational humanized IgG4 monoclonal antibody with high affinity and specificity for PD-1, was engineered to minimize binding to FcγR on macrophages in order to abrogate antibody-dependent phagocytosis, a mechanism of T-cell clearance and potential resistance to anti-PD-1 therapy. It has shown robust antitumor activity and was generally well tolerated in patients with advanced solid tumors and with classical Hodgkin lymphoma (cHL). The objectives of this analysis were to develop a population pharmacokinetics (PopPK) model and quantify the impact of demographic and disease characteristics on tislelizumab PK

OBJECTIVES

- Perform PopPK analysis of tislelizumab using the data collected from three clinical studies (BGB-A317-001, BGB-A317-102, and BGB-A317-203) and estimate typical values and interpatient variability of PK parameters in cancer patients
- Determine the effects of demographic, pathophysiologic, and disease-related covariates on the PK of tislelizumab to better understand clinical factors that might affect exposure in individual patients

METHODS

Analysis Dataset

- The final PopPK model was developed from a dataset of 798 subjects (with 5,935 samples) enrolled in three clinical studies to quantitatively describe the clinical PK of tislelizumab and identify sources of interindividual variability (Table 1)

Table 1: Summary of Studies Included in PopPK Analyses

Study Region	Design and Indication	Treatment	Patients (N)	PK Observations (N)
BGB-A317-001 Global Phase 1a/1b	**Indication:** Advanced solid tumors **Design:** Open-label, two-part, dose escalation and expansion	0.5 mg/kg Q2W	3	69
		2 mg/kg Q2W	26	359
		5 mg/kg Q2W	26	370
		10 mg/kg Q2W	7	99
		2 mg/kg Q3W	21	444
		5 mg/kg Q3W	341	2145
		200 mg Q3W	12	153
BGB-A317-102 China Phase 1/2	**Indication:** Advanced solid tumors **Design:** Open-label, two-part, dose escalation and expansion	200 mg Q3W	292	1961
BGB-A317-203 China Phase 2	**Indication:** Relapsed or refractory classical Hodgkin lymphoma **Design:** Open-label, single-arm, multi-center, and multinational	200 mg Q3W	70	335

Abbreviations: PK, pharmacokinetic; PopPK, population pharmacokinetics; Q2W, every 2 weeks; Q3W, every 3 weeks.

Modeling Approach

- A nonlinear mixed effects modeling approach with the first-order conditional estimation with interaction (FOCEI) method in NONMEM, version 7.3.0 (ICON, Maryland) was used for the PopPK analysis

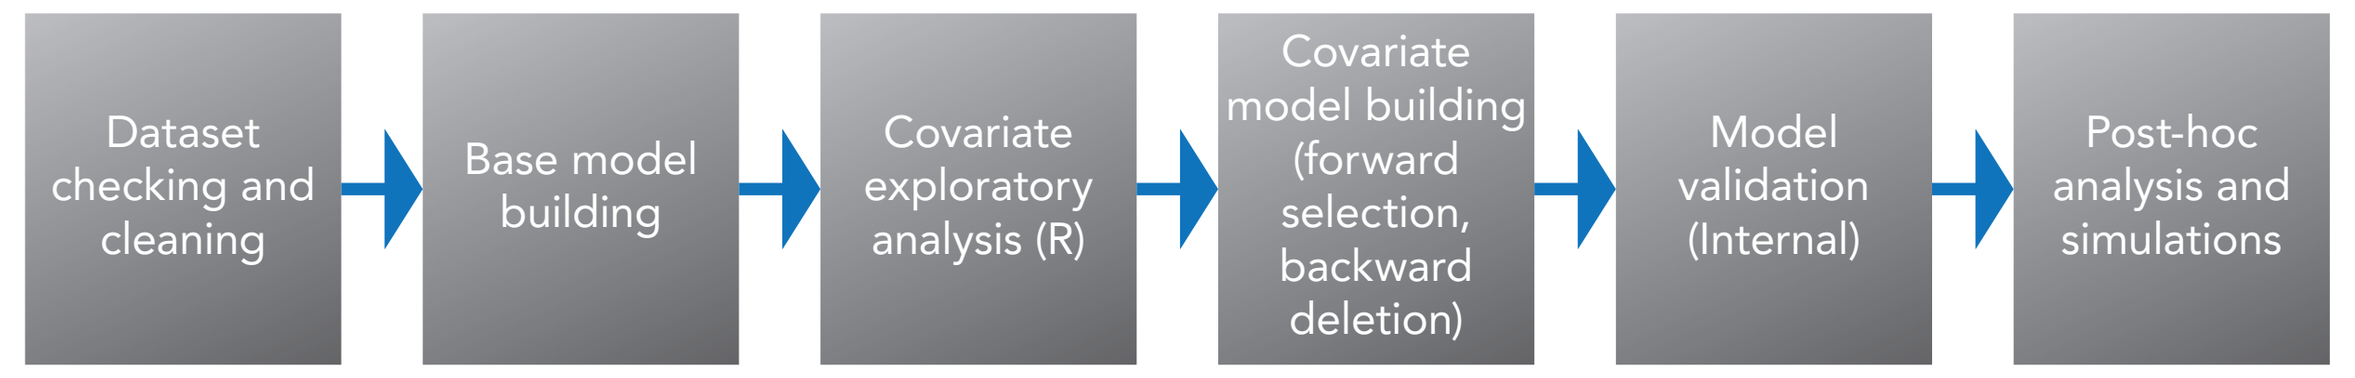
Dataset checking and cleaning
Base model building
Covariate exploratory analysis (R)
Covariate model building (forward selection, backward deletion)
Model validation (Internal)
Post-hoc analysis and simulations

Covariate Screening

- The final model was developed by incorporating the effect of relevant covariates on key structural model parameters of the base model. Covariates were selected based on clinical judgment, mechanistic plausibility, and prior knowledge, using a forward addition and backward elimination method (based on the significance levels of *P*<0.01 and *P*<0.001, respectively).These covariates were as follows:

Demographics

- Baseline age (AGE)
- Baseline body weight (WT)
- Sex (SEX)
- Race (RACE; Asian vs Other)

Hepatic and Renal Function Related Covariates

- Baseline estimated glomerular filtration rate (eGFR)
- Baseline bilirubin (BIL)
- Baseline alanine aminotranferase (ALT)
- Baseline aspartate aminotransferase (AST)

Disease Related Covariates

- Baseline albumin (ALB)
- Tumor type (TUMTP)
- Tumor size at baseline (TUMSZ for solid tumor and SUMPPD for cHL)
- Lactate dehydrogenase (LDH)
- Baseline Eastern Cooperative Oncology Group (ECOG) status (0 vs ≥1)

Molecule Related Covariate

- Positive treatment-emergent anti-drug antibody status (ADA)

RESULTS

- The PK of tislelizumab in the dose range tested was best described by a three-compartment model with first order elimination from the central compartment, and redistribution into the peripheral compartments, as illustrated in Figure 1. The PopPK model was parameterized in terms of clearance from the central compartment (CL), volume of the central compartment (V_c), clearance of distribution from the central to the peripheral compartment (Q_2 and Q_3), and volume of the peripheral compartment (V_2 and V_3). No time-varying CL was identified following tislelizumab treatment
- Tables 2 and 3 list all the covariates included in stepwise covariate analysis, Table 4 lists the population PK parameters
- The general goodness-of-fit plots of the final PopPK model are shown in Figures 2 and 3, where a good agreement between the predicted concentrations and the observed concentrations was observed and no apparent bias was observed in the residual plots over time and across predicted concentrations
- Baseline age, race, ALT, AST, BIL, LDH, eGFR, ECOG status, and ADA did not show statistically significant impact on the PK of tislelizumab
- Baseline albumin, TUMSZ, and TUMTP were identified as significant covariates on CL. Baseline body weight, sex, and TUMTP were identified as significant covariates on V_c
- The following statistically significant parameter-covariate relationships were identified:

$$CL_i\ (L/day) = 0.164\times e^{\left(0.0794\times\ln\left(\frac{TUMSZ}{68}\right)-0.579\times\log\left(\frac{ALB}{40}\right)-0.224_{cHL}+0.135_{GC}+\eta_{CL,i}\right)}$$

$$V_{C_i}\ (L) = 2.92\times e^{\left(0.423\times\ln\left(\frac{WT}{66}\right)-0.111_{Female}-0.0269_{cHL}+0.181_{GC}+\eta_{Vc,i}\right)}$$

TUMSZ: Baseline tumor size for solid tumors
ALB: Baseline albumin
cHL: Classical Hodgkin lymphoma , yes (=x1) or no (=x0)
GC: Gastric cancer, yes (=x1) or no (=x0)
Female: Yes (=x1) or no (=x0)
WT: Baseline body weight

Sensitivity Analysis

- The sensitivity analysis (Figure 4) showed that tumor type and baseline albumin were the most influential covariates on tislelizumab PK
- Although these covariates tested statistically significant, the model predicted steady-state exposures are still in the comparable range, which are not expected to have a clinically meaningful impact on tislelizumab considering the overall variability of the population

Figure 1: PopPK Model Diagram for Tislelizumab

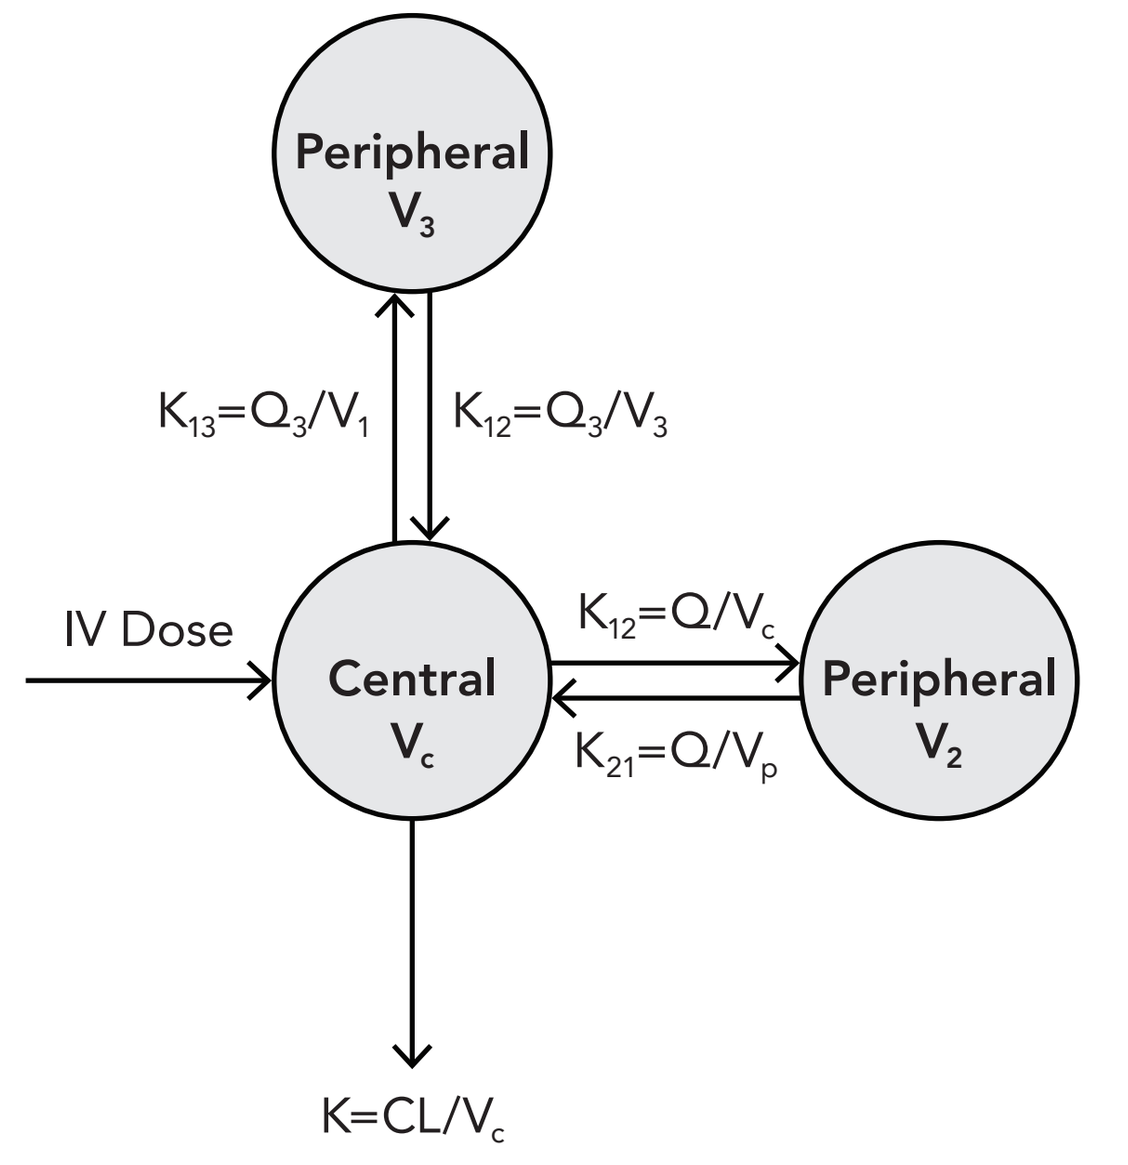
Peripheral V3
K13=Q3/V1
K12=Q3/V3
IV Dose
Central Vc
K12=Q/Vc
K21=Q/Vp
Peripheral V2
K=CL/Vc

$$\frac{dX_1}{dt} = K_{21}\times X_2 + K_{31} - (K + K_{12} + K_{13})\times X_1$$

$$\frac{dX_2}{dt} = K_{12}\times X_1 - K_{21}\times X_2$$

$$\frac{dX_{23}}{dt} = K_{13}\times X_1 - K_{31}\times X_3$$

Abbreviations: CL, clearance; K, first-order elimination rate constant; K_{12}, rate constant from central to peripheral 2; K_{13}, rate constant from central to peripheral 3; K_{21}, rate constant from peripheral 2 to central; K_{31}, rate constant from peripheral 3 to central; Q_2, inter-compartmental clearance 2; Q_3, inter-compartmental clearance 3; V_1, peripheral volume 1; V_2, peripheral volume 2; V_3, peripheral volume 3; V_c, central volume; X_1, quantity of drug in central compartment; X_2, quantity of drug in peripheral compartment 2; X_3, quantity of drug in peripheral compartment 3.

Figure 2: Predicted Versus Observed Concentration Diagnostic Plots for the Final PopPK Model

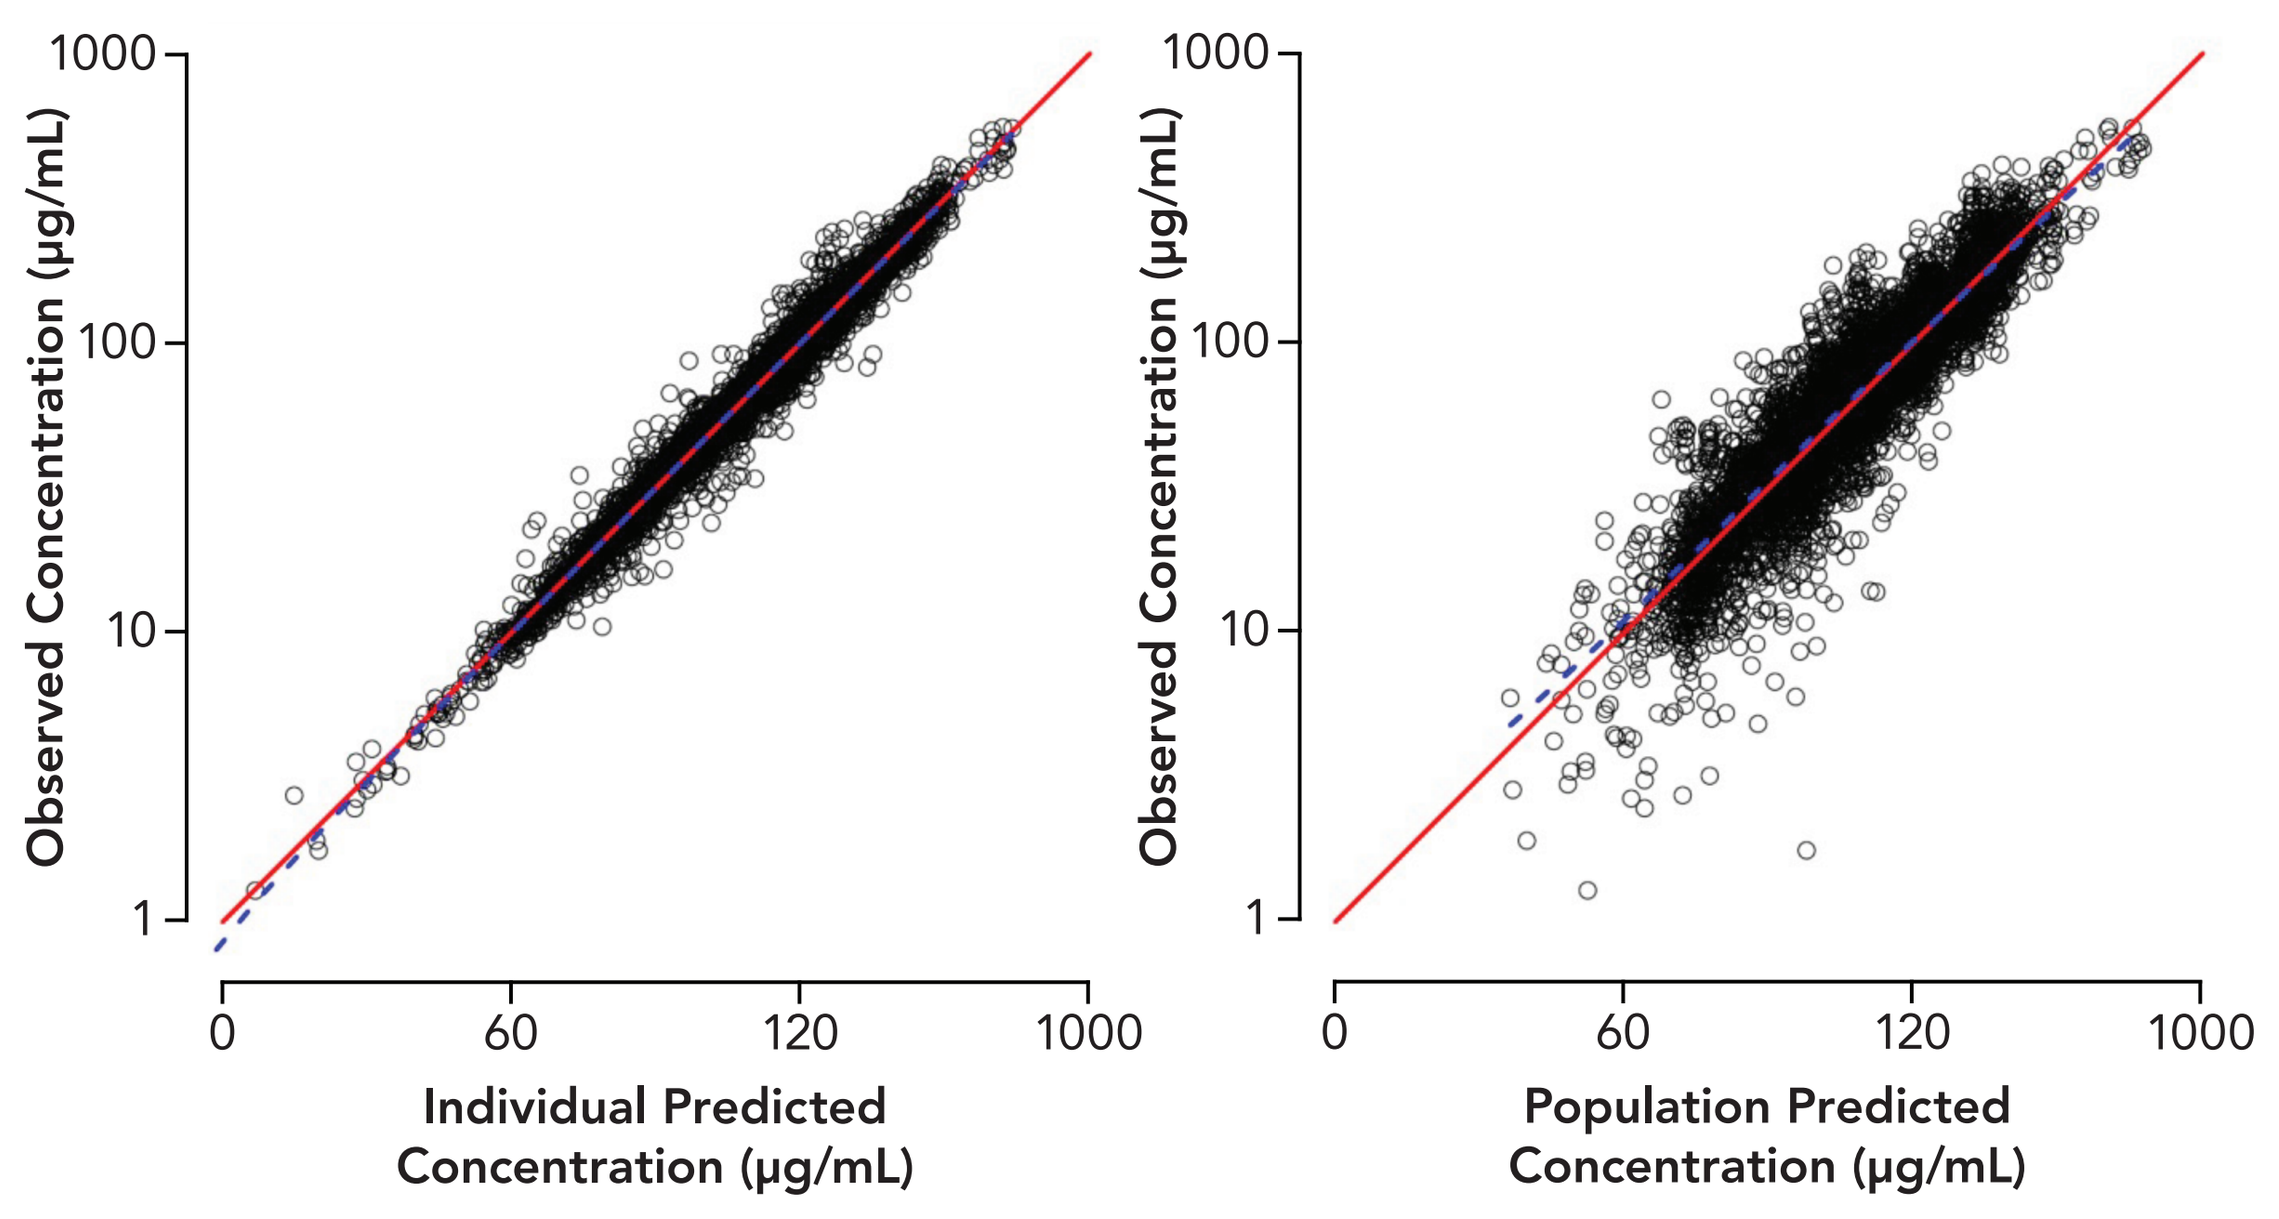
Observed Concentration (µg/mL)
1000
100
10
1
0
60
120
1000
Individual Predicted Concentration (µg/mL)
Population Predicted Concentration (µg/mL)

Observed versus individual predicted concentrations (left) and observed versus population predicted concentrations (right) for the final PopPK model. Points are individual data and red lines represent the unit diagonal. The blue dashed lines are smooth curves (lowess) showing the relationship between two variables.
Abbreviations: PopPK, population pharmacokinetics.

Figure 3: Residual Diagnostic Plots for the Final PopPK Model

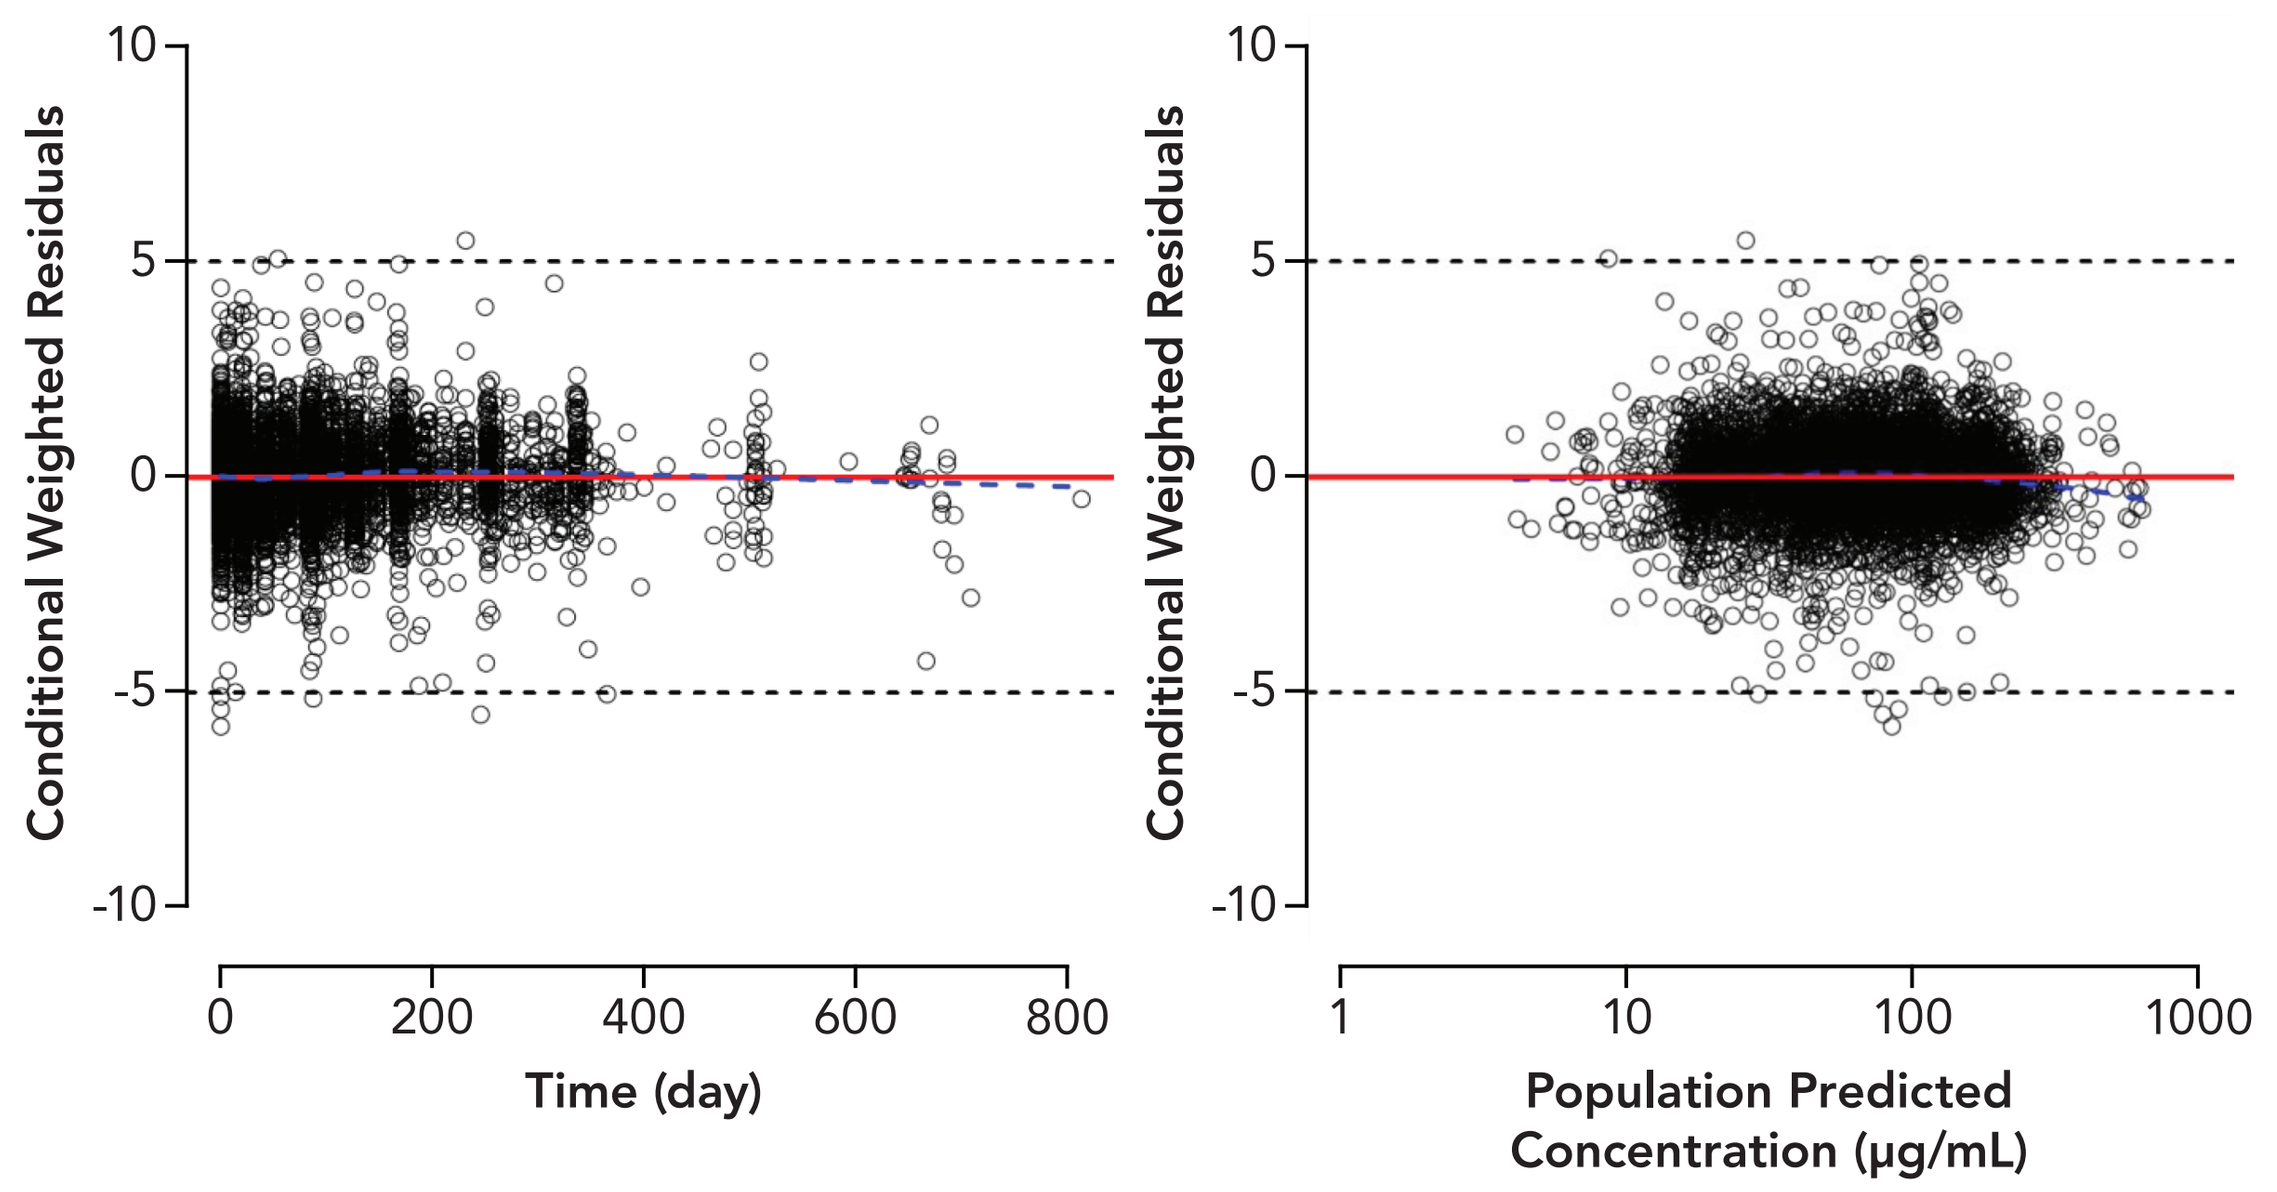
Conditional Weighted Residuals
10
5
0
-5
-10
0
200
400
600
800
Time (day)
1
10
100
1000
Population Predicted Concentration (µg/mL)

CWRES versus time (left) and PRED (right). Points are individual data. Red solid lines represent the unit line at zero. Black dotted lines represent |CWRES| of 5. The blue dashed lines are smooth curves (lowess) showing the relationship between two variables.
Abbreviations: CWRES, conditional weighted residuals; PopPK, population pharmacokinetics; PRED, population predicted value.

Figure 4: Sensitivity Plot Comparing the Effect of Covariates on Tislelizumab Steady-State Exposure

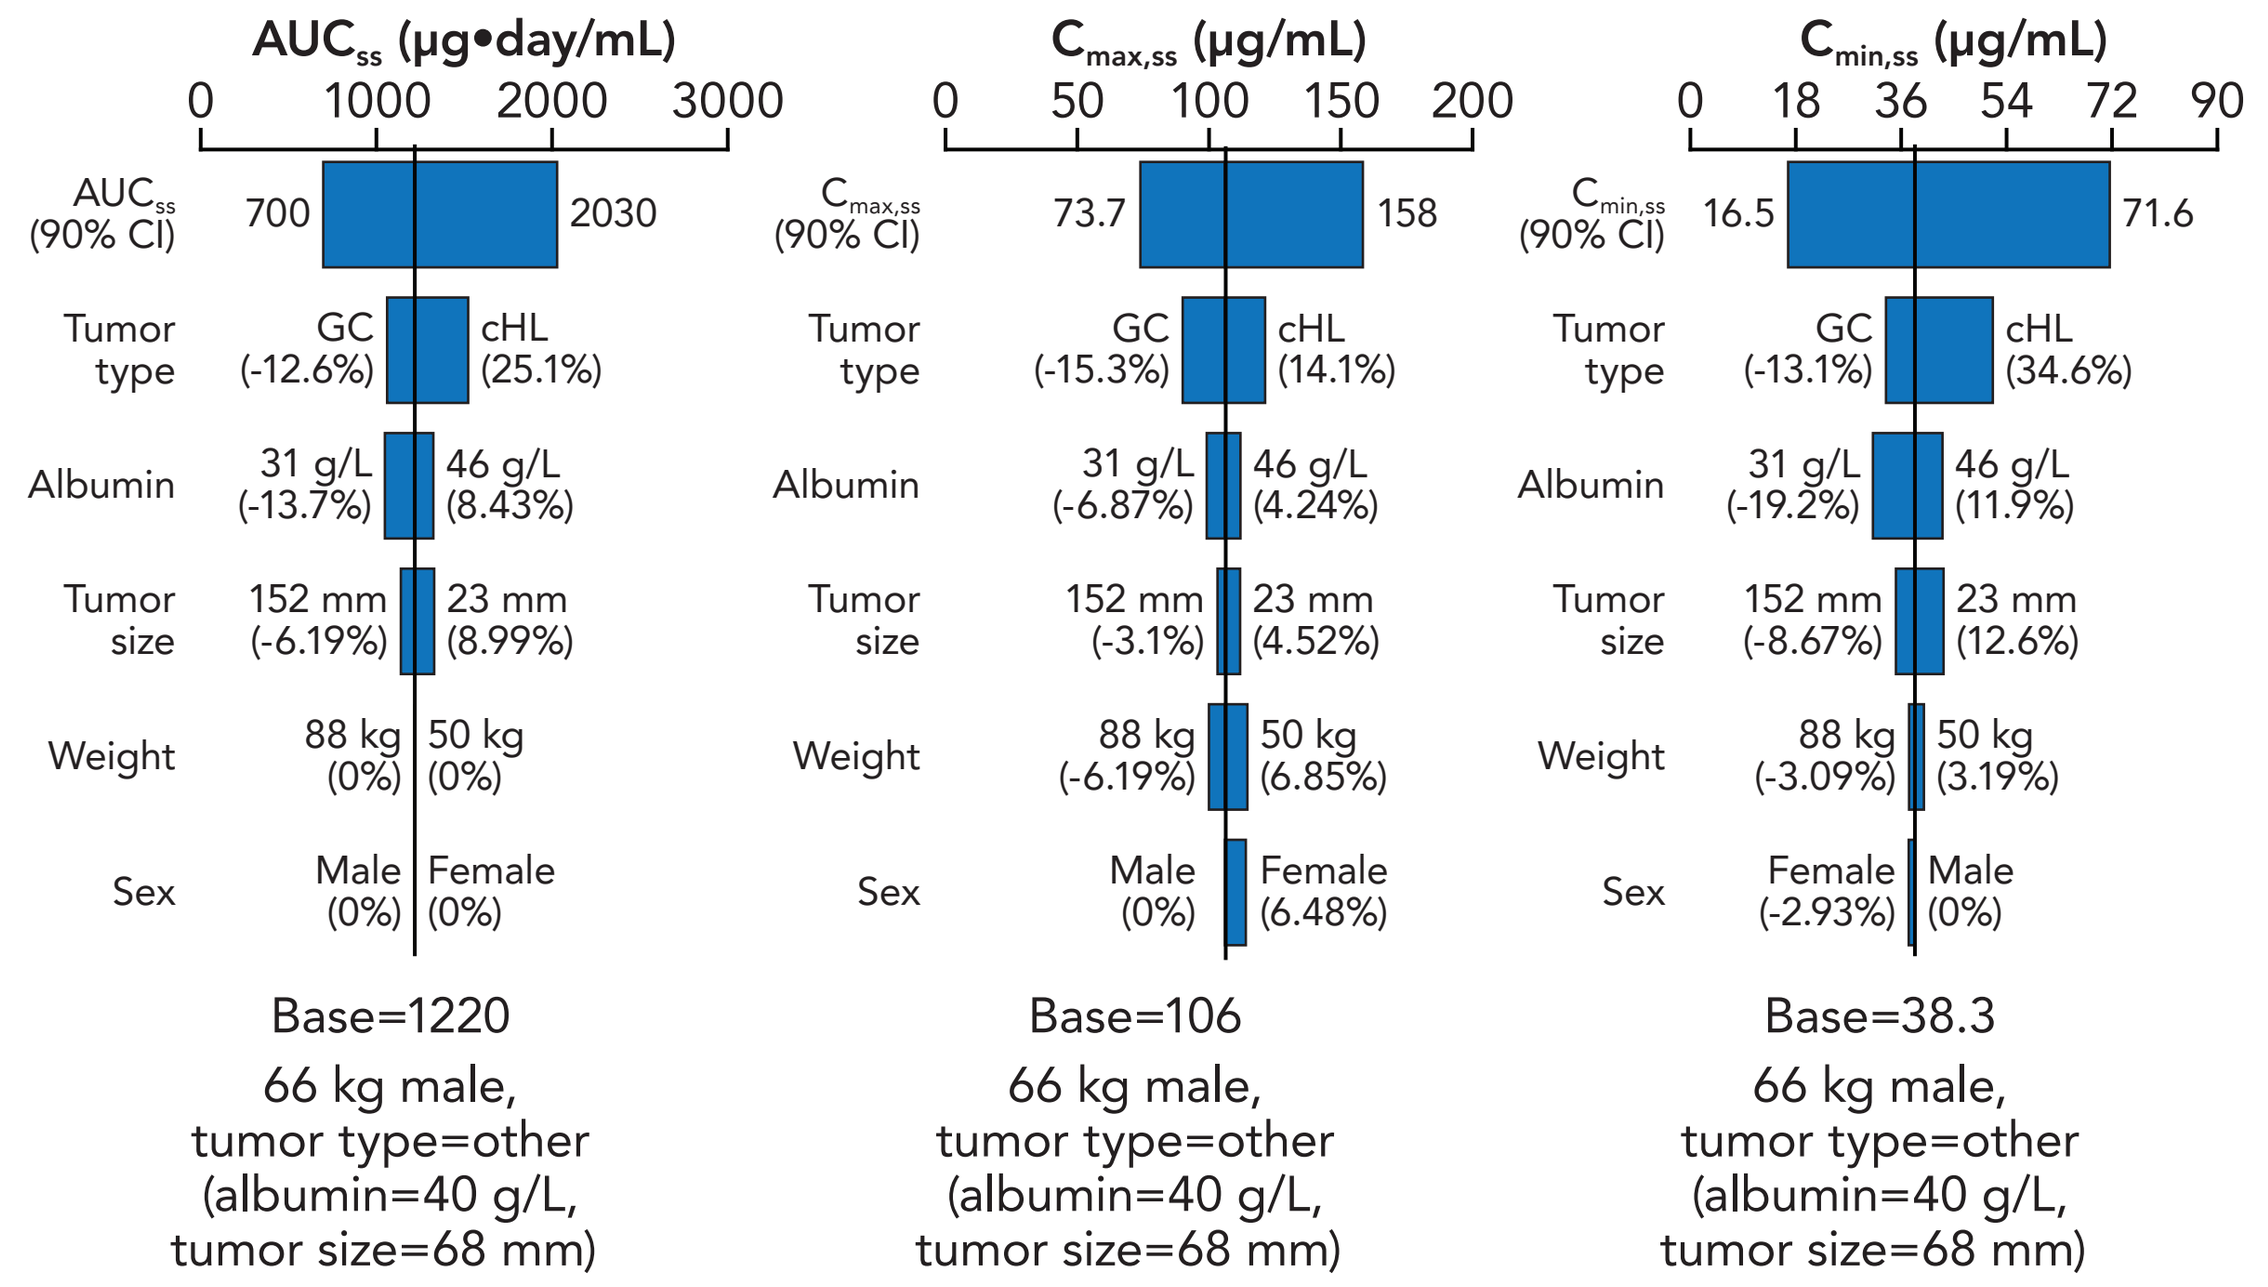
AUCss (µg•day/mL)
0 1000 2000 3000
AUCss (90% CI) 700 2030
Tumor type GC (-12.6%) cHL (25.1%)
Albumin 31 g/L (-13.7%) 46 g/L (8.43%)
Tumor size 152 mm (-6.19%) 23 mm (8.99%)
Weight 88 kg (0%) 50 kg (0%)
Sex Male (0%) Female (0%)
Base=1220
66 kg male, tumor type=other (albumin=40 g/L, tumor size=68 mm)
Cmax,ss (µg/mL)
0 50 100 150 200
Cmax,ss (90% CI) 73.7 158
Tumor type GC (-15.3%) cHL (14.1%)
Albumin 31 g/L (-6.87%) 46 g/L (4.24%)
Tumor size 152 mm (-3.1%) 23 mm (4.52%)
Weight 88 kg (-6.19%) 50 kg (6.85%)
Sex Male (0%) Female (6.48%)
Base=106
66 kg male, tumor type=other (albumin=40 g/L, tumor size=68 mm)
Cmin,ss (µg/mL)
0 18 36 54 72 90
Cmin,ss (90% CI) 16.5 71.6
Tumor type GC (-13.1%) cHL (34.6%)
Albumin 31 g/L (-19.2%) 46 g/L (11.9%)
Tumor size 152 mm (-8.67%) 23 mm (12.6%)
Weight 88 kg (-3.09%) 50 kg (3.19%)
Sex Female (-2.93%) Male (0%)
Base=38.3
66 kg male, tumor type=other (albumin=40 g/L, tumor size=68 mm)

Base, as represented by the black vertical line and values, refers to the predicted exposure (AUC_{ss}, $C_{max,ss}$, $C_{min,ss}$) of tislelizumab in a typical male patient after repeated 10 doses of 200 mg Q3W. The black shaded bar with value at each end shows the 5th to 95th percentile exposure range across the entire population. Each blue shaded bar represents the influence of covariates on the exposure. The label at left end of the bar represents the covariate being evaluated. The upper and lower values for each covariate capture 80% of the plausible range in the population. The length of each bar describes the potential impact of the covariates on tremelimumab exposure, with the percentage value in the parentheses at each end representing the percent change of exposure from the base. The most influential covariates are at the top of the plot for each exposure parameter.
Abbreviations: AUC_{ss}, steady-state area-under-the-curve; $C_{max,ss}$, steady-state peak concentration; $C_{min,ss}$, steady-state trough concentration.

Table 2: Summary of Continuous Covariates Included in Stepwise Covariate Analysis

Covariate (units)	Patients, (N)	Mean (SD)	Median	Range	Missing
Age (years)	802	55.9 (13.5)	58	18-82	—
Weight (kg)	802	67.5 (14.9)	65.9	31.9-129	—
Bilirubin (µmol/L)	802	10.2 (6.36)	9	2-96	—
AST (IU/L)	802	30.0 (21.8)	24	5-338	—
ALT (IU/L)	802	23.7 (23.4)	18	2.5-340	—
Albumin (g/L)	802	38.9 (5.83)	39.5	17-53.2	—
Creatine (µmol/L)	802	73.4 (21.4)	70	21.5-186	—
eGFR (mL/min/m²)	793	92.4 (21.3)	93.4	30.7-163	—
LDH (µkat/L)	800	5.08 (5.28)	3.79	1.45-100	2 (0.249%)
Tumor size (mm)	732	79.1 (54.7)	68	10-355	—
SUMPPD (mm²)	70	1540 (1400)	1160	136-6380	—

Abbreviations: ALT, alanine aminotranferase; AST, aspartate aminotransferase; eGFR, estimated glomerular filtration rate; LDH, lactate dehydrogenase; SUMPPD, Sum of Products of Perpendicular Diameters.

Table 3: Summary of Categorical Covariates Covariates Included in Stepwise Covariate Analysis

Covariate		Patients, N (%)
ADA	Missing	94 (11.72)
	0	600 (74.81)
	1	108 (13.47)
Race	White	281 (35.04)
	Asian	491 (61.22)
	Other	30 (3.74)
Sex	Male	485 (60.47)
	Female	317 (39.53)
ECOG	0	294 (36.66)
	1	508 (63.34)
Tumor type	Gastric cancer	75 (9.35)
	Ovarian cancer	49 (6.11)
	Colorectal cancer	50 (6.23)
	Hepatocellular carcinoma	66 (8.23)
	Esophageal carcinoma	74 (9.23)
	Urothelial bladder cancer	39 (4.86)
	Non-small cell lung cancer	107 (13.34)
	Classical Hodgkin Lymphoma	70 (8.73)
	Other	272 (33.92)

Abbreviations: ADA, anti-drug antibody; ECOG, Eastern Cooperative Oncology Group.

CONCLUSIONS

- Tislelizumab PK was confirmed linear in the dose range tested and can be adequately described by a three-compartment disposition model with linear clearance. No time-varying CL was observed in this analysis
 - For a typical male subject with tumors except cHL and GC, body weight of 66 kg, ALB of 40 g/L, and tumor size of 68 mm, the estimated CL was 0.164 L/day, V_c was 2.92 L, Q_2 was 0.713 L/day, V_2 was 0.928 L, Q_3 was 0.146 L/day, V_3 was 1.39 L, and elimination half-life was 25.9 days
- The covariates tested did not have a clinically meaningful impact on tislelizumab exposure. Sensitivity analysis results support the use of the current clinical dose of 200 mg Q3W and no dose adjustment is necessary based on patients' age, body weight, race, sex, tumor type, and tumor size

Table 4: Tislelizumab Population PK Parameters

Parameter Description	Population Estimate (% SE)	Interindividual Variability (% SE)
Clearance, CL (L/day)	0.164 (1.60%)	32.2 (6.39%)
Influence of tumor size on CL	0.0794 (23.9%)	—
Influence of albumin on CL	-0.579 (15.4%)	—
Influence of cHL on CL	-0.224 (20.1%)	—
Influence of GC on CL	0.135 (35.7%)	—
Central volume, V_c (L)	2.92 (0.98%)	16.7 (7.1%)
Influence of weight on V_c	0.423 (7.88%)	—
Influence of sex on V_c	-0.111 (12.9%)	—
Influence of cHL on V_c	-0.0269 (97.1%)	—
Influence of GC on V_c	0.181 (13.8%)	—
Inter-compartmental clearance, Q_2 (L/day)	0.713 (14.9%)	—
Peripheral volume, V_2 (L)	0.928 (8.15%)	56.6 (21.9%)
Inter-compartmental clearance, Q_3 (L/day)	0.146 (9.01%)	61.1 (20.8%)
Peripheral volume, V_3 (L)	1.39 (7.03%)	94.2 (13.7%)
Covariance (CL, V_c)	0.0199 (14.5%)	—
Residual error (%)	14.3 (2.27%)	

Abbreviations: cHL, classical Hodkins Lymphoma; CL, clearance; GC, gastric cancer; PK, pharmacokinetic; Q_2, inter-compartmental clearance 2; Q3, inter-compartmental clearance 3; V_1, peripheral volume 1; V_2, peripheral volume 2; V_3, peripheral volume 3; V_c, central volume.

REFERENCES

1. U.S. Department of Health and Human Services. Food and Drug Administration. Population pharmacokinetics: guidance for industry. https://www.fda.gov/media/128793/download. Published July 2019. Accessed August 22, 2019.
2. European Medicines Agency. Guideline on reporting the results of population pharmacokinetic analyses. https://www.ema.europa.eu/en/documents/scientific-guideline/draft-guideline-reporting-results-population-pharmacokinetic-analyses_en.pdf. Published June 28, 2006. Accessed August 22, 2019.
3. Sheng J, Srivastava S, Sanghavi K, et al. Clinical pharmacology considerations for the development of immune checkpoint inhibitors. *J Clin Pharmacol.* 2017;57(suppl 10):S26-S42.
4. Kinnunen M, Piirainen P, Kokki H, et al. Updated clinical pharmacokinetics and pharmacodynamics of oxycodone. *Clin Pharmacokinet.* 2019;58:835-857.

CONFLICTS OF INTEREST

CW: Employee- BeiGene USA, Inc; **TT:** Employee- BeiGene USA, Inc; **LL:** Employee- Shanghai Qiang shi information Technology; **YG:** Employee- Shanghai Qiang shi information Technology; **YB:** Employee- BeiGene USA, Inc.; **SS:** Employee- BeiGene USA, Inc.

ACKNOWLEDGMENTS

This study was sponsored by BeiGene, Ltd. Editorial assistance was provided by Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL), and funded by the sponsor.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com