T-CELL-, MHC I-, AND TUMOR-RELATED GENE SIGNATURES PREDICT CLINICAL BENEFIT AND RESISTANCE TO TISLELIZUMAB MONOTHERAPY IN PRETREATED PD-L1–HIGH UROTHELIAL CARCINOMA

Dingwei Ye[1], Aiping Zhou[2], Qing Zou[3], Hanzhong Li[4], Cheng Fu[5], Hailong Hu[6], Jian Huang[7], Wei Shen[8], Yun Zhang[8], Xiaopeng Ma[8], Pei Zhang[8], Ruiqi Huang[8], Xiusong Qiu[8], Lilin Zhang[8], Feng Bi[9]

[1]Fudan University Shanghai Cancer Center, Shanghai, China; [2]Cancer Hospital, Chinese Academy of Medical Sciences, Beijing, China; [3]Jiangsu Cancer Hospital, Nanjing, China; [4]Peking Union Medical College Hospital, Beijing, China; [5]Liaoning Cancer Hospital & Institute, Shenyang, China; [6]The Second Hospital of Tianjin Medical University, Tianjin, China; [7]Sun Yat-sen University, Sun Yat-Sen Memorial Hospital, Guangzhou, China; [8]BeiGene (Beijing) Co., Ltd., Beijing, China; [9]West China Hospital, Sichuan University, Chengdu, China

Poster: 78
Society for Immunotherapy of Cancer
November 9-14, 2020, Virtual Congress

BACKGROUND

- Urothelial carcinoma (UC) is the most common histologic type of bladder cancer and has a low 5-year survival rate when diagnosed at an advanced stage[1]
 - In China, UC is the sixth most common cancer in men and the thirteenth most common in women[2]
- While several FDA-approved anti-programmed cell death protein-1/programmed death-ligand 1 (PD-1/PD-L1) therapies have exhibited improved efficacy in late-line UC,[3] the mechanisms of response or resistance are not clear and warrant further exploration
- Tislelizumab, a humanized IgG4 monoclonal antibody with high affinity and binding specificity for PD-1[4] is the first PD-1/PD-L1 blockade approved in PD-L1–high UC[5] by the National Medical Products Administration (NMPA) in China (April 2020)
- Here, retrospective analyses of gene expression profiles correlating with response and resistance to tislelizumab treatment are presented

METHODS

Study Design

- This phase 2 trial (BGB-A317-204; NCT04004221) conducted in China and Korea assessed the safety, tolerability, and efficacy of tislelizumab (200 mg every 3 weeks) in patients with PD-L1–high UC previously treated with ≥1 platinum-containing therapy
- Patients were considered PD-L1–high if ≥25% of tumor/immune cells expressed PD-L1 as assessed by the VENTANA™ PD-L1 (SP263) assay
- One hundred Chinese patients had evaluable gene expression profiling (GEP) data and were further analyzed

Gene Expression Profiling

- GEP analysis was conducted on baseline tumor samples (formalin-fixed, paraffin-embedded) using HTG EdgeSeq Precision Immuno-oncology panel
- Gene signature scores were calculated using the Gene Set Variation Analysis package and publicly available gene signatures
- Non-responder subgroups were hierarchically clustered by Euclidean distance metrics with average linkage by columns

Statistical Analysis

- Objective response rate (ORR) and Clopper-Pearson 95% confidence intervals (CIs) were calculated
- The Kaplan-Meier method was used to estimate median overall survival (OS), progression-free survival (PFS), and 95% CIs
- Differential gene expression (DEG) analysis was performed between responders and non-responders using the Wilcoxon rank-sum test; the Bonferroni method was used to adjust for multiple comparisons
- Kaplan-Meier curves of biomarker subgroups were compared using the log-rank test; hazard ratios were estimated using the Cox proportional hazard model

RESULTS

Patient Characteristics

- Baseline characteristics and clinical outcomes of the 100 GEP-evaluable patients were comparable with the overall population in this trial (Table 1)

T-cell and MHC I Signatures Were Associated With Response and Survival Benefits

- T-cell[6] (*CD3D, CD3E, CD3G, CD6, SH2D1A, TRAT1*) and major histocompatibility complex class I (MHC I) signatures[7] (*HLA.A, TAP1*) were enriched in responders (Figure 1A and 1B)
- Using median signature score as the cutoff:
 - Significant improvement of OS (median OS: 13.5 vs 7.3 months, *P*=0.013, Figure 1E) and a trend of longer PFS (median PFS: 3.8 vs 2.1 months, *P*=0.125, Figure 1C) was observed in the T-cell–high group versus the T-cell–low group
 - Significant improvement of PFS (median PFS: 4.1 vs 2.1 months, *P*=0.016, Figure 1D) and a trend of longer OS (median OS: 11.4 vs 8.5 months, *P*=0.184, Figure 1F) was observed in the MHC I–high group versus the MHC I–low group

Table 1: Baseline Characteristics and Clinical Outcomes

Characteristic		Overall (n=113)	GEP Evaluable (n=100)
Age, n (%)	<65	44 (38.9)	34 (34.0)
	≥65	69 (61.1)	66 (66.0)
Sex, n (%)	Male	84 (74.3)	75 (75.0)
	Female	29 (25.7)	25 (25.0)
Known metastasis, n (%)	Lymph node only	25 (22.3)	23 (23.0)
	Liver metastasis	33 (29.5)	29 (29.0)
	Visceral metastasis	87 (77.7)	77 (77.0)
Number of previous regimens, n (%)	1	69 (61.1)	61 (61.0)
	2	37 (32.7)	32 (32.0)
	≥3	7 (6.2)	7 (7.0)
Response, n (%)	CR	10 (8.8)	10 (10.0)
	PR	15 (13.2)	13 (13.0)
	SD	15 (13.2)	12 (12.0)
	PD	51 (45.1)	47 (47.0)
	Non-CR/Non-PD	4 (3.5)	3 (3.0)
	NE	18 (15.9)	15 (15.0)
ORR, % (95% CI)[a]		22 (14.9, 30.9)	23 (15.2, 32.5)
Median PFS, months (95% CI)		2.2 (2.1, 3.9)	2.1 (2.1, 4.0)
Median OS, months (95% CI)		9.8 (7.5, 12.5)	9.8 (7.5, 12.2)

[a]Objective response rate was the proportion of patients with confirmed complete/partial responses using RECIST v1.1 in the overall /GEP-evaluable population.
Abbreviations: CI, confidence interval; CR, complete response; GEP, gene expression profile; NE, not estimable; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; SD, stable disease.

Figure 1: DEG Analysis by Clinical Response and Kaplan-Meier Curves for PFS and OS by T-cell and MHC I Signatures

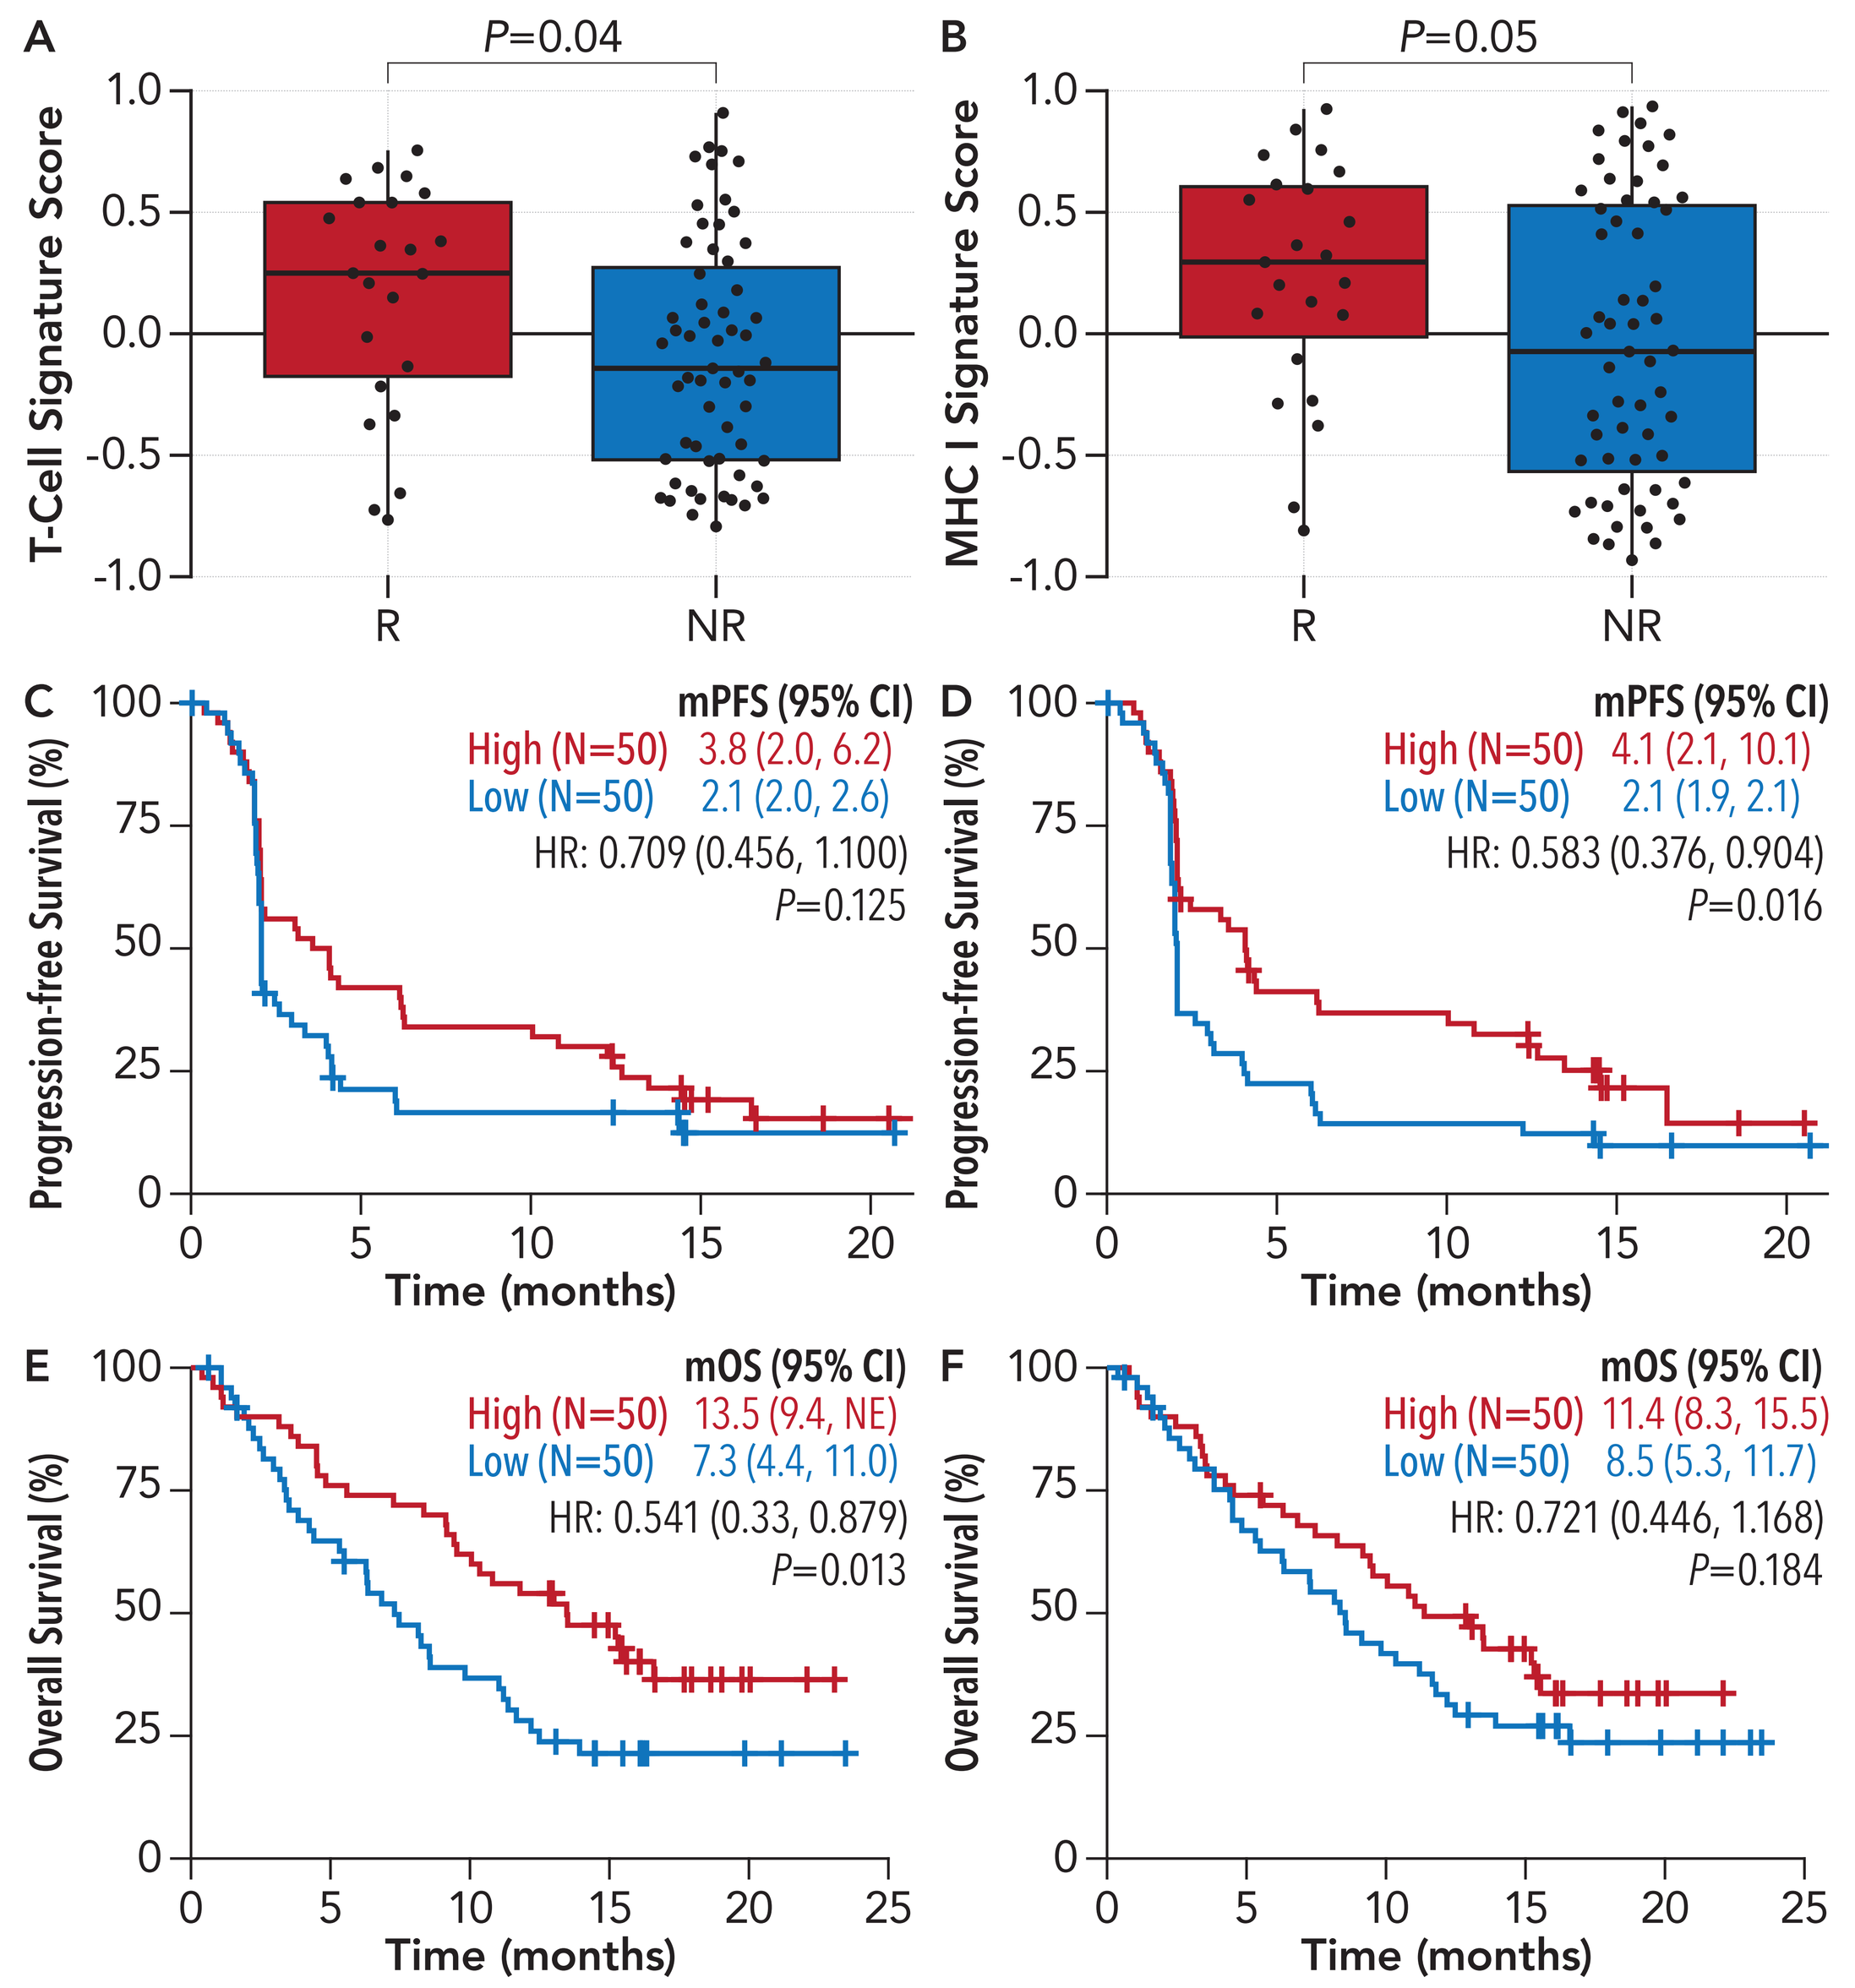
A
P=0.04
T-Cell Signature Score
R
NR
B
P=0.05
MHC I Signature Score
C
mPFS (95% CI)
High (N=50) 3.8 (2.0, 6.2)
Low (N=50) 2.1 (2.0, 2.6)
HR: 0.709 (0.456, 1.100)
P=0.125
Progression-free Survival (%)
Time (months)
D
mPFS (95% CI)
High (N=50) 4.1 (2.1, 10.1)
Low (N=50) 2.1 (1.9, 2.1)
HR: 0.583 (0.376, 0.904)
P=0.016
E
mOS (95% CI)
High (N=50) 13.5 (9.4, NE)
Low (N=50) 7.3 (4.4, 11.0)
HR: 0.541 (0.33, 0.879)
P=0.013
Overall Survival (%)
F
mOS (95% CI)
High (N=50) 11.4 (8.3, 15.5)
Low (N=50) 8.5 (5.3, 11.7)
HR: 0.721 (0.446, 1.168)
P=0.184

Abbreviations: CI, confidence interval; HR, hazard ratio; MHC I, major histocompatibility complex class I; mOS, median overall survival; mPFS, median progression-free survival; NE, not estimable; NR, non-responder; OS, overall survival; PFS, progression-free survival; R, responder.

Double-High Signature Score Subgroup Showed the Highest ORR and the Longest Survival Benefit

- Correlation of T-cell and MHC I signatures were low (Figure 2A)
- Using the median as a cutoff, three subgroups were identified by combining both signatures: double high (30/100), single high (40/100), and double low (30/100)
- The double-high subgroup (T-cell–high and MHC I–high groups; ORR=40%) had higher ORR compared with either the single-high (ORR=20%, *P*=0.03) or double-low (ORR=10%, *P*=0.01) subgroups (Figure 2B)
- Significantly longer PFS (*P*=0.04) and OS (*P*=0.05) were observed for patients with double-high signature score (Figure 2C and 2D)

Figure 2: DEG Analysis by Clinical Response and Kaplan-Meier Curves for PFS and OS by T-cell Signature and MHC I Signature

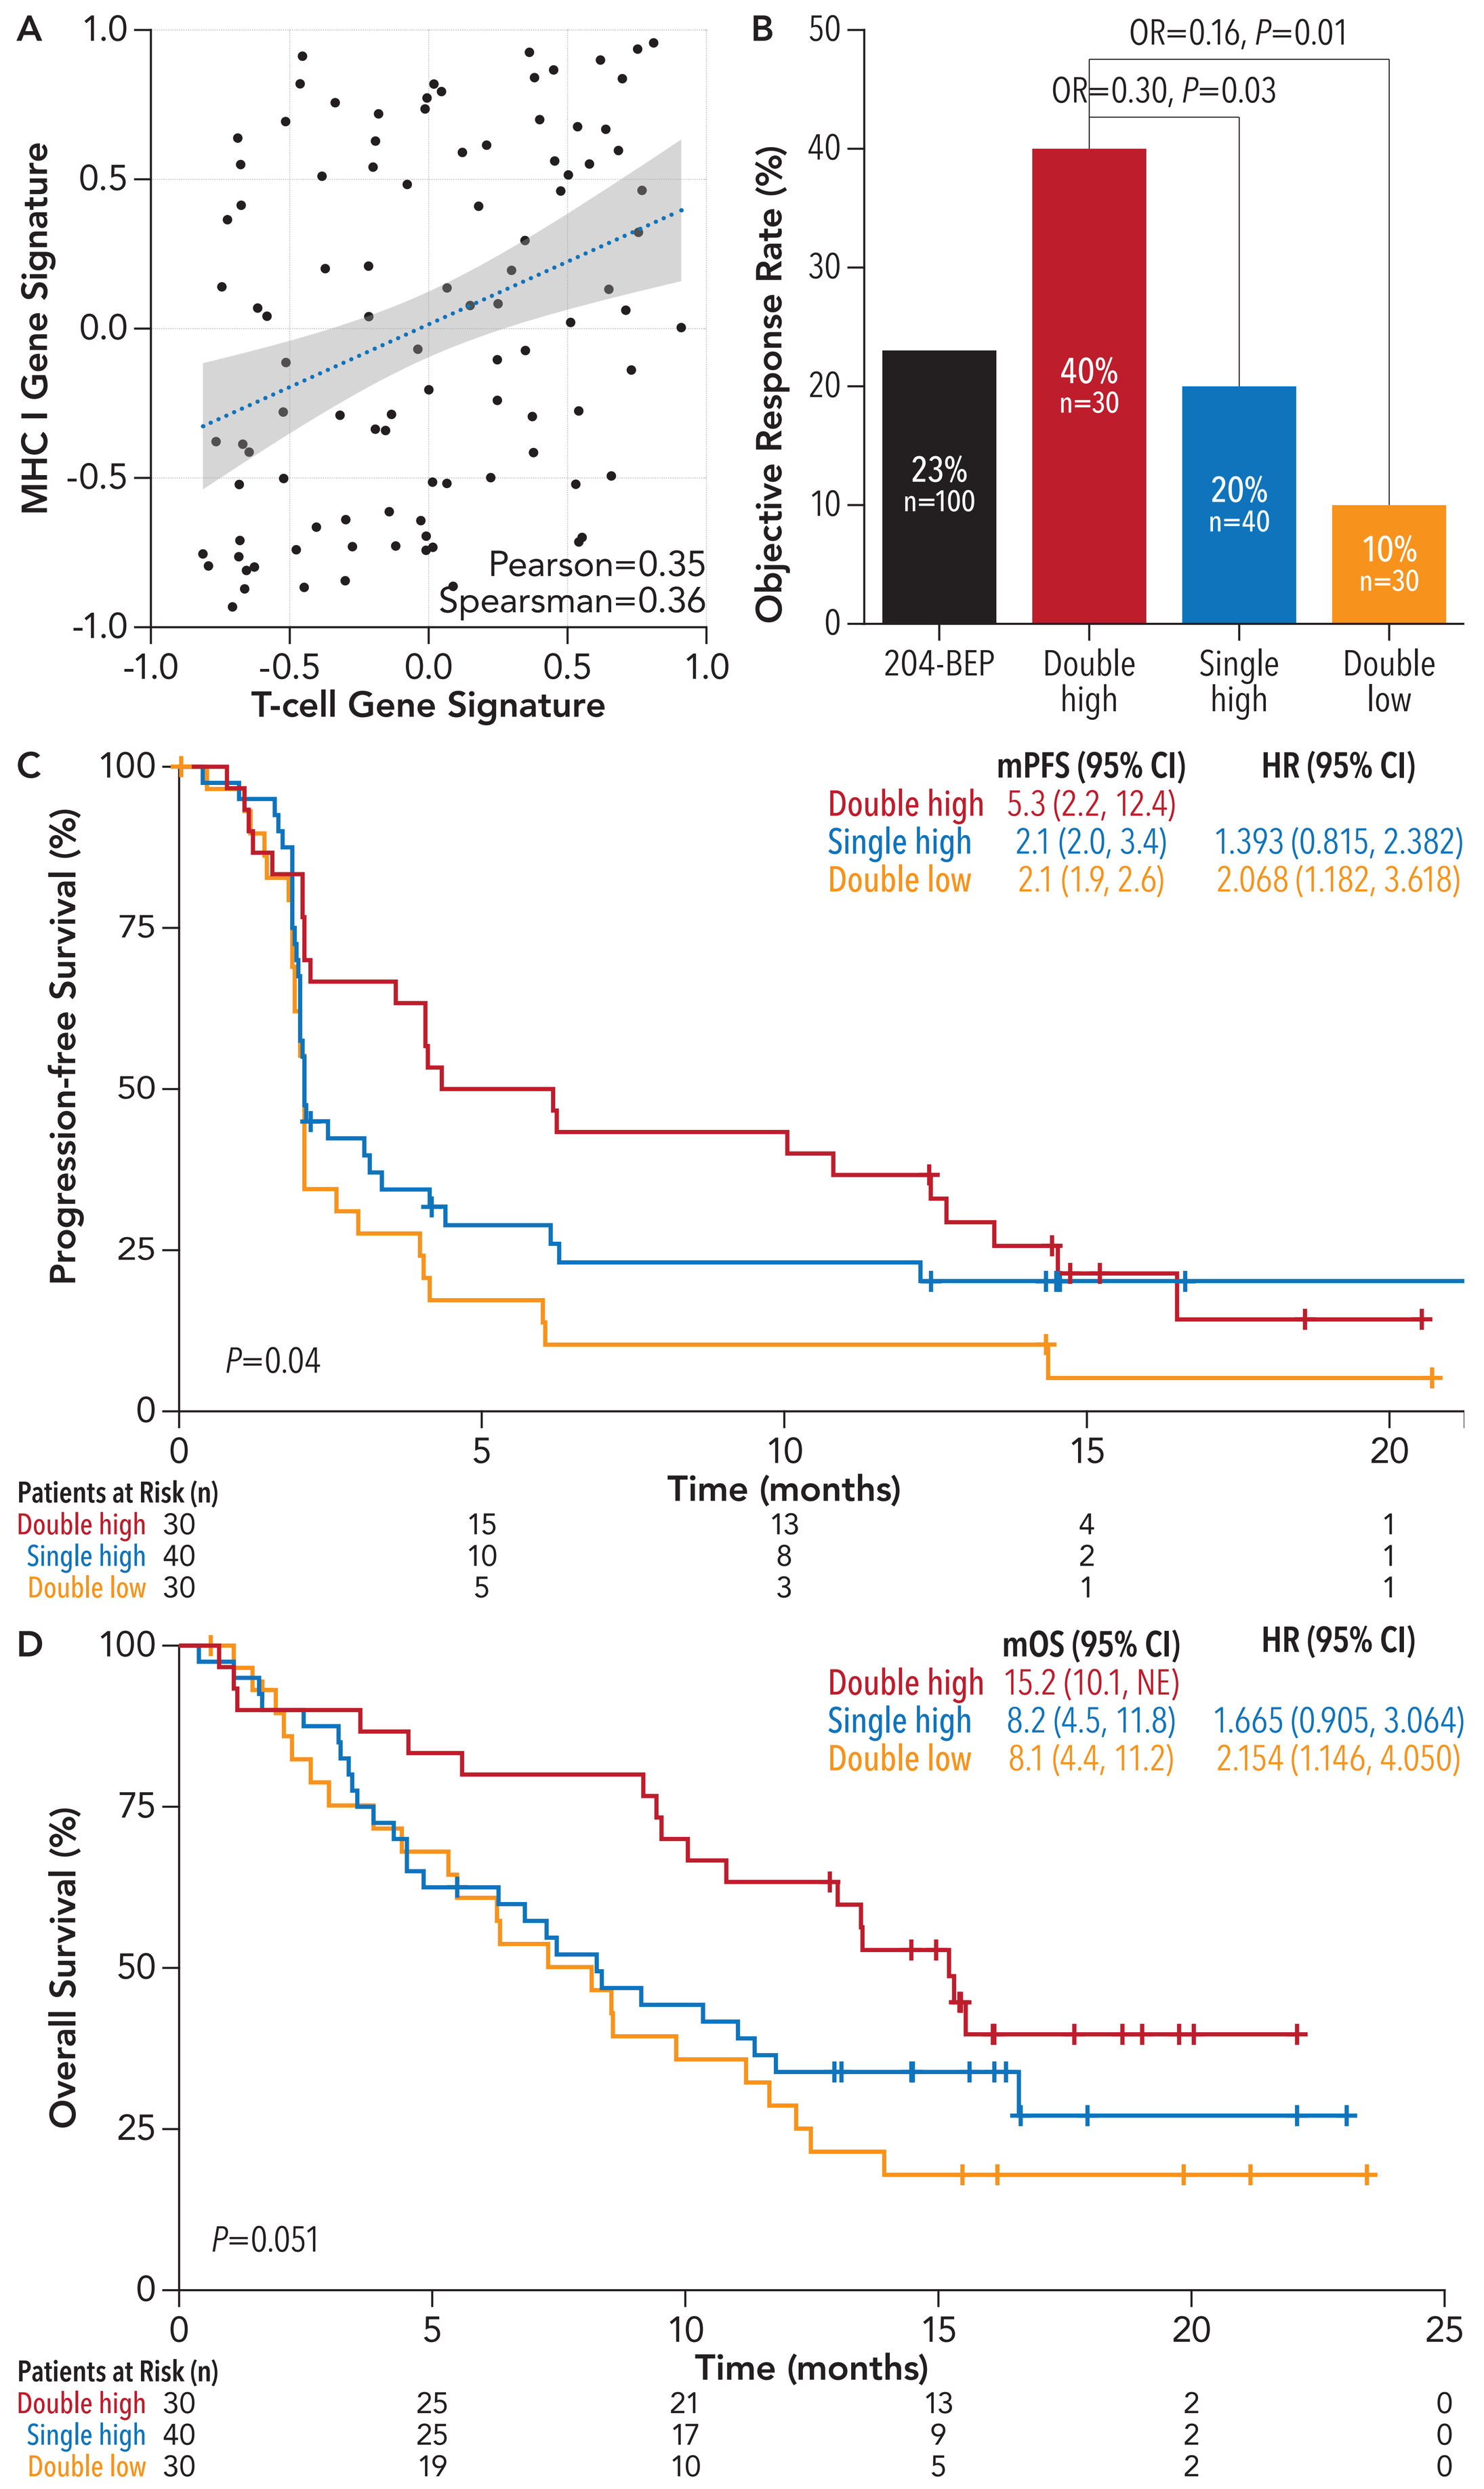
A
MHC I Gene Signature
T-cell Gene Signature
Pearson=0.35
Spearsman=0.36
B
OR=0.16, P=0.01
OR=0.30, P=0.03
Objective Response Rate (%)
23% n=100
40% n=30
20% n=40
10% n=30
204-BEP
Double high
Single high
Double low
C
mPFS (95% CI) HR (95% CI)
Double high 5.3 (2.2, 12.4)
Single high 2.1 (2.0, 3.4) 1.393 (0.815, 2.382)
Double low 2.1 (1.9, 2.6) 2.068 (1.182, 3.618)
Progression-free Survival (%)
P=0.04
Time (months)
Patients at Risk (n)
Double high 30 15 13 4 1
Single high 40 10 8 2 1
Double low 30 5 3 1 1
D
mOS (95% CI) HR (95% CI)
Double high 15.2 (10.1, NE)
Single high 8.2 (4.5, 11.8) 1.665 (0.905, 3.064)
Double low 8.1 (4.4, 11.2) 2.154 (1.146, 4.050)
Overall Survival (%)
P=0.051
Time (months)
Patients at Risk (n)
Double high 30 25 21 13 2 0
Single high 40 25 17 9 2 0
Double low 30 19 10 5 2 0

In panel B, odds ratios compared the ORR between subgroups. In panel C and D, hazard ratios tested comparisons between subgroups for PFS (C) and OS (D) by utilizing the double high group as the reference.
Abbreviations: BEP, biomarker evaluable population; CI, confidence interval; HR, hazard ratio; MHC I, major histocompatibility complex class I; mOS, median overall survival; mPFS, median progression-free survival; NE, not estimable; OR, odds ratio; ORR, objective response rate; OS, overall survival.

Differential Signature and Single Gene Expression Between Responders and Non-responders

- Non-responders were clustered into three distinct subgroups (NR1, NR2, and NR3) according to immune- and tumor-related gene signatures (Table 2)
- With a comparable tumor inflammation signature (TIS) to responders, NR1 had significantly higher expression of T-cell co-inhibition signatures (Figure 3A, *P*=0.04)
- NR2 was enriched for TGFβ signaling (Figure 3C, *P*=0.004)
- NR3 exhibited non-inflamed features (decreased TIS [Supplemental Figure 1, *P*=0.01]), but was enriched for cell cycle signatures (Figure 3E, *P*=0.0003)
- Key genes enriched in each non-responder group are shown in Figure 3B, 3D, and 3F

Figure 3: Gene Signatures and Single Gene Expression In Responders and Non-responders

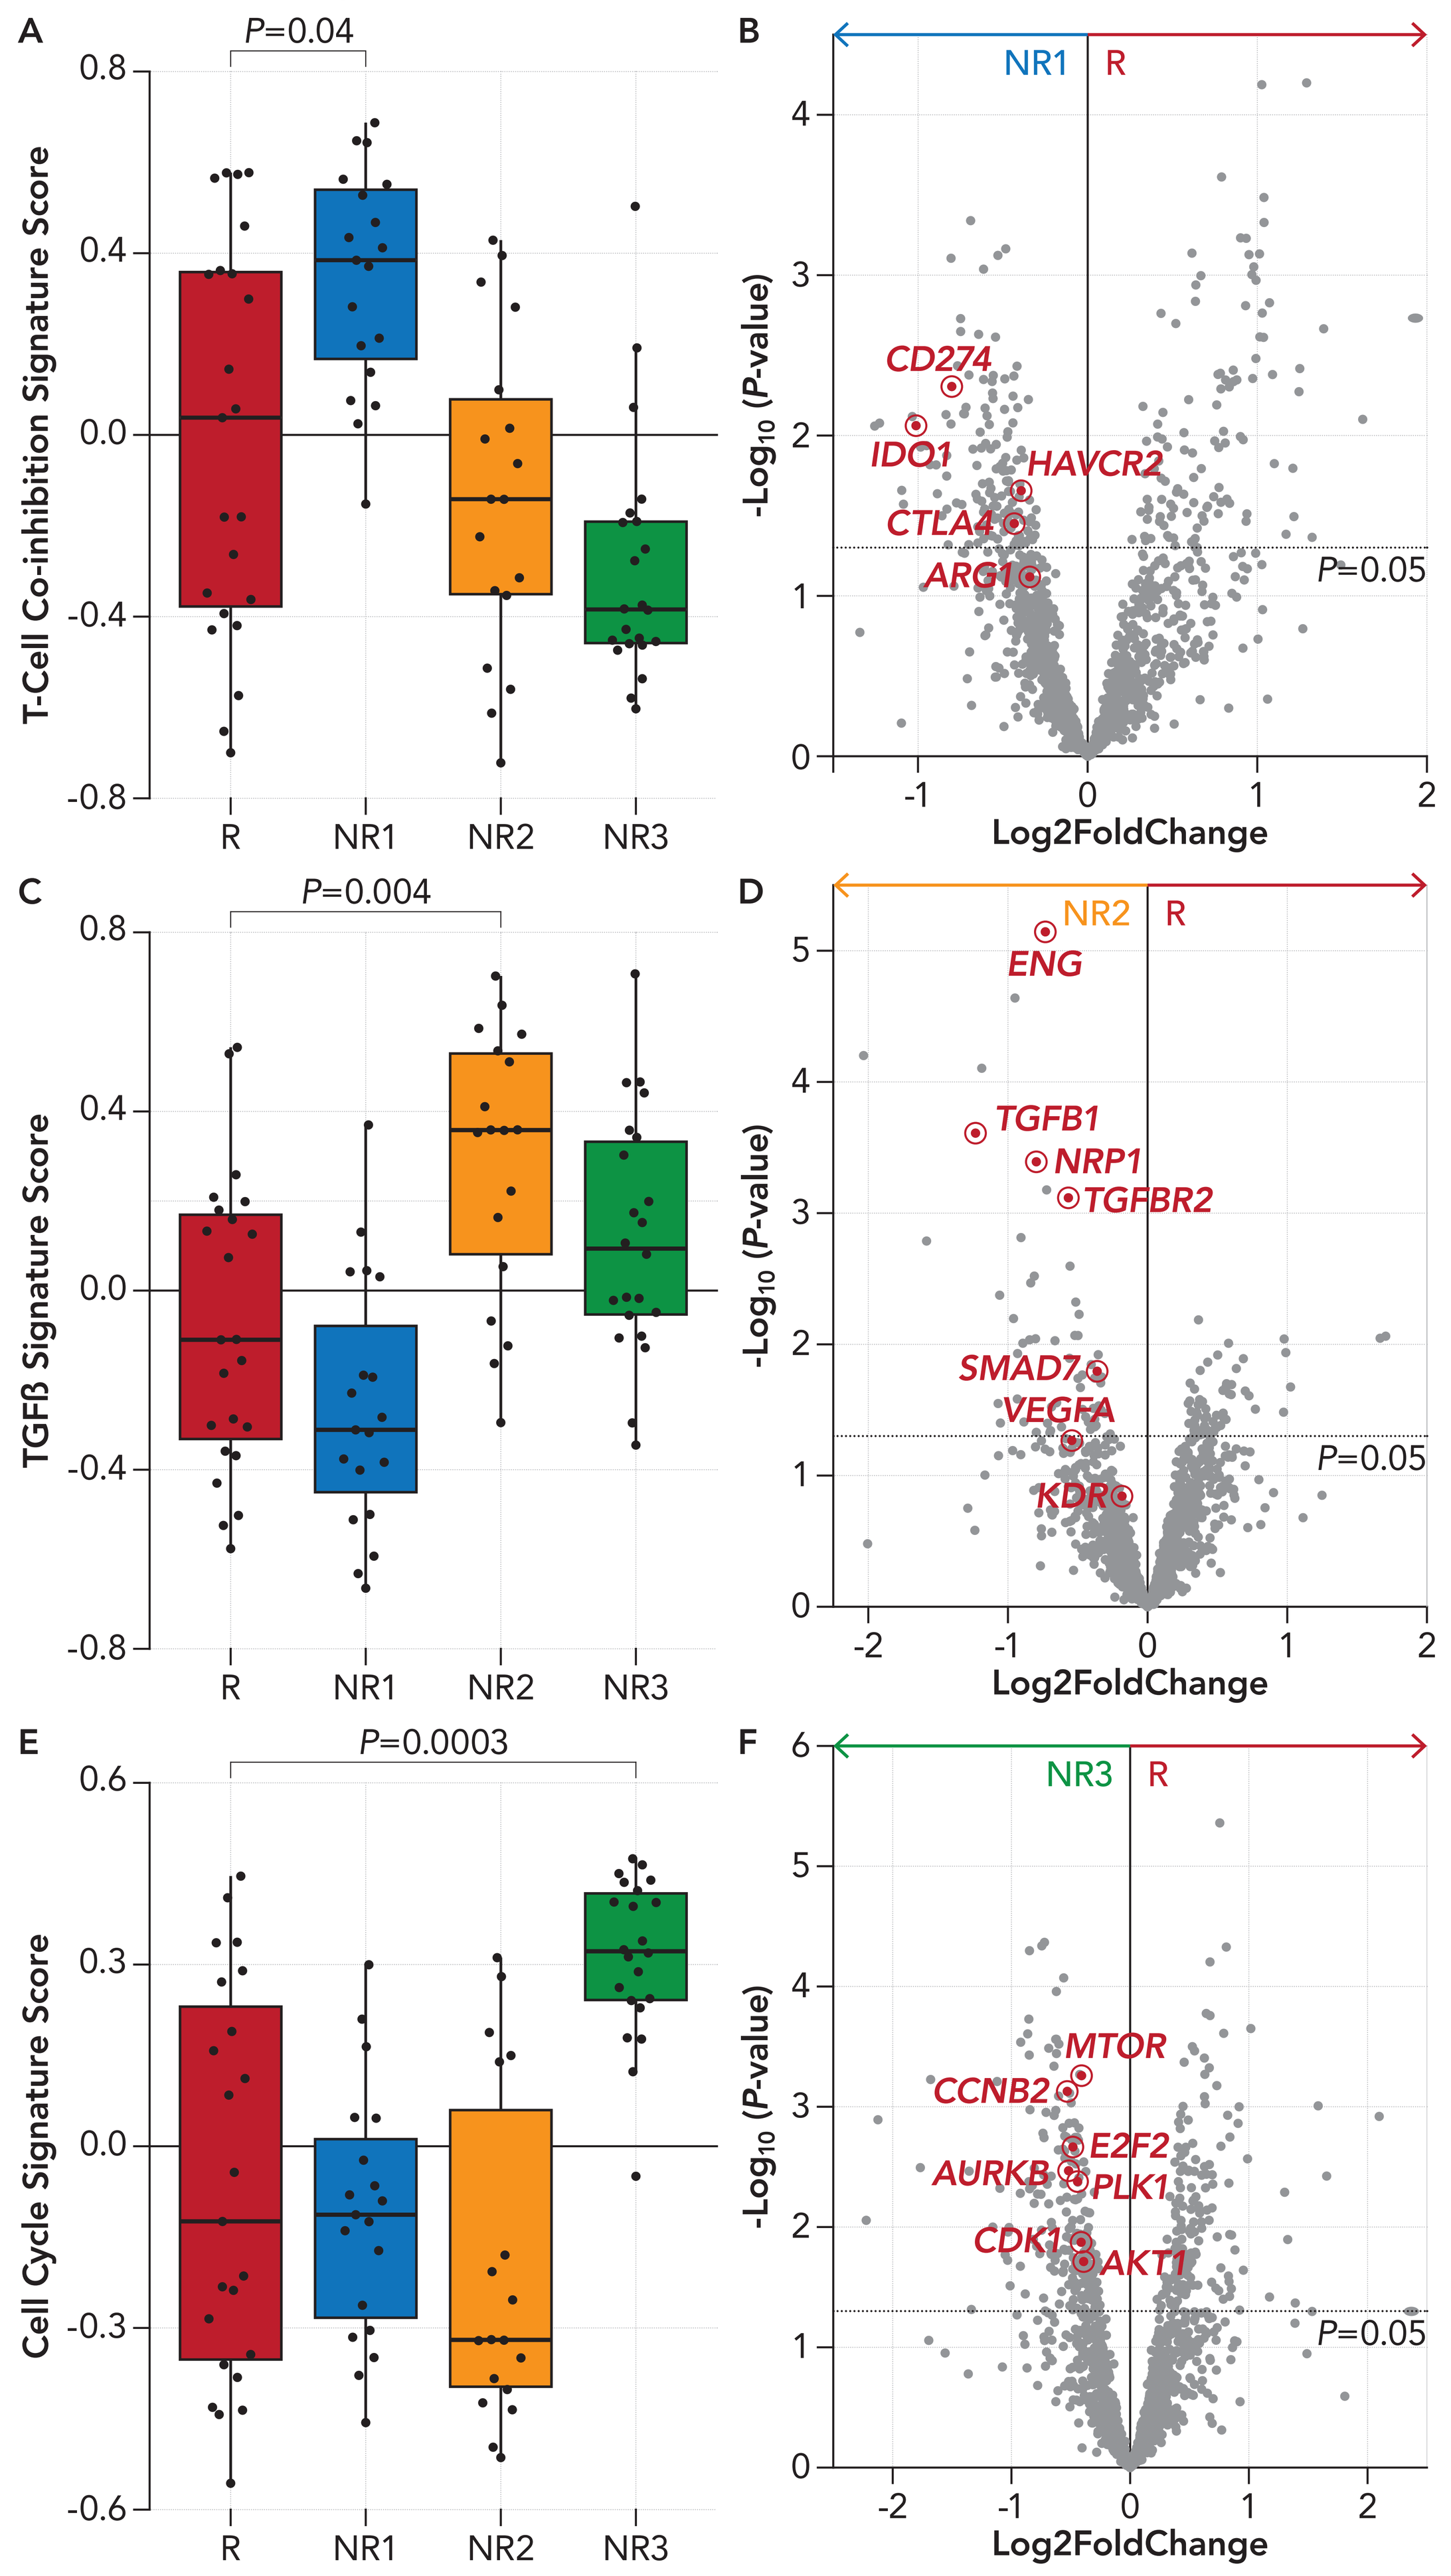
A
P=0.04
T-Cell Co-inhibition Signature Score
R NR1 NR2 NR3
B
NR1 R
CD274
IDO1
HAVCR2
CTLA4
ARG1
P=0.05
-Log10 (P-value)
Log2FoldChange
C
P=0.004
TGFβ Signature Score
D
NR2 R
ENG
TGFB1
NRP1
TGFBR2
SMAD7
VEGFA
KDR
E
P=0.0003
Cell Cycle Signature Score
F
NR3 R
MTOR
CCNB2
E2F2
AURKB
PLK1
CDK1
AKT1

Abbreviations: NR, nonresponder; R, responder; TIS, tumor inflammation signature.

CONCLUSIONS

- T-cell and MHC I signatures were associated with better clinical response and longer PFS and OS after tislelizumab treatment
- Combining high T-cell signatures with high MHC I signatures identified a subgroup with improved clinical benefit (40% ORR, 5.26-month median PFS and 15.2-month median OS)
- Distinct gene expression profiles were negatively correlated with clinical benefit, indicating the presence of a potential resistance signal:
 - Elevated immune checkpoint and suppressive immune cell signatures in inflamed non-responders
 - High expression of TGFβ signaling and angiogenesis-related genes
 - Low tumor inflammation signature with enriched cell cycle signature
- The association between gene expression biomarkers and clinical benefit could enhance our understanding of the process of adaptive antitumor immunity and also indicate a potential novel combination treatment strategy
- Due to the limitations associated with a single-arm study, the potential predictive role of the gene signatures discussed above warrant further exploration in an ongoing phase 3 study (RATIONALE 310; BGB-A317-310; NCT03967977)

Non-responders Could Be Clustered Into Three Distinct Subgroups According to the Immune- and Tumor-related Gene Signatures

Table 2: Immune and Tumor Gene Signatures Utilized for Non-responder Subgroups Clustering

Antitumor Activity	Immune Cells		Feature of Tumor
TIS	Macrophage	CD8 T cell	Endothelial cells
Cytotoxic activity	M1 Macrophage	T cell	Focal adhesion
IFNγ	M2 Macrophage	DC	EMT
MHC class I	Mast cell	NK	Angiogenesis
Checkpoint-T cell	Neutrophils	MDSC	TGFβ signaling
Checkpoint-APC		B cell	Cell cycle

Abbreviations: APC, antigen presentation cell; DC, dendritic cell; EMT, epithelial-mesenchymal transition, MDSC, myeloid-derived suppressor cell; MCH, major histocompatibility complex; NK, natural killer cell; TIS, tumor inflammation signature.

REFERENCES

1. National Cancer Institute. SEER Stat Fact Sheets: Bladder Cancer. Available at: http://seer.cancer.gov/statfacts/html/urinb.html. Accessed October 18, 2019.
2. Pang C, Guan Y, Li H, et al. Urologic cancer in China. *Jpn J Clin Oncol.* 2016;46(6):497-501.
3. Stenehjem DD, Tran D, Nkrumah MA, et al. PD1/PDL1 inhibitors for the treatment of advanced urothelial bladder cancer. *Onco Targets Ther.* 2018;11:5973-5989.
4. Zhang T, Song X, Xu L, et al. The binding of an anti-PD-1 antibody to FcγRI has a profound impact on its biological functions. *Cancer Immunol Immunother.* 2018;67(7):1079-1090.
5. Ye D, Liu J, Zhou A, et al. First report of efficacy and safety from a phase 2 trial of tislelizumab, an anti-PD-1 antibody, for the treatment of PD-L1+ locally advanced or metastatic urothelial carcinoma (UC) in Asian patients. *Ann Oncol.* 2019;30(suppl 5):v356-v402.
6. Danaher P, Warren S, Dennis L, et al. Gene expression markers of tumor infiltrating leukocytes. *J Immunother Cancer.* 2017;5:18.
7. Rooney MS, Shukla SA, Wu CJ, et al. Molecular and genetic properties of tumors associated with local immune cytolytic activity. *Cell.* 2015;160(1-2):48-61.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative centers' study staff and study patients, and to recognize those from BeiGene, Ltd. who have substantially contributed to the development of this presentation. This study was sponsored by BeiGene, Ltd. Editorial assistance was provided by Stephan Lindsey, PhD, and Elizabeth Hermans, PhD (OPEN Health Medical Communications, Chicago, IL), and funded by the study sponsor.

Copies of this poster and the supplemental appendix obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com